Phase 1b/2 Trial of BGB-A445 (OX40 Agonist) With or Without Tislelizumab in Urothelial Carcinoma, Renal Cell Carcinoma, and Melanoma

1980P

Bin Lian,[1] Qingyun Zhang,[2] Shuang Zhang,[3] Xiubao Ren,[4] Yu Chen,[5] Jiasheng Bian,[6] Jiayuan Zhang,[7] Rui Liu,[8] Fan Bai,[7] Chuanliang Cui,[1] Jun Guo[1]

[1]Department of Melanoma and Sarcoma, Peking University Cancer Hospital & Institute, Beijing, China; [2]Cancer Hospital of Guangxi Medical University, Guangxi, China; [3]West China Hospital, Sichuan, China; [4]Tianjin Medical University Cancer Institute and Hospital, Tianjin, China; [5]Fujian Cancer Hospital, Fuzhou, China; [6]Shandong Cancer Hospital and Institute, Jinan, China; [7]BeOne Medicines, Ltd, Shanghai, China; [8]BeOne Medicines, Ltd, Beijing, China

CONCLUSIONS

- BGB-A445 was generally well tolerated as monotherapy and in combination with tislelizumab
- Although antitumor activity was limited with BGB-A445 monotherapy, antitumor activity was observed with BGB-A445 plus tislelizumab combination therapy, including among patients who switched from BGB-A445 monotherapy
- Immune cell biomarker findings indicated the potential functional activity of BGB-A445, consistent with its mechanism of action

INTRODUCTION

- OX40 is a costimulatory receptor expressed on intratumoral T cells that has been shown to enhance antigen-stimulated T-cell responses and prevent tumor-induced immune tolerance in preclinical models[1,2]
- In mouse tumor models, OX40 agonism depleted intratumoral Tregs, which correlated with tumor regression[3]
- BGB-A445 is a humanized immunoglobulin G (IgG) 1 monoclonal antibody agonist of OX40
- Here, we report the final results of the phase 1b/2 trial (NCT05661955) evaluating BGB-A445 alone and in combination with tislelizumab in advanced urothelial carcinoma (UC), renal cell carcinoma (RCC), and melanoma

METHODS

- This open-label, multicenter, parallel-assignment trial enrolled patients with UC (cohorts A, B, G), RCC (cohorts C, D), or melanoma (cohorts E, F, H, I) (**Figure 1**)
- Patients received intravenous BGB-A445 alone or BGB-A445 plus tislelizumab on day 1 of each 21-day cycle
- Treatment continued until disease progression, intolerable toxicity, or completion of 2 years of study treatment
- Patients assigned to BGB-A445 monotherapy who experienced disease progression were given the option to switch to a combination of BGB-A445 plus tislelizumab
- Safety/tolerability profile was assessed among patients assigned to their initial cohort (Period 1) and separately among patients who switched from monotherapy to combination therapy (Period 2)
 - Treatment-emergent adverse events (TEAEs) were classified by MedDRA v27.0 and graded per CTCAE v5.0
- Antitumor activity was assessed per RECIST v1.1 and reported in terms of confirmed responses
- Multiplex immunohistochemistry using seven cell markers (CD3, CD4, CD8, Foxp3, OX40, Ki67, PanCK) was performed to assess the pharmacodynamic effect of BGB-A445 monotherapy, with paired baseline and cycle 3 day 1 tumor samples available from six patients in cohort E

Figure 1. Study Design

KEY ELIGIBILITY

Adults with UC, RCC, or melanoma

- UC
 - ECOG PS ≤2
 - Cohort A: 1-3 prior LoT
 - Cohort B: 1-3 prior LoT, CPI naïve
 - Cohort G: No prior LoT, cisplatin-ineligible
- RCC
 - ECOG PS ≤1
 - Cohort C: 1-3 prior LoT
 - Cohort D: 1-3 prior LoT, CPI naïve
- Melanoma
 - ECOG PS ≤1
 - Cohort E: 1-3 prior LoT
 - Cohort F: 1-3 prior LoT, CPI naïve
 - Cohort H: Non-mucosal, no prior LoT
 - Cohort I: Non-mucosal, 1-2 prior LoT including CPI

Phase 1b/2 (NCT05661955)

Cohorts A, C, E
BGB-A445
IV Q3W

Cohorts B, G, D, F, H, I
BGB-A445+TIS 200 mg
IV Q3W

ENDPOINTS

Primary
- ORR by investigator per RECIST v1.1[a]

Secondary
- DCR and CBR by investigator per RECIST v1.1[a]
- Safety/tolerability[b]

Exploratory
- Pharmacodynamic biomarker analysis

[a]Efficacy analyses included all patients who received ≥1 dose of study drug(s), had evaluable disease at baseline, and had ≥1 evaluable postbaseline tumor response assessment unless clinical progressive disease or death occurred before the first planned postbaseline tumor assessment. ORR is the proportion of patients who achieve a CR or PR. Disease control rate is the percentage of patients with CR, PR, or SD. Clinical benefit rate is the proportion of patients who achieved CR, PR, or ≥24 weeks of SD [b]Safety analyses included all patients who received ≥1 dose of study drug(s).
Abbreviations: CBR, clinical benefit rate; CPI, checkpoint inhibitor; CR, complete response; DCR, disease control rate; ECOG PS, Eastern Cooperative Oncology Group performance status; IV, intravenous; LoT, lines of therapy; ORR, objective response rate; PR, partial response; Q3W, every 3 weeks; RCC, renal cell carcinoma; SD, stable disease; TIS, tislelizumab; UC, urothelial carcinoma.

RESULTS

Overall Study Population

- At the end of the trial (May 27, 2025), 113 patients were enrolled across cohorts A through H (**Table 1**)
 - Median follow-up was 9.92 (range, 3.76-15.92) months
 - Sixty patients were assigned to monotherapy, and 53 patients were assigned to combination therapy
- Baseline characteristics were generally comparable across tumor types
- No patients were enrolled in cohort I because the trial was stopped by the sponsor
 - The decision to terminate the study was not related to any safety concerns with BGB-A445 or tislelizumab
- At study completion, all patients were discontinued from treatment; progressive disease (PD) was the most common (68%) reason for discontinuation

Table 1. Baseline Characteristics and Patient Disposition

	UC			RCC		Melanoma			
	Mono	Combo		Mono	Combo	Mono	Combo		
Prior LoT	1-3	1-3	0	1-3	1-3	1-3	1-3	0	
Cohort	A (n=20)	B (n=18)	G (n=1)	C (n=20)	D (n=15)	E (n=20)	F (n=7)	H (n=12)	Total (N=113)
Patients treated, n (%)	20 (100.0)	18 (100.0)	1 (100.0)	20 (100.0)	15 (100.0)	20 (100.0)	7 (100.0)	12 (100.0)	113 (100.0)
Median age, yrs (range)	62.5 (45-71)	63.5 (41-79)	67.0 (67-67)	58.0 (44-75)	59.0 (43-79)	61.0 (40-75)	58.0 (38-71)	63.0 (47-76)	61.0 (38-79)
Age ≥65, n (%)	9 (45.0)	7 (38.9)	1 (100.0)	5 (25.0)	2 (13.3)	5 (25.0)	1 (14.3)	4 (33.3)	34 (30.1)
Male, n (%)	15 (75.0)	14 (77.8)	1 (100.0)	13 (65.0)	11 (73.3)	11 (55.0)	5 (71.4)	9 (75.0)	79 (69.9)
ECOG PS, n (%)									
0	7 (35.0)	1 (5.6)	0	2 (10.0)	4 (26.7)	14 (70.0)	3 (42.9)	6 (50.0)	37 (32.7)
1	13 (65.0)	15 (83.3)	1 (100.0)	18 (90.0)	11 (73.3)	6 (30.0)	4 (57.1)	6 (50.0)	74 (65.5)
2	0	2 (11.1)	0	0	0	0	0	0	2 (1.8)
Discontinued, n (%)	20 (100.0)	18 (100.0)	1 (100.0)	20 (100.0)	15 (100.0)	20 (100.0)	7 (100.0)	12 (100.0)	113 (100.0)
Progressive disease	14 (70.0)	9 (50.0)	0 (0.0)	13 (65.0)	10 (66.7)	16 (80.0)	6 (85.7)	9 (75.0)	77 (68.1)
Withdrawal	1 (5.0)	1 (5.6)	0	4 (20.0)	0	2 (10.0)	0	2 (16.7)	10 (8.8)
Study terminated	0	4 (22.2)	1 (100.0)	0	2 (13.3)	0	0	1 (8.3)	8 (7.1)
Adverse event	2 (10.0)	2 (11.1)	0	1 (5.0)	1 (6.7)	0	0	0	6 (5.3)
Physician decision	1 (5.0)	1 (5.6)	0	1 (5.0)	1 (6.7)	0	1 (14.3)	0	5 (4.4)
Treatment completion	0	0	0	0	0	2 (10.0)	0	0	2 (1.8)
Other	2 (10.0)	1 (5.6)	0	1 (5.0)	1 (6.7)	0	0	0	5 (4.4)

Abbreviations: combo, combination therapy with BGB-A445 + tislelizumab; ECOG PS, Eastern Cooperative Oncology Group performance status; LoT, lines of therapy; mono, monotherapy with BGB-A445; RCC, renal cell carcinoma; UC, urothelial carcinoma.

- In patients with the same tumor type, exposure to BGB-A445 in the combination therapy cohorts (B, G, D, F, H, I) was generally higher than in the monotherapy cohorts (A, C, E)
- Median relative dose intensity was >98% for both BGB-A445 and tislelizumab in all cohorts, indicating high levels of patient adherence

Antitumor Activity

- Although no patient achieved a partial response (PR) or complete response (CR) in the monotherapy cohorts (A, C, E), responses were observed in the combination therapy cohorts (**Table 2**)
 - In the combination therapy cohorts where only checkpoint inhibitor–naive patients were enrolled, overall response rate was 29.4%, 20.0%, and 16.7% (B, D, F, respectively); among these patients, one CR (5.9%) was reported in cohort B
 - Among treatment-naïve patients in cohorts G and H, PR was observed in each cohort

Table 2. Investigator-confirmed Antitumor Activity of BGB-A445 With or Without Tislelizumab

	UC			RCC		Melanoma		
	Mono	Combo		Mono	Combo	Mono	Combo	
Prior LoT	1-3	1-3	0	1-3	1-3	1-3	1-3	0
Cohort	A (n=18)	B (n=17)	G (n=1)	C (n=19)	D (n=15)	E (n=19)	F (n=6)	H (n=10)
ORR, n (%)[a]	0	5 (29.4)	1 (100.0)	0	3 (20.0)	0	1 (16.7)	1 (10.0)
CR	0	1 (5.9)	0	0	0	0	0	0
PR	0	4 (23.5)	1 (100.0)	0	3 (20.0)	0	1 (16.7)	1 (10.0)
SD	6 (33.3)	7 (41.2)	0	7 (36.8)	5 (33.3)	5 (26.3)	3 (50.0)	1 (10.0)
PD	9 (50.0)	5 (29.4)	0 (0.0)	12 (63.2)	7 (46.7)	14 (73.7)	2 (33.3)	8 (80.0)
DCR, n (%)	6 (33.3)	12 (70.6)	1 (100)	7 (36.8)	8 (53.3)	5 (26.3)	4 (66.7)	2 (20.0)
CBR, n (%)	0	7 (41.2)	1 (100)	3 (15.8)	4 (26.7)	1 (5.3)	2 (33.3)	1 (10.0)

[a]Response data represent results before patients switched therapy.
Abbreviations: CBR, clinical benefit rate; combo, combination therapy with BGB-A445 + tislelizumab; CR, complete response; DCR, disease control rate; LoT, lines of therapy; mono, monotherapy with BGB-A445; ORR, objective response rate; PD, progressive disease; PR, partial response; RCC, renal cell carcinoma; SD, stable disease; UC, urothelial carcinoma.

- Among 15 patients with melanoma who switched from BGB-A445 monotherapy to BGB-A445 + tislelizumab after PD, four had an unconfirmed PR, including two with durable responses lasting 15.1 and 20.8 months and maximum tumor shrinkage of 53.5% and 86.5%, respectively

Safety and Tolerability

- Period 1 safety assessment included 113 patients in the combination therapy cohorts (cohorts B, D, F, G, H) and those in monotherapy cohorts (cohorts A, C, E) who did not switch to combination therapy or prior to their switching to combination therapy (**Table 3**)
- Most (90.3%) patients reported a TEAE, with a similar proportion across tumor types and treatment type
- More than half of the patients (67.3%) reported an AE considered related to treatment by the investigator (TRAE); however, most were mild to moderate in severity
 - Any-grade BGB-A445-related AEs were reported in 76 (67.3%) patients, and any-grade tislelizumab-related AEs were reported in 45 (39.8%) patients (**Table 3**)
 - Grade ≥3 TRAEs were reported in 23 (20%) patients; anemia (3.5%) was the most commonly reported grade ≥3 TRAE (**Table 4**)
- Three TRAEs led to treatment discontinuation in cohorts A (n=1), B (n=1), and D (n=1)
- The incidence of grade ≥3 immune-mediated AEs was low and similar across tumor and treatment types (**Table 3** and **Table 4**)
- One grade 5 TRAE of cerebral infarction occurred in a patient in cohort A with UC and extensive metastatic disease to the kidney, lung, and lymph nodes

Table 3. AEs Across All Cohorts in Period 1

	UC			RCC		Melanoma			
	Mono	Combo		Mono	Combo	Mono	Combo		
Prior LoT	1-3	1-3	0	1-3	1-3	1-3	1-3	0	
Cohort	A (n=20)	B (n=18)	G (n=1)	C (n=20)	D (n=15)	E (n=20)	F (n=7)	H (n=12)	Total (N=113)
Any TEAE, n (%)	16 (80.0)	17 (94.4)	1 (100.0)	18 (90.0)	14 (93.3)	18 (90.0)	7 (100.0)	11 (91.7)	102 (90.3)
BGB-A445-related[a]	10 (50.0)	16 (88.9)	1 (100.0)	10 (50.0)	13 (86.7)	11 (55.0)	7 (100.0)	8 (66.7)	76 (67.3)
TIS-related[a]	0	16 (88.9)	1 (100.0)	0	13 (86.7)	0	7 (100.0)	8 (66.7)	45 (39.8)
Serious TRAEs, n (%)[a]	2 (10.0)	2 (11.1)	1 (100.0)	1 (5.0)	1 (6.7)	0	2 (28.6)	1 (8.3)	10 (8.8)
Grade ≥3 TRAEs, n (%)[a]	2 (10.0)	8 (44.4)	1 (100.0)	2 (10.0)	4 (26.7)	1 (5.0)	3 (42.9)	2 (16.7)	23 (20.4)
Any imAE, n (%)	3 (15.0)	5 (27.8)	1 (100.0)	2 (10.0)	7 (46.7)	1 (5.0)	3 (42.9)	1 (8.3)	23 (20.4)
Grade ≥3	1 (5.0)	2 (11.1)	1 (100.0)	0	1 (6.7)	0	0	1 (8.3)	6 (5.3)
Serious	0	2 (11.1)	1 (100.0)	0	0	0	0	1 (8.3)	4 (3.5)

[a]TRAEs include those events considered by the investigator to be related or with missing assessment of the causal relationship.
Abbreviations: AE, adverse event; combo, combination therapy with BGB-A445 + tislelizumab; imAE, immune-mediated adverse event; LoT, lines of therapy; mono, monotherapy with BGB-A445; RCC, renal cell carcinoma; TEAE, treatment-emergent adverse event; TIS, tislelizumab; TRAE, treatment-related adverse event; UC, urothelial carcinoma.

Table 4. Grade ≥3 TRAEs and All Grade imAEs Across All Cohorts in Period 1

	UC			RCC		Melanoma			
	Mono	Combo		Mono	Combo	Mono	Combo		
Prior LoT	1-3	1-3	0	1-3	1-3	1-3	1-3	0	
Cohort	A (n=20)	B (n=18)	G (n=1)	C (n=20)	D (n=15)	E (n=20)	F (n=7)	H (n=12)	Total (N=113)
Grade ≥3 TRAEs[a]									
Anemia	0	2 (11.1)	0	0	2 (13.3)	0	0	0	4 (3.5)
ALT increased	0	1 (5.6)	0	0	0	0	2 (28.6)	0	3 (2.7)
Lipase increased	0	1 (5.6)	0	0	1 (6.7)	0	1 (14.3)	0	3 (2.7)
Hypercalcemia	2 (10.0)	0	0	0	1 (6.7)	0	0	0	3 (2.7)
Amylase increased	0	0	0	0	1 (6.7)	0	1 (14.3)	0	2 (1.8)
AST increased	0	1 (5.6)	0	0	0	0	1 (14.3)	0	2 (1.8)
Neutrophil count decreased	0	0	0	2 (10.0)	0	0	0	0	2 (1.8)
Immune-mediated hepatitis	0	1 (5.6)	1 (100.0)	0	0	0	0	0	2 (1.8)
Any imAEs									
Rash	1 (5.0)	1 (5.6)	1 (100.0)	0	3 (20.0)	0	2 (28.6)	0	8 (7.1)
Hypothyroidism	1 (5.0)	2 (11.1)	0	0	3 (20.0)	0	0	0	6 (5.3)
Hyperthyroidism	0	0	0	0	2 (13.3)	0	0	0	2 (1.8)
Immune-mediated hepatitis	0	1 (5.6)	1 (100.0)	0	0	0	0	0	2 (1.8)

[a]TRAEs include those events considered by the investigator to be related or with missing assessment of the causal relationship.
Abbreviations: ALT, alanine aminotransferase; AST, aspartate aminotransferase; combo, combination therapy with BGB-A445 + tislelizumab; mono, monotherapy with BGB-A445; LoT, lines of therapy; RCC, renal cell carcinoma; TRAE, treatment-related adverse event; UC, urothelial carcinoma.

- Period 2 safety assessment included 37 patients in the monotherapy cohorts (cohorts A, C, E) whose AEs were assessed after switching to combination therapy (**Table 5**)
 - Most (n=32/37; 86.5%) patients reported at least one TEAE during Period 2
 - The safety profile in Period 2 was generally consistent with that observed in Period 1

Table 5. AEs in Monotherapy Cohorts (Cohorts A, C, E) Who Switched to Combination Treatment (Period 2)

	UC	RCC	Melanoma	
	Combination	Combination	Combination	
Prior LoT	1-3	1-3	1-3	
Cohort	A (n=7)	C (n=15)	E (n=15)	Total (N=37)
Any TEAEs, n (%)	4 (57.1)	14 (93.3)	14 (93.3)	32 (86.5)
BGB-A445-related[a]	2 (28.6)	7 (46.7)	9 (60.0)	18 (48.6)
Tislelizumab-related[a]	2 (28.6)	8 (53.3)	9 (60.0)	19 (51.4)
Serious TRAEs, n (%)[a]	1 (14.3)	2 (13.3)	0 (0.0)	3 (8.1)
Grade ≥3 TRAEs, n (%)[a]	3 (42.9)	4 (26.7)	1 (6.7)	8 (21.6)
Any imAE, n (%)	1 (14.3)	2 (13.3)	3 (20.0)	6 (16.2)
Grade ≥3	1 (14.3)	1 (6.7)	0	2 (5.4)
Serious	0	1 (6.7)	0	1 (2.7)

[a]TRAEs include those events considered by the investigator to be related or with missing assessment of the causal relationship.
Abbreviations: combination, combination therapy with BGB-A445 + tislelizumab; imAE, immune-mediated adverse event; LoT, lines of therapy; RCC, renal cell carcinoma; TEAE, treatment-emergent adverse event; TRAE, treatment-related adverse event; UC, urothelial carcinoma.

Pharmacodynamics

- Multiplex immunohistochemistry of paired baseline and posttreatment tumor samples from cohort E demonstrated an upward trend in $OX40^{+}CD4^{+}Foxp3^{-}$ effector T cells and a downward trend in $Ki67^{+}CD4^{+}Foxp3^{+}$ Treg cells compared with baseline

REFERENCES

1. Gough MJ, Ruby CE, Redmond WL, et al. OX40 agonist therapy enhances CD8 infiltration and decreases immune suppression in the tumor. *Cancer Res.* 2008;68(13):5206-5215.
2. Jensen SM, Maston LD, Gough MJ, et al. Signaling through OX40 enhances antitumor immunity. *Semin Oncol.* 2010;37(5):524-532.
3. Bulliard Y, Jolicoeur R, Zhang J, et al. OX40 engagement depletes intratumoral Tregs via activating FcγRs, leading to antitumor efficacy. *Immunol Cell Biol.* 2014;92(6):475-480.

DISCLOSURES

BL: no conflicts of interest relevant to this work.

ACKNOWLEDGMENTS

The authors thank the participants and their families, investigators, co-investigators, and the study teams at each of the participating centers. This study was sponsored by BeOne Medicines, Ltd. Medical writing and editorial assistance were provided by Michael Franklin, Jose Casasnovas Nieves, and Elizabeth Hermans, PhD, employees of BeOne Medicines, Ltd.

PRESENTER: Bin Lian
CORRESPONDENCE: Jun Guo, guoj307@126.com

Presented at the 2026 European Society for Medical Oncology Annual Meeting; October 23-27, 2026; Madrid, Spain