HEALTHCARE COSTS AND RESOURCE UTILIZATION ASSOCIATED WITH PARP INHIBITORS IN OVARIAN CANCER PATIENTS INITIALLY TREATED WITH PLATINUM-BASED THERAPY

Soraya Azmi[1], Jason C. Allaire[2], Mark Balk[1], Jane Dennison[1], Boxiong Tang[1]

[1]BeiGene, Ltd.; [2]Generativity Health Economics and Outcomes Research

ISPOR Europe 2020
16-19 November 2020, Virtual Conference

INTRODUCTION

- Ovarian cancer (OC) is the eighth most common type of cancer and the fifth leading cause of cancer death in the EU[1].
- The economic burden of OC is substantial with annual all-cause total costs ranging from \$134,025-\$146,267 with annual ovarian cancer-related total costs ranging from \$82,349-\$91,508[2].
- First-line treatment generally involves surgery and/or interval debulking followed by platinum-containing chemotherapy (carboplatin and paclitaxel)[3].
- Similar to the NCCN Guidelines[4], the recently updated ESMO guidelines suggest that PARP inhibitors (PARPi) (olaparib, niraparib or rucaparib) be used for maintenance therapy following a response to platinum-based therapy in patients with recurrent platinum-sensitive high-grade ovarian cancer [5].
- Few published studies have yet to examine the real-world healthcare-related costs and utilization associated with PARPi use.
 - One study reported a comparison of mean drug costs per month. However, the costs included only drug costs calculated over the time that the PARPi was being prescribed[6].
 - Information about the medical costs including hospitalizations and emergency room visits is limited to a cost-effectiveness analysis which utilized cost estimates[7].
 - A community practice-based study showed toxicity profiles of the existing PARPis were similar to clinical trials, but only reported average cost per cycle[8].
- The objective of this analysis was to explore the real-world costs and healthcare utilization in OC patients undergoing PARPi treatment following first-line platinum-based therapy using U.S. administrative claims data.

MATERIALS

Data Source and Cohort Creation

- OC patients initiating a PARPi after first-line platinum-based therapy were identified in the IBM Marketscan database (1/1/2009 to 7/31/2019).
- Eligible patients met the following criteria:
 - At least one claim for OC (ICD-9-CM: 183.x; ICD-10-CM: c56.1, c56.2, c56.9)
 - Continuous enrollment at least 90 days prior to and 60 days after first OC claim
 - Began first-line platinum-based chemotherapy within 30 days of first OC claim
- The date of the first PARPi claim after platinum-based therapy was the index date; the follow-up period ended at the end of available data.
- Patients were categorized into 3 cohorts (olaparib, rucaparib or niraparib) based on the first PARPi they received following platinum therapy.

Key Variables and Analytic Approach

- Pharmacy, medical (nonpharmacy) and total costs were calculated on a per-member per-month (PMPM) basis from index date through the follow-up period.
- Healthcare utilization (outpatient visits [OP], emergency department [ED] visits, and hospitalization) were calculated on a PMPM basis from index date through the end of the follow-up period.
- Mean differences in age and duration of PARPi therapy between cohorts were examined using analysis of variance with pairwise post-hoc tests.
- Due to their non-normal distributions, differences in medians were examined for the costs and resource utilization variables using the Kruskal–Wallis non-parametric tests.

RESULTS

Patient Characteristics

- A total of 276 patients were identified with 48% receiving olaparib (n = 133), 35% niraparib (n =96), and 17% rucaparib (n = 47) (Table 1).
- There was a significant difference in age of the cohorts with the olaparib cohort being significantly younger than the niraparib and rucaparib cohorts.
- There was no difference among the cohorts for median follow-up time or initiation time of PARPi.
- Olaparib and rucaparib patients received treatment over a significantly longer time than those on niraparib.

Table 1. Baseline Characteristics (n = 276)

	Olaparib	Niraparib	Rucaparib	P-value
Number, % of patients	133 (48%)	96 (35%)	47 (17%)	
Age, mean (SD)	56.49 (8.17)	59.03 (8.90)	59.11 (7.95)	0.04
34-54, n %	49, 36.8%	27, 28.1%	16, 34.0%	
55-60, n %	43, 32.3%	28, 29.2%	11, 23.4%	
61-82, n %	41, 30.8%	41, 42.7%	20, 42.6%	
Median follow-up time, days	286.00	300.50	253.00	0.29
Median time to PARPi initiation, days	781.00	731.00	779.00	0.80
Median time on PARPi, days	149.11	94.00	141.50	0.03

Healthcare Costs

- As can been seen in Table 2 and Figure 1, there was a significant difference in Pharmacy Costs.
 - Follow-up tests indicated that median Pharmacy Costs were significantly higher in olaparib (\$10,312) and rucaparib (\$9,290) patients relative to niraparib patients (\$6,561) ($p<0.05$).
- A trend ($p=0.06$) was found for medical costs with niraparib being higher (\$5,231) than the other two PARPi (olaparib, \$3,194; rucaparib, \$2,197).

Table 2. Median Costs PMPM by PARPi Cohort

	Olaparib	Niraparib	Rucaparib	P-value
Number, % of patients	133 (48%)	96 (35%)	47 (17%)	
All Costs (\$) PMPM				
Median	**15,506**	**13,365**	**15,465**	0.08
Mean	16,958	15,421	17,848	
(SD)	(7,954)	(11,364)	(11,365)	
Medical Costs (\$) PMPM				
Median	**3,194**	**5,231**	**2,197**	0.06
Mean	6,657	8,297	7,689	
(SD)	(8,374)	(8,762)	(11,554)	
Pharmacy Costs (\$) PMPM				
Median	**10,312**	**6,561**	**9,290**	0.00
Mean	10,301	7,124	10,159	
(SD)	(5,560)	(4,567)	(6,089)	

Figure 1. Median Costs PMPM by PARPi Cohort

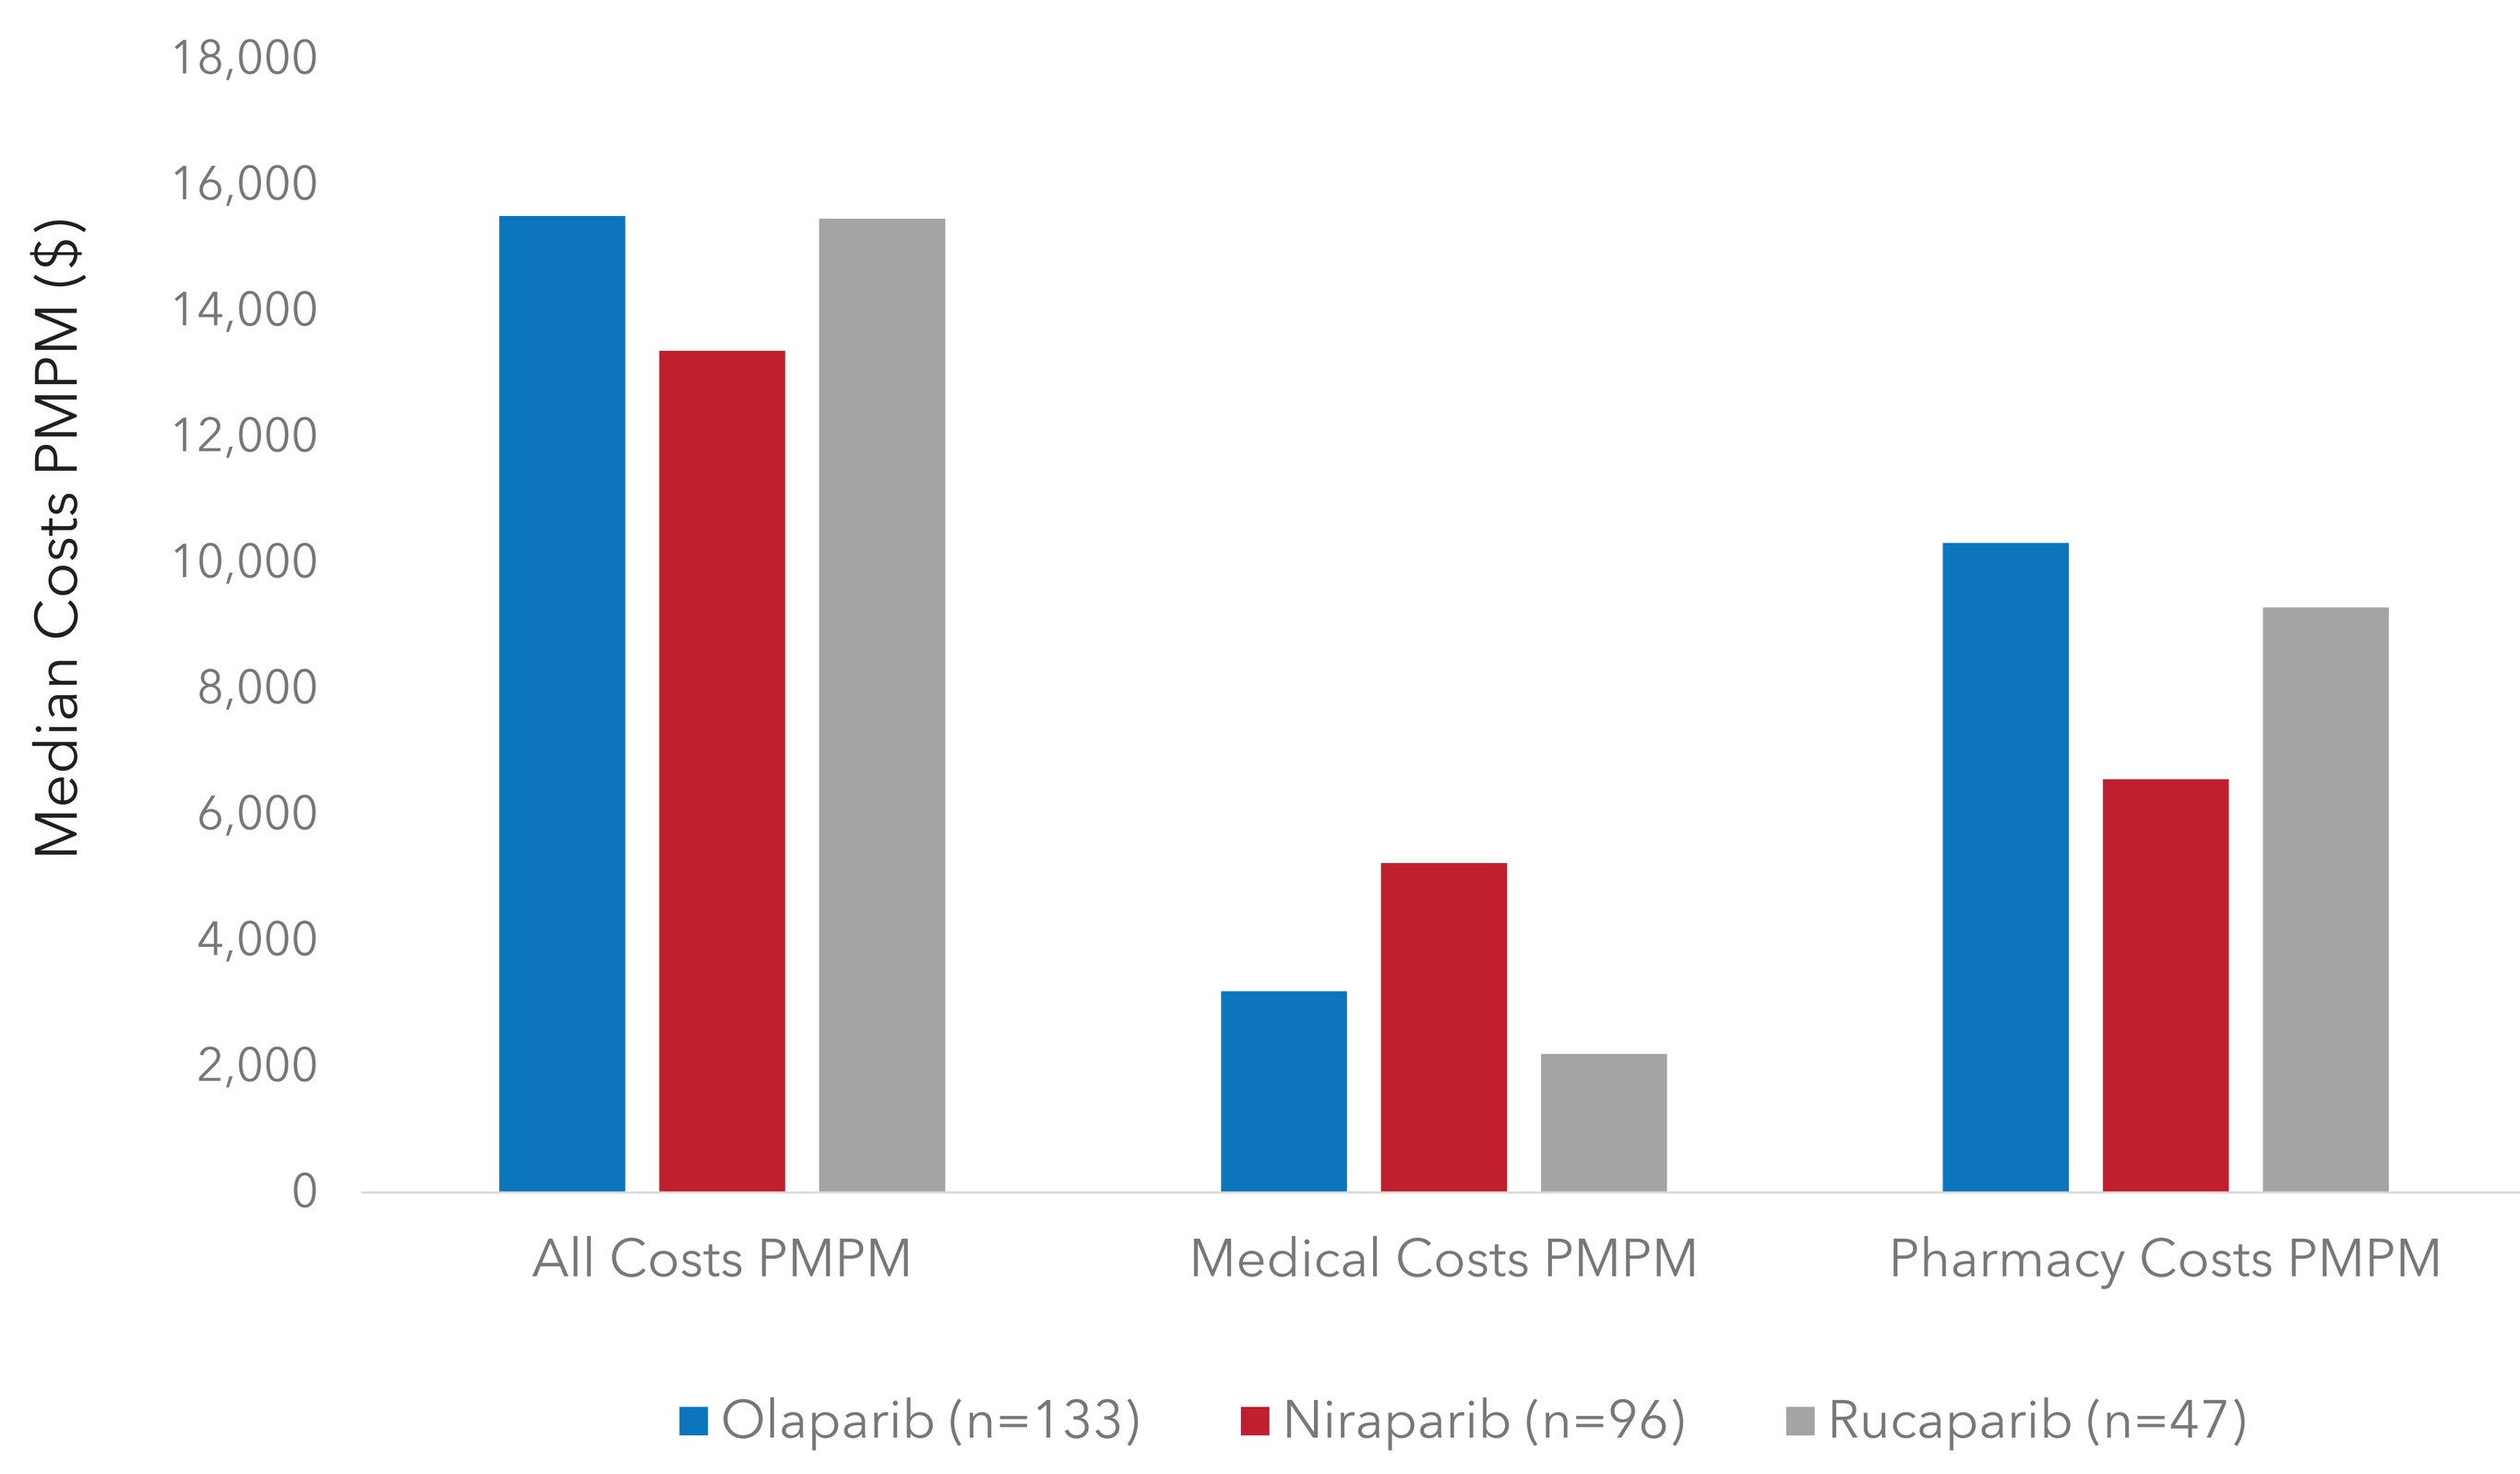
18,000
16,000
14,000
12,000
10,000
8,000
6,000
4,000
2,000
0
Median Costs PMPM (\$)
All Costs PMPM
Medical Costs PMPM
Pharmacy Costs PMPM
Olaparib (n=133)
Niraparib (n=96)
Rucaparib (n=47)

Healthcare Resource Utilization

- Resource utilization (OP visits, ER visits and hospital stay) showed a trend for higher proportion of patients on niraparib requiring OP and ED visits (Table 3).
- Though not statistically significant, the number of ER days and the length of hospital stays was higher in the rucaparib patients than the other two cohorts.

Table 3. Healthcare Utilization

	Olaparib	Niraparib	Rucaparib	P-value
Outpatient Visits PMPM				
Median	**1.34**	**1.51**	**1.30**	0.35
Mean	1.52	1.75	1.44	
(SD)	(.96)	(1.09)	(.77)	
ER Visit (YES/NO), n (%)	53 (39.8%)	43 (44.8%)	17 (36.2%)	0.58
# ER days				
Median	**1.22**	**1.64**	**3.98**	0.26
Mean	2.80	3.36	4.14	
(SD)	(4.49)	(7.21)	(4.59)	
Hospital Stay (YES/NO), n (%)	42 (31.6%)	14 (29.8%)	36 (39.1%)	0.55
# Days in Hospital PMPM				
Median	**0.76**	**0.82**	**1.60**	0.27
Mean	1.18	1.46	1.99	
(SD)	(1.45)	(2.03)	(2.14)	

CONCLUSIONS

- Overall costs during the entire follow up period, were highest for olaparib and rucaparib patients. However, these higher costs seem to be primarily driven by the significantly higher pharmacy costs.
- Despite the shorter time on treatment, niraparib patients had a trend toward higher medical costs than those on olaparib and rucaparib. However, niraparib patients had significantly lower pharmacy costs. The opposite was found with olaparib and rucaparib (significantly higher pharmacy costs, but lower medical costs).
- These findings suggest that pharmacy costs might serve as a strong driver of the total treatment costs among PARPis.
 - Pricing differences for various PARPi will have a significant impact on overall pharmacy costs.
- Limitations for this study are that:
 - As an exploratory study, these results do not differentiate between lines of therapy, or by treatment versus maintenance.
 - The analysis performed did not address confounding or bias. Therefore, caution should be applied in interpreting this data.
 - We explored only medical and pharmacy costs, while other economic and humanistic outcomes could also be considered.
 - Specific drivers of medical costs were not available in this claims database but these data could prove useful in choosing between agents.
- Future research should examine cost and utilization by line of therapy and treatment versus maintenance.
- Increased understanding about the clinical outcome and economic cost drivers among currently available PARPi will provide greater insight toward improved clinical and payer decision-making.

REFERENCES

1. The European Cancer Information System (ECIS); https://ecis.jrc.ec.europa.eu/. Accessed September 10, 2020.
2. Chin L, Hansen RN, Carlson JJ. Economic Burden of Metastatic Ovarian Cancer in a Commercially Insured Population: A Retrospective Cohort Analysis. *Journal of Managed Care & Specialty Pharmacy*. 2020;26(8):962-970.
3. Ledermann JA, Raja FA, Fotopoulou C, Gonzalez-Martin A, Colombo N, Sessa C. Newly diagnosed and relapsed epithelial ovarian carcinoma: ESMO Clinical Practice Guidelines for diagnosis, treatment and follow-up. *Annals of oncology* : official journal of the European Society for Medical Oncology. 2018;29(Suppl 4):iv259.
4. National Comprehensive Cancer Network. Ovarian Cancer (Version 2.2019). http://www.nccn.org/professionals/physician_gls/pdf/ovarian.pdf. Accessed September 10, 2020.
5. ESMO Guidelines Committee. eUpdate – Relapsed Epithelial Ovarian Carcinoma Treatment Recommendations 1 April 2020. ESMO. https://www.esmo.org/guidelines/gynaecological-cancers/newly-diagnosed-and-relapsed-epithelial-ovarian-carcinoma/eupdate-ovarian-cancer-treatment-recommendations. Accessed 13 September, 2020.
6. McBride A, Perhanidis J, Gibson C, Wetzstein G. HSR20-099: Cost Comparison of PARP Inhibitors in Women With Ovarian Cancer (OC) in the US HealthCare Market. 2020;18(3.5):HSR20-099.
7. Guy H, Walder L, Fisher M. Cost-Effectiveness of Niraparib Versus Routine Surveillance, Olaparib and Rucaparib for the Maintenance Treatment of Patients with Ovarian Cancer in the United States. *PharmacoEconomics*. 2019;37(3):391-405.
8. Eakin C, Ewongwo A, Pendleton L, Monk J, Chase D. Real world experience of poly (ADP-ribose) polymerase inhibitor use in a community oncology practice. Gynecol Oncol. 2020 Oct;159(1):112-117.

FUNDING SOURCE: This study was funded by BeiGene, Ltd.