A PHASE 1B STUDY TO ASSESS SAFETY, TOLERABILITY, PHARMACOKINETICS, AND PRELIMINARY ANTITUMOR ACTIVITY OF SITRAVATINIB IN COMBINATION WITH TISLELIZUMAB IN PATIENTS WITH ADVANCED SOLID TUMORS

Jeffrey Goh,[1] Bo Gao,[2] Ben Markman,[3] Hui Gan,[4] Mark Voskoboynik,[5] Jermaine Coward,[1] Yi-Long Wu,[6] Jun Guo,[7] Qing Zhou,[6] Jun Zhao,[7] Cheng Chen,[8] Xiao Xiang,[9] Jingjun Qiu,[9] Yingying Xu,[9] Liu Yang,[8] Michael Millward[10]

[1]ICON Cancer Foundation, South Brisbane, and University of Queensland, St Lucia, QLD, Australia; [2]Blacktown Cancer and Haematology Centre, Blacktown, NSW, Australia; [3]Monash Health, Melbourne, VIC, Australia; [4]Austin Hospital, Heidelberg, VIC, Australia; [5]Nucleus Network, Melbourne, VIC, Australia; [6]Guangdong General Hospital, Guangzhou, Guangdong, China; [7]Beijing Cancer Hospital, Beijing, China; [8]BeiGene (Shanghai) Co., Ltd., Shanghai, China; [9]BeiGene (Beijing) Co., Ltd., Beijing, China; [10]University of Western Australia, Crawley, and Linear Clinical Research, Perth, WA, Australia

POSTER CT167 / 11
AACR Annual Meeting
2 April 2019, Atlanta, GA

BACKGROUND

- Sitravatinib is an investigative, orally bioavailable, spectrum-selective receptor tyrosine kinase (RTK) inhibitor with potential antitumor activity[1] that has been shown to potently inhibit split kinase receptors (eg, VEGFR2, KIT) and TAM receptors (eg, AXL, MER), which are dysregulated in many cancers[2]
- Inhibition of RTKs by sitravatinib may also modulate effects on the tumor microenvironment to overcome resistance to checkpoint inhibitors, including enhanced M1/suppressed M2 macrophage cytokine response, abrogated negative regulation of antitumor natural killer cell activity, and depleted regulatory T- and myeloid-derived suppressor cells with enhanced antitumor cytotoxic T-cell activity[3] (**Figure 1**)
- Tislelizumab (BGB-A317) is an investigational, humanized, immunoglobulin 4 monoclonal antibody that has been shown to have high affinity for and binding specificity to programmed cell death protein-1 (PD-1)[4]
- Tislelizumab was specifically engineered to minimize binding to FcγR on macrophages in order to abrogate antibody-dependent phagocytosis,[4] a mechanism of T-cell clearance and potential resistance to anti-PD-1 therapy[5] (**Figure 2**)
- Combining an agent that has both immune modulatory and antitumor properties with an immunotherapeutic PD-1 checkpoint inhibitor could enhance the antitumor efficacy observed with either agent alone[6,7]

METHODS

Overall Design and Study Objective

- This is an open-label, multicenter, nonrandomized, phase 1b clinical trial (NCT03666143) designed to assess the safety and tolerability of sitravatinib combined with tislelizumab in patients with advanced solid tumors

Study Population

- Adult patients with histologically or cytologically confirmed, locally advanced or metastatic, non-small cell lung cancer, renal cell carcinoma, or ovarian cancer

Figure 1: Sitravatinib in the Tumor Microenvironment

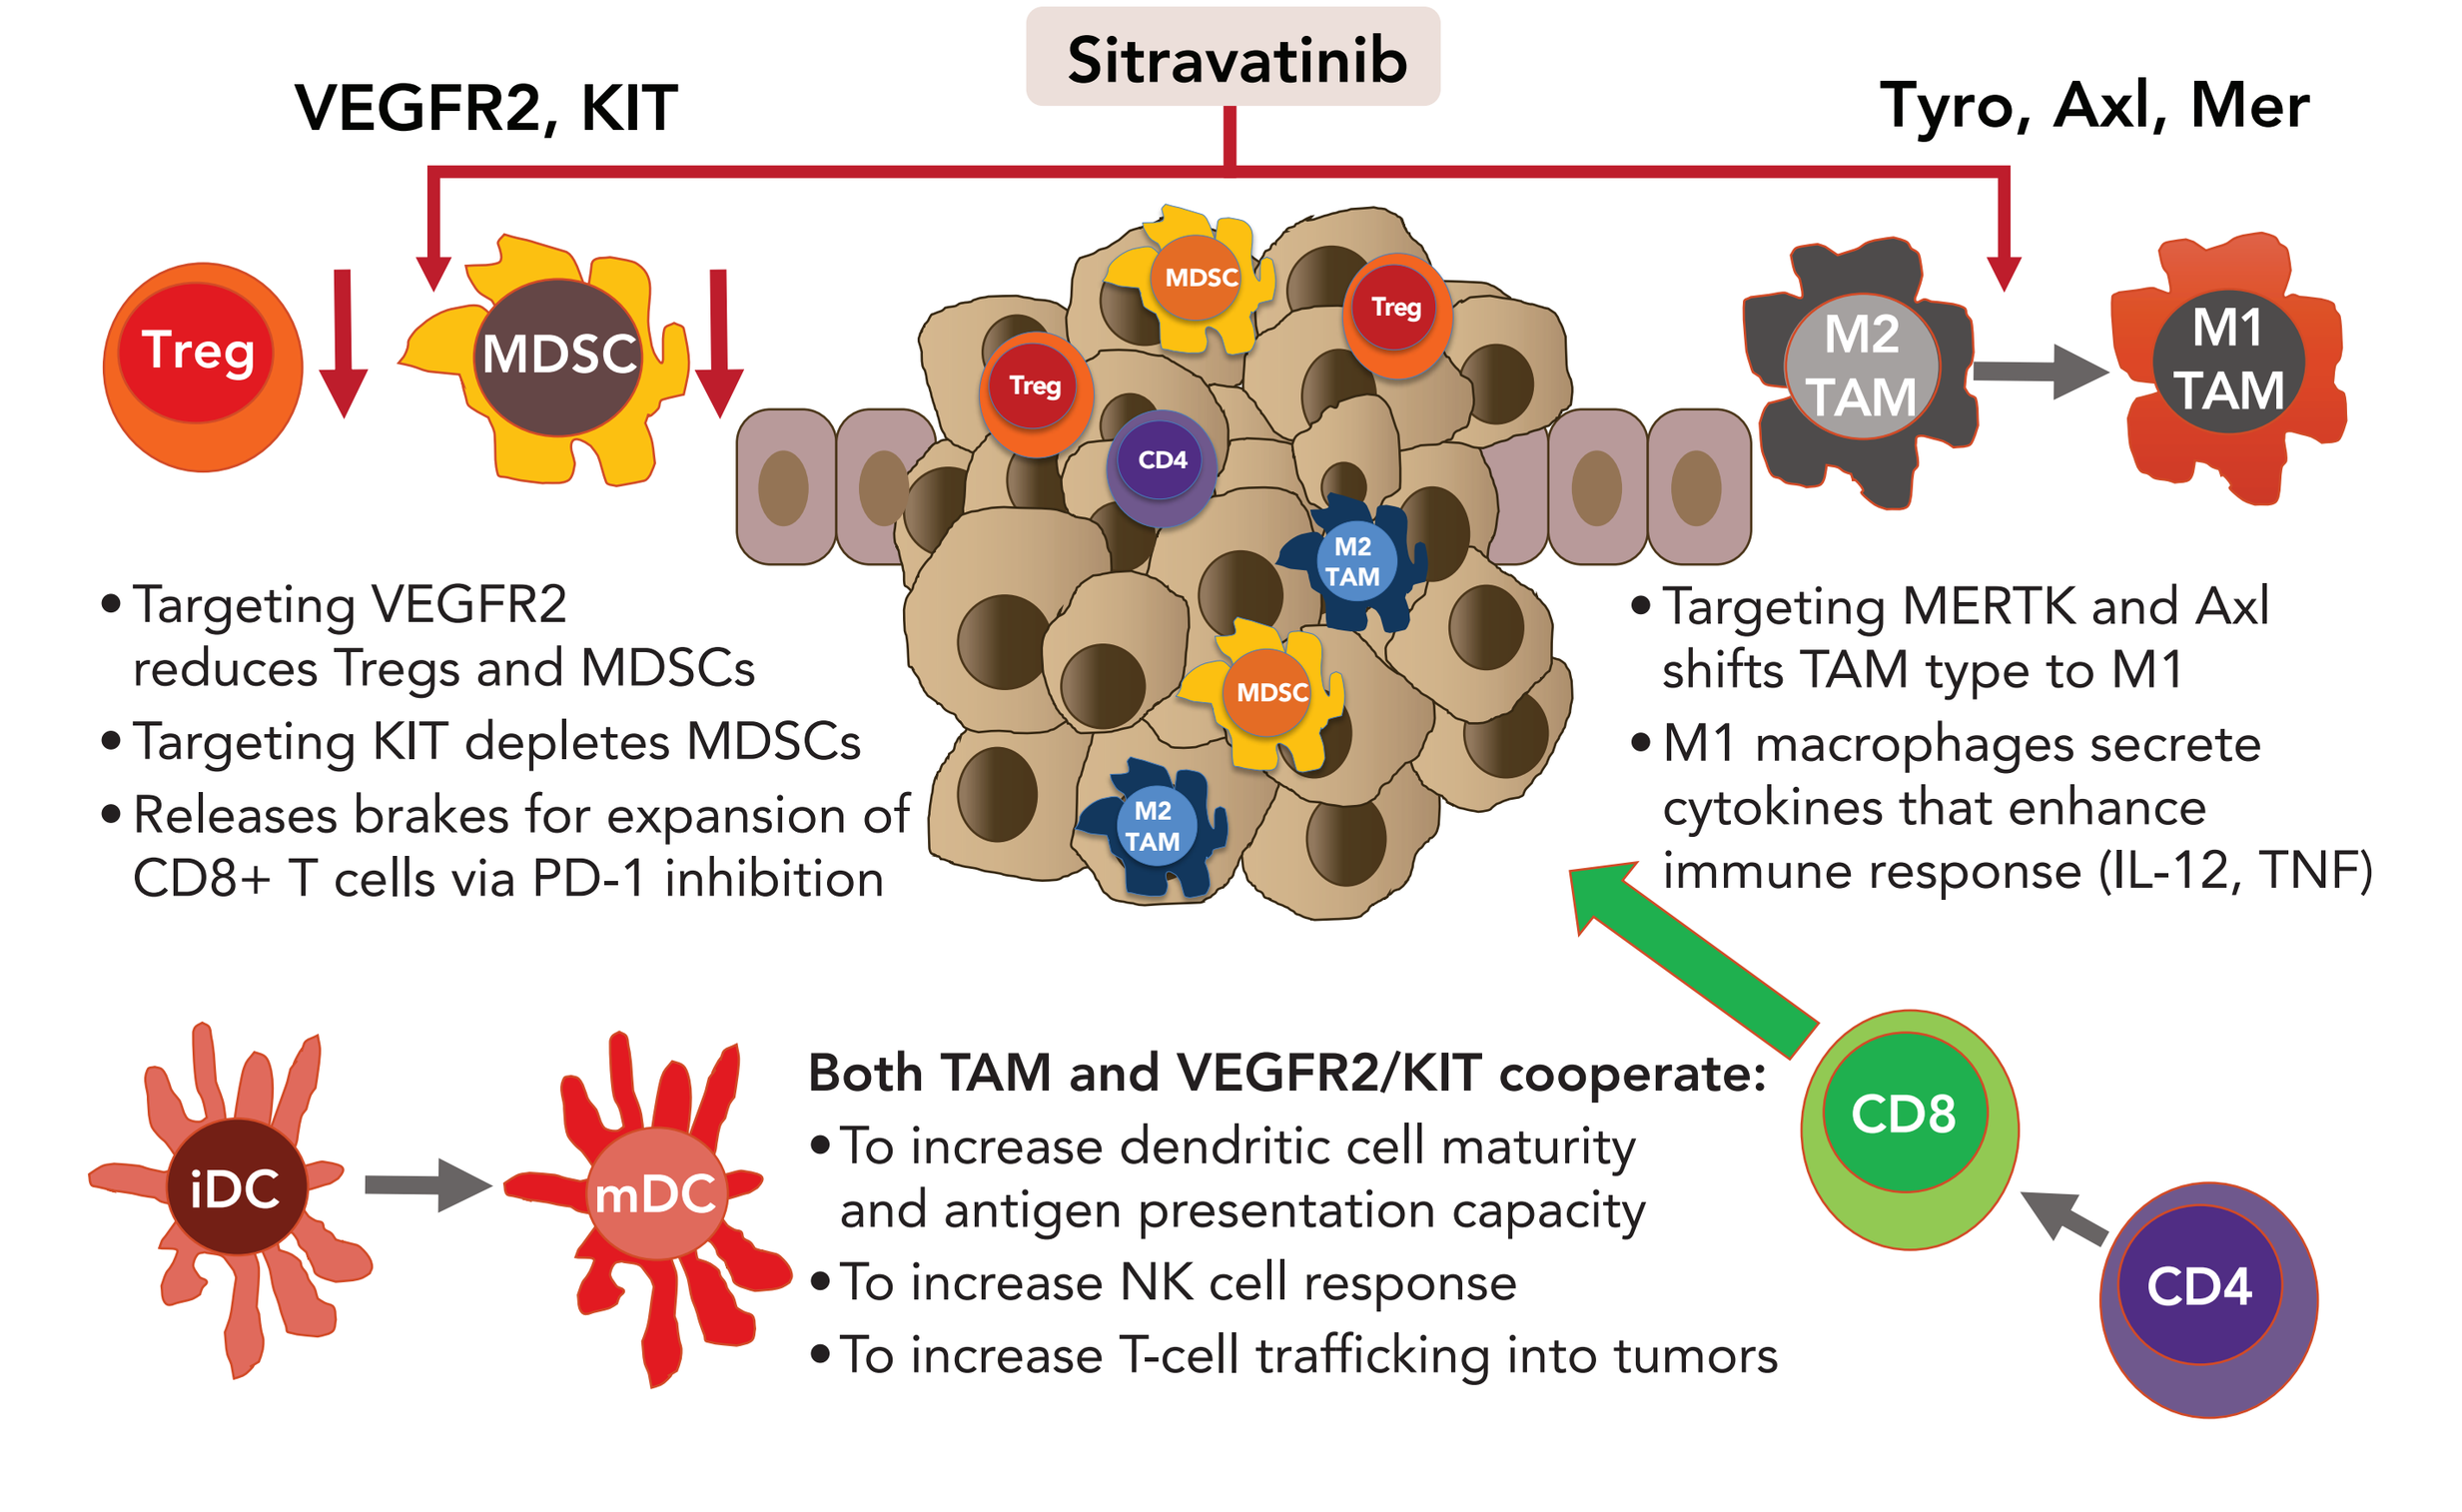
Sitravatinib
VEGFR2, KIT
Tyro, Axl, Mer
Treg
MDSC
M2 TAM
M1 TAM
CD4
CD8
iDC
mDC
• Targeting VEGFR2 reduces Tregs and MDSCs
• Targeting KIT depletes MDSCs
• Releases brakes for expansion of CD8+ T cells via PD-1 inhibition
• Targeting MERTK and Axl shifts TAM type to M1
• M1 macrophages secrete cytokines that enhance immune response (IL-12, TNF)
Both TAM and VEGFR2/KIT cooperate:
• To increase dendritic cell maturity and antigen presentation capacity
• To increase NK cell response
• To increase T-cell trafficking into tumors

iDC=immature dendritic cell, IL-12=interleukin 12, mDC=mature dendritic cell, MDSC=myeloid-derived suppressor cell, NK=natural killer, PD-1=programmed cell death protein-1, TAM=tumor-associated macrophage, TNF=tumor necrosis factor, Treg=T regulatory cell, VEGFR2=vascular endothelial growth factor receptor 2

- Eligibility criteria include Eastern Cooperative Oncology Group Performance Status ≤1 and adequate end-organ function
- Approximately 100 patients are expected to be enrolled in 5 cohorts (n=20 patients per cohort) (**Figure 3**)

TREATMENT

- All patients will receive sitravatinib 120 mg given orally once daily in combination with tislelizumab 200 mg given intravenously every 3 weeks
- Treatment will be administered until progressive disease, unacceptable toxicity, death, withdrawal of consent, or study termination by the sponsor

STUDY ASSESSMENTS AND STATISTICAL ANALYSIS

- The primary endpoint is assessment of safety and tolerability of the combination of sitravatinib with tislelizumab by monitoring adverse events (AEs) and serious AEs

Figure 2: Anti-PD-1 Monoclonal Antibodies With and Without FcγR Binding

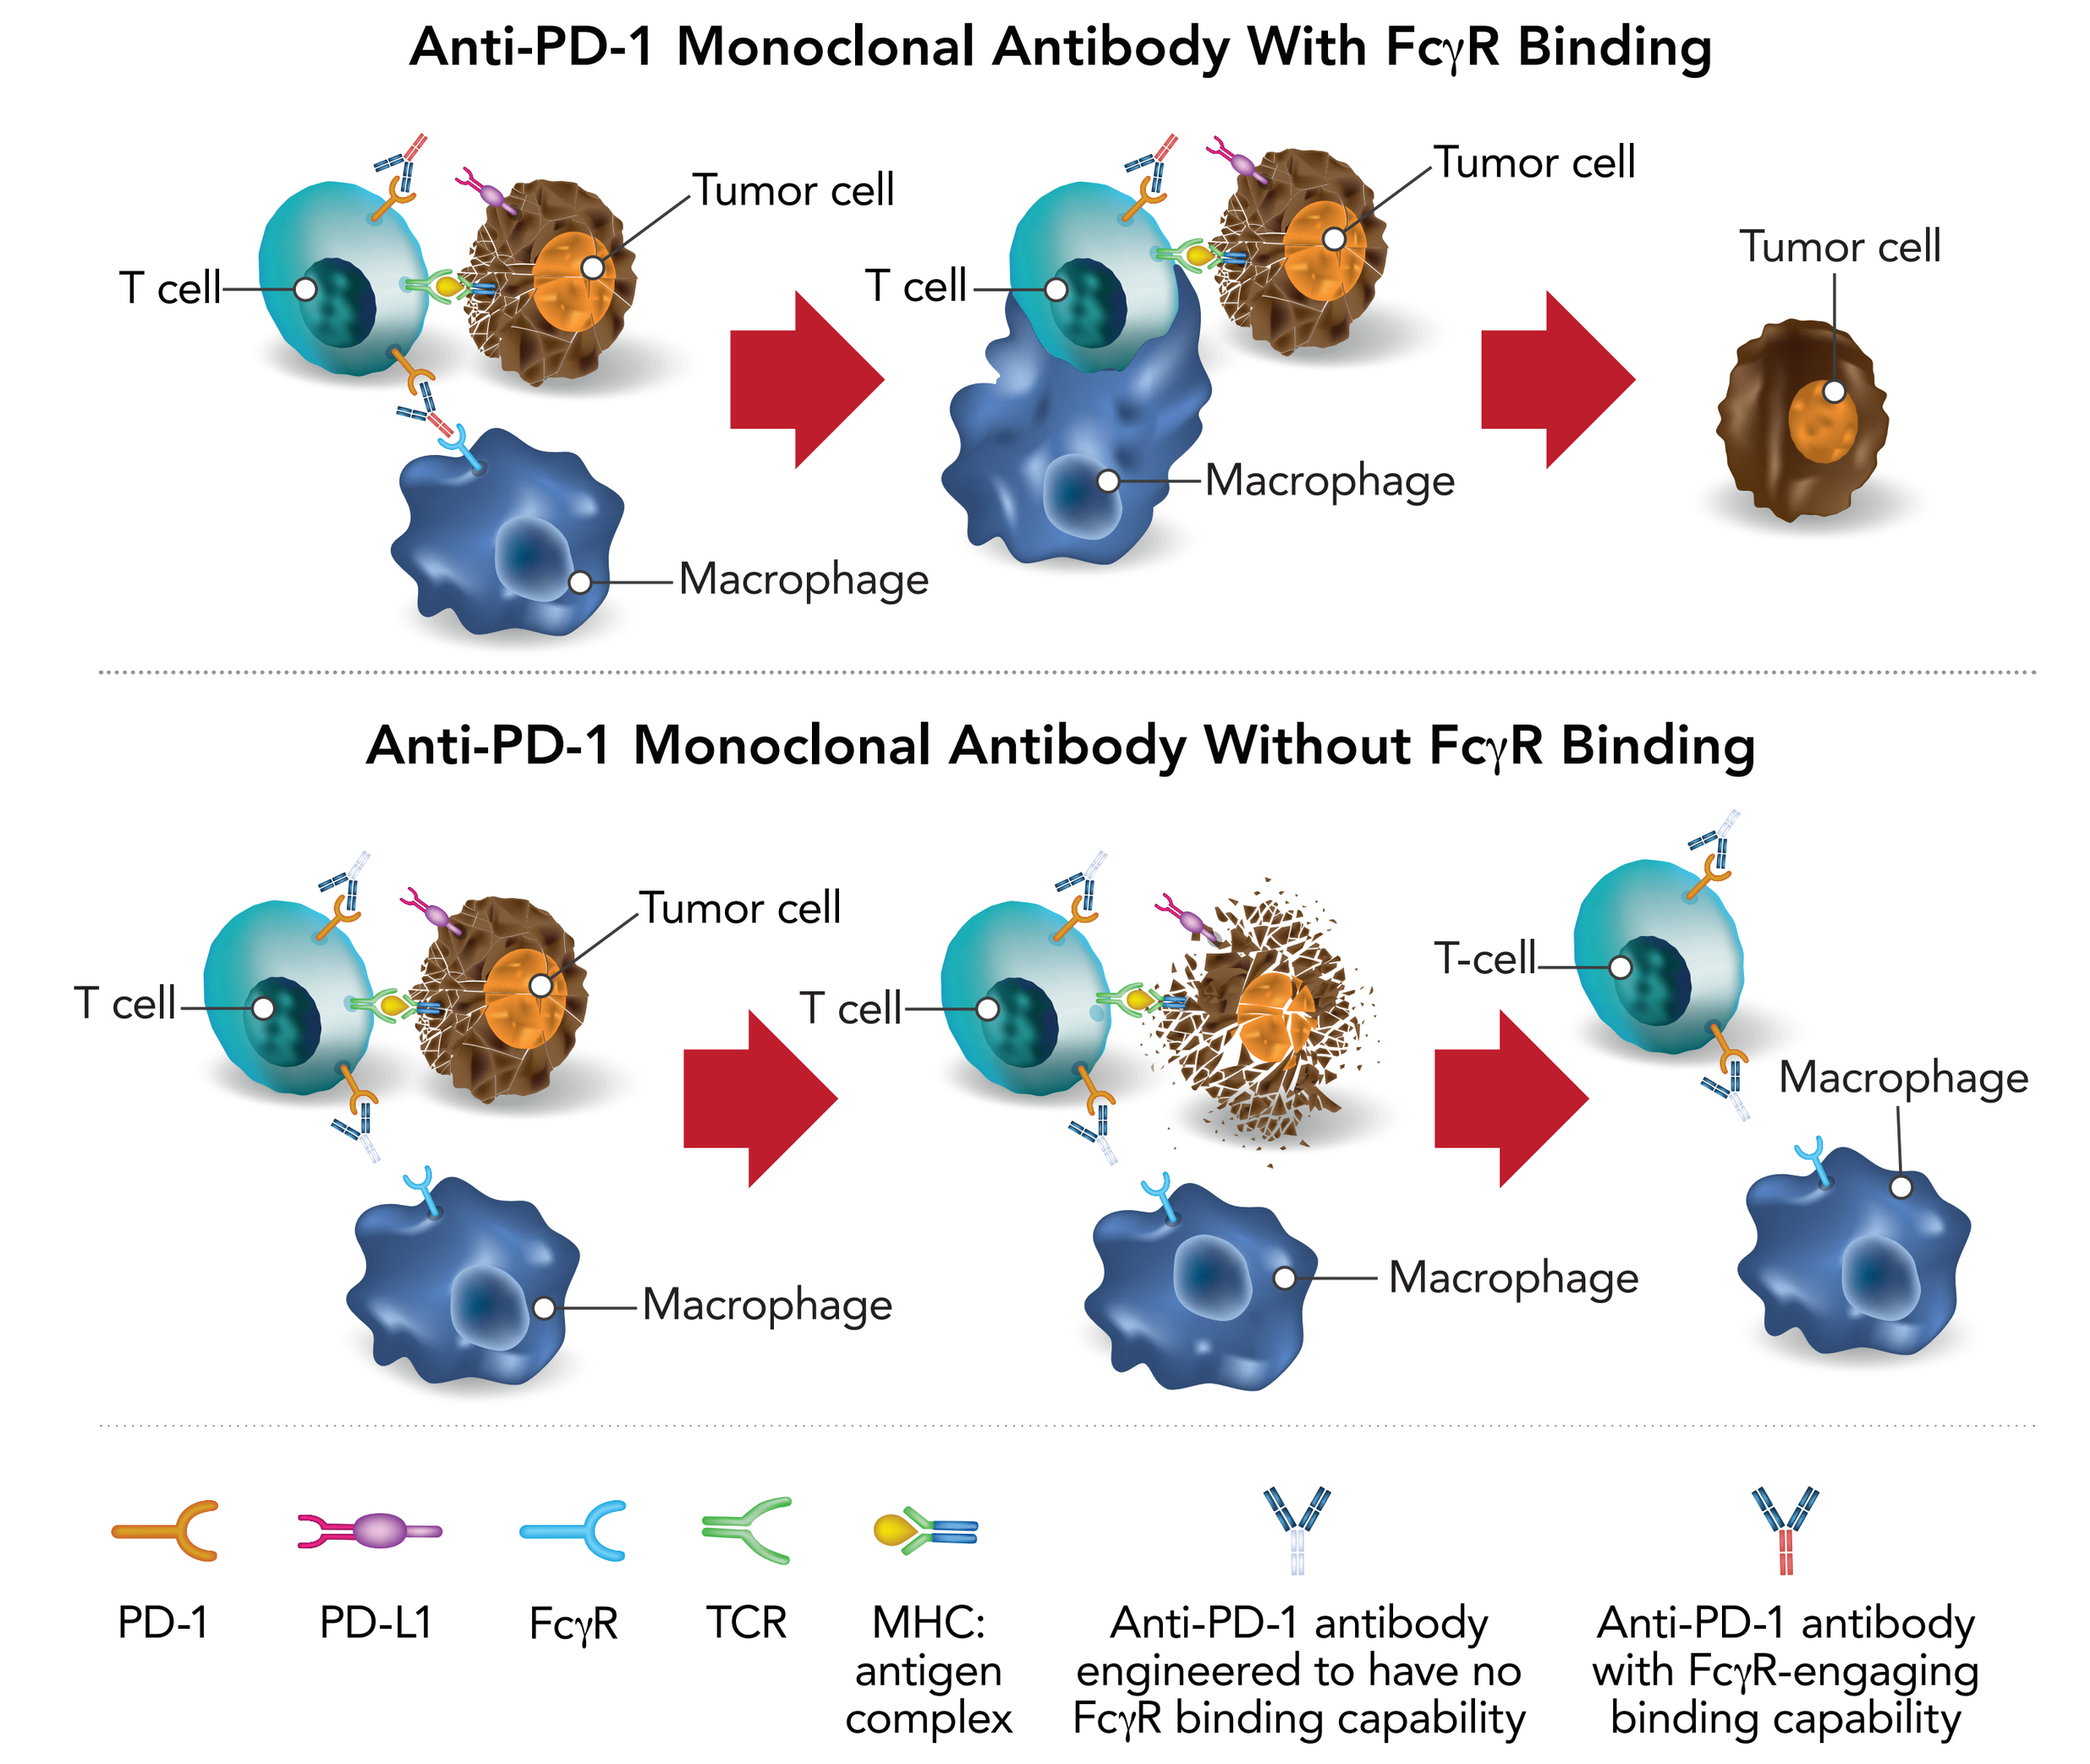
Anti-PD-1 Monoclonal Antibody With FcγR Binding
T cell
Tumor cell
Macrophage
Anti-PD-1 Monoclonal Antibody Without FcγR Binding
T-cell
PD-1
PD-L1
FcγR
TCR
MHC: antigen complex
Anti-PD-1 antibody engineered to have no FcγR binding capability
Anti-PD-1 antibody with FcγR-engaging binding capability

FcγR=Fc gamma receptor, MHC=major histocompatibility complex, PD-1=programmed cell death protein-1, PD-L1=programmed cell death ligand-1, TCR=T-cell receptor

- Secondary endpoints include overall response rate, duration of response, disease control rate, progression-free survival assessed by Response Evaluation Criteria In Solid Tumors (RECIST) version 1.1 (v1.1), and plasma concentrations and derived pharmacokinetic parameters of single- and repeated-dose sitravatinib
- Safety and tolerability will be assessed by monitoring AEs, including immune-related AEs, and through physical examinations, electrocardiograms, and laboratory assessments
- Tumor response will be evaluated every 6 weeks during Year 1 and every 9 weeks from Year 2 onwards, in accordance with RECIST v1.1
 – For ovarian cancer patients, response will be assessed using RECIST v1.1 and Gynecologic Cancer InterGroup criteria

Figure 3: Study Design

Cohort	Description	Eligibility	Planned (n=20/cohort) Currently enrolled (as of: 28 Feb 2019)
A	Anti-PD-(L)1 antibody-refractory/-resistant, metastatic, nonsquamous **NSCLC**	• Radiographic progression per RECIST v1.1 on/after anti-PD-1/PD-L1 as most recent treatment • ≤2 lines of therapy • No other prior immunotherapies, including, but not limited to, anti-CTLA-4, anti-OX40, and anti-CD137	1
B	Anti-PD-(L)1 antibody-naive, metastatic, nonsquamous **NSCLC**	• Radiographic progression per RECIST on/after chemotherapy for metastatic NSCLC without prior exposure to anti-PD-1/PD-L1 agent • ≤2 lines of therapy	1
C	Anti-PD-(L)1 antibody-refractory/-resistant, metastatic, clear cell **RCC**	• Radiographic progression per RECIST on/after anti-PD-1/PD-L1 as most recent treatment • ≤2 lines of therapy • No other prior immunotherapies, including, but not limited to, anti-CTLA-4, anti-OX40, and anti-CD137	1
D	Metastatic or advanced **RCC** without prior systemic therapy*	• Systemic therapy not available, is considered not suitable, or has been refused by the patient • Prior neoadjuvant/adjuvant therapy for RCC is acceptable if completed >12 months prior to enrollment	1
E	Anti-PD(L)1 antibody-naive, recurrent, platinum-resistant, epithelial **OC**†	• No platinum-refractory disease (PD <1 month of last dose of platinum-based chemotherapy) • No prior exposure to anti-PD-1/PD-L1 agent	17

→ **Sitravatinib 120 mg PO QD + Tislelizumab 200 mg IV Q3W** →

Primary Endpoint
- **Safety and tolerability**

Secondary Endpoints
- **ORR**
- **DOR**
- **DCR**
- **PFS by RECIST v1.1**
- **Plasma concentrations and derived PK parameters of single- and repeated-dose sitravatinib**

*Cohort D will only enroll in China. †Resistance to platinum-based therapy is defined as relapse 1–6 months after last dose of platinum-based treatment.
DCR=disease control rate, DOR=duration of response, IV=intravenous, NSCLC=non-small cell lung cancer, OC=ovarian cancer, ORR=overall response rate, PD=progressive disease, PD-1=programmed cell death protein-1, PD-L1=programmed cell death ligand-1, PFS=progression-free survival, PK=pharmacokinetics, PO=per oral, Q3W=every 3 weeks, QD=once daily, RCC=renal cell carcinoma, RECIST=Response Evaluation Criteria In Solid Tumors, v1.1=version 1.1

TRIAL STATUS

- 21 patients have enrolled as of 28 February 2019 at the study sites in Australia and China

REFERENCES

1. Patwardhan PP et al. Oncotarget. 2016;7:4093-4109.
2. Blume-Jensen P, Hunter T. Nature. 2001;411:355-365.
3. Du W et al. JCI Insight. 2018;3:e124184.
4. Zhang T et al. Cancer Immunol Immunother. 2018;67:1079-1090.
5. Dahan R et al. Cancer Cell. 2015;28:285-295.
6. Leal TA et al. Ann Oncol. 2018;29(suppl 8):viii400-vii441.
7. Leal T et al. IASLC 18th World Conference on Lung Cancer; 15-18 October 2017; Yokohama, Japan; abstract MA 02.01.

ACKNOWLEDGMENTS

We would like to thank the investigators, site support staff, and especially the patients for participating in this study. This study is sponsored by BeiGene. Editorial support funded by BeiGene was provided by Ira Mills, PhD, and Shannon Davis of Ashfield Healthcare Communications.

Copies of this poster obtained through Quick Response Code are for personal use only and may not be reproduced without permission from the author of this poster.