Comparative Efficacy of Zanubrutinib Versus Fixed-Duration Acalabrutinib Plus Venetoclax for First-Line Treatment of Chronic Lymphocytic Leukemia (CLL): A Matching-Adjusted Indirect Comparison (MAIC)

CLL-1032

Talha Munir,[1] Keri Yang,[2] Sheng Xu,[3] Rhys Williams,[2] Mazyar Shadman[4]

[1]Leeds Teaching Hospital NHS Trust, Leeds, West Yorkshire, UK; [2]BeOne Medicines Ltd., USA, San Carlos, CA, USA; [3]BeOne Medicines Ltd., Shanghai, China; [4]Fred Hutchinson Cancer Center and University of Washington, Seattle, WA, USA

Conclusions

- This MAIC examined the relative efficacy of zanubrutinib versus acalabrutinib plus the B-cell lymphoma-2 inhibitor venetoclax (AV) and demonstrated a significant progression-free survival (PFS) advantage for zanubrutinib over AV regimen
- Results should be interpreted with considerations of typical MAIC model assumptions. Future analyses upon trial data maturation are warranted

Background

- In treatment-naïve (TN) CLL, the efficacy of continuous zanubrutinib has been investigated in the phase 3 SEQUOIA trial (NCT03336333)[1,2]
- Efficacy of fixed duration combination regimen AV was evaluated in the phase 3 AMPLIFY trial (NCT03836261), with interim analysis results first presented in Dec 2024[3] and published in Feb 2025[4]

Objective

- In the absence of head-to-head clinical trials, an anchored matching-adjusted indirect comparison (MAIC) was conducted to investigate the comparative efficacy of zanubrutinib and AV in patients with low-risk TN CLL (without del(17p) or *TP53* mutations)

Methods

- This MAIC was conducted using datasets with similar median follow-ups (SEQUOIA, 43.7 months; AMPLIFY, 41.0 months)
- With the assumption of bendamustine plus rituximab (BR) and fludarabine plus cyclophosphamide and rituximab (FCR)/BR treated as common control arms, SEQUOIA and AMPLIFY can be linked through FCR/BR and the comparison of zanubrutinib and AV was conducted in an anchored MAIC
- Individual patient data of low-risk (without del(17p) or *TP53* mutations) zanubrutinib patients in SEQUOIA were re-weighted to match the key population characteristics of AMPLIFY (**Figure 1**)
- Population adjustments considered prognostic factors or effect modifiers, including age, sex, Eastern Cooperative Oncology Group Performance Status (ECOG PS), disease stage, del(11q), and immunoglobulin heavy chain variable (IGHV) gene mutation status (**Table 1**)
- Reconstructed individual patient data for AMPLIFY were generated from digitized Kaplan-Meier (KM) curves of progression-free survival (PFS)
- Weighted Cox proportional hazard regression was used to derive relative treatment effect estimates for PFS
- Sensitivity analyses were conducted in model scenarios of different matching variables
- At the time of this abstract submission, an anchored MAIC was conducted based on data availability of interim analysis of AMPLIFY[3] that reported only independent review committe (IRC)-assessed PFS (IRC-PFS) and common control arm of FCR/BR. Based on data availability of the AMPLIFY publication from 2025,[4] this poster presents analysis of INV-PFS, as well as additional sensitivity analysis of IRC-PFS, and an unanchored MAIC without the FCR/BR common control arm assumption

Figure 1. Overall Methodology Details

SEQUOIA
(ITT, n=479)

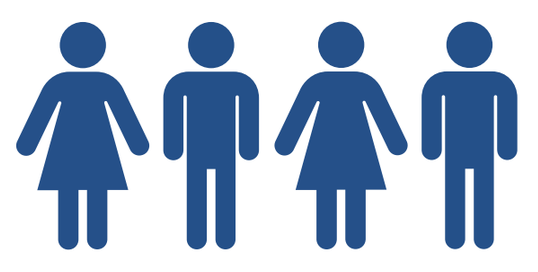

Individual patient-level data (IPD)
(Median follow-up: 43.7 months)

AMPLIFY
(AV, n=291; FCR/BR, n=290)

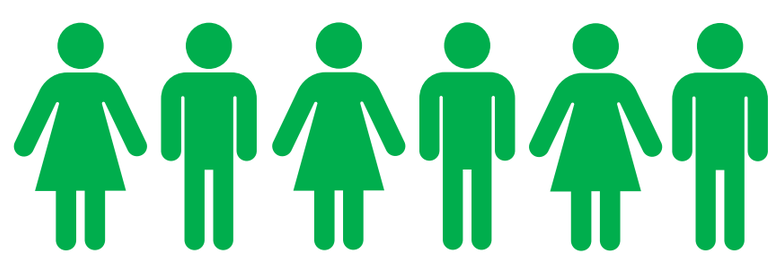

Published aggregate data
(Median follow-up: 41.0 months)

Variables identified as potential treatment effect modifiers or prognostic factors for matching

Age, sex, ECOG PS, disease stage, del(11q), IGHV gene mutation status, geographic region, complex karyotype, CIRS, creatinine clearance

Sensitivity analyses of model scenarios to consider impact of different matching values, IRC-PFS, and unanchored MAIC of FCR and BR

Matching, reweighting, and adjusting variables

- Zanubrutinib low-risk population (SEQUOIA), n=389
- After population adjustments, ESS=126 for SEQUOIA

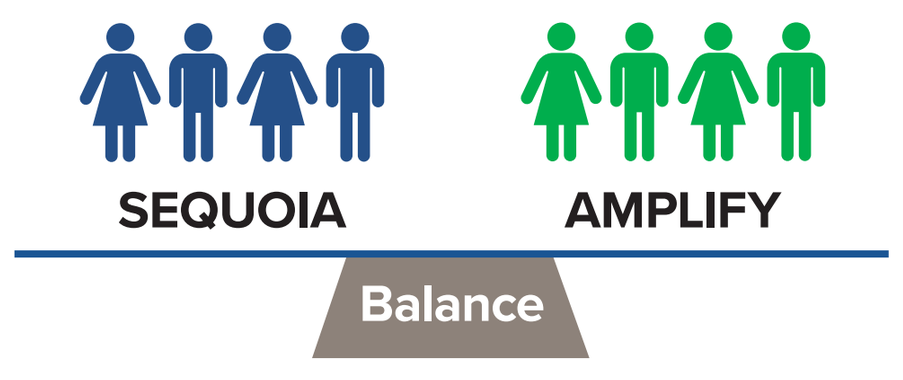
SEQUOIA
AMPLIFY
Balance

Outcome

INV-PFS

HRs for INV-PFS: Weighted Cox proportional hazard regression

AV, acalabrutinib plus venetoclax; BR, bendamustine + rituximab; CIRS, Cumulative Illness Rating Scale; del(11q), chromosome 11q deletion; ECOG PS, Eastern Cooperative Oncology Group Performance Status; ESS, effective sample size; FCR, fludarabine-cyclophosphamide-rituximab; HR, hazard ratio; IGHV, immunoglobulin heavy-chain variable; INV-PFS, investigator-assessed progression-free survival; IRC-PFS, independent review committee-assessed progression-free survival; ITT, intent-to-treat; MAIC, matching-adjusted indirect comparison.

Table 1. Variables Matched in the Base Case and Sensitivity Analyses

Variables	Main analysis: Unadjusted population	Main analysis: Base case adjusted population	S1	S2	S3	S4	S5	S6	S7	S8	S9	S10	S11	S12	S13
Sample size for SEQUOIA, zanubrutinib	N=389	ESS=126	ESS: 82	125	38	120	291	55	155	343	127	147	126	129	126
Age >65 (vws ≤65)		√	√	√	√	√		√	√		√	√	√	√	√
Male		√	√	√	√	√				√	√	√	√	√	
ECOG PS=0-1 (vs 2)		√	√	√	√	√				√		√	√	√	√
Binet stage AB or Rai 0-II (vs C or III-IV)		√	√	√	√	√				√	√		√	√	√
Del(11q)		√	√	√	√	√				√	√	√		√	√
IGHV unmutated		√	√	√	√	√				√	√	√	√		√
Geographic region			√	√											
Complex karyotype ≥3 abnormalities			√												
CIRS >6					√		√	√							
Creatinine clearance <60 mL/min						√									

CIRS, Cumulative Illness Rating Scale; del(11q), chromosome 11q deletion; ECOG PS, Eastern Cooperative Oncology Group performance status; ESS, effective sample size; IGHV, immunoglobulin heavy chain variable.

Results

Base Case

- After population adjustment, the effective sample size (ESS) for SEQUOIA was 126 (**Table 2**)

Table 2. Baseline Characteristics of Low-Risk Patients in SEQUOIA Pre- and Post-Matching and in AMPLIFY

Population characteristic	AMPLIFY N=581	SEQUOIA Pre-matching N=389	SEQUOIA Post-matching ESS=126
Age >65 (vs ≤65)	26.8%	78.7%	26.8%
Male	64.5%	61.7%	64.5%
ECOG PS=0-1 (vs 2)	91.8%	93.3%	91.8%
Binet stage AB or Rai 0-II (vs C or III-IV)	56.2%	70.2%	56.2%
Del(11q)	17.6%	19.5%	17.6%
IGHV unmutated	58.6%	52.7%	58.6%

Note: Unweighted population included only patients with non-missing baseline characteristics regarding all selected matching factors.
del(11q), chromosome 11q deletion; ECOG PS, Eastern Cooperative Oncology Group performance status; ESS, effective sample size; IGHV, immunoglobulin heavy chain variable.

INV-PFS

- The unadjusted comparison of INV-PFS for zanubrutinib in SEQUOIA vs AV in AMPLIFY demonstrated a significant treatment benefit for zanubrutinib with hazard ratio (HR) of 0.47 (95% confidence interval [CI]: 0.28-0.77; *P*=.003)
- Population-adjusted INV-PFS for zanubrutinib vs AV indicated superior INV-PFS in favor of zanubrutinib with HR of 0.26 (95% CI: 0.13-0.54; *P*=.0003) (**Figure 2**)
- The 36-month PFS rate for zanubrutinib was 85.6% before matching and 88.5% after matching in the base case, compared with 76.5% for AV (**Table 3**)

Sensitivity Analyses

- Sensitivity analyses showed consistent results in model scenarios of different matching variables (**Table 4**) as well as IRC-PFS (HR=0.23, 95% CI: 0.12-0.48, *P*<.0001) and in unanchored MAIC without the common FCR/BR control arm assumption (HR=0.44, 95%CI: 0.21-0.89, *P*=.0220)

Figure 2. KM Plot for Reweighted SEQUOIA and AMPLIFY

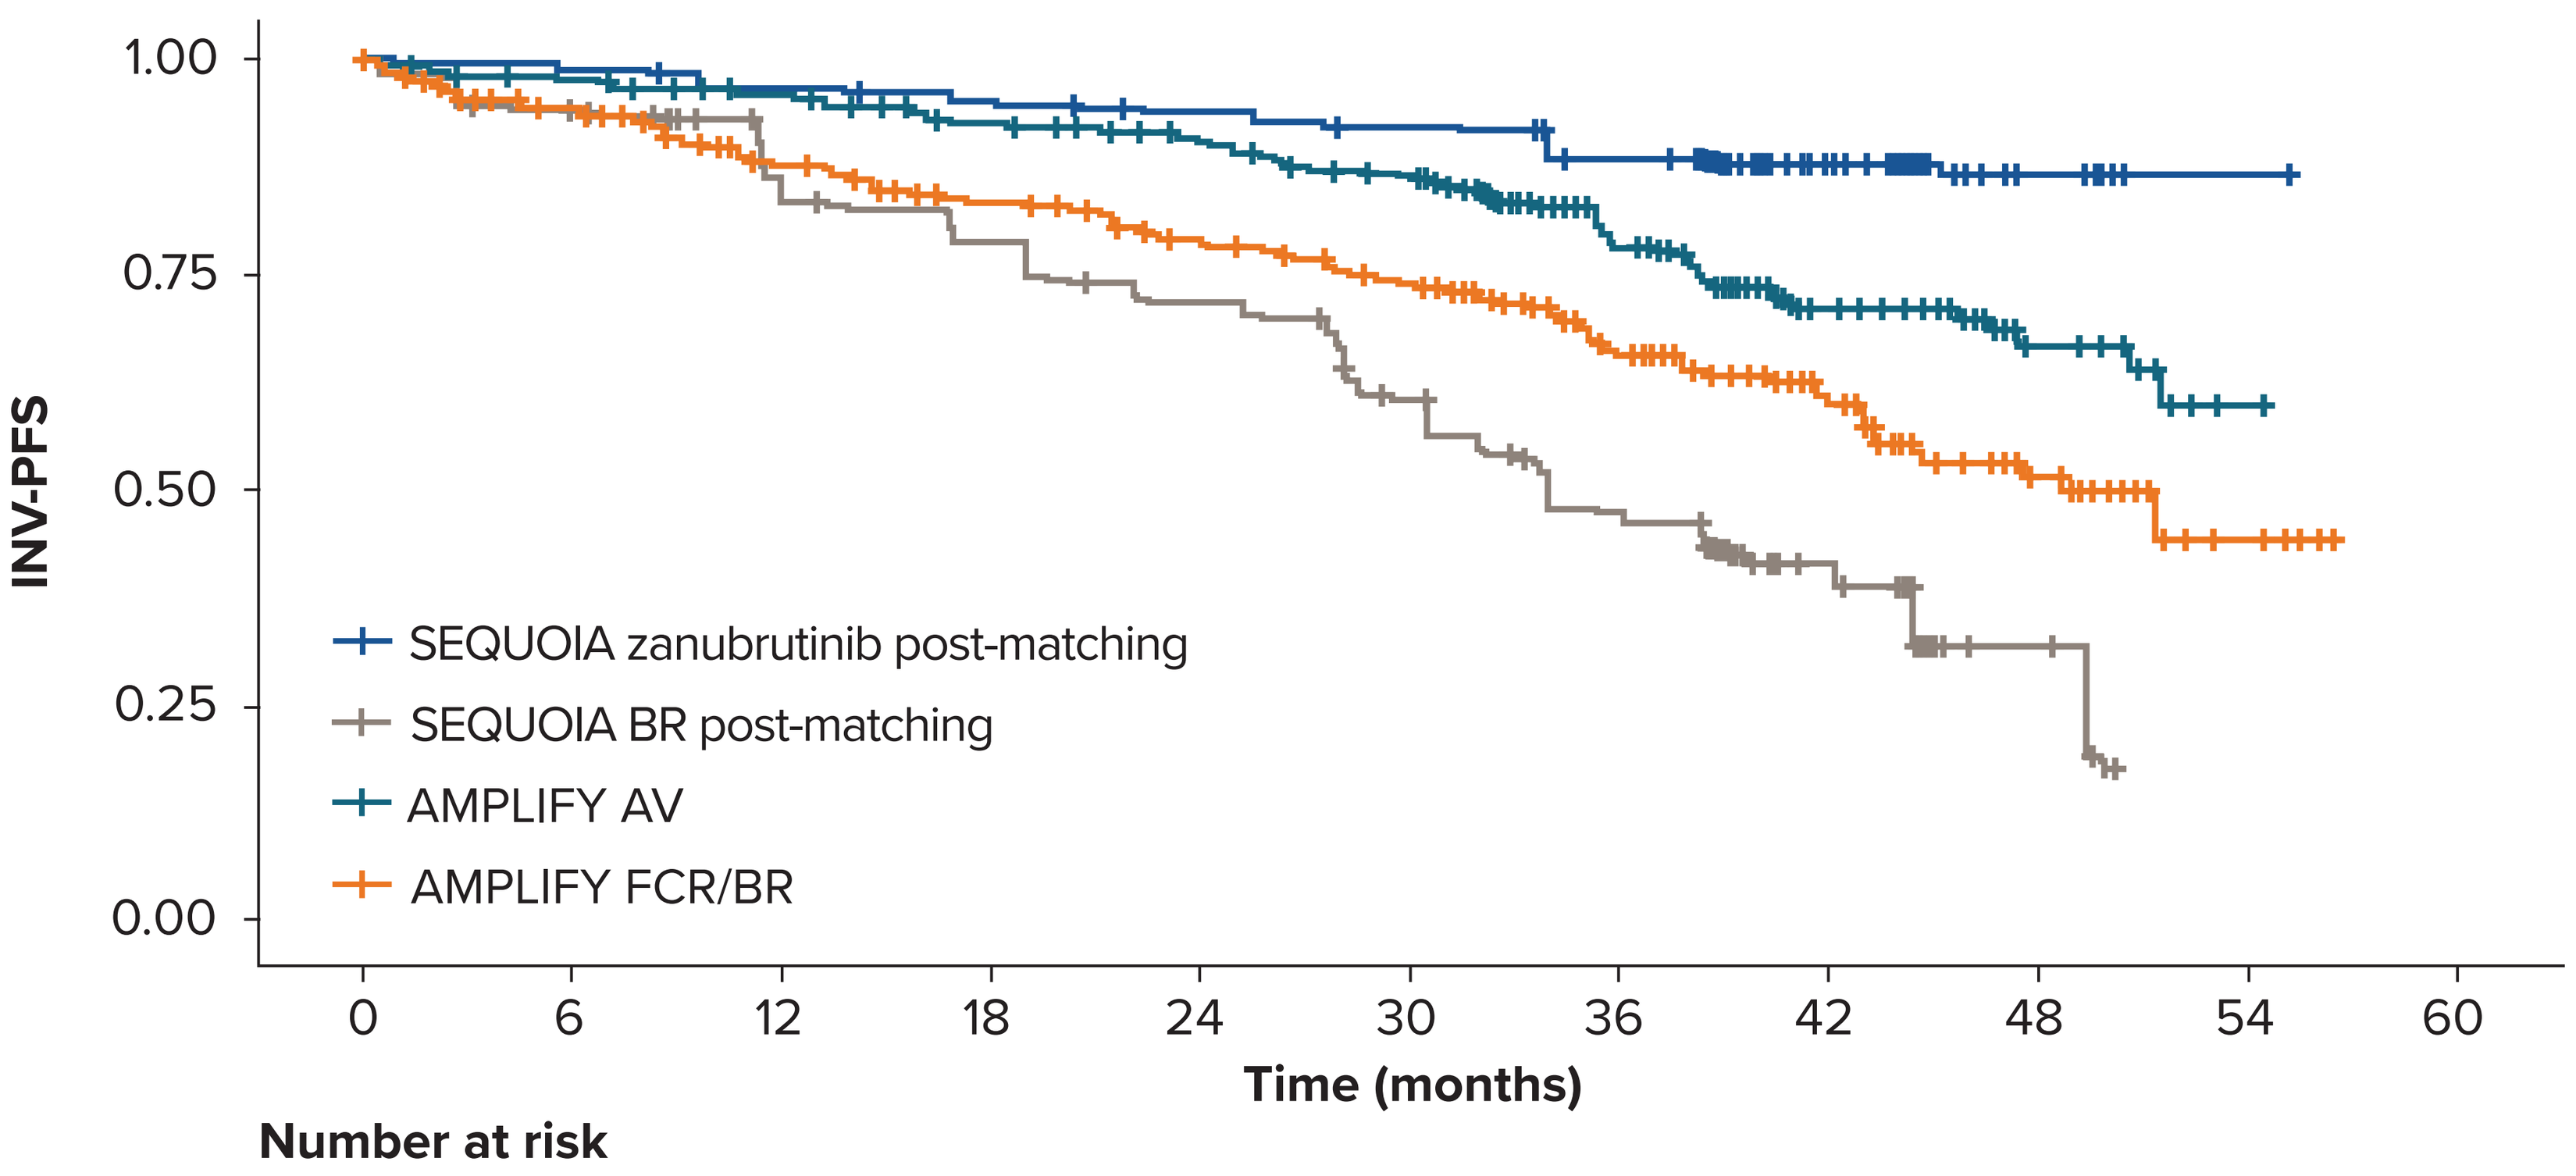
1.00
0.75
0.50
0.25
0.00
INV-PFS
SEQUOIA zanubrutinib post-matching
SEQUOIA BR post-matching
AMPLIFY AV
AMPLIFY FCR/BR
0
6
12
18
24
30
36
42
48
54
60
Time (months)

Number at risk

	0	6	12	18	24	30	36	42	48	54	60
SEQUOIA zanubrutinib post-matching	60	58	55	54	53	50	46	24	6	1	0
SEQUOIA BR post-matching	66	60	50	46	42	34	25	13	2	0	0
AMPLIFY AV	291	282	272	258	246	231	182	105	34	2	0
AMPLIFY FCR/BR	290	236	210	192	176	161	130	68	31	8	0

Time (months)

AV, acalabrutinib plus venetoclax; BR, bendamustine + rituximab; FCR, fludarabine-cyclophosphamide-rituximab; INV-PFS, investigator-assessed progression-free survival; KM, Kaplan-Meier.

Table 3. Landmark PFS Rates

Time (months)	SEQUOIA zanubrutinib (INV-PFS)	SEQUOIA BR (INV-PFS)	AMPLIFY AV[a] (INV-PFS)	AMPLIFY FCR/BR[a] (INV-PFS)	AMPLIFY AV[a] (IRC-PFS)	AMPLIFY FCR/BR[a] (IRC-PFS)
12	97%	83.6%	96%	88%	95%	88%
24	94.2%	71.8%	91%	79%	88%	79%
36	88.5%	47.8%	79%	66%	77%	67%
48	86.7%	32.2%	67%	52%	64%	49%

[a]Estimates are calculated from digitized KM curves.
AV, acalabrutinib plus venetoclax; BR, bendamustine + rituximab; FCR, fludarabine-cyclophosphamide-rituximab; INV-PFS, investigtor-assessed progression-free survival; IRC-PFS, independent review committee-assessed progression-free survival; KM, Kaplan-Meier; PFS, progression-free survival.

Table 4. INV-PFS HR Summary Table

	HR INV-PFS zanubrutinib vs AV (95% CI, *P* value)
Main analysis	
Unadjusted (low-risk)	**0.47** (0.28-0.77, *P*=.003)
Base case	**0.26** (0.13-0.54, *P*=.0003)
Sensitivity analyses	
S1	**0.15** (0.07-0.34, *P*<.0001)
S2	**0.26** (0.13-0.54, *P*=.0003)
S3	0.73 (0.29-1.84, *P*=.4992)
S4	**0.22** (0.11-0.47, *P*=.0001)
S5	**0.49** (0.27-0.86, *P*=.014)
S6	0.43 (0.15-1.21, *P*=.1092)
S7	**0.30** (0.15-0.61, *P*=.0009)
S8	**0.45** (0.26-0.76, *P*=.0033)
S9	**0.27** (0.13-0.56, *P*=.0004)
S10	**0.29** (0.14-0.59, *P*=.0008)
S11	**0.27** (0.13-0.54, *P*=.0003)
S12	**0.27** (0.14-0.54, *P*=.0002)
S13	**0.27** (0.13-0.54, *P*=.0002)

Note: bolded values indicate *P*<.05.
AV, acalabrutinib plus venetoclax; CI, confidence interval; HR, hazard ratio; INV-PFS, investigator-assessed progression-free survival.

Discussion

- In the absence of head-to-head comparative trials, the indirect comparison statistical analyses were applied to compare efficacy of zanubrutinib vs AV
- Results should be interpreted with considerations of inherent limitations of indirect comparison, such as MAIC model assumptions, ie, the assumption that cross-trial differences in patient populations can be entirely explained by matching variables

Disclosures

TM: Consulting or advising roles for Janssen-Cilag, AstraZeneca, BeOne, SOBI, Roche, AbbVie, Alexion Pharmaceuticals, and Lilly; speakers' bureau participation for AbbVie, Janssen-Cilag, Gilead Sciences, Alexion Pharmaceuticals, AstraZeneca, and SOBI; and travel, accommodations, or expenses from Janssen-Cilag, AbbVie, Alexion Pharmaceuticals, and AstraZeneca. **KY**: Employment and equity holder in BeOne. **SX**: Employment and equity holder in BeOne. **RW**: Employment and equity holder in BeOne. **MS**: Employment with Bristol Myers Squibb; consulting or advising roles for AbbVie, Genentech, AstraZeneca, Pharmacyclics, BeOne, Bristol Myers Squibb/Celgene, MorphoSys, Kite (a Gilead company), Fate Therapeutics, Lilly, Genmab, Merck, Nurix, and ADC Therapeutics; research funding from Pharmacyclics, Acerta Pharma, Merck, TG Therapeutics, BeOne, Celgene, Genentech, MustangBio, AbbVie, Sunesis Pharmaceuticals, Bristol Myers Squibb/Celgene, Genmab, and Vincerx Pharma; and stock options in Koi Biotherapeutics.

References

1. Tam CS, et al Lancet Oncol. 2022;23:1031-1043.
2. Munir T, et al Poster session presented at: EHA 2023; 2023 June 8-11; Frankfurt, Germany.
3. Brown JR, et al Blood. 2024;144(Supplement 1):1009.
4. Brown JR, et al N Engl J Med. 2025;392(8):748-762.

Acknowledgments

Medical writing support provided by SNELL Medical Communication, Inc. This study was funded by BeOne Medicines Ltd.

Correspondence: Talha Munir; tmunir@nhs.net

Presented at SOHO 2025; September 3-6, 2025; Houston, Texas