Consideration of Adjusted Ideal Body Weight Dosing in BG-C9074 (B7-H4–Targeting ADC) From Pharmacokinetics, Efficacy, and Safety Perspectives

3029

Hugh Giovinazzo,[1] Ramil Abdrashitov,[2] Yaogeng Wang,[3] Garret Winkler,[1] Shuai Yuan,[3] Saana D'Alonzo,[4] Aleksandra Kozicka,[4] William D. Hanley,[1] Nageshwar Budha,[1] Yen Lin Chia[1]

[1]BeOne Medicines, Ltd, San Carlos, CA, USA; [2]BeOne Medicines, Ltd, Gaithersburg, MD, USA; [3]BeOne Medicines, Ltd, Shanghai, China; [4]BeOne Medicines Switzerland GmbH, Basel, Switzerland

CONCLUSIONS

- AiBW dosing effectively reduced PK variability compared with TBW dosing, resulting in more consistent exposure across body weight ranges
- Higher ADC exposure was associated with increased efficacy, but also with a higher incidence of manageable TRAEs
- AiBW dosing optimizes the risk–benefit profile of BG-C9074 across body weight ranges

INTRODUCTION

- B7-H4, a transmembrane glycoprotein in the B7 superfamily, has limited expression in normal tissue but is upregulated in solid tumors, including breast cancer, ovarian cancer (OC), endometrial cancer (EC), and cholangiocarcinoma (CCA)[1,2]
- BG-C9074 is an investigational topoisomerase I inhibitor antibody-drug conjugate (ADC) that targets B7-H4 with an innovative drug-linker design, a drug-to-antibody ratio of 6, and strong bystander effect
- Here, we present results of the pharmacokinetic (PK) and exposure–response (ER) analyses supporting the implementation of adjusted ideal body weight (AiBW)-based dosing in the ongoing phase 1 study (NCT06233942)

METHODS

Trial Design

- This dose-escalation component of the phase 1a/b, open label, multicenter study comprised two parts (**Figure 1**)

Figure 1. Study Design

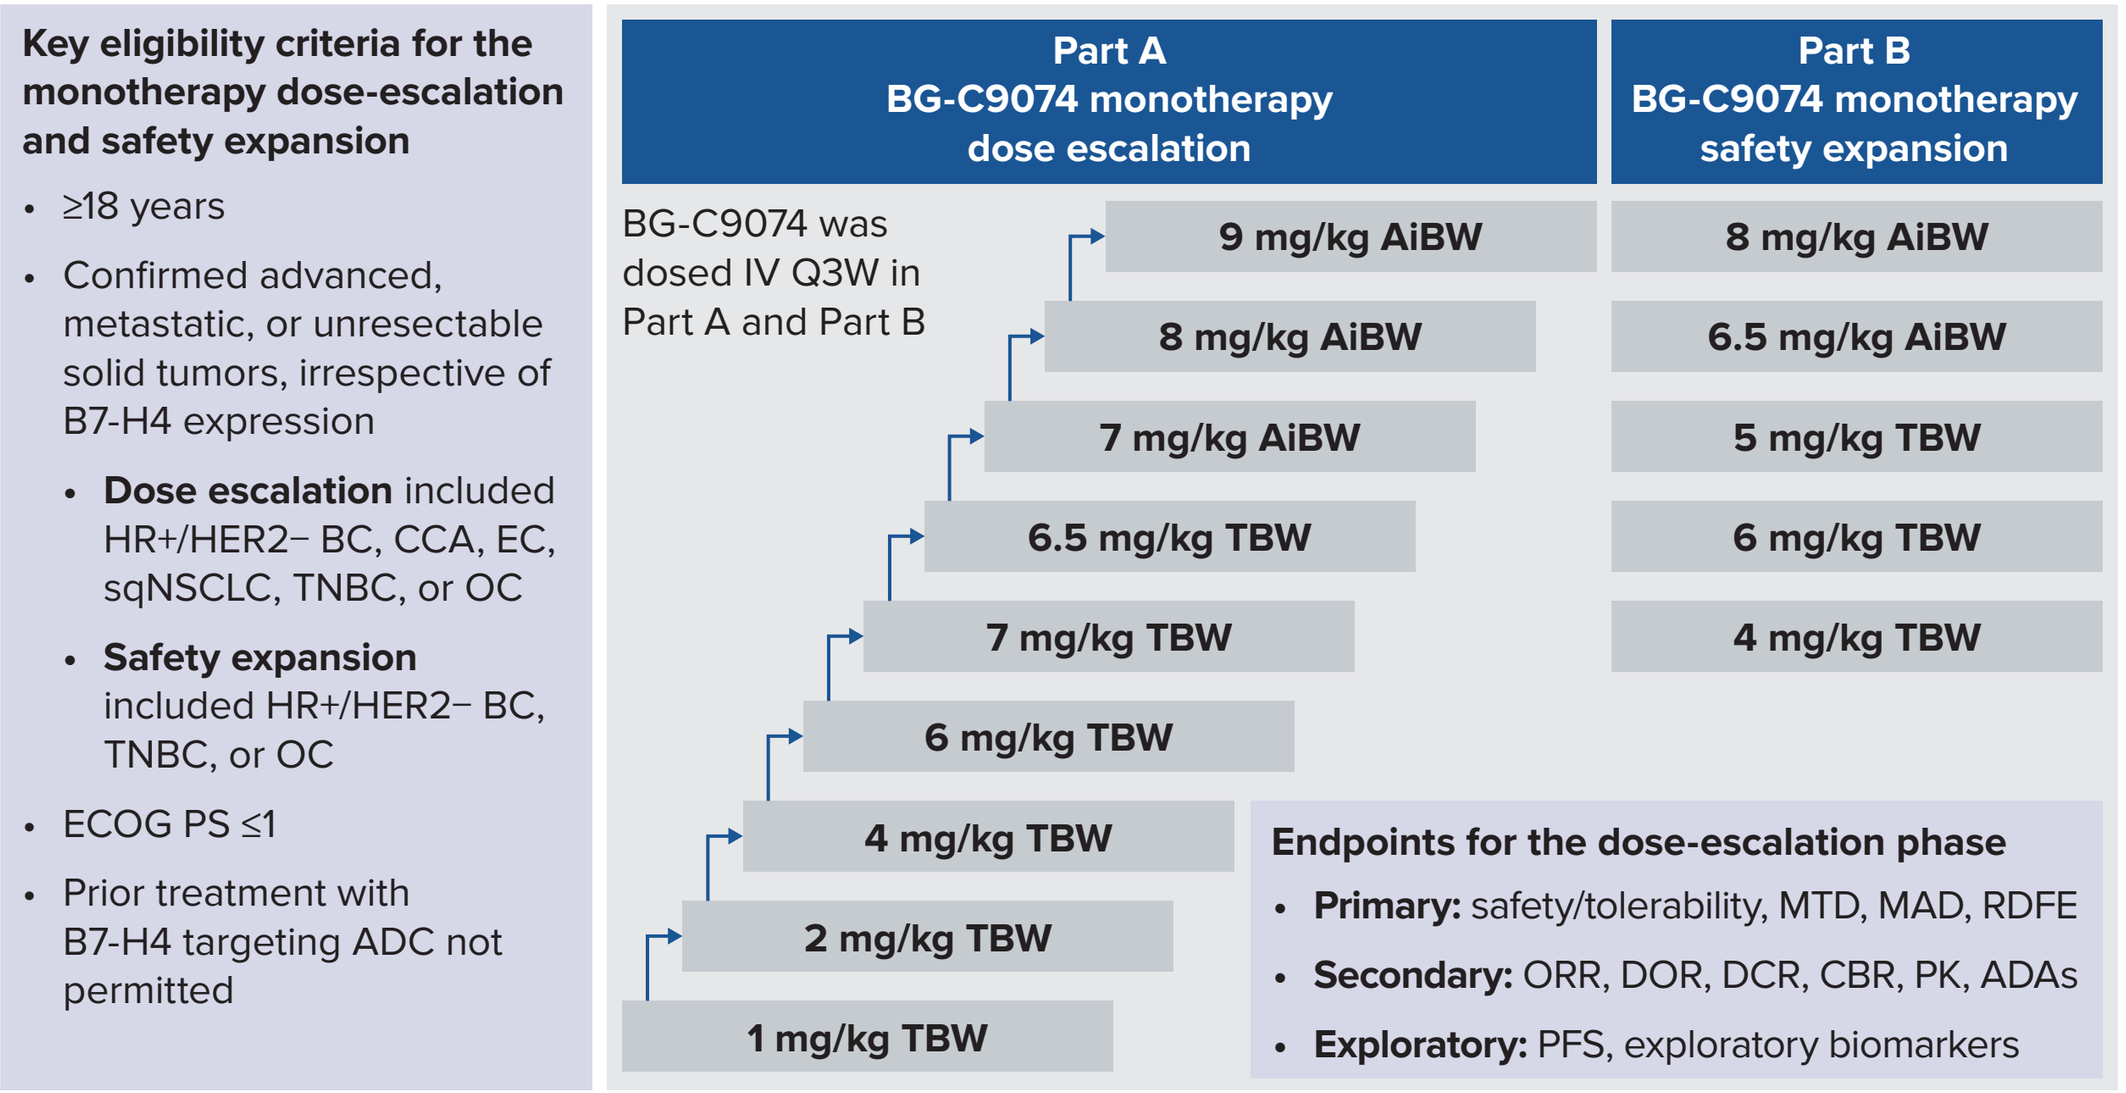
Key eligibility criteria for the monotherapy dose-escalation and safety expansion
≥18 years
Confirmed advanced, metastatic, or unresectable solid tumors, irrespective of B7-H4 expression
Dose escalation included HR+/HER2− BC, CCA, EC, sqNSCLC, TNBC, or OC
Safety expansion included HR+/HER2− BC, TNBC, or OC
ECOG PS ≤1
Prior treatment with B7-H4 targeting ADC not permitted
Part A BG-C9074 monotherapy dose escalation
BG-C9074 was dosed IV Q3W in Part A and Part B
9 mg/kg AiBW
8 mg/kg AiBW
7 mg/kg AiBW
6.5 mg/kg TBW
7 mg/kg TBW
6 mg/kg TBW
4 mg/kg TBW
2 mg/kg TBW
1 mg/kg TBW
Part B BG-C9074 monotherapy safety expansion
8 mg/kg AiBW
6.5 mg/kg AiBW
5 mg/kg TBW
6 mg/kg TBW
4 mg/kg TBW
Endpoints for the dose-escalation phase
Primary: safety/tolerability, MTD, MAD, RDFE
Secondary: ORR, DOR, DCR, CBR, PK, ADAs
Exploratory: PFS, exploratory biomarkers

Abbreviations: ADA, anti-drug antibody; AiBW, adjusted ideal body weight; BC, breast cancer; CBR, clinical benefit rate; CCA, cholangiocarcinoma; DCR, disease control rate; DOR, duration of response; EC, endometrial cancer; ECOG PS, Eastern Cooperative Oncology Group performance status; HR+/HER2−, hormone receptor positive/human epidermal growth factor receptor 2 negative; IV, intravenous; MAD, multiple ascending dose; MTD, maximum tolerated dose; OC, ovarian cancer; ORR, objective response rate; PFS, progression-free survival; PK, pharmacokinetics; Q3W, every 3 weeks; RDFE, recommended dose for expansion; sqNSCLC, squamous non-small cell lung cancer; TBW, total body weight; TNBC, triple-negative breast cancer.

AiBW Calculation

$$AiBW = 0.4 * (\text{actual weight} - IBW) + IBW$$

where

$$IBW\ (\text{men}) = 50\ \text{kg} + 2.3\ \text{kg} * (\text{height, in} - 60)$$

$$IBW\ (\text{women}) = 45.5\ \text{kg} + 2.3\ \text{kg} * (\text{height, in} - 60)$$

- Rich PK sampling was performed in Cycle 1 and again at steady state in Cycle 5

Analysis and Statistical Methods

- Population PK models for conjugated P1021 (ADC) and free payload analyses were developed using Monolix
- Simulated PK metrics of area under curve (AUC), C_{max} and C_{avg} time-to-event endpoints were calculated using Monolix and R
- ER analyses for efficacy and safety were evaluated using incidence of events by exposure quantiles and logistic regression

RESULTS

Baseline Characteristics and Patient Disposition

- Wide ranges of body weight and body mass index (BMI) values were observed across dose cohorts
- Higher body weight and BMI were associated with increased exposure and a corresponding increase in adverse events (AEs)
- An AiBW dosing strategy was introduced for all newly enrolled patients, which started at 6.5 mg/kg AiBW and enabled dose escalation up to 9 mg/kg AiBW

PK

- Approximately dose-proportional PK was observed for both the ADC and the payload
- T_{max} occurred at 15 minutes to 4 hours for the ADC and 2-6 hours for the free payload
- The ADC half-life was approximately 7 days (**Figure 2**)
- BG-C9074 ADC PK exhibited declining clearance over time, leading to reduced formation of the free P1021 payload. The PK was well described by a two-compartment population PK model with a time-varying component for both the ADC and the payload
- AIBW dosing allowed reduction of PK variability across patients with different BMI

Figure 2. Concentration-Time Plots for ADC and Free Payload

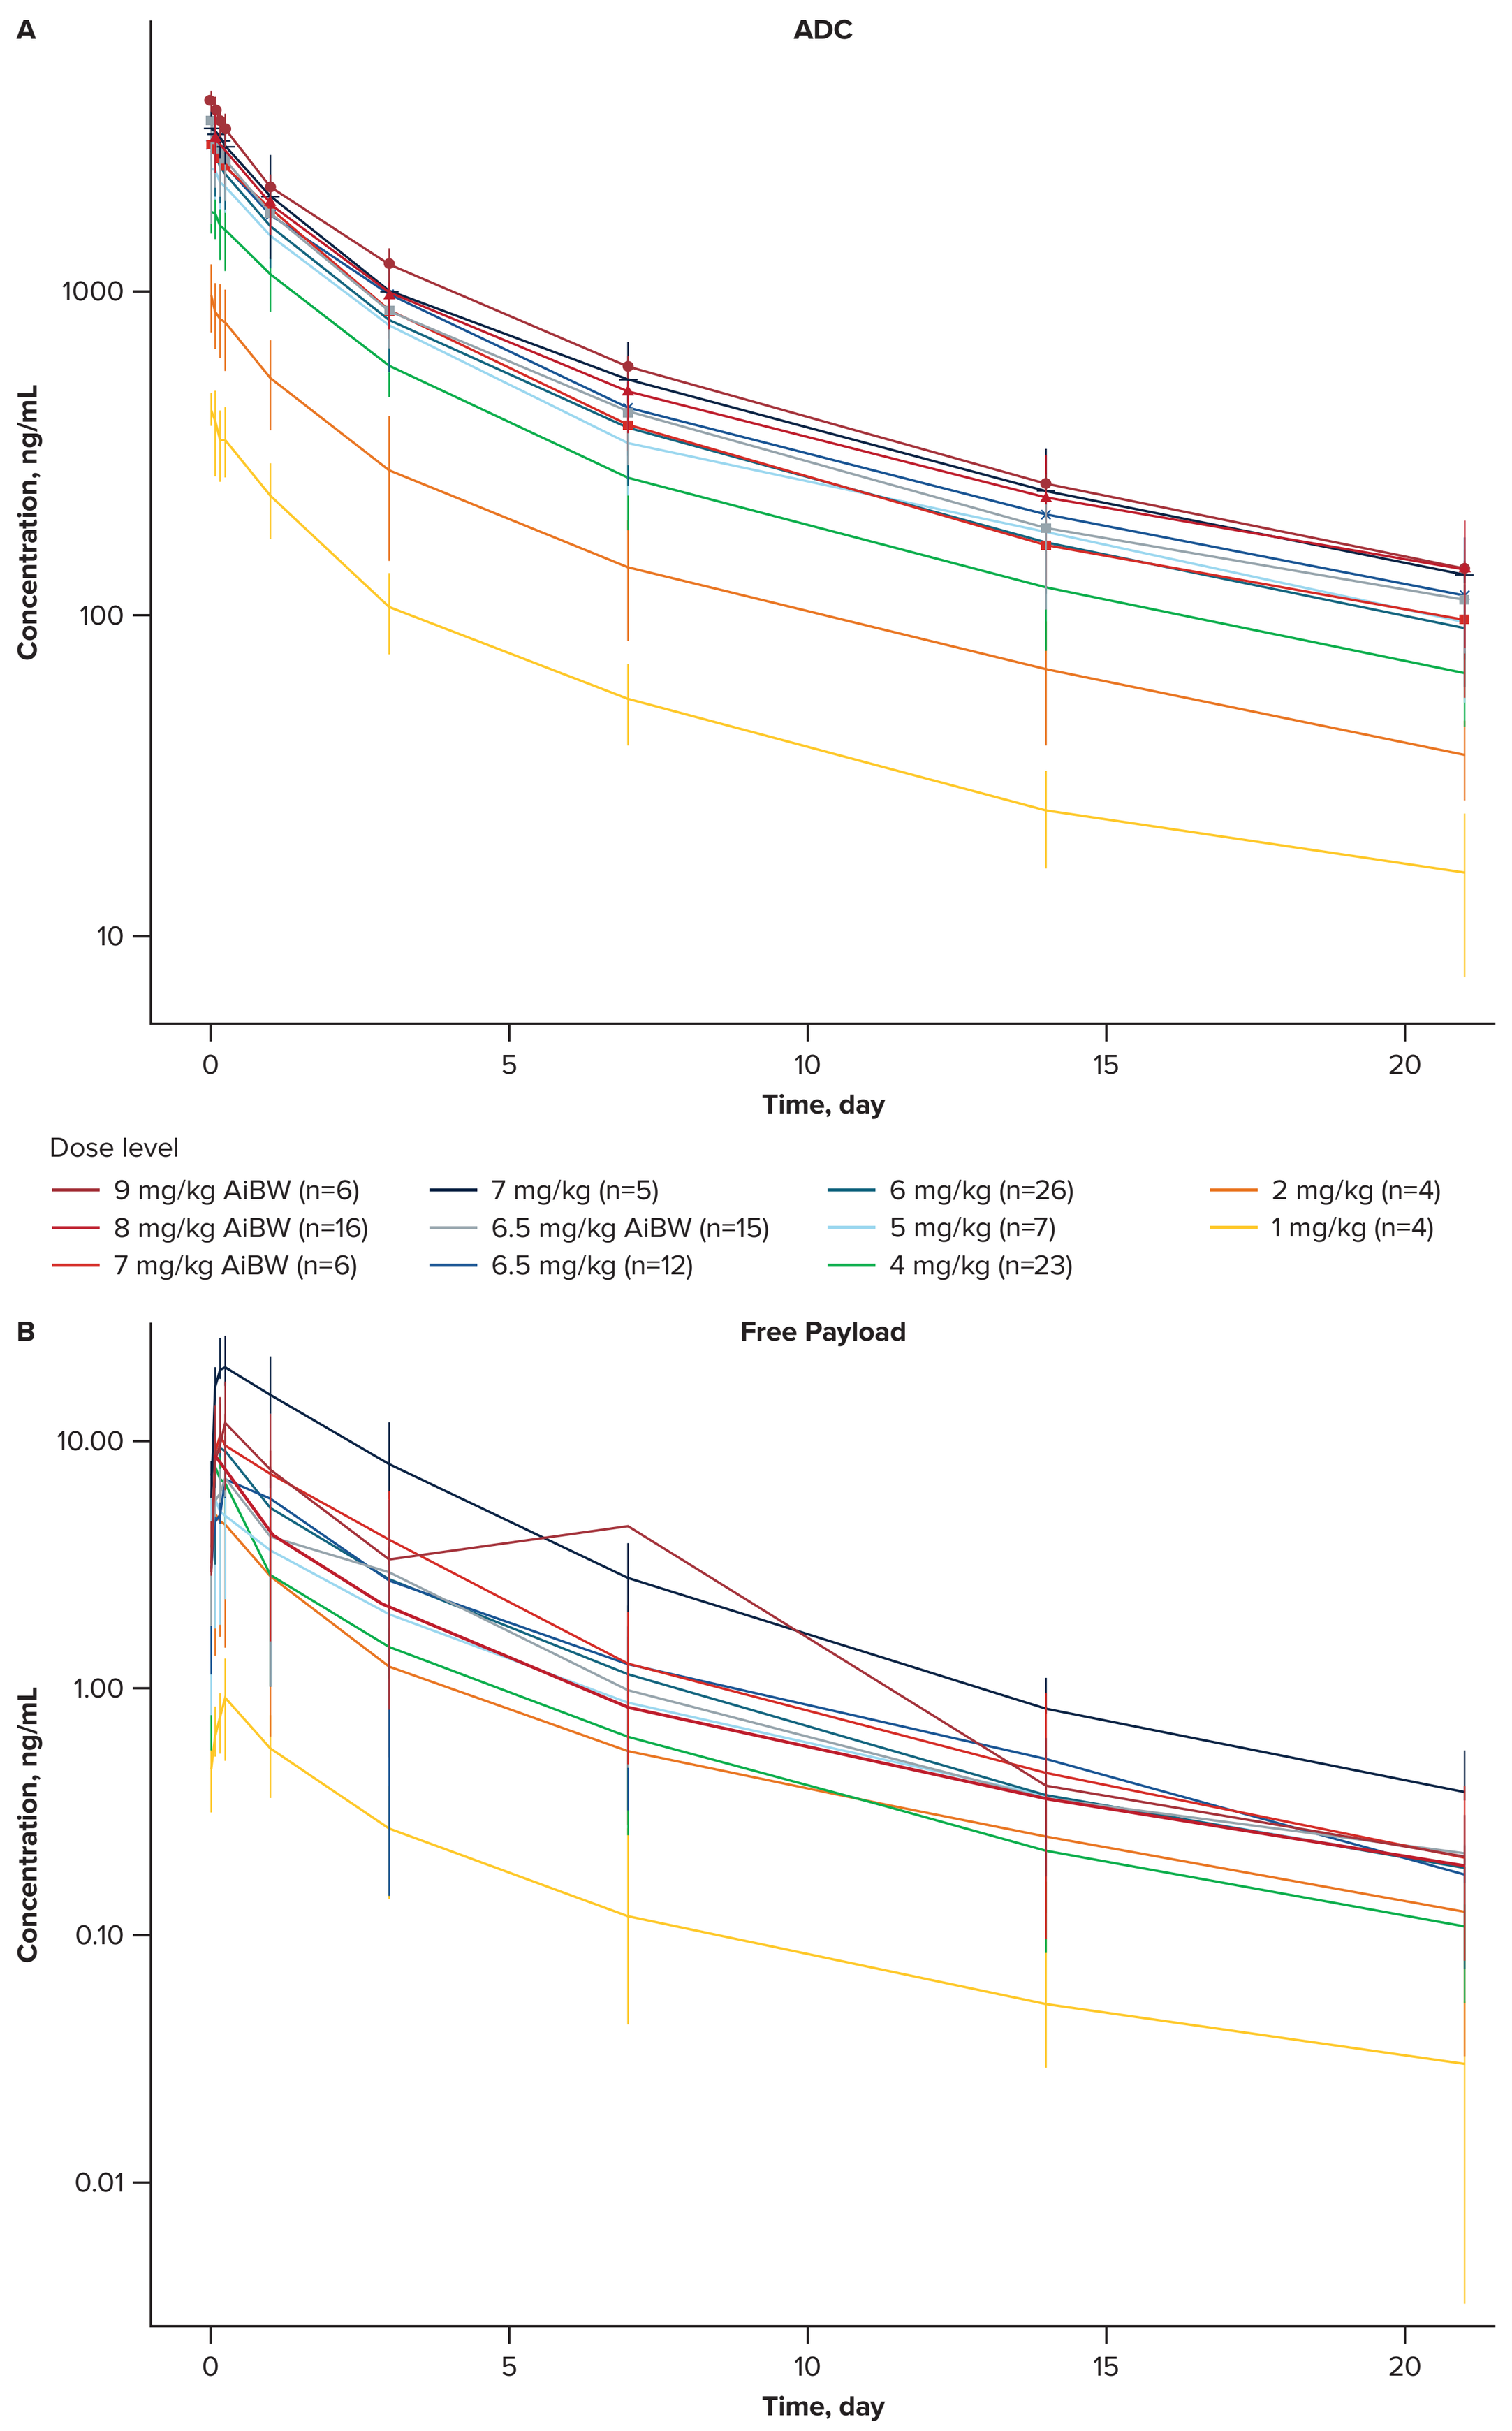
A
ADC
Concentration, ng/mL
1000
100
10
0
5
10
15
20
Time, day
Dose level
9 mg/kg AiBW (n=6)
8 mg/kg AiBW (n=16)
7 mg/kg AiBW (n=6)
7 mg/kg (n=5)
6.5 mg/kg AiBW (n=15)
6.5 mg/kg (n=12)
6 mg/kg (n=26)
5 mg/kg (n=7)
4 mg/kg (n=23)
2 mg/kg (n=4)
1 mg/kg (n=4)
B
Free Payload
Concentration, ng/mL
10.00
1.00
0.10
0.01
Time, day

Abbreviations: ADC, antibody-drug conjugate; AiBW, adjusted ideal body weight.

Safety

- Increased incidence of grade ≥3 treatment-related adverse events (TRAEs) and grade ≥3 neutropenia with BMI observed with TBW dosing and similar trends observed in body weight led to implementation of AiBW dosing (**Figure 3**)

Figure 3. TBW Dosing Leads to Increased Incidence of Grade ≥3 TRAEs and Grade ≥3 Neutropenia With BMI

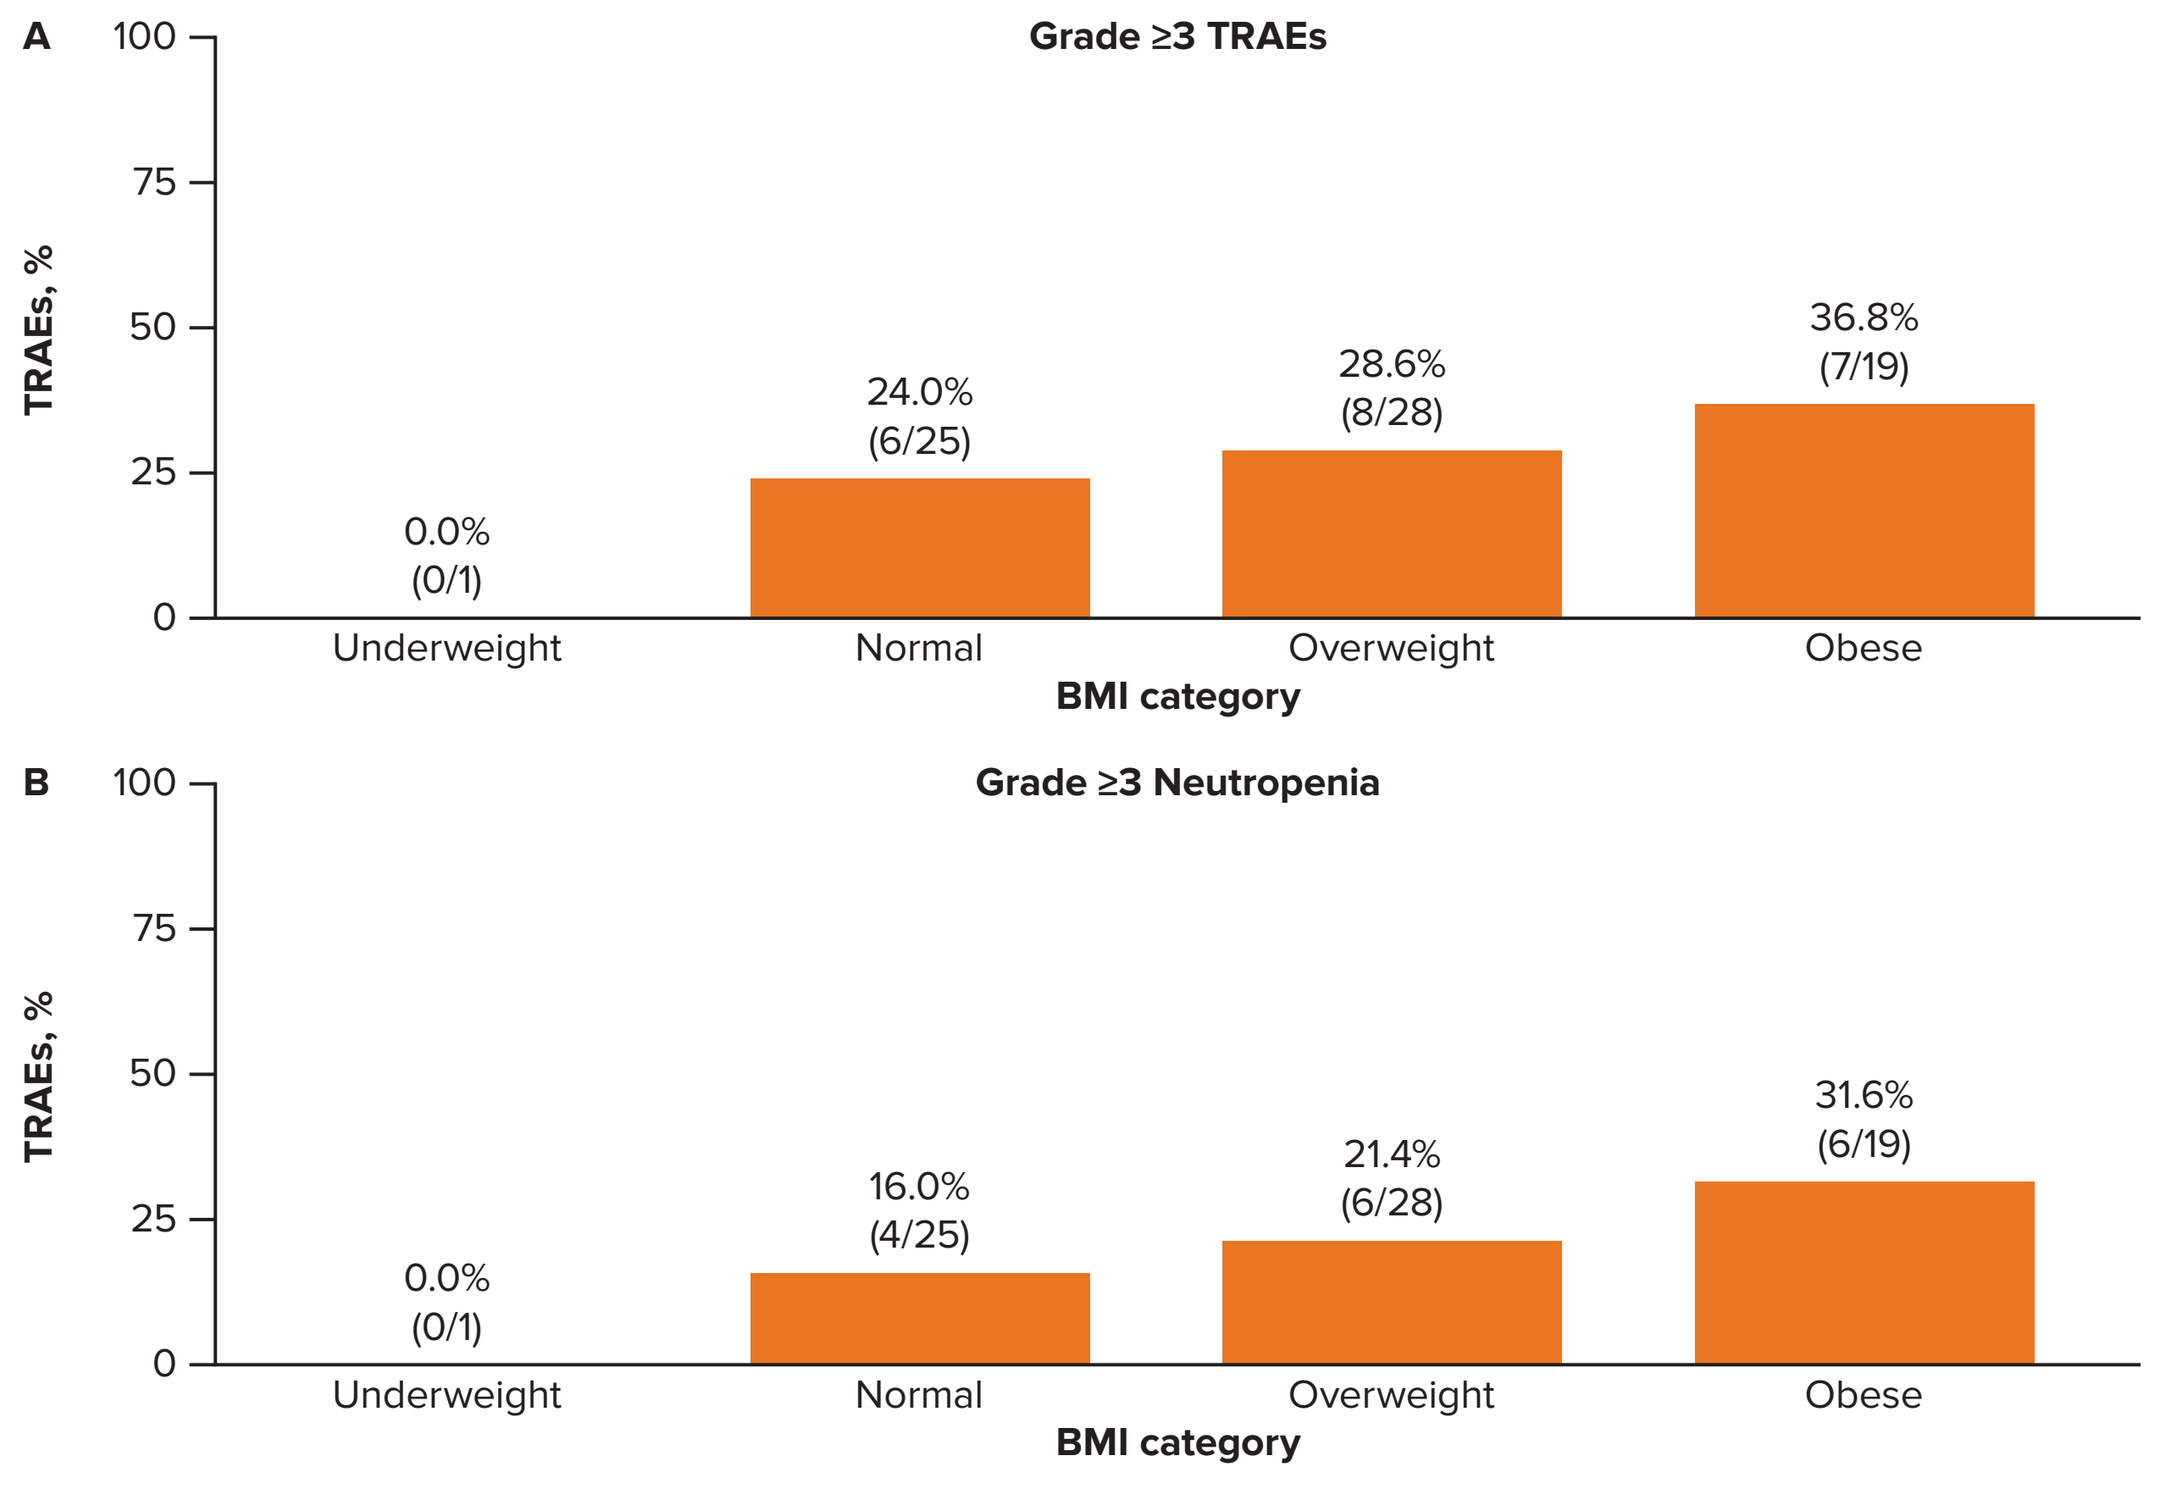
A
Grade ≥3 TRAEs
TRAEs, %
0.0% (0/1)
24.0% (6/25)
28.6% (8/28)
36.8% (7/19)
Underweight
Normal
Overweight
Obese
BMI category
B
Grade ≥3 Neutropenia
0.0% (0/1)
16.0% (4/25)
21.4% (6/28)
31.6% (6/19)

Abbreviations: BMI, body mass index; TBW, total body weight; TRAE, treatment-related adverse event.

- TBW dosing led to increased ADC C_{avg} and Payload C_{avg} with higher BMI (**Figure 4A**)
- Implementation of AiBW resulted in more narrow ADC C_{avg} exposure range across BMI groups (**Figure 4B**) and allowed increase to 8 mg/kg AiBW during dose escalation
- AiBW dosing allowed dose escalation to 8 mg/kg AiBW while maintaining the exposure of high BMI/body weight in the tolerable exposure range (**Figure 4C**)

Figure 4. Reduction in PK Variability Observed With Implementation of AiBW Dosing

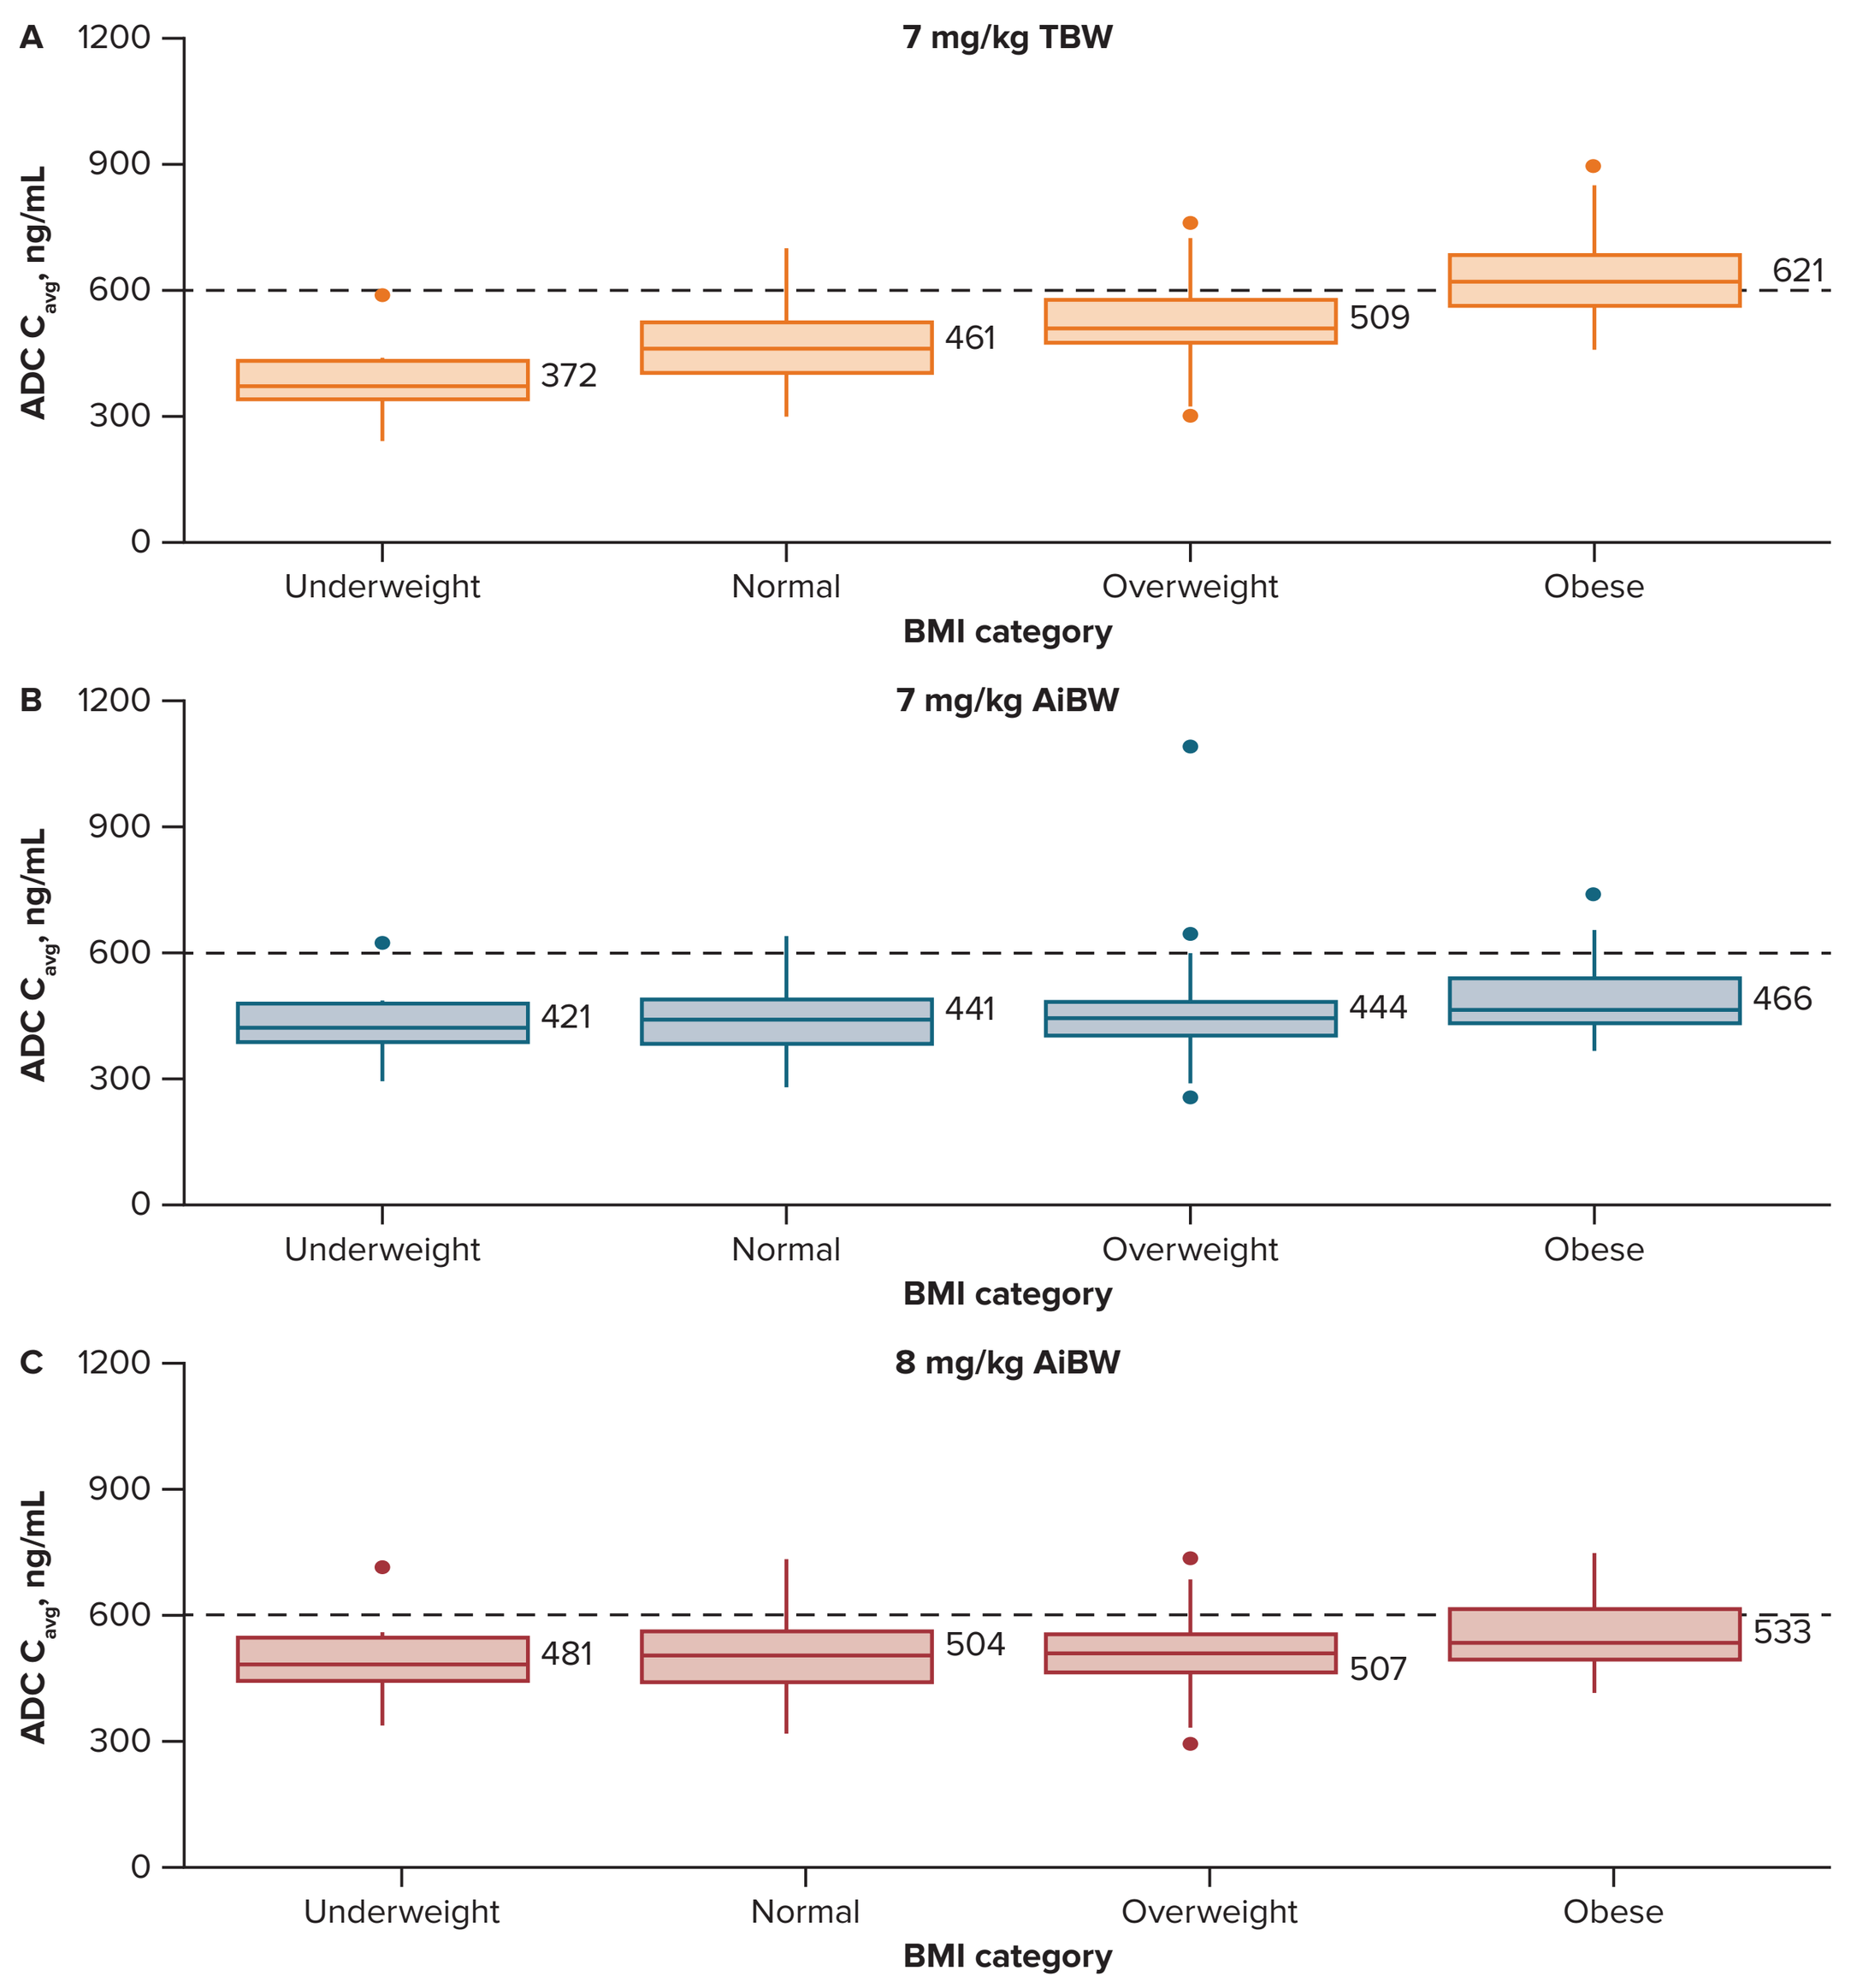
A
7 mg/kg TBW
ADC Cavg, ng/mL
372
461
509
621
Underweight
Normal
Overweight
Obese
BMI category
B
7 mg/kg AiBW
421
441
444
466
C
8 mg/kg AiBW
481
504
507
533

Abbreviations: ADC, antibody-drug conjugate; AiBW, adjusted ideal body weight; BMI, body mass index; TBW, total body weight.

- Thus, implementation of AiBW-based dosing reduced the incidence of grade ≥3 TRAEs, grade ≥3 neutropenia, AEs leading to dose modification, and both all grade and grade ≥3 nausea and vomiting across BMI (**Figure 5**)

Figure 5. AiBW Dosing Leads to Lower and Consistent Incidence of Grade ≥3 TRAEs and Grade ≥3 Neutropenia With BMI

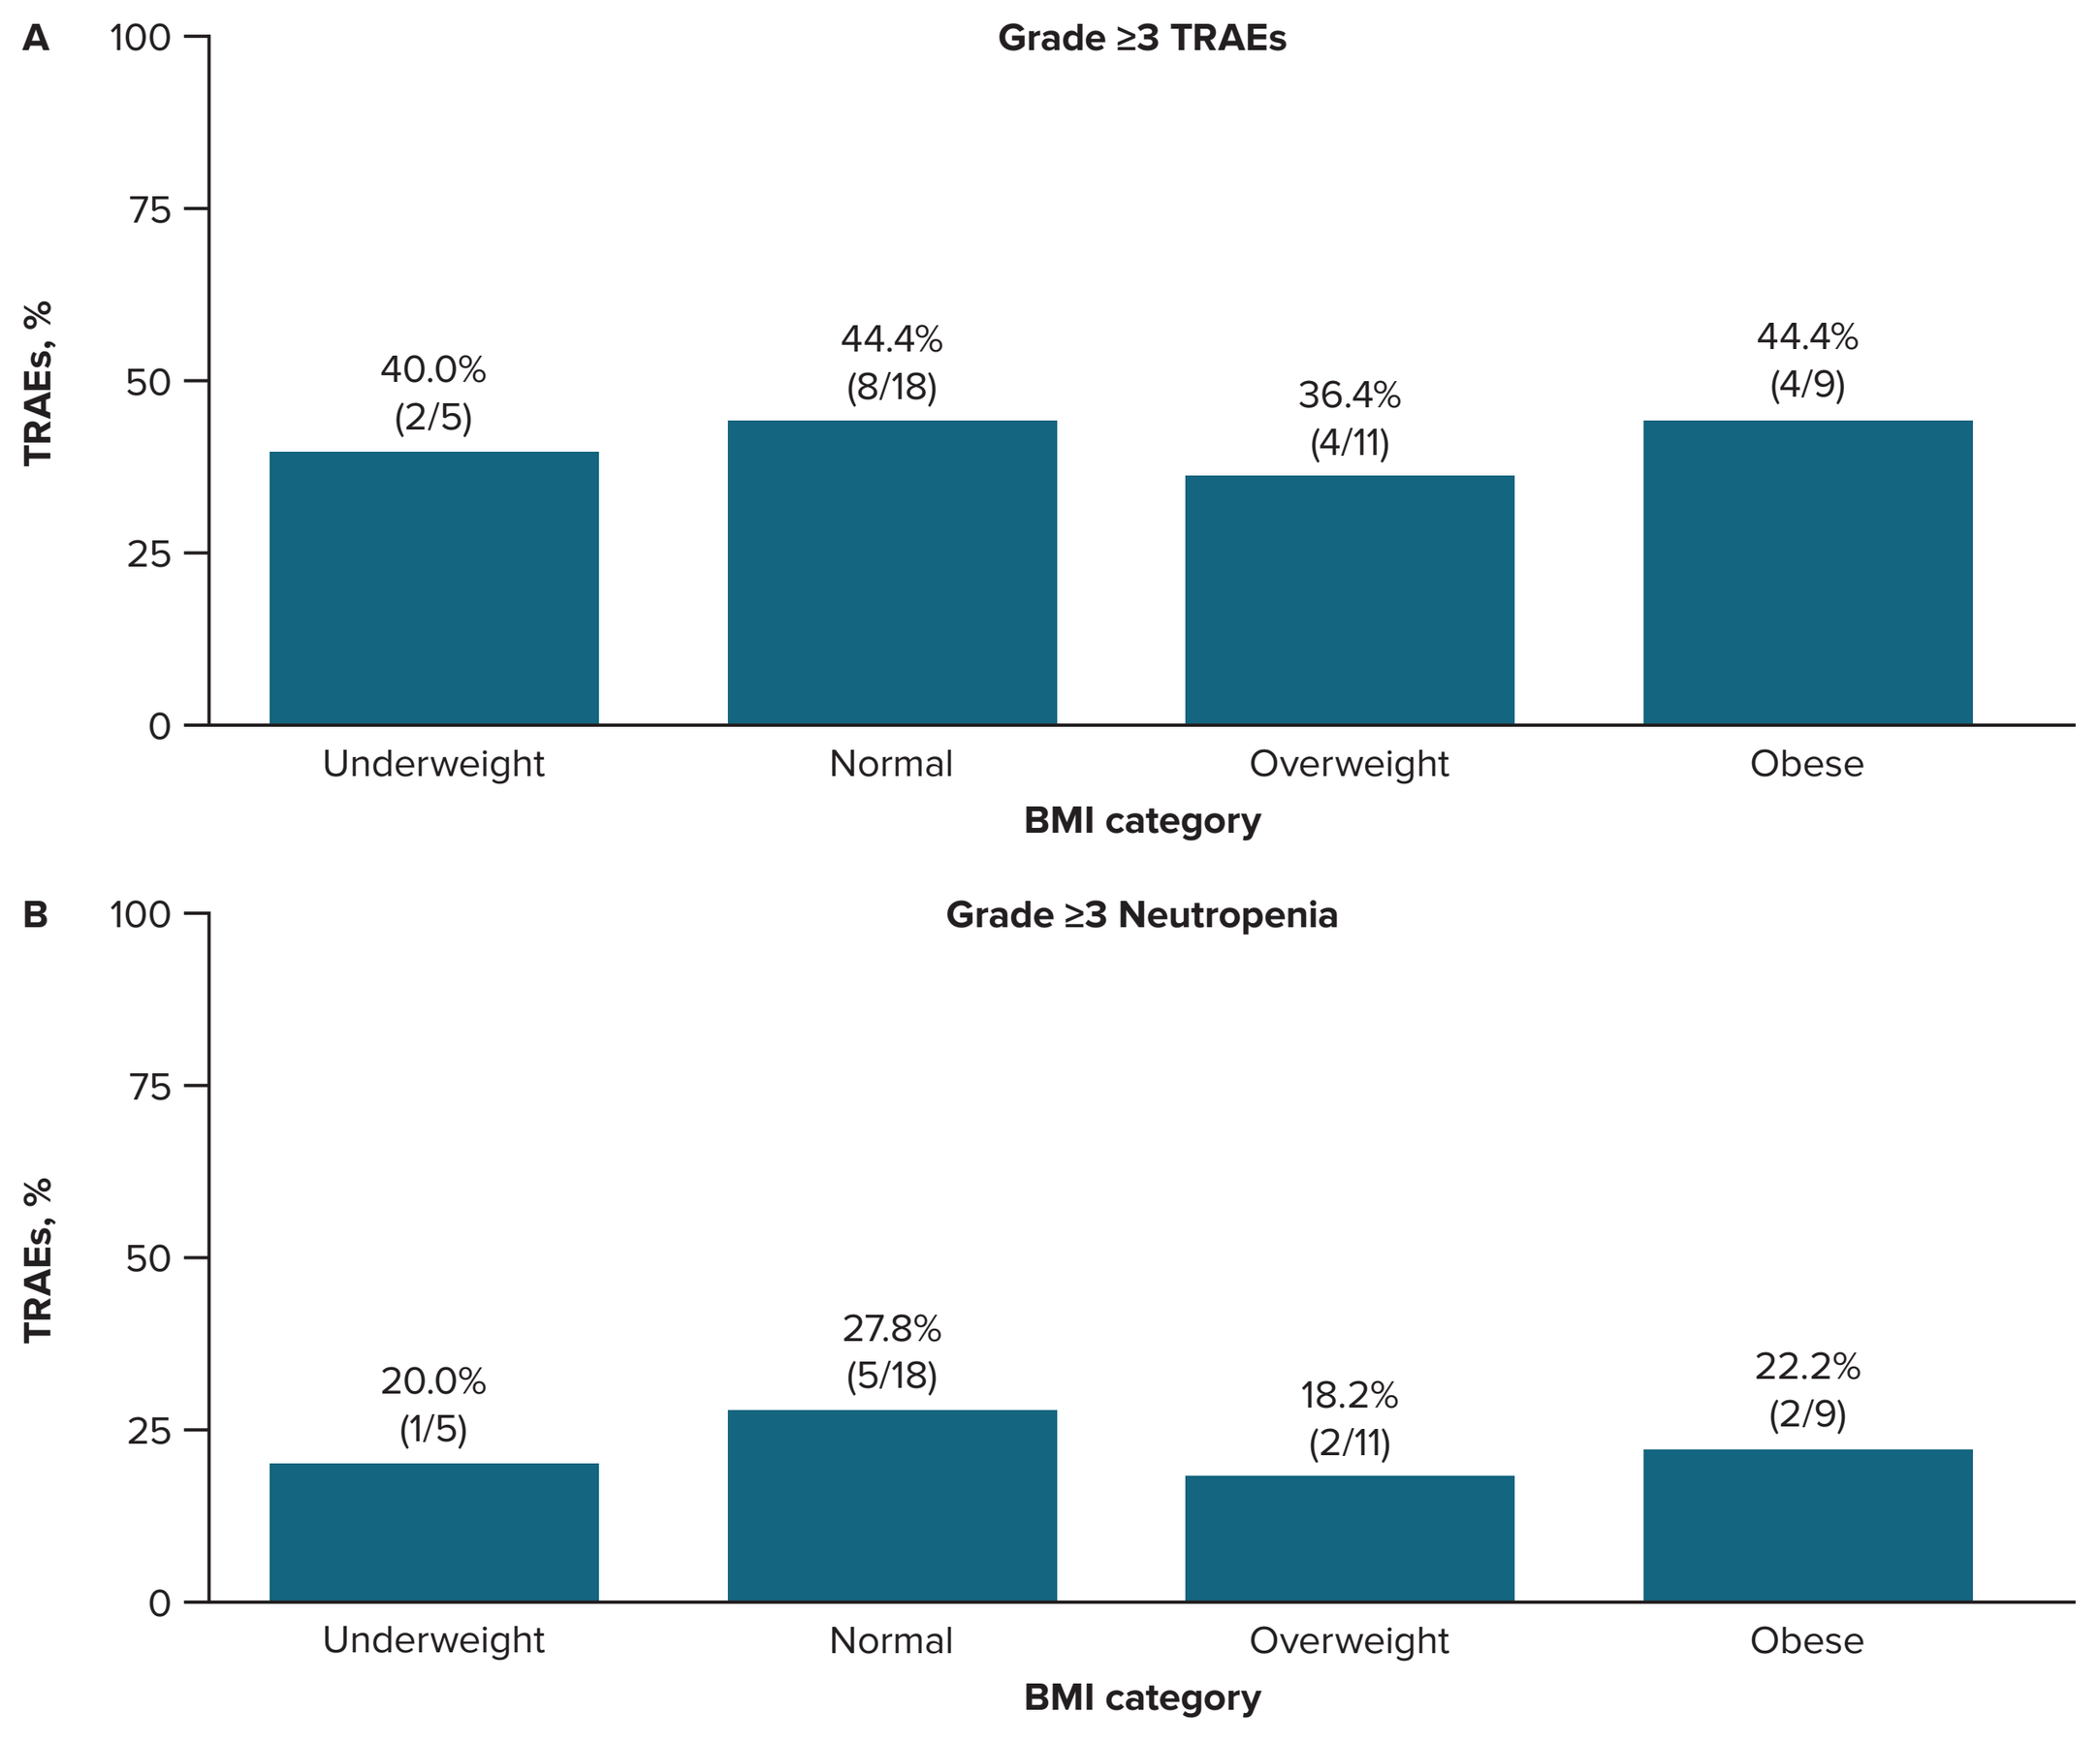
A
Grade ≥3 TRAEs
TRAEs, %
40.0% (2/5)
44.4% (8/18)
36.4% (4/11)
44.4% (4/9)
Underweight
Normal
Overweight
Obese
BMI category
B
Grade ≥3 Neutropenia
20.0% (1/5)
27.8% (5/18)
18.2% (2/11)
22.2% (2/9)

Abbreviations: AiBW, adjusted ideal body weight; BMI, body mass index; TRAE, treatment-related adverse event.

- A relationship between ADC exposure and grade ≥3 TRAEs, grade ≥3 neutropenia, and AEs leading to dose modification was observed, with a similar pattern seen for the free payload (**Figures 6 and 7**)

Figure 6. Implementation of AiBW Dosing Results in Reduced Incidence of Grade ≥3 TRAEs and Grade ≥3 Neutropenia Across Cohorts

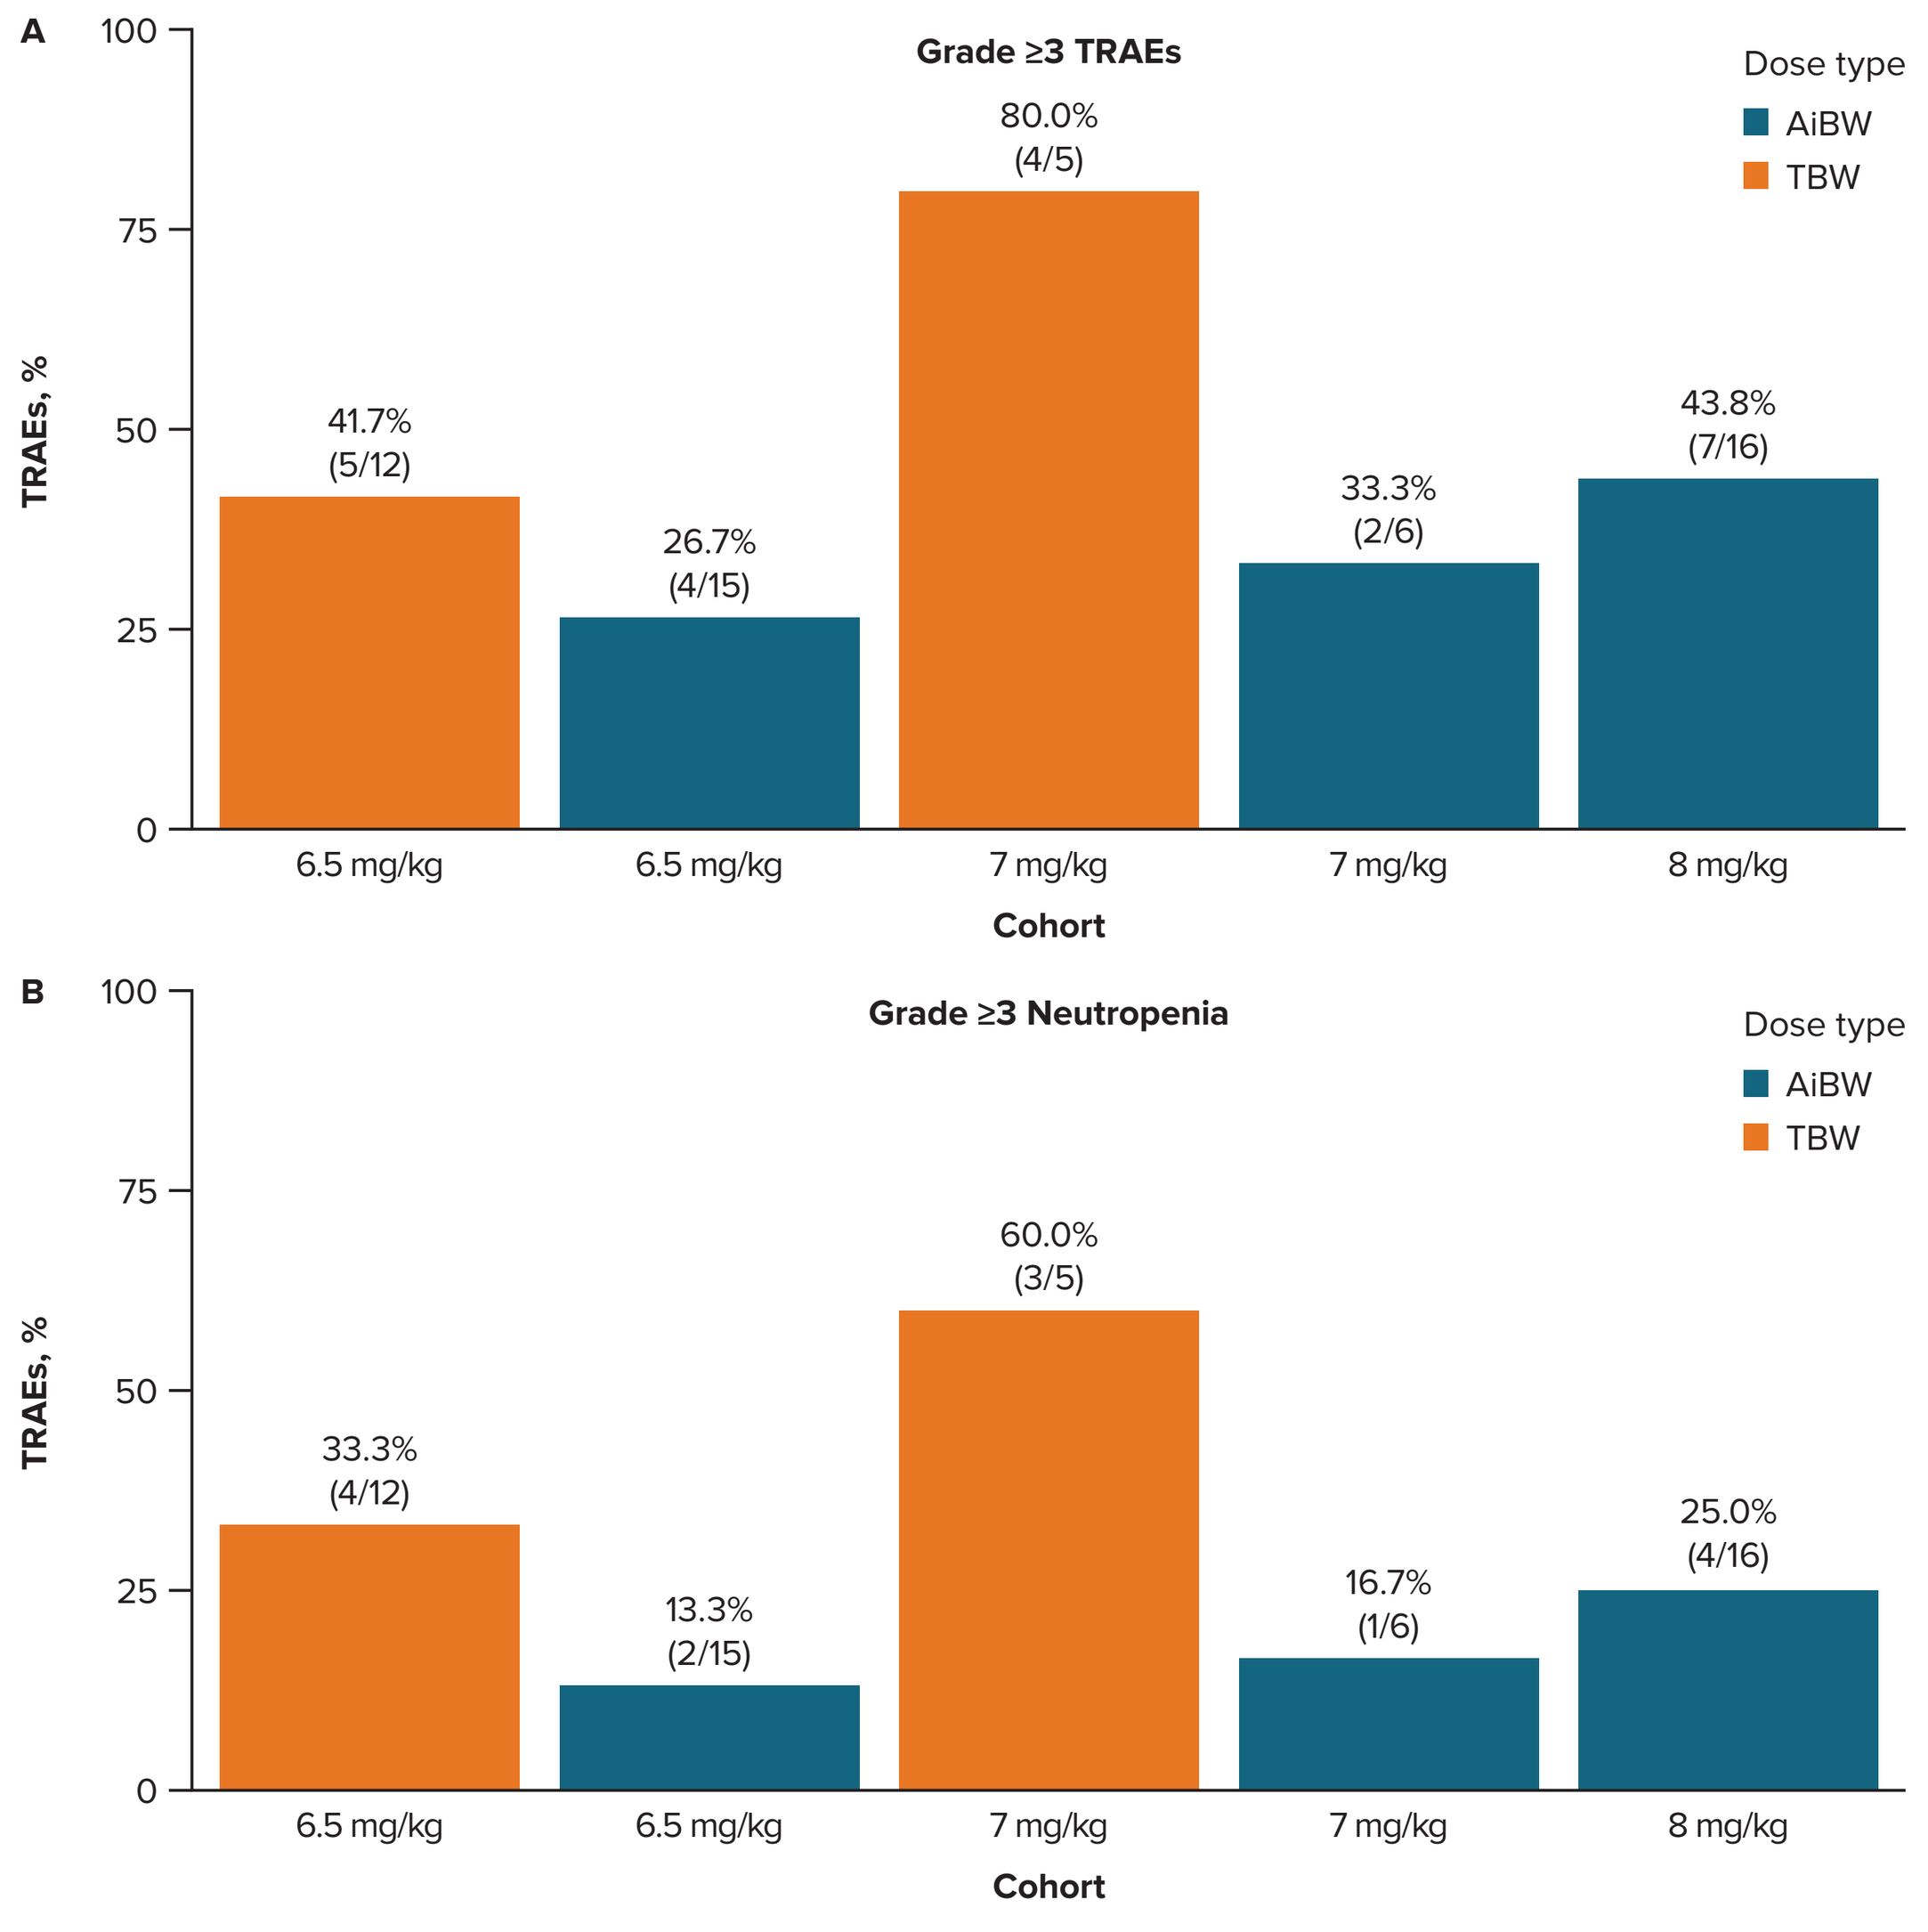
A
Grade ≥3 TRAEs
Dose type
AiBW
TBW
TRAEs, %
41.7% (5/12)
26.7% (4/15)
80.0% (4/5)
33.3% (2/6)
43.8% (7/16)
6.5 mg/kg
6.5 mg/kg
7 mg/kg
7 mg/kg
8 mg/kg
Cohort
B
Grade ≥3 Neutropenia
33.3% (4/12)
13.3% (2/15)
60.0% (3/5)
16.7% (1/6)
25.0% (4/16)

Abbreviations: AiBW, adjusted ideal body weight; TBW, total bodyweight; TRAE, treatment-related adverse event.

Figure 7. Exposure Safety Relationships Observed For Grade ≥3 TRAEs and Grade ≥3 Neutropenia

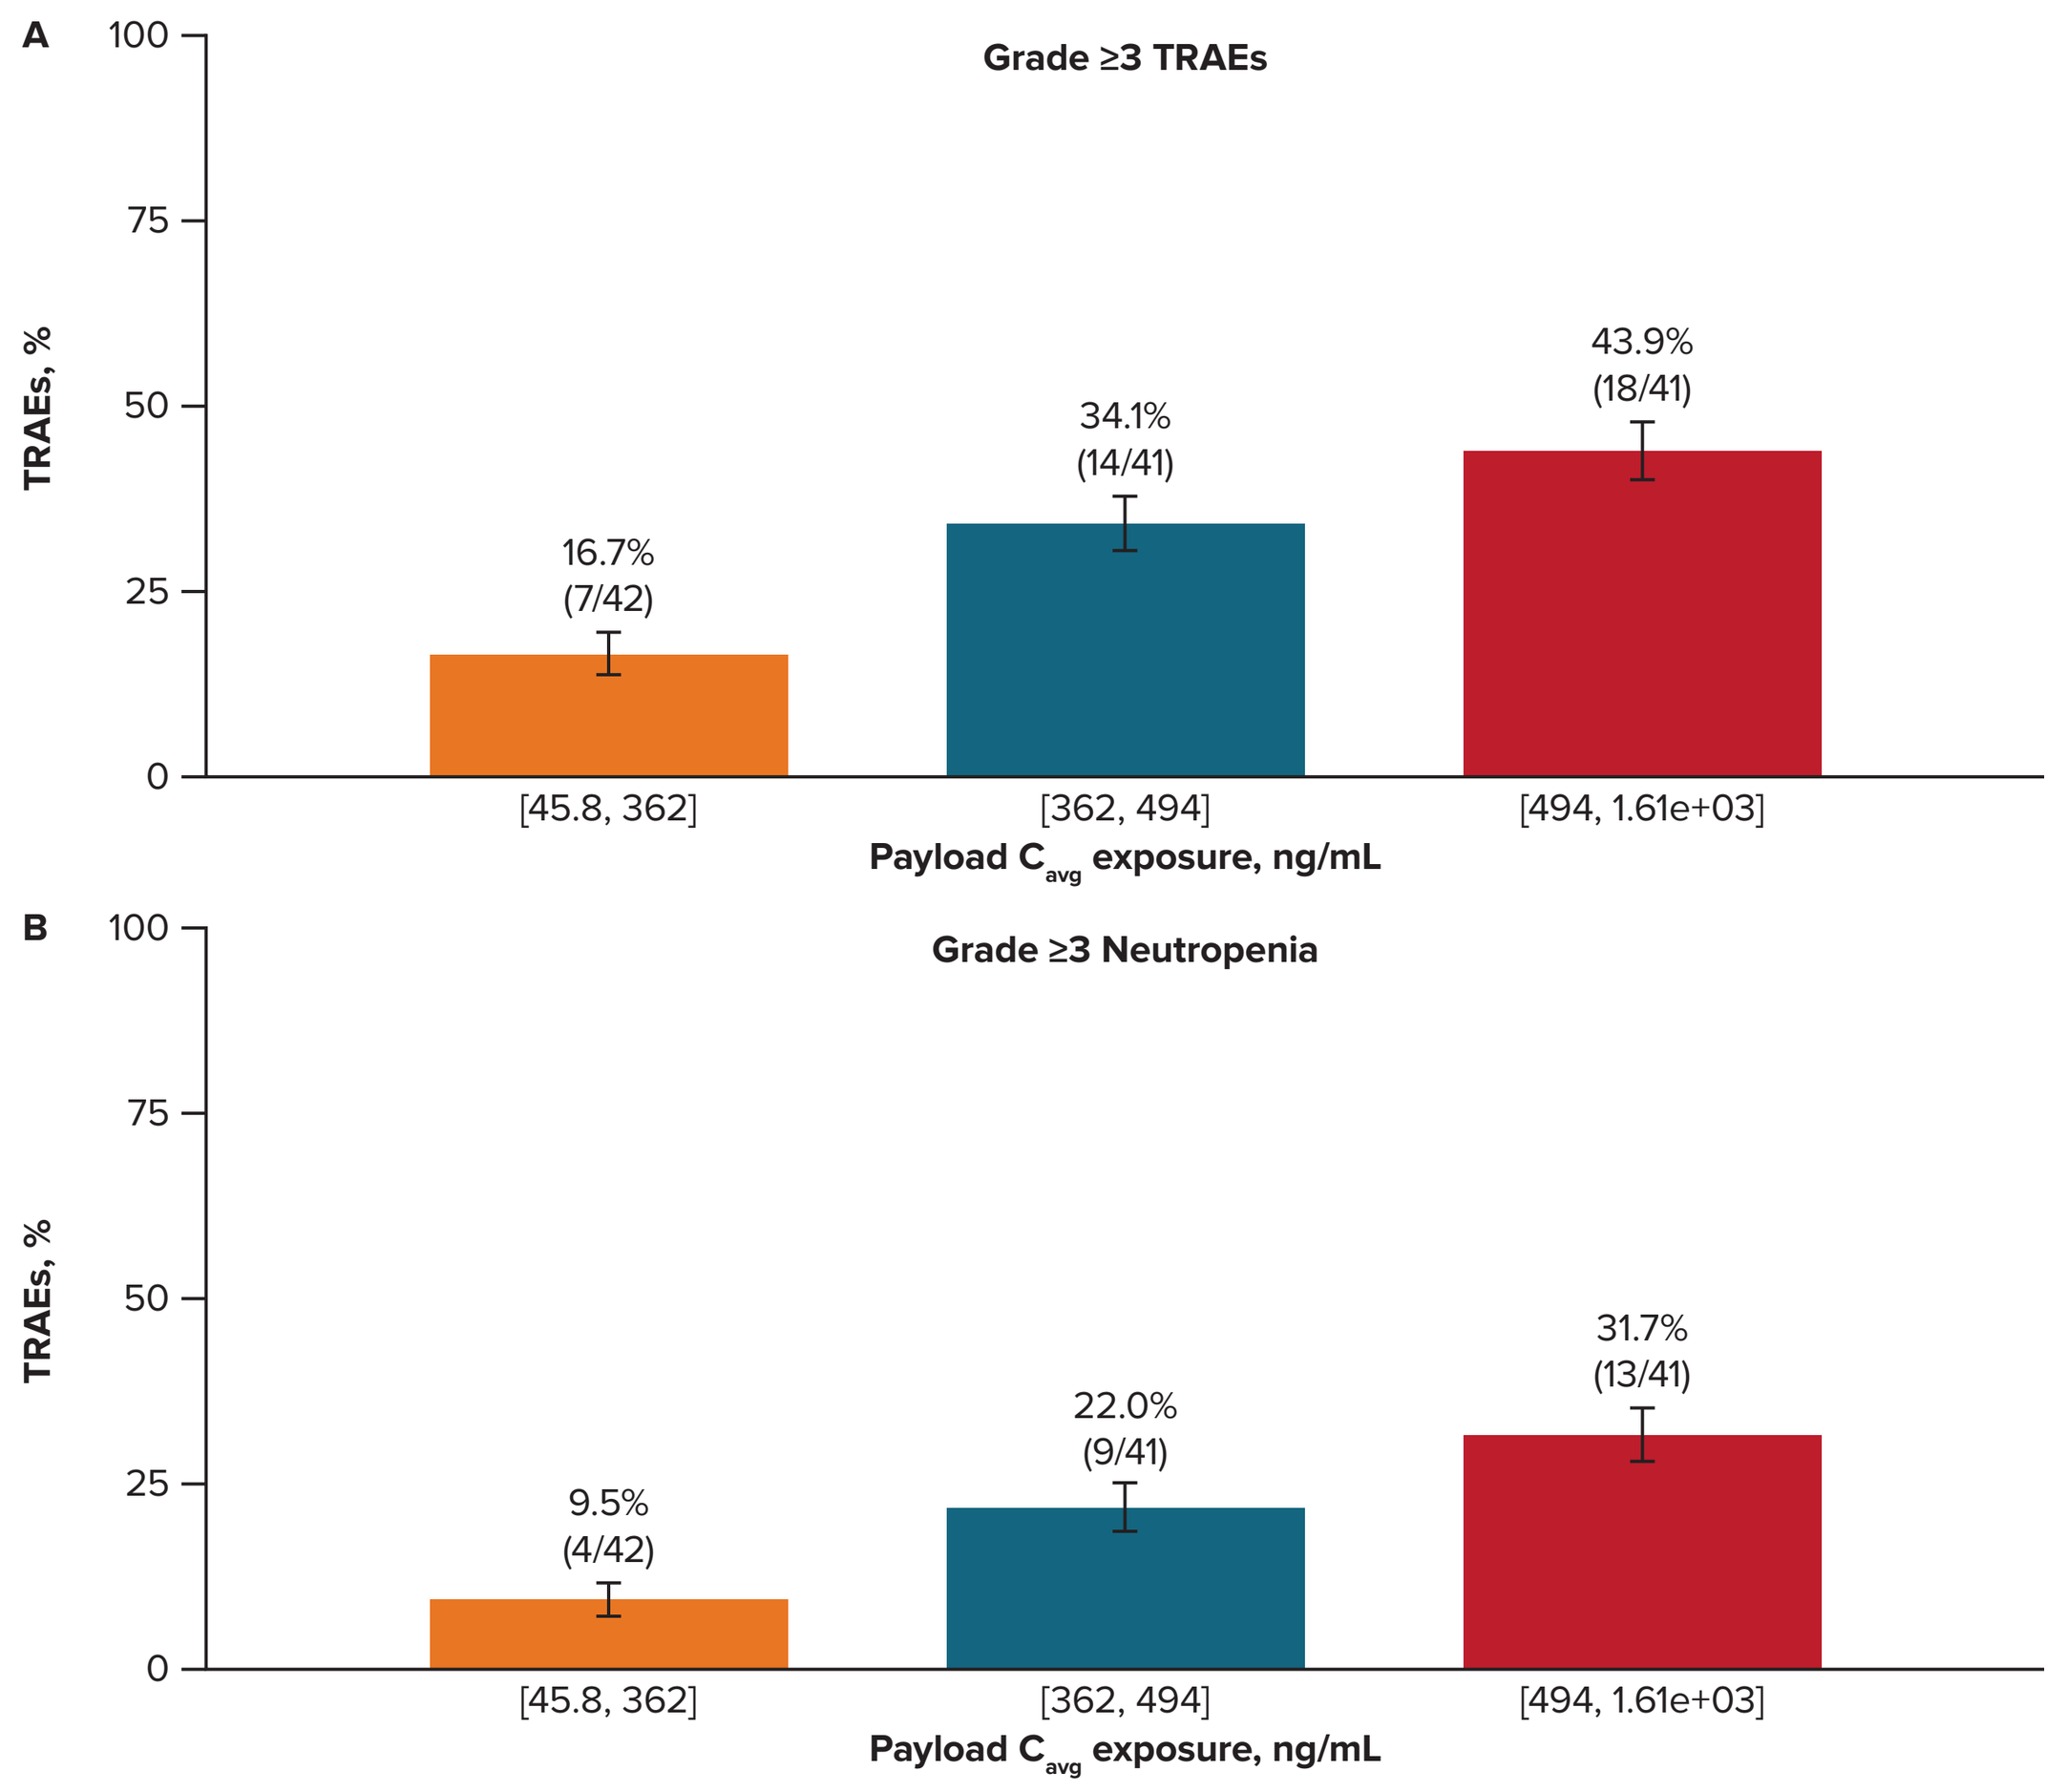
A
Grade ≥3 TRAEs
TRAEs, %
16.7% (7/42)
34.1% (14/41)
43.9% (18/41)
[45.8, 362]
[362, 494]
[494, 1.61e+03]
Payload Cavg exposure, ng/mL
B
Grade ≥3 Neutropenia
9.5% (4/42)
22.0% (9/41)
31.7% (13/41)

Abbreviations: TRAE, treatment-related adverse event.

Efficacy

- A positive relationship between ADC C_{avg} exposure and ORR was observed in the overall population, including patients in the OC subgroup of interest (**Figure 8**)

Figure 8. Increased Maximum Tumor Size Change From Baseline With Dose in Patients With OC

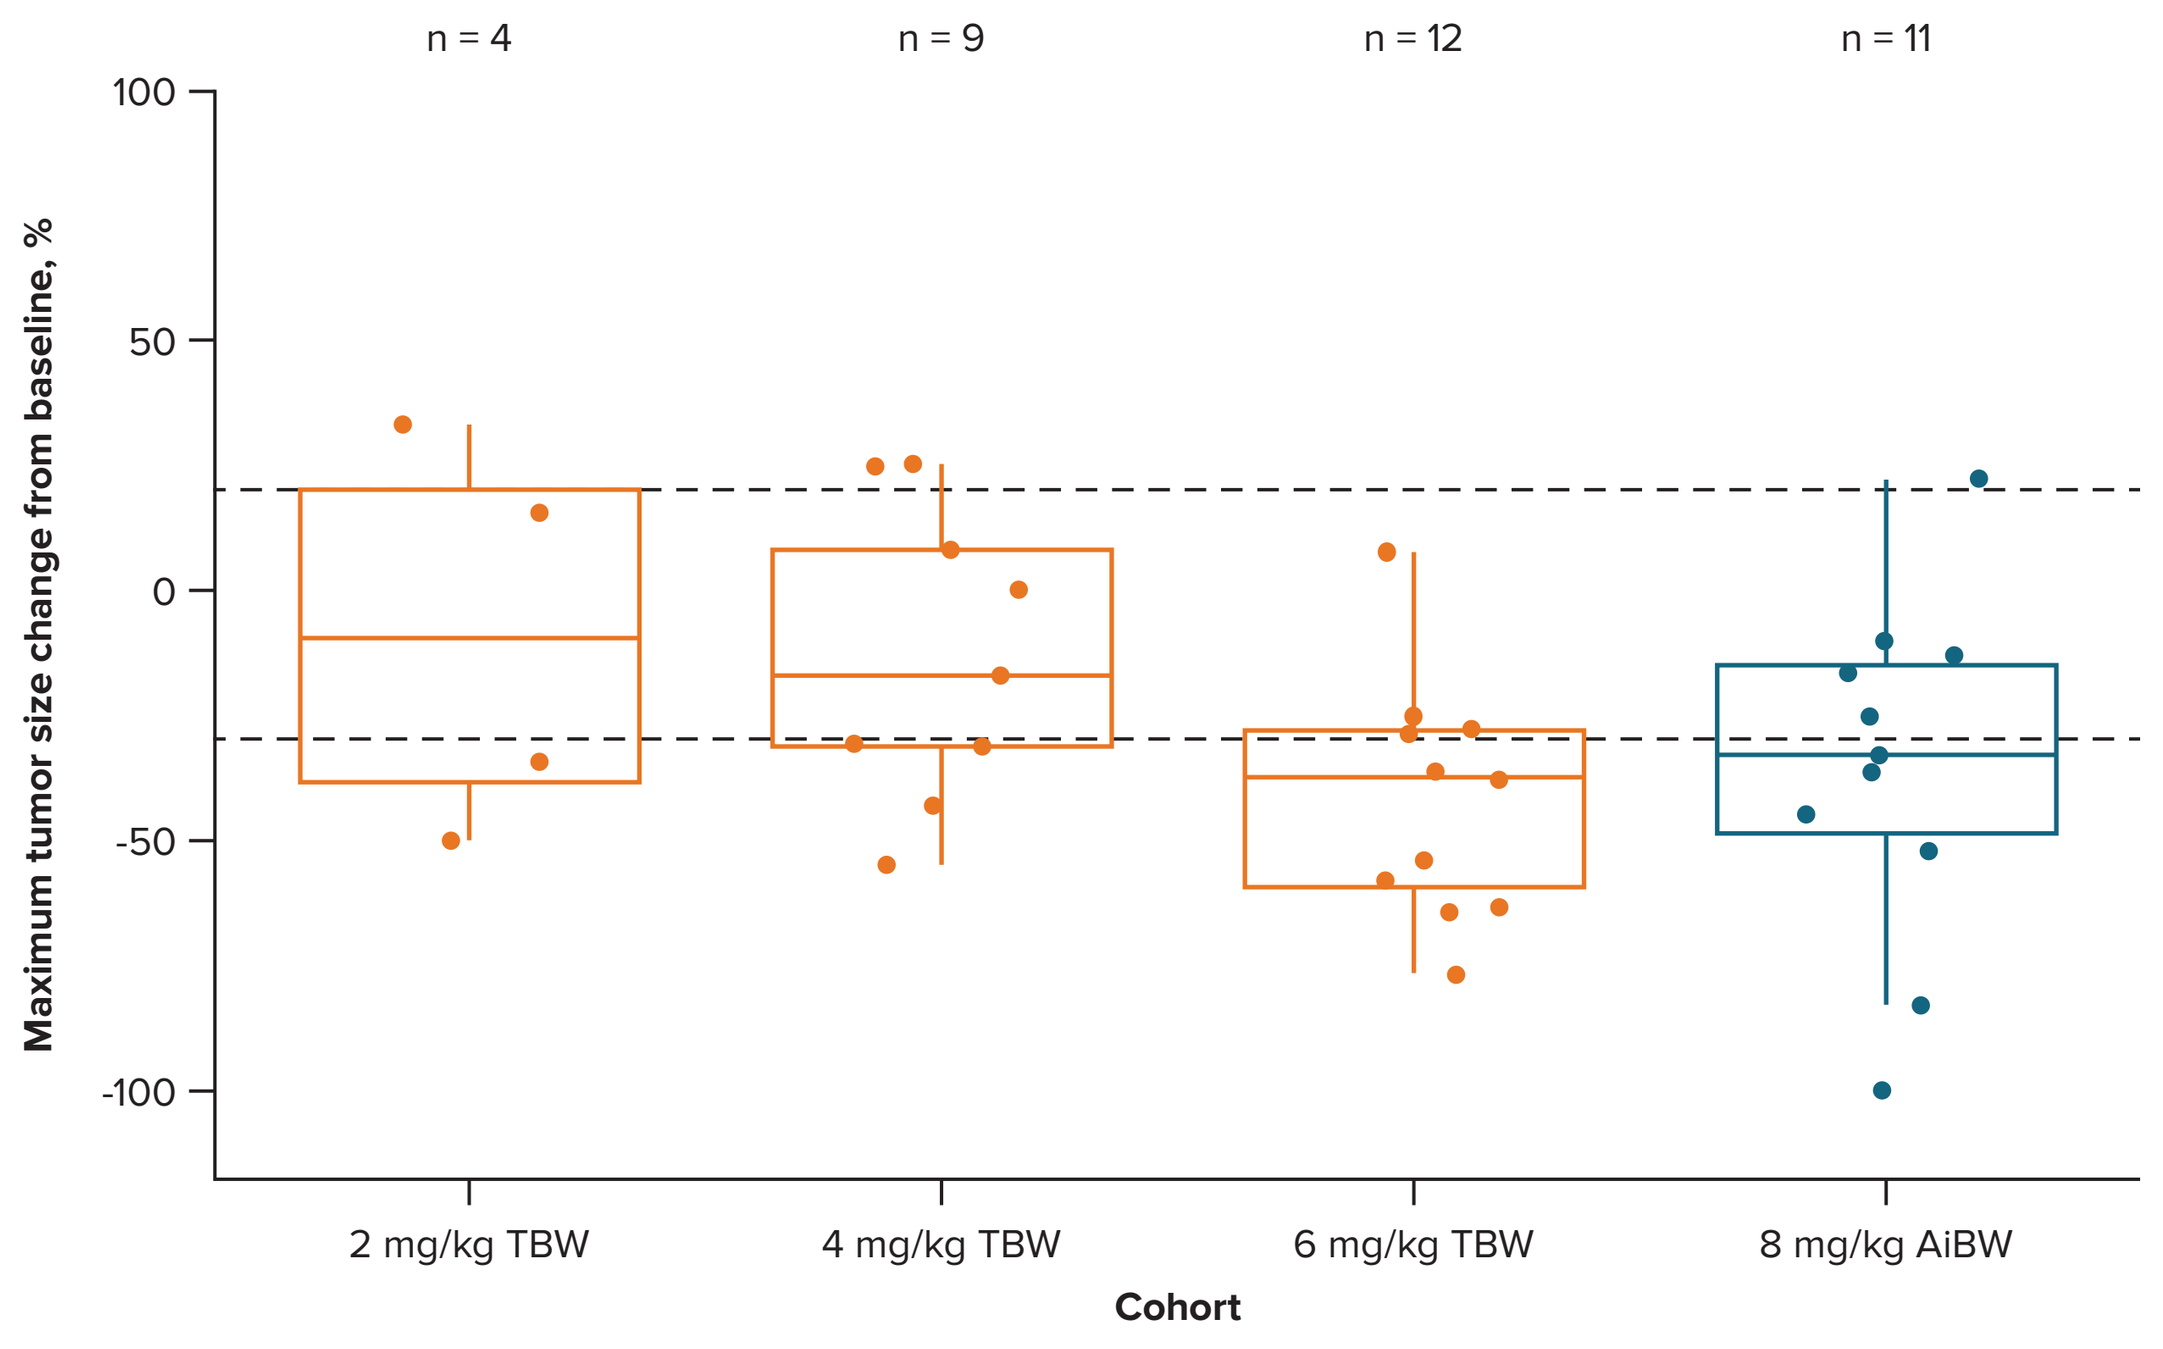
n = 4
n = 9
n = 12
n = 11
Maximum tumor size change from baseline, %
2 mg/kg TBW
4 mg/kg TBW
6 mg/kg TBW
8 mg/kg AiBW
Cohort

Abbreviations: AiBW, adjusted ideal body weight; OC, ovarian cancer; TBW, total bodyweight.

REFERENCES

1. Dawidowicz M et al. *Cancers.* 2024;16:2519.
2. Zhou L et al. *Front Immunol.* 2024;15:1426050.

DISCLOSURES

HG: Employment: BeOne Medicines, Ltd; Stock: BeOne Medicines, Ltd; Patents, royalties, or other intellectual property: BeOne Medicines, Ltd.

ACKNOWLEDGMENTS

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This study was sponsored by BeOne Medicines, Ltd. Editorial assistance was provided by Amiculum, and supported by BeOne Medicines.

PRESENTER: Hugh Giovinazzo
CORRESPONDENCE: Hugh Giovinazzo, hugh.giovinazzo@beonemed.com and Yen Lin Chia, yenlin.chia@beonemed.com

Presented at ASCO Annual Meeting; May 29-June 2, 2026; Chicago, IL, USA