RATIONALE-305: Post-Treatment Immuno-Oncology Adjustment Analysis to Assess Tislelizumab Treatment Effect vs Placebo

2931P

Markus Moehler,[1] Sun Young Rha,[2] Rui-Hua Xu,[3] Elizabeth Smyth,[4] Xuan (Leo) Kong,[5] Na Zhao,[5] Maria Alsina[6]

[1]Department of Medicine, Johannes Gutenberg-University Clinic, Mainz, Germany; [2]Yonsei Cancer Center, Yonsei University College of Medicine, Seoul, Republic of Korea; [3]Sun Yat-sen University Cancer Center, State Key Laboratory of Oncology in South China, Collaborative Innovation Center of Cancer Medicine, Guangzhou, China; [4]Oxford University Hospitals NHS Foundation Trust, Oxford, UK; [5]BeOne Medicines, Ltd, Shanghai, China; [6]Department of Oncology, Hospital Universitario de Navarra, Pamplona, Spain

CONCLUSIONS

- In patients with human epidermal growth factor receptor 2 (HER2)-negative unresectable locally advanced or metastatic gastric/gastroesophageal junction adenocarcinoma (GC/GEJC) enrolled in the phase 3 RATIONALE-305 trial, adjustment for subsequent immunotherapy (IO) use strengthened the estimated overall survival (OS) treatment effect favoring first-line tislelizumab + chemotherapy (CT) over placebo + CT, suggesting that subsequent IO use may have partially obscured the treatment benefit in the primary analysis
 - Observed OS: 15.0 months vs 12.9 months, respectively; stratified HR, 0.79 (95% CI, 0.69-0.90)
 - Adjusted OS: 14.3 months vs 12.1 months, respectively; stratified HR, 0.77 (95% CI, 0.67-0.88)
- Subsequent IO use was associated with longer survival, irrespective of randomized treatment; however, subsequent IO use was more prevalent in Asia and among patients with a lower disease burden, indicating potential selection bias and limiting interpretation of these findings as evidence of the efficacy of subsequent IO

INTRODUCTION

- Immune checkpoint inhibitor + CT is a standard first-line treatment option for patients with HER2-negative unresectable locally advanced or metastatic GC/GEJC[1-3]
- Tislelizumab is a high-affinity immune checkpoint inhibitor targeting programmed cell death protein 1 engineered to minimize binding to Fcγ receptors on macrophages[3,4]
- In the phase 3 RATIONALE-305 trial (NCT03777657), first-line tislelizumab + chemotherapy (capecitabine + oxaliplatin [CAPOX] or cisplatin + 5-fluorouracil [5-FU]) significantly improved median OS (mOS) vs placebo + CT in patients with HER2-negative unresectable locally advanced or metastatic GC/GEJC[3]
- Patients in the placebo + CT arm were more likely to receive subsequent IO after disease progression than those in the tislelizumab + CT arm (19.8% vs 13.0%, respectively)[5]
- Subsequent IO after disease progression may attenuate the observed OS difference between treatment and control arms by prolonging survival in patients initially assigned to the control arm and obscuring the treatment effect, as demonstrated in ATTRACTION-4[6]
- This post hoc analysis evaluated the impact of subsequent-IO use on OS in patients enrolled in RATIONALE-305

METHODS

- In the RATIONALE-305 trial, patients were randomized 1:1 to receive tislelizumab (200 mg intravenous every 3 weeks) or placebo + CAPOX or cisplatin + 5-FU for up to six cycles, followed by maintenance tislelizumab or placebo, with optional capecitabine continuation after CAPOX (**Figure 1**)
- In this post hoc analysis, patients who had post-progression survival were stratified according to subsequent IO use in the second- or third-line setting (yes vs no)
- A two-stage adjustment method was applied to estimate the effect of subsequent IO on post-progression survival and subsequently to estimate counterfactual OS assuming that no patients received subsequent IO after progression; adjusted treatment effect was estimated using a cox model stratified by the prespecified randomization factor; 95% CIs were obtained from 2000 bootstrap samples

Figure 1. Study Design

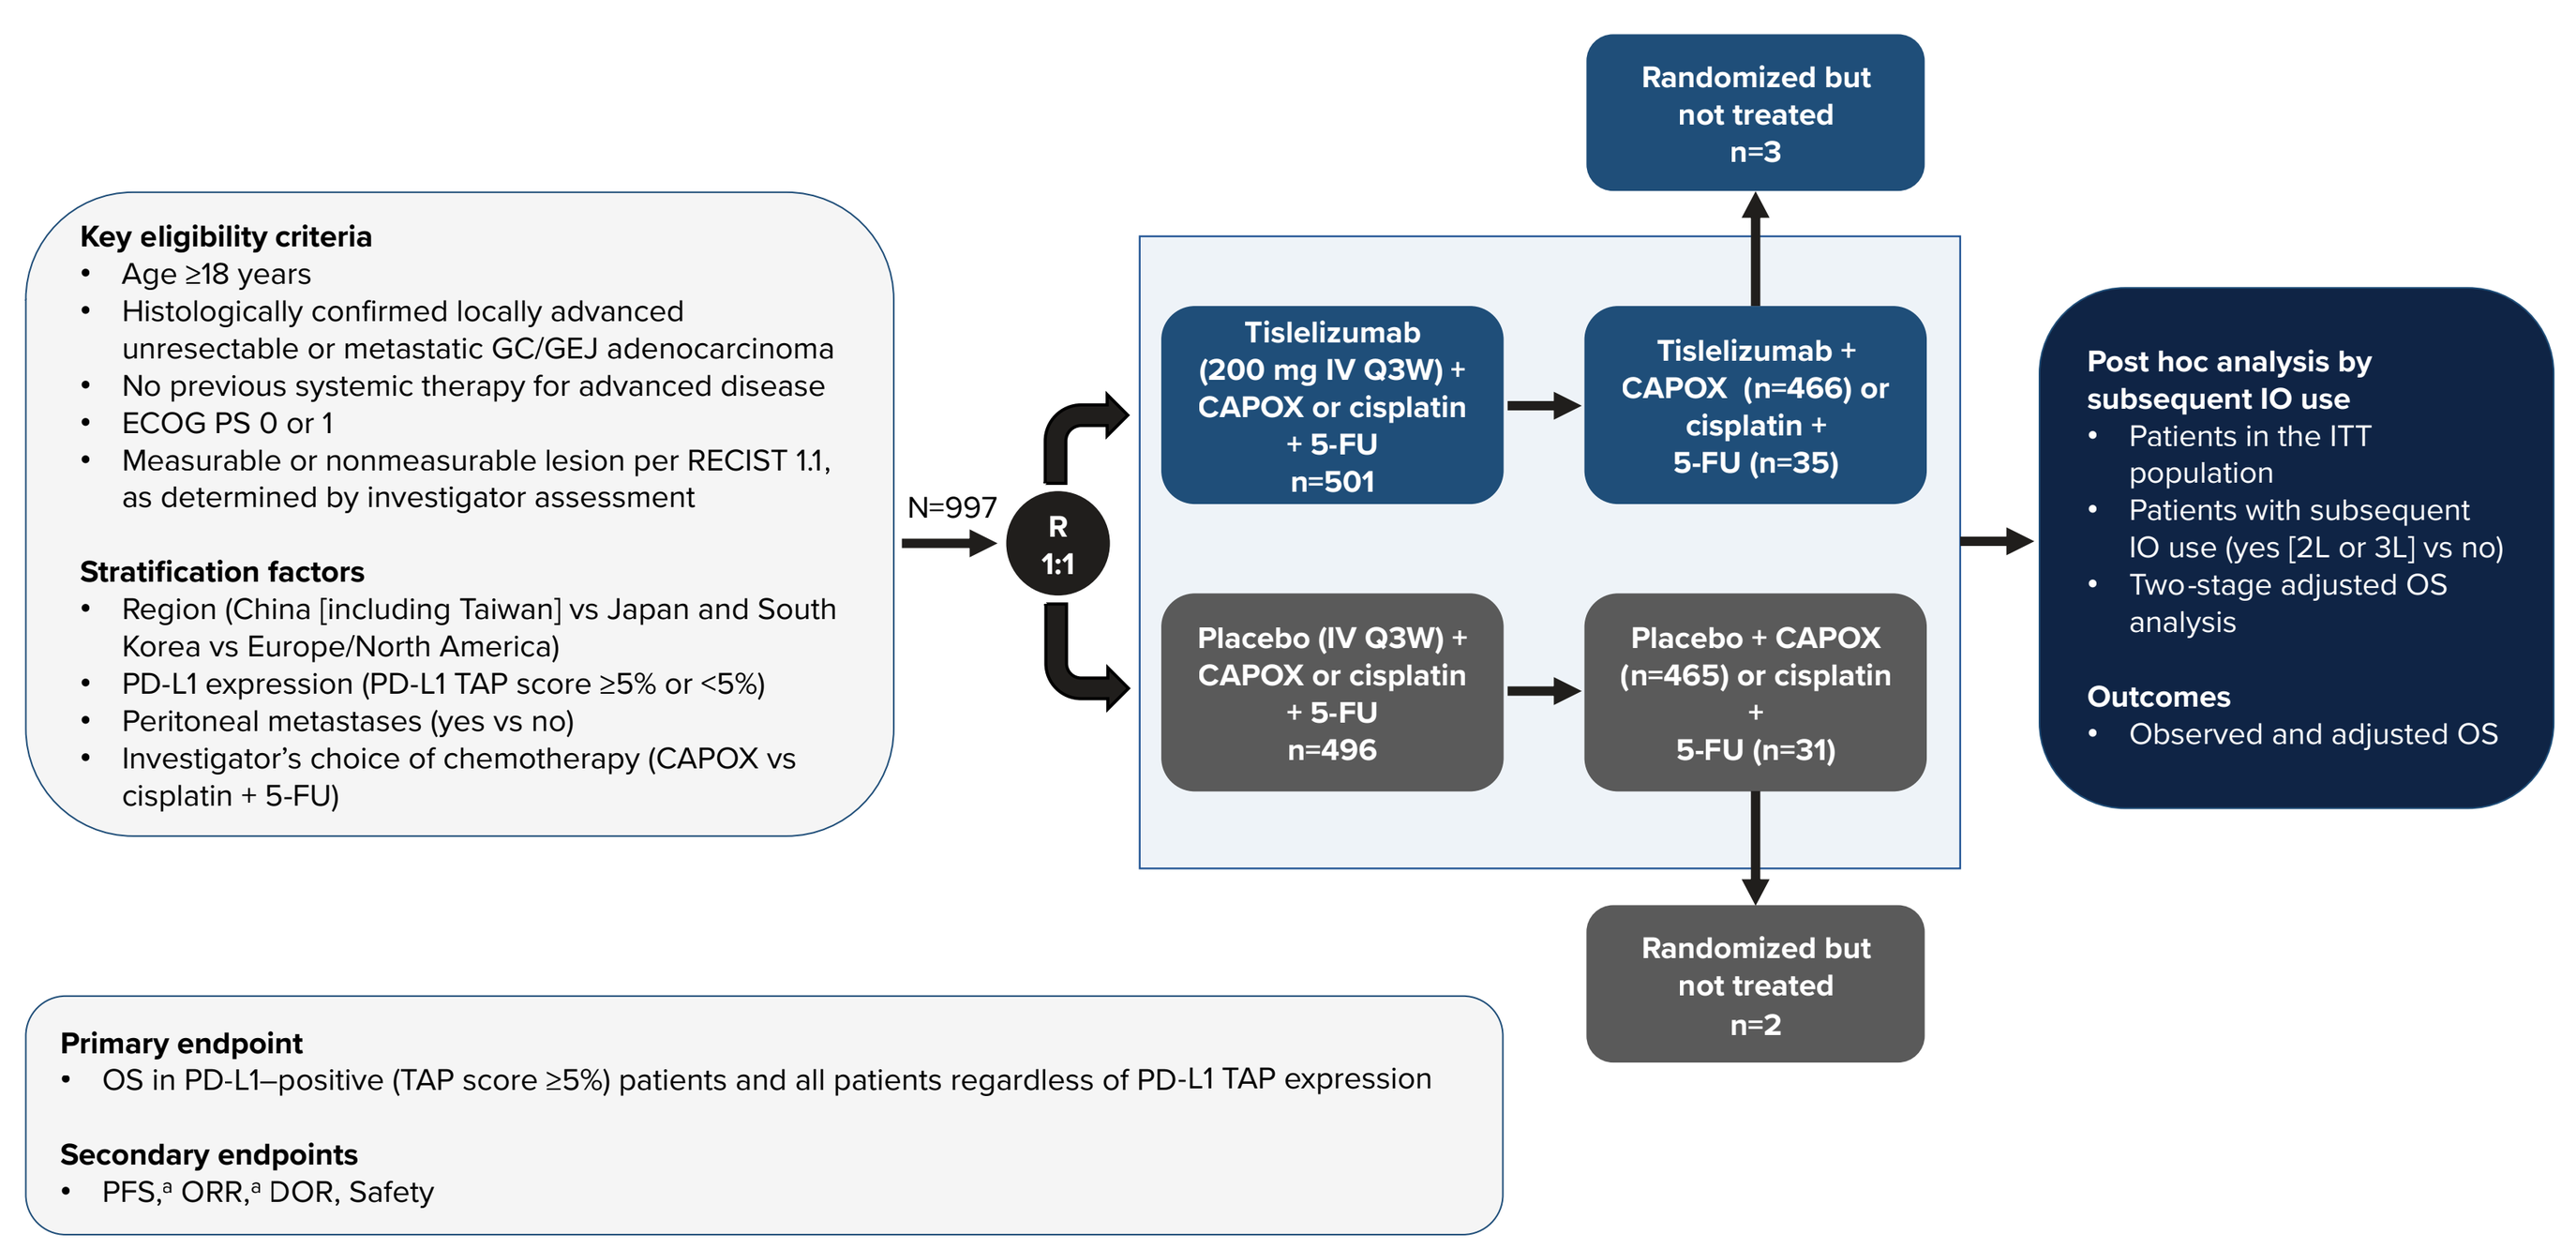
Key eligibility criteria
Age ≥18 years
Histologically confirmed locally advanced unresectable or metastatic GC/GEJ adenocarcinoma
No previous systemic therapy for advanced disease
ECOG PS 0 or 1
Measurable or nonmeasurable lesion per RECIST 1.1, as determined by investigator assessment
Stratification factors
Region (China [including Taiwan] vs Japan and South Korea vs Europe/North America)
PD-L1 expression (PD-L1 TAP score ≥5% or <5%)
Peritoneal metastases (yes vs no)
Investigator's choice of chemotherapy (CAPOX vs cisplatin + 5-FU)
N=997
R 1:1
Tislelizumab (200 mg IV Q3W) + CAPOX or cisplatin + 5-FU n=501
Tislelizumab + CAPOX (n=466) or cisplatin + 5-FU (n=35)
Randomized but not treated n=3
Placebo (IV Q3W) + CAPOX or cisplatin + 5-FU n=496
Placebo + CAPOX (n=465) or cisplatin + 5-FU (n=31)
Randomized but not treated n=2
Post hoc analysis by subsequent IO use
Patients in the ITT population
Patients with subsequent IO use (yes [2L or 3L] vs no)
Two-stage adjusted OS analysis
Outcomes
Observed and adjusted OS
Primary endpoint
OS in PD-L1–positive (TAP score ≥5%) patients and all patients regardless of PD-L1 TAP expression
Secondary endpoints
PFS,a ORR,a DOR, Safety

[a]Investigator assessed per RECIST v1.1.
Abbreviations: 2L, second-line; 3L, third-line; 5-FU, 5-fluorouracil; CAPOX, capecitabine and oxaliplatin; DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; GC, gastric cancer; GEJ, gastroesophageal junction; IO, immunotherapy; ITT, intent-to-treat; IV, intravenous; ORR, overall response rate; OS, overall survival; PD-L1, programmed cell death 1 ligand 1; PFS, progression-free survival; Q3W, every 3 weeks; R, randomization; RECIST, Response Evaluation Criteria in Solid Tumors; TAP, tumor area positivity.

RESULTS

- As of February 28, 2024, 894 patients survived beyond disease progression and therefore had an opportunity to receive subsequent treatment (**Table 1**)
- Patients who received subsequent IO were more frequently from East Asia than those who did not receive subsequent IO (**Table 1**)
- Objective response rates were 48.1%, 52.1%, 40.3%, and 47.6% in patients who received tislelizumab + CT without subsequent IO, tislelizumab + CT with subsequent IO, placebo + CT without subsequent IO, and placebo + CT with subsequent IO, respectively

Table 1. Baseline Characteristics

Characteristic	Tislelizumab No subsequent-IO n=389	Tislelizumab Subsequent-IO[a] n=48	Placebo No subsequent-IO n=375	Placebo Subsequent-IO[a] n=82
Age, median, y	60.0	55.0	61.0	61.0
Age, n (%)				
<65, years	267 (68.6)	35 (72.9)	238 (63.5)	56 (68.3)
≥65, years	122 (31.4)	13 (27.1)	137 (36.5)	26 (31.7)
Sex, n (%)				
Male	269 (69.2)	28 (58.3)	267 (71.2)	55 (67.1)
Female	120 (30.8)	20 (41.7)	108 (28.8)	27 (32.9)
Race, n (%)				
Asian	290 (74.6)	46 (95.8)	274 (73.1)	75 (91.5)
White	94 (24.2)	1 (2.1)	89 (23.7)	3 (3.7)
Region, n (%)				
East Asia	290 (74.6)	46 (95.8)	274 (73.1)	75 (91.5)
Rest of World	99 (25.4)	2 (4.2)	101 (26.9)	7 (8.5)
ECOG PS, n (%)				
0	131 (33.7)	22 (45.8)	111 (29.6)	35 (42.7)
1	258 (66.3)	26 (54.2)	264 (70.4)	47 (57.3)
TAP Score, n (%)				
≥5%	217 (55.8)	22 (45.8)	201 (53.6)	48 (58.5)
<5%	172 (44.2)	26 (54.2)	174 (46.4)	34 (41.5)
Metastatic sites, n (%)				
0-2 sites	262 (67.4)	30 (62.5)	NA	61 (74.4)
≥3 sites	127 (32.6)	18 (37.5)	NA	21 (25.6)
Liver metastasis, n (%)	155 (39.8)	12 (25.0)	142 (37.9)	31 (37.8)
Peritoneal metastasis, n (%)	164 (42.2)	28 (58.3)	164 (43.7)	33 (40.2)
Primary tumor location, n (%)				
GEJ	73 (18.8)	7 (14.6)	70 (18.7)	19 (23.2)
Stomach	316 (81.2)	41 (85.4)	304 (81.1)	63 (76.8)
Prior (neo)adjuvant tx or surgery, n (%)	120 (30.8)	24 (50.0)	125 (33.3)	31 (37.8)

[a]For second or third line.
Abbreviations: ECOG PS, Eastern Cooperative Oncology Group performance status; GEJ, gastroesophageal junction; IO, immunotherapy; Tx, treatment.

Survival Outcomes in the Intent-to-Treat Population

- In the overall population, tislelizumab + CT significantly improved mOS vs placebo + CT (**Figure 2**)
 - mOS: 15.0 months vs 12.9 months, respectively (HR, 0.79; 95% CI, 0.69-0.90), with higher 4-year OS rates (16.4% vs 8.3%)

Figure 2. OS by Treatment Arm

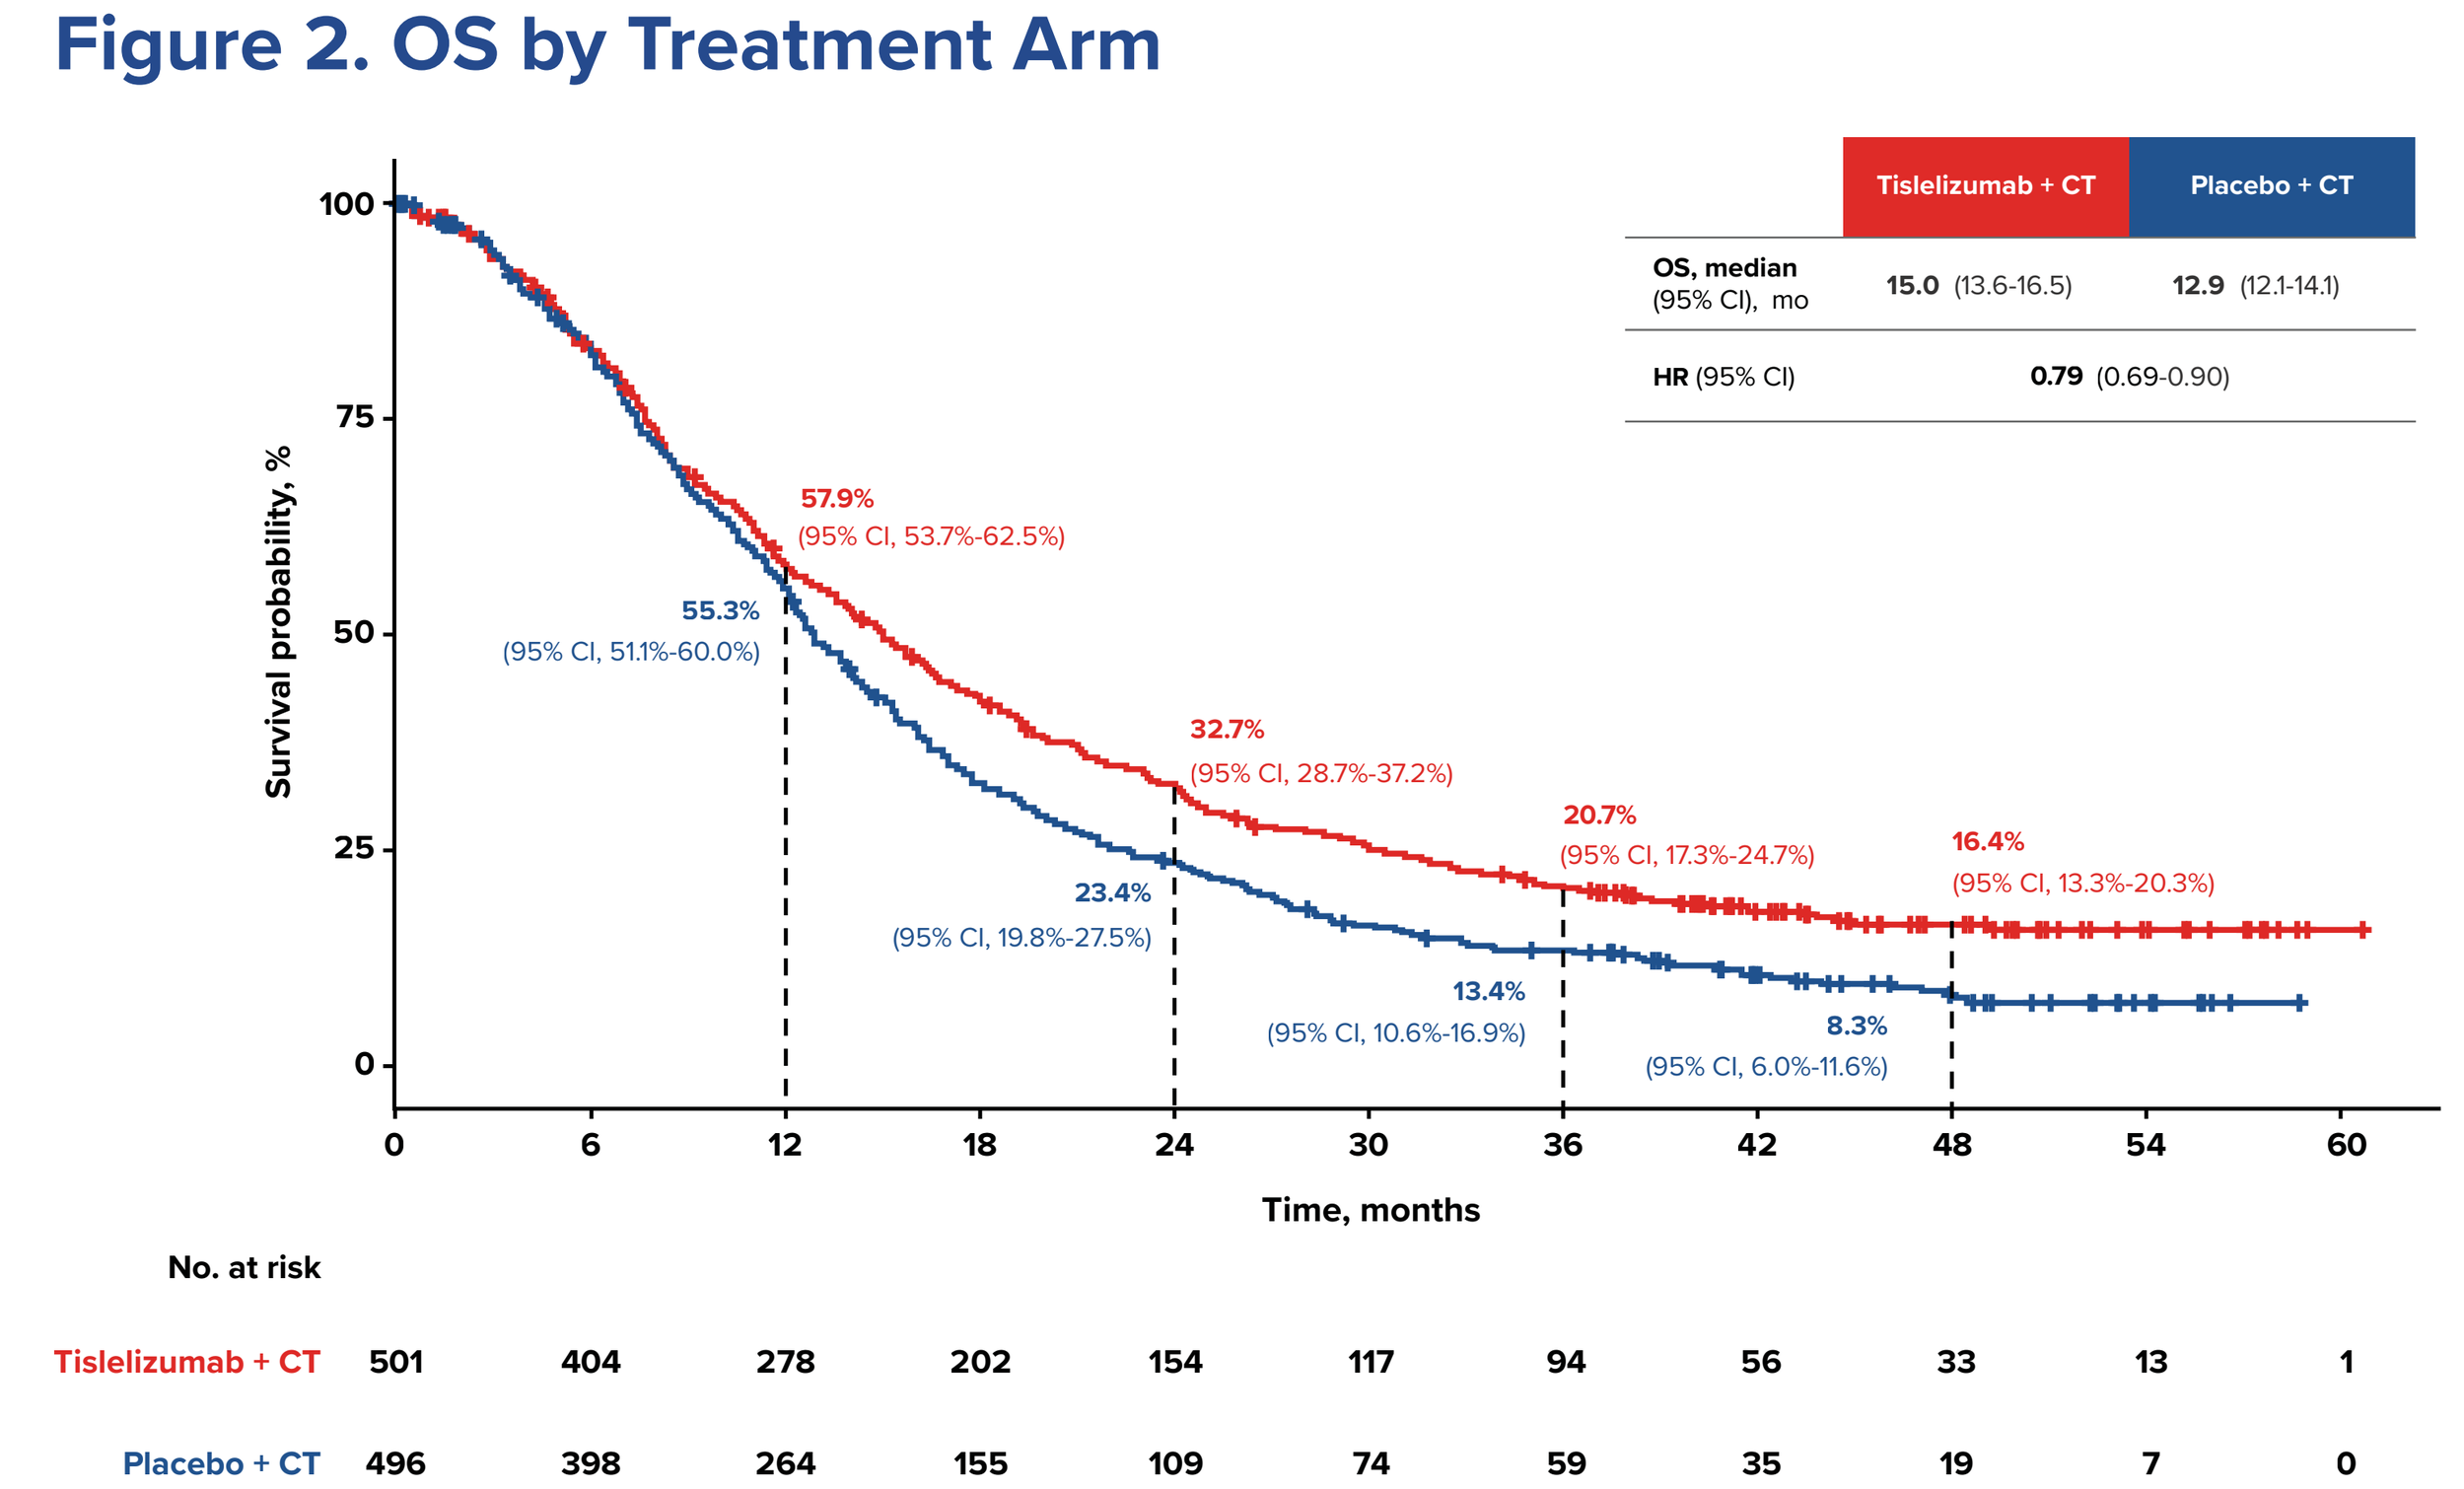
Tislelizumab + CT
Placebo + CT
OS, median (95% CI), mo 15.0 (13.6-16.5) 12.9 (12.1-14.1)
HR (95% CI) 0.79 (0.69-0.90)
Survival probability, %
57.9% (95% CI, 53.7%-62.5%)
55.3% (95% CI, 51.1%-60.0%)
32.7% (95% CI, 28.7%-37.2%)
23.4% (95% CI, 19.8%-27.5%)
20.7% (95% CI, 17.3%-24.7%)
13.4% (95% CI, 10.6%-16.9%)
16.4% (95% CI, 13.3%-20.3%)
8.3% (95% CI, 6.0%-11.6%)
Time, months
No. at risk
Tislelizumab + CT 501 404 278 202 154 117 94 56 33 13 1
Placebo + CT 496 398 264 155 109 74 59 35 19 7 0

A two-stage Weibull accelerated failure time model was used.
Abbreviations: CT, chemotherapy; HR, hazard ratio; OS, overall survival.

Survival Outcomes by Treatment and Subsequent IO Use

- Patients who received subsequent IO had longer mOS than those who did not in both treatment arms (**Figure 3**)
 - With subsequent IO: tislelizumab + CT, 23.2 months (95% CI, 17.2-26.4) vs placebo + CT, 16.9 months (95% CI, 14.7-23.4)
 - Without subsequent IO: tislelizumab + CT, 15.6 months (95% CI, 14.0-17.1) vs placebo + CT, 12.7 months (95% CI, 11.8-14.1)

Figure 3. OS by Treatment and Subsequent IO Use

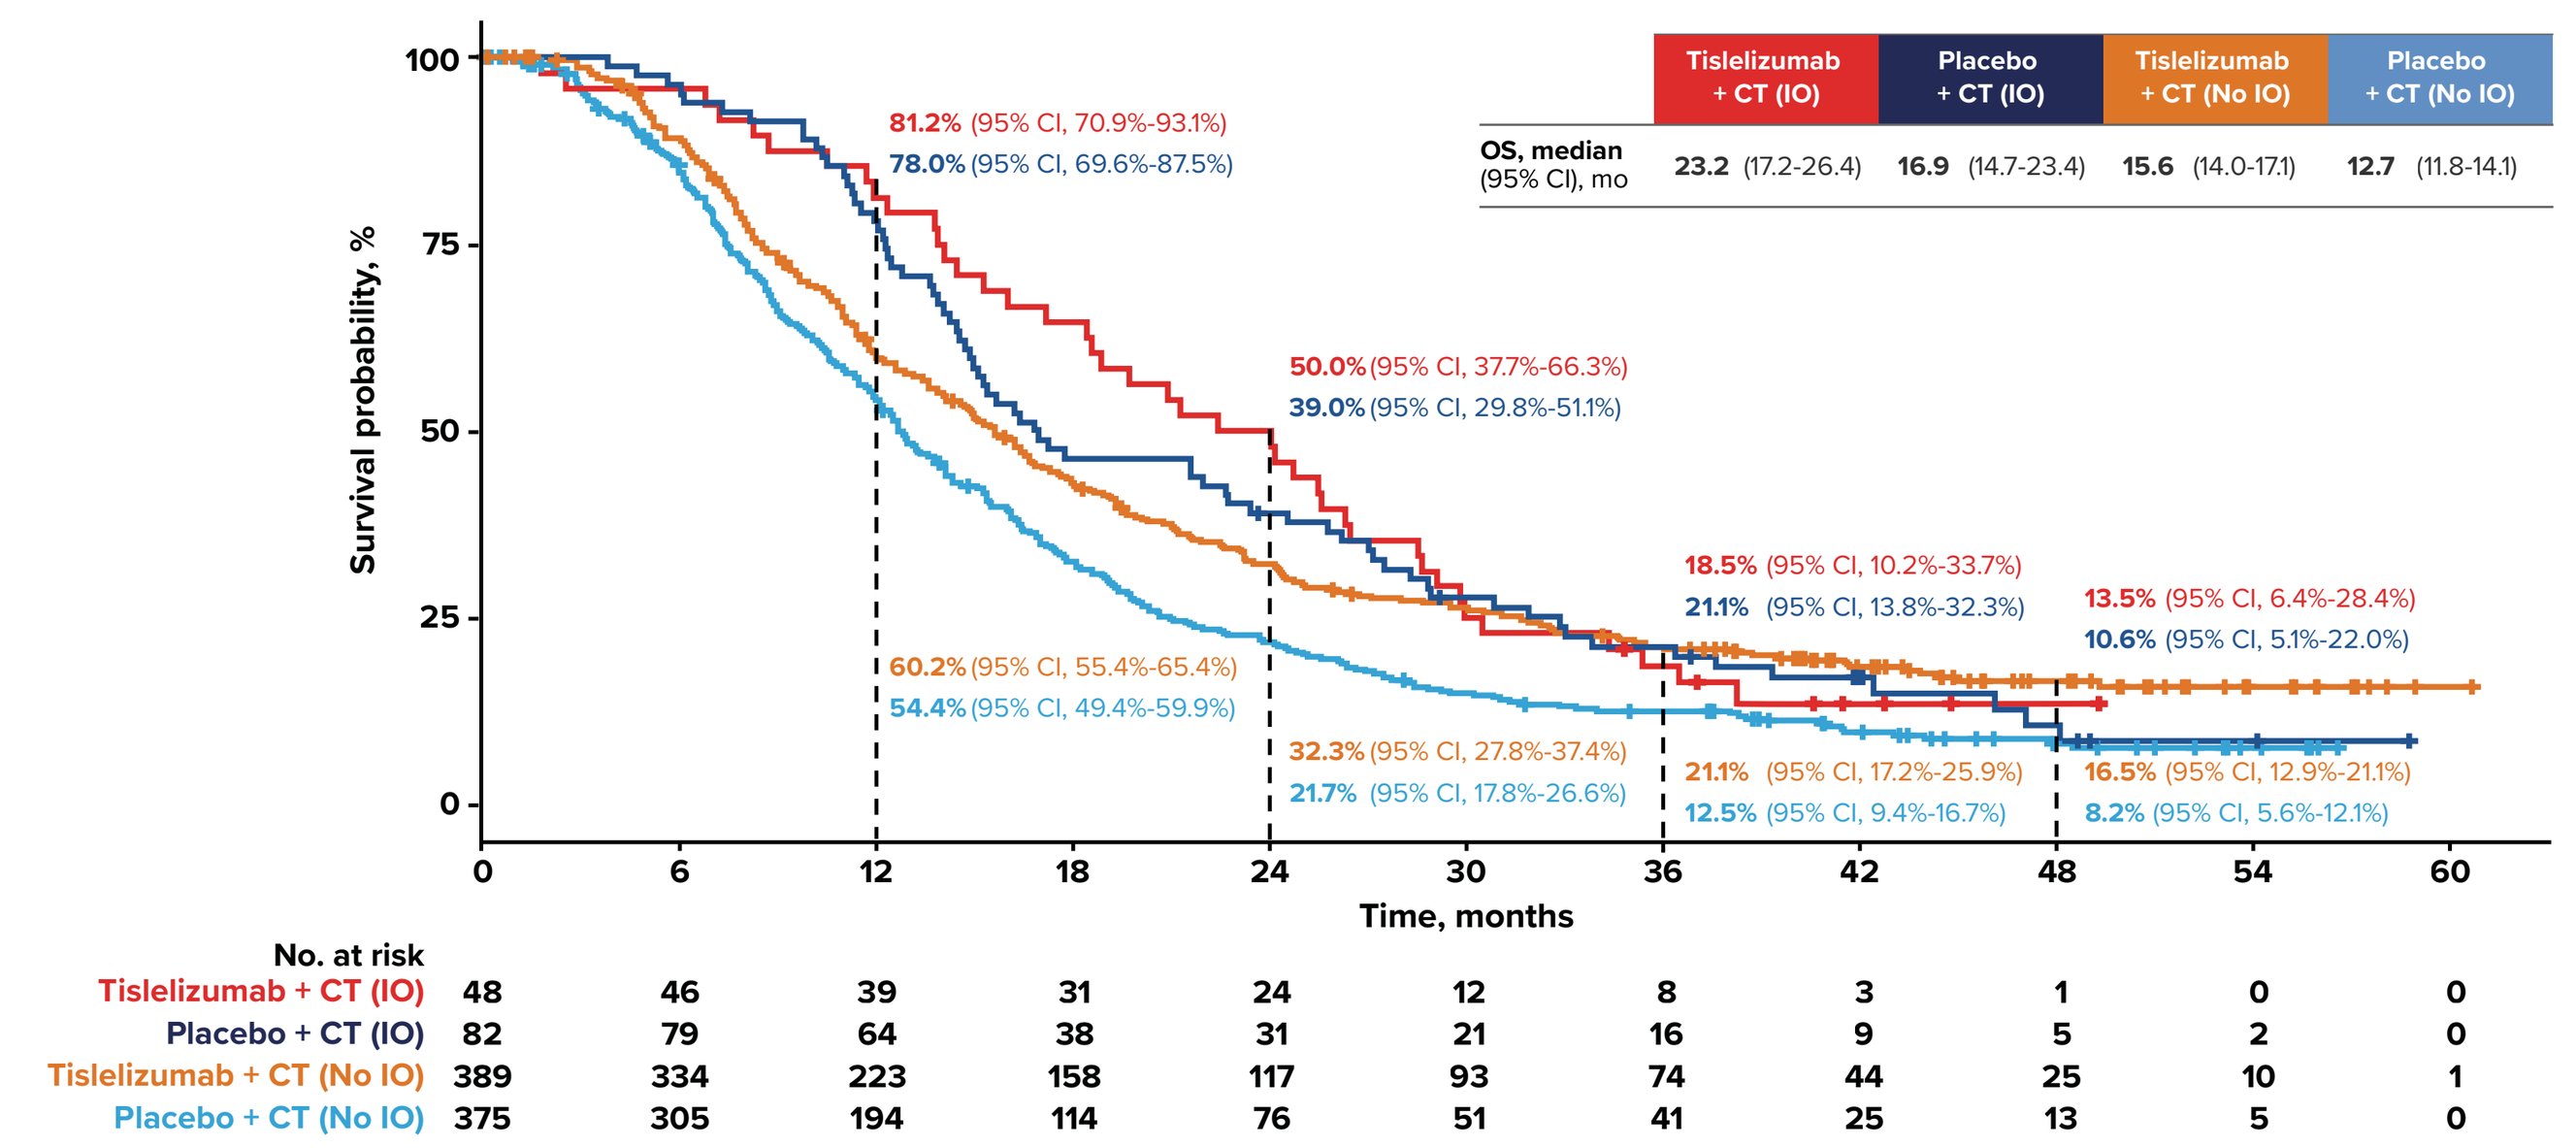
Tislelizumab + CT (IO)
Placebo + CT (IO)
Tislelizumab + CT (No IO)
Placebo + CT (No IO)
OS, median (95% CI), mo 23.2 (17.2-26.4) 16.9 (14.7-23.4) 15.6 (14.0-17.1) 12.7 (11.8-14.1)
Survival probability, %
81.2% (95% CI, 70.9%-93.1%)
78.0% (95% CI, 69.6%-87.5%)
60.2% (95% CI, 55.4%-65.4%)
54.4% (95% CI, 49.4%-59.9%)
50.0% (95% CI, 37.7%-66.3%)
39.0% (95% CI, 29.8%-51.1%)
32.3% (95% CI, 27.8%-37.4%)
21.7% (95% CI, 17.8%-26.6%)
18.5% (95% CI, 10.2%-33.7%)
21.1% (95% CI, 13.8%-32.3%)
21.1% (95% CI, 17.2%-25.9%)
12.5% (95% CI, 9.4%-16.7%)
13.5% (95% CI, 6.4%-28.4%)
10.6% (95% CI, 5.1%-22.0%)
16.5% (95% CI, 12.9%-21.1%)
8.2% (95% CI, 5.6%-12.1%)
Time, months
No. at risk
Tislelizumab + CT (IO) 48 46 39 31 24 12 8 3 1 0 0
Placebo + CT (IO) 82 79 64 38 31 21 16 9 5 2 0
Tislelizumab + CT (No IO) 389 334 223 158 117 93 74 44 25 10 1
Placebo + CT (No IO) 375 305 194 114 76 51 41 25 13 5 0

A two-stage Weibull accelerated failure time model was used.
Abbreviations: CT, chemotherapy; HR, hazard ratio; IO, immunotherapy; OS, overall survival.

Survival Adjusted for Subsequent IO Switching

- Adjustment for subsequent IO use resulted in a greater estimated mOS treatment effect of tislelizumab + CT vs placebo + CT (**Figure 4**)
 - mOS: 14.3 months vs 12.1 months, respectively (HR, 0.77; 95% CI, 0.67-0.88), with higher 4-year OS rates (16.1% vs 7.8%)

Figure 4. OS Adjusted for Subsequent IO Switching

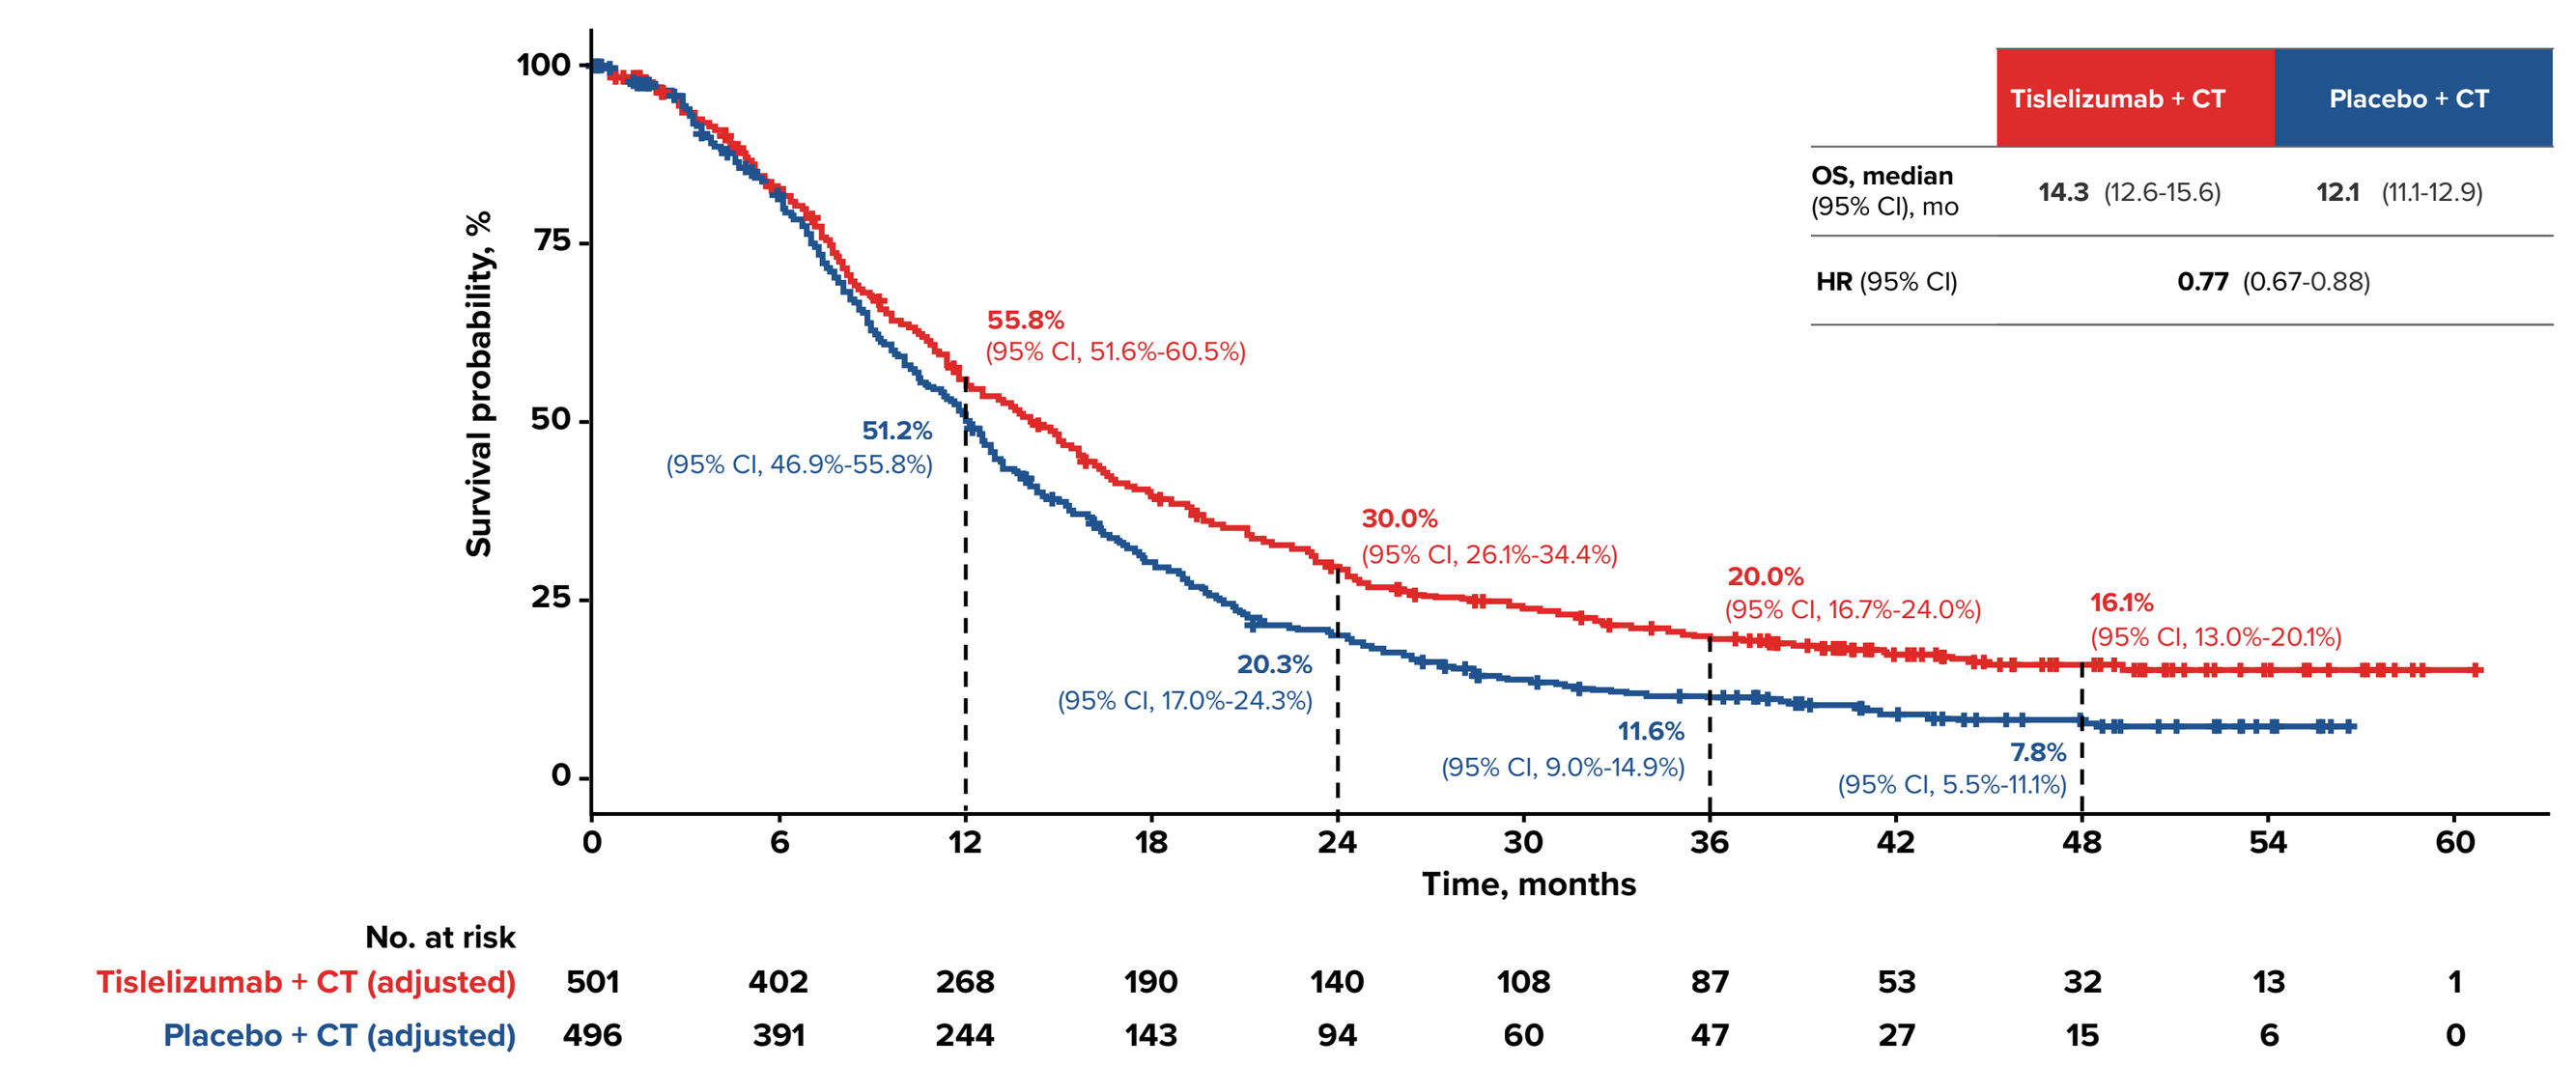
Tislelizumab + CT
Placebo + CT
OS, median (95% CI), mo 14.3 (12.6-15.6) 12.1 (11.1-12.9)
HR (95% CI) 0.77 (0.67-0.88)
Survival probability, %
55.8% (95% CI, 51.6%-60.5%)
51.2% (95% CI, 46.9%-55.8%)
30.0% (95% CI, 26.1%-34.4%)
20.3% (95% CI, 17.0%-24.3%)
20.0% (95% CI, 16.7%-24.0%)
11.6% (95% CI, 9.0%-14.9%)
16.1% (95% CI, 13.0%-20.1%)
7.8% (95% CI, 5.5%-11.1%)
Time, months
No. at risk
Tislelizumab + CT (adjusted) 501 402 268 190 140 108 87 53 32 13 1
Placebo + CT (adjusted) 496 391 244 143 94 60 47 27 15 6 0

A two-stage Weibull accelerated failure time model was used.
Abbreviations: CT, chemotherapy; HR, hazard ratio; OS, overall survival.

REFERENCES

1. Janjigian YY, et al. *Lancet.* 2021;398(10294):27-40.
2. Rha SY, et al. *Lancet Oncol*. 2023;24(11):1181-1195.
3. Qiu MZ, et al. *BMJ.* 2024;385:e078876.
4. Zhang T, et al. *Cancer Immunol Immunother*. 2018;67:1079-1090.
5. Correa MC, et al. *Ann Oncol*. 2024;35(suppl 2):S893-S894.
6. Kang YK, et al. *Lancet Oncol.* 2022;23(2):234-247.

DISCLOSURES

MM: Advisory board: BMS, Servier, Amgen, Lilly, Bayer, BeOne Medicines, Ltd, Novartis, Taiho, Daiichi Sankyo, MD; Honoraria: MSD, BMS, Falk foundation, AIO, BeOne Medicines, Ltd, Amgen; Expert testimony: Sanofi; Other: EORTC, Chair.

ACKNOWLEDGMENTS

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. The authors also thank Yaling Xu for her contributions to the development of this work. This study was sponsored by BeOne Medicines, Ltd. Medical writing support was provided by Colton Smith, PhD of Nucleus Global, an Inizio company, and was supported by BeOne Medicines.

Presented at the ESMO Congress 2026; October 23-27, 2026; Madrid, Spain