DP071

CELESTIAL-TNCLL: An Ongoing, Open-Label, Multiregional, Phase 3 Study of Sonrotoclax (BGB-11417) + Zanubrutinib vs Venetoclax + Obinutuzumab for Treatment-Naive CLL

Antonio Cuneo,[1] Piers E.M. Patten,[2,3] Arnon P. Kater,[4] Jennifer A. Woyach,[5] Jianyong Li,[6] Talha Munir,[7] Tommi Salmi,[8] Patrick Phuong,[9] Tian Tian,[9] Mazyar Shadman[10,11]

[1]Hematology Section, St. Anna University Hospital, Ferrara, Italy; [2]Comprehensive Cancer Centre, King's College London, London, UK; [3]King's College Hospital, London, UK; [4]Lymphoma and Myeloma Research Amsterdam, Amsterdam, the Netherlands; [5]Ohio State University Comprehensive Cancer Center, Columbus, OH, USA; [6]The First Affiliated Hospital of Nanjing Medical University, Jiangsu Province Hospital, Nanjing, China; [7]Leeds Teaching Hospitals NHS Trust, Leeds, UK; [8]BeiGene International GmbH, Basel, Switzerland; [9]BeiGene USA, Inc, San Mateo, CA, USA; [10]Fred Hutchinson Cancer Research Center, Seattle, WA, USA; [11]University of Washington, Seattle, WA, USA

INTRODUCTION

- The combination of venetoclax, the first-generation BCL2 inhibitor, and ibrutinib, a BTK inhibitor, has demonstrated efficacy in patients with CLL[1]
- However, the toxicity profile of this regimen suggests a need for a more tolerable BTK/BCL2 inhibitor combination
- Sonrotoclax, a next-generation BCL2 inhibitor, is a more selective and more pharmacologically potent inhibitor of BCL2 than venetoclax[2]
- Zanubrutinib, a next-generation BTK inhibitor, significantly improved PFS and had a more tolerable safety profile, including fewer cardiac AEs, vs ibrutinib in a randomized, head-to-head study of patients with CLL/SLL[3]
- In a phase 1 study in patients with treatment-naive (TN) CLL, treatment with sonrotoclax + zanubrutinib was generally well-tolerated, with low rates of TEAEs, and resulted in high ORRs and deep responses[4]
- Presented here is the study design of CELESTIAL-TNCLL, a phase 3 trial of sonrotoclax + zanubrutinib in TN CLL

STUDY STATUS

- Enrollment for CELESTIAL-TNCLL began in December 2023, and the study is currently recruiting
- Approximately 230 study sites in 20 countries are planned (**Figure 2**), with an estimated enrollment of 640 patients. In the Americas, there are approximately 50 sites in the US, 6 in Brazil and 15 in Canada

Figure 2. CELESTIAL-TNCLL Study Sites (Planned)

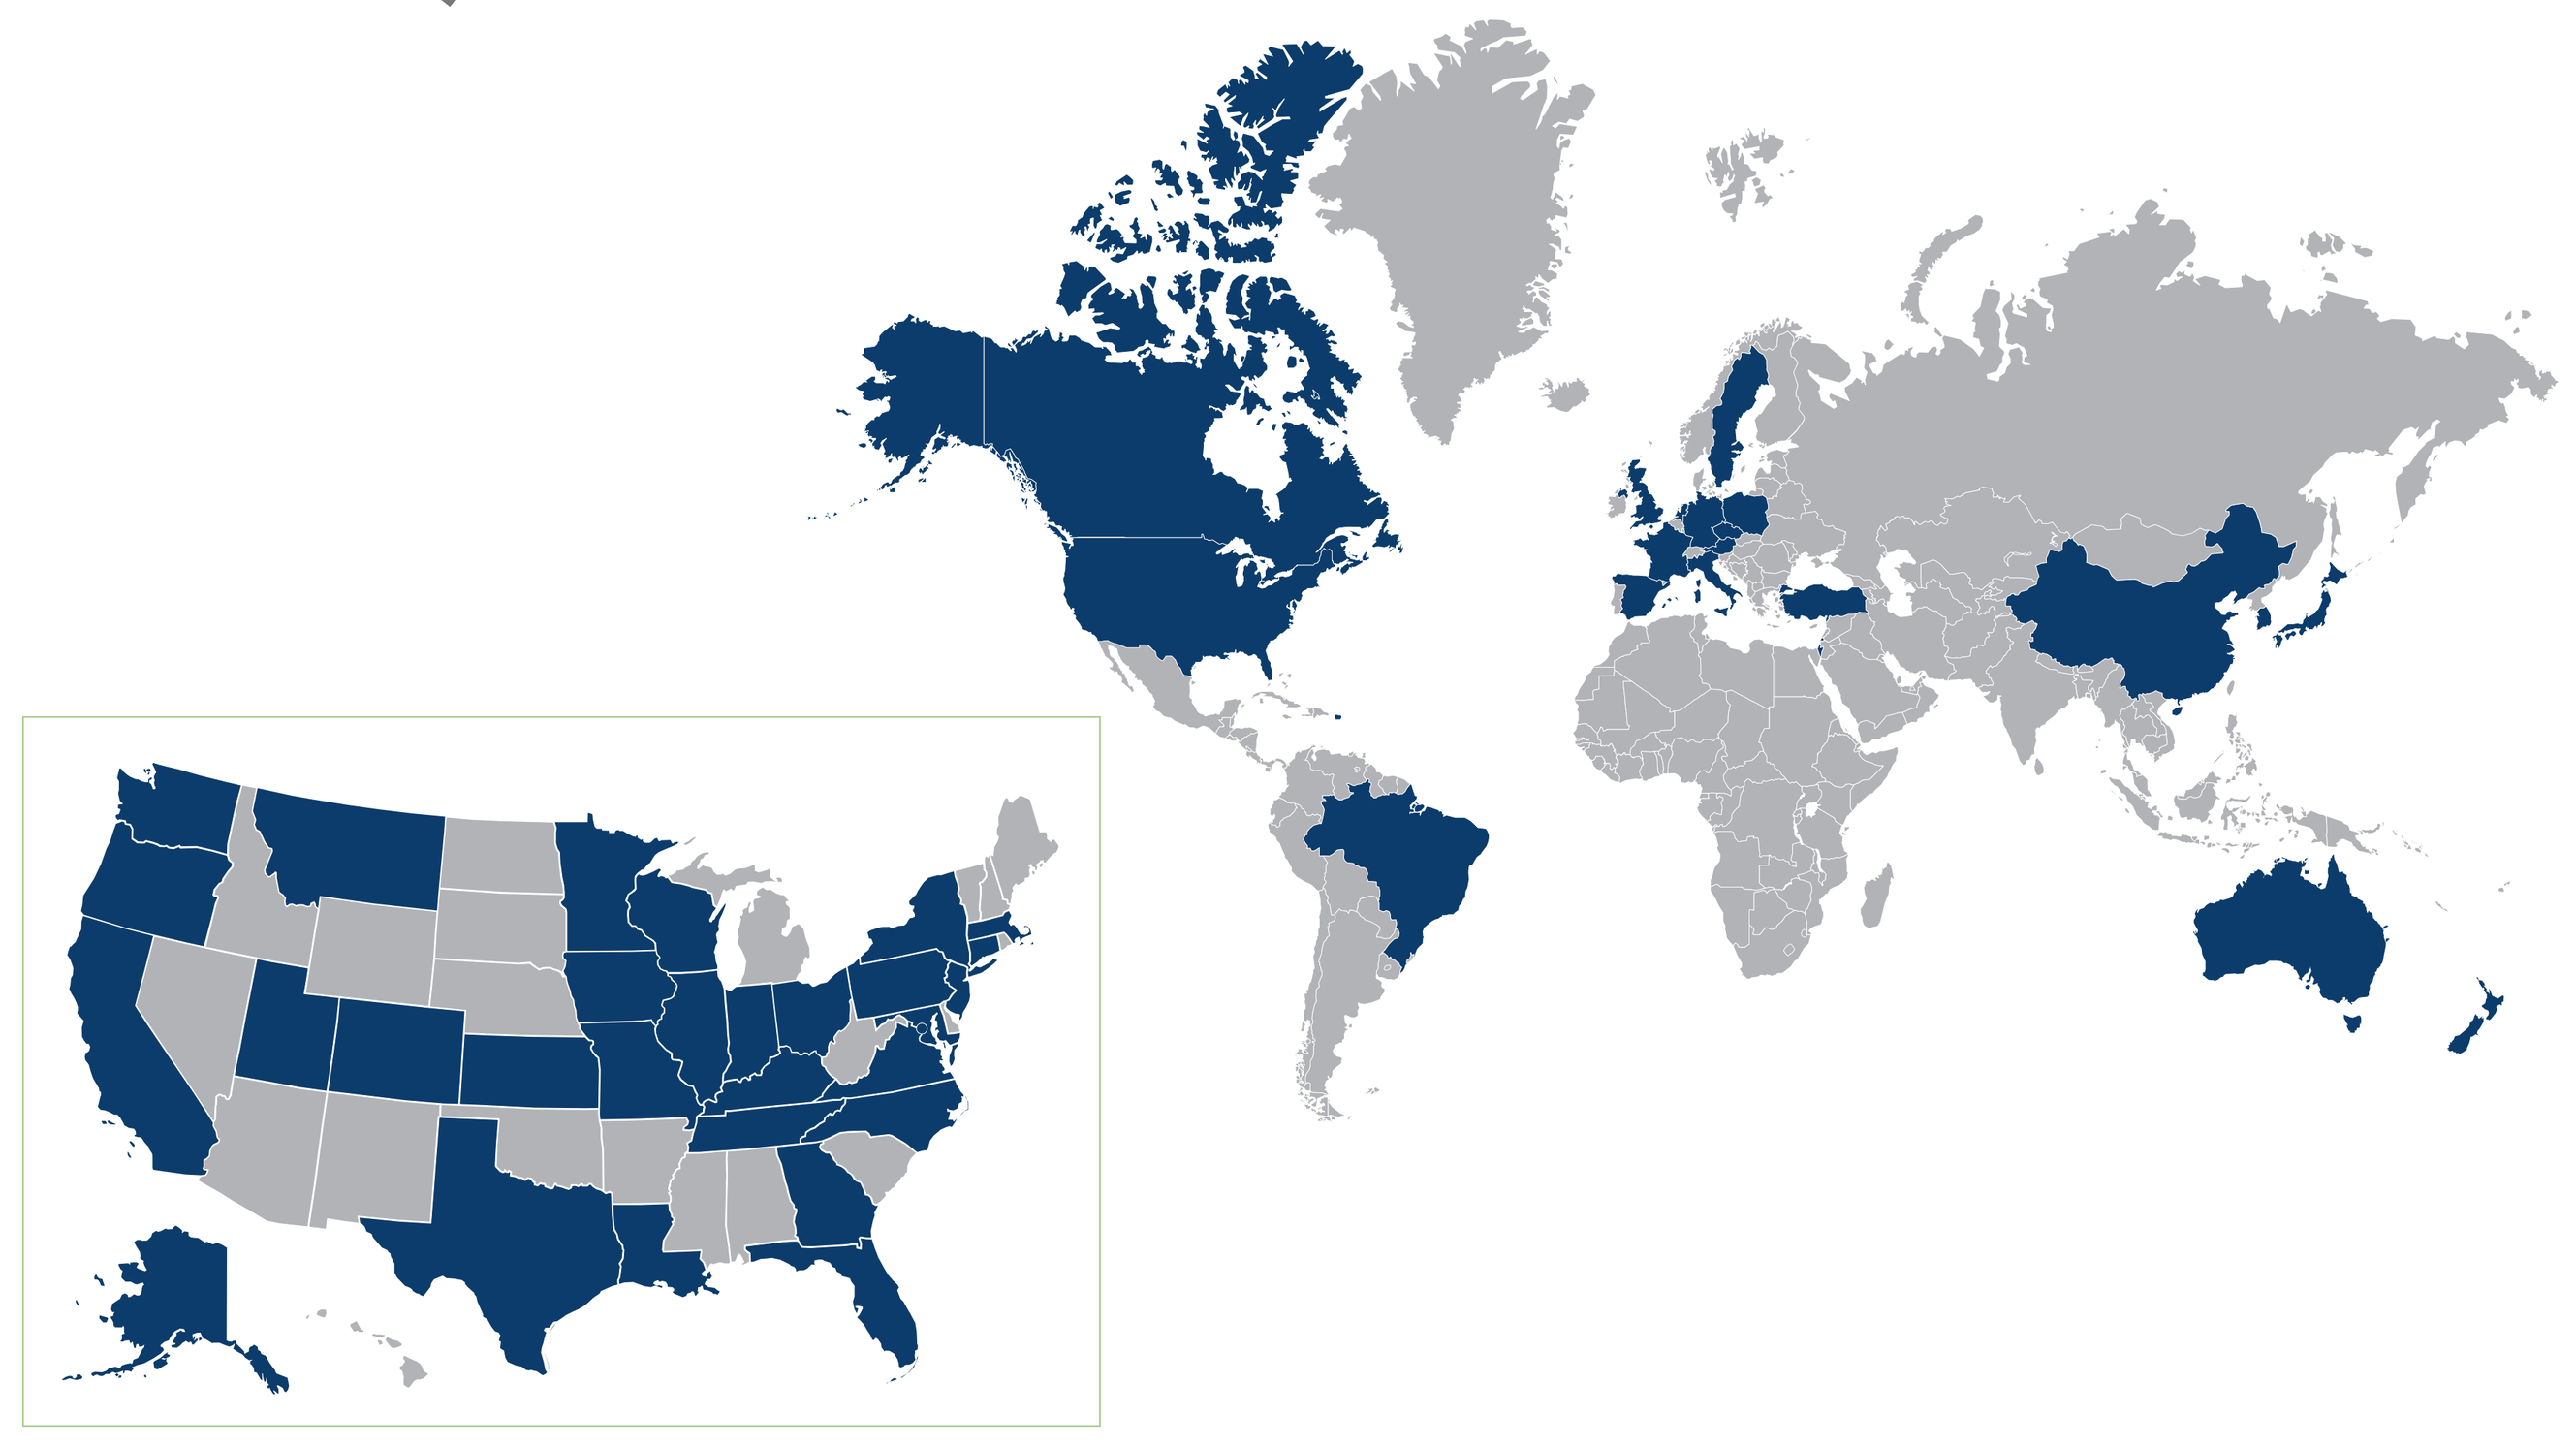

METHODS

- CELESTIAL-TNCLL (BGB-11417-301; NCT06073821) is a randomized, open-label, phase 3 study comparing the efficacy of sonrotoclax + zanubrutinib vs venetoclax + obinutuzumab in patients with TN CLL (**Figure 1**)
- Efficacy will be assessed in accordance with 2018 iwCLL guidelines[5] with modification of treatment-related lymphocytosis for patients with CLL[6]

Figure 1. CELESTIAL-TNCLL Study Design

Key Eligibility Criteria

- Age 18 years and above
- Confirmed CLL diagnosis, no previous treatment
- Measurable disease by CT/MRI
- ECOG PS of 0-2
- Adequate BM and organ function
- No history of, or currently suspected, Richter's transformation

Study Design

Previously untreated CLL (N~640) → **R** 1:1

Arm A: **Zanubrutinib lead-in (3 cycles) followed by Sonrotoclax + Zanubrutinib (12 cycles)** (n~320)

Arm B: **Venetoclax (12 cycles) + Obinutuzumab (6 cycles)** (n~320)

Randomization stratified by:
- Age (<65 vs ≥65 years)
- IGHV status
- Del(17p)/*TP53* mutation status

Endpoints

Primary
- PFS (IRC; iwCLL 2018[5])

Secondary
- CRR[a] (IRC and INV)
- Rates of uMRD4 (BM and PB)[b]
- OS
- PFS (INV)
- ORR (IRC and INV)
- DOR (IRC and INV)
- Patient-reported outcomes
- Safety and tolerability

[a] Defined as CR or CR with incomplete recovery. [b] At $<10^{-4}$ sensitivity at the first post-treatment follow-up based on next-generation sequencing by clonoSEQ® and flow cytometry.
BM, bone marrow; CRR, complete response rate; DOR, duration of response; INV, assessed by investigator; IRC, assessed by independent review committee; PB, peripheral blood; R, randomized; TN, treatment naive; uMRD4, undetectable measurable residual disease.

REFERENCES

1. Wierda WG, et al. *J Clin Oncol.* 2021;29:3853-3865.
2. Hu N, et al. AACR 2020. Abstract 3077.
3. Brown JR, et al. *N Engl J Med.* 2023;388:319-332.
4. Tam CS, et al. ASH 2023. Abstract 327.
5. Hallek M, et al. *Blood.* 2018;131:2745-2760.
6. Cheson BD, et al. *J Clin Oncol.* 2012;30:2820-2822.

DISCLOSURES

AC: Fees for participation at congresses and advisory boards: AbbVie, AstraZeneca, BeiGene, Janssen, and Lilly. **PEMP:** Research funding: AstraZeneca, AbbVie, BeiGene, Janssen, Novartis; Honoraria: AstraZeneca, AbbVie, BeiGene; Travel, accommodations, expenses: AbbVie, BeiGene, Janssen. **APK:** Research funding: Janssen, Roche, Genentech, AbbVie, AstraZeneca, BMS; Honoraria: AbbVie; Consulting role: Janssen, AbbVie, Genentech, AstraZeneca, BMS, Lava Therapeutics; Travel, accommodations, expenses: AbbVie, Janssen. **JAW:** Consulting: AbbVie, AstraZeneca, BeiGene, Genentech, LOXO@Lilly, Merck, Newave, Pharmacyclics; Research funding: AbbVie, Janssen, BeiGene, Pharmacyclics. **JL:** Nothing to disclose. **TM:** Honoraria: BeiGene, AstraZeneca, Sobi, Roche, Janssen, AbbVie, Lilly; Consulting: AbbVie, BeiGene, Sobi, Alexion, Novartis, Janssen, AstraZeneca, Lilly, Roche; Research grants: Janssen, AbbVie; Travel, accommodations, expenses: Alexion, BeiGene, AbbVie, Janssen, AstraZeneca; Advisory board: AbbVie, BeiGene, AstraZeneca, Janssen. **TS:** Employment: BeiGene Switzerland GmbH; Equity holder: BeiGene Ltd. **PP, TT:** Employment: BeiGene. **MS:** Consultancy: AbbVie, Genentech, AstraZeneca, Genmab, Janssen, BeiGene, BMS, MorphoSys/Incyte, Kite Pharma, Eli Lilly, Fate Therapeutics, Nurix, Merck; Research funding: Mustang Bio, Genentech, AbbVie, BeiGene, AstraZeneca, Genmab, Morphosys/Incyte, Vincerx; Stock options: Koi Biotherapeutics; Employment: BMS (spouse).

ACKNOWLEDGMENTS

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This study was sponsored by BeiGene, Ltd.
Medical writing support was provided by Angela R. Eder, PhD, of Nucleus Global, an Inizio company, and supported by BeiGene.

CORRESPONDENCE: Antonio Cuneo, antonio.cuneo@unife.it

Presented at the 51st SIE National Congress; September 23-25, 2024; Milano, Italy
Data originally presented at the 2024 ASCO Annual Meeting; May 31-June 4, 2024; Chicago, IL, USA. Abstract TPS7087