Anti-CCR8 mediates long-lasting antitumor immunological memory and enhances anti-tumor immunity in immune-cold cancer through the combination with chemotherapy

Shuiqing Hu, Minjuan Deng, Wei Tan, Rui Liu, Amin Zhang, Lu Tan, Mengqian Chen, Pu Sun, Li Luo, Wen Li, Juan Yang, Jiyuan Zhang, Wei Jin, Zhirong Shen

Authors' Affiliation: BeiGene Global Research, P.R. China; Correspondence: shuiqing.hu@beigene.com, zhirong.shen@beigene.com

BeiGene

Abstract No. 4874

Abstract

Tumor-infiltrating regulatory T cells (Tregs) suppress CD8+ T cells and promote tumor progression. Tumor Tregs are elevated in various malignancies and are associated with poor prognosis. Targeting Tregs is an appealing strategy in cancer immunotherapy. Recent research has identified that the chemokine receptor CCR8 is preferentially expressed by tumor Tregs and labels highly suppressive Tregs. Thus, using anti-CCR8 mAbs to deplete tumor Tregs could be an effective and safe strategy that is being widely explored.

Although the anti-tumor efficacy of anti-CCR8 mAb has been reported, the dynamics and specific contributions of Tregs and CD8+ T cells to anti-tumor activity still need to be fully elucidated. Here, we explored the dynamic changes of Tregs and CD8+ T cells in the tumor microenvironment (TME) and evaluated their contributions to anti-tumor efficacy using a surrogate anti-CCR8. Immune-cold tumors are typically not sensitive to anti-PD1 treatment. Given that chemotherapy is the standard of care for most immune-cold tumors, we further evaluated the combination of chemotherapy drugs with surrogate anti-CCR8 in various syngeneic tumor models. We also assessed lymphocyte infiltration in the TME and found that the combination treatment can decrease Tregs and tumor-associated macrophages (TAMs), increase CD8+ T cells, and activate tumor-associated DCs (TADCs). These findings support the clinical application of combined CCR8 mAb and chemotherapy.

Dynamic Changes of Tumor Infiltrating Tregs and CD8+ T Cells upon Anti-CCR8 Treatment

Figure 1. CT26 and EMT6 tumor cells were inoculated and anti-CCR8 mAb was administered once via intraperitoneal injection. The percentages of tumor infiltrating Tregs and CD8+ T cells were analyzed by flow cytometry.

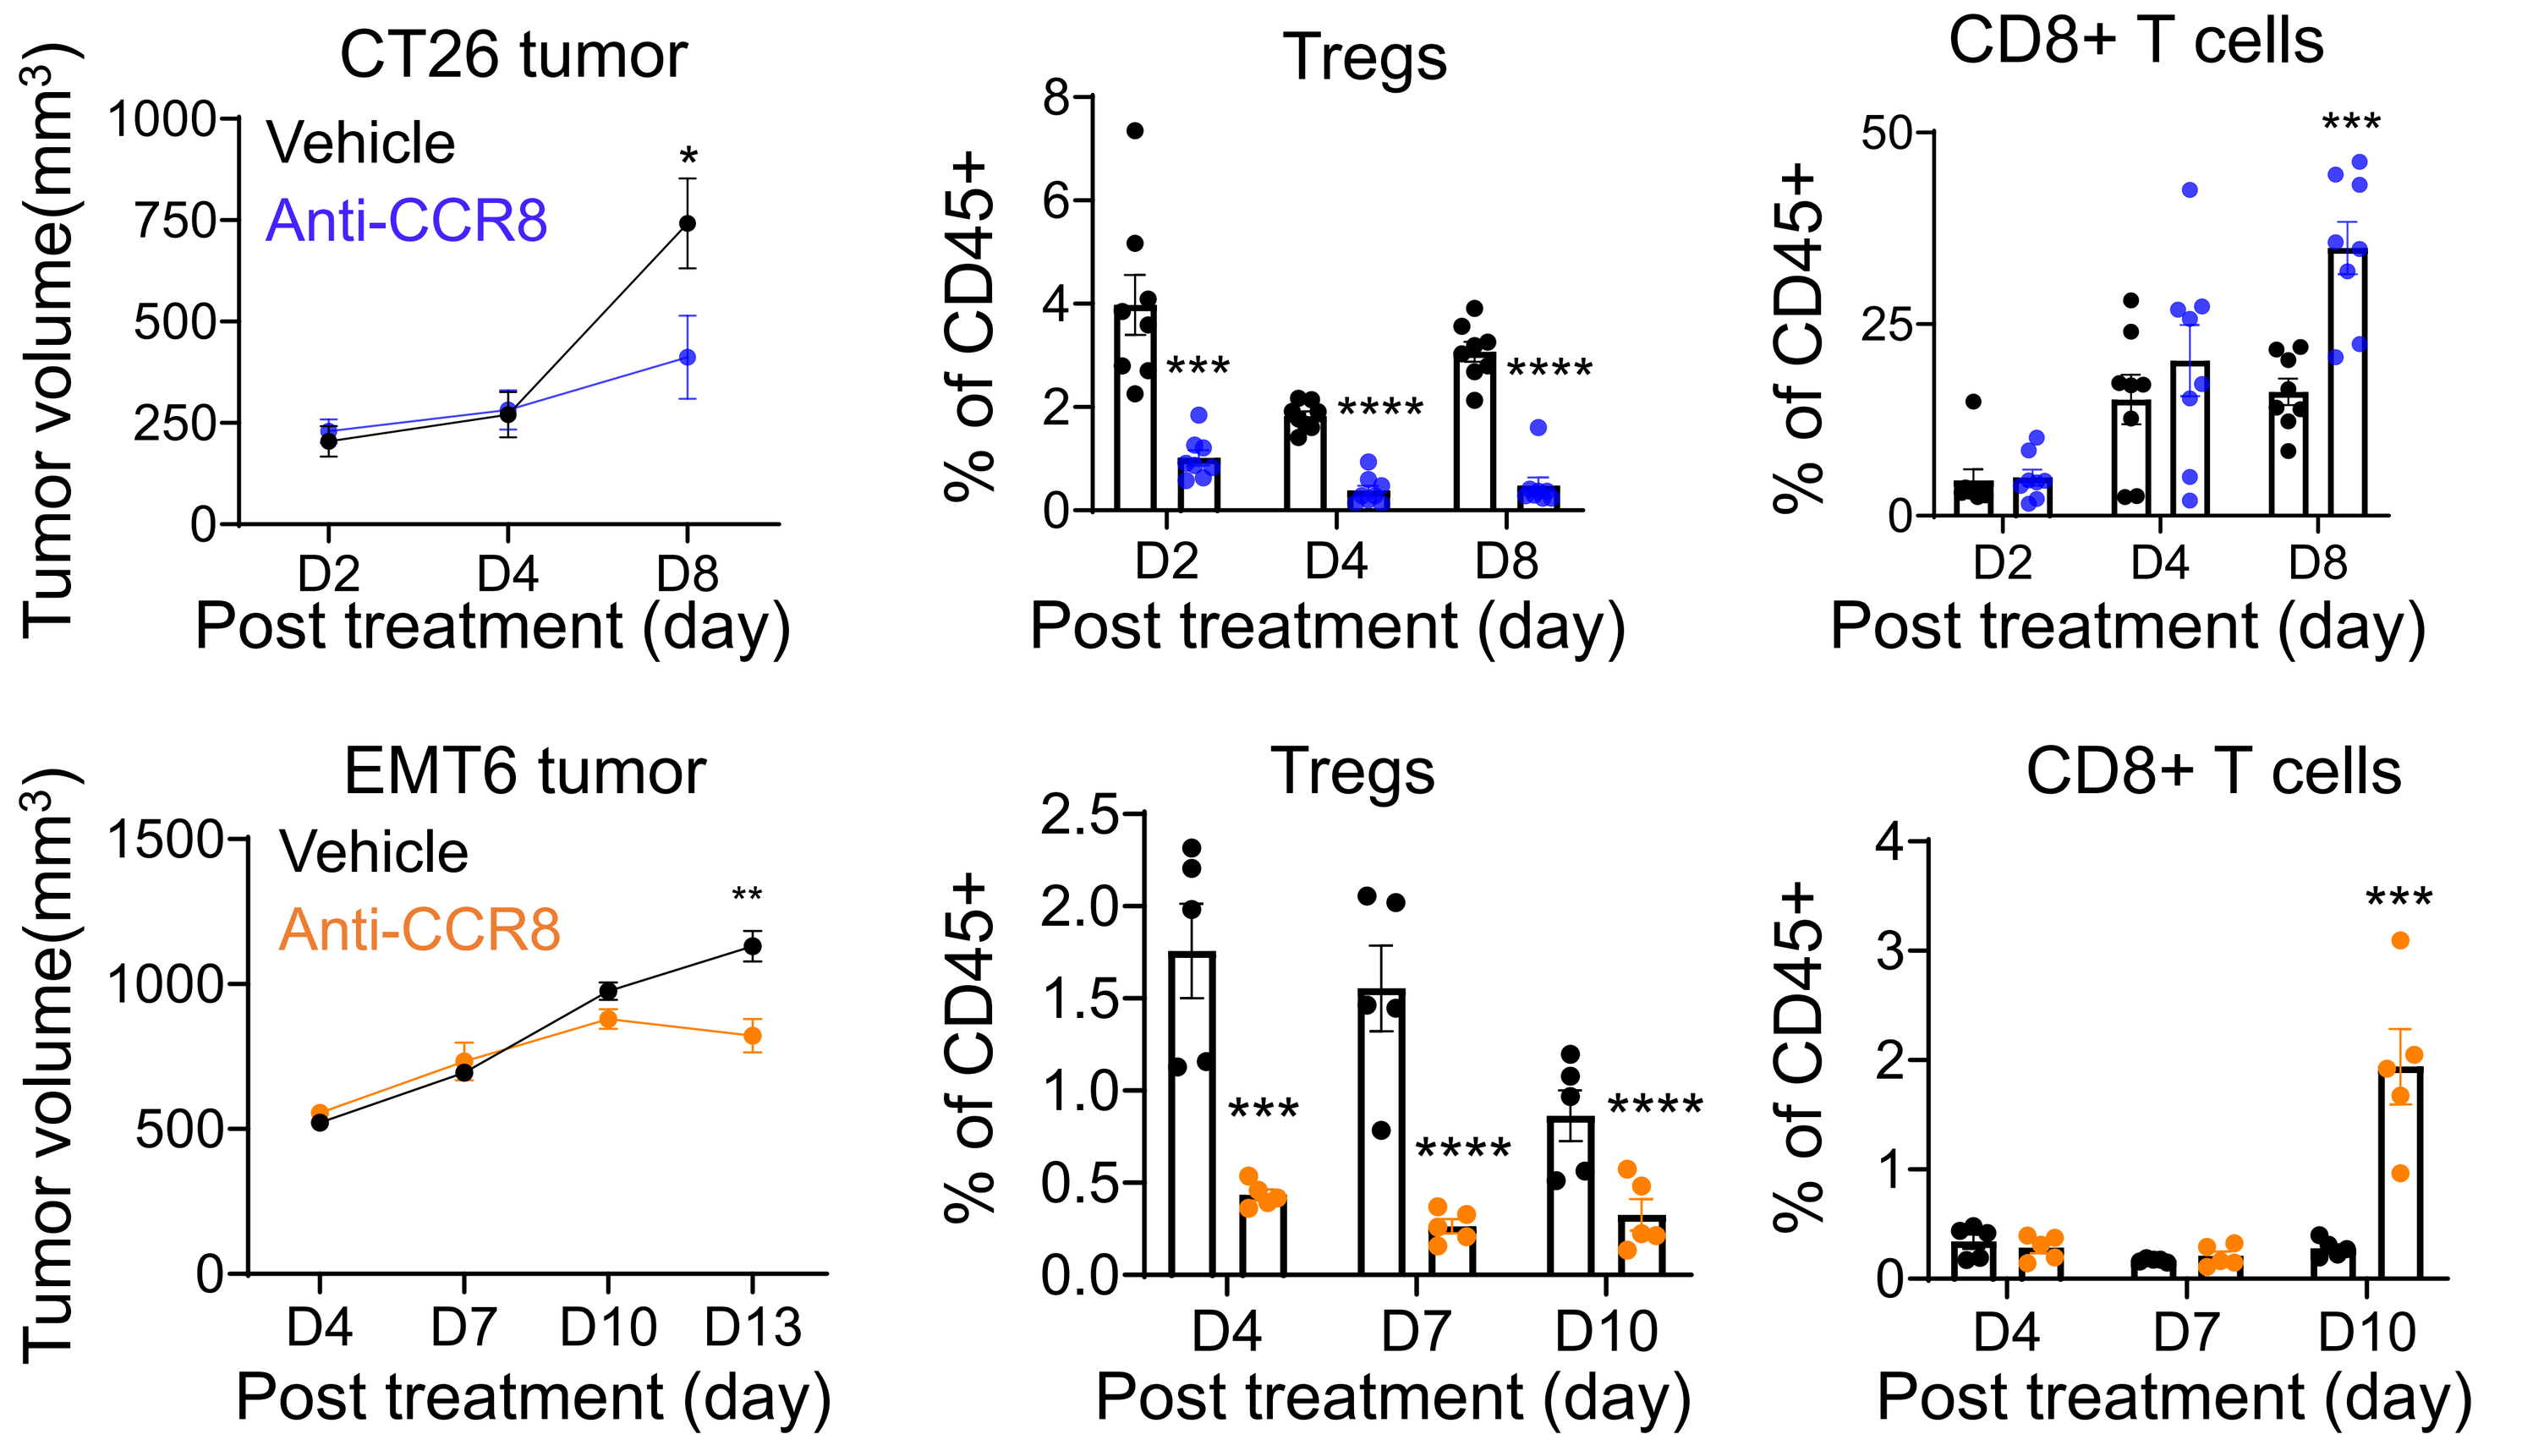
CT26 tumor
Tregs
CD8+ T cells
EMT6 tumor
Vehicle
Anti-CCR8
Tumor volume(mm³)
% of CD45+
Post treatment (day)
D2
D4
D8
D7
D10
D13

Correlation Analysis Showed that the Presence of CD8+ T cells Was Negatively Associated with Tumor Volume Reduction

Figure 2. Correlation analysis was performed between tumor-infiltrating CD8+ T cells and Tregs, and tumor volume in the EMT6 tumor model following treatment with anti-CCR8 mAb.

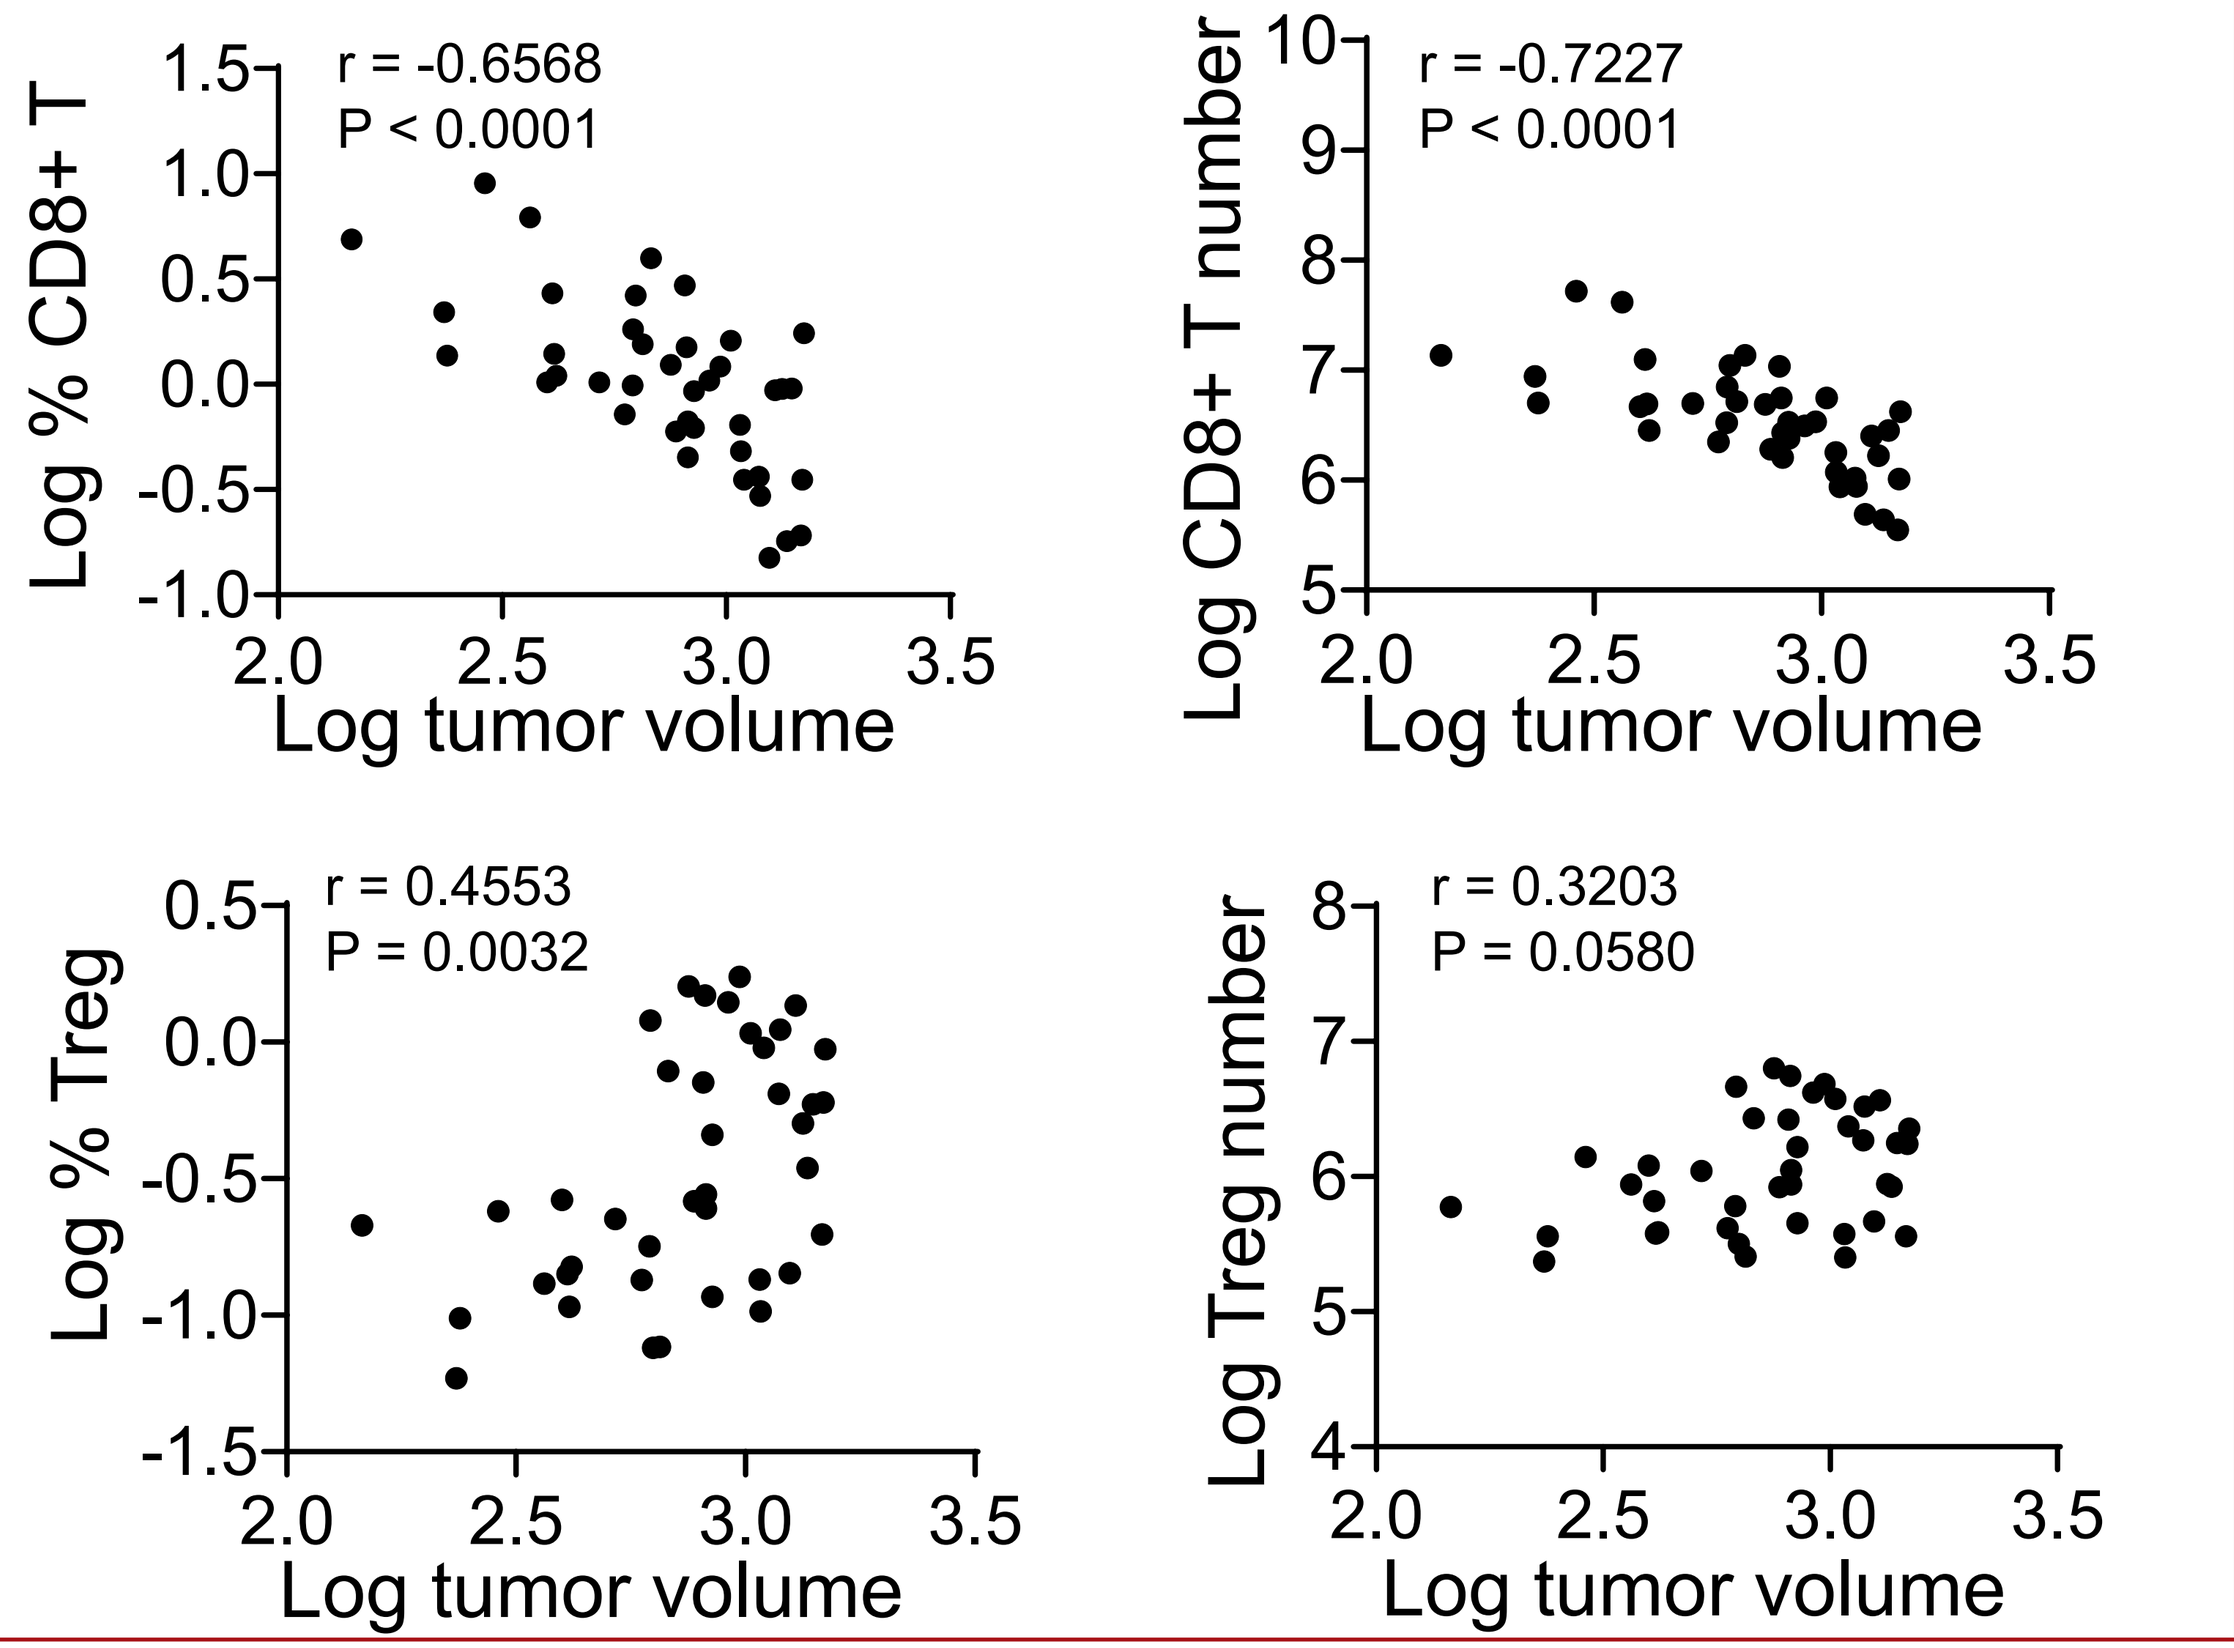
r = -0.6568
P < 0.0001
r = -0.7227
P < 0.0001
r = 0.4553
P = 0.0032
r = 0.3203
P = 0.0580
Log % CD8+ T
Log CD8+ T number
Log % Treg
Log Treg number
Log tumor volume

Depletion of CCR8+ Tregs can Induce Long-lasting Antitumor Immune Memory

Figure 3. (A) Induction of anti-tumor immune memory via anti-CCR8 therapy. Mice were inoculated with CT26 cells on the right flank and treated with anti-CCR8 (0.1, 1, and 3 mg/kg) or vehicle for a total of 3 times. Tumor volumes were recorded. Mice that achieved complete response (CR) were maintained for two months and then rechallenged with 50-fold more CT26 cells on the opposite side (left side) without any treatment. Meanwhile, two groups of naive mice were injected with the same number of CT26 cells as controls. When the tumor size reached 80 mm³, one group was treated with anti-CCR8 and the other group with vehicle. Tumor volumes were recorded, and tumor size were presented in the figure. (B) Induction of memory CD8+ T cells by anti-CCR8 treatment. Blood memory CD8+ T cells were analyzed in CT26 rechallenged CR mice (immune memory formed) and in naive CT26-bearing mice treated with either vehicle or anti-CCR8 (3 mg/kg). The CR mice exhibited significantly higher levels of memory CD8+ T cells compared to other groups, supporting the induction of anti-tumor immune memory by anti-CCR8.

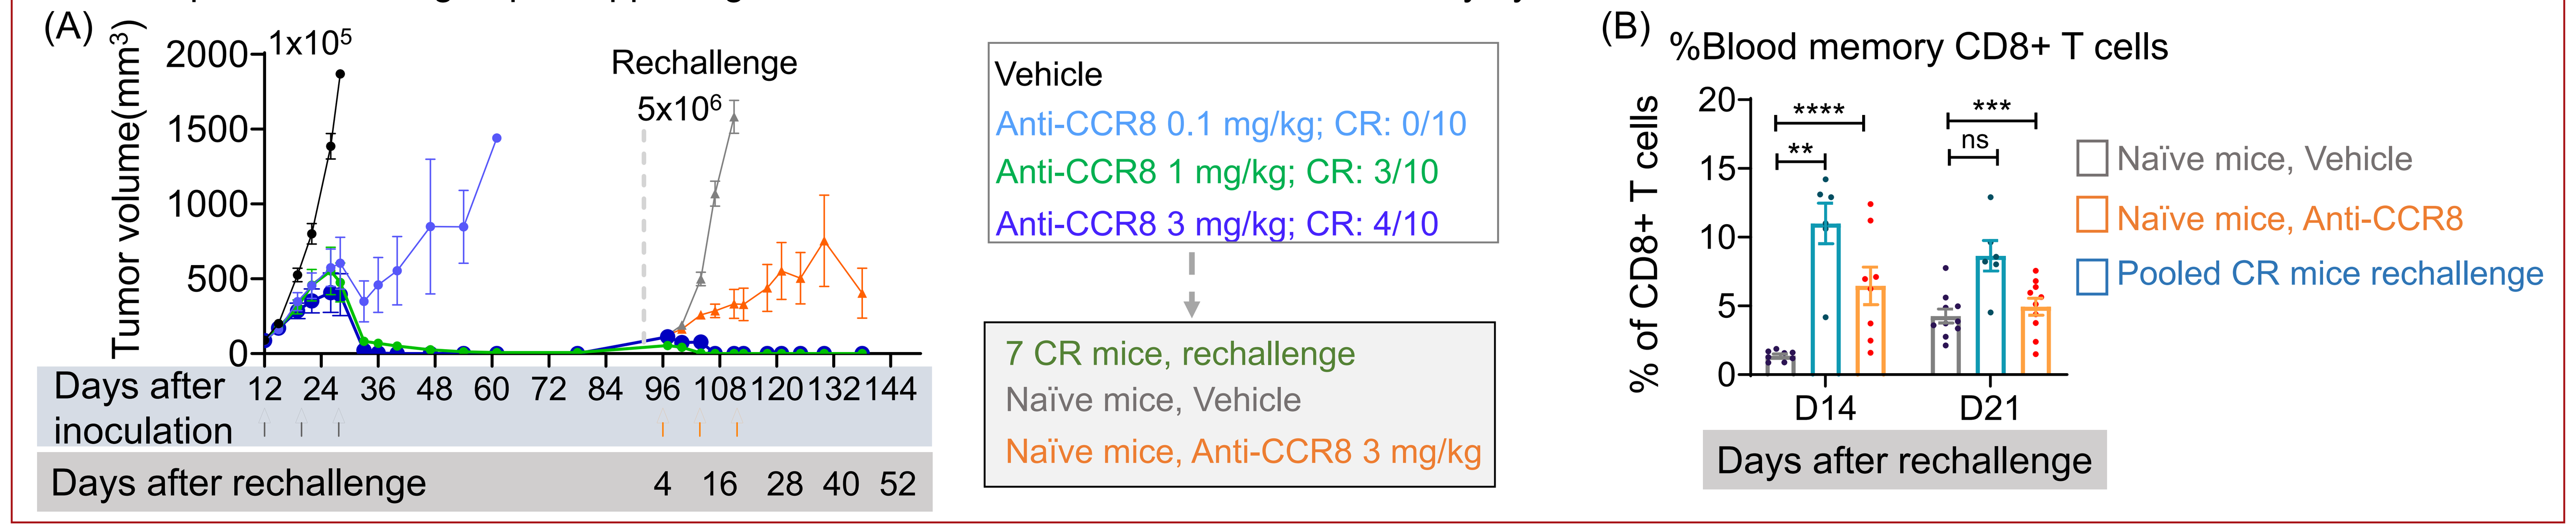
(A)
1x10⁵
Rechallenge
5x10⁶
Tumor volume(mm³)
Days after inoculation
Days after rechallenge
Vehicle
Anti-CCR8 0.1 mg/kg; CR: 0/10
Anti-CCR8 1 mg/kg; CR: 3/10
Anti-CCR8 3 mg/kg; CR: 4/10
7 CR mice, rechallenge
Naïve mice, Vehicle
Naïve mice, Anti-CCR8 3 mg/kg
(B)
%Blood memory CD8+ T cells
% of CD8+ T cells
D14
D21
Days after rechallenge
Naïve mice, Vehicle
Naïve mice, Anti-CCR8
Pooled CR mice rechallenge

Enhanced Antitumor Activity by Combining anti-CCR8 with Various Chemotherapies

Figure 4. Murine breast cancer 4T1 cells (A) and kidney cancer Renca cells (B) were subcutaneously implanted into Balb/c mice. Murine pancreatic cancer Pan02 cells (C) were subcutaneously implanted into C57BL/6J mice. When the tumor volume reached approximately 60~90 mm³, mice were randomly assigned to different treatment groups and administered the indicated treatments. Anti-mouse CCR8 mAb (3 mg/kg), Oxaliplatin (1.5 mg/kg) and Gemcitabine (100 mg/kg) were administered once every week via intraperitoneal injection. Docetaxel (30 mg/kg) was administered twice every week via intraperitoneal injection. The treatment dates are indicated by vertical arrows. Tumor growth curves are expressed as mean ± SEM, and tumor growth inhibition is shown in the table.

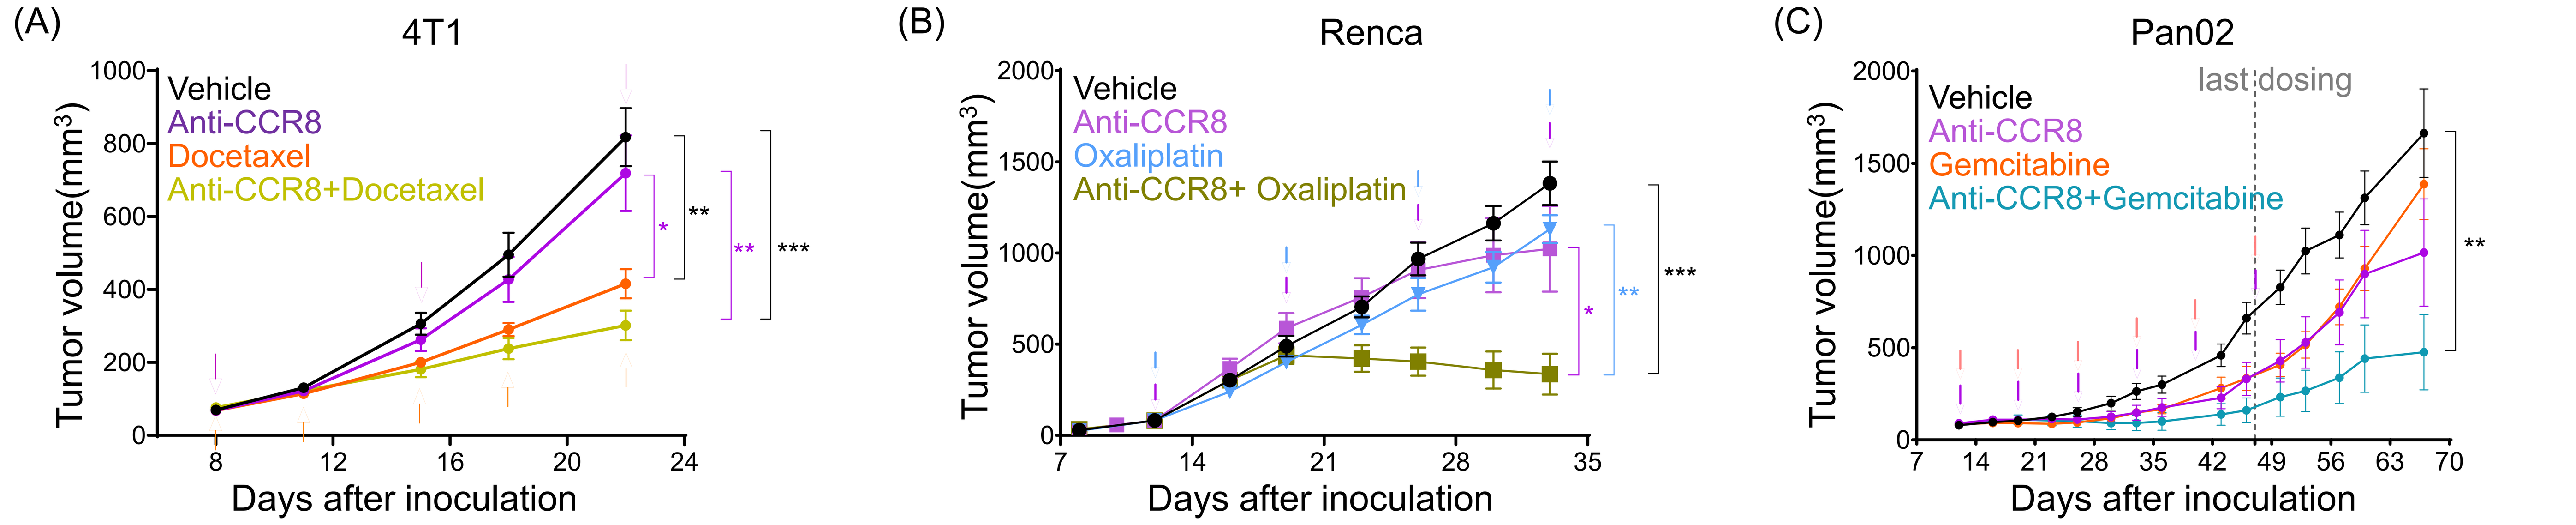
(A)
4T1
Vehicle
Anti-CCR8
Docetaxel
Anti-CCR8+Docetaxel
(B)
Renca
Vehicle
Anti-CCR8
Oxaliplatin
Anti-CCR8+ Oxaliplatin
(C)
Pan02
last dosing
Vehicle
Anti-CCR8
Gemcitabine
Anti-CCR8+Gemcitabine
Tumor volume(mm³)
Days after inoculation

4T1	TGI at Day 22
Vehicle	-
Anti-CCR8	13.83%
Docetaxel	60.01%
Anti-CCR8 + Docetaxel	71.13%

Renca	TGI at Day 33
Vehicle	-
Anti-CCR8	26.32%
Oxaliplatin	18.32%
Anti-CCR8 + Oxaliplatin	77.65%

Pan02	TGI at Day 67
Vehicle	-
Anti-CCR8	41.53%
Gemcitabine	17.77%
Anti-CCR8 + Gemcitabine	75.47%

Decreased Tumor-infiltrating Tregs and TAMs, Increased CD8+ T Cells, and Activated TADCs Following Combined Treatment with Anti-CCR8 and Gemcitabine

Figure 5. Murine pancreatic cancer Pan02 cells were subcutaneously implanted into C57BL/6J mice. When tumor size was around 90 mm³, Pan02 tumor-bearing mice were randomly assigned to different treatment groups and treated with vehicle, anti-CCR8 (3 mg/kg), Gemcitabine (100 mg/kg), or the combination once weekly via intraperitoneal injection for a total of four doses. Pan02 tumors were collected, and tumor infiltrating lymphocytes were analyzed by flow cytometry.

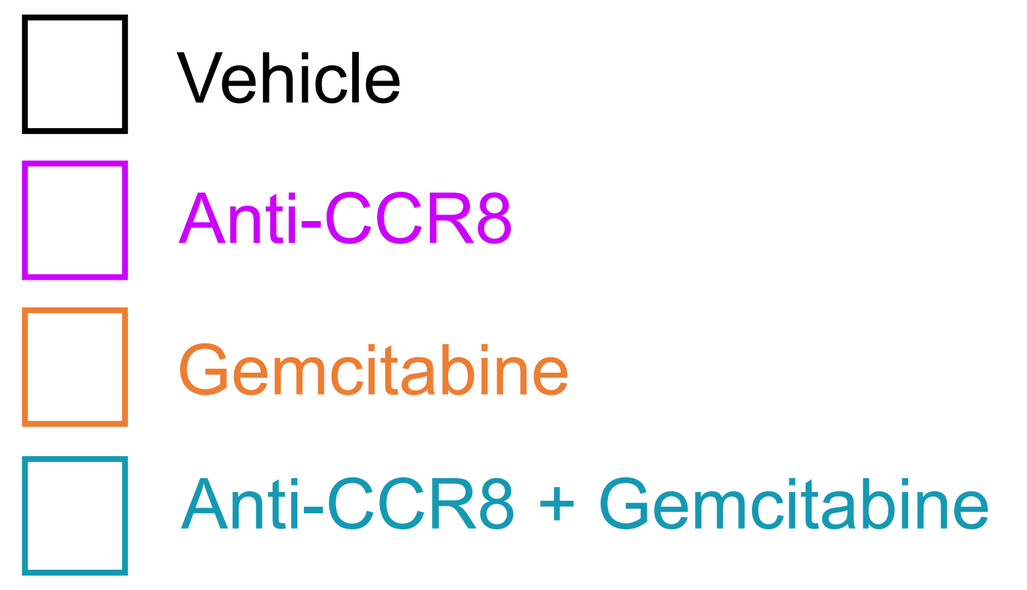
Vehicle
Anti-CCR8
Gemcitabine
Anti-CCR8 + Gemcitabine

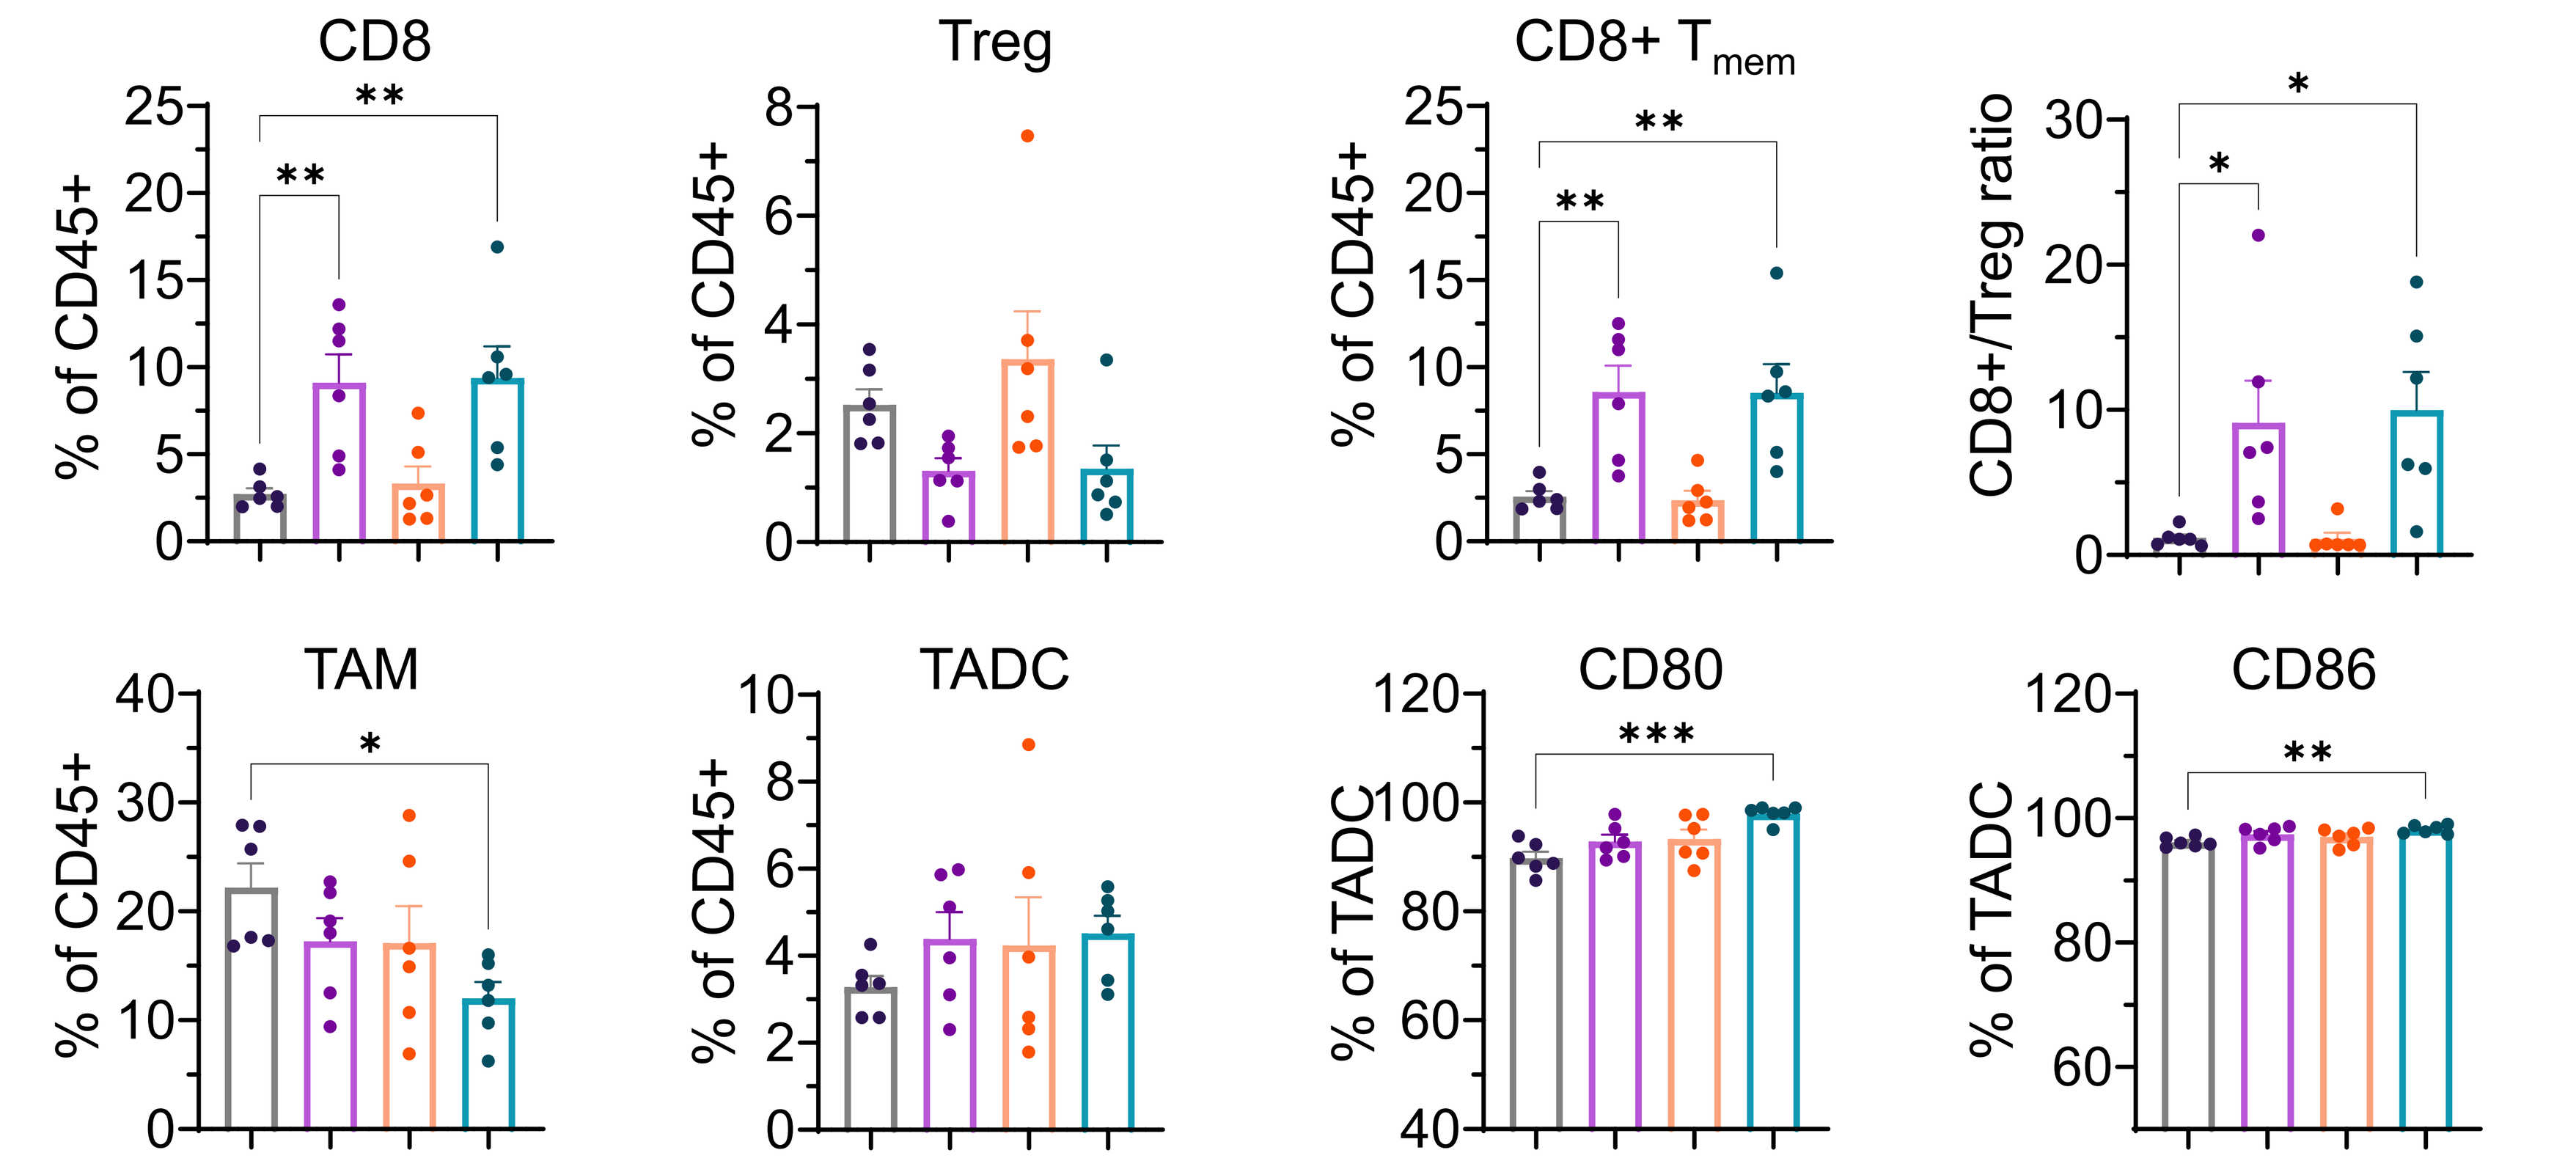
CD8
Treg
CD8+ Tmem
CD8+/Treg ratio
TAM
TADC
CD80
CD86
% of CD45+
% of TADC

Conclusion

Our study elucidated the underlying anti-tumor mechanisms of anti-CCR8 mAb and provided strategies for its clinical application.

- Tumor-infiltrating Tregs were rapidly depleted within two days, and CD8+ T cells increased after one week following treatment with anti-CCR8, which coincided with the observed tumor growth inhibition.
- Tumor volume was negatively correlated with the percentage and number of tumor-infiltrating CD8+ T cells, suggesting that an increased presence of CD8+ T cells in the tumor may indicate an active anti-tumor immune response.
- Anti-CCR8 can induce long-lasting immune memory, as evidenced by the active rejection of rechallenged tumor cells and a significant increase in memory CD8+ T cells in blood.
- Combining anti-CCR8 with chemotherapies results in synergistic anti-tumor efficacy in various immune-cold tumor models, supporting its potential for clinical application.
- Combined treatment with anti-CCR8 and gemcitabine in Pan02 model led to a decrease in Tregs and macrophages within the tumor, while CD8+ T cells increased and TADCs exhibited an activated phenotype. These pharmacodynamic changes support the combination of anti-CCR8 and chemotherapy.

AACR Annual Meeting 2025 April 25-30, 2025. Chicago, Illinois

AACR 2025