A Phase 1 Study of BGB-A3055 (Anti-CCR8) With or Without Tislelizumab (Anti-PD-1) in Patients With Solid Tumors

2523

Judith Raimbourg,[1] Martin Gutierrez,[2] Antoine Italiano,[3] Shiraj Sen,[4] Afaf Abed,[5] Jermaine Coward,[6] Muhammad Furqan,[7] Dingzhi Huang,[8] Rongbo Lin,[9] Pu Sun,[10] Zhaoyin Zhu,[11] Rachel Stern,[12] Andongfac Florinas,[12] Hua-Xin Gao,[11] Christophe Le Tourneau[13]

[1]Institut de Cancerologie de l'Ouest, Saint-Herblain, France; [2]John Theurer Cancer Center, Hackensack University Medical Center, Hackensack, NJ, USA; [3]Centre de Lutte Contre Le Cancer – Institut Bergonie, Bordeaux, France; [4]Next Oncology, San Antonio, TX, USA; [5]Linear Clinical Research, Nedlands, WA, Australia; [6]Icon Cancer Centre South Brisbane, Brisbane, QLD, Australia; [7]University of Iowa, Iowa City, IA, USA; [8]Tianjin Medical University Cancer Institute & Hospital, Tianjin, China; [9]Fujian Cancer Hospital, Fuzhou, China; [10]BeOne Medicines, Ltd, Shanghai, China; [11]BeOne Medicines, Ltd, Cambridge, MA, USA; [12]BeOne Medicines, Ltd, San Carlos, CA, USA; [13]Gustave Roussy, Paris, France

CONCLUSIONS

- BGB-A3055 as monotherapy or combined with tislelizumab demonstrated a safety profile consistent with CCR8 on-target effects and showed promising preliminary antitumor activity in heavily pretreated patients with advanced solid tumors

INTRODUCTION

- C-C motif chemokine receptor 8 (CCR8) is a G-protein-coupled receptor expressed in regulatory T cells (Tregs) and type 2 helper T (Th2) cells in the blood,[1,2] as well as the thymus and spleen[2-4]
 - CCR8 is upregulated in different types of solid tumors,[5-7] with high CCR8 expression often associated with poor prognosis[5,6,8]
- BGB-A3055 is a humanized monoclonal antibody (mAb) that targets CCR8
 - Preclinical studies have shown that BGB-A3055 is a highly potent anti-CCR8 afucosylated antibody that induces strong antibody-dependent cellular cytotoxicity activity of CCR8-expressing Tregs[9]
- Here, we present results from a phase 1a dose-escalation trial of BGB-A3055 alone (Part A) or combined with the anti-programmed cell death protein-1 mAb tislelizumab (Part B) in patients with advanced or metastatic solid tumors (NCT05935098)

METHODS

Trial Design

- This dose-escalation stage of the phase 1, open-label, multicenter, dose-escalation/dose-expansion trial was conducted across sites in Australia, the United States, France, and China, reflecting a globally enrolled patient population (**Figure 1**)

Figure 1. Study Design

Phase 1a: Dose Escalation (N=55 to 158)

Key eligibility criteria for phase 1a
- Adults ≥18 years
- Histologically/cytologically confirmed advanced or metastatic solid tumors associated with high CCR8 expression
- Previously received adequate available standard systemic therapy or for whom treatment is not available or not tolerated and had not received any prior therapy targeting CCR8
- ECOG PS ≤1
- ≥1 measurable lesion per RECIST v1.1

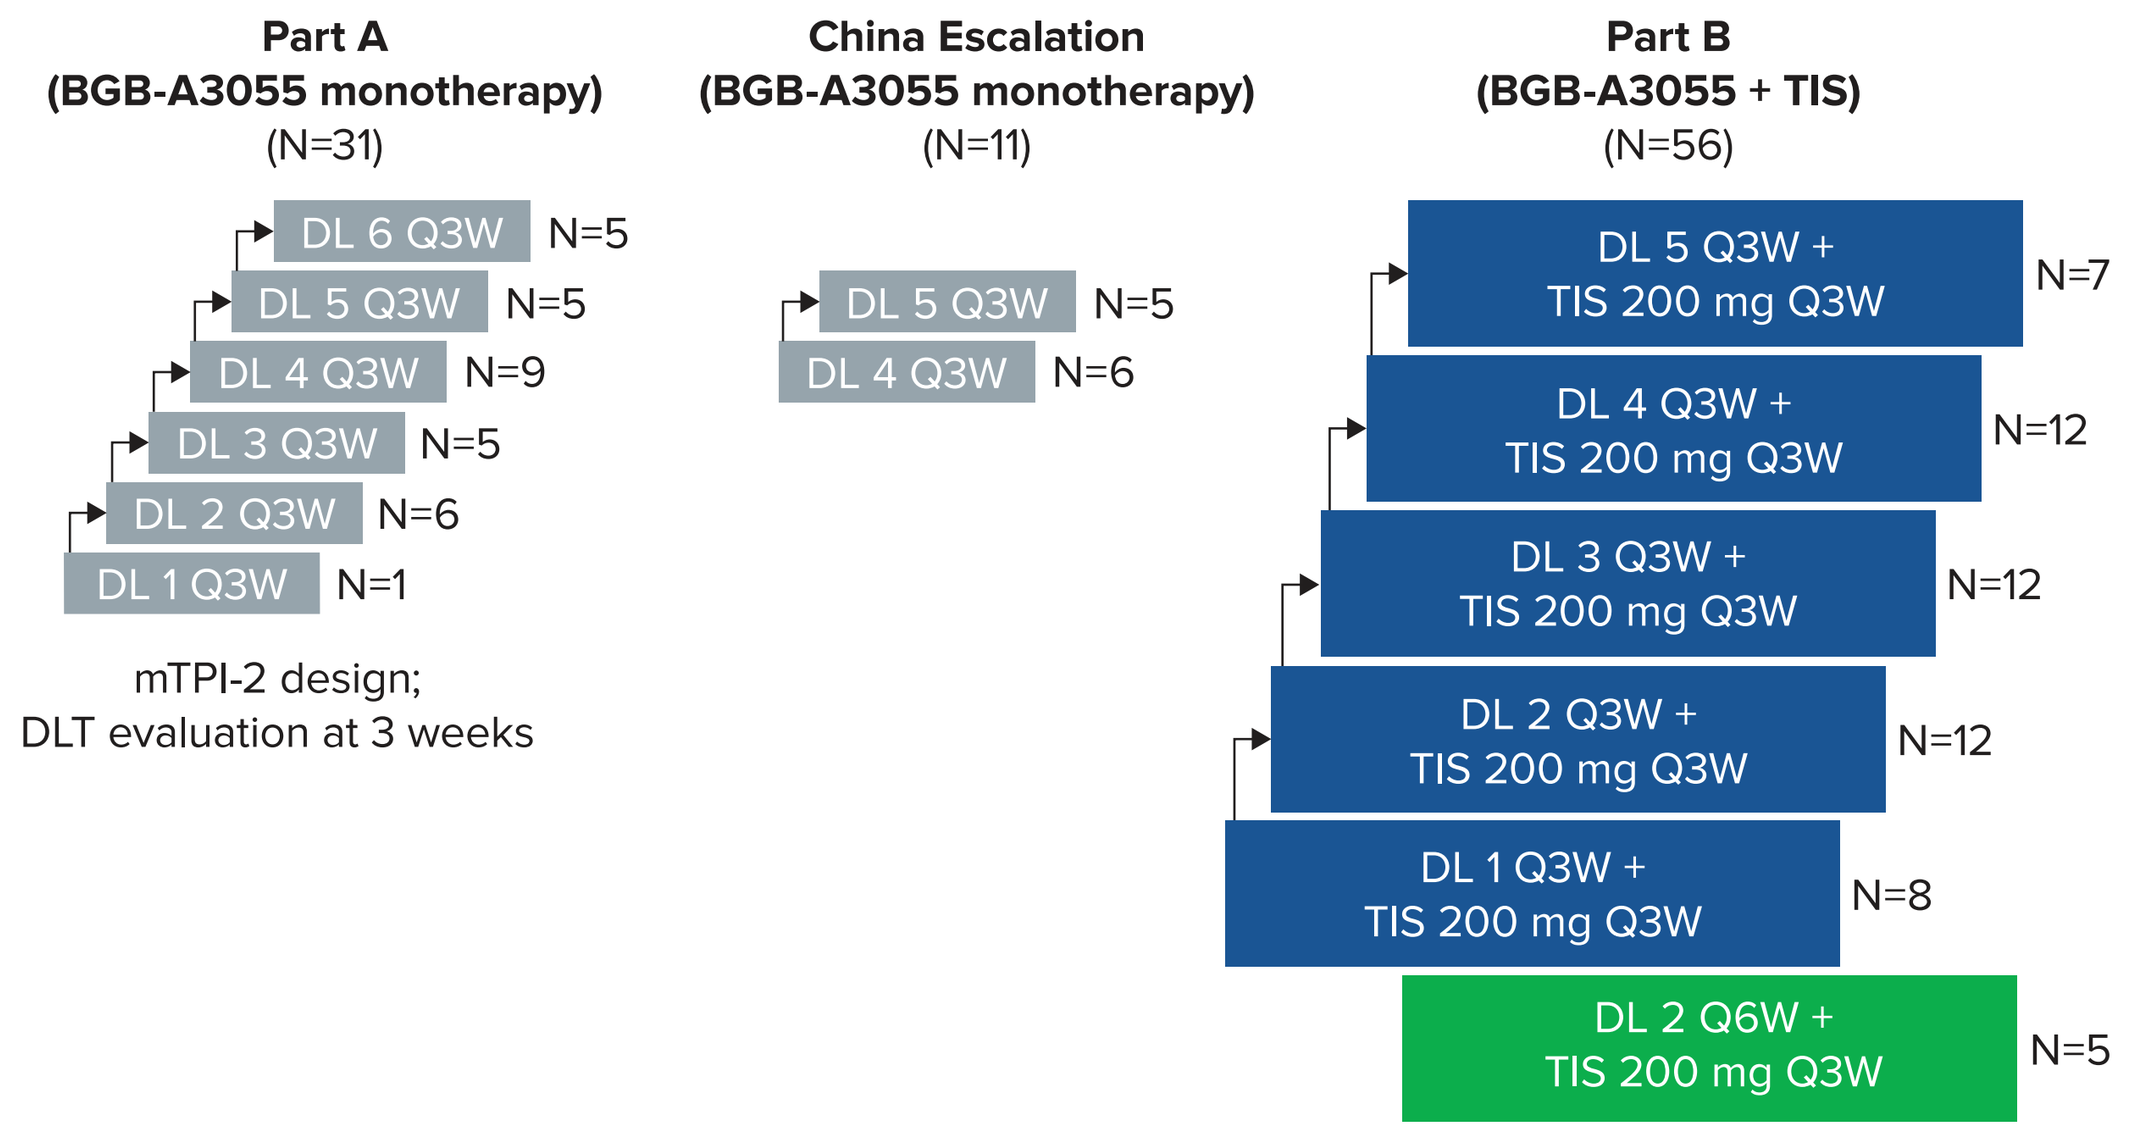
Part A
(BGB-A3055 monotherapy)
(N=31)
DL 6 Q3W N=5
DL 5 Q3W N=5
DL 4 Q3W N=9
DL 3 Q3W N=5
DL 2 Q3W N=6
DL 1 Q3W N=1
mTPI-2 design;
DLT evaluation at 3 weeks
China Escalation
(BGB-A3055 monotherapy)
(N=11)
DL 5 Q3W N=5
DL 4 Q3W N=6
Part B
(BGB-A3055 + TIS)
(N=56)
DL 5 Q3W + TIS 200 mg Q3W N=7
DL 4 Q3W + TIS 200 mg Q3W N=12
DL 3 Q3W + TIS 200 mg Q3W N=12
DL 2 Q3W + TIS 200 mg Q3W N=12
DL 1 Q3W + TIS 200 mg Q3W N=8
DL 2 Q6W + TIS 200 mg Q3W N=5

Endpoints
Primary: Safety and tolerability; MTD or MAD and RDFE
Secondary: ORR, TTR, DoR, DCR, and CBR; PKs; host immunogenicity to BGB-A3055; pharmacodynamics

The mTPI-2 method was used to guide dose escalation with a target toxicity rate for the MTD of Φ=0.30.
Abbreviations: CBR, clinical benefit rate; DCR, disease control rate; DL, dose level; DLT, dose-limiting toxicity; ECOG PS, Eastern Cooperative Oncology Group Performance Status; MAD, maximum administered dose; MTD, maximum tolerated dose; mTPI-2, modified toxicity probability interval-2; ORR, overall response rate; PK, pharmacokinetic; Q3W, every 3 weeks; RDFE, recommended dose for expansion; RECIST, Response Evaluation Criteria in Solid Tumors; TIS, tislelizumab; TTR, time to response.

Analysis and Statistical Methods

- The safety analysis set included all patients who received ≥1 dose of study drug(s)
- The efficacy-evaluable analysis set included all patients who received ≥1 dose of study drug(s) and had radiologically evaluable disease at baseline, and ≥1 evaluable radiological tumor response assessment post-baseline unless any clinical progressive disease (PD) or death occurred before the first post-baseline tumor assessment
- Safety was assessed by the type, frequency, and severity (as graded by the National Cancer Institute Common Terminology Criteria for Adverse Events [NCI-CTCAE] v5.0) of adverse events (AEs) and American Society for Transplantation and Cellular Therapy (ASTCT) consensus grading system for cytokine release syndrome
- Data from phase 1a were summarized by DL and total

Biomarker Methods

- The CCR8 receptor occupancy on Treg and Treg change in whole blood post-treatment was assessed by flow cytometry using peripheral blood samples collected at cycle 1 day 1 (C1D1), C1D2, C1D8, C2D1, C3D1, and C5D1
- Tumor Treg change post-treatment was assessed by multiplex immunohistochemistry in pre- and post-treatment (C2D8 or later time point) biopsies

RESULTS

Baseline Characteristics and Patient Disposition

- As of the data cutoff date of January 27, 2026, 98 patients were treated (42 in Part A and 56 in Part B)
- Median (range) study follow-up was 3.8 (0.9-21.2) months in Part A and 4.7 (0.5-17.0) months in Part B
- Patients were treated at DLs 1-6 in Part A, and DLs 1-5 in Part B
- Baseline characteristics are shown in **Table 1**

Table 1. Baseline Characteristics (Safety Analysis Set)

	Part A BGB-A3055 monotherapy (N=42)	Part B BGB-A3055 + TIS (N=56)
Median (range) age, years	64.5 (39.0-79.0)	59.0 (33.0-81.0)
Sex, n (%)		
Male	26 (61.9)	26 (46.4)
Female	16 (38.1)	30 (53.6)
Race,[a] n (%)		
Asian	13 (31.0)	2 (3.6)
White	16 (38.1)	24 (42.9)
Not reported	5 (11.9)	5 (8.9)
Unknown	3 (7.1)	1 (1.8)
Other	0 (0)	1 (1.8)
ECOG PS, n (%)		
0	9 (21.4)	21 (37.5)
1	33 (78.6)	35 (62.5)
Type of cancer at initial diagnosis, n (%)		
Colorectal cancer	11 (26.2)	7 (12.5)
Non-small cell lung cancer	7 (16.7)	5 (8.9)
Head and neck cancer	6 (14.3)	6 (10.7)
Breast cancer	4 (9.5)	4 (7.1)
Pancreatic cancer	3 (7.1)	11 (19.6)
Gastric cancer	2 (4.8)	0 (0)
Kidney cancer	2 (4.8)	1 (1.8)
Duodenal adenocarcinoma	1 (2.4)	0 (0)
Esophageal cancer	1 (2.4)	1 (1.8)
Gastric or gastroesophageal junction cancer	1 (2.4)	0 (0)
Hepatocellular carcinoma	1 (2.4)	0 (0)
Ovarian cancer	1 (2.4)	13 (23.2)
Small cell lung cancer	1 (2.4)	0 (0)
Urothelial cancer	1 (2.4)	1 (1.8)
Cervical cancer	0 (0)	5 (8.9)
Nasopharyngeal cancer	0 (0)	2 (3.6)

[a]Race is not listed for 5 patients in Part A and 23 patients in Part B from France, as France does not allow the collection of racial demographics.

Safety and Tolerability

- BGB-A3055 as monotherapy or combined with tislelizumab had a safety profile consistent with selective CCR8 on-target effects (**Table 2**)
 - The most common serious TEAE was immune-mediated enterocolitis (4.8% [2/42] in Part A and 7.1% [4/56] in Part B)
 - No treatment emergent AEs (TEAEs) leading to discontinuation occurred in more than one patient in Part A; immune-mediated enterocolitis, diarrhea, and colitis occurred as TEAEs leading to discontinuation in more than one patient in Part B
 - The most common treatment-related TEAEs were neutrophil count decreased (21.4% [9/42]) in Part A and pyrexia (21.4% [12/56]) in Part B (**Table 3**)
 - The most common immune-mediated AEs (imAEs) were rash, rash maculo-papular, and hypothyroidism in Part A (7.1% [3/43] each) and rash maculo-papular in Part B (17.9% [10/56])
- DLTs occurred in two patients in Part A (immune-mediated hepatitis in one patient and febrile neutropenia and platelet count decreased in the other patient) and in four patients in Part B (colitis, drug-induced liver injury, nephrotic syndrome, and rash maculo-papular in a single patient each)
- MAD was DL 6 for Part A and DL 5 for Part B; MTD was not reached for both Part A and B

Table 2. Overall Safety Summary (Safety Analysis Set)

	Part A BGB-A3055 monotherapy (N=42)	Part B BGB-A3055 + TIS (N=56)
Any TEAE, n (%)	41 (97.6)	55 (98.2)
Grade ≥3	26 (61.9)	47 (83.9)
Serious	17 (40.5)	37 (66.1)
Leading to death	1 (2.4)	3 (5.4)
Leading to treatment discontinuation	9 (21.4)	23 (41.1)
Treatment-related TEAE, n (%)	35 (83.3)	54 (96.4)
Grade ≥3	17 (40.5)	31 (55.4)
Serious	5 (11.9)	20 (35.7)
Leading to death	0 (0.0)	0 (0.0)
Leading to treatment discontinuation	5 (11.9)	19 (33.9)
Any imAE, n (%)	15 (35.7)	33 (58.9)

AEs were graded for severity using CTCAE v5.0. Treatment-related TEAEs include those events considered by the investigator to be related or with missing assessment of the causal relationship.

Table 3. Treatment-related TEAEs in ≥10% of Patients in Arms A or B (Safety Analysis Set)

	Part A BGB-A3055 monotherapy (N=42)	Part B BGB-A3055 + TIS (N=56)
Any treatment-related TEAE, n (%)	35 (83.3)	54 (96.4)
Pyrexia	5 (11.9)	12 (21.4)
Diarrhea	6 (14.3)	11 (19.6)
Nausea	6 (14.3)	11 (19.6)
Chills	7 (16.7)	8 (14.3)
Fatigue	5 (11.9)	10 (17.9)
Asthenia	6 (14.3)	8 (14.3)
Decreased appetite	7 (16.7)	5 (8.9)
Rash	3 (7.1)	9 (16.1)
Neutrophil count decreased	9 (21.4)	2 (3.6)
Rash maculo-papular	3 (7.1)	8 (14.3)
Anemia	6 (14.3)	4 (7.1)
Vomiting	3 (7.1)	7 (12.5)
Pruritus	2 (4.8)	8 (14.3)
ALT increased	2 (4.8)	8 (14.3)
AST increased	2 (4.8)	7 (12.5)
Hypoalbuminemia	5 (11.9)	3 (5.4)
Gamma-glutamyltransferase increased	5 (11.9)	2 (3.6)
White blood cell count decreased	5 (11.9)	0 (0)

AEs were classified based on MedDRA v28.0. Patients with multiple events for a given preferred term were counted once at the preferred term level. Events were sorted by overall decreasing frequency.
Abbreviations: ALT, alanine aminotransferase; AST, aspartate aminotransferase.

Antitumor Activity

- ORR (95% confidence interval [CI]) was 5.0% (0.6-16.9, confirmed) and 7.5% (1.6-20.4, unconfirmed) in Part A, and 16.4% (7.8-28.8, confirmed) and 18.2% (9.1-30.9, unconfirmed) in Part B. Confirmed responses are listed in **Table 4**
 - There were two patients with confirmed partial responses (PRs) in Part A; both received prior immunotherapies, and one had a DoR of 421 days with BGB-A3055 monotherapy
 - There was one patient with a confirmed complete response (CR) and nine PRs (eight with confirmed response) in Part B, of which four received prior immunotherapies. One of them had a DoR of 385 days
- There were two patients in Part A and two in Part B that had durable stable disease (SD; ≥6 months)
- Best percent change from baseline in target lesion sum of diameters is presented in **Figure 2**

Table 4. Efficacy Data (Efficacy Evaluable Analysis Set)

	Part A BGB-A3055 monotherapy (N=40)	Part B BGB-A3055 + TIS (N=55)
ORR, n (% [95% CI[a]])	2 (5.0 [0.6-16.9])	9 (16.4 [7.8-28.8])
BOR, n (%)		
CR	0 (0)	1 (1.8)
PR	2 (5.0)	8 (14.5)
SD	12 (30.0)	22 (40.0)
PD	25 (62.5)	16 (29.1)
NE	1 (2.5)	8 (14.5)
DCR, n (% [95% CI[a]])	14 (35.0 [20.6-51.7])	31 (56.4 [42.3-69.7])
CBR, n (% [95% CI[a]])	4 (10.0 [2.8-23.7])	11 (20.0 [10.4-33.0])

[a]95% CI was estimated using the Clopper–Pearson method.
Abbreviations: BOR, best overall response; NE, not evaluable.

Figure 2. Best Percent Change from Baseline in Target Lesion Sum of Diameters for Part A (A) and Part B (B) (Efficacy Evaluable Analysis Set)

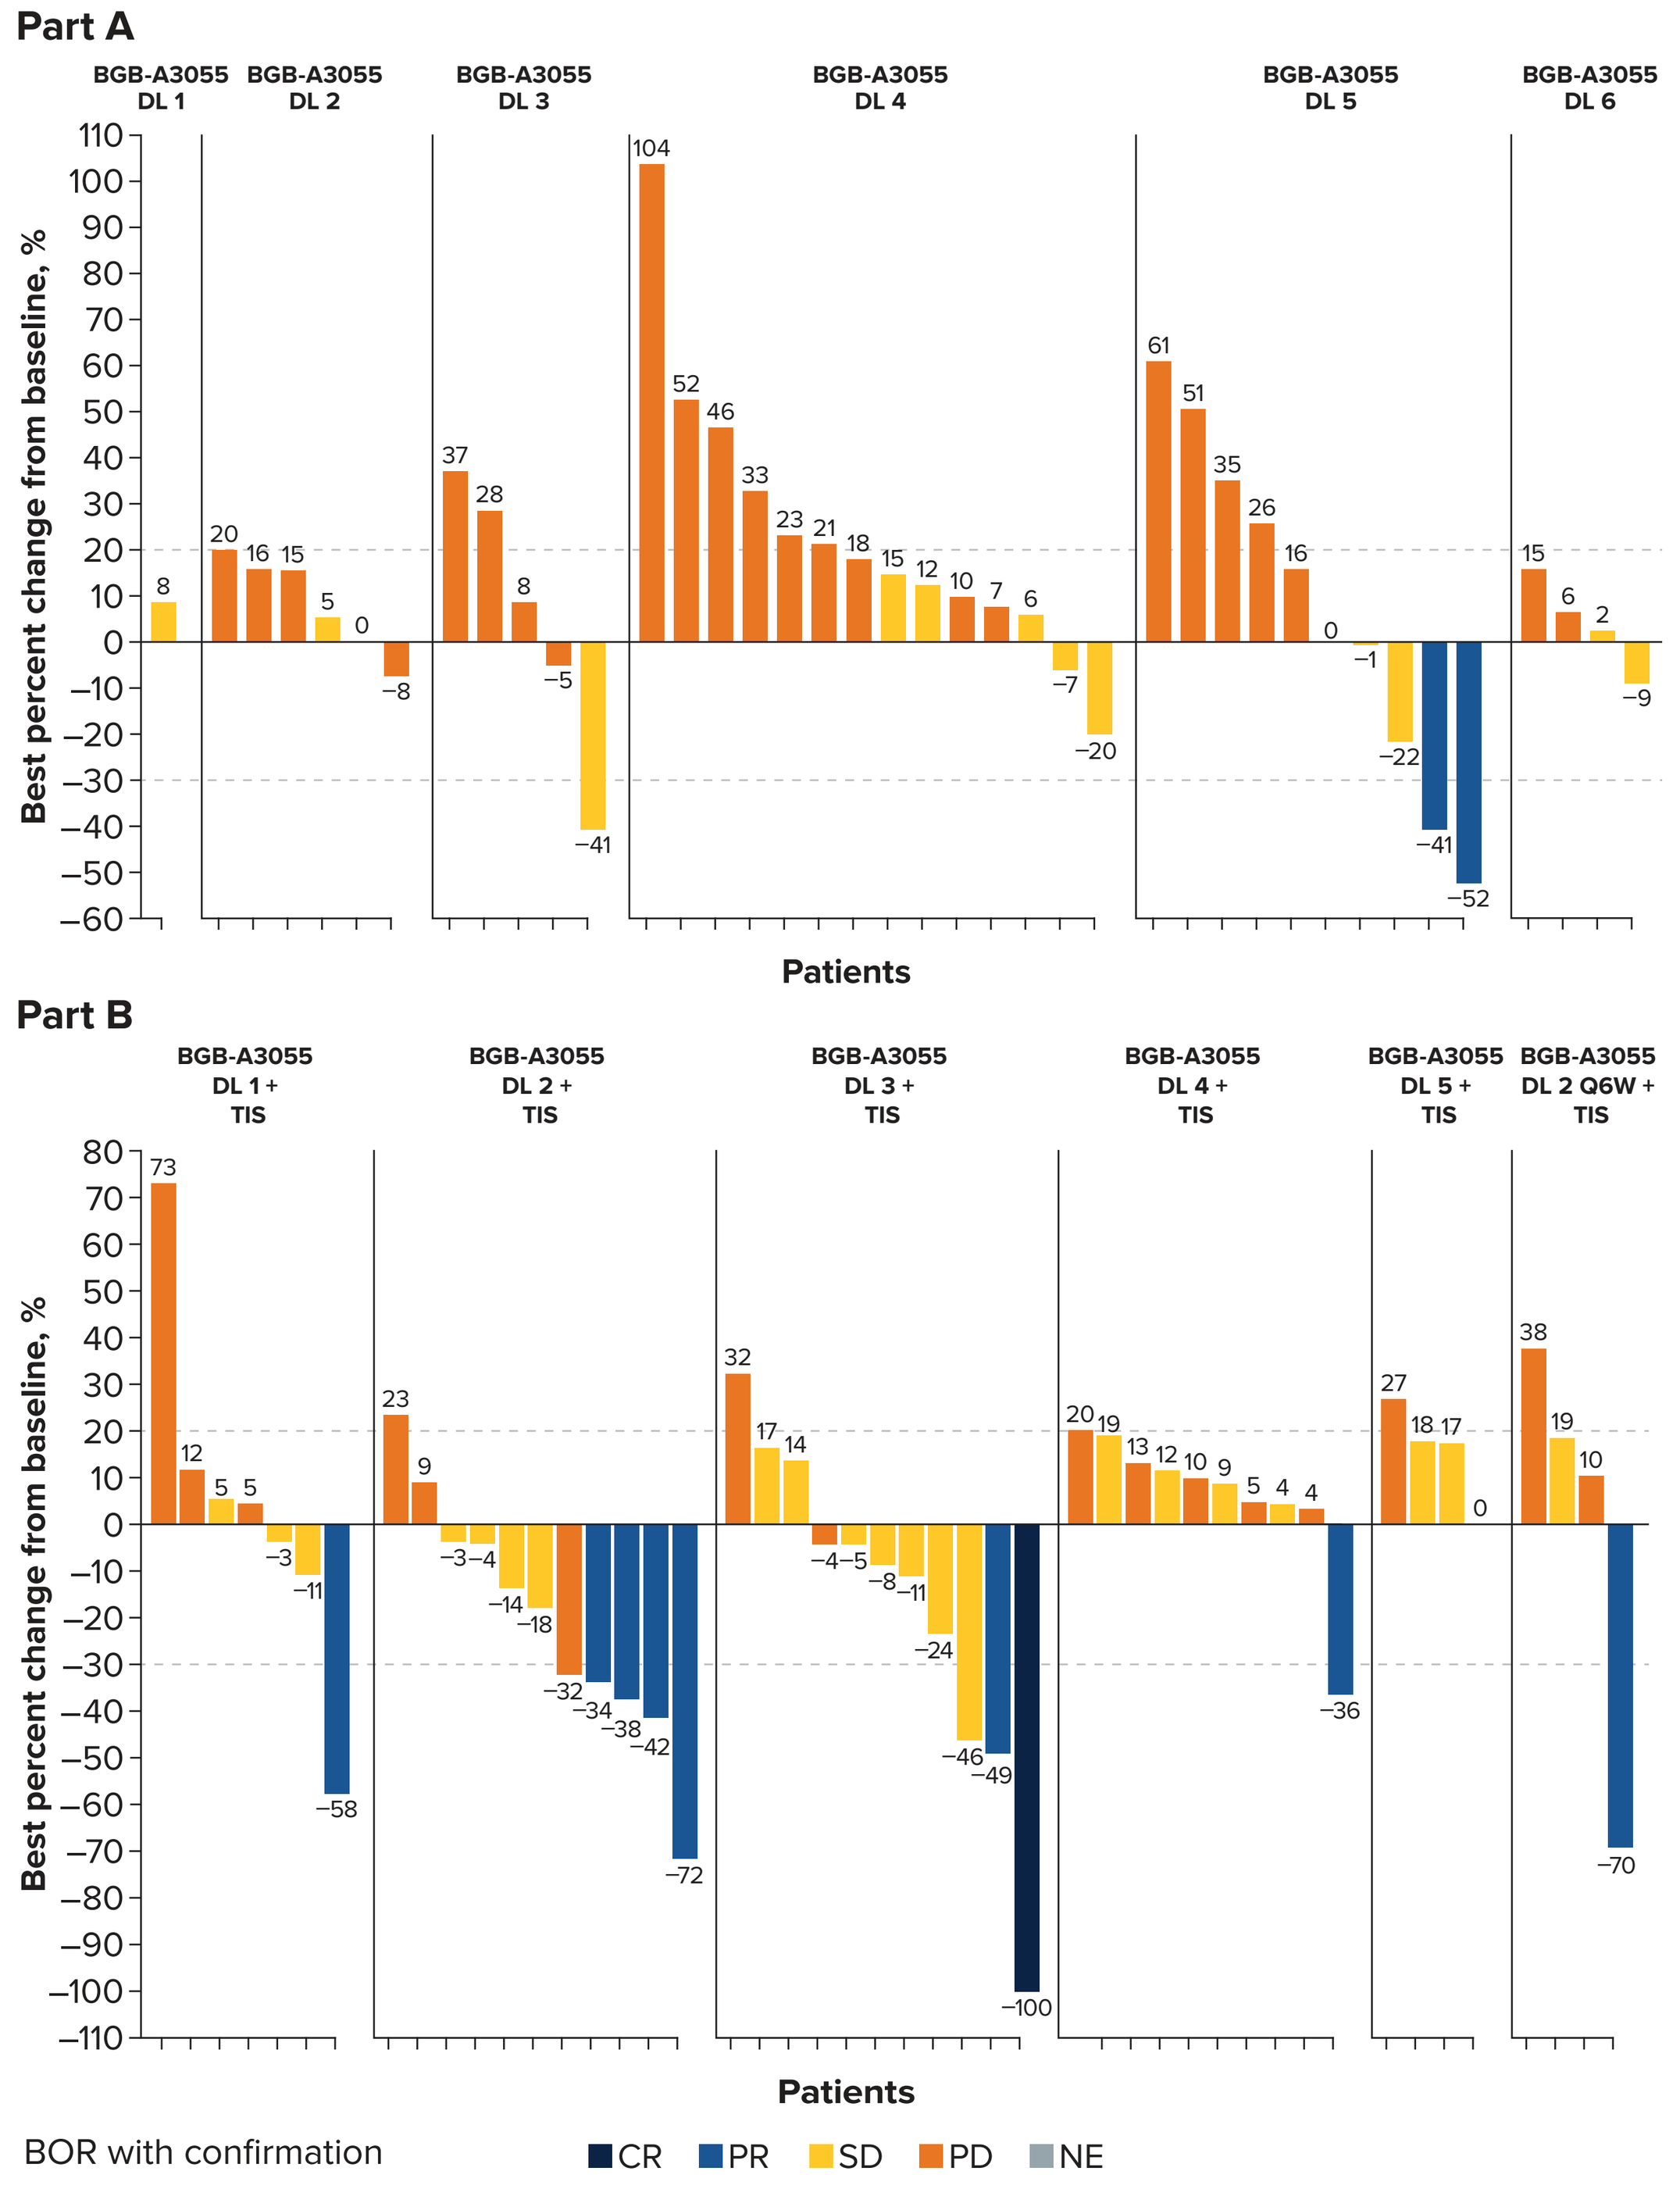
Part A
BGB-A3055 DL 1
BGB-A3055 DL 2
BGB-A3055 DL 3
BGB-A3055 DL 4
BGB-A3055 DL 5
BGB-A3055 DL 6
Best percent change from baseline, %
8
20 16 15 5 0 −8
37 28 8 −5 −41
104 52 46 33 23 21 18 15 12 10 7 6 −7 −20
61 51 35 26 16 0 −1 −22 −41 −52
15 6 2 −9
Patients
Part B
BGB-A3055 DL 1 + TIS
BGB-A3055 DL 2 + TIS
BGB-A3055 DL 3 + TIS
BGB-A3055 DL 4 + TIS
BGB-A3055 DL 5 + TIS
BGB-A3055 DL 2 Q6W + TIS
Best percent change from baseline, %
73 12 5 5 −3 −11 −58
23 9 −3 −4 −14 −18 −32 −34 −38 −42 −72
32 17 14 −4 −5 −8 −11 −24 −46 −49 −100
20 19 13 12 10 9 5 4 4 −36
27 18 17 0
38 19 10 −70
Patients
BOR with confirmation
CR PR SD PD NE

Biomarkers

- A decrease in peripheral Treg levels and complete CCR8 receptor occupancy on cluster of differentiation 4 (CD4+) Tregs after treatment with BGB-A3055 was achieved across all DLs (**Figure 3A/B**)
- Decrease in tumor Tregs upon BGB-A3055 dosing were observed in patients in both Parts A and B, supporting on-target pharmacodynamic activity in agreement with the mechanism of action (**Figure 3C**)

Figure 3. Treg Reduction Was Observed in Peripheral Blood and Tumor Tissue Post-BGB-A3055 Treatment

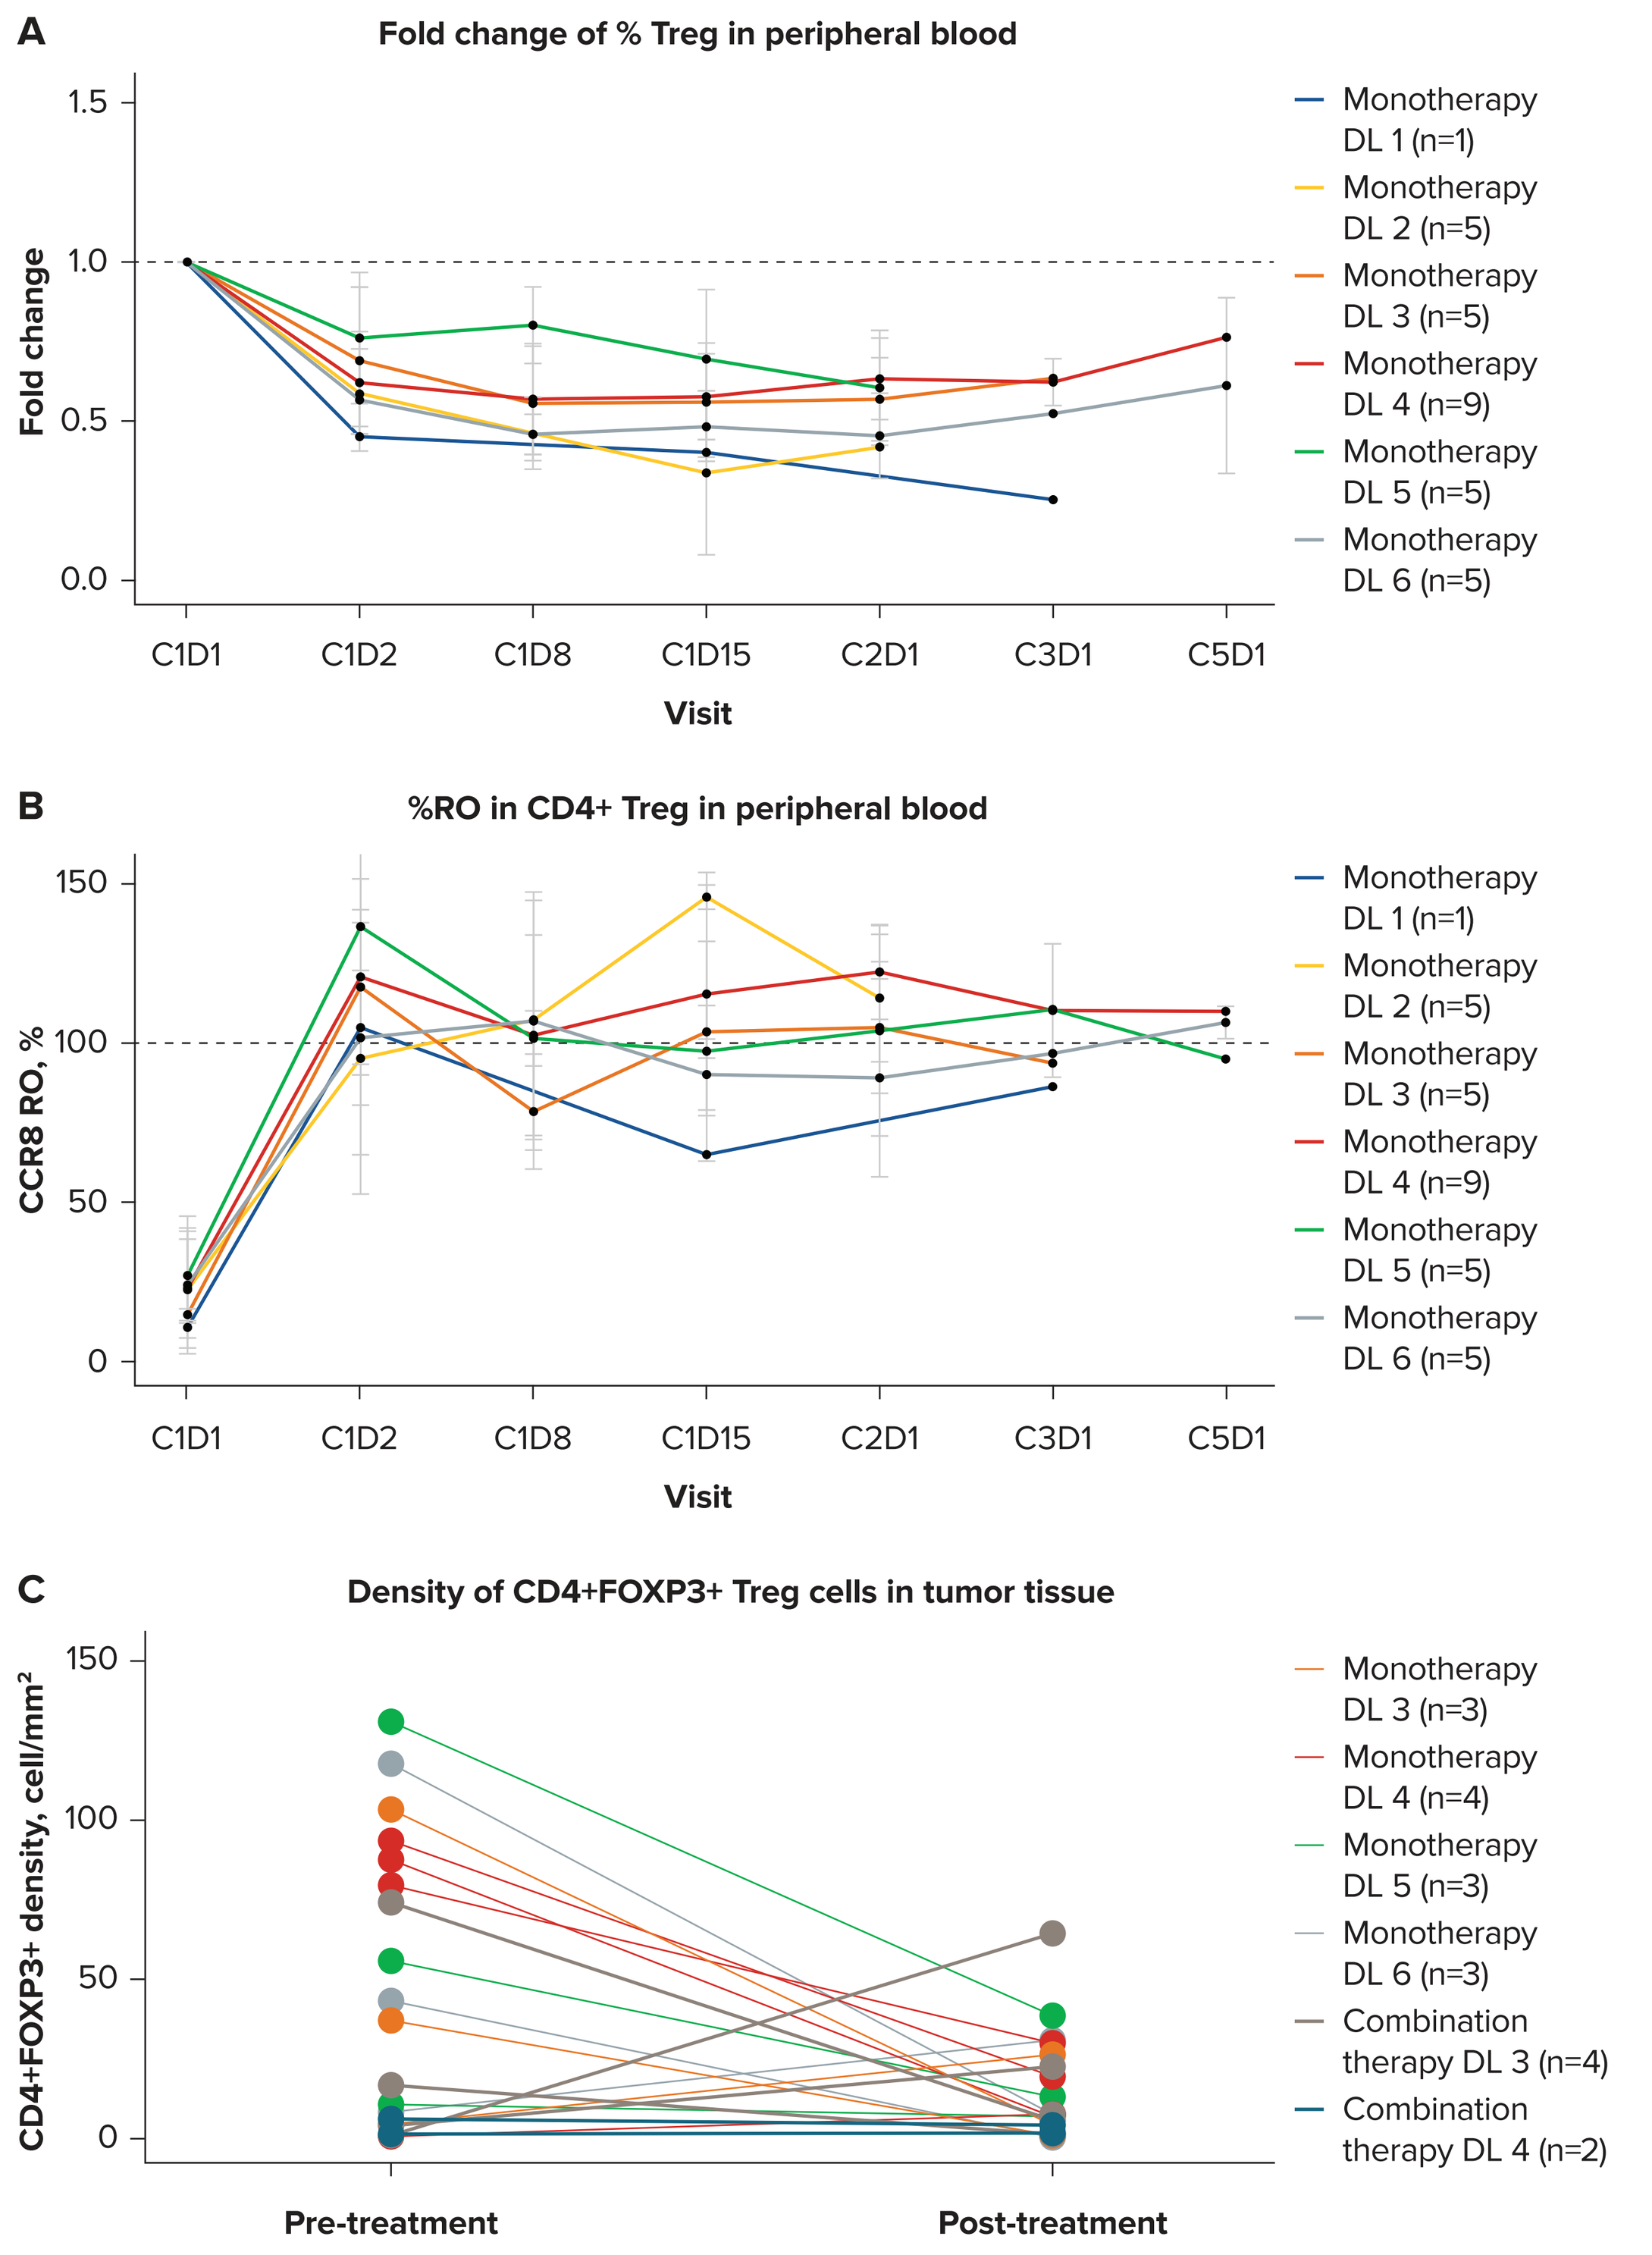
A
Fold change of % Treg in peripheral blood
Fold change
Visit
C1D1 C1D2 C1D8 C1D15 C2D1 C3D1 C5D1
Monotherapy DL 1 (n=1)
Monotherapy DL 2 (n=5)
Monotherapy DL 3 (n=5)
Monotherapy DL 4 (n=9)
Monotherapy DL 5 (n=5)
Monotherapy DL 6 (n=5)
B
%RO in CD4+ Treg in peripheral blood
CCR8 RO, %
Visit
C1D1 C1D2 C1D8 C1D15 C2D1 C3D1 C5D1
Monotherapy DL 1 (n=1)
Monotherapy DL 2 (n=5)
Monotherapy DL 3 (n=5)
Monotherapy DL 4 (n=9)
Monotherapy DL 5 (n=5)
Monotherapy DL 6 (n=5)
C
Density of CD4+FOXP3+ Treg cells in tumor tissue
CD4+FOXP3+ density, cell/mm²
Pre-treatment
Post-treatment
Monotherapy DL 3 (n=3)
Monotherapy DL 4 (n=4)
Monotherapy DL 5 (n=3)
Monotherapy DL 6 (n=3)
Combination therapy DL 3 (n=4)
Combination therapy DL 4 (n=2)

Abbreviations: FOXP3, forkhead box P3; RO, receptor occupancy.

REFERENCES

1. Schaerli P, et al. *J Exp Med*. 2004;199:1265-1275.
2. Soler D, et al. *J Immunol*. 2006;177:6940-6951.
3. Annunziato F, et al. *J Exp Med*. 2002;196:379-387.
4. Lee JH, et al. *J Immunol*. 2007;178:301-311.
5. Plitas G, et al. *Immunity*. 2016;45:1122-1134.
6. Wang T, et al. *Cancer Immunol Immunother*. 2020;69:1855-1867
7. Van Damme H, et al. *J Immunother Cancer*. 2021;9:e001749.
8. Alvisi G, et al. *J Clin Invest*. 2020;130:3137-3150.
9. Wang X, et al. *Cancer Res*. 2025;85(8_Suppl_1):4787.

DISCLOSURES

JR: Travel, accommodation, or expenses: Daiichi Sankyo, Janssen, Pierre Fabre, Pfizer, and Roche.

ACKNOWLEDGMENTS

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This study was sponsored by BeOne Medicines, Ltd. Medical writing was provided by Lee Blackburn, MSc of Amiculum, and supported by BeOne Medicines.

CORRESPONDENCE: Judith Raimbourg, judith.raimbourg@ico.unicancer.fr

Presented at ASCO Annual Meeting; May 29-June 2, 2026; Chicago, IL, USA