303

Real-World Chronic Lymphocytic Leukemia–Specific Biomarker Testing Patterns and Frontline Treatment Patterns in Community Oncology Patients With Chronic Lymphocytic Leukemia/Small Lymphocytic Lymphoma: an Electronic Health Record Study

Michael McGovern,[1] Da Sol Kim,[1] Beverly Fuerte,[1] Timothy Reynolds,[1] Linda Chen,[1] Archana Sagar,[1] Mohammad Eskandari,[1] Xiaoliang Wang,[2] Lee Ding,[2] Gregory A. Maglinte,[2] Paul J. Godley[1]

[1]Baylor Scott & White Health, Temple, TX, USA; [2]BeOne Medicines, Ltd, San Carlos, CA, USA

CONCLUSIONS

- Despite high uptake of del(17p) testing (85.2%) in this US cohort of 223 patients, critical molecular assessments, including *TP53* (10.8%) and IGHV (20.6%) sequencing were markedly underutilized
- Only 6.3% of patients received NCCN-recommended comprehensive testing (all three biomarkers)
- Testing gaps appeared largely systemic rather than driven by access disparities, with no significant differences by sex, insurance coverage, rural–urban location, or social vulnerability. Testing rates were modestly lower in older patients and varied across racial and ethnic groups; however, these differences may be attributable to small subgroup sample sizes
- Among patients with confirmed del(17p) loss, 72% (18/25) received guideline-concordant targeted therapy (second-generation BTK inhibitors or BCL2 inhibitors)

INTRODUCTION

- Chronic lymphocytic leukemia (CLL) is the most common leukemia in adults in the United States, with around 21,000 cases diagnosed annually[1]
- National Comprehensive Cancer Network (NCCN) Guidelines recommend prognostic testing before initiating frontline therapy[2]:
 - del(17p) by fluorescence in situ hybridization (FISH) analysis
 - *TP53* mutation sequencing
 - Immunoglobulin heavy chain variable (IGHV) mutation sequencing
- These biomarkers guide therapy selection, particularly identifying high-risk patients who benefit from novel targeted agents (second-generation Bruton tyrosine kinase [BTK] inhibitors, B-cell lymphoma 2 [BCL2] inhibitors) over chemoimmunotherapy
- However, real-world adoption of comprehensive testing remains poorly characterized[3,4]
- Here we assess biomarker testing frequency, associated factors, and frontline treatment patterns in patients with CLL/small lymphocytic lymphoma (SLL) initiating therapy at Baylor Scott & White Health, a large integrated health system in Texas

METHODS

Study Design

- A retrospective cohort chart review at Baylor Scott & White Health, a 53-hospital integrated delivery network in Texas (**Figure 1**)

Figure 1. Study Timeline

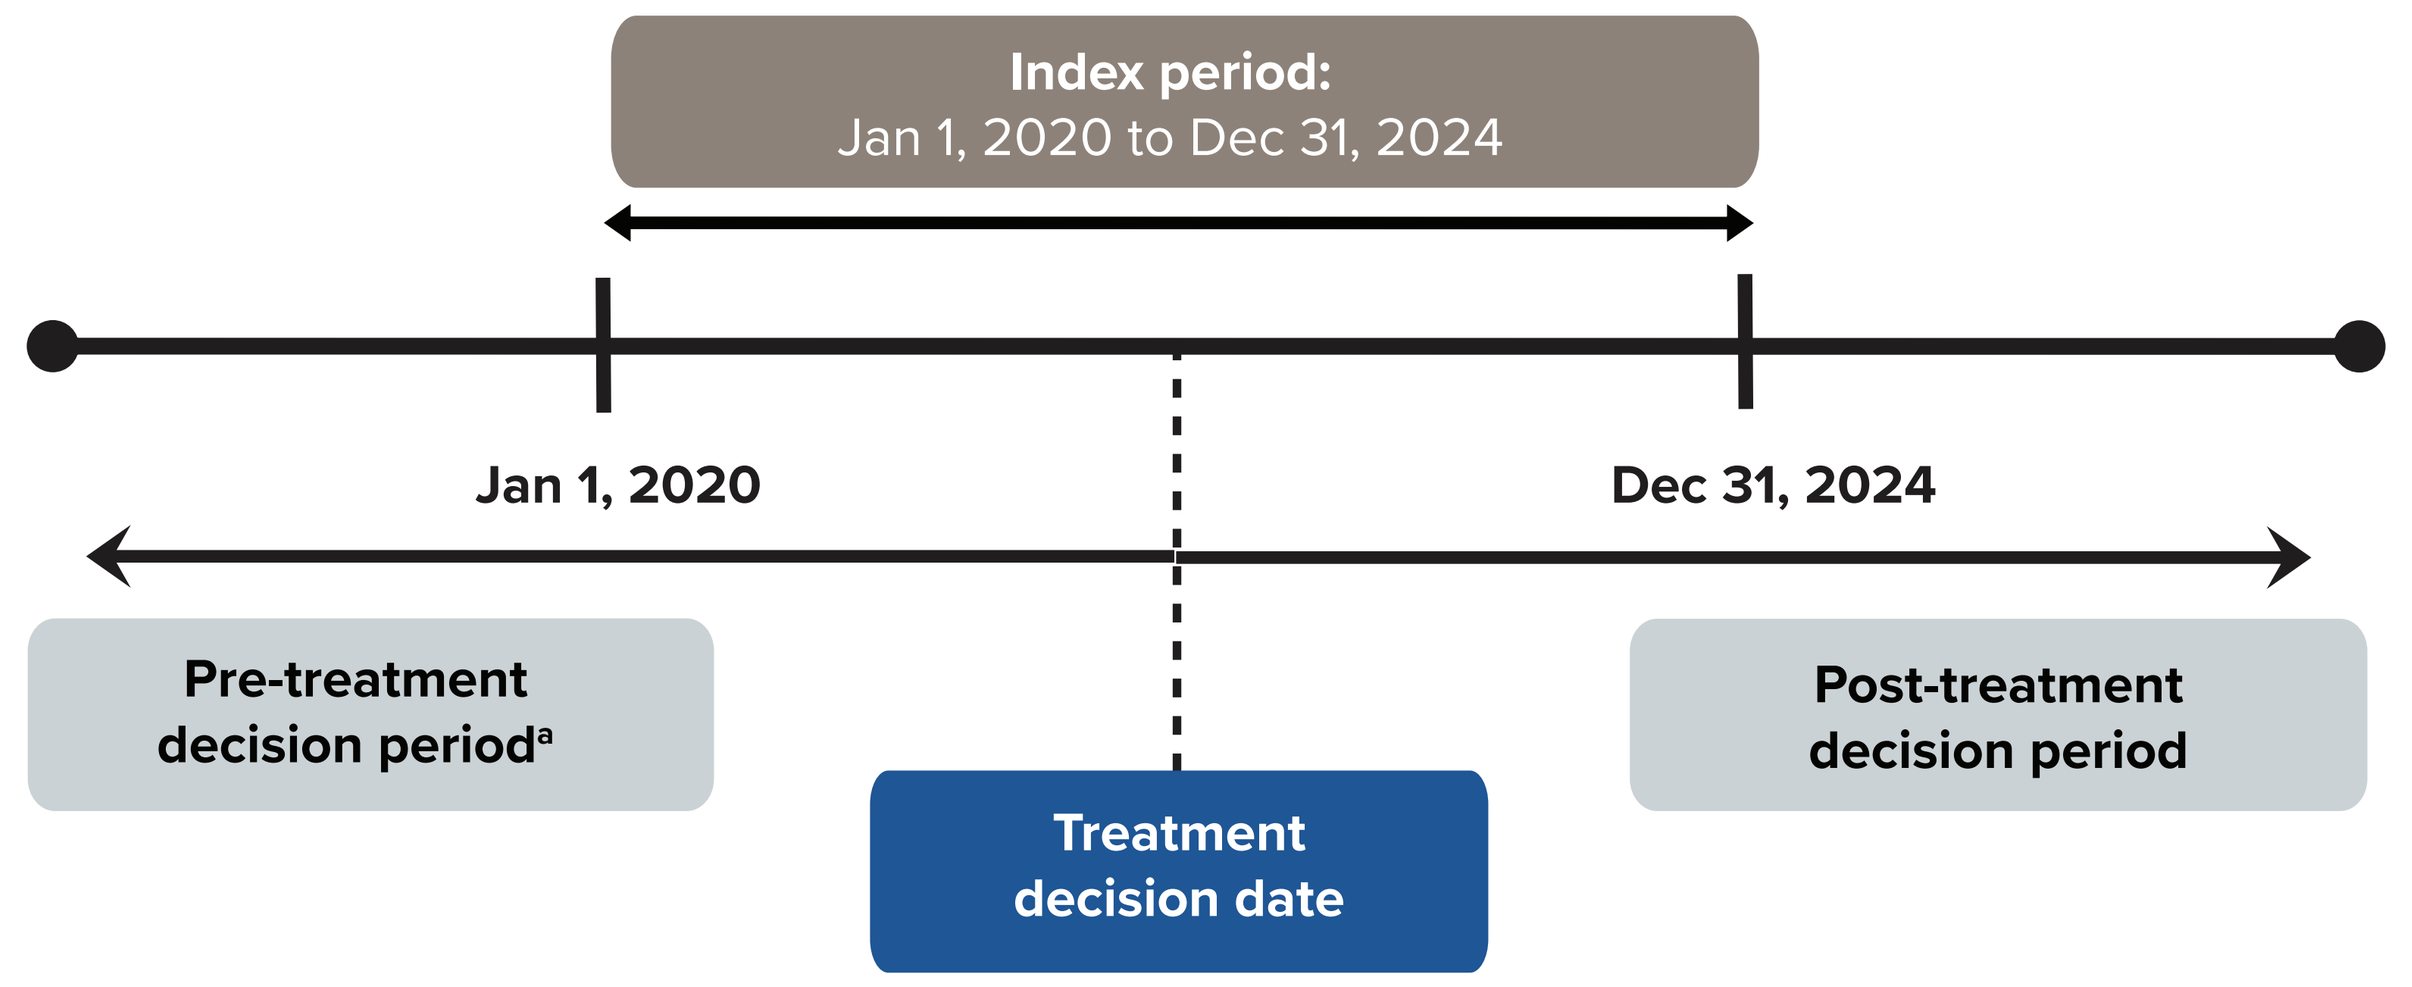
Index period:
Jan 1, 2020 to Dec 31, 2024
Jan 1, 2020
Dec 31, 2024
Pre-treatment decision period[a]
Treatment decision date
Post-treatment decision period

[a]Treatment decision date defined as initial frontline medication order.

Study Population

- Patients were included if they were:
 - aged 18 years or older at CLL/SLL diagnosis
 - initiated frontline therapy between 2020 and 2024
 - did not participate in a clinical trial
 - did not have Richter's transformation at frontline
- Eligible patients were identified via structured medical records, and confirmed via chart review
- See **Figure 2** for cohort selection information

Figure 2. Cohort Selection

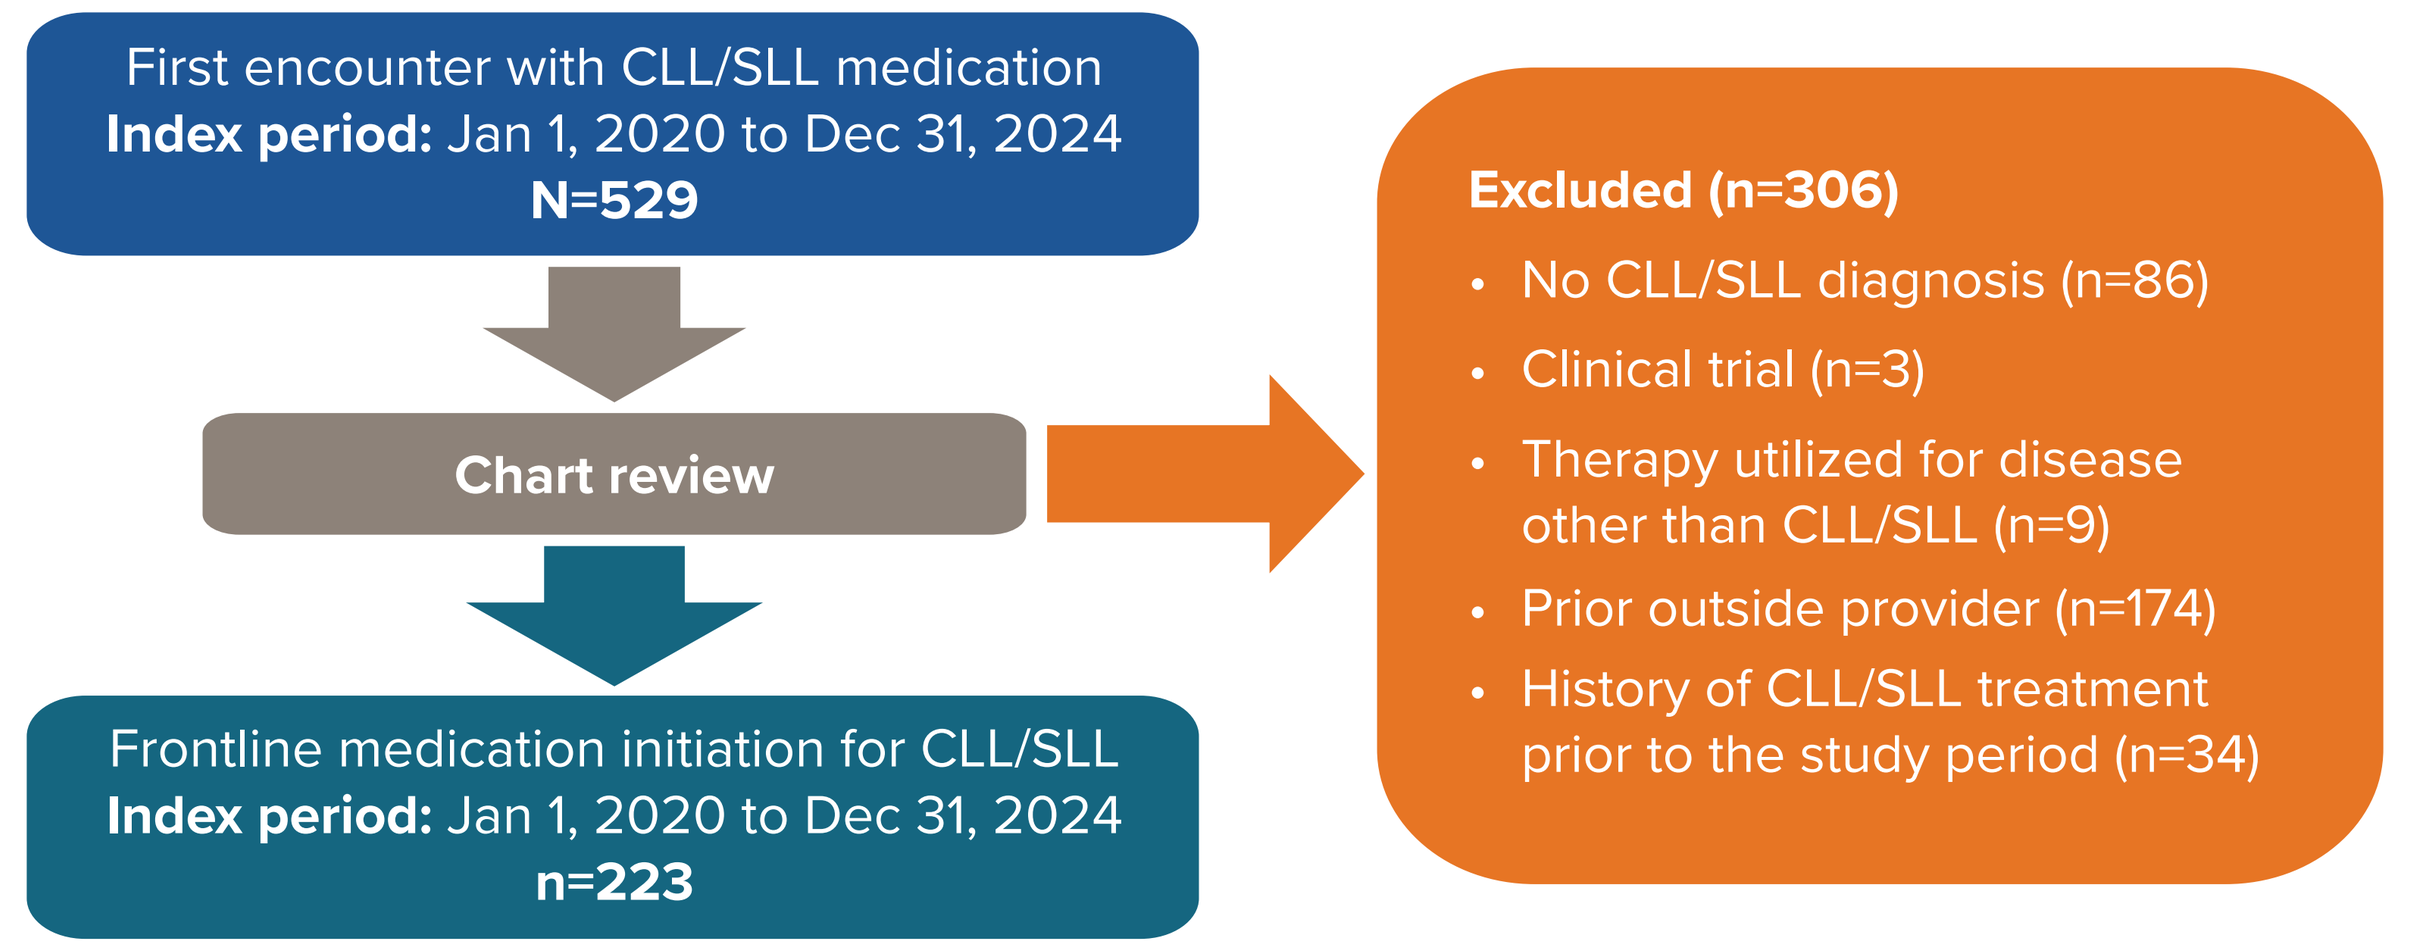
First encounter with CLL/SLL medication
Index period: Jan 1, 2020 to Dec 31, 2024
N=529
Chart review
Frontline medication initiation for CLL/SLL
Index period: Jan 1, 2020 to Dec 31, 2024
n=223
Excluded (n=306)
No CLL/SLL diagnosis (n=86)
Clinical trial (n=3)
Therapy utilized for disease other than CLL/SLL (n=9)
Prior outside provider (n=174)
History of CLL/SLL treatment prior to the study period (n=34)

Abbreviations: CLL, chronic lymphocytic leukemia; SLL, small lymphocytic lymphoma.

Data Collection and Statistical Analysis

- Demographic characteristics were collected via structured data; biomarker testing and frontline treatments were confirmed via chart review
- Descriptive statistics were used to summarize testing and treatment patterns
- Associations with del(17p) testing were evaluated for demographic, insurance, Social Vulnerability Index, and Rural Urban Commuting Area (RUCA) variables
- Group comparisons used χ^2, Fisher's exact, *t*, or Wilcoxon rank-sum tests, as appropriate

RESULTS

Patient Characteristics

- In the 223 patients included, the median age was 73 years, and 64.1% were male (**Table 1**)
 - Most patients had Medicare insurance (74.4%) and resided in urban areas, defined as RUCA scores 1-3 (80.7%)

Table 1. Patient Characteristics

Characteristic	All (N=223)	del(17p) tested (n=190)	del(17p) not tested (n=33)	*P* value
Age, median (IQR)	73.0 (14.5)	72.0 (14.0)	79.0 (20.0)	**0.018**[a]
Male, n (%)	143 (64.1)	125 (65.8)	18 (54.5)	0.295[b]
Race and ethnicity, n (%)				**0.047**[c]
Non-Hispanic White	182 (81.6)	157 (82.6)	25 (75.8)	
Non-Hispanic Black	24 (10.8)	21 (11.1)	3 (9.1)	
Hispanic	10 (4.5)	7 (3.7)	3 (9.1)	
Other	7 (3.1)	5 (2.6)	2 (6.1)	
Insurance, n (%)				
Medicare	166 (74.4)	140 (73.7)	26 (78.8)	0.667[c]
Commercial	47 (21.1)	41 (21.6)	6 (18.2)	0.833[b]
Medicaid	3 (1.3)	2 (1.1)	1 (3.0)	0.383[c]
Other Government	7 (3.1)	6 (3.2)	1 (3.0)	1.000[c]
Social Vulnerability Index, n (%)				0.518[c]
Lowest (≤0.2499)	43 (19.3)	34 (17.9)	9 (27.3)	
Medium low (0.2500-0.4999)	57 (25.6)	51 (26.8)	6 (18.2)	
Medium high (0.5000-0.7499)	43 (19.3)	38 (20.0)	5 (15.2)	
Highest (0.7500-1.0000)	53 (23.8)	43 (22.6)	10 (30.3)	
Unknown	27 (12.1)	24 (12.6)	3 (9.1)	
RUCA score, n (%)				0.680[b]
Urban (RUCA 1-3)	180 (80.7)	152 (80.0)	28 (84.8)	
Rural (RUCA 4-10)	43 (19.3)	38 (20.0)	5 (15.2)	

[a]Wilcoxon rank-sum test (asymptotic). [b]Pearson χ^2 test (with Yates' continuity correction for 2×2 tables). [c]Fisher's exact test (used when any expected cell count <5).
Abbreviation: RUCA, Rural-Urban Commuting Area.

Biomarker Testing Rates and Timing

- del(17p) testing was performed most commonly and typically prior to the initiation of frontline therapy (154 patients [69.1%]) (**Figure 3**)
- *TP53* and IGHV sequencing rates were low, and the majority of patients remained untested before the initiation of frontline therapy (199 patients [89.2%] and 177 patients [79.4%], respectively)
- Overall, 6.3% (14/223) of patients received all three recommended biomarkers (del17p, *TP53*, and IGHV), with 71.4% (10/14) completing all testing prior to therapy

Figure 3. Biomarker Testing Rates and Timing Relative to Frontline Therapy

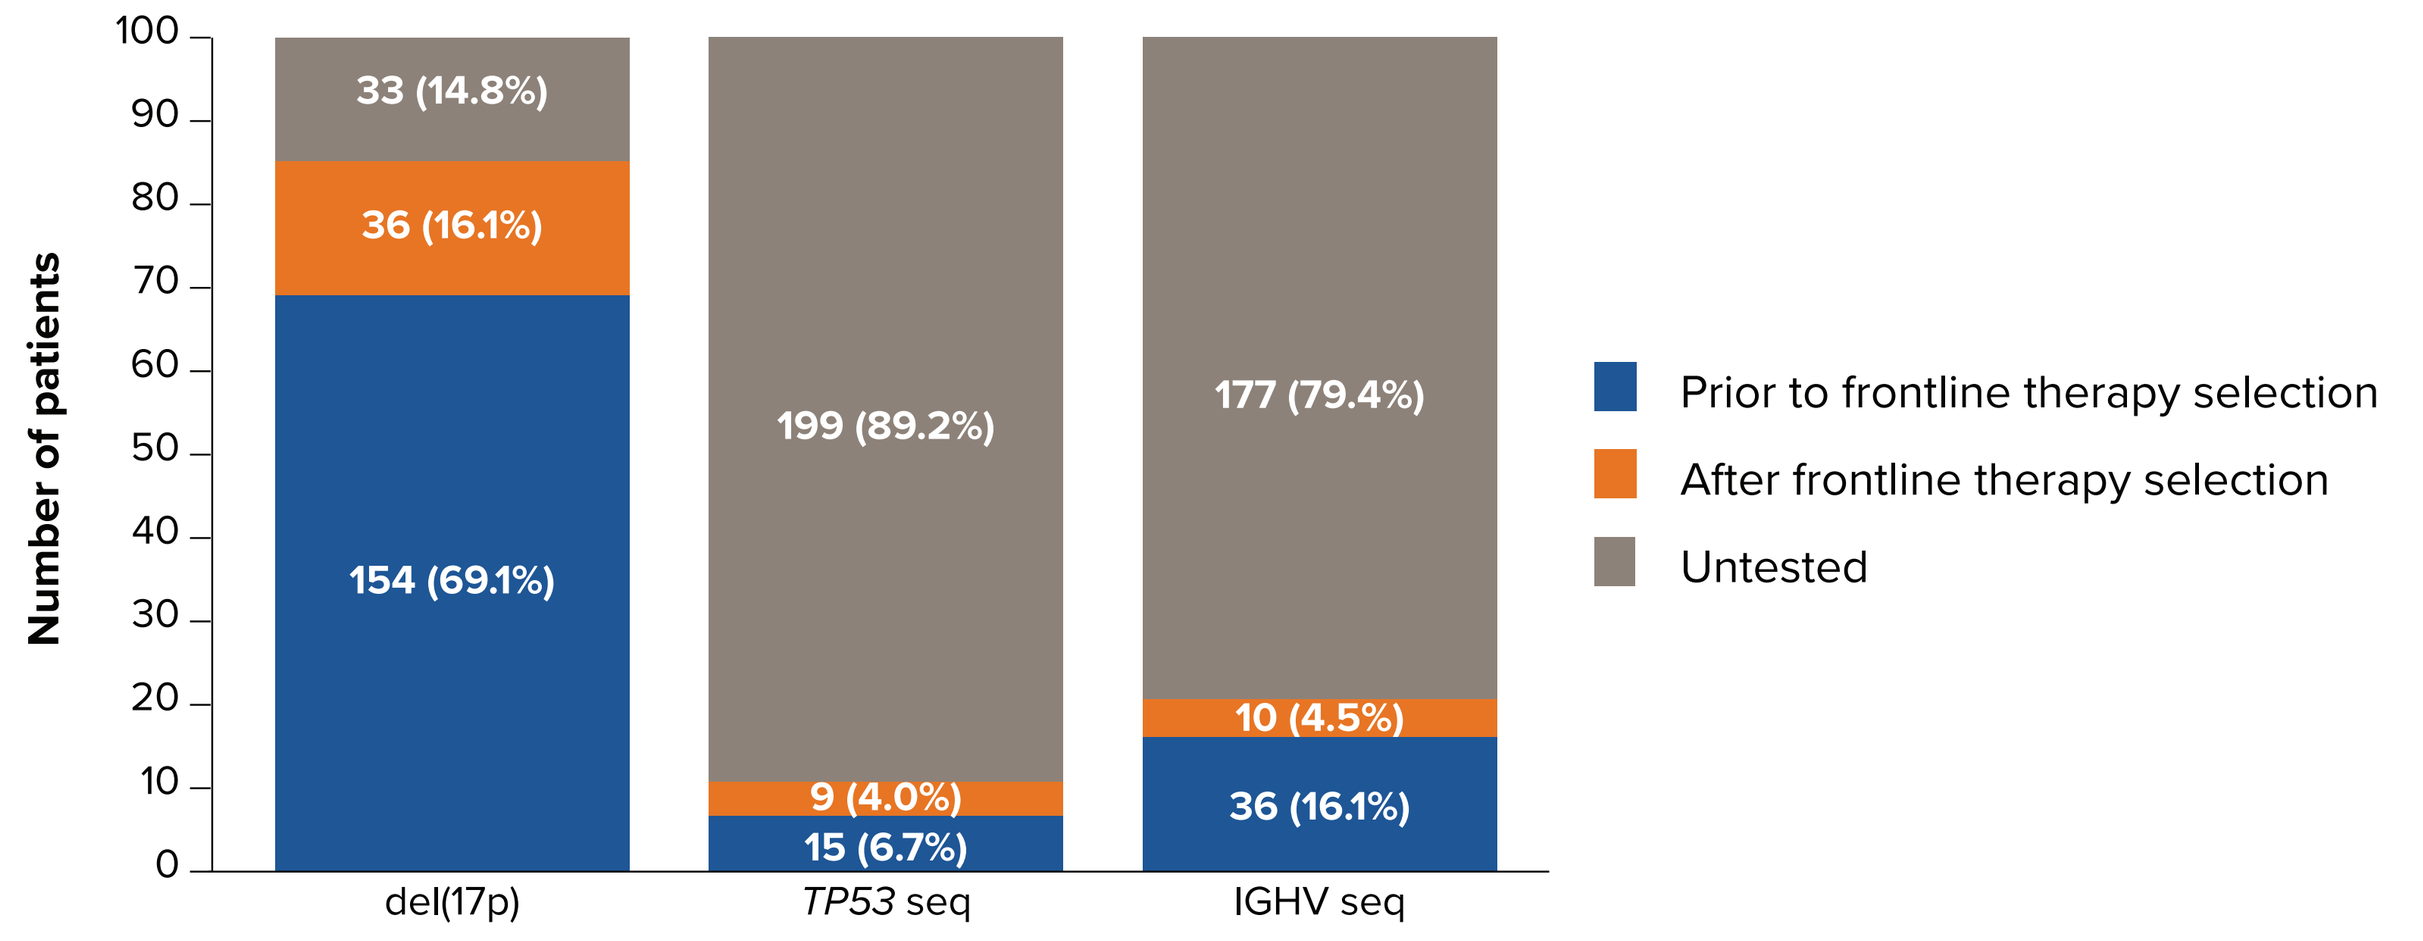
Number of patients
100
90
80
70
60
50
40
30
20
10
0
33 (14.8%)
36 (16.1%)
154 (69.1%)
199 (89.2%)
9 (4.0%)
15 (6.7%)
177 (79.4%)
10 (4.5%)
36 (16.1%)
del(17p)
TP53 seq
IGHV seq
Prior to frontline therapy selection
After frontline therapy selection
Untested

Abbreviations: IGHV, immunoglobulin heavy chain variable; seq, sequencing.

Mutation Rates

- Among patients with biomarker testing results, del(17p) loss was present in 13.2%, *TP53* mutations in 4.2%, and unmutated IGHV in 45.7% (**Table 2**)

Table 2. Mutation Results Among Tested Patients

Biomarker test	Result	n/N (%)
del(17p) FISH	Normal	165/190 (86.8)
	Loss (high risk)	**25/190 (13.2)**
***TP53* sequencing**	Absent	23/24 (95.8)
	Present	1/24 (4.2)
IGHV sequencing	Mutated (favorable)	21/46 (45.7)
	Unmutated (poor)	**21/46 (45.7)**
	Undetermined	4/46 (8.7)

Abbreviations: FISH, fluorescence in situ hybridization; IGHV, immunoglobulin heavy chain variable.

Treatment Patterns

- In the frontline setting, second-generation BTK inhibitors were most frequently given to patients (80 patients [35.9%]), followed by first-generation BTK inhibitors (42 patients [18.8%]) (**Figure 4**)

Figure 4. Distribution of Frontline Treatment Classes

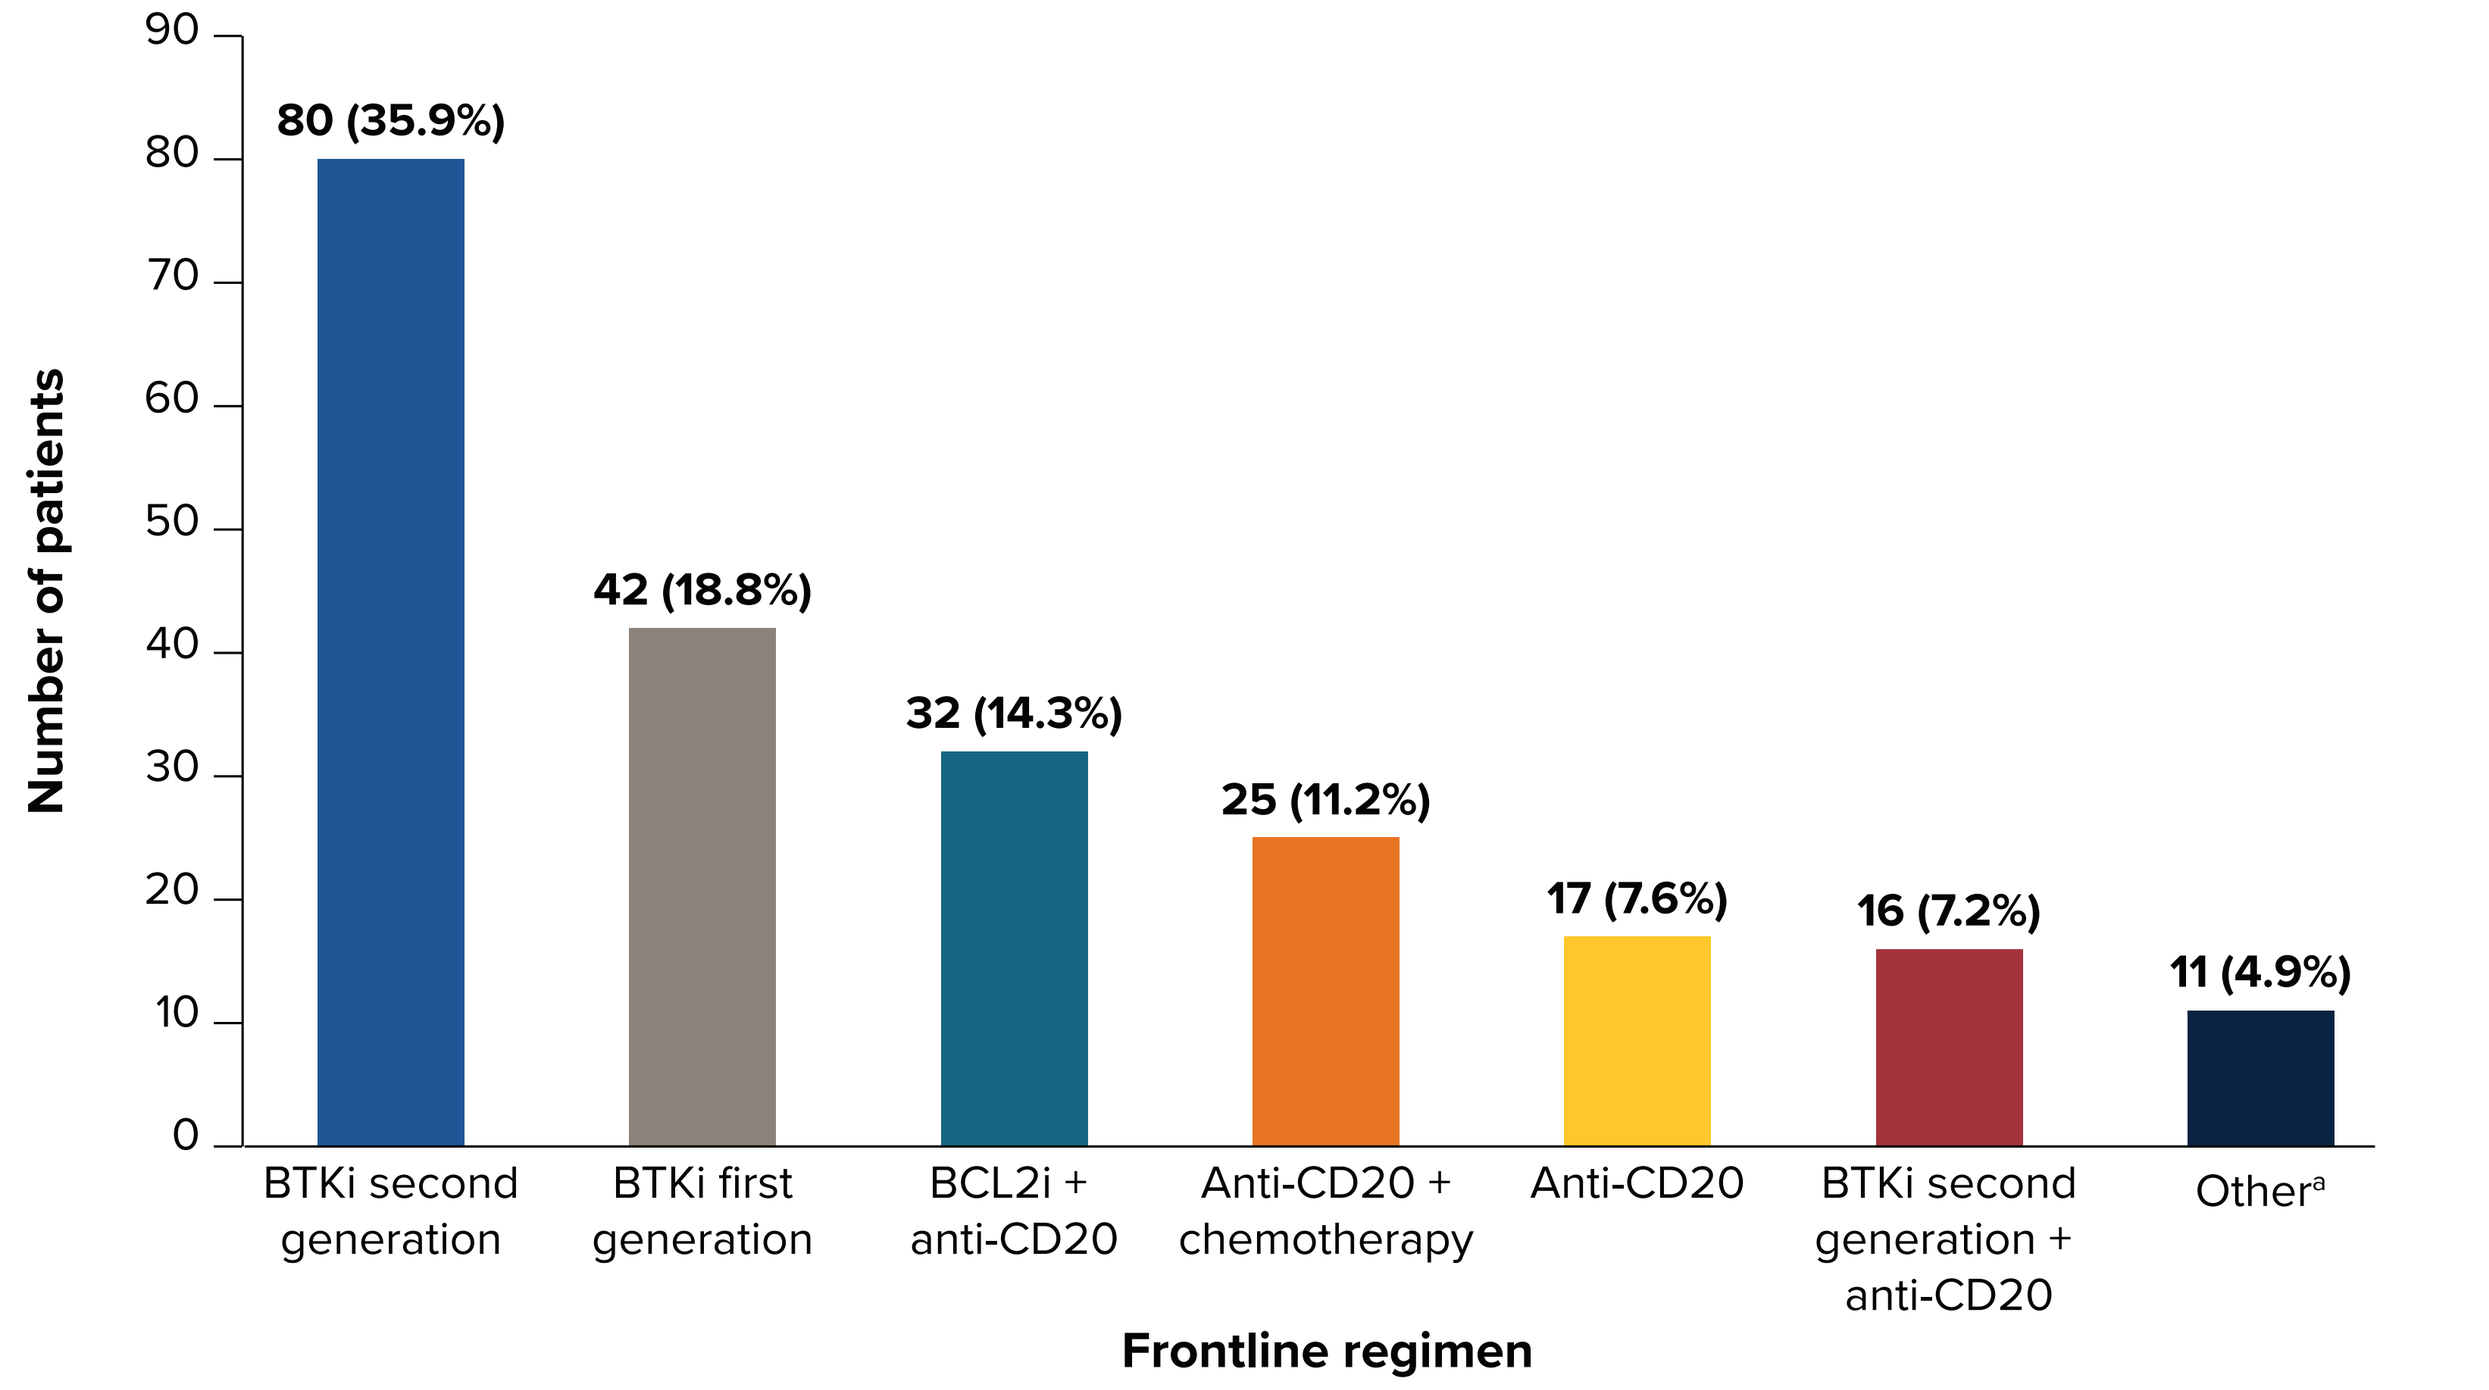
Number of patients
90
80
70
60
50
40
30
20
10
0
80 (35.9%)
42 (18.8%)
32 (14.3%)
25 (11.2%)
17 (7.6%)
16 (7.2%)
11 (4.9%)
BTKi second generation
BTKi first generation
BCL2i + anti-CD20
Anti-CD20 + chemotherapy
Anti-CD20
BTKi second generation + anti-CD20
Other[a]
Frontline regimen

[a]Other regimens: chemotherapy, immunomodulatory therapy, first-generation BTK inhibitor–based combinations (± anti-CD20, ± chemotherapy, or with BCL2i); and BCL2i-based therapy.
Abbreviations: BCL2i, B-cell lymphoma 2 inhibitor; BTKi, Bruton tyrosine kinase inhibitor.

- When comparing treatment patterns in patients with del(17p) loss vs patients with normal del(17p) (**Figure 5**):
 - Second-generation BTK inhibitors were the most common frontline therapy given; patients with del(17p) loss were more likely to receive second-generation BTK inhibitors as frontline therapy (56.0%) compared with patients with normal del(17p) (41.8%)
 - First-generation BTK inhibitor use was lower in patients with del(17p) loss (12.0%) than in patients with del(17p) normal (22.4%)
 - The use of BCL2 inhibitor–based regimens and other regimens was similar between patients with del(17p) loss and patients with del(17p) normal

Figure 5. Treatment Regimen Distribution by del(17p) Status (Loss vs Normal)

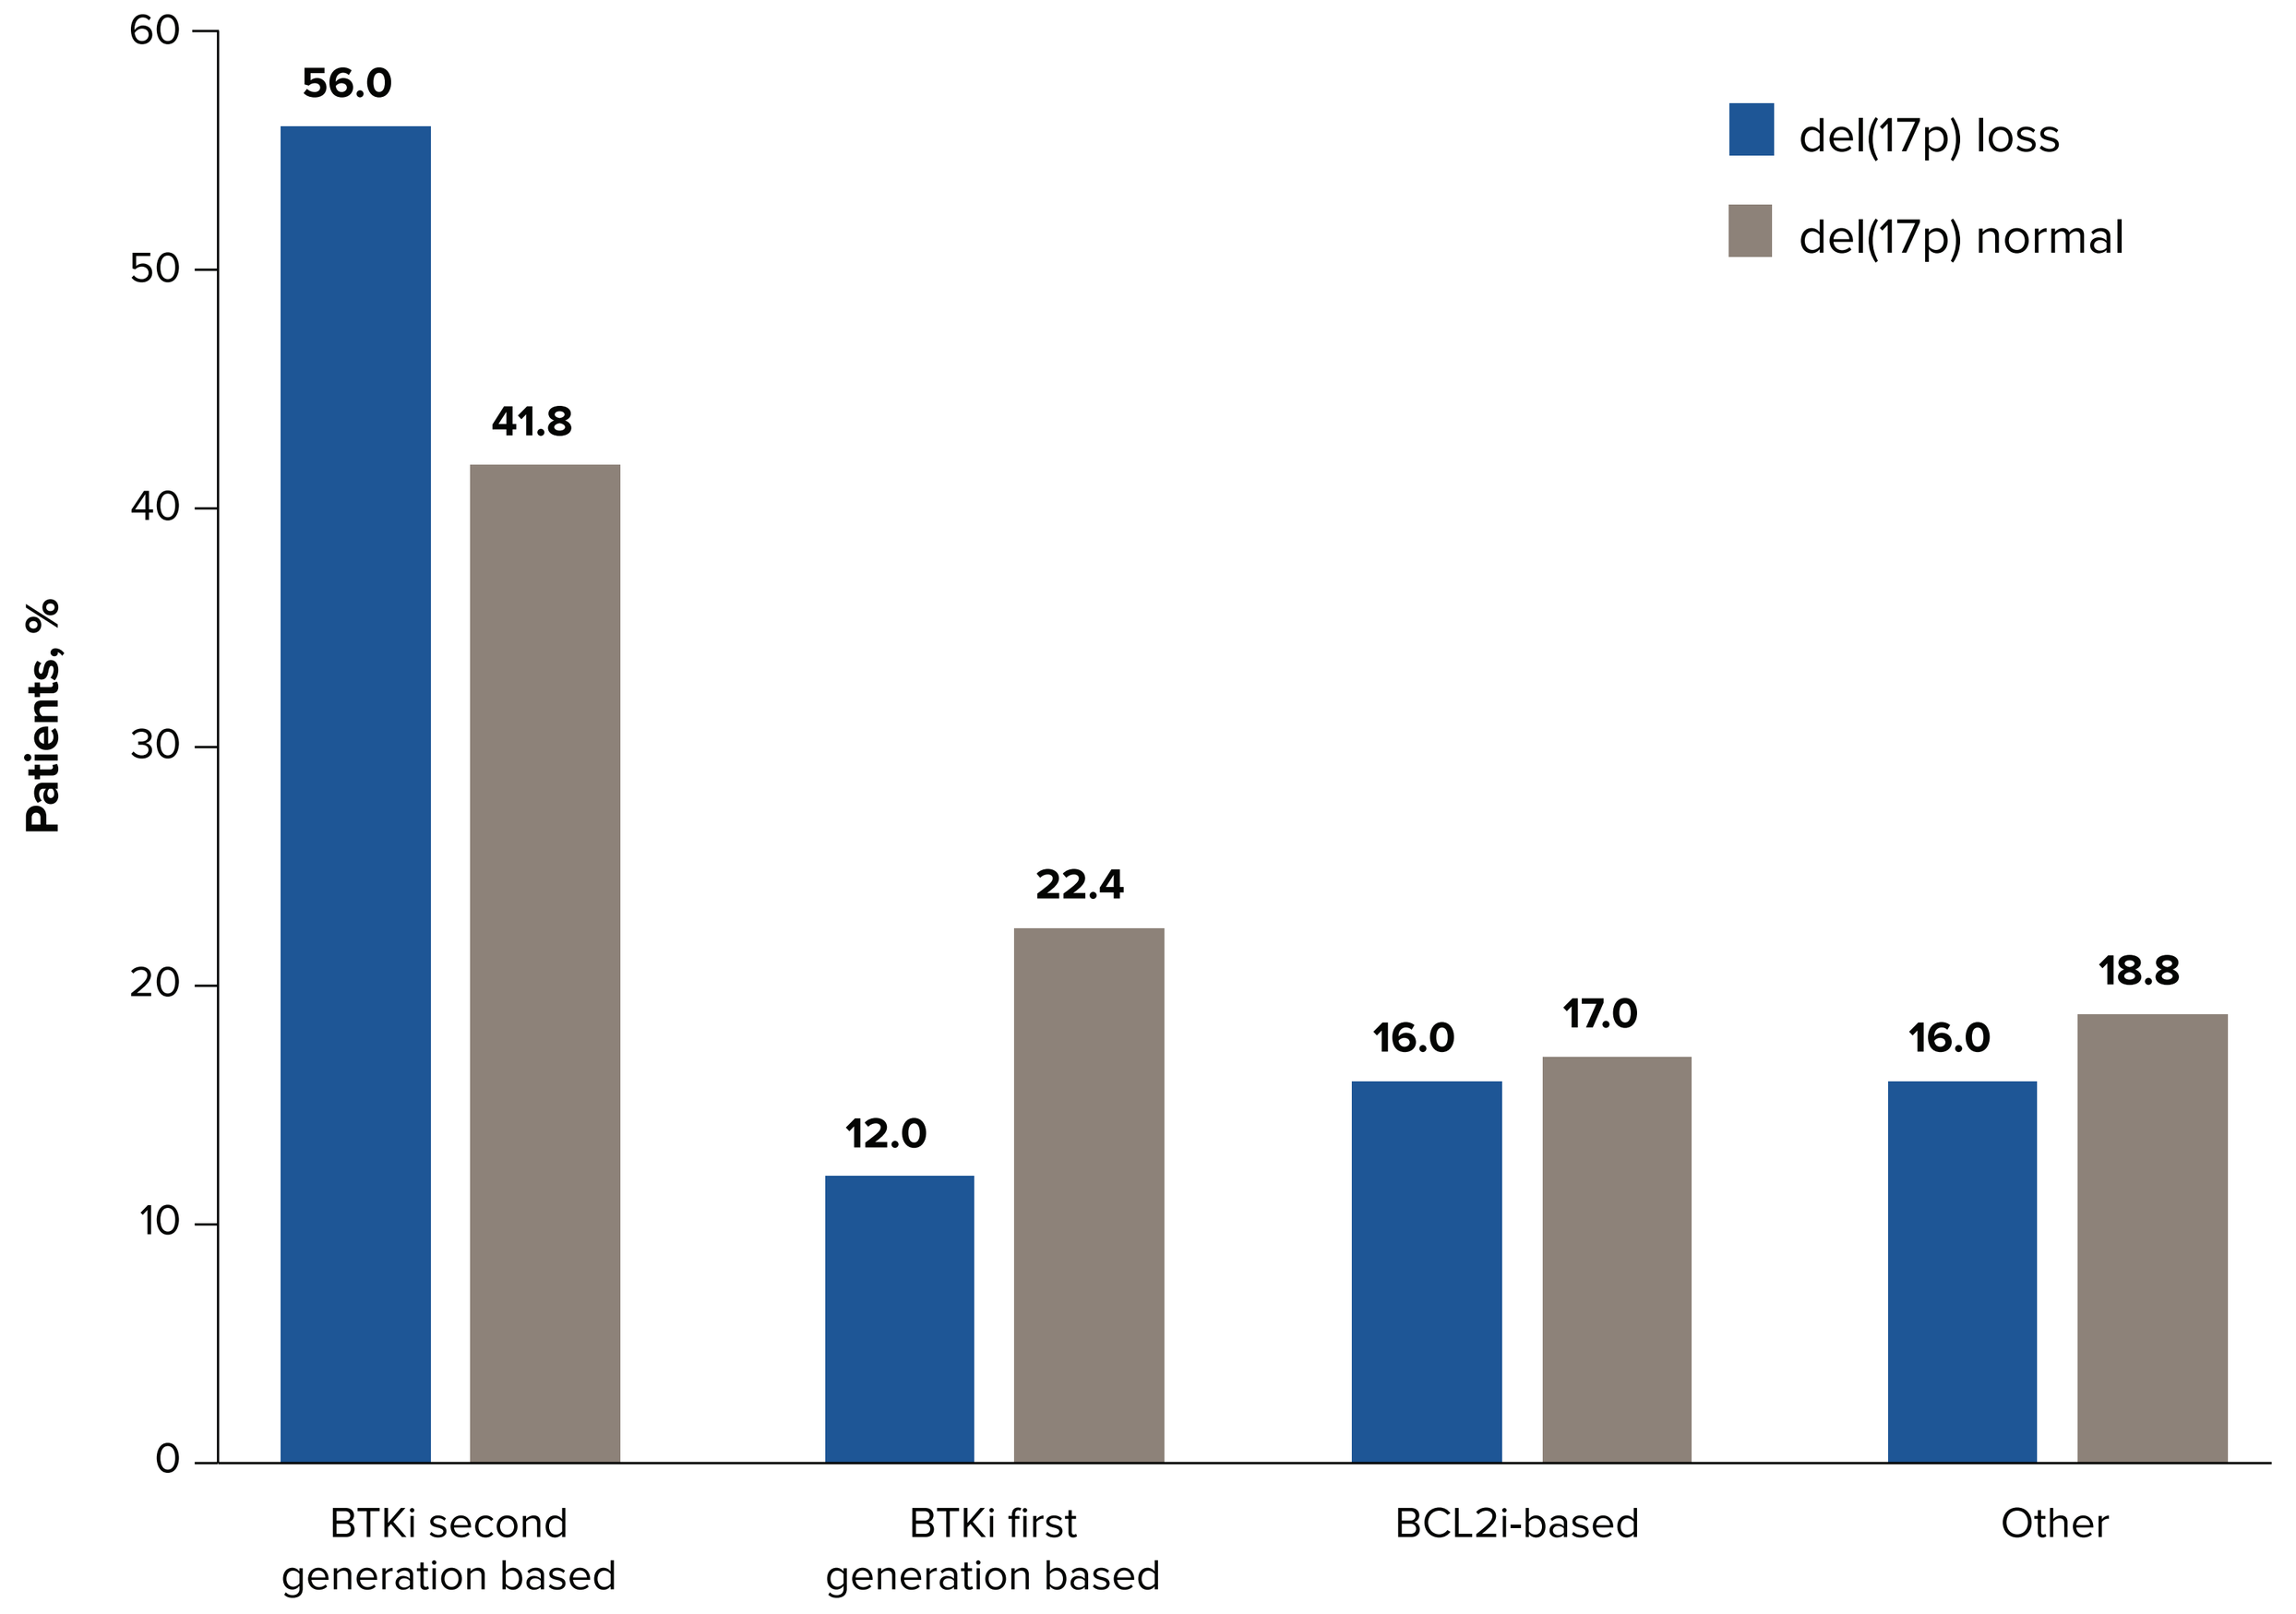
Patients, %
60
50
40
30
20
10
0
56.0
41.8
12.0
22.4
16.0
17.0
16.0
18.8
BTKi second generation based
BTKi first generation based
BCL2i-based
Other
del(17p) loss
del(17p) normal

Abbreviations: BCL2i, B-cell lymphoma 2 inhibitor; BTKi, Bruton tyrosine kinase inhibitor.

STRENGTHS AND LIMITATIONS

- A key strength of this analysis is that biomarker testing and treatment information were verified via chart review, allowing for comprehensive and accurate capture of these data
- Limitations include that the sample size was limited for subgroup analysis or multivariate analysis. Furthermore, care delivered outside Baylor Scott & White Health may not be captured with the same accuracy as care received from internal providers, which may limit documentation completeness

REFERENCES

1. Leukemia and Lymphoma Society. Accessed February 5, 2026. https://cllsociety.org/cll-sll-patient-education-toolkit/understanding-cll-sll/
2. Wierda WG, et al. *J Natl Compr Canc Netw*. 2024;22(3):175-204.
3. Mato AR, et al. *Blood Adv*. 2023;7(17):4760-4764.
4. Chanan-Khan A, et al. ASH 2021. Abstract 4078.

ACKNOWLEDGMENTS

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This study was sponsored by BeOne Medicines, Ltd. Editorial support was provided by Nucleus Global, an Inizio company, and supported by BeOne Medicines.

DISCLOSURES

MM, DSK, BF: Consultant: BeOne Medicines, Ltd, Sanofi. **TR, LC, PJG**: Consultant: BeOne Medicines, Ltd, Sanofi, Pfizer; **XW, LD, GAM**: Employment: BeOne Medicines, Ltd. **AS, ME**: No disclosures.

CORRESPONDENCE: Michael McGovern, michael.McGovern@BSWHealth.org

Presented at the AMCP 2026 Annual Meeting; April 13-16, 2026; Nashville, TN, USA