Pathological Response to Neoadjuvant Tislelizumab (TIS) Plus Platinum-Doublet (PtDb) Chemotherapy (CT) in Resectable Stage II-IIIA NSCLC Patients (pts) in the Phase 3 (Ph3) RATIONALE-315 Trial

Federico Cappuzzo,[1] Dongsheng Yue,[2] Wenxiang Wang,[3] Hongxu Liu,[4] Qixun Chen,[5] Chun Chen,[6] Jun Zhang,[7] Fan Bai,[8] Changli Wang[2]

[1]Istituto Nazionale Tumori IRCCS Regina Elena, Roma, Italy; [2]Tianjin Medical University Cancer Institute and Hospital, Tianjin, China; [3]Hunan Cancer Hospital, Hunan, China; [4]Liaoning Cancer Hospital and Institute, Shenyang, China; [5]Zhejiang Cancer Hospital, Hangzhou, China; [6]Fujian Medical University Union Hospital, Fuzhou, China; [7]BeiGene USA, Inc., San Mateo, CA, USA; [8]BeiGene (Shanghai) Co., Ltd., Shanghai, China

Poster No: B09 Presented at XXVI Italian Medical Oncology Association (AIOM); November 8-10, 2024; Rome, Italy

Conclusions

- **Tislelizumab (TIS) + chemotherapy (CT) showed statistically significant and clinically meaningful improvements in major pathological response (MPR) and pathological complete response (pCR) rates versus placebo (PBO) + CT as neoadjuvant treatment**
- **The safety profile of TIS + CT is manageable and consistent with previous reports, further supporting this treatment combination for patients with resectable stage II or IIIA non-small cell cancer (NSCLC)**
- **The RATIONALE-315 study is ongoing; a subsequent interim analysis showed significant improvement in event-free survival (EFS) in the TIS arm (these data will be shared at a future meeting)**

Background

- Lung cancer is the second most diagnosed cancer globally and the leading cause of cancer-related mortality worldwide[1]
 - NSCLC is the predominant subtype of lung cancer, accounting for nearly 85% of lung cancer cases[2]
- Surgery offers the highest likelihood of curing patients with early stage NSCLC,[3] but approximately 30% to 55% of patients experience disease recurrence after curative surgery[4]
- (Neo)adjuvant CT has been recommended for patients with resectable stage II-IIIA NSCLC[5]
 - Studies have shown promising pathological response rates (ie, MPR, pCR) with neoadjuvant anti-programmed cell death protein-1 or programmed death-ligand 1 (PD-[L]1) monoclonal antibodies (mAbs) ± CT[5]
 - However, post-operative recurrence remains a concern[5]
- RATIONALE-315 (NCT04379635) is investigating the efficacy and safety of neoadjuvant TIS (anti-PD-1 mAb) + CT or PBO + CT, then adjuvant TIS or PBO, in patients with resectable stage II-IIIA NSCLC in China
- Here we present the MPR and pCR results at the data cutoff of February 20, 2023

Methods

- RATIONALE-315 is a randomized, double-blind, placebo-controlled, phase 3 study (**Figure 1**)
- Patients were required to have resectable stage II or IIIA NSCLC, be eligible for R0 resection, and have an ECOG PS of 0 or 1
- Eligible patients stratified by histology, disease stage, and PD-L1 expression were randomized 1:1 to either Arm A (TIS + CT) or Arm B (PBO + CT)
- The surgical procedure was performed within 4 to 6 weeks after the last dose of neoadjuvant treatment
- Eligible patients entered the adjuvant phase 2 to 8 weeks after surgery

Figure 1. RATIONALE-315 Study Design

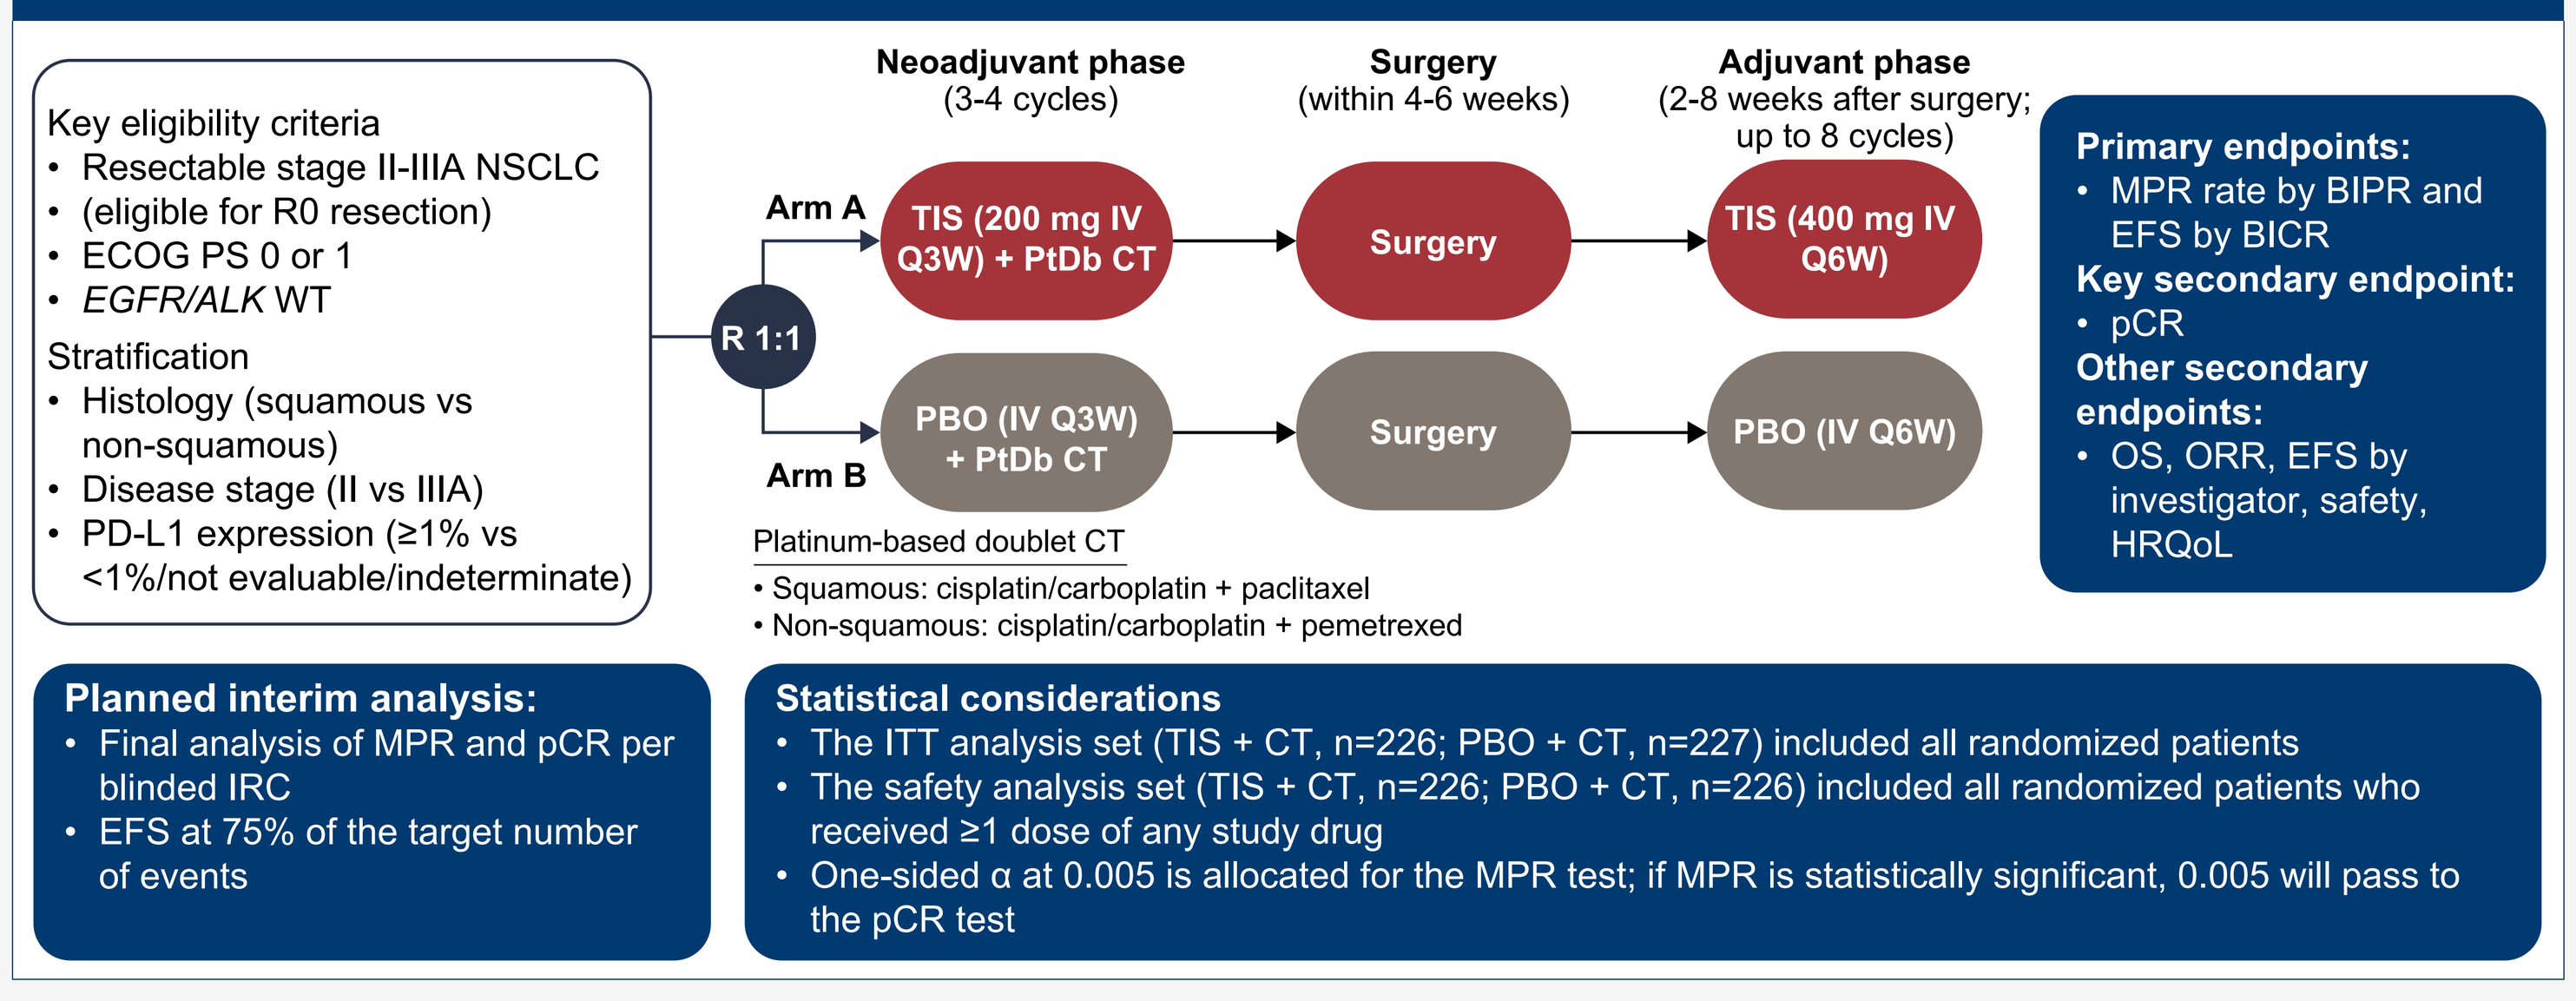
Key eligibility criteria
• Resectable stage II-IIIA NSCLC
• (eligible for R0 resection)
• ECOG PS 0 or 1
• EGFR/ALK WT
Stratification
• Histology (squamous vs non-squamous)
• Disease stage (II vs IIIA)
• PD-L1 expression (≥1% vs <1%/not evaluable/indeterminate)
Neoadjuvant phase (3-4 cycles)
Surgery (within 4-6 weeks)
Adjuvant phase (2-8 weeks after surgery; up to 8 cycles)
R 1:1
Arm A
TIS (200 mg IV Q3W) + PtDb CT
Surgery
TIS (400 mg IV Q6W)
Arm B
PBO (IV Q3W) + PtDb CT
Surgery
PBO (IV Q6W)
Platinum-based doublet CT
• Squamous: cisplatin/carboplatin + paclitaxel
• Non-squamous: cisplatin/carboplatin + pemetrexed
Primary endpoints:
• MPR rate by BIPR and EFS by BICR
Key secondary endpoint:
• pCR
Other secondary endpoints:
• OS, ORR, EFS by investigator, safety, HRQoL
Planned interim analysis:
• Final analysis of MPR and pCR per blinded IRC
• EFS at 75% of the target number of events
Statistical considerations
• The ITT analysis set (TIS + CT, n=226; PBO + CT, n=227) included all randomized patients
• The safety analysis set (TIS + CT, n=226; PBO + CT, n=226) included all randomized patients who received ≥1 dose of any study drug
• One-sided α at 0.005 is allocated for the MPR test; if MPR is statistically significant, 0.005 will pass to the pCR test

ALK, anaplastic lymphoma kinase; BICR, blinded independent central review; BIPR, blinded independent pathology review; CT, chemotherapy; ECOG PS, Eastern Cooperative Oncology Group performance status; EFS, event-free survival; EGFR, epidermal growth factor receptor; HRQoL, health-related quality of life; IRC, independent review committee; ITT, intent-to-treat; IV, intravenously; MPR, major pathological response; NSCLC, non-small cell lung cancer; ORR, objective response rate; OS, overall survival; PBO, placebo; pCR, pathological complete response; PD-L1, programmed death-ligand 1; PtDb, platinum-based doublet; Q3W, once every 3 weeks; Q6W, once every 6 weeks; R0, pathological complete resection of the primary tumor; TIS, tislelizumab; WT, wild type.

Results

- A total of 453 patients were randomized to receive TIS + CT or PBO + CT (**Figure 2**)
- At data cutoff (February 20, 2023), median study follow up time was 16.8 months
- In Arm A, all 226 randomized patients received TIS + CT in the neoadjuvant phase, of whom 93.4% completed treatment; 190 patients had definitive surgery and eligible patients received adjuvant TIS
- In Arm B, 226 of the 227 randomized patients received PBO + CT in the neoadjuvant phase, of whom 92.5% completed treatment; 173 patients had definitive surgery and eligible patients received PBO as adjuvant treatment
- Demographics and baseline characteristics were similar between the two arms (**Table 1**)

Figure 2. Patient Disposition

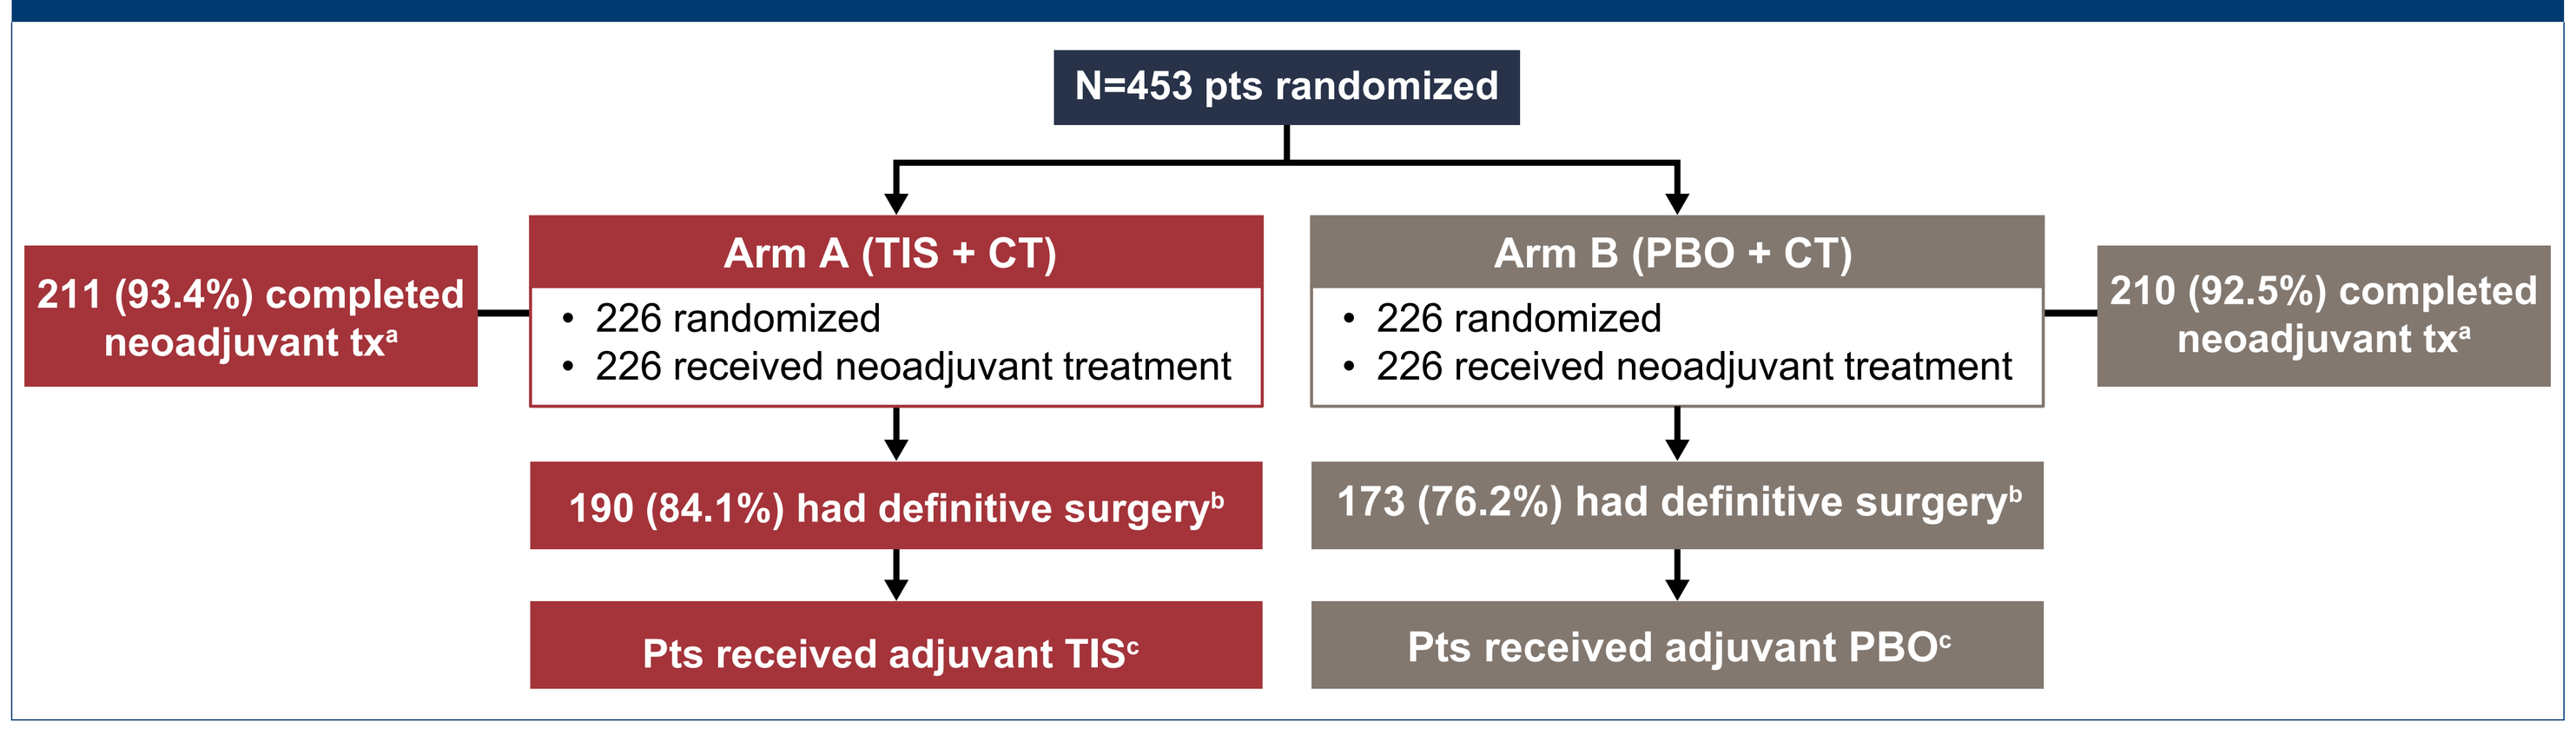
N=453 pts randomized
211 (93.4%) completed neoadjuvant tx[a]
Arm A (TIS + CT)
• 226 randomized
• 226 received neoadjuvant treatment
Arm B (PBO + CT)
• 226 randomized
• 226 received neoadjuvant treatment
210 (92.5%) completed neoadjuvant tx[a]
190 (84.1%) had definitive surgery[b]
173 (76.2%) had definitive surgery[b]
Pts received adjuvant TIS[c]
Pts received adjuvant PBO[c]

[a]Reasons for not completing neoadjuvant treatment included withdrawal by subject (TIS + CT, 2.2%; PBO + CT, 4.0%), AE (TIS + CT, 3.1%; PBO + CT, 0.9%), PD (TIS + CT, 0.9%; PBO + CT, 1.8%), and physician decision (TIS + CT, 0.4%; PBO + CT, 0.9%). [b]Denominator based on randomized patients. Reasons for cancelled surgeries included withdrawal by subject (TIS + CT, 8.8%; PBO + CT, 12.3%), PD (TIS + CT, 2.2%; PBO + CT, 5.3%), physician decision (TIS + CT, 1.8%; PBO + CT, 5.3%), AE (TIS + CT, 2.7%; PBO + CT, 0.9%), and other reasons (TIS + CT, 0.4%). [c]Not all patients who completed surgery entered the adjuvant phase.
AE, adverse event; CT, chemotherapy; PBO, placebo; PD, progressive disease; pts, patients; TIS, tislelizumab; tx, treatment.

Table 1. Demographics and Baseline Characteristics[a]

	TIS + CT (n=226)	PBO + CT (n=227)
Median age, years (range)	62.0 (30-80)	63.0 (36-78)
Sex, male	205 (90.7)	205 (90.3)
Race, Asian	226 (100.0)	227 (100.0)
ECOG performance status[b]		
0	142 (62.8)	154 (67.8)
1	83 (36.7)	73 (32.2)
Smoking status		
Current/former	192 (85.0)	188 (82.8)
Never	34 (15.0)	39 (17.2)
Histology[c]		
Squamous	179 (79.2)	175 (77.1)
Non-squamous	45 (19.9)	50 (22.0)
Disease stage[d]		
II	93 (41.2)	93 (41.0)
IIIA	132 (58.4)	132 (58.1)
PD-L1 expression[e]		
<1%	89 (39.4)	84 (37.0)
≥1%	130 (57.5)	131 (57.7)

All data are n (%) unless otherwise stated. [a]ITT analysis set. [b]One pt in the TIS + CT arm had a missing ECOG PS. [c]Histology by CRF; not shown in table: pts with mixed histology (n=2 in each arm) were categorized as "other". [d]Disease stage by CRF, per AJCC 8th edition; not shown in table: 1 pt (TIS + CT arm) and 2 pts (PBO + CT arm) had stage IIIB disease. [e]PD-L1 expression from Central Lab; excluded pts with PD-L1 results that were not evaluable/indeterminate and/or missing.
AJCC, American Joint Committee on Cancer; CRF, case report form; CT, chemotherapy; ECOG, Eastern Cooperative Oncology Group; ITT, intent-to-treat; PBO, placebo; PD-L1, programmed death-ligand 1; pts, patients; TIS, tislelizumab.

Major Pathological Response

- The MPR rate was significantly improved with TIS + CT versus PBO + CT (*P*<0.0001) in patients with resectable stage II-IIIA NSCLC; there was a 41.1% difference in MPR between the two arms (**Figure 3**)
- Improvement in MPR rate with TIS + CT compared with PBO + CT was consistent across subgroups

Figure 3. Major Pathological Response

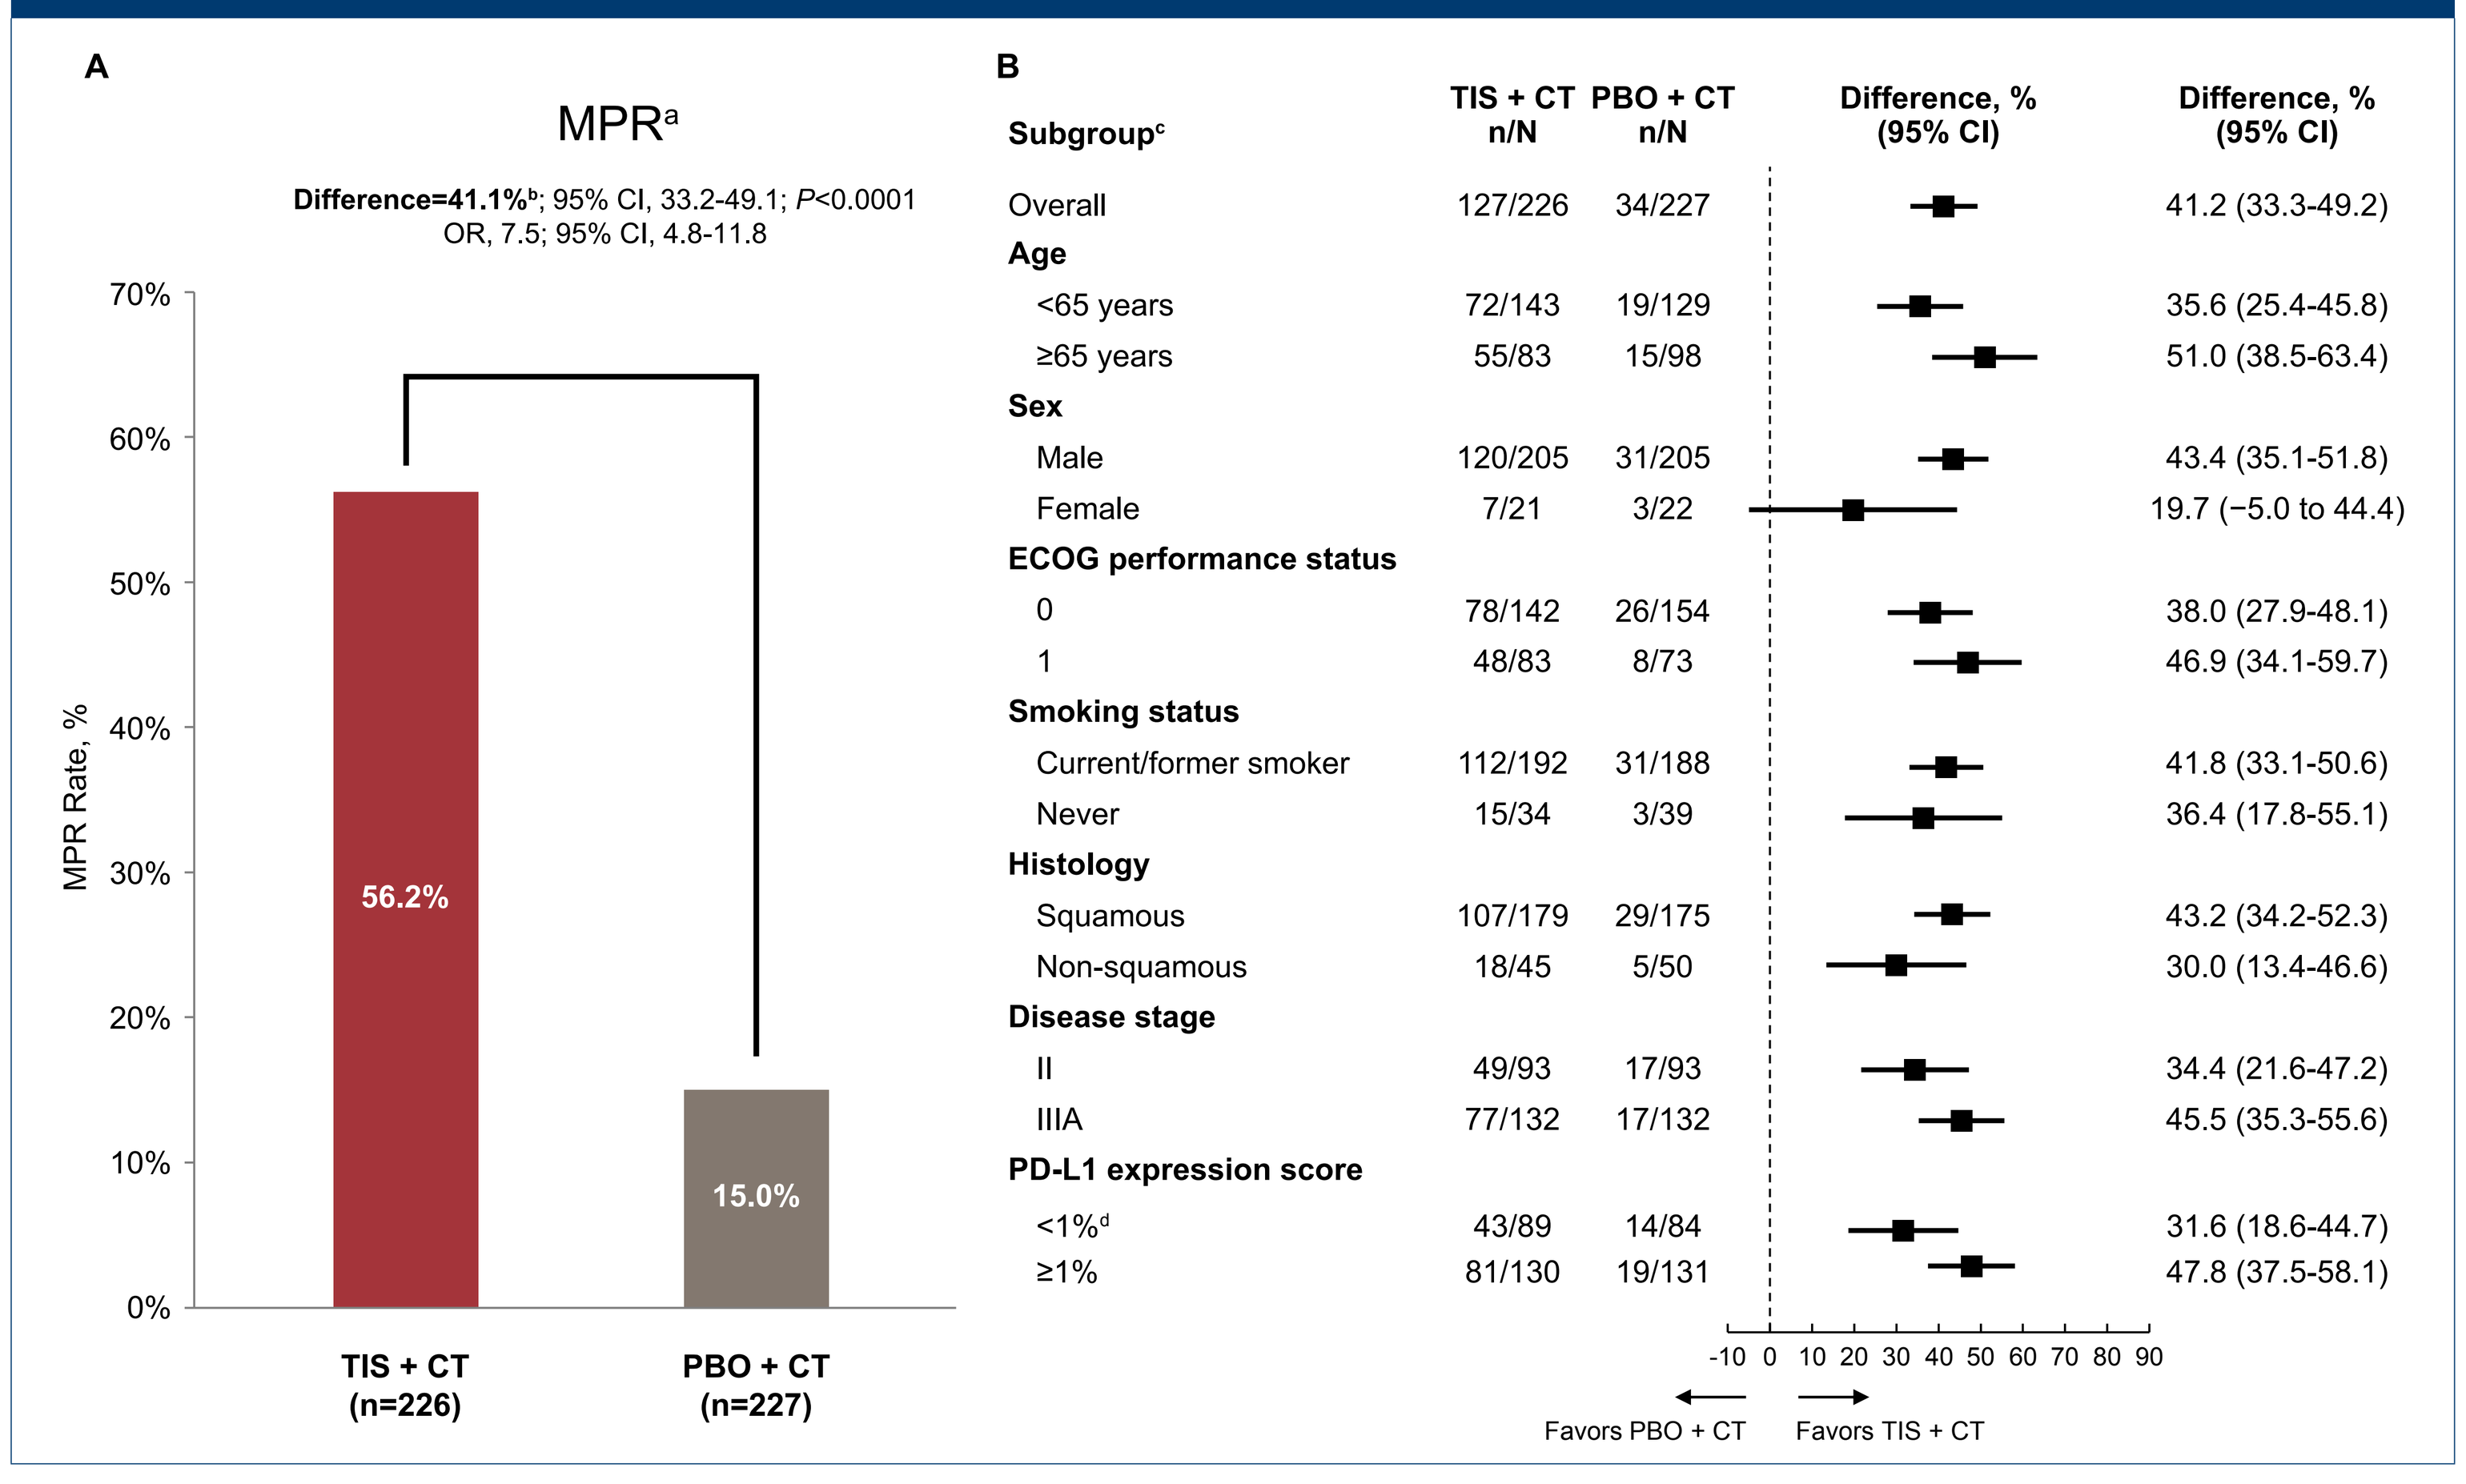
A
MPR[a]
Difference=41.1%[b]; 95% CI, 33.2-49.1; P<0.0001
OR, 7.5; 95% CI, 4.8-11.8
MPR Rate, %
0% 10% 20% 30% 40% 50% 60% 70%
56.2%
15.0%
TIS + CT (n=226)
PBO + CT (n=227)
B
Subgroup[c] | TIS + CT n/N | PBO + CT n/N | Difference, % (95% CI)
Overall 127/226 34/227 41.2 (33.3-49.2)
Age
<65 years 72/143 19/129 35.6 (25.4-45.8)
≥65 years 55/83 15/98 51.0 (38.5-63.4)
Sex
Male 120/205 31/205 43.4 (35.1-51.8)
Female 7/21 3/22 19.7 (−5.0 to 44.4)
ECOG performance status
0 78/142 26/154 38.0 (27.9-48.1)
1 48/83 8/73 46.9 (34.1-59.7)
Smoking status
Current/former smoker 112/192 31/188 41.8 (33.1-50.6)
Never 15/34 3/39 36.4 (17.8-55.1)
Histology
Squamous 107/179 29/175 43.2 (34.2-52.3)
Non-squamous 18/45 5/50 30.0 (13.4-46.6)
Disease stage
II 49/93 17/93 34.4 (21.6-47.2)
IIIA 77/132 17/132 45.5 (35.3-55.6)
PD-L1 expression score
<1%[d] 43/89 14/84 31.6 (18.6-44.7)
≥1% 81/130 19/131 47.8 (37.5-58.1)
-10 0 10 20 30 40 50 60 70 80 90
Favors PBO + CT
Favors TIS + CT

[a]MPR rate was defined as the proportion of pts with ≤10% residual viable tumor in the resected primary tumor and all resected lymph nodes after completion of neoadjuvant tx as assessed by BIPR in an ITT analysis set. Pts who did not receive surgical resection were considered as nonresponders in the analysis. MPR was compared between TIS + CT and PBO + CT using Cochran-Mantel-Haenszel chi-square test methodology. [b]Mantel-Haenszel common risk difference was estimated, along with its 95% CIs constructed by a normal approximation and Sato's variance estimator stratified by stratification factors. [c]In the subgroup analyses, risk difference and its 95% CI were estimated using the same method without stratification factors. [d]Excludes pts who were not evaluable/indeterminate.
BIPR, blinded independent pathology review; ECOG PS, Eastern Cooperative Oncology Group Performance Status; ITT, intent-to-treat; MPR, major pathological response; NSCLC, non-small cell lung cancer; OR, odds ratio; PBO, placebo; PD-L1, programmed death-ligand 1; pts, patients; TIS, tislelizumab; tx, treatment.

Pathological Complete Response

- The pCR rate was significantly improved with TIS + CT versus PBO + CT (*P*<0.0001) in patients with resectable stage II-IIIA NSCLC; there was a 35% difference in pCR between the two arms (**Figure 4**)
- Improvements in pCR rate were consistent across subgroups

Figure 4. Pathological Complete Response

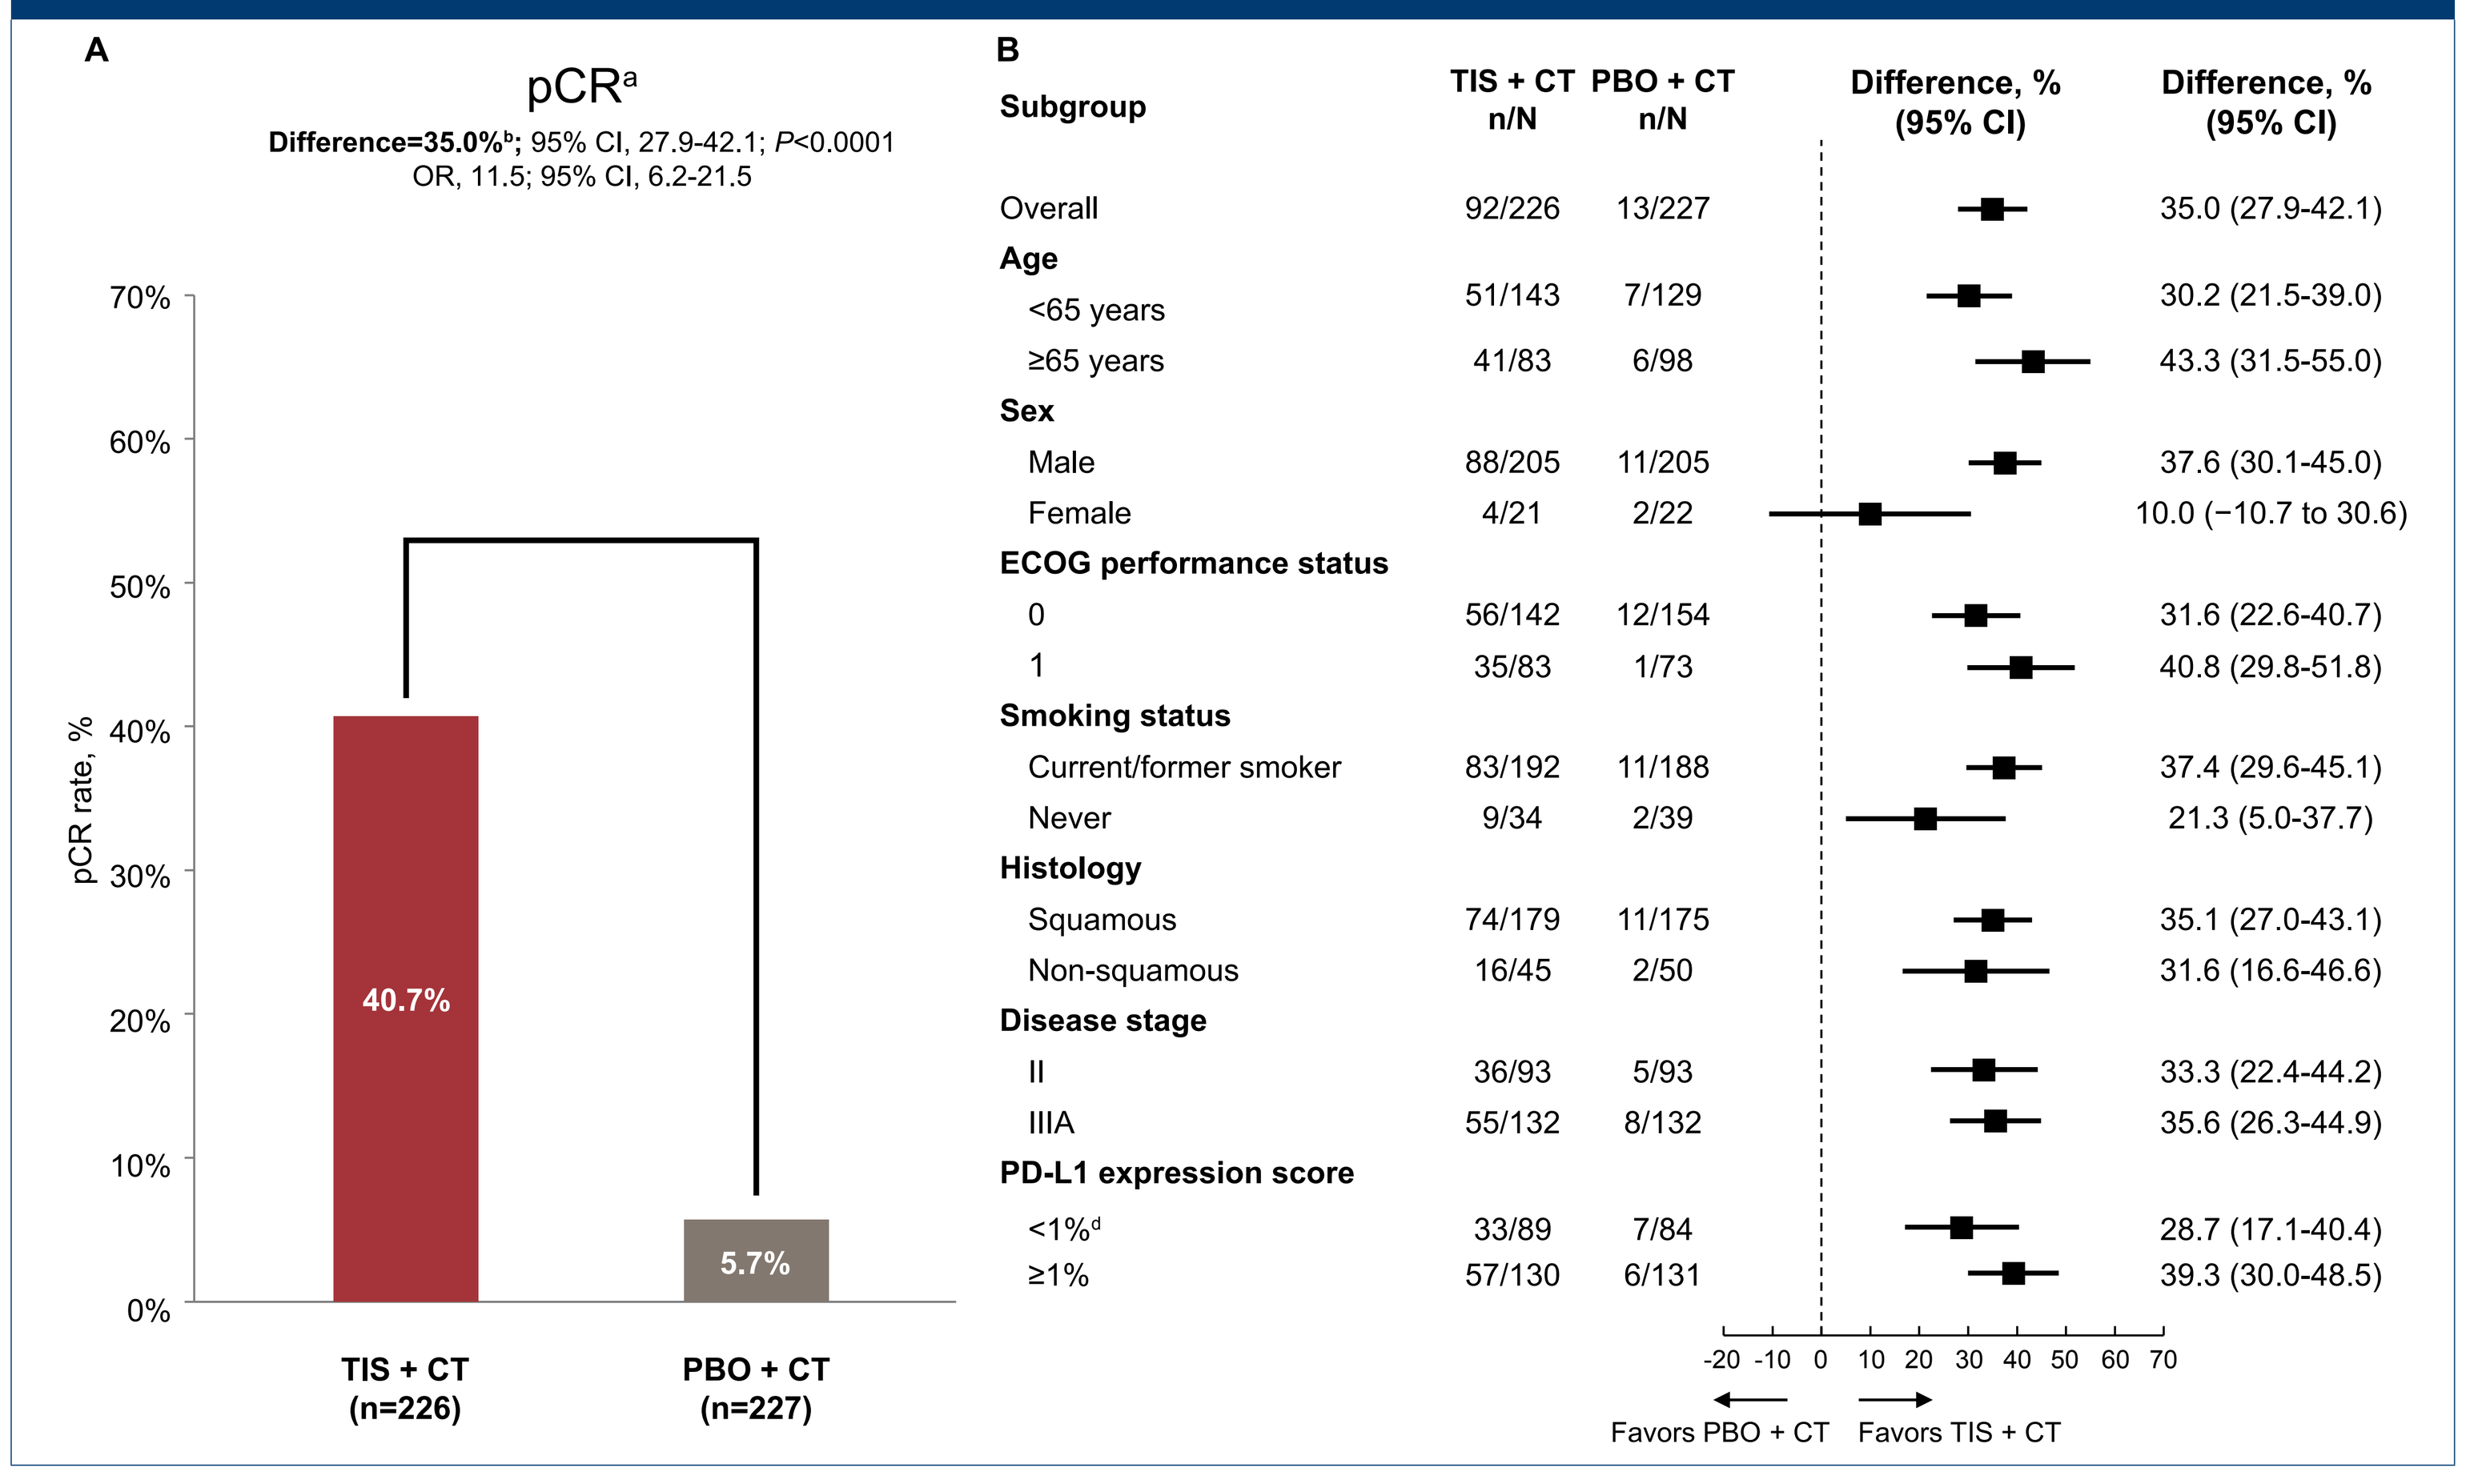
A
pCR[a]
Difference=35.0%[b]; 95% CI, 27.9-42.1; P<0.0001
OR, 11.5; 95% CI, 6.2-21.5
pCR rate, %
0% 10% 20% 30% 40% 50% 60% 70%
40.7%
5.7%
TIS + CT (n=226)
PBO + CT (n=227)
B
Subgroup | TIS + CT n/N | PBO + CT n/N | Difference, % (95% CI)
Overall 92/226 13/227 35.0 (27.9-42.1)
Age
<65 years 51/143 7/129 30.2 (21.5-39.0)
≥65 years 41/83 6/98 43.3 (31.5-55.0)
Sex
Male 88/205 11/205 37.6 (30.1-45.0)
Female 4/21 2/22 10.0 (−10.7 to 30.6)
ECOG performance status
0 56/142 12/154 31.6 (22.6-40.7)
1 35/83 1/73 40.8 (29.8-51.8)
Smoking status
Current/former smoker 83/192 11/188 37.4 (29.6-45.1)
Never 9/34 2/39 21.3 (5.0-37.7)
Histology
Squamous 74/179 11/175 35.1 (27.0-43.1)
Non-squamous 16/45 2/50 31.6 (16.6-46.6)
Disease stage
II 36/93 5/93 33.3 (22.4-44.2)
IIIA 55/132 8/132 35.6 (26.3-44.9)
PD-L1 expression score
<1%[d] 33/89 7/84 28.7 (17.1-40.4)
≥1% 57/130 6/131 39.3 (30.0-48.5)
-20 -10 0 10 20 30 40 50 60 70
Favors PBO + CT
Favors TIS + CT

[a]pCR rate was defined as the proportion of pts absent of residual viable tumor in the resected primary tumor and all resected lymph nodes after completion of neoadjuvant tx as assessed by BIPR in an ITT analysis set. Pts who do not receive surgical resection were considered as non-responders in the analysis. pCR was compared between TIS + CT and PBO + CT using Cochran-Mantel-Haenszel chi-square test methodology. [b]Mantel-Haenszel common risk difference was estimated, along with its 95% CIs constructed by a normal approximation and Sato's variance estimator stratified by stratification factors. cIn the subgroup analyses, risk difference and its 95% CI were estimated using the same method without stratification factors. [d]Excludes pts who were not evaluable/indeterminate.
BIPR, blinded independent pathology review; ECOG PS, Eastern Cooperative Oncology Group Performance Status; ITT, intent-to-treat; NSCLC, non-small cell lung cancer; nsq, nonsquamous; OR, odds ratio; PBO, placebo; pCR, pathological complete response; PD-L1, programmed death-ligand 1; pts, patients; sq, squamous; TIS, tislelizumab; tx, treatment.

Safety (Neoadjuvant Phase)

- The safety profile of TIS + CT was consistent with the known risks of each treatment and was well tolerated in patients with resectable stage II-IIIA NSCLC (**Table 2**)

Table 2. Safety (Neoadjuvant Phase)

	Study Drug Exposure	
	TIS + CT (n=226)	PBO + CT (n=226)
Median duration of treatment, weeks (range)	9.6 (1.6-18.0)	9.4 (3.0-18.1)
No. of cycles received, n (%)		
≤2	19 (8.4)	17 (7.5)
3	129 (57.1)	118 (52.2)
4	78 (34.5)	91 (40.3)
	Overall Safety Profiles[a]	
Pts with ≥1 TEAE, n (%)	224 (99.1)	225 (99.6)
Grade ≥3	157 (69.5)	148 (65.5)
Treatment-related	223 (98.7)	225 (99.6)
Serious	25 (11.1)	24 (10.6)
Related to TIS/PBO	11 (4.9)	7 (3.1)
Leading to death	3 (1.3)	0
Related to TIS/PBO	2 (0.9)	0
Leading to treatment discontinuation	20 (8.8)	19 (8.4)
TIS/PBO	7 (3.1)	2 (0.9)
Any component of CT	17 (7.5)	19 (8.4)
Leading to dose modification	70 (31.0)	69 (30.5)
TIS/PBO[b]	36 (15.9)	37 (16.4)
Any component of CT[c]	66 (29.2)	66 (29.2)

[a]The safety analysis set only included pts in the neoadjuvant phase. [b]Dose modifications for TIS/PBO included dose interruption, dose delay and infusion rate decrease. [c]Dose modifications for CT included dose reduction, dose interruption, dose delay and infusion rate decrease.
CT, chemotherapy; PBO, placebo; pts, patients; TEAE, treatment-emergent adverse event; TIS, tislelizumab.

References

1. Sung H, et al. *CA Cancer J Clin*. 2021;71(3):209-249.
2. Ganti AK, et al. *Jama Oncol*. 2021;7(12):1824-1832.
3. Ettinger DS, et al. Non-Small Cell Lung Cancer, Version 3.2023. NCCN Clinical Practice Guidelines in Oncology. 2023. nscl.pdf (nccn.org).
4. Forde PM, et al. *N Engl J Med*. 2022;386(21):1973-1985.
5. Heymach JV, et al. *Clin Lung Cancer*. 2022;23(3):e247-e251.

Acknowledgments

We would like to thank the investigators, site support staff, and especially the patients for participating in this study. This study was sponsored by BeiGene, Ltd. Medical writing support, under the direction of the authors, was provided by Apurva Davé, PhD, of Medical Expressions, an Inizio company, and was funded by BeiGene. Editorial support, under the direction of the authors, was provided by Smitha Reddy, PhD, of Envision Pharma Inc., and was funded by BeiGene. Reused with permission from the European Society for Medical Oncology (ESMO). This abstract was accepted and previously presented by Dongsheng Yue et al. at the ESMO Congress 2023, FPN (Final Publication Number): LBA58, *Annals of Oncology*, Volume 34, 2023 Supplement 2. All rights reserved.

Presenter Disclosures

Federico Cappuzzo received consulting fees, honoraria, and participated on Data Safety Monitoring Board or Advisory Board for Roche, AstraZeneca, BMS, Pfizer, Takeda, Lilly, Bayer, Amgen, Sanofi, Pharmamar, Novocure, Mirati, Galecto, OSE, ILLUMINA, Thermofisher, BeiGene, and MSD; served on Advisory Board for Roche, AstraZeneca, BMS, Pfizer, Takeda, Lilly, Bayer, Amgen, Sanofi, Pharmamar, Novocure, Mirati, Galecto, OSE, ILLUMINA, Thermofisher, BeiGene, and MSD.

Contact: f.cappuzzo@gmail.com (Federico Cappuzzo)