Sitravatinib + tislelizumab in patients with anti-PD-(L)1 refractory/resistant metastatic non-small cell lung cancer

Bo Gao*,[1] Zhiyong Ma,[2] Xinmin Yu,[3] Dingzhi Huang,[4] Jun Zhao,[5] Daphne Day,[6] Amy Louise Body,[6] Qing Zhou,[7] Qian Chu,[8] Hongming Pan,[9] Jiuwei Cui,[10] Hui Li,[11] Jingchao Sun,[11] Juan Zhang,[11] Cong Fei,[11] Yi-Long Wu[12]

[1]Blacktown Cancer and Hematology Centre, Blacktown, NSW, Australia; [2]The Affiliated Cancer Hospital of Zhengzhou University/Henan Cancer Hospital, Zhengzhou, China; [3]Department of Medical Oncology, Cancer Hospital of University of Chinese Academy of Sciences & Zhejiang Cancer Hospital; [4]Tianjin Medical University Cancer Institute and Hospital, Tianjin Key Laboratory of Cancer Prevention and Therapy, National Clinical Research Center for Cancer; [5]Key Laboratory of Carcinogenesis and Translational Research (Ministry of Education, Beijing), Department of Thoracic Medical Oncology, Peking University Cancer Hospital & Institute, Beijing, China; [6]Monash Health and Monash University, Melbourne, Australia; [7]Guangdong Lung Cancer Institute, Guangdong Provincial People's Hospital and Guangdong Academy of Medical Sciences Guangdong, South China University of Technology, Guangzhou, China; [8]Department of Oncology, Tongji Hospital of Tongji Medical College, Huazhong University of Science and Technology; [9]Sir Run Run Shaw Hospital, Zhejiang University, Hangzhou, China; [10]The First Hospital of Jilin University, Changchun, China; [11]BeiGene (Beijing) Co., Ltd., Beijing, China; [12]Guangdong Provincial Key Laboratory of Translational Medicine in Lung Cancer, School of Medicine, Guangdong Lung Cancer Institute, Guangdong Provincial People's Hospital and Guangdong Academy of Medical Sciences, South China University of Technology, Guangzhou 510080, China

*Author contact details: Bo.Gao@health.nsw.gov.au

Poster No: 210

Introduction

- Programmed death protein (ligand)-1 (PD-[L]1) inhibitors are effective first-line treatments for advanced non-small cell lung cancer (NSCLC).[1] Despite this, many patients ultimately relapse and treatment options are limited for patients with metastatic NSCLC that is refractory/resistant (R/R) to anti-PD-(L)1 therapies[2,3]
- Tislelizumab is an anti-PD-1 antibody designed to minimize binding to FcγR on macrophages in order to abrogate antibody-dependent phagocytosis, a mechanism of T cell clearance and anti-PD-1 resistance[4,5]
- Sitravatinib is an oral spectrum-selective tyrosine kinase inhibitor targeting TAM (TYRO3, AXL, MER) receptors and split tyrosine-kinase domain-containing receptors (VEGFR2, KIT) that can reduce the number of myeloid-derived suppressor cells, regulatory T cells, increase the ratio of M1/M2 polarized macrophages, and may augment antitumor immune responses[6]
- Combining a PD-1 inhibitor and an agent with immune modulatory and antitumor properties may enhance antitumor activity beyond that provided by either agent alone[7]
- Sitravatinib in combination with tislelizumab is currently being investigated in several solid tumor types (NCT03666143)
 - We report safety, tolerability, and antitumor activity results for cohorts with squamous or non-squamous metastatic NSCLC that is R/R to anti-PD-(L)1 therapy

Methods

- An open-label, multicenter, non-randomized, multi-cohort, Phase 1b trial was conducted (NCT03666143)
- Study design and endpoints are summarized in **Figure 1**
- Cohorts reported herein included patients with non-squamous (cohort A) or squamous (cohort F) metastatic NSCLC that is R/R to anti-PD-(L)1 therapy
 - Resistant disease was defined as partial response, complete response, or stable disease for ≥12 weeks per RECIST v1.1, followed by radiographic disease progression
 - Refractory disease was defined as radiographic disease progression <12 weeks after initiation of treatment

Figure 1. Study design

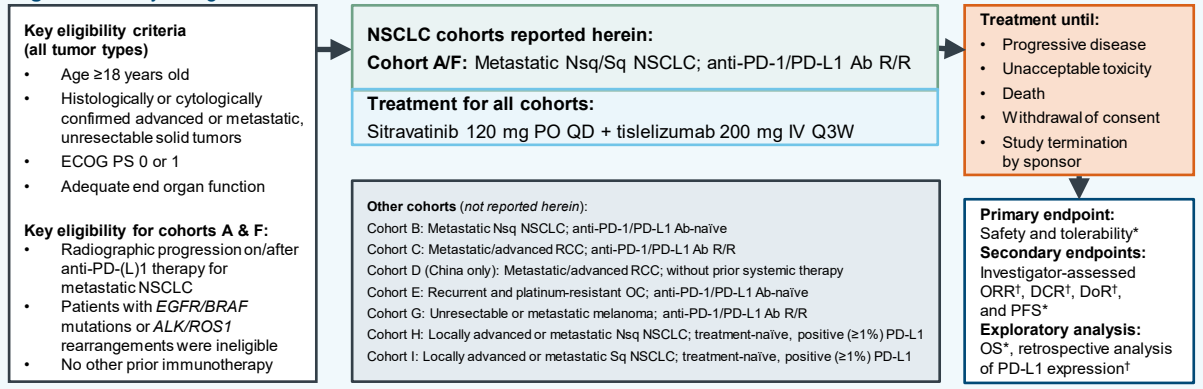
Key eligibility criteria (all tumor types)
• Age ≥18 years old
• Histologically or cytologically confirmed advanced or metastatic, unresectable solid tumors
• ECOG PS 0 or 1
• Adequate end organ function
Key eligibility for cohorts A & F:
• Radiographic progression on/after anti-PD-(L)1 therapy for metastatic NSCLC
• Patients with EGFR/BRAF mutations or ALK/ROS1 rearrangements were ineligible
• No other prior immunotherapy
NSCLC cohorts reported herein:
Cohort A/F: Metastatic Nsq/Sq NSCLC; anti-PD-1/PD-L1 Ab R/R
Treatment for all cohorts:
Sitravatinib 120 mg PO QD + tislelizumab 200 mg IV Q3W
Other cohorts (not reported herein):
Cohort B: Metastatic Nsq NSCLC; anti-PD-1/PD-L1 Ab-naïve
Cohort C: Metastatic/advanced RCC; anti-PD-1/PD-L1 Ab R/R
Cohort D (China only): Metastatic/advanced RCC; without prior systemic therapy
Cohort E: Recurrent and platinum-resistant OC; anti-PD-1/PD-L1 Ab-naïve
Cohort G: Unresectable or metastatic melanoma; anti-PD-1/PD-L1 Ab R/R
Cohort H: Locally advanced or metastatic Nsq NSCLC; treatment-naïve, positive (≥1%) PD-L1
Cohort I: Locally advanced or metastatic Sq NSCLC; treatment-naïve, positive (≥1%) PD-L1
Treatment until:
• Progressive disease
• Unacceptable toxicity
• Death
• Withdrawal of consent
• Study termination by sponsor
Primary endpoint:
Safety and tolerability*
Secondary endpoints:
Investigator-assessed ORR†, DCR†, DoR†, and PFS*
Exploratory analysis:
OS*, retrospective analysis of PD-L1 expression†

*Safety, tolerability, PFS, and OS were assessed using the safety analysis set (all patients receiving ≥1 dose of study drug); †Tumor responses were assessed using the efficacy evaluable analysis set (all dosed patients who had measurable disease at baseline per RECIST v1.1 and who had ≥1 evaluable post-baseline tumor assessment unless treatment was discontinued due to disease progression or death before tumor assessment)
Ab, antibody; ALK, anaplastic lymphoma kinase; BRAF, v-raf murine sarcoma viral oncogene homolog B1; DCR, disease control rate; DoR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; EGFR, epidermal growth factor receptor; IV, intravenously; NSCLC, non-small cell lung cancer; Nsq, non-squamous; OC, ovarian cancer; ORR, objective response rate; OS, overall survival; PD-1, programmed death protein-1; PD-L1, programmed death ligand-1; PFS, progression-free survival; PO, orally; QD, once-daily; Q3W, once every three weeks; R/R, resistant/refractory; RCC, renal cell carcinoma; RECIST, Response Evaluation Criteria in Solid Tumors; ROS1, c-ros oncogene 1; Sq, squamous

Results

- From December 2018–June 2020, 47 patients with non-squamous (n=24) and squamous (n=23) NSCLC were enrolled
- Median follow-up at the time of data cut-off (October 13, 2020) was 7.8 months (range: 0.4 to 18.1) and four patients (8.5%) remained on treatment
- Median age was 60 years and 72.3% of patients had received ≥2 prior lines of therapy (**Table 1**)

Table 1. Baseline demographic and disease characteristics

		Total (N=47)
Age, years	Median (range)	60.0 (25–79)
Sex, n (%)	Male	36 (76.6)
	Female	11 (23.4)
Race, n (%)	Asian	36 (76.6)
	White	11 (23.4)
ECOG PS, n (%)	0	13 (27.7)
	1	34 (72.3)
Histology at diagnosis, n (%)	Squamous	23 (48.9)
	Non-squamous	24 (51.1)
Prior lines of anticancer therapy, n (%)	1	13 (27.7)
	≥2	34 (72.3)
Duration of last therapy, months	Median (range)	4.21 (0.7–24.9)

ECOG PS, Eastern Cooperative Oncology Group performance status

Conclusions

- **Treatment with sitravatinib + tislelizumab had a manageable safety and tolerability profile in patients with metastatic NSCLC that is R/R to prior anti-PD-(L)1 therapy**
- **The combination demonstrated promising antitumor activity: patients achieved an ORR of 13.6%, DCR of 86.4%, and a median PFS of 5.2 months**
- **These findings support sitravatinib in combination with tislelizumab as a potential treatment option for patients with metastatic NSCLC that is R/R to prior anti-PD-(L)1 therapy, and further investigation is warranted**

Safety

- The median duration of exposure was 17.9 weeks (range: 1.3 to 53.9) for sitravatinib and 18 weeks (range: 3.0 to 51.1) for tislelizumab
- Mean relative dose intensity was 77.8% (SD: 21.6) for sitravatinib and 94.3% (SD: 10.4) for tislelizumab
- All patients had ≥1 treatment-emergent adverse event (TEAE) and ≥Grade 3 TEAEs were reported in 68.1% of patients (**Table 2**)
 - Hypertension was the most reported ≥Grade 3 TEAE (in 9 patients [19.1%], **Table 2**), which was well managed with anti-hypertensives
 - One patient had hypertension that led to sitravatinib dose reduction, four patients had hypertension (grouped terms) that led to sitravatinib dose interruption, and one patient had hypertension that led to tislelizumab dose modification
- All patients had ≥1 treatment-related adverse event (TRAE) and ≥Grade 3 TRAEs were reported in 19 patients (40.4%, **Table 2**)
- TRAEs leading to death were reported in three patients, including one case each of cardiac failure with pneumonia and respiratory failure (related to tislelizumab), one case of ischemic stroke (related to sitravatinib), and one case of unspecified death (related to sitravatinib + tislelizumab)

Table 2. Summary of TEAE and TRAE incidence (safety analysis set)

Patients, n (%)	Total (N=47)	
	TEAE	TRAE
Any AE	47 (100.0)	47 (100.0)
≥Grade 3 AE	32 (68.1)	19 (40.4)
Serious AE	24 (51.1)	15 (31.9)
≥Grade 3 serious AE	21 (44.7)	8 (17.0)
AE leading to death	8 (17.0)	3 (6.4)
AE leading to treatment discontinuation	9 (19.1)	9 (19.1)
AE leading to tislelizumab dose modification*	18 (38.3)	17 (36.2)
AE leading to sitravatinib dose modification†	35 (74.5)	34 (72.3)
≥Grade 3 AEs reported in ≥5% of patients‡		
Hypertension	9 (19.1)	8 (17.0)
Death	4 (8.5)	1 (2.1)
Stomatitis	3 (6.4)	3 (6.4)

**AE leading to tislelizumab dose modification includes dose delay and/or interruption; †AE leading to sitravatinib dose modification includes dose reduction and/or interruption; ‡Incidences reported by preferred term for any TEAE or TRAE reported in ≥5% of patients*
All AEs are treatment-emergent and graded based on National Cancer Institute–Common Terminology Criteria for Adverse Events (version 5.0)
AE, adverse event; TEAE, treatment-emergent AE; TRAE; treatment-related AE

Efficacy: Tumor response

- Treatment with sitravatinib + tislelizumab demonstrated antitumor activity, with an objective response rate of 13.6% (**Table 3**)
 - The median duration of response was 6.9 months (**Table 3**)
- Median time to response was 2.7 months (range: 1.4 to 5.5 months)
- Confirmed partial response was reported in 6 patients (13.6%) (**Table 3 and Figure 2)**
- Disease control was achieved in the majority of patients (86.4%, **Table 3**)

Table 3. Analysis of confirmed disease response per RECIST v1.1 (efficacy evaluable analysis set)

Clinical activity	Total (N=44)
Confirmed ORR, % (95% CI)	13.6 (5.2, 27.4)
Best overall response, n (%)	
Complete response	0 (0.0)
Partial response	6 (13.6)
Stable disease	32 (72.7)
Progressive disease	3 (6.8)
NE	3 (6.8)*
DCR†, % (95% CI)	86.4 (72.7, 94.8)
Median DoR, months (95% CI)	6.90 (3.06, NE)

**Includes two patients who died early with no post-baseline tumor assessment and one patient with an NE tumor response; †DCR = complete response + partial response + stable disease*
CI, confidence interval; DCR, disease control rate; DoR, duration of response; NE, non-evaluable, ORR, objective response rate; RECIST, Response Evaluation Criteria in Solid Tumors

Figure 2. Best change in target lesion size from baseline by confirmed best overall response (efficacy evaluable analysis set)

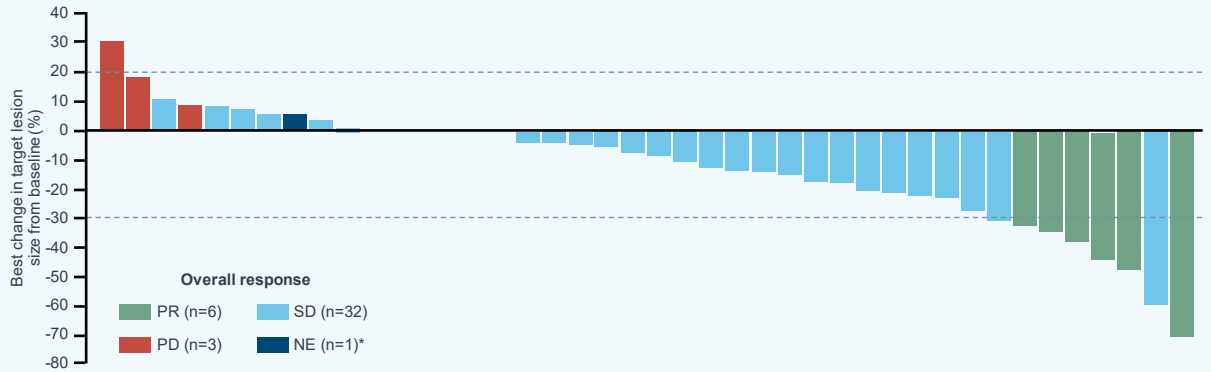
Best change in target lesion size from baseline (%)
40
30
20
10
0
-10
-20
-30
-40
-50
-60
-70
-80
Overall response
PR (n=6)
SD (n=32)
PD (n=3)
NE (n=1)*

**Two patients with no post-baseline tumor assessment due to early death were not included in this figure; Tumor responses assessed by investigators per RECIST v1.1*
NE, non-evaluable; PD, disease progression; PR, partial response; SD, stable disease

Efficacy: Survival

- Median progression-free survival (PFS) was 5.2 months (95% CI: 4.1, 5.9) (**Figure 3A**)
 - 6- and 12-month PFS rates were 33.9% (95% CI: 19.0, 49.4) and 6.4% (95% CI: 0.5, 23.5), respectively
- Median overall survival (OS) was 10.1 months (95% CI: 6.1, 18.1) (**Figure 3B**)
 - OS data are not mature (median follow-up duration was 12.4 months)

Figure 3. PFS and OS (safety analysis set)

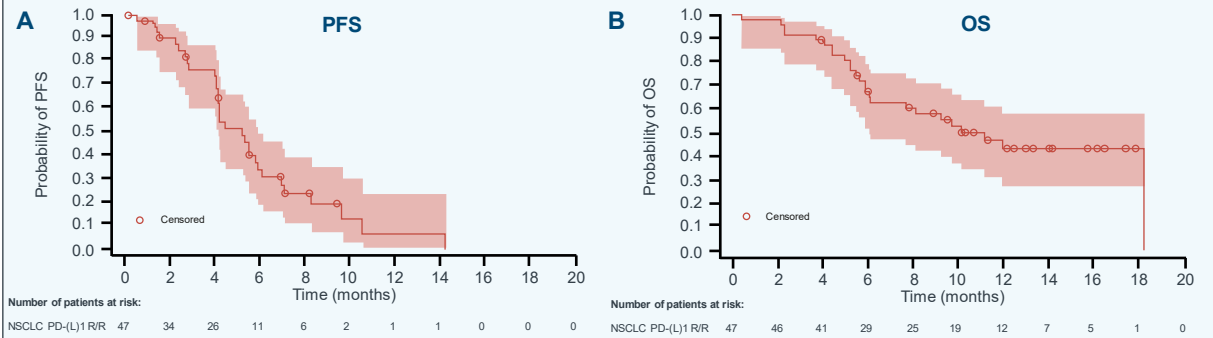
A
PFS
Probability of PFS
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
Censored
0 2 4 6 8 10 12 14 16 18 20
Time (months)
Number of patients at risk:
NSCLC PD-(L)1 R/R 47 34 26 11 6 2 1 1 0 0 0
B
OS
Probability of OS
Censored
0 2 4 6 8 10 12 14 16 18 20
Time (months)
Number of patients at risk:
NSCLC PD-(L)1 R/R 47 46 41 29 25 19 12 7 5 1 0

NSCLC, non-small cell lung cancer; OS, overall survival; PD-1, programmed death protein-1; PFS, progression-free survival; R/R relapsed/refractory

Efficacy: PD-L1 expression and tumor response

- Defined cut-offs for PD-L1 tumor cell or immune cell expression were used to investigate whether there was an association between PD-L1 expression and tumor response
 - Based on current results, no association was observed (**Figure 4**) and further exploration is required in a larger population

Figure 4. Subgroup analysis of ORR per TC and IC PD-L1 expression (efficacy evaluable analysis set*)

PD-L1 expression level of TC	n	Response
Total	44	6
TC < or ≥1%		
<1%	10	2
≥1%	12	1
TC < or ≥50%		
<50%	16	2
≥50%	6	1
NA†	22	3

PD-L1 expression level of IC	n	Response
Total	44	6
IC < or ≥10%		
<10%	14	2
≥10%	8	1
IC < or ≥30%		
<30%	18	2
≥30%	4	1
NA†	22	3

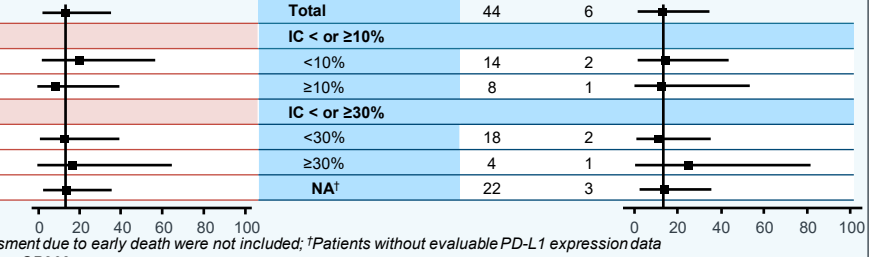
ORR, % (95% CI)
0 20 40 60 80 100
ORR, % (95% CI)
0 20 40 60 80 100

**Two patients with no post-baseline tumor assessment due to early death were not included; †Patients without evaluable PD-L1 expression data*
PD-L1 expression was assessed using the Ventana SP263 assay
CI, confidence interval; IC, immune cell; NA, not applicable; ORR, objective response rate; PD-L1, programmed death ligand-1; TC, tumor cell

References

1. Wagner G, et al. Oncoimmunology 2020;9:1774314
2. Pathak R, et al. Cancers 2020;12:3851
3. Planchard D, et al. Ann Oncol 2018;29(Suppl 4):iv192–iv237 [Updated September 2020]
4. Dahan R, et al. Cancer Cell 2015;28:285–295
5. Zhang T, et al. Cancer Immunol Immunother 2018;67:1079–90
6. Du W, et al. JCI Insight 2018;3:e124184
7. Marshall HT, Djamgoz MBA. Front Oncol. 2018;8:315

Acknowledgements

Medical writing support for the development of this poster and associated abstract, under direction of the authors, was provided by Claire White, PhD, of Ashfield MedComms, an Ashfield Health company, and was funded by BeiGene, Ltd.

ESMO Targeted Anticancer Therapies Congress; March 7-8, 2022