296

Treatment Patterns and Healthcare Resource Utilization in Advanced Esophageal Cancer Across Racial and Ethnic Groups in the Immunotherapy Era: A Retrospective Claims Data Analysis in the United States

Bassel El-Rayes,[1] Albert Craig Lockhart,[2] Wenxi Tang,[3] Rachel Delinger,[4] Dong Yuan,[3] Kaitlyn Esselman,[4] Wes Furnback,[4] Qianhong Fu,[3] Lin Zhan[3]

[1]University of Alabama at Birmingham, Birmingham, AL, USA; [2]Medical University of South Carolina, Charleston, SC, USA; [3]BeOne Medicines, Ltd, Emeryville, CA, USA; [4]Real Chemistry, New York, NY, USA

CONCLUSIONS

- Overall uptake of PD-1 inhibitors has increased but remains limited among EC patients
- No significant differences in the utilization of PD-1 inhibitors by race were observed in claims data, suggesting equitable access to effective treatments among treated populations
- Higher HCRU among minority patients, including EC-related inpatient admissions, suggests potential disparities and unmet needs and underscores the need for further research to address underlying causes and improve treatment outcomes in these patient groups

INTRODUCTION

- An estimated 22,530 patients in the United States (US) will be newly diagnosed with esophageal cancer (EC) in 2026, resulting in approximately 16,290 deaths[1]
- Immune checkpoint inhibitors, including programmed death protein 1 (PD-1) inhibitors, first received FDA approval for second-line (2L) EC in 2019 and first-line (1L) EC in 2021, and have since become the standard of care for advanced EC[2-4]
- Since the introduction of anti-PD-1 therapies, limited real-world studies have examined treatment patterns among EC patients and across racial and ethnic groups
- The objective of this study was to assess treatment patterns and healthcare resource utilization (HCRU) across racial and ethnic groups among US patients with advanced or metastatic EC receiving 1L treatment using administrative claims data

METHODS

- This was a retrospective cohort study that analyzed Symphony Integrated Dataverse (IDV®) claims data
 - Study period: July 1, 2018–June 30, 2025
 - Index period: July 1, 2019–March 31, 2025
 - The index date was defined as the date of 1L treatment initiation
 - Pre-index period: 12 months prior to the index date
 - Post-index period: 3 months post-index date
 - Continuous enrollment was required in the form of ≥1 medical or pharmacy claim in each of the four quarters during the pre-index period and ≥1 medical or pharmacy claim in each of the two quarters during the post-index period
 - Patient inclusion/exclusion criteria are outlined in **Table 1**
- Patients were stratified into sub-cohorts according to the following race/ethnicities: non-Hispanic Black (NHB), Hispanic, non-Hispanic White (NHW), Other, or Unknown
 - **Figure 1** illustrates the stepwise application of inclusion/exclusion criteria and the resulting cohort sizes
- Descriptive statistics were reported for baseline characteristics, 1L treatment distribution, duration of therapy (DoT), and treatment sequencing from 1L to 2L
 - Line of therapy information is not explicitly captured in Symphony IDV claims data, and was defined using an algorithm based on treatment regimens
 - Patients were considered discontinued if they initiated a next line of therapy, or if the gap between the line end date and the loss of continuous enrollment was >60 days; otherwise, they were censored at the earlier of the loss of continuous enrollment or the end of the study period
 - The Kaplan-Meier method was used to estimate DoT
- HCRU, including outpatient visits and inpatient admissions, were recorded for all patients and cohorts by race and reported as per-patient-per-month (PPPM)
 - HCRU claims were categorized as all-cause or EC-related. All-cause included all HCRU claims; EC-related utilization included HCRU claims with an EC-related ICD-10 code
 - Both descriptive and adjusted analyses were conducted for HCRU Poisson regression was used to assess all-cause and EC-related HCRU during 1L treatment, with incidence rate ratios (IRRs) and *P*-values reported at a 0.05 significance level

Table 1. Patient Inclusion/Exclusion Criteria

Inclusion Criteria	Exclusion Criteria
• Age ≥18 years on index date • Presence of ≥2 claims for EC (ICD-10: C15.X) on different days during the study period • Initiation of 1L treatment during the index period • Continuous enrollment during 12-month pre-index period and 3-month post-index period	• Enrollment in a in clinical trial during the study period • Presence of ≥1 claims for other primary cancer diagnoses (not including indolent cancers)

Abbreviations: 1L, first line; EC, esophageal cancer.

RESULTS

Patient Characteristics

Figure 1. Patient Selection

Patients with ≥2 claims on different days during the study period for services including any of the following ICD-10 codes: C15.3, C15.4, C15.5, C15.8, C15.9
N=210,625
↓
Initiation of a 1L EC treatment regimen during the index period (07/01/2019-03/31/2025)
N=20,699
↓
Patients ≥18 years of age at index date
N=20,699
↓
Exclude patients participating in clinical trials during study period
N=20,134
↓
Exclude patients with ≥1 claim(s) for other primary cancer diagnoses during the study period
N=7662
↓
Patients ≥1 medical OR pharmacy claim during 12-month period prior to index date
N=5266
↓
Patients ≥1 medical OR pharmacy claim during first 3-month and second 3-month period after index date
N=4371
↓
Non-Hispanic Black (N=304; 7%) | Hispanic (N=163; 4%) | Non-Hispanic White (N=2850; 65%)

Note: Two percent of patients had a classified race as "Other"; 23% of patients had missing race information.
Abbreviations: 1L, first line; EC, esophageal cancer.

- A total of 4371 patients with advanced or metastatic EC were included (**Figure 1**)
 - Among the study population, 7% were NHB (n=304), 4% were Hispanic (n=163), and 65% were NHW (n=2850)
- A majority of patients were male (79.9%) and commercially insured (59.8%); the mean age was 67.6 years (**Table 2**)
- Patients had a mean Charlson Comorbidity Index (CCI) score at index of 5.7
 - The most prevalent comorbidities at baseline were gastrointestinal disease (70.4%, eg, esophageal obstruction, diverticulosis of large intestine, constipation), cardiovascular disease (65.3%), and hypertension (56.1%)
- Hispanic and NHB patients were younger compared to NHW patients (median age: 67 vs 70) and had a slightly higher CCI score (mean: 5.9-6.5 vs 5.6; **Table 2**)

Table 2. Demographics and Clinical Characteristics During the Baseline Period

Variable	1L Cohort Overall (N=4371)	NHB (n=304)	Hispanic (n=163)	NHW (n=2850)
Sex, n (%)				
Male	3494 (79.9)	228 (75.0)	126 (77.3)	2328 (81.7)
Female	877 (20.1)	76 (25.0)	37 (22.7)	522 (18.3)
Age (at index)				
Mean	67.6 (8.7)	65.9 (8.6)	66.2 (9.3)	68.3 (8.4)
Median	69	67	67	70
Payer type, n (%)				
Medicaid	154 (3.5)	14 (4.6)	9 (5.5)	75 (2.6)
Medicare	1472 (33.7)	84 (27.6)	37 (22.7)	1035 (36.3)
Commercial	2615 (59.8)	200 (65.8)	115 (70.6)	1654 (58)
Other	58 (1.3)	2 (0.7)	1 (0.6)	42 (1.5)
Unknown	66 (1.5)	4 (1.3)	1 (0.6)	38 (1.3)
Region, n (%)				
Northeast	970 (22.2)	43 (14.1)	29 (17.8)	647 (22.7)
Midwest	1022 (23.4)	51 (16.8)	17 (10.4)	748 (26.2)
South	1522 (34.8)	200 (65.8)	51 (31.3)	993 (34.8)
West	808 (18.5)	9 (3.0)	65 (39.9)	452 (15.9)
Index year, n (%)				
2019	322 (7.4)	29 (9.5)	15 (9.2)	203 (7.1)
2020	628 (14.4)	50 (16.4)	13 (8.0)	422 (14.8)
2021	700 (16.0)	55 (18.1)	25 (15.3)	459 (16.1)
2022	843 (19.3)	58 (19.1)	40 (24.5)	543 (19.1)
2023	904 (20.7)	53 (17.4)	32 (19.6)	581 (20.4)
2024	796 (18.2)	47 (15.5)	31 (19.0)	527 (18.5)
2025	178 (4.1)	12 (3.9)	7 (4.3)	115 (4.0)
Charlson Comorbidity Index, Mean (SD)	5.7 (3.6)	6.5 (3.6)	5.9 (3.6)	5.6 (3.6)

Abbreviations: 1L, first line; NHB, non-Hispanic Black; NHW, non-Hispanic White; SD, standard deviation.

Treatment Patterns

- Systemic chemotherapy only was utilized by 61.6% of 1L-treated patients; 38.3% of patients utilized anti-PD-1 therapies (**Figure 2**)
- The utilization rate of 1L anti-PD-1 treatment, including both monotherapy and combination therapy, increased during the study period from 8.1% of treated patients in 2019 to 52.2% in 2025 (**Figure 3**)
- Anti-PD-1 use was highest among Hispanic patients (44.2%), followed by NHB (38.5%) and NHW (36.4%) patients (**Figure 2**)

Figure 2. Treatment Regimen Distribution in 1L

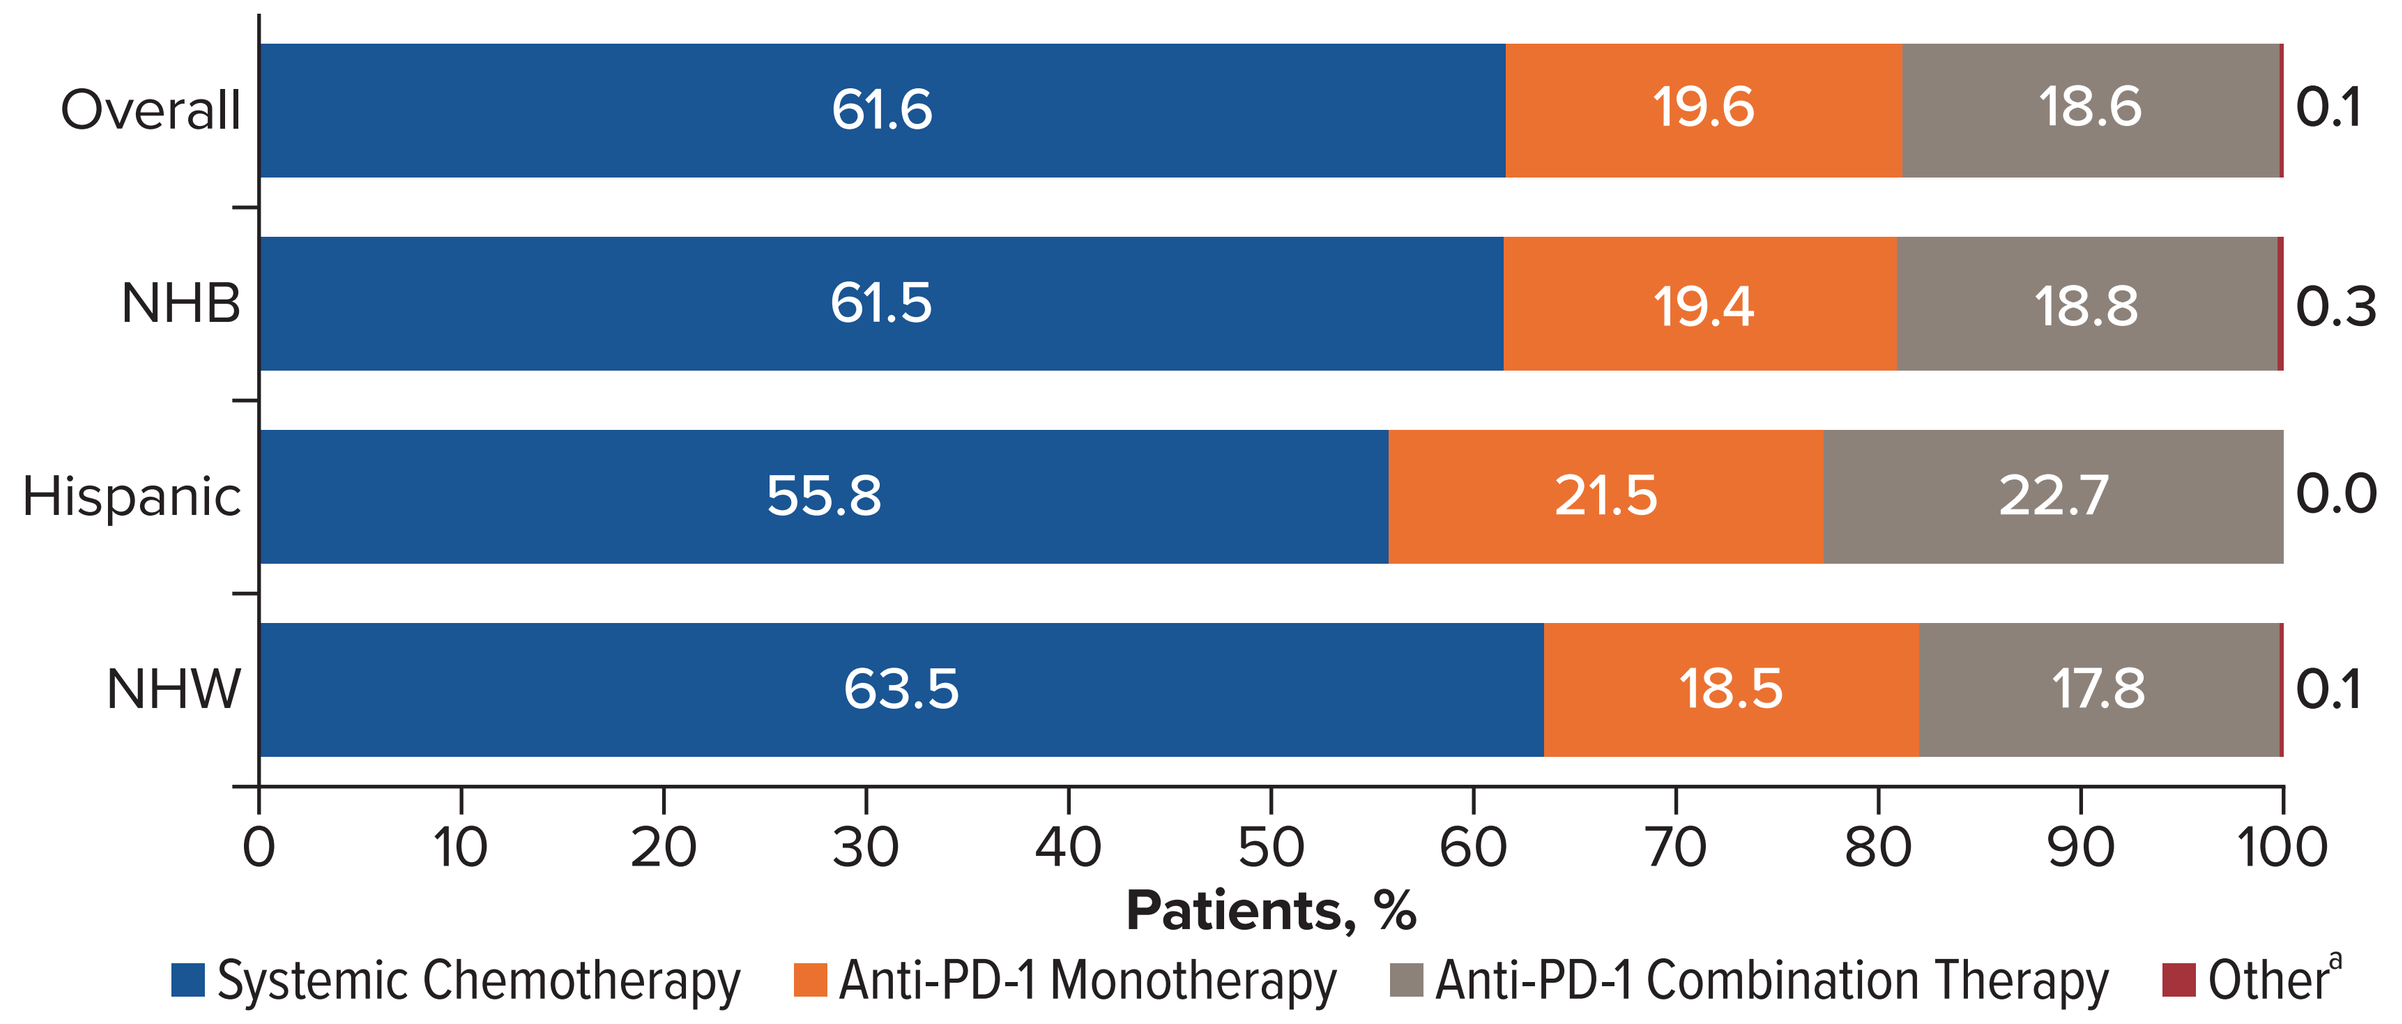
Overall
61.6
19.6
18.6
0.1
NHB
61.5
19.4
18.8
0.3
Hispanic
55.8
21.5
22.7
0.0
NHW
63.5
18.5
17.8
0.1
0
10
20
30
40
50
60
70
80
90
100
Patients, %
Systemic Chemotherapy
Anti-PD-1 Monotherapy
Anti-PD-1 Combination Therapy
Other[a]

*Other regimens include BRAF, NTRK.
Abbreviations: 1L, first line; NHB, non-Hispanic Black; NHW, non-Hispanic White; PD-1, programmed death protein 1.

Figure 3. Treatment Usage During Study Period

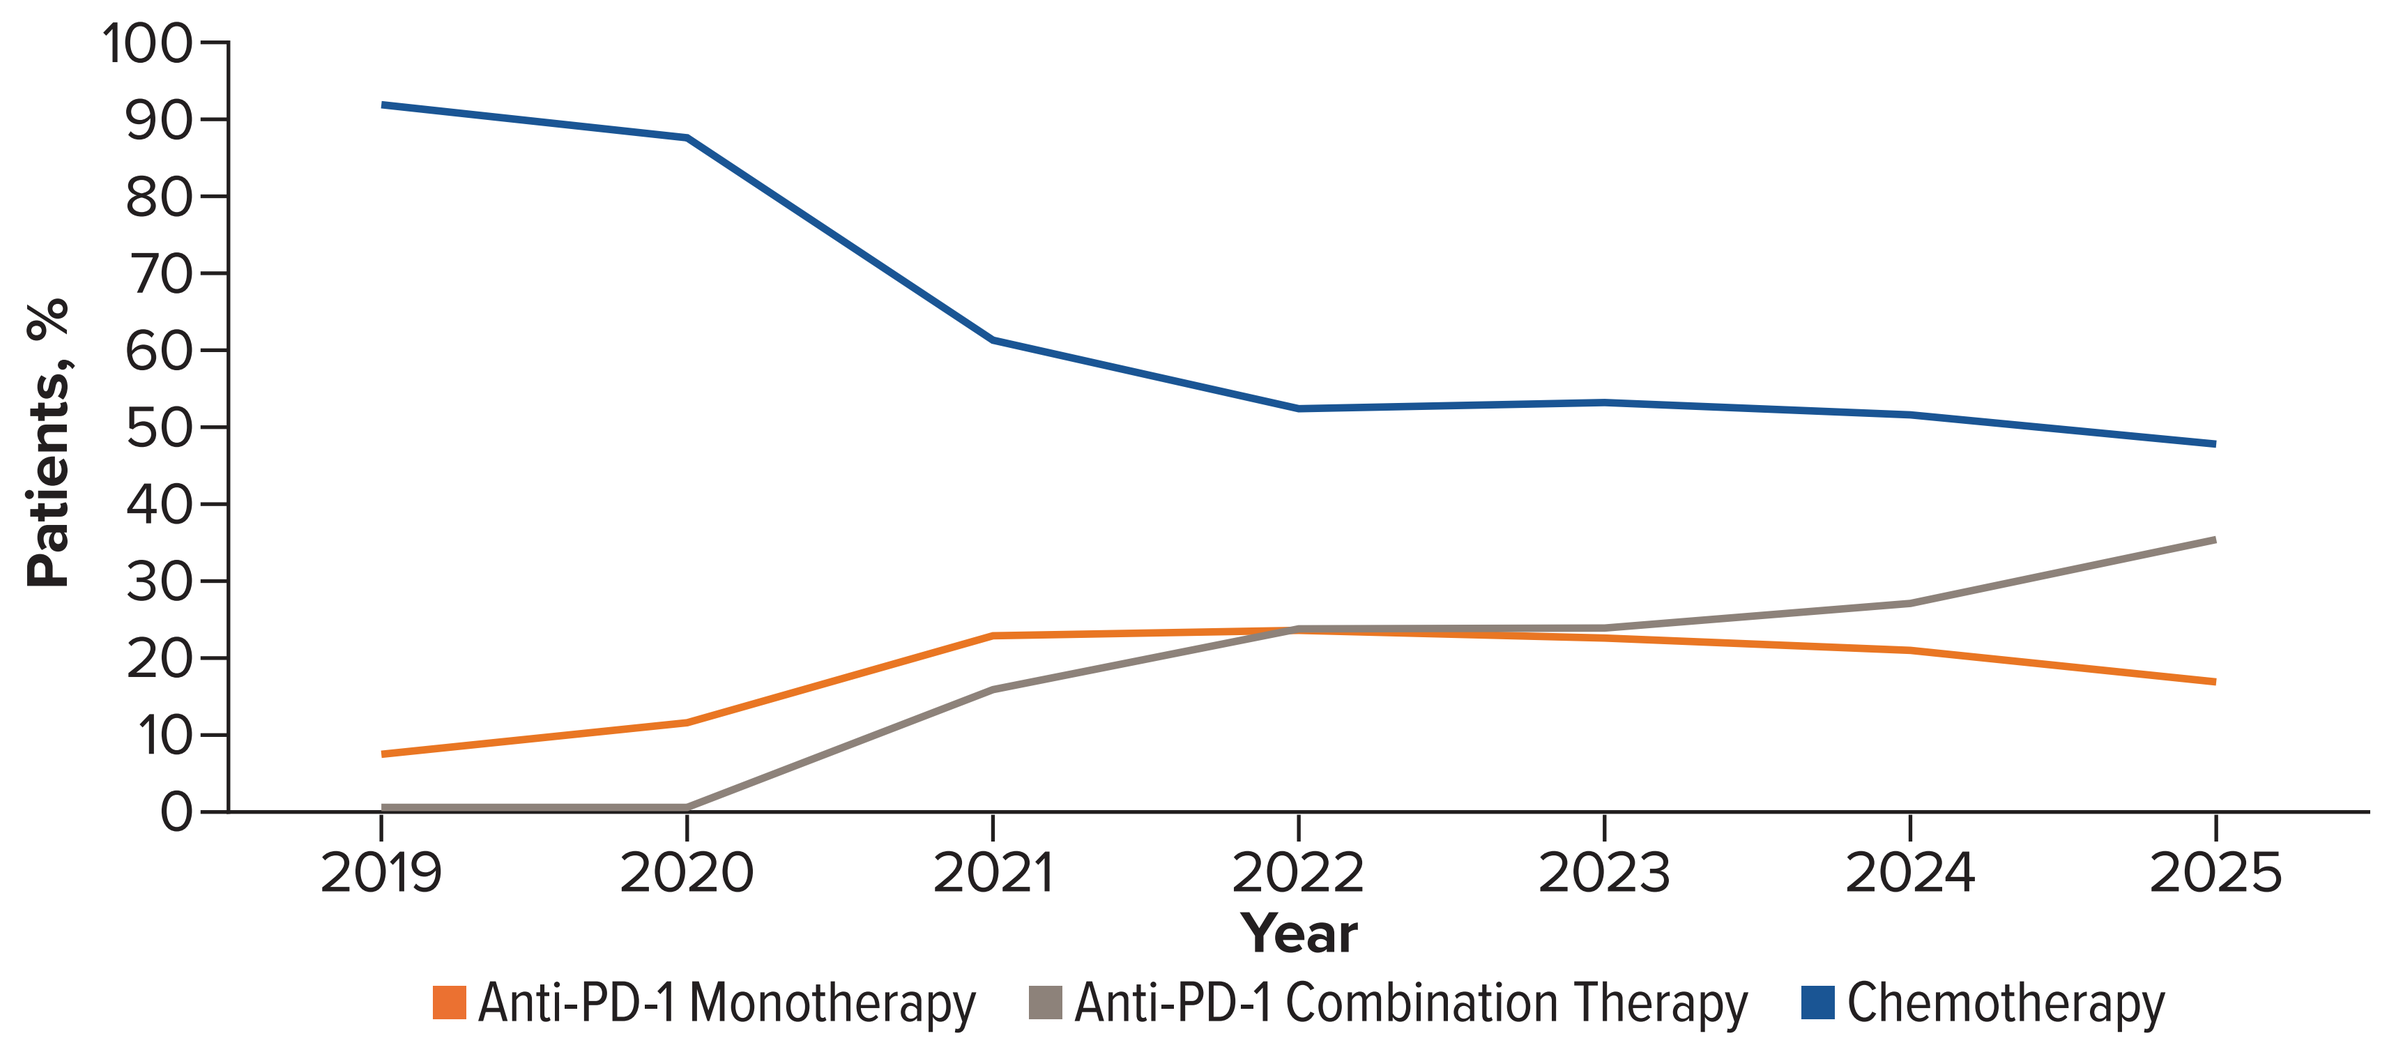
Patients, %
100
90
80
70
60
50
40
30
20
10
0
2019
2020
2021
2022
2023
2024
2025
Year
Anti-PD-1 Monotherapy
Anti-PD-1 Combination Therapy
Chemotherapy

Abbreviation: PD-1, programmed death protein 1.

- The median 1L DoT was longest among NHW patients (3.8 months or 115 days) compared to Hispanic and NHB patients (3.5 months or 106 days; **Table 3**)

Table 3. Duration of 1L Treatment by Racial Group

	Overall (N=4371)	NHB (n=304)	Hispanic (n=163)	NHW (n=2850)
Follow-up, days, mean (SD)	558.2 (464.2)	592.4 (495.2)	495.5 (407.4)	570.8 (476.7)
DoT, days median (95% CI)	120 (113.0-126.0)	106.0 (92.0-130.0)	106.0 (82.0-126.0)	115.0 (106.0-122.0)

Abbreviations: 1L, first line; CI, confidence interval; DoT, duration of therapy; NHB, non-Hispanic Black; NHW, non-Hispanic White; SD, standard deviation.

- The median 1L DoT was longer among patients who initiated anti-PD-1 monotherapy (7.9 months or 239 days) and anti-PD-1 combination therapies (7.4 months or 225 days) compared to those who were treated with systemic chemotherapy (2.3 months or 70 days; **Table 4**)

Table 4. Duration of 1L Treatment by Index Treatment Regimen

	Systemic Chemotherapy (n=2694)	Anti-PD-1 Monotherapy (n=857)	Anti-PD-1 Combination (n=813)
Follow-up, days, mean (SD)	599.7 (507.6)	578.7 (417.2)	400.7 (300.3)
DoT, days median (95% CI)	70.0 (65.0-72.0)	239.0 (218.0-260.0)	225.0 (207.0-246.0)

Abbreviations: 1L, first line; CI, confidence interval; DoT, duration of therapy; PD-1, programmed death protein 1; SD, standard deviation.

- Among all treated 1L EC patients, 24.0% received 2L treatment (**Figure 4**)
 - Among patients treated with systemic chemotherapy in 1L, 26.5% received 2L treatment; among those treated with anti-PD-1 therapies, 20.3% received 2L treatment
- Among patients who initiated chemotherapy in 1L, 8.4% switched to PD-1 treatment in 2L

Figure 4. Class-level Sankey Diagram

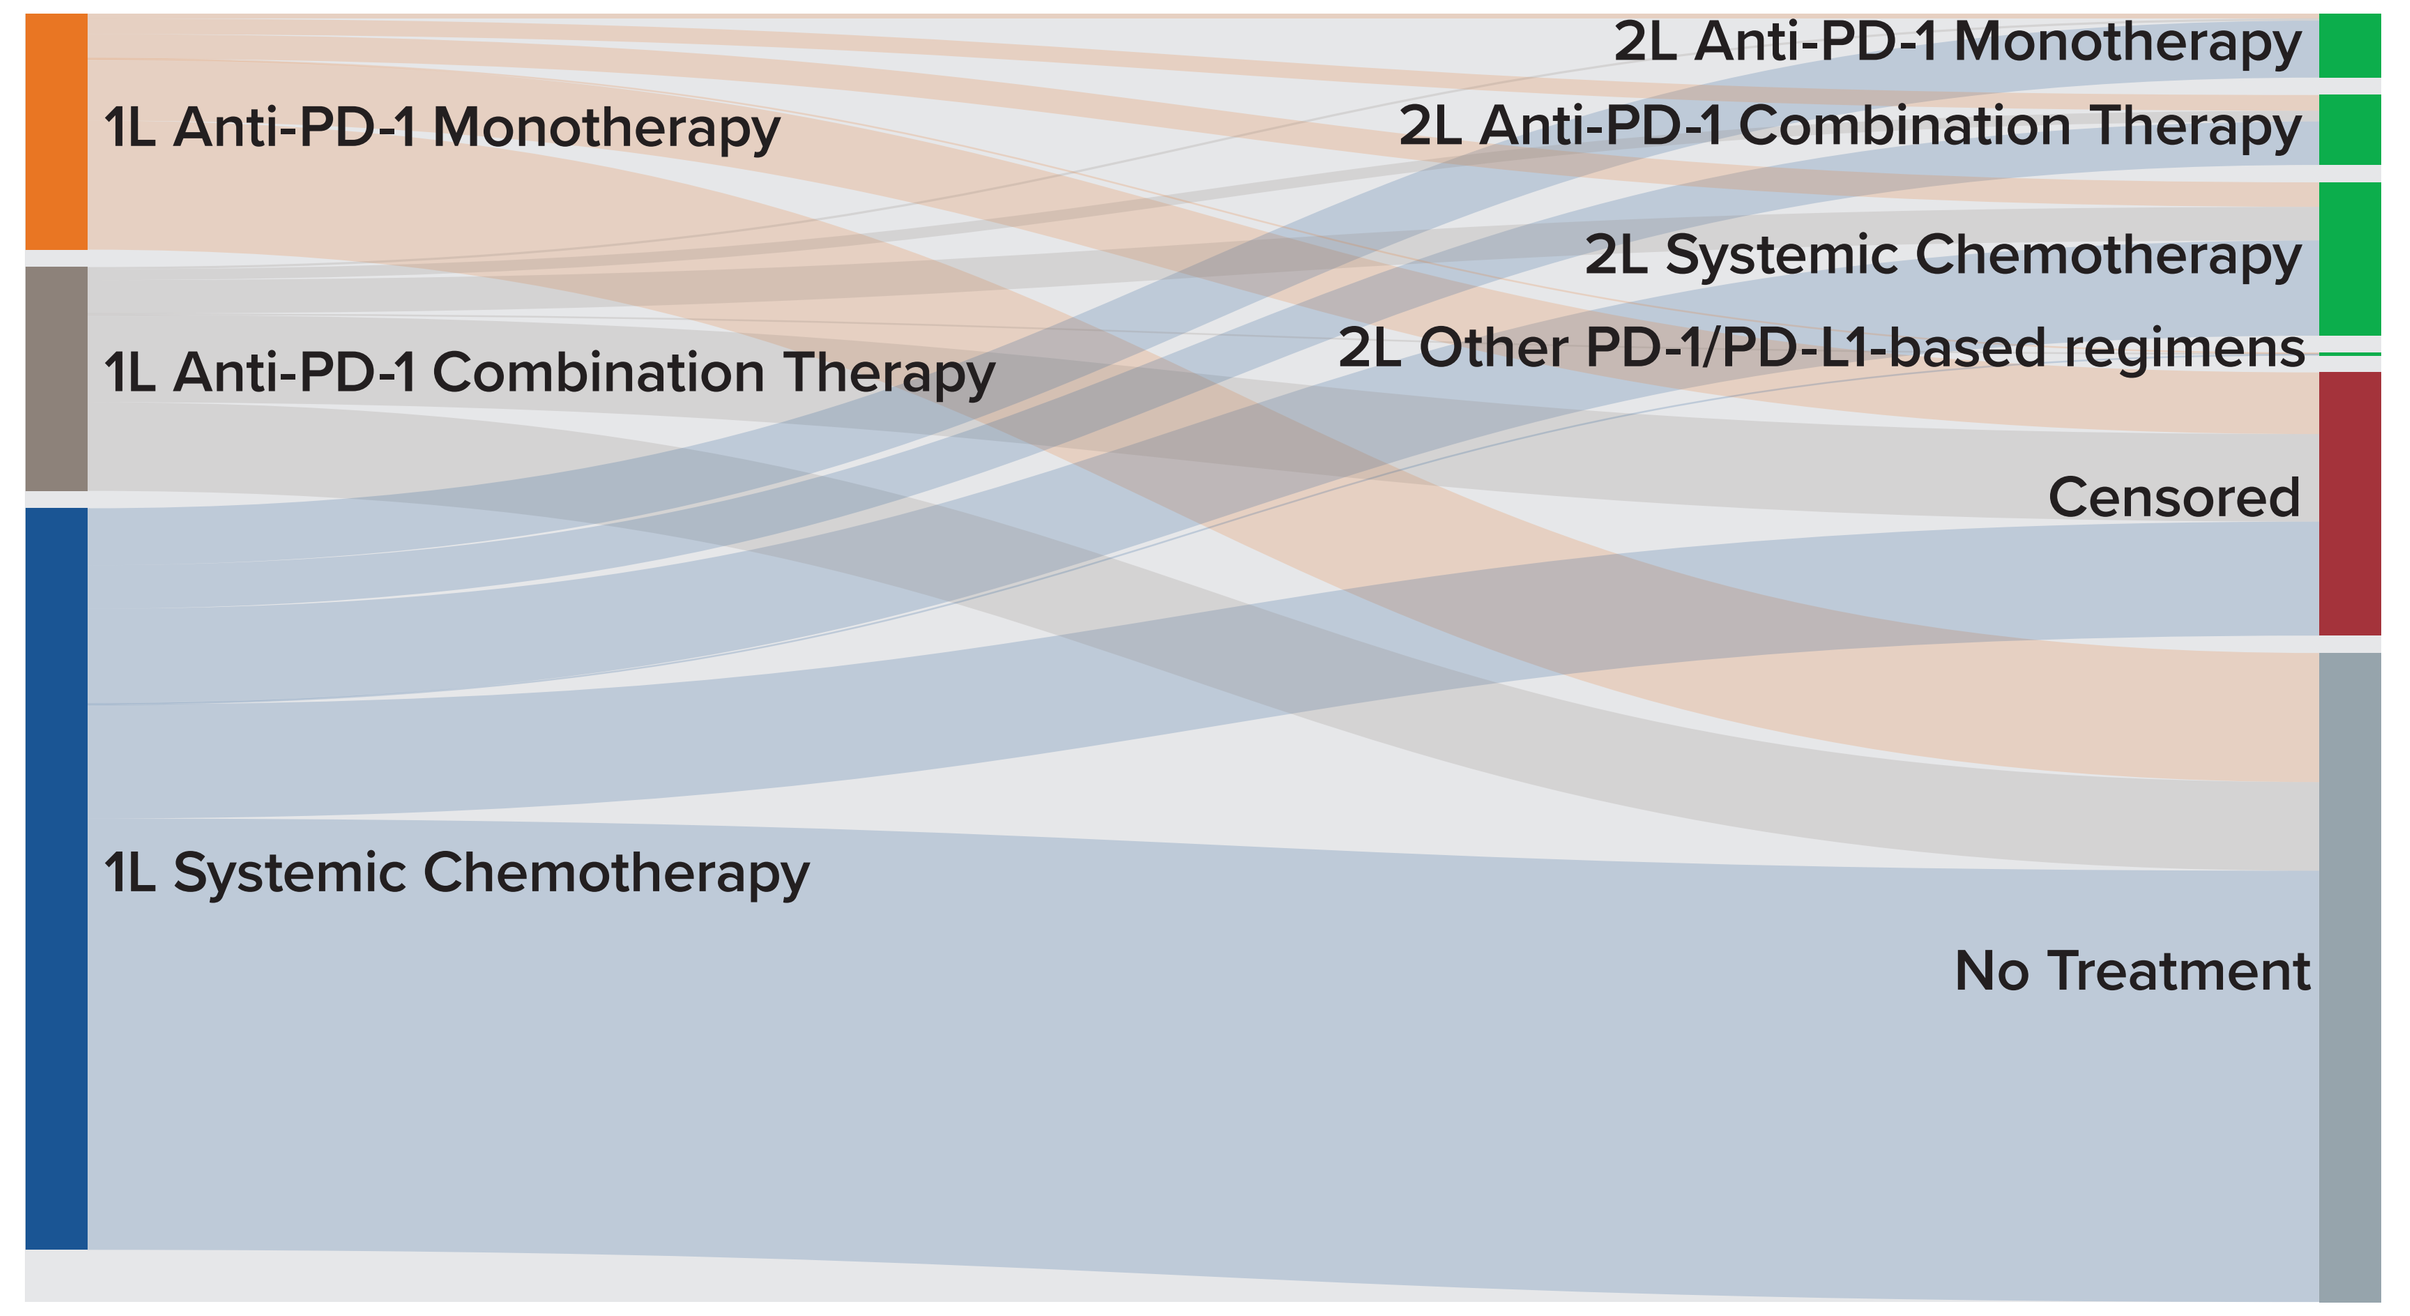
1L Anti-PD-1 Monotherapy
1L Anti-PD-1 Combination Therapy
1L Systemic Chemotherapy
2L Anti-PD-1 Monotherapy
2L Anti-PD-1 Combination Therapy
2L Systemic Chemotherapy
2L Other PD-1/PD-L1-based regimens
Censored
No Treatment

Abbreviations: 1L, first line; 2L, second line; PD-1, programmed death protein 1.

Healthcare Resource Utilization

- During 1L treatment, 27% of NHB patients had ≥1 EC-related inpatient admission, compared to 26% of Hispanic and 22% of NHW patients
- During 1L treatment, NHB patients (PPPM: 2.05 vs 1.50; adjusted IRR: 1.06; *P*=0.0007) and Hispanic patients (PPPM: 1.93 vs 1.50; adjusted IRR: 1.16; *P*<0.0001) had significantly higher EC-related outpatient visits compared to NHW patients (**Figure 5A**)
- NHB patients (PPPM: 0.08 vs 0.06; adjusted IRR: 1.19; *P*=0.025) and Hispanic patients (PPPM: 0.08 vs 0.06; adjusted IRR: 1.44; *P*=0.0004) had significantly higher EC-related inpatient admissions compared to NHW patients (**Figure 5B**)

Figure 5. Outpatient Versus Inpatient HCRU in 1L Treatment by Race

A. Outpatient Visits

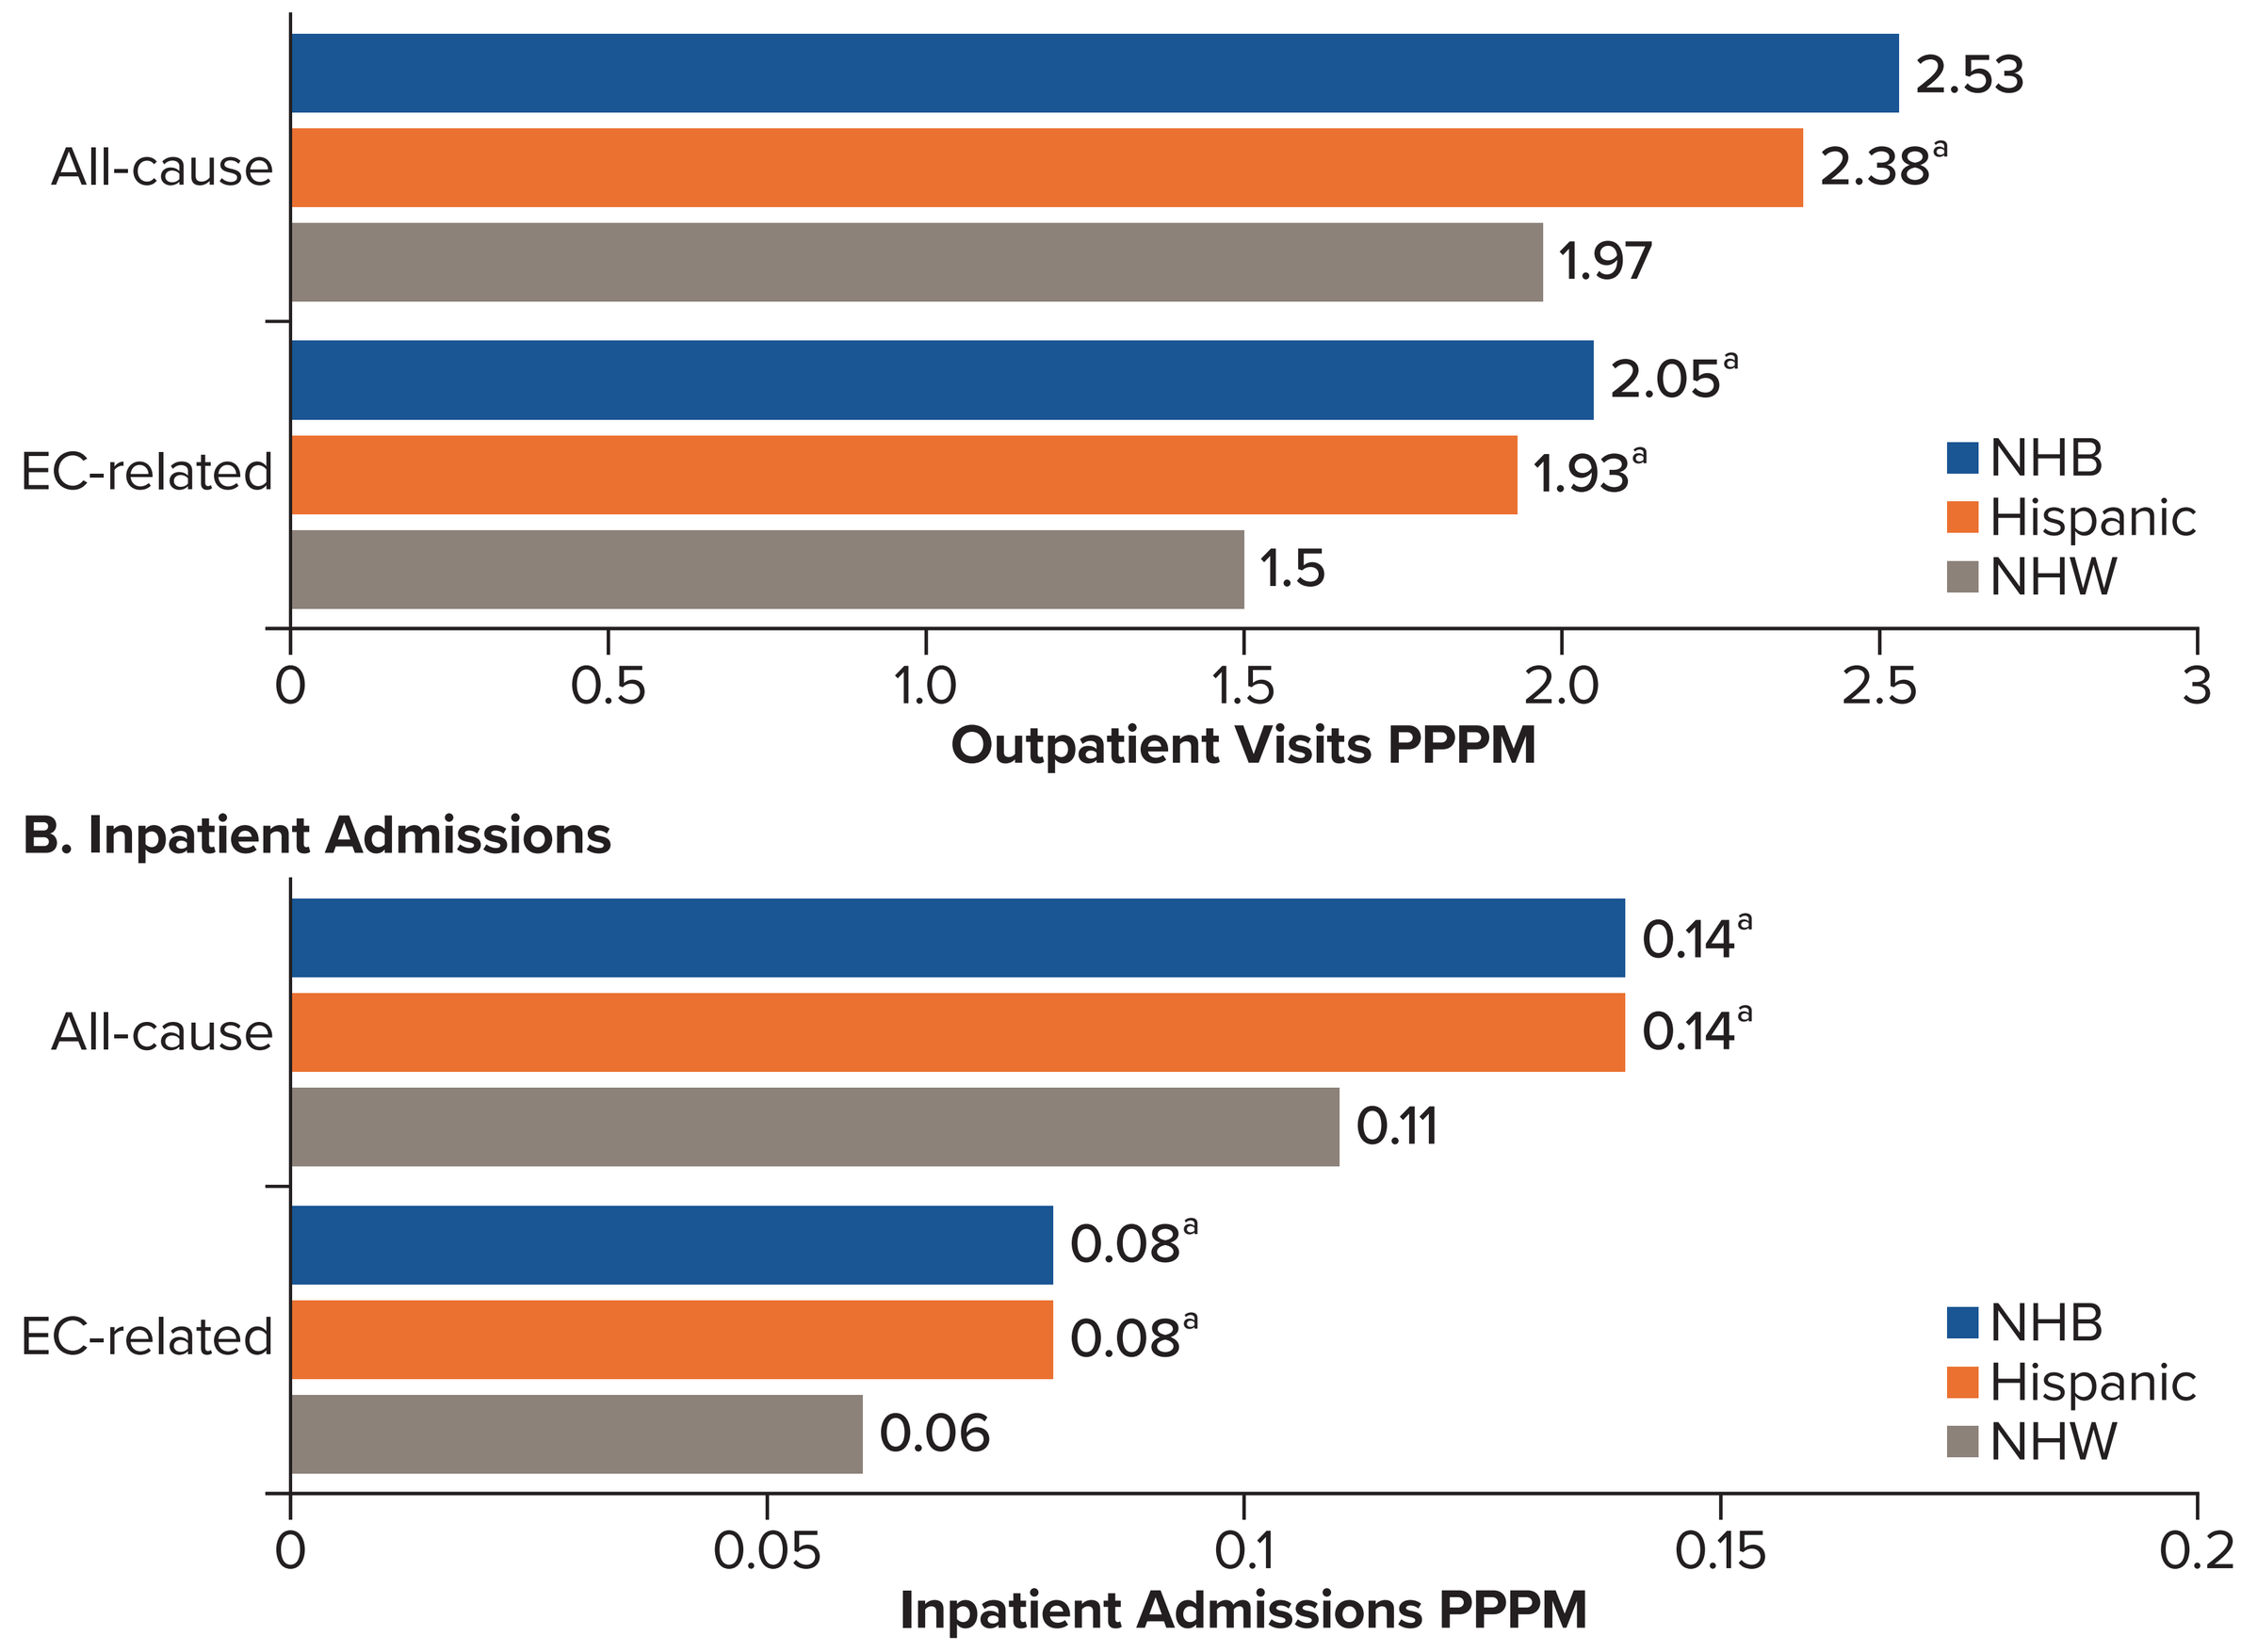
All-cause
2.53
2.38[a]
1.97
EC-related
2.05[a]
1.93[a]
1.5
NHB
Hispanic
NHW
0
0.5
1.0
1.5
2.0
2.5
3
Outpatient Visits PPPM
B. Inpatient Admissions
All-cause
0.14[a]
0.14[a]
0.11
EC-related
0.08[a]
0.08[a]
0.06
NHB
Hispanic
NHW
0
0.05
0.1
0.15
0.2
Inpatient Admissions PPPM

[a]Denotes statistically significant *P*-value compared to NHW patients after adjusting for age, sex, comorbidity, and 1L treatment.
Abbreviations: 1L, first line; EC, esophageal cancer; HCRU healthcare resource utilization; NHB, non-Hispanic Black; NHW, non-Hispanic White, PPPM, per-patient per-month.

LIMITATIONS

- This analysis was based on retrospective real-world data, which may be subject to coding errors and incomplete or miscategorized capture of clinical information
- Esophageal adenocarcinoma and esophageal squamous cell carcinoma cannot be distinguished using ICD-10 codes, though they are different histological cancer types and may differ in epidemiology and treatment response
- The Symphony IDV database lacks detailed clinical variables such as death/progression (time to next treatment or survival outcomes were not summarized due to data limitation), disease stage at diagnosis, tumor biomarkers (eg, PD-L1 expression), and performance status
- Treatment intent and reasons for therapy selection or discontinuation could not be determined
- Results may not be generalizable to populations outside the dataset or to uninsured patients

REFERENCES

1. Shiravand, et al. Immune checkpoint inhibitors in cancer therapy. *Curr Oncol.* 2022;29(5):3044-3060.
2. KEYTRUDA® (pembrolizumab). Prescribing information. Merck & Co; 2025.
3. OPDIVO® (nivolumab). Prescribing information. Bristol Myers Squibb; 2025.
4. TEVIMBRA® (tislelizumab-jsgr). Prescribing information. BeOne Medicines, Ltd; 2025.

DISCLOSURES

BE: received consulting fees from BeOne Medicines, Ltd. **ACL:** reports grants or contracts from AbbVie and Apollo Therapeutics; received consulting fees from BeOne Medicines, Ltd; received payment or honoraria from BeOne Medicines, Ltd and Ipsen; received payment for expert testimony from BeOne Medicines, Ltd; and participated on a data safety monitoring board or advisory board as a DSMC member for Revolution Medicines. **WT, DY, QF, LZ:** employees and equity holders in BeOne Medicines, Ltd. **RD, KE, WF:** are employees of Real Chemistry, which received consulting fees from BeOne Medicines, Ltd.

ACKNOWLEDGMENTS

This study is sponsored by BeOne Medicines, Ltd.

CORRESPONDENCE: Wenxi Tang, wenxi.tang@beonemed.com

Presented at the 2026 Academy of Managaed Care Pharmacy Annual Meeting; April 13–16, 2026; Nashville, TN