Alternative Dosing Regimens of Tislelizumab Proposed Using Modeling and Simulation

Ahsan Rizwan,[1*] Tian Yu,[1†] Yuying Gao,[2] Kun Wang,[2] Fengyan Xu,[2] Ya Wan,[3] Jun Wang,[4] Srikumar Sahasranaman,[1†] Marcia Campbell,[5] Patrick Schnell,[6] Ramil Abdrashitov,[7] William D. Hanley,[1] Nageshwar Budha[1]

[1]Clinical Pharmacology and Pharmacometrics, BeiGene USA, Inc., San Mateo, CA, USA; [2]Pharmacometrics, Shanghai Qiangshi Information Technology Co., Ltd., Shanghai, China; [3]Scientific Programming, BeiGene (Beijing) Co., Ltd., Beijing, China; [4]Global Statistics and Data Sciences, BeiGene (Beijing), Co., Ltd., Beijing, China; [5]Regulatory Affairs, BeiGene (Canada) ULC, Toronto, Ontario, Canada; [6]Product Safety, BeiGene USA, Inc., Ridgefield Park, NJ, USA; [7]Clinical Development, BeiGene USA, Inc., Fulton, MD, USA. *Lead and presenting author. †Affiliation at the time the analysis was conducted.

Poster No: 394 presented at ASCO GI, San Francisco, CA, January 23-25, 2025

Conclusions

The clinical pharmacology characteristics of tislelizumab (BGB-A317), pharmacometrics-based analyses, and clinical evidence presented here collectively form the basis of proposing alternative doses of tislelizumab.

Alternative dosing regimens of tislelizumab at 150 mg every 2 weeks (Q2W), 300 mg Q4W, and 400 mg Q6W are expected to result in similar safety and efficacy profiles as the 200 mg Q3W reference dosing regimen and may be used interchangeably for indications where 200 mg Q3W is approved.

The alternative dosing regimens of tislelizumab have the potential to offer patients flexible treatment options that are compatible with background chemotherapy and reduce the number of infusion visits while maintaining the same therapeutic benefit.

Background

- Tislelizumab, a humanized immunoglobulin G4 monoclonal anti-programmed cell death protein-1 (PD-1) antibody which binds to PD-1 with high affinity and specificity,[1] is approved for the treatment of multiple solid tumors globally, either as a monotherapy and/or in combination with other therapies at 200 mg Q3W
- Alternative dosing regimens of tislelizumab have the potential to provide flexible treatment options that are compatible with background chemotherapy and to reduce the number of infusion visits for patients
- In clinical trials, tislelizumab did not have any dose-limiting toxicities up to 10 mg/kg Q2W[2-4]
- Previous model-based analyses have demonstrated that the pharmacokinetic (PK) profile of tislelizumab is similar across ethnicities and tumor types, and that tislelizumab has a relatively flat exposure–response relationship across a broad range of exposures[2,3]
- Therefore, we evaluated alternative dosing regimens of tislelizumab at 150 mg Q2W, 300 mg Q4W, and 400 mg Q6W using a model-based approach, with the aim of alleviating patient burden by providing longer dosing intervals and/or treatment flexibility compatible with background chemotherapy to meet the needs of patients and healthcare practitioners

Methods

- A previously developed population PK model[2,3] was used for simulating PK exposure of the proposed alternative intravenous tislelizumab dosing regimens (150 mg Q2W, 300 mg Q4W, and 400 mg Q6W), which were selected by exposure-matching to the reference dose of 200 mg Q3W
- PK-based criteria (peak serum concentration [C_{max}] within 25% and average serum concentration [$C_{average}$] and trough serum concentration [C_{trough}] within 20% of the reference dose) based on Food and Drug Administration guidance[5] were used
- Alternative dosing regimen exposures in the first least-common time interval and at steady state were compared with the reference dose of 200 mg Q3W
- Deviations from PK-based criteria were bridged using appropriate safety and efficacy references and exposure–response analyses using a pool of 4 phase 1, 2, and 3 clinical trials of tislelizumab in patients with solid tumors, including gastric cancer and esophageal squamous cell carcinoma
- Objective response rate (ORR) was the efficacy endpoint in the exposure–response efficacy analysis; the model-predicted $C_{average}$ of the first dose ($C_{avg,dose1}$) was used as the primary exposure endpoint in the exposure–response efficacy analysis, while the model-predicted C_{max} at steady state ($C_{max,ss}$) was used as the primary exposure endpoint in the exposure–response safety analysis

Results

External Model Validation and Selection of Doses for the Alternative Dosing Regimens

- Simulated PK exposures of the 300 mg Q4W and 400 mg Q6W tislelizumab regimens were consistent with observed data
- The $C_{max,ss}$ for the 150 mg Q2W dosing regimen overlapped, and C_{trough} was maintained above that of the 200 mg Q3W reference (**Figure 1A**)
- The C_{max} for the 300 mg Q4W and 400 mg Q6W dosing regimens were greater than the 200 mg Q3W reference; however, they were lower than the 5 mg/kg Q3W dose of the tislelizumab safety reference, tested clinically in approximately 355 patients (phase 1A and phase 1B of BGB-A317-001) (**Figure 1B and 1C**)
- Although the C_{trough} of the longer dosing interval 400 mg Q6W regimen trended slightly lower than the 200 mg Q3W reference (not shown), it was maintained above the 2 mg/kg Q3W tislelizumab efficacy reference (**Figure 1D**)

Figure 1. Steady State PK Profiles for the (A) 150 mg Q2W, (B) 300 mg Q4W, and (C, D) 400 mg Q6W Alternative Tislelizumab Regimens vs the 200 mg Q3W Reference Regimen and Safety/Efficacy References

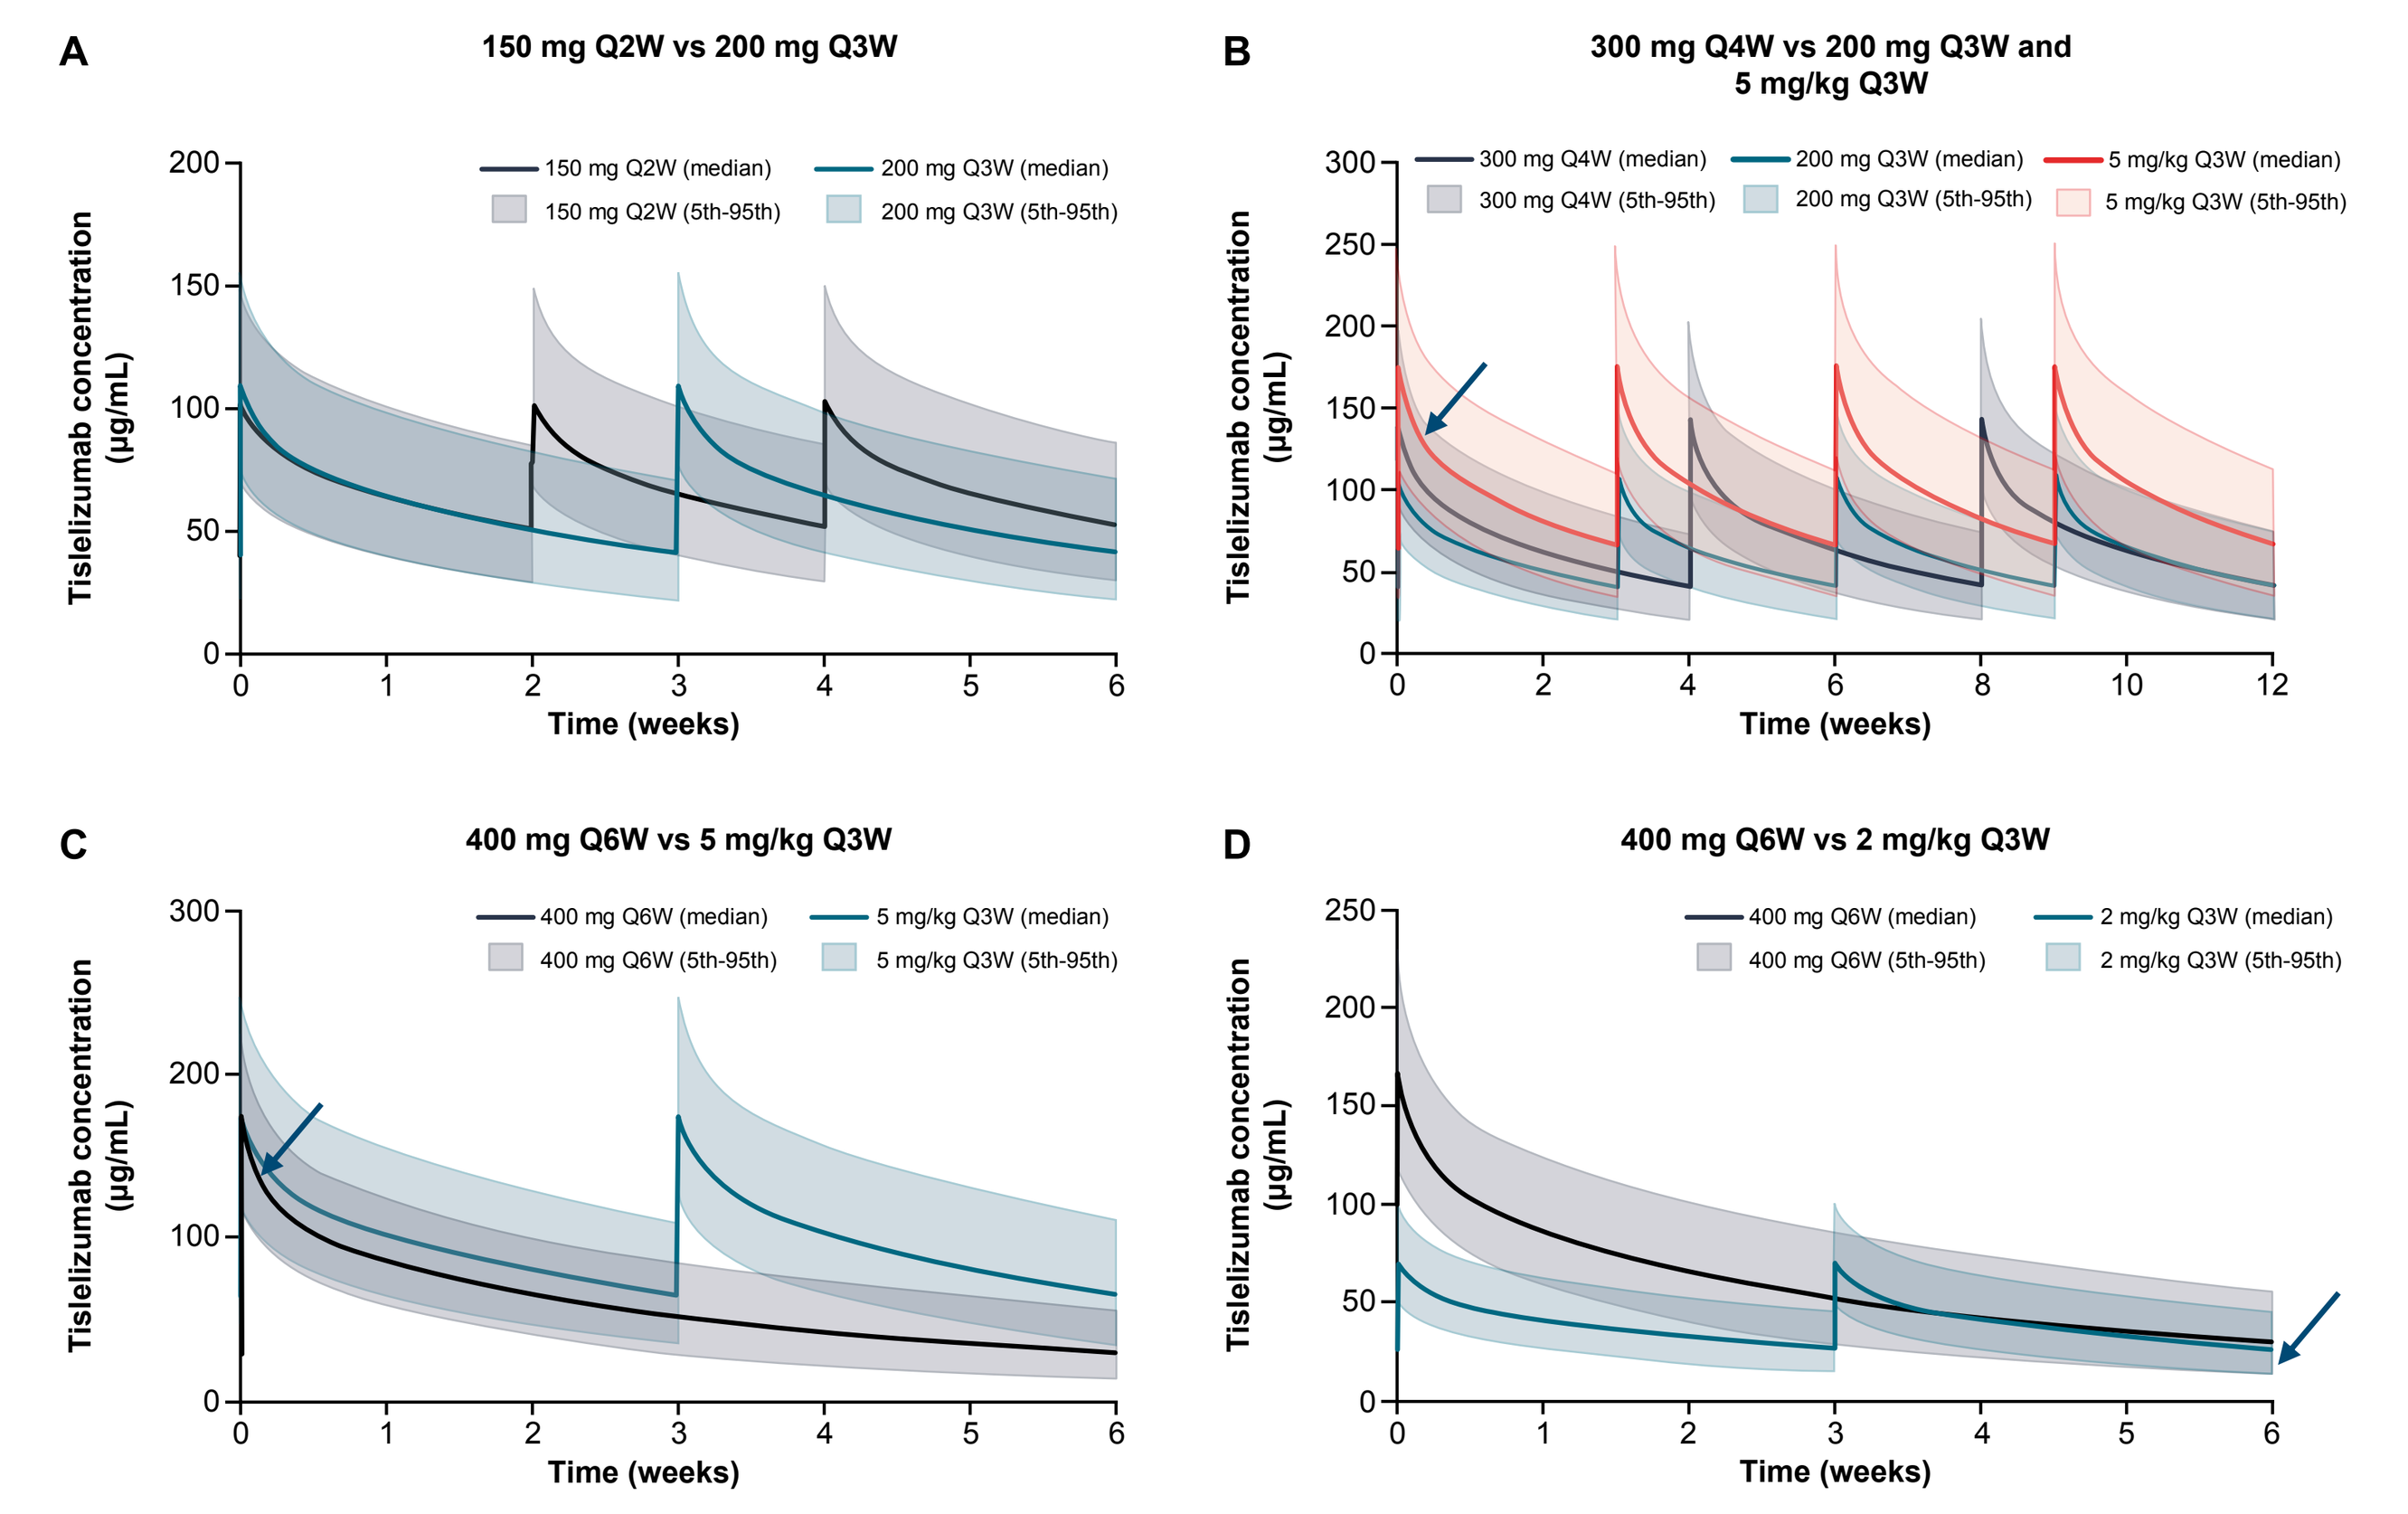
A
150 mg Q2W vs 200 mg Q3W
150 mg Q2W (median)
150 mg Q2W (5th-95th)
200 mg Q3W (median)
200 mg Q3W (5th-95th)
B
300 mg Q4W vs 200 mg Q3W and 5 mg/kg Q3W
300 mg Q4W (median)
300 mg Q4W (5th-95th)
200 mg Q3W (median)
200 mg Q3W (5th-95th)
5 mg/kg Q3W (median)
5 mg/kg Q3W (5th-95th)
C
400 mg Q6W vs 5 mg/kg Q3W
400 mg Q6W (median)
400 mg Q6W (5th-95th)
5 mg/kg Q3W (median)
5 mg/kg Q3W (5th-95th)
D
400 mg Q6W vs 2 mg/kg Q3W
400 mg Q6W (median)
400 mg Q6W (5th-95th)
2 mg/kg Q3W (median)
2 mg/kg Q3W (5th-95th)
Tislelizumab concentration (μg/mL)
Time (weeks)

The solid line represents the median and the shaded area represents the 90% prediction interval of steady-state pharmacokinetic exposures. Blue arrows indicate where clinical references were used for bridging the alternative doses. The Y-axes show simulated tislelizumab PK concentrations, and X-axes show the least common dose interval to the 200 mg Q3W reference regimen (6 weeks for the 150 mg Q2W and 400 mg Q6W regimens and 12 weeks for the 300 mg Q4W regimen).
Abbreviation: Q#W, every # weeks.

Simulation of Exposures for Alternative Dosing Regimens

150 mg Q2W

- All PK-based criteria were met for the tislelizumab 150 mg Q2W regimen. The $C_{average}$ and C_{trough} were not lower than those of the 200 mg Q3W regimen and the $C_{max,ss}$ was not higher than that of the 200 mg Q3W regimen (**Table 1**)

300 mg Q4W

- $C_{average}$ and C_{trough} met the prespecified PK-based criteria. Although C_{max} was 6.4% higher than the recommended limit of 25%, it was 18.7% lower when compared with the safety reference dosing regimen of 5 mg/kg Q3W (**Table 1**)

400 mg Q6W

- $C_{average}$ met the prespecified PK-based criteria. However, C_{trough} was 28.4% lower and $C_{max,ss}$ was 52.2% higher than the 200 mg Q3W regimen and did not meet the PK-based criteria of 20% and 25%, respectively (**Table 1**)
- Results demonstrate that the 400 mg Q6W dosing regimen would maintain $C_{trough,ss}$ levels 10.7% higher than the 2 mg/kg Q3W efficacy reference dosing regimen, and the predicted $C_{max,ss}$ was below that of the 5 mg/kg Q3W safety reference (25.1% lower). Therefore, the higher $C_{max,ss}$ for the 400 mg Q6W regimen is unlikely to be associated with an unacceptable clinical safety profile

Exposure–Response Analysis for Efficacy

- Median $C_{avg,dose1}$ values were similar between responders and nonresponders in the population pool of solid tumor types, which included esophageal squamous cell carcinoma, non-small cell lung cancer, and gastric cancer, among others (**Figure 2A**)
- Logistic regression modeling showed no significant association between the ORR and $C_{avg,dose1}$ (P=0.118) in the predicted exposure range of the 150 mg Q2W, 300 mg Q4W, and 400 mg Q6W regimens (**Figure 2B**)

Exposure–Response Analysis for Safety

- There were no apparent relationships between tislelizumab exposure and any of the safety endpoints evaluated. The incidence of treatment-emergent adverse events was comparable across studies (**Table 2**)
- Overall, the safety profiles were comparable and consistent between patients with a predicted $C_{max,ss}$ greater than the predicted geometric mean C_{max} for the 150 mg Q2W, 300 mg Q4W, and 400 mg Q6W dosing regimens and patients with a $C_{max,ss}$ less than or equal to the predicted geometric mean C_{max}

Table 1. Steady-state PK Exposure Metrics for Alternative Tislelizumab Doses vs Reference Doses

Parameter	150 mg Q2W	300 mg Q4W		400 mg Q6W		
	vs 200 mg Q3W	vs 200 mg Q3W	vs 5 mg/kg Q3W[a]	vs 200 mg Q3W	vs 5 mg/kg Q3W[a]	vs 2 mg/kg Q3W[b]
C_{max}	−5.8 (Yes)	31.4 (No)	−18.7 (Yes)	52.2 (No)	–25.1 (Yes)	–
$C_{average}$	12.3 (Yes)	12.6 (Yes)	–	0.4 (Yes)	–	–
C_{trough}	27.2 (Yes)	0.1 (Yes)	–	−28.4 (No)	–	10.7 (Yes)

Data are presented as difference (% = [GMtest − GMreference/GMreference] × 100) vs the 200 mg Q3W reference, [a]safety reference, or [b]efficacy reference (meets regulatory criteria, Yes/No).
Abbreviations: GM, geometric mean; Q#W, every # weeks.

Figure 2. Exposure–Response Analysis Between (A) Tislelizumab Exposure and ORR and (B) the Predicted Probability of Patients Achieving an Objective Response in the Context of the Exposures of the 200 mg Q3W, 150 mg Q2W, 300 mg Q4W, and 400 mg Q6W Dosing Regimens

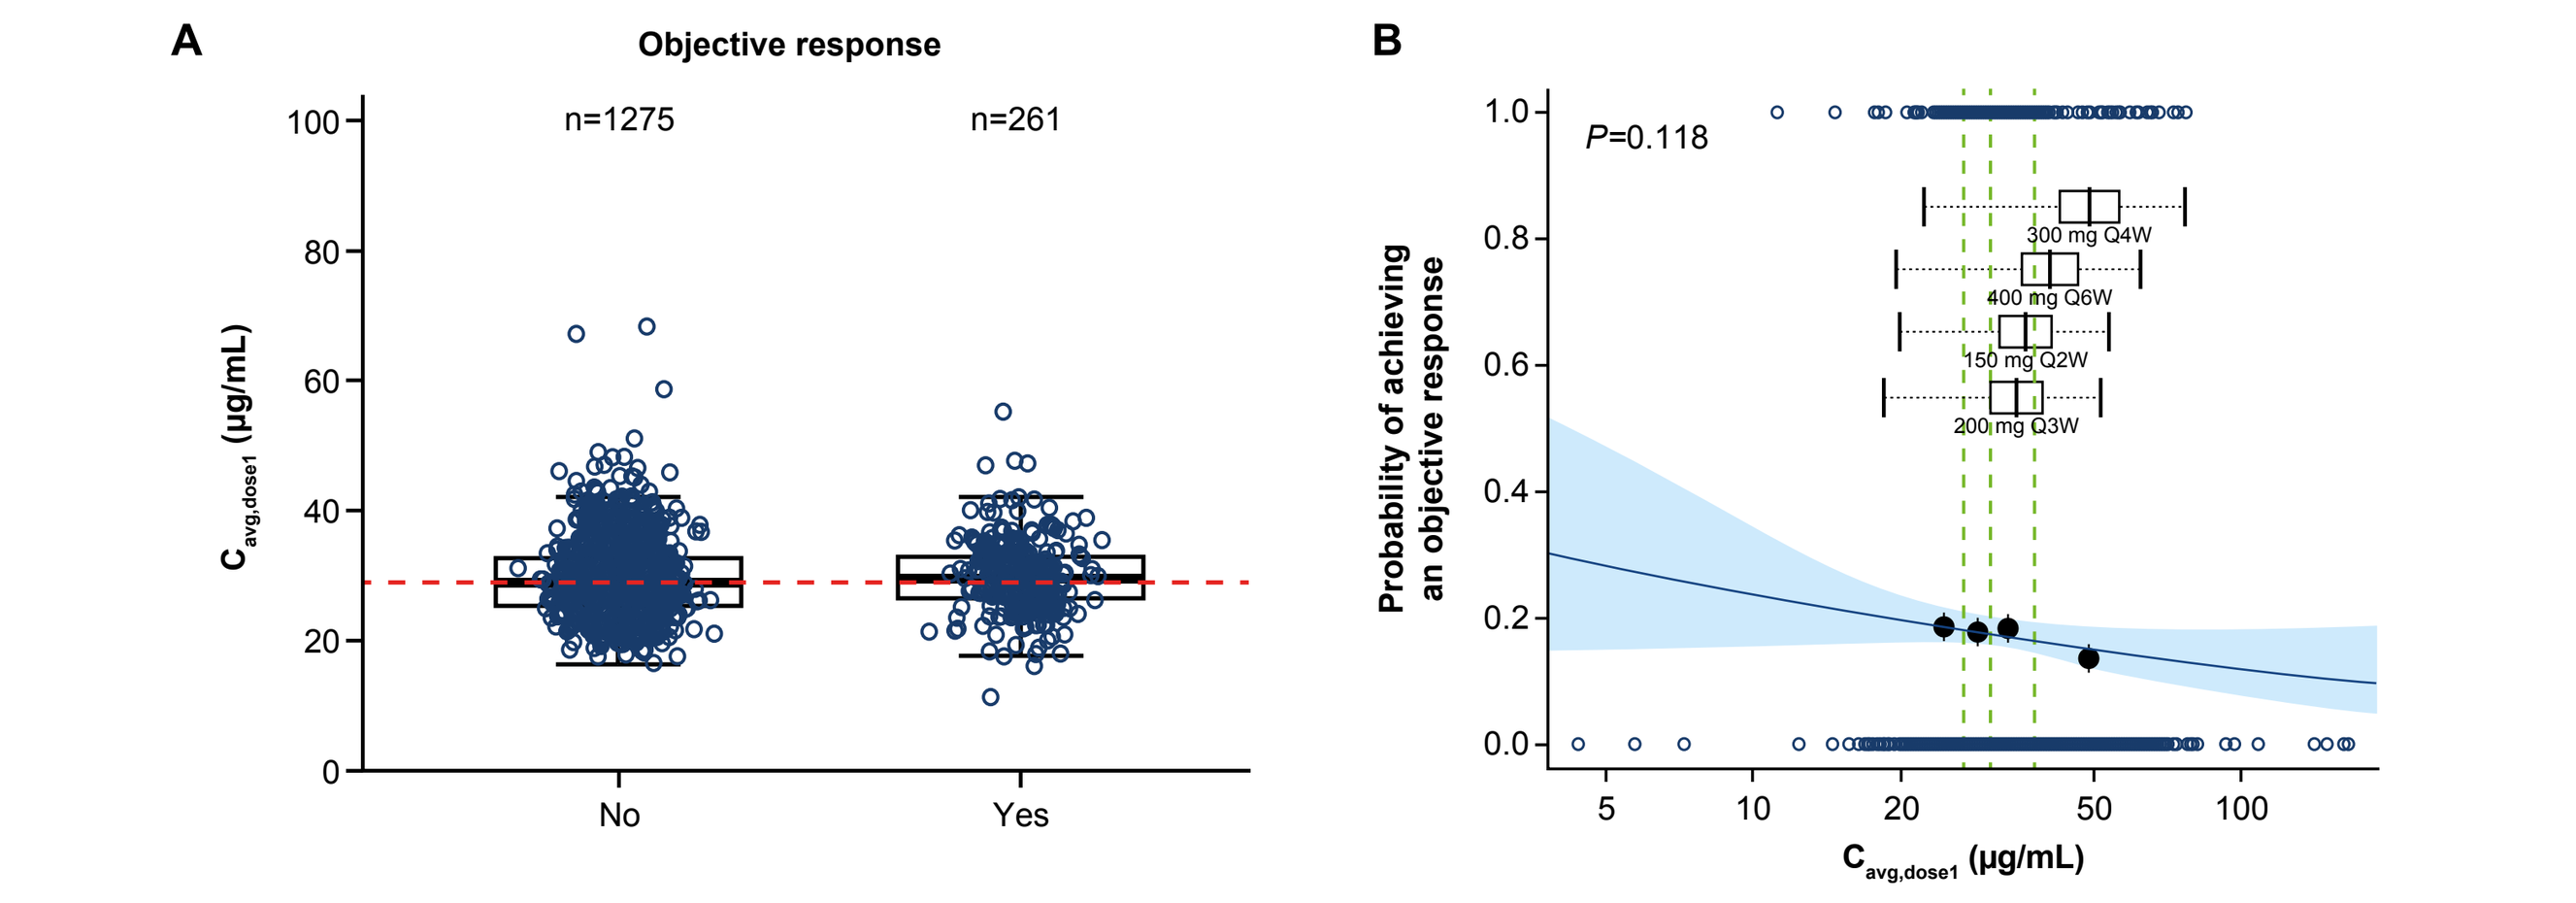
A
Objective response
n=1275
n=261
$C_{avg,dose1}$ (μg/mL)
No
Yes
B
P=0.118
300 mg Q4W
400 mg Q6W
150 mg Q2W
200 mg Q3W
Probability of achieving an objective response
$C_{avg,dose1}$ (μg/mL)

Abbreviations: $C_{avg,dose1}$, average serum concentration of the first dose; ORR, objective response rate; Q#W, every # weeks.

Table 2. Summary of Selected Safety Outcomes in the Exposure–Response Dataset

Safety endpoints, n (%)	BGB-A317-001 (n=450)[a]	BGB-A317-102 (n=300)[a]	BGB-A317-302 (n=254)[b]	BGB-A317-303 (n=532)[c]	Overall (N=1536)
Grade ≥3 TEAEs	207 (46.0)	137 (45.7)	123 (48.4)	230 (43.2)	697 (45.4)
Immune-mediated TEAEs	131 (29.1)	89 (29.7)	87 (34.3)	186 (35.0)	493 (32.1)
TEAEs leading to treatment discontinuation	39 (8.7)	34 (11.3)	49 (19.3)	65 (12.2)	187 (12.2)
Infusion-related reactions	55 (12.2)	7 (2.3)	11 (4.3)	4 (0.8)	77 (5.0)
TEAEs leading to dose modification	129 (28.7)	94 (31.3)	59 (23.2)	142 (26.7)	424 (27.6)
TEAEs of special interest	156 (34.7)	89 (29.7)	90 (35.4)	188 (35.3)	523 (34.0)
Serious TEAEs	169 (37.6)	85 (28.3)	109 (42.9)	189 (35.5)	552 (35.9)

[a]Patients with solid tumors. [b]Patients with ESCC. [c]Patients with NSCLC.
Abbreviations: ESCC, esophageal squamous cell carcinoma; NSCLC, non-small cell lung cancer; TEAE, treatment-emergent adverse event.

References

1. Zhang T, et al. *Cancer Immunol Immunother.* 2018;67:1079-1090.
2. Budha N, et al. *CPT Pharmacometrics Syst Pharmacol.* 2023;12:95-109.
3. Yu T, et al. *Clin Transl Sci.* 2024;17:e13769.
4. Yu T, et al. *Clin Transl Sci.* 2024. In submission.
5. US Food and Drug Administration. https://www.fda.gov/media/151745/download. Accessed August 15, 2024.

Acknowledgments

We would like to thank the investigators, the site support staff, and especially the patients for participating in this study. This study was sponsored by BeiGene, Ltd. Medical writing support was provided Russell Craddock, PhD, of Parexel, with funding provided by BeiGene, Ltd.

Disclosures

All listed authors are currently or were employees of BeiGene, Ltd. at the time the analysis was conducted, and may own shares in BeiGene, Ltd.

Author contact details: nageshwar.budha@beigene.com (Nageshwar Budha)

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.