First-Line Tislelizumab Plus Chemotherapy in Gastric/Gastroesophageal Junction Cancer: RATIONALE-305 Asian Subgroup

Keun-Wook Lee,[1] Ken Kato,[2] Yuxian Bai,[3] Jianhua Shi,[4] Jufeng Wang,[5] Hongming Pan,[6] Sun Young Rha,[7] Ruixing Zhang,[8] Hidekazu Hirano,[2] Kensei Yamaguchi,[9] Zengqing Guo,[10] Yi Ba,[11] Lei Yang,[12] Hiroshi Tsukuda,[13] Yaling Xu,[14] Tao Sheng,[15] Silu Yang,[14] Liyun Li,[14] Do-Youn Oh,[16] Rui-Hua Xu[17]

[1]Department of Internal Medicine, Seoul National University College of Medicine, Seoul National University Bundang Hospital, Seongnam, Republic of Korea; [2]Department of Gastrointestinal Medical Oncology, National Cancer Center Hospital, Tokyo, Japan; [3]Department of Gastrointestinal Oncology, Harbin Medical University Cancer Hospital, Harbin, China; [4]Department of Oncology, Linyi Cancer Hospital, Linyi, China; [5]Department of Medical Oncology, Henan Cancer Hospital, Zhengzhou, China; [6]Department of Oncology, Sir Run Run Shaw Hospital, Zhejiang University School of Medicine, Hangzhou, China; [7]Department of Internal Medicine, Yonsei Cancer Center, Yonsei University Health System, Seoul, Republic of Korea; [8]Department of Gastroenterology and Hepatology, Fourth Hospital of Hebei Medical University, Shijiazhuang, China; [9]Department of Gastroenterological Chemotherapy, Cancer Institute Hospital of JFCR, Tokyo, Japan; [10]Department of Medical Oncology, Fujian Cancer Hospital, Fuzhou, China; [11]Department of GI Medical Oncology, Tianjin Medical University Cancer Institute & Hospital, Tianjin, China; [12]Department of Oncology, Nantong Cancer Hospital, Nantong, China; [13]Department of Medical Oncology, Izumi Municipal Hospital, Izumi, Japan; [14]Department of Clinical Development, BeiGene (Beijing) Co., Ltd, Beijing, China; [15]Department of Biostatistics, BeiGene, Ltd, Boston, MA, United States; [16]Department of Internal Medicine, Seoul National University Hospital, Cancer Research Institute, Seoul National University College of Medicine, Seoul, Republic of Korea; [17]Department of Medical Oncology, Sun Yat-sen University Cancer Center State Key Laboratory of Oncology in South China, Collaborative Innovation Center of Cancer Medicine, Guangzhou, China.

Abstract no. ABST-000186. Presented at the 17th Annual Meeting of the Korean Society of Medical Oncology (KSMO) & 2024 International Conference; September 26-27, 2024; Seoul, Korea

Conclusions

- **In Asian patients in the RATIONALE-305 study**
 - **Tislelizumab plus chemotherapy showed an improvement in overall survival (OS) versus placebo plus chemotherapy in both the intent-to-treat (ITT) and programmed death-ligand 1 (PD-L1) score ≥5% populations**
 - **Additionally, tislelizumab plus chemotherapy demonstrated improved progression-free survival (PFS), favorable response rates, and more durable antitumor responses versus placebo plus chemotherapy**
 - **Tislelizumab plus chemotherapy also showed a manageable safety profile, with no new safety signals identified**
- **The efficacy and safety results in the Asian patient subgroup were consistent with the results in the overall study population, suggesting this combination can be a first-line treatment option for Asian patients with advanced gastric/gastroesophageal junction cancer (GC/GEJC)[1]**

Introduction

- Gastric cancer is one of the leading causes of cancer-related deaths worldwide; Asia has a notably higher incidence and mortality rate of GC, and the disease is of particular concern in China, South Korea, and Japan[2-5]
- Prior to the introduction of immunotherapy, platinum plus fluoropyrimidine chemotherapy was the standard first-line therapy for advanced GC/GEJC, with a median OS of less than 12 months[6-8]
- The RATIONALE-305 study (NCT03777657) met its primary endpoint, showing significant improvement in OS with tislelizumab plus chemotherapy versus placebo plus chemotherapy in the PD-L1 ≥5% population at interim analysis, and in the ITT population at final analysis, with favorable PFS[1,9]
- Overall, results of the final analysis supported tislelizumab plus chemotherapy as a potential first-line treatment option for patients with advanced GC/GEJC[1]
- Here, we present results from the Asian patient subgroup of the RATIONALE-305 study at final analysis

Methods

- Of 997 randomized patients, 748 (tislelizumab with chemotherapy: n=376; placebo with chemotherapy: n=372) were enrolled from Asia, of whom 403 had a PD-L1 score of ≥5% (**Figure 1**)
- The Asian subgroup comprised patients from China (including Taiwan), Japan, and South Korea
- As of the data cutoff for the final analysis (February 28, 2023), median study follow-up in the Asian subgroup was 14.5 months (range, 0.1-50.1) and minimum study follow-up in this subgroup was 24.6 months

Figure 1. Study Design and Patient Population

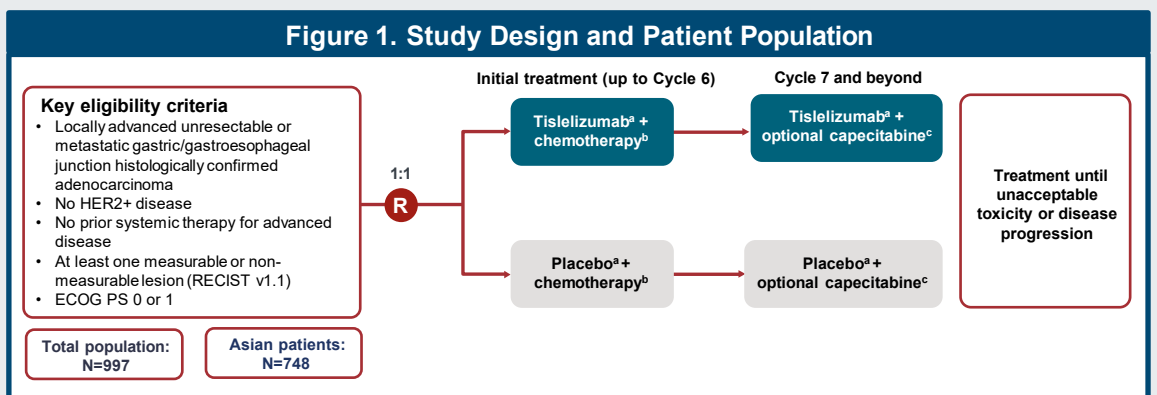
Initial treatment (up to Cycle 6)
Cycle 7 and beyond
Key eligibility criteria
Locally advanced unresectable or metastatic gastric/gastroesophageal junction histologically confirmed adenocarcinoma
No HER2+ disease
No prior systemic therapy for advanced disease
At least one measurable or non-measurable lesion (RECIST v1.1)
ECOG PS 0 or 1
1:1
R
Tislelizumab[a] + chemotherapy[b]
Tislelizumab[a] + optional capecitabine[c]
Placebo[a] + chemotherapy[b]
Placebo[a] + optional capecitabine[c]
Treatment until unacceptable toxicity or disease progression
Total population: N=997
Asian patients: N=748

Stratification factors
- Regions of enrollment
- Peritoneal metastasis
- PD-L1 expression score (≥5% vs <5%)[d]
- Investigator-chosen chemotherapy (XELOX or FP)

Endpoints
- **Primary endpoint:** OS in PD-L1 score ≥5% and ITT populations
- **Secondary endpoints:** Progression-free survival, objective response rate, duration of response, and safety

[a]Tislelizumab 200 mg or placebo Q3W (day 1). [b]Oxaliplatin 130 mg/m² IV (day 1) and oral capecitabine 1000 mg/m² twice daily (14 consecutive days from day 1) Q3W (XELOX), or cisplatin 80 mg/m² IV (day 1) and FP 800 mg/m²/day IV (days 1-5) Q3W. [c]Capecitabine as maintenance therapy was optional and only for XELOX-treated patients. [d]PD-L1 score was determined using the VENTANA PD-L1 (SP263) assay by tumor area positivity score.
ECOG PS, Eastern Cooperative Oncology Group Performance Status; FP, 5-fluorouracil; HER2, human epidermal growth factor receptor 2; ITT, intent-to-treat; IV, intravenous; OS, overall survival; PD-L1, programmed death-ligand 1; Q3W, once every 3 weeks; R, randomized; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1.

Results

- Improved OS differences of 2.1 and 4.6 months in favor of tislelizumab plus chemotherapy versus placebo plus chemotherapy were observed in the overall ITT and PD-L1 score ≥5% populations at final and interim analysis, respectively (**Figure 2**)[9]

Figure 2. Overall Survival: Overall Population

ITT Population at Final Analysis

	Events, n (%)	Median OS (95% CI), Months[a]	HR (95% CI)[a] P Value[b]
TIS + chemo (n=501)	370 (73.9)	15.0 (13.6-16.5)	0.80 (0.70-0.92)
PBO + chemo (n=496)	406 (81.9)	12.9 (12.1-14.1)	0.0011

PD-L1 Score ≥5% Population at Interim Analysis[9]

	Events, n (%)	Median OS (95% CI), Months[c]	HR (95% CI)[c] P Value[b]
TIS + chemo (n=274)	130 (47.4)	17.2 (13.9-21.3)	0.74 (0.59-0.94)
PBO + chemo (n=272)	161 (59.2)	12.6 (12.0-14.4)	0.0056

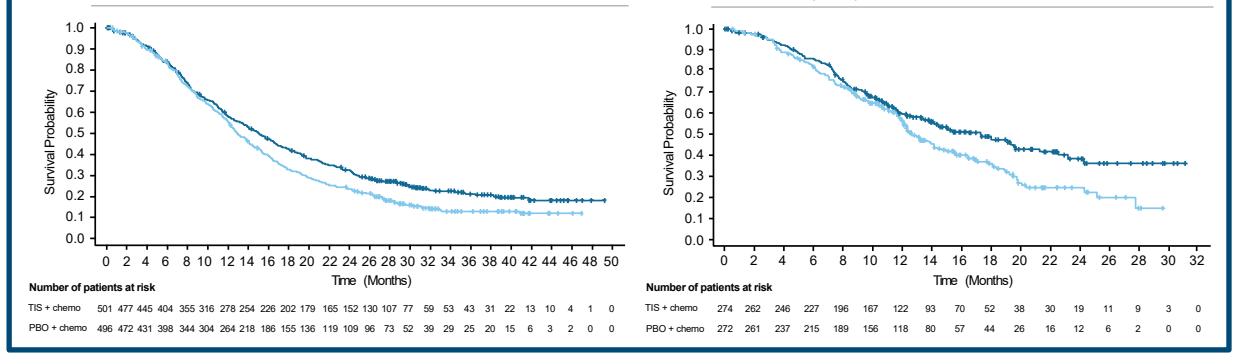
Survival Probability
Time (Months)
Number of patients at risk
TIS + chemo 501 477 445 404 355 316 278 254 226 202 179 165 152 130 107 77 59 53 43 31 22 13 10 4 1 0
PBO + chemo 496 472 431 398 344 304 264 218 186 155 136 119 109 96 73 52 39 29 25 20 15 6 3 2 0 0
TIS + chemo 274 262 246 227 196 167 122 93 70 52 38 30 19 11 9 3 0
PBO + chemo 272 261 237 215 189 156 118 80 57 44 26 16 12 6 2 0 0

[a]Cox regression model was stratified by regions (east Asia vs rest of the world), PD-L1 expression, and presence of peritoneal metastasis. [b]*P* values are one-sided and based on the stratified log-rank test. [c]Cox regression model was stratified by regions (east Asia vs rest of the world) and presence of peritoneal metastasis.
Chemo, chemotherapy; HR, hazard ratio; ITT, intent-to-treat; OS, overall survival; PBO, placebo; PD-L1, programmed death-ligand 1; TIS, tislelizumab.

- Demographics and baseline characteristics of patients in the Asian subgroup were similar to those of the overall ITT population (**Table 1**)

Table 1. Demographics and Baseline Disease Characteristics

	Asian Subgroup ITT Population		Overall ITT Population	
	Tislelizumab + Chemotherapy (n=376)	Placebo + Chemotherapy (n=372)	Tislelizumab + Chemotherapy (n=501)	Placebo + Chemotherapy (n=496)
Age, median (range), years	59.0 (23.0-86.0)	61.0 (25.0-83.0)	60.0 (23.0-86.0)	61.0 (25.0-86.0)
Sex, male, n (%)	258 (68.6)	261 (70.2)	346 (69.1)	346 (69.8)
ECOG PS, n (%)				
0	120 (31.9)	102 (27.4)	169 (33.7)	154 (31.0)
1	256 (68.1)	270 (72.6)	332 (66.3)	342 (69.0)
Primary tumor location, n (%)[a]				
Stomach	329 (87.5)	319 (85.8)	405 (80.8)	395 (79.6)
GEJ	47 (12.5)	52 (14.0)	96 (19.2)	100 (20.2)
PD-L1 TAP score, n (%)				
≥5%	202 (53.7)	201 (54.0)	274 (54.7)	272 (54.8)
<5%	174 (46.3)	171 (46.0)	227 (45.3)	224 (45.2)
Metastatic disease, n (%)	373 (99.2)	369 (99.2)	494 (98.6)	490 (98.8)
Peritoneal metastasis, n (%)	165 (43.9)	160 (43.0)	220 (43.9)	214 (43.1)
Investigator-chosen chemo, n (%)				
Oxaliplatin/capecitabine	370 (98.4)	367 (98.7)	466 (93.0)	465 (93.8)
Cisplatin/5-fluorouracil	6 (1.6)	5 (1.3)	35 (7.0)	31 (6.3)

Data cutoff: February 28, 2023.
[a]The diagnosis of 1 patient was updated from gastric adenocarcinoma to pancreatic cancer after randomization and the patient remained in the ITT population.
Chemo, chemotherapy; ECOG PS, Eastern Cooperative Oncology Group Performance Status; GEJ, gastroesophageal junction; ITT, intent-to-treat; PBO, placebo; PD-L1, programmed death-ligand 1; TAP, tumor area positivity; TIS, tislelizumab.

- An improvement in OS was observed with tislelizumab plus chemotherapy versus placebo plus chemotherapy in the ITT population (**Figure 3**)

Figure 3. Overall Survival: Asian Subgroup (ITT Population)

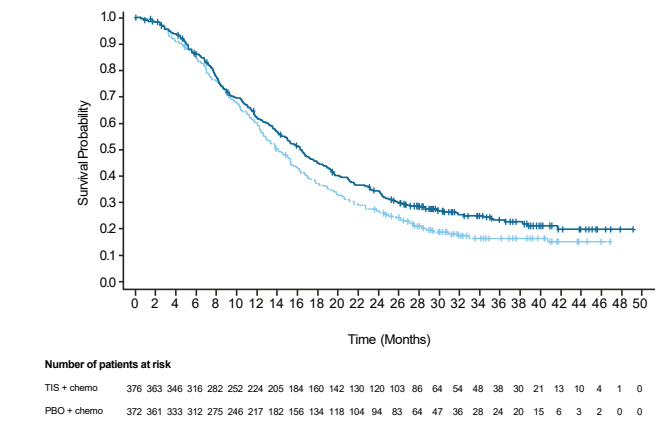
Survival Probability
Time (Months)
Number of patients at risk
TIS + chemo 376 363 346 316 282 252 224 205 184 160 142 130 120 103 86 64 54 48 38 30 21 13 10 4 1 0
PBO + chemo 372 361 333 312 275 246 217 182 156 134 118 104 94 83 64 47 36 28 24 20 15 6 3 2 0 0

	Events, n (%)	Median OS (95% CI), Months[a]	HR (95% CI)[a]
TIS + chemo (n=376)	274 (72.9)	16.4 (14.4-18.0)	0.83 (0.70-0.97)
PBO + chemo (n=372)	298 (80.1)	14.1 (12.8-15.4)	

Data cutoff: February 28, 2023.
[a]HR and 95% CIs were estimated from an unstratified Cox regression model including treatment as a covariate.
Chemo, chemotherapy; HR, hazard ratio; ITT, intent-to-treat; OS, overall survival; PBO, placebo; TIS, tislelizumab.

- An improvement in OS was also observed with tislelizumab plus chemotherapy versus placebo plus chemotherapy in the PD-L1 score ≥5% population (**Figure 4**)

Figure 4. Overall Survival: Asian Subgroup (PD-L1 Score ≥5% Population)

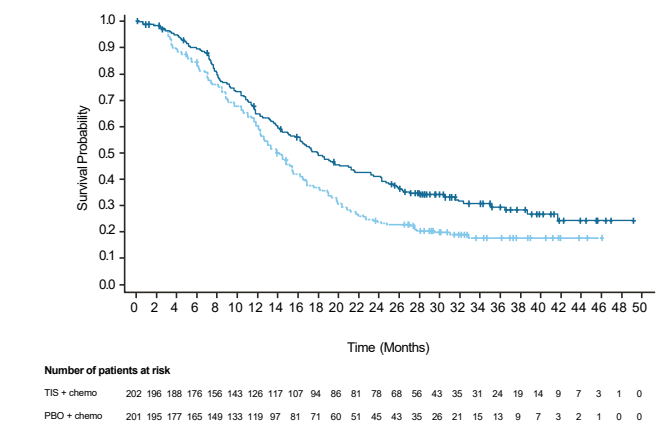
Survival Probability
Time (Months)
Number of patients at risk
TIS + chemo 202 196 188 176 156 143 126 117 107 94 86 81 78 68 56 43 35 31 24 19 14 9 7 3 1 0
PBO + chemo 201 195 177 165 149 133 119 97 81 71 60 51 45 43 35 26 21 15 13 9 7 3 2 1 0 0

	Events, n (%)	Median OS (95% CI), Months[a]	HR (95% CI)[a]
TIS + chemo (n=202)	135 (66.8)	18.0 (15.0-21.6)	0.71 (0.56-0.89)
PBO + chemo (n=201)	157 (78.1)	14.0 (12.3-15.5)	

Data cutoff: February 28, 2023.
[a]HR and 95% CIs were estimated from an unstratified Cox regression model including treatment as a covariate.
Chemo, chemotherapy; CI, confidence interval; HR, hazard ratio; OS, overall survival; PBO, placebo; PD-L1, programmed death-ligand 1; TIS, tislelizumab.

- Improved PFS was observed with tislelizumab plus chemotherapy versus placebo plus chemotherapy in the ITT and PD-L1 score ≥5% populations (**Figure 5**)

Figure 5. Progression-Free Survival: Asian Subgroup

ITT Population

	Events, n (%)	Median PFS (95% CI), Months	HR (95% CI)[a]
TIS + chemo (n=376)	266 (70.7)	7.1 (5.8-8.3)	0.76 (0.64-0.90)
PBO + chemo (n=372)	293 (78.8)	6.9 (5.8-7.2)	

PD-L1 Score ≥5% Population

	Events, n (%)	Median PFS (95% CI), Months	HR (95% CI)[a]
TIS + chemo (n=202)	134 (66.3)	8.2 (6.9-9.7)	0.67 (0.53-0.84)
PBO + chemo (n=201)	157 (78.1)	6.8 (5.6-8.1)	

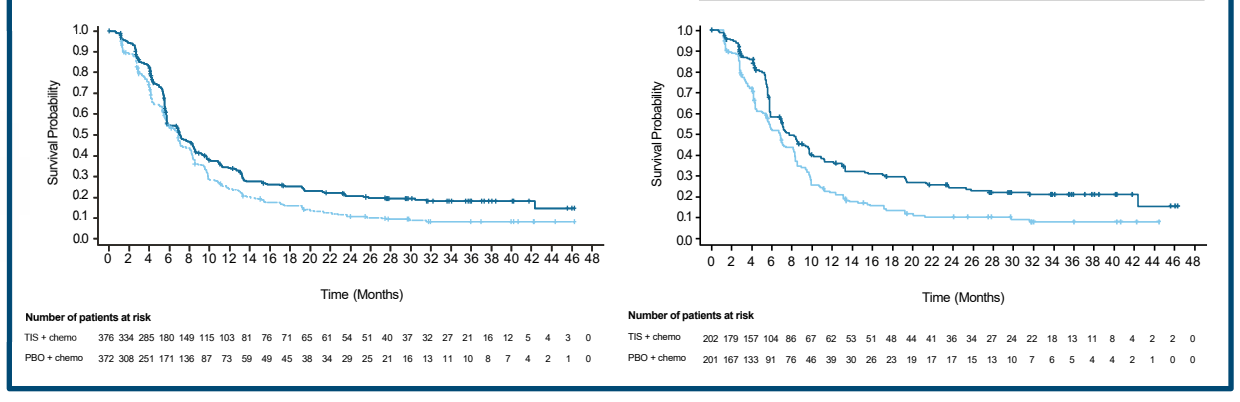
Survival Probability
Time (Months)
Number of patients at risk
TIS + chemo 376 334 285 180 149 115 103 81 76 71 65 61 54 51 40 37 32 27 21 16 12 5 4 3 0
PBO + chemo 372 308 251 171 136 87 73 59 49 45 38 34 29 25 21 16 13 11 10 8 7 4 2 1 0
TIS + chemo 202 179 157 104 86 67 62 53 51 48 44 41 36 34 27 24 22 18 13 11 8 4 2 2 0
PBO + chemo 201 167 133 91 76 46 39 30 26 23 19 17 17 15 13 10 7 6 5 4 4 2 1 0 0

Data cutoff: February 28, 2023.
[a]HR was based on an unstratified Cox regression model.
Chemo, chemotherapy; HR, hazard ratio; ITT, intent-to-treat; PBO, placebo; PD-L1, programmed death-ligand 1; PFS, progression-free survival; TIS, tislelizumab.

- Tislelizumab plus chemotherapy showed a favorable objective response rate and duration of response compared with placebo plus chemotherapy in the ITT and PD-L1 score ≥5% populations (**Figure 6**)

Figure 6. Disease Response and Duration of Response: Asian Subgroup

ITT Population

	ORR % (95% CI)[a,b]	Median DoR (95% CI), Months[c]
TIS + chemo (n=376)	51.1 (45.9-56.2)	9.5 (8.1-11.5)
PBO + chemo (n=372)	43.5 (38.4-48.8)	8.2 (6.9-9.9)

PD-L1 Score ≥5% Population

	ORR % (95% CI)[a,b]	Median DoR (95% CI), Months[d]
TIS + chemo (n=202)	56.9 (49.8-63.9)	10.3 (8.2-18.0)
PBO + chemo (n=201)	45.3 (38.3-52.4)	8.0 (6.5-9.9)

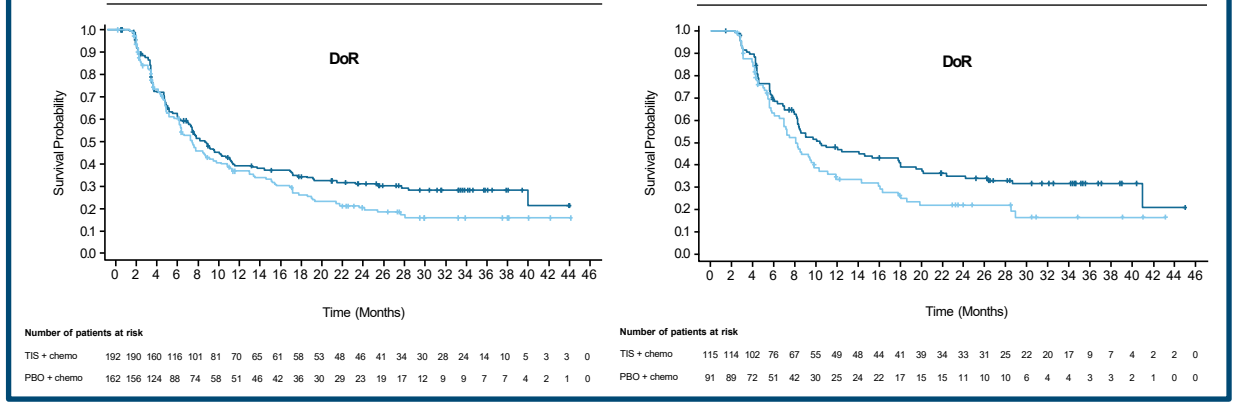
DoR
Survival Probability
Time (Months)
Number of patients at risk
TIS + chemo 192 190 160 116 101 81 70 65 61 58 53 48 46 41 34 30 28 24 14 10 5 3 3 0
PBO + chemo 162 156 124 88 74 58 51 46 42 36 30 29 23 19 17 12 9 9 7 7 4 2 1 0
TIS + chemo 115 114 102 76 67 55 49 48 44 41 39 34 33 31 25 22 20 17 9 7 4 2 2 0
PBO + chemo 91 89 72 51 42 30 25 24 22 17 15 15 11 10 10 6 4 4 3 3 2 1 0 0

Data cutoff: February 28, 2023.
[a]ORR was calculated using the unstratified Cochran-Mantel-Haenszel method. [b]ORR is defined as the proportion of patients with a confirmed complete response or partial response. [c]DoR analysis performed on 192 patients in the TIS + chemo arm and 162 patients in the PBO + chemo arm. [d]DoR analysis performed on 115 patients in the TIS + chemo arm and 91 patients in the PBO + chemo arm.
Chemo, chemotherapy; DoR, duration of response; ORR, objective response rate; PBO, placebo; PD-L1, programmed death-ligand 1; TIS, tislelizumab.

- No new safety signals with tislelizumab plus chemotherapy were identified in the Asian subgroup (**Figure 7**)

Figure 7. Safety Summary: Asian Subgroup (Safety Analysis Population)

	Tislelizumab + Chemotherapy (n=375)	Placebo + Chemotherapy (n=370)
Patients with at least one TRAE	366 (97.6)	360 (97.3)
Grade ≥3 TRAEs	208 (55.5)	185 (50.0)
Serious TRAEs	97 (25.9)	55 (14.9)
TRAE leading to death	9 (2.4)	3 (0.8)
TRAE leading to treatment discontinuation	64 (17.1)	33 (8.9)

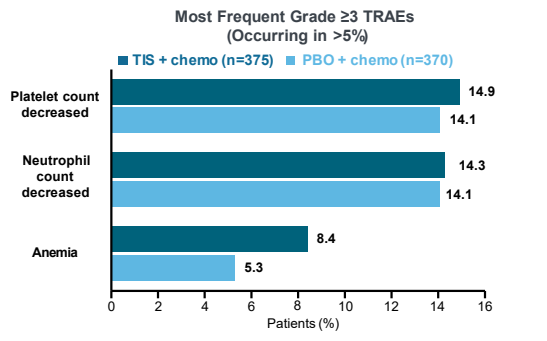
Most Frequent Grade ≥3 TRAEs
(Occurring in >5%)
TIS + chemo (n=375)
PBO + chemo (n=370)
Platelet count decreased
14.9
14.1
Neutrophil count decreased
14.3
14.1
Anemia
8.4
5.3
Patients (%)

Data cutoff: February 28, 2023. Data are n (%).
Chemo, chemotherapy; PBO, placebo; TIS, tislelizumab; TRAE, treatment-related adverse event.

References

1. Xu et al. *Ann Oncol*. 2023;34(suppl 2):S1320-S1321.
2. Sung et al. *CA Cancer J Clin*. 2021;71(3):209-249.
3. Shin et al. *Cancers (Basel)*. 2023;15(9):2639.
4. Yan et al. *Chin J Cancer Res*. 2023;35(2):81-91.
5. Sekiguchi et al. *Digestion*. 2022;103(1):22-28.
6. Cheng et al. *Ther Adv Med Oncol*. 2019;11:1758835919877726.
7. Lordick et al. *Ann Oncol*. 2022;33(10):1005-1020.
8. Catenacci et al. *Oncologist*. 2021;26(10):e1704-e1729.
9. Moehler et al. *J Clin Oncol*. 2023;41(suppl 4):286.

Acknowledgments

We would like to thank the investigators, site support staff, and especially the patients for participating in this study. This study is sponsored by BeiGene, Ltd. Medical writing support, under the direction of the authors, was provided by Steven Moore, PhD, and Camile Semighini Grubor, PhD, of Envision Pharma Inc., and was funded by BeiGene.

Presenter Disclosures

K-WL reports grants or contracts from ABLBIO, ALX Oncology, Amgen, Astellas, AstraZeneca, BeiGene, Bolt Therapeutics, Daiichi Sankyo, Elevar Therapeutics, Exelixis, Genome & Company, Green Cross Corp, Idience, InventisBio, Leap Therapeutics, Macrogenics, MedPacto, Merck KGaA, Merck Sharp and Dohme, Metafines, Oncologie, Ono Pharmaceutical, Pfizer, Pharmacyclics, Roche, Seagen, Taiho Pharmaceutical, Trishula Therapeutics, Y-BIOLOGICS, and Zymeworks.

Contact: imdoctor@snu.ac.kr (Keun-Wook Lee)