SEQUOIA 5-year Follow-up in Arm C: Frontline Zanubrutinib Monotherapy in Patients with del(17p) and Treatment-naive Chronic Lymphocytic Leukemia/Small Lymphocytic Lymphoma

PS1565

Constantine S. Tam,[1] Paolo Ghia,[2,3] Mazyar Shadman,[4,5] Talha Munir,[6] Stephen S. Opat,[7] Patricia A. Walker,[8] Masa Lasica,[9] Ian W. Flinn,[10] Tian Tian,[11] Stephanie Agresti,[11] Jamie Hirata,[11] Jennifer R. Brown[12]

[1]Alfred Hospital and Monash University, Melbourne, VIC, Australia; [2]Università Vita-Salute San Raffaele, Milano, Italy; [3]IRCCS Ospedale San Raffaele, Milano, Italy; [4]Fred Hutchinson Cancer Center, Seattle, WA, USA; [5]University of Washington, Seattle, WA, USA; [6]Leeds Teaching Hospitals NHS Trust, Leeds, UK; [7]Lymphoma Research Group, School of Clinical Sciences at Monash Health, Monash University, Clayton, VIC, Australia; [8]Peninsula Health and Peninsula Private Hospital, Melbourne, VIC, Australia; [9]St Vincent's Hospital Melbourne, Melbourne, VIC, Australia; [10]Tennessee Oncology/OneOncology, Nashville, TN, USA; [11]BeOne Medicines Ltd, San Carlos, CA, USA; [12]Dana-Farber Cancer Institute, Boston, MA, USA

CONCLUSIONS

- SEQUOIA Arm C reports on the largest prospective cohort of uniformly treated patients with del(17p) TN CLL/SLL
- With a median follow-up of 5-years, zanubrutinib demonstrates durable efficacy in patients with del(17p)
 - The estimated 60-month PFS with zanubrutinib was 72.2%, similar to that observed in patients without del(17p)[6], highlighting that zanubrutinib overcomes the negative prognostic impact of del(17p)
- The benefit of zanubrutinib in patients with del(17p) was also demonstrated in the phase 3 ALPINE study, which demonstrated PFS superiority of zanubrutinib over ibrutinib[4]
- Long-term follow-up from SEQUOIA confirms the impressive zanubrutinib efficacy and good tolerability in patients with TN CLL/SLL with or without del(17p)

INTRODUCTION

- Zanubrutinib is a highly potent and selective next-generation Bruton tyrosine kinase (BTK) inhibitor that was designed to provide complete and sustained target inhibition and is the only BTK inhibitor to demonstrate superiority over ibrutinib in a head-to-head phase 3 trial[1-4]
- SEQUOIA (NCT03336333) is a registrational phase 3, open-label, randomized study that evaluated zanubrutinib in broad range of treatment naive (TN) chronic lymphocytic leukemia (CLL) patients, including those with high-risk features[5-7]
 - In Arms A and B, zanubrutinib monotherapy (Arm A) demonstrated superior PFS compared with bendamustine + rituximab (Arm B) in patients without del(17p) at 26.2-month follow-up and sustained PFS benefit at 5-year follow-up (Arm A: 75.8%)[5,6]
 - In Arm C, patients with del(17p) treated with zanubrutinib monotherapy have achieved high overall response rates and PFS, despite being at high risk for disease progression and death[7]
- Here, we present updated results from SEQUOIA Arm C after approximately 5 years of follow-up in a historically difficult to treat del(17p) patient population
 - To our knowledge, this reports the largest cohort of uniformly treated patients with del(17p) TN CLL/SLL

METHODS

Study design

- Arm C is a nonrandomized cohort of SEQUOIA patients with del(17p) that received zanubrutinib monotherapy; key study endpoints are shown in **Figure 1**
- ORR was assessed by investigator per the 2008 International Workshop on Chronic Lymphocytic Leukemia (iwCLL) guidelines[8] with modification for treatment-related lymphocytosis[9] for patients with CLL and per Lugano criteria[10] for patients with SLL
- ORR was defined as achievement of partial response with lymphocytosis (PR-L) or better

Assessments

- Sensitivity analyses were performed for PFS and OS with deaths due to COVID-19 infection, censored at the time of death if no prior progression was observed
- Response assessments were performed every 12 weeks after the first dose of study drug for 96 weeks, then every 24 weeks until progressive disease
- Adverse events (AEs) were graded by NCI-CTCAE version 4.03 and documented from the time of first dose of study drug, until 30 days after the last dose of study drug, or until disease progression (whichever occurred later) or until the first day of a new CLL/SLL treatment

Figure 1. Study Design

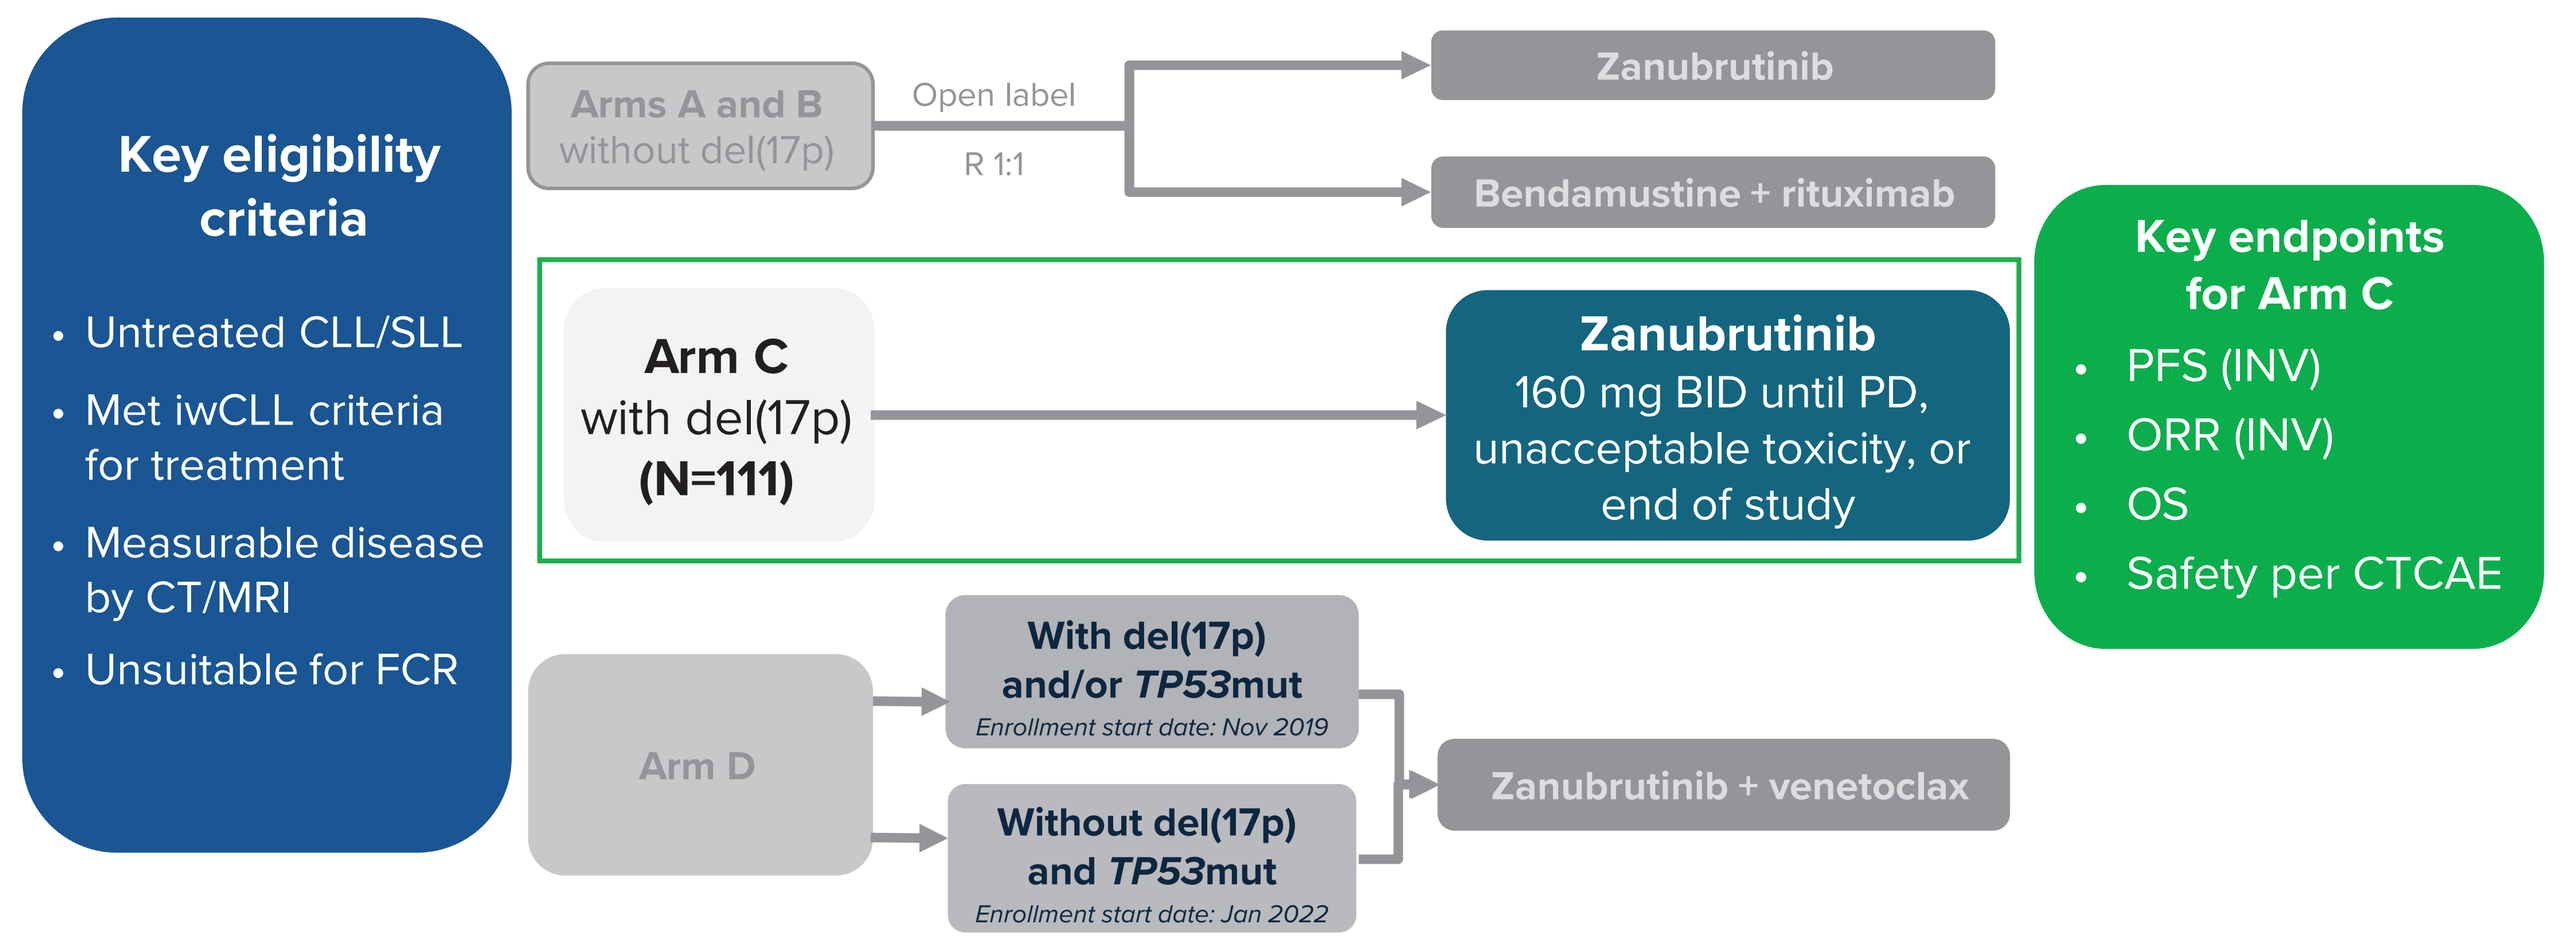
Key eligibility criteria
Untreated CLL/SLL
Met iwCLL criteria for treatment
Measurable disease by CT/MRI
Unsuitable for FCR
Arms A and B without del(17p)
Open label
R 1:1
Zanubrutinib
Bendamustine + rituximab
Arm C with del(17p) (N=111)
Zanubrutinib 160 mg BID until PD, unacceptable toxicity, or end of study
Arm D
With del(17p) and/or TP53mut
Enrollment start date: Nov 2019
Without del(17p) and TP53mut
Enrollment start date: Jan 2022
Zanubrutinib + venetoclax
Key endpoints for Arm C
PFS (INV)
ORR (INV)
OS
Safety per CTCAE

Abbreviations: BID, twice daily; CT, computed tomography; CLL, chronic lymphocytic leukemia; FCR, fludarabine, cyclophosphamide, and rituximab; INV, investigator-assessed; iwCLL, International Workshop on Chronic Lymphocytic Leukemia; MRD, minimal residual disease; MRI, magnetic resonance imaging; Mut, mutation; ORR, overall response rate; OS, overall survival; PD, progressive disease; PFS, progression-free survival; R, randomized; SLL, small lymphocytic lymphoma.

RESULTS

Disposition and baseline characteristics

- Between Feb 2018 and Mar 2019, 111 treatment-naive patients with del(17p) were enrolled to receive zanubrutinib
- As of April 30, 2024, at a median follow-up of 65.8 months (range, 5.0-75.0), 69 patients (62.2%) remained on treatment; the most common causes for treatment discontinuation were AEs (17.1%) and progressive disease (15.3%)
- Baseline demographic and disease characteristics are shown in **Table 1**

Table 1. Baseline Demographics and Clinical Characteristics

	All patients (N=111)
Age, median (range), years	71 (42-87)
≥65 years, n (%)	95 (85.6)
Male, n (%)	79 (71.2)
ECOG PS 0-1, n (%)	97 (87.3)
CLL, n (%)	100 (90.1)
SLL, n (%)	11 (9.9)
Binet stage C, n (%)[a]	37 (37.0)
Bulky disease, n (%)	
LDi ≥5 cm	44 (39.6)
LDi ≥10 cm	12 (10.8)
Median time from initial diagnosis, months	21.39
***TP53* mutated, n (%)**	47 (42.3)
del(17p), n (%)	110 (99.1)
del(17)p and *TP53* mutated, n (%)	47 (42.3)
IGHV mutated, n (%)	36 (32.4)
IGHV unmutated, n (%)	67 (60.4)
Complex karyotype, n (%)	
≥3 abnormalities	31 (27.9)
≥5 abnormalities	21 (18.9)

[a]Binet stage was assessed at study entry for patients with CLL.
Abbreviations: CLL, chronic lymphocytic leukemia; ECOG PS, Eastern Cooperative Oncology Group performance status; IGHV, immunoglobulin heavy-chain variable region; LDi, longest diameter; SLL, small lymphocytic lymphoma.

Efficacy

- Median PFS for zanubrutinib was not reached
- Estimated 60-month PFS rate (95% CI) was 72.2% (62.4-79.8) (**Figure 2A**)
 - When adjusted for COVID-19 impact, estimated 60-month PFS rate (95% CI) was 73.0% (63.3-80.6) (**Figure 2B**)
 - In patients with mutated and unmutated IGHV, estimated 60-month PFS rate (95% CI) was 74.6% (56.9-85.9) and 70.7% (57.4-80.6), respectively (**Figure 2C**)
- In total, 18 deaths occurred in the study and median OS was not reached
- Estimated 60-month OS rate (95% CI) was 85.1% (76.9-90.6) (**Figure 3A**)
 - When adjusted for COVID-19 the estimated 60-month OS rate (95% CI) was 87.0% (79.0-92.1) (**Figure 3B**)

Figure 2. PFS, COVID-19 Adjusted PFS, and PFS by IGHV Mutation Status

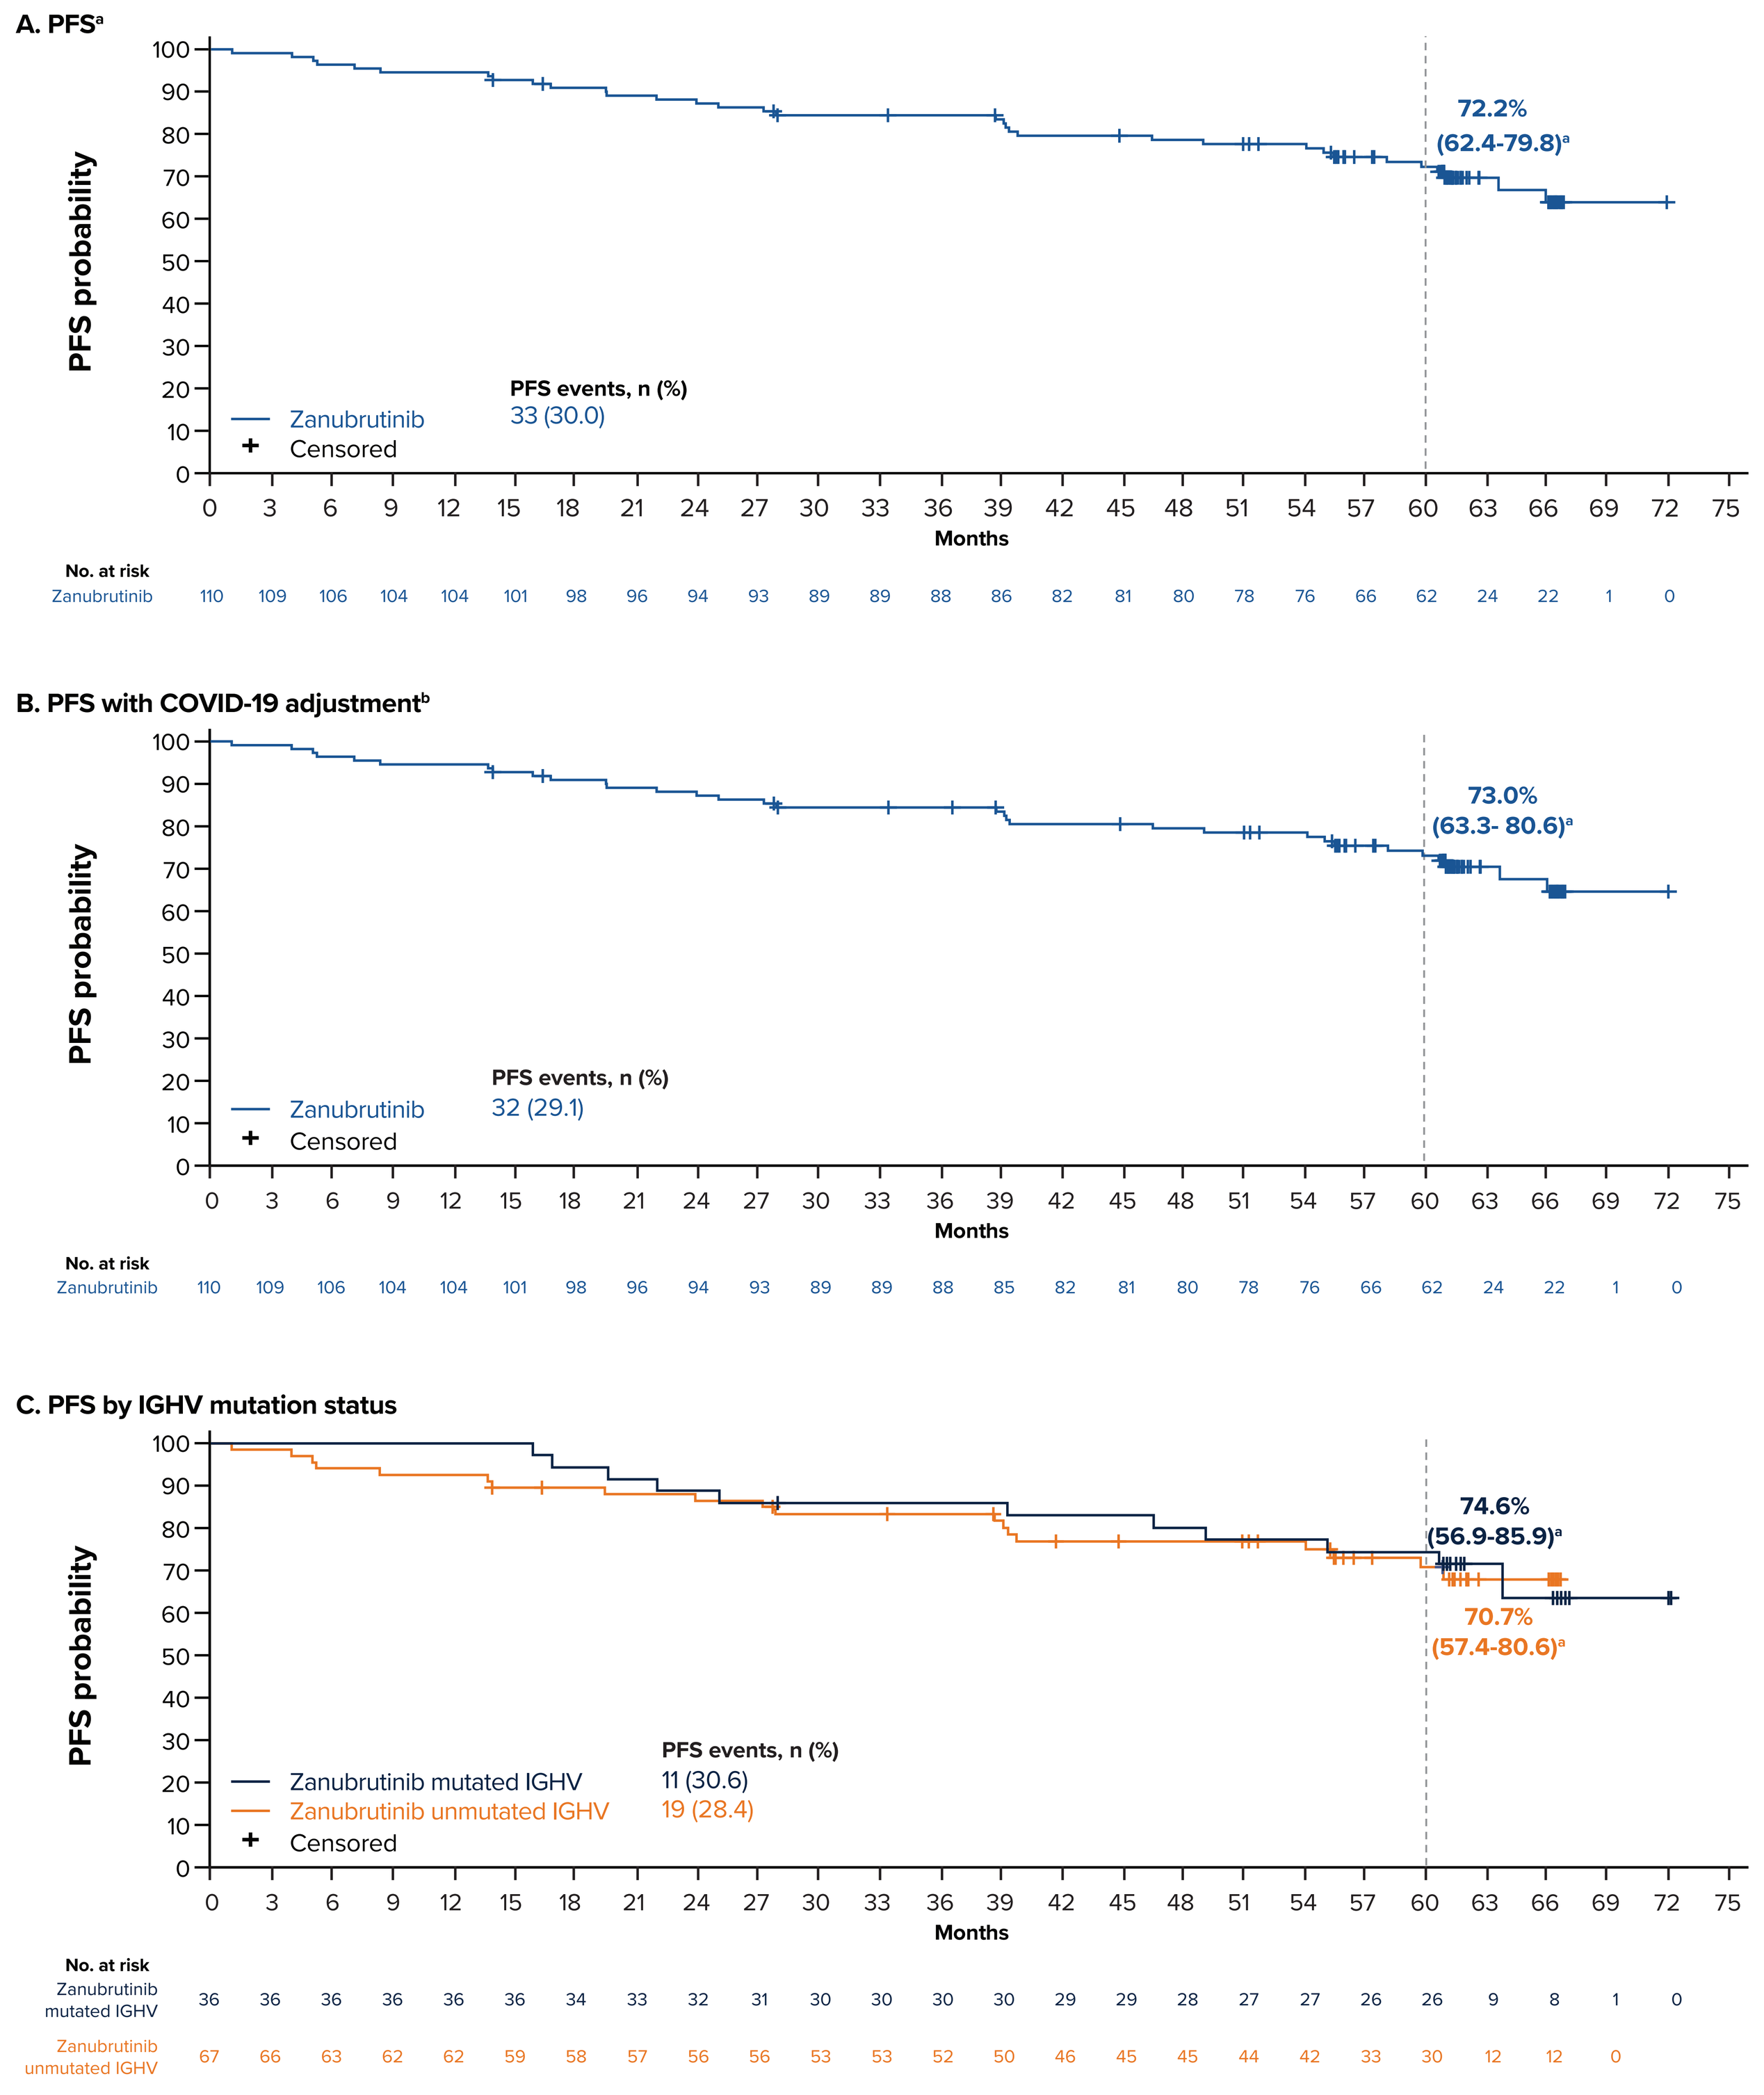
A. PFS[a]
PFS probability
72.2% (62.4-79.8)[a]
Zanubrutinib
Censored
PFS events, n (%)
33 (30.0)
Months
No. at risk
Zanubrutinib 110 109 106 104 104 101 98 96 94 93 89 89 88 86 82 81 80 78 76 66 62 24 22 1 0
B. PFS with COVID-19 adjustment[b]
73.0% (63.3- 80.6)[a]
PFS events, n (%)
32 (29.1)
No. at risk
Zanubrutinib 110 109 106 104 104 101 98 96 94 93 89 89 88 85 82 81 80 78 76 66 62 24 22 1 0
C. PFS by IGHV mutation status
74.6% (56.9-85.9)[a]
70.7% (57.4-80.6)[a]
Zanubrutinib mutated IGHV
Zanubrutinib unmutated IGHV
Censored
PFS events, n (%)
11 (30.6)
19 (28.4)
No. at risk
Zanubrutinib mutated IGHV 36 36 36 36 36 36 34 33 32 31 30 30 30 30 29 29 28 27 27 26 26 9 8 1 0
Zanubrutinib unmutated IGHV 67 66 63 62 62 59 58 57 56 56 53 53 52 50 46 45 45 44 42 33 30 12 12 0

[a]95% CI values. [b]Data is presented in patients with del(17p), confirmed by central laboratory (N=110).
Abbreviations: PFS, progression-free survival; IGHV, immunoglobulin heavy-chain variable region.

Figure 3: OS and COVID-19 Adjusted OS

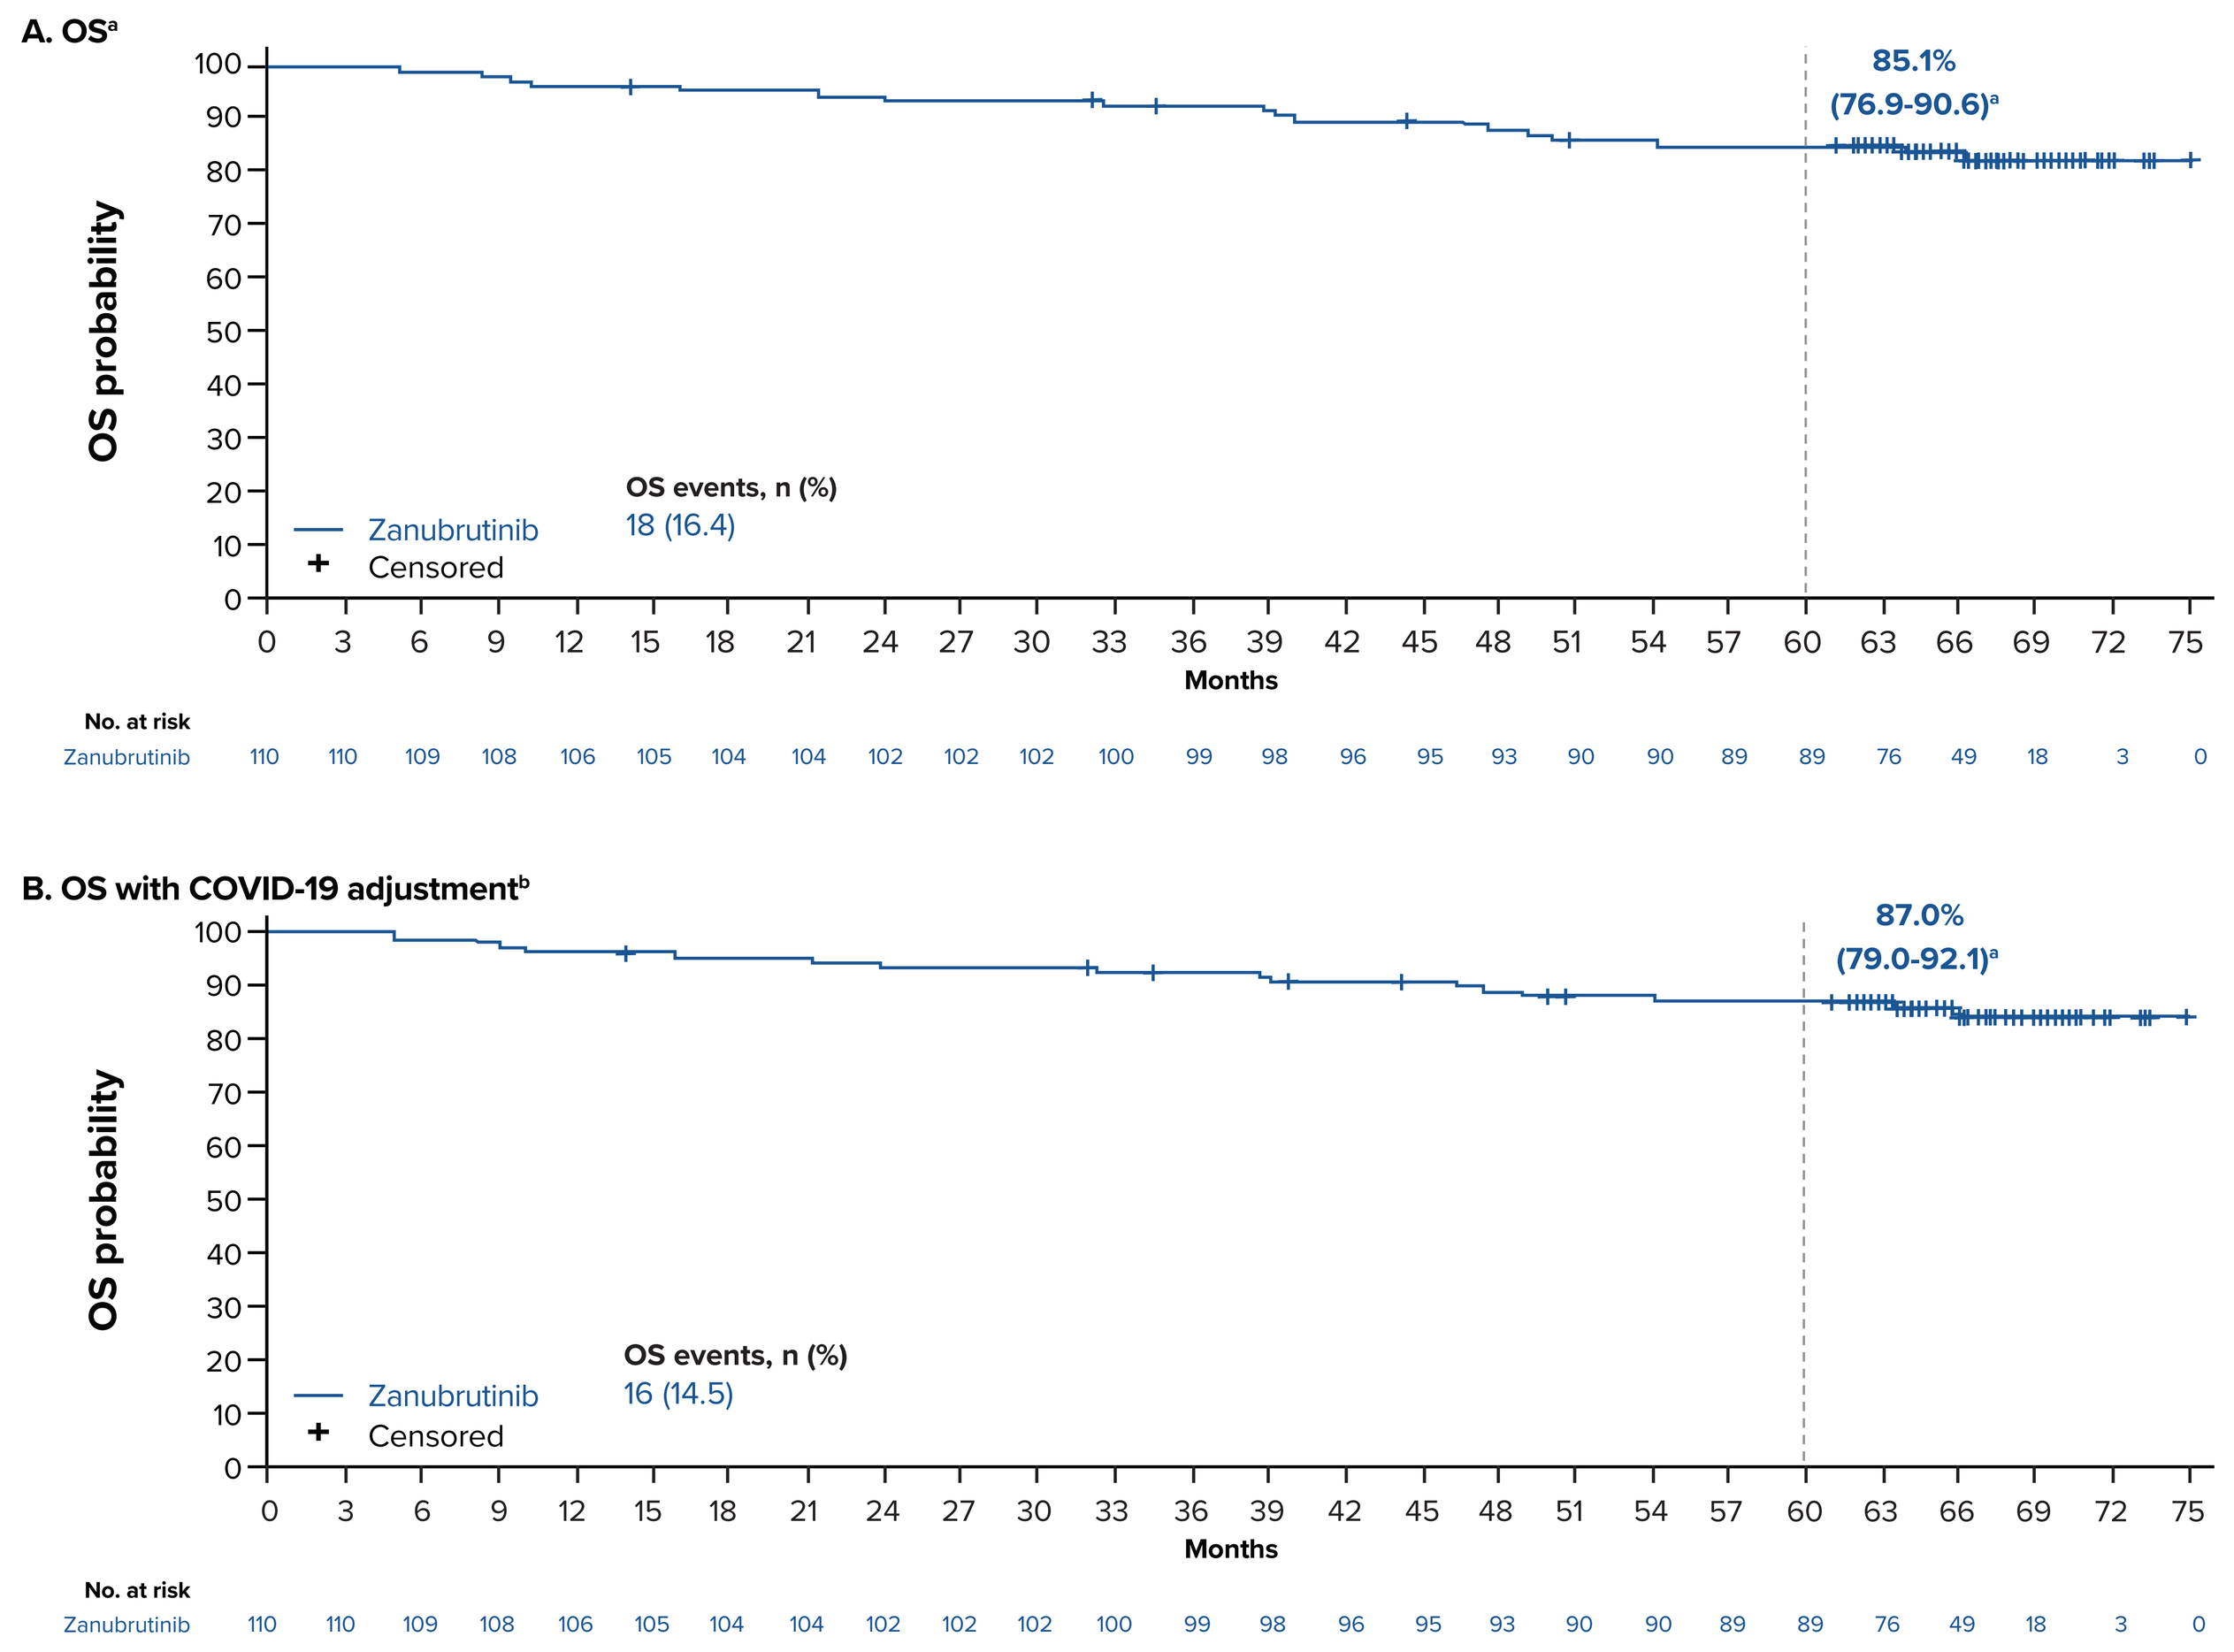
A. OS[a]
OS probability
85.1% (76.9-90.6)[a]
Zanubrutinib
Censored
OS events, n (%)
18 (16.4)
Months
No. at risk
Zanubrutinib 110 110 109 108 106 105 104 104 102 102 102 100 99 98 96 95 93 90 90 89 89 76 49 18 3 0
B. OS with COVID-19 adjustment[b]
87.0% (79.0-92.1)[a]
OS events, n (%)
16 (14.5)
No. at risk
Zanubrutinib 110 110 109 108 106 105 104 104 102 102 102 100 99 98 96 95 93 90 90 89 89 76 49 18 3 0

[a]95% CI values. [b]Data is presented in patients with del(17p), confirmed by central laboratory (N=110).
Abbreviations: OS, overall survival.

Best overall response

- The ORR was 97.3%, and the combined complete response/complete response with incomplete hematologic recovery rate (CR/CRi) was 18.2% (**Table 2**)

Table 2: Best Overall Response Rate

	Zanubrutinib (N=110)[a]
ORR, n (%)	107 (97.3)
Best overall response, n (%)	
CR/CRi	20 (18.2)
nPR	3 (2.7)
PR	84 (76.4)
PR-L	0
SD	2 (1.8)
PD	1 (0.9)

[a]Patients with del(17p), confirmed by central laboratory.
Abbreviations: CR, complete response; CRi, complete response with incomplete hematopoietic recovery; ORR, overall response rate; nPR, nodular partial response; PD, progressive disease; PR, partial response; PR-L, partial response with lymphocytosis; SD, stable disease.

Safety

- The most common treatment-emergent adverse events (TEAEs) and TEAEs of special interest are presented in **Figure 4**
- AEs led to death in 6 patients (5.4%)

Figure 4. TEAEs and TEAEs of Special Interest

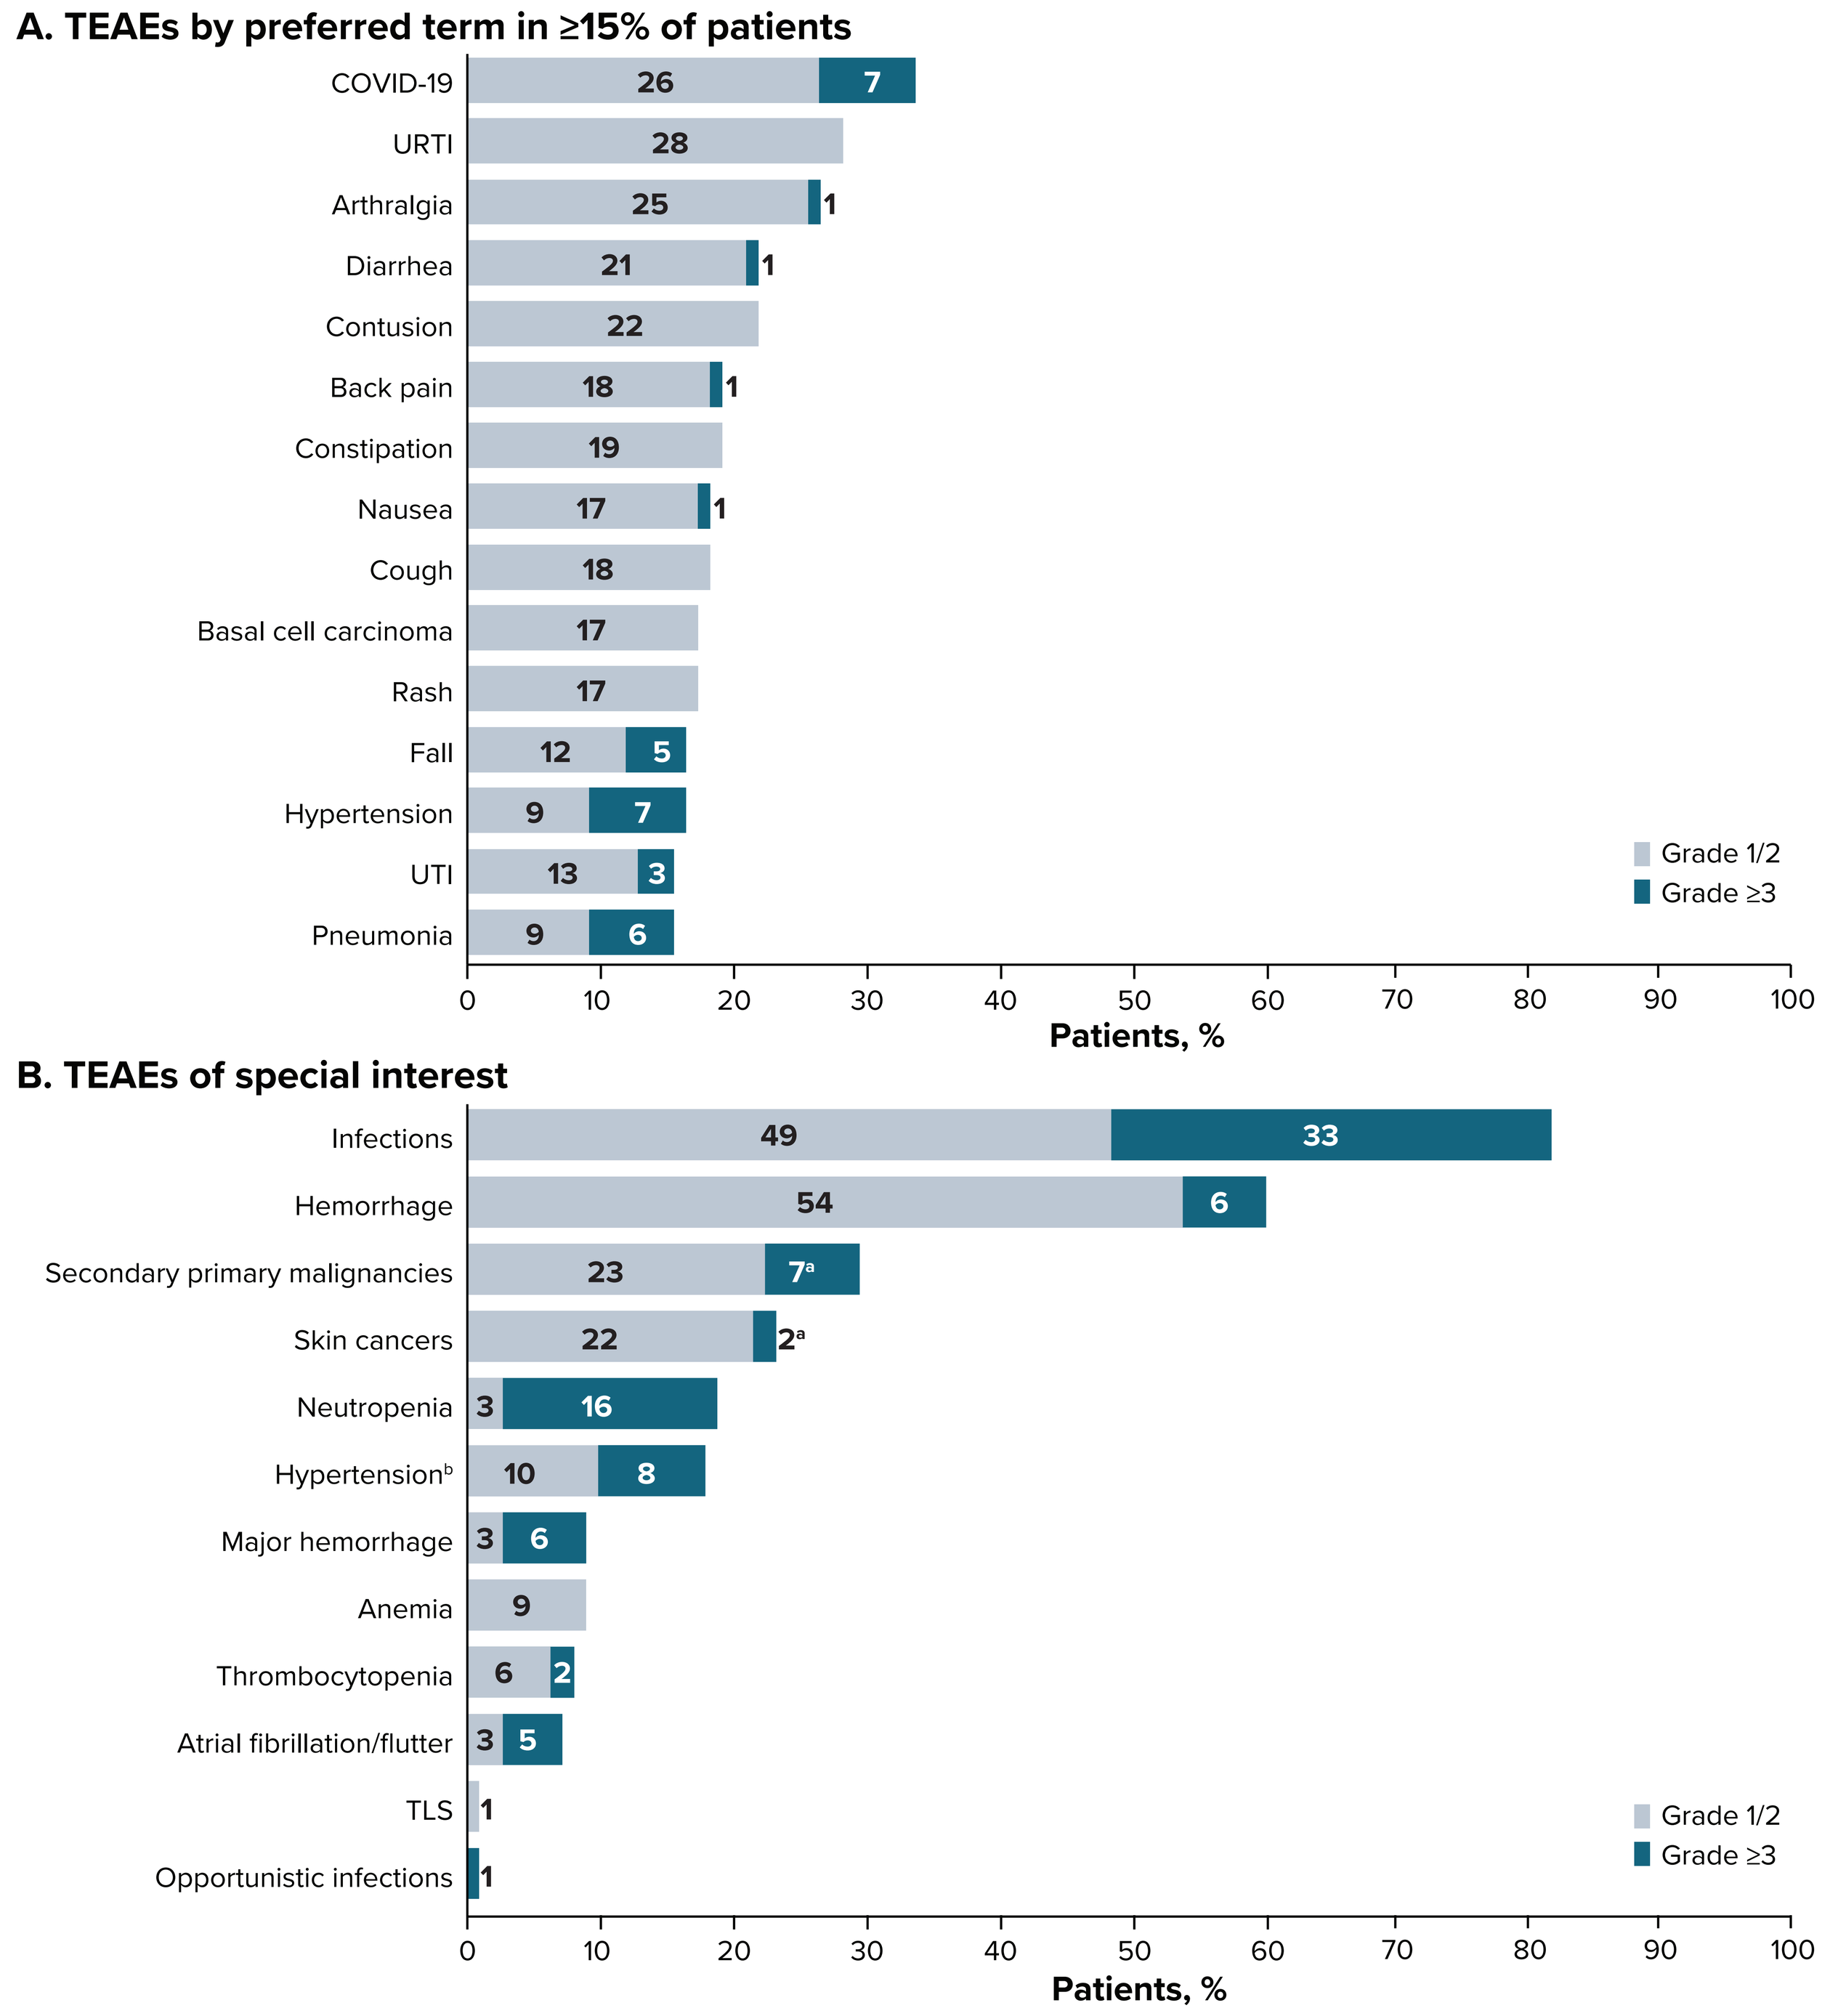
A. TEAEs by preferred term in ≥15% of patients
COVID-19 26 7
URTI 28
Arthralgia 25 1
Diarrhea 21 1
Contusion 22
Back pain 18 1
Constipation 19
Nausea 17 1
Cough 18
Basal cell carcinoma 17
Rash 17
Fall 12 5
Hypertension 9 7
UTI 13 3
Pneumonia 9 6
Grade 1/2
Grade ≥3
Patients, %
B. TEAEs of special interest
Infections 49 33
Hemorrhage 54 6
Secondary primary malignancies 23 7[a]
Skin cancers 22 2[a]
Neutropenia 3 16
Hypertension[b] 10 8
Major hemorrhage 3 6
Anemia 9
Thrombocytopenia 6 2
Atrial fibrillation/flutter 3 5
TLS 1
Opportunistic infections 1
Grade 1/2
Grade ≥3
Patients, %

[a]Includes two patients with malignant melanoma. [b]Includes hypertension, increased blood pressure, hypertensive crisis and hypertensive heart disease.
Abbreviations: AE, adverse event; TEAE, treatment-emergent adverse event; TLS, tumor lysis syndrome; URTI, upper respiratory tract infection; UTI, urinary tract infection.

REFERENCES

1. Guo Y, et al. *J Med Chem.* 2019;62(17):7923-7940.
2. Brukinsa (zanubrutinib). Prescribing information. BeiGene USA; 2024.
3. Brukinsa (zanubrutinib). Summary of product characteristics. BeiGene Ireland Limited; 2024.
4. Brown J, et al. *Blood.* 2024;144(26):2706-2717.
5. Tam CS, et al. *Lancet Oncol.* 2022;23(8):1031-1043.
6. Shadman M, et al. *J Clin Oncol.* 2025;43(7):780-787.
7. Tam CS, et al. *Haematologica.* 2021;106(9):2354-2363.
8. Hallek M, et al. *Blood.* 2008;111(12):5446-5456.
9. Cheson BD, et al. *J Clin Oncol.* 2012;30(23):2820-2822.
10. Cheson BD, et al. *J Clin Oncol.* 2014;32(27):3059-3967.

ACKNOWLEDGMENTS

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers.
This study was sponsored by BeOne Medicines Ltd. Medical writing support was provided by Manoshi Nath, MSc, of Nucleus Global, an Inizio company, and supported by BeOne Medicines.

DISCLOSURES

CST: Honoraria: AbbVie, Janssen, BeOne Medicines Ltd, AstraZeneca. **PG:** Honoraria: AbbVie, AstraZeneca, BeOne Medicines Ltd, BMS, Janssen, Galapagos, Lilly/Loxo, MSD, Roche; Research funding: AbbVie, AstraZeneca, BMS, Janssen. **MS:** Consultant: AbbVie, Genentech, AstraZeneca, Genmab, Janssen, BeOne Medicines Ltd, BMS, MorphoSys/Incyte, Kite Pharma, Lilly, Fate Therapeutics, Nurix, Merck; Research funding: Mustang Bio, Genentech, AbbVie, BeOne Medicines Ltd, AstraZeneca, Genmab, Morphosys/Incyte, Vincerx; Stock: Koi Biotherapeutics; Employment: BMS (spouse). **TM:** Honoraria: BeOne Medicines Ltd, AstraZeneca, Sobi, Roche, Janssen, AbbVie, Lilly; Consultant: AbbVie, BeOne Medicines Ltd, Sobi, Alexion, Novartis, Janssen, AstraZeneca, Lilly, Roche; Research grants: Janssen, AbbVie; Travel, accommodations, or expenses: Alexion, BeOne Medicines Ltd, AbbVie, Janssen, AstraZeneca; Advisory board: AbbVie, BeOne Medicines Ltd, AstraZeneca, Janssen. **SSO:** Current employment: Monash Health; Consultancy (includes expert testimony): AbbVie, AstraZeneca, BeOne Medicines Ltd, Janssen, Novartis; Research funding: AbbVie, AstraZeneca, BeOne Medicines Ltd, Gilead, Janssen, Novartis, Pharmacyclics, Roche, Takeda; Honoraria: AbbVie, AstraZeneca, Gilead, Janssen, Merck, Roche, Takeda. PAW: **PW:** No disclosure. **ML:** Honoraria: Janssen, AbbVie; Advisory board: Janssen, Sobi, AbbVie, Recordati. **IWF:** Employment: OneOncology; Research grants (all payments made to institution): AbbVie, AstraZeneca, BeOne Medicines Ltd, BMS, Celgene, City of Hope National Medical Center, Epizyme, Fate Therapeutics, Genentech, Gilead Sciences, IGM Biosciences InnoCare Pharma, Incyte, Janssen, Kite Pharma, Loxo, Marker Therapeutics, Merck, MorphoSys, Myeloid Therapeutics, Novartis, Nurix, Pfizer, Roche, Seattle Genetics, TG Therapeutics, Vincerx Pharma, 2seventy bio; Consultant fees (all payments made to physician): Abbvie, BeOne Medicines Ltd, Genentech, Genmab, KITE, Vincerx; Board of Directors or advisory committee: OneOncology, Vincerx Adv Committee. **TT:** Employment and may own stock: BeOne Medicines Ltd. **SA:** Employment, may own stock, travel, accommodations, or expenses: BeOne Medicines Ltd. **JH:** Employment: BeOne Medicines Ltd, Genentech; Stocks or other ownership: BeOne Medicines Ltd, Roche. **JRB:** Consultant: Abbvie, Acerta/AstraZeneca, Alloplex Biotherapeutics, BeOne Medicines Ltd, BMS, EcoR1, Galapagos NV, Genentech/Roche, Grifols Worldwide Operations, InnoCare Pharma Inc, Kite Pharma, Loxo/Lilly, Magnet Biomedicine, Merck, Pharmacyclics; Research funding: BeOne Medicines Ltd, Gilead, iOnctura, Loxo/Lilly, MEI Pharma, TG Therapeutics; Royalties: UpToDate; Data Safety Monitoring Board for Grifols Therapeutics.

Presented at the EHA2025 Congress; June 12-15, 2025; Milan, Italy
Data originally presented at the 2025 ASCO Annual Meeting; May 30-June 3, 2025; Chicago, IL, USA. Abstract 7011