PF601

Subsequent Therapies and Time to Second Progression-Free Survival Events in Treatment-Naive Chronic Lymphocytic Leukemia/ Small Lymphocytic Lymphoma Previously Treated With Zanubrutinib or Bendamustine-Rituximab in SEQUOIA: Long-Term Follow-Up

Constantine S. Tam,[1] Brad S. Kahl,[2] Talha Munir,[3] Tadeusz Robak,[4] Jennifer R. Brown,[5] Paolo Ghia,[6] Stephen S. Opat,[7] Patricia A. Walker,[8] Masa Lasica,[9] Ian W. Flinn,[10] Wojciech Jurczak,[11] Jiayi Shen,[12] Marcus Lefebure,[13] Jamie Hirata,[12] Mazyar Shadman[14]

[1]Alfred Hospital and Monash University, Melbourne, VIC, Australia; [2]Siteman Cancer Center, Washington University School of Medicine, St. Louis, MO, USA; [3]Leeds Teaching Hospitals NHS Trust, Leeds, UK; [4]Copernicus Memorial Hospital, Medical University of Lodz, Lodz, Poland; [5]Dana-Farber Cancer Institute, Boston, MA, USA; [6]Università Vita-Salute San Raffaele, Milano, Italy, and Comprehensive Cancer Center, IRCCS Ospedale San Raffaele, Milano, Italy; [7]Lymphoma Research Group, School of Clinical Sciences at Monash Health, Monash University, Clayton, VIC, Australia; [8]Peninsula Health and Peninsula Private Hospital, Melbourne, VIC, Australia; [9]St Vincent's Hospital Melbourne, Melbourne, VIC, Australia; [10]Tennessee Oncology/OneOncology, Nashville, TN, USA; [11]Maria Sklodowska-Curie National Research Institute of Oncology, Kraków, Poland; [12]BeOne Medicines, Ltd, San Carlos, CA, USA; [13]BeOne Medicines, Ltd, London, UK; [14]Fred Hutchinson Cancer Center and University of Washington, Seattle, WA, USA

Supplemental materials can be found here

CONCLUSIONS

- To our knowledge, the long-term PFS rates observed with zanubrutinib are among the highest reported to date versus approved BTK inhibitor monotherapies and fixed-duration venetoclax-based regimens
- PFS2 remained superior with zanubrutinib, despite the broad use of effective novel subsequent therapies (including crossover) in patients originally treated with BR
- Time to symptomatic progression requiring treatment was prolonged with zanubrutinib versus BR, as reflected by improvements in TTNT and TTNT-D
- Responses to subsequent BCL2 inhibitor–based therapy were favorable after more than 3 years of follow-up, indicating that prior zanubrutinib exposure does not compromise sensitivity to later targeted agents
- Collectively, these data reaffirm zanubrutinib as a leading frontline treatment option in CLL, delivering durable PFS while maintaining responsiveness to subsequent lines of therapy. These findings further support the early use of zanubrutinib to achieve sustained disease control while preserving future treatment options

INTRODUCTION

- Zanubrutinib is a highly potent and selective next-generation Bruton tyrosine kinase (BTK) inhibitor that was designed to provide complete and sustained target inhibition, and is the only BTK inhibitor to demonstrate superiority over ibrutinib in a head-to-head phase 3 trial[1-3]
- Zanubrutinib has continuous exposure coverage above its IC50 compared with ibrutinib and acalabrutinib which is expected to lead to more sustained and complete BTK inhibition to improve efficacy[1]
- SEQUOIA (NCT03336333) is a registrational phase 3 study with four treatment arms that evaluated zanubrutinib in a broad range of patients with treatment-naive (TN) chronic lymphocytic leukemia/small lymphocytic lymphoma (CLL/SLL)[1,4-6]
 - In Arms A and B, zanubrutinib monotherapy (Arm A) demonstrated superior progression-free survival (PFS) compared with bendamustine-rituximab (BR; Arm B) in patients without del(17p) at 72.8 months of follow-up (hazard ratio [HR], 0.28; 95% CI, 0.20-0.38; *P*<.0001)[7]
 - Consistent efficacy and a favorable safety profile have also been observed in older patients, supporting the use of zanubrutinib in a broad, representative TN population[8]
- CLL/SLL remains incurable, and most patients will require multiple lines of therapy; therefore, frontline treatment selection must prioritize durable efficacy, appropriate sequencing, and optimal outcomes for high-risk subgroups, including ensuring patients with unmutated IGHV receive the most effective therapy upfront to maximize long-term benefit and subsequent treatment options
- However, long-term evidence describing outcomes with subsequent therapies following disease progression remains limited
- Here, we present 7-year follow-up data from Arms A and B of SEQUOIA, including time to second progression-free survival event (PFS2), to characterize long-term outcomes and treatment sequencing following frontline zanubrutinib

METHODS

- Patients without del(17p) were randomized to receive continuous zanubrutinib or six cycles of BR; patients who received BR could cross over to receive zanubrutinib after disease progression and prior to the start of any other therapy for CLL/SLL (**Figure 1**)
- All patients were followed up post disease progression for details on any subsequent CLL/SLL treatments
- This analysis evaluated PFS2, time to next treatment (TTNT) and TTNT or death (TTNT-D), and overall survival (OS)
- *P* values are descriptive only

Figure 1. SEQUOIA Study Design

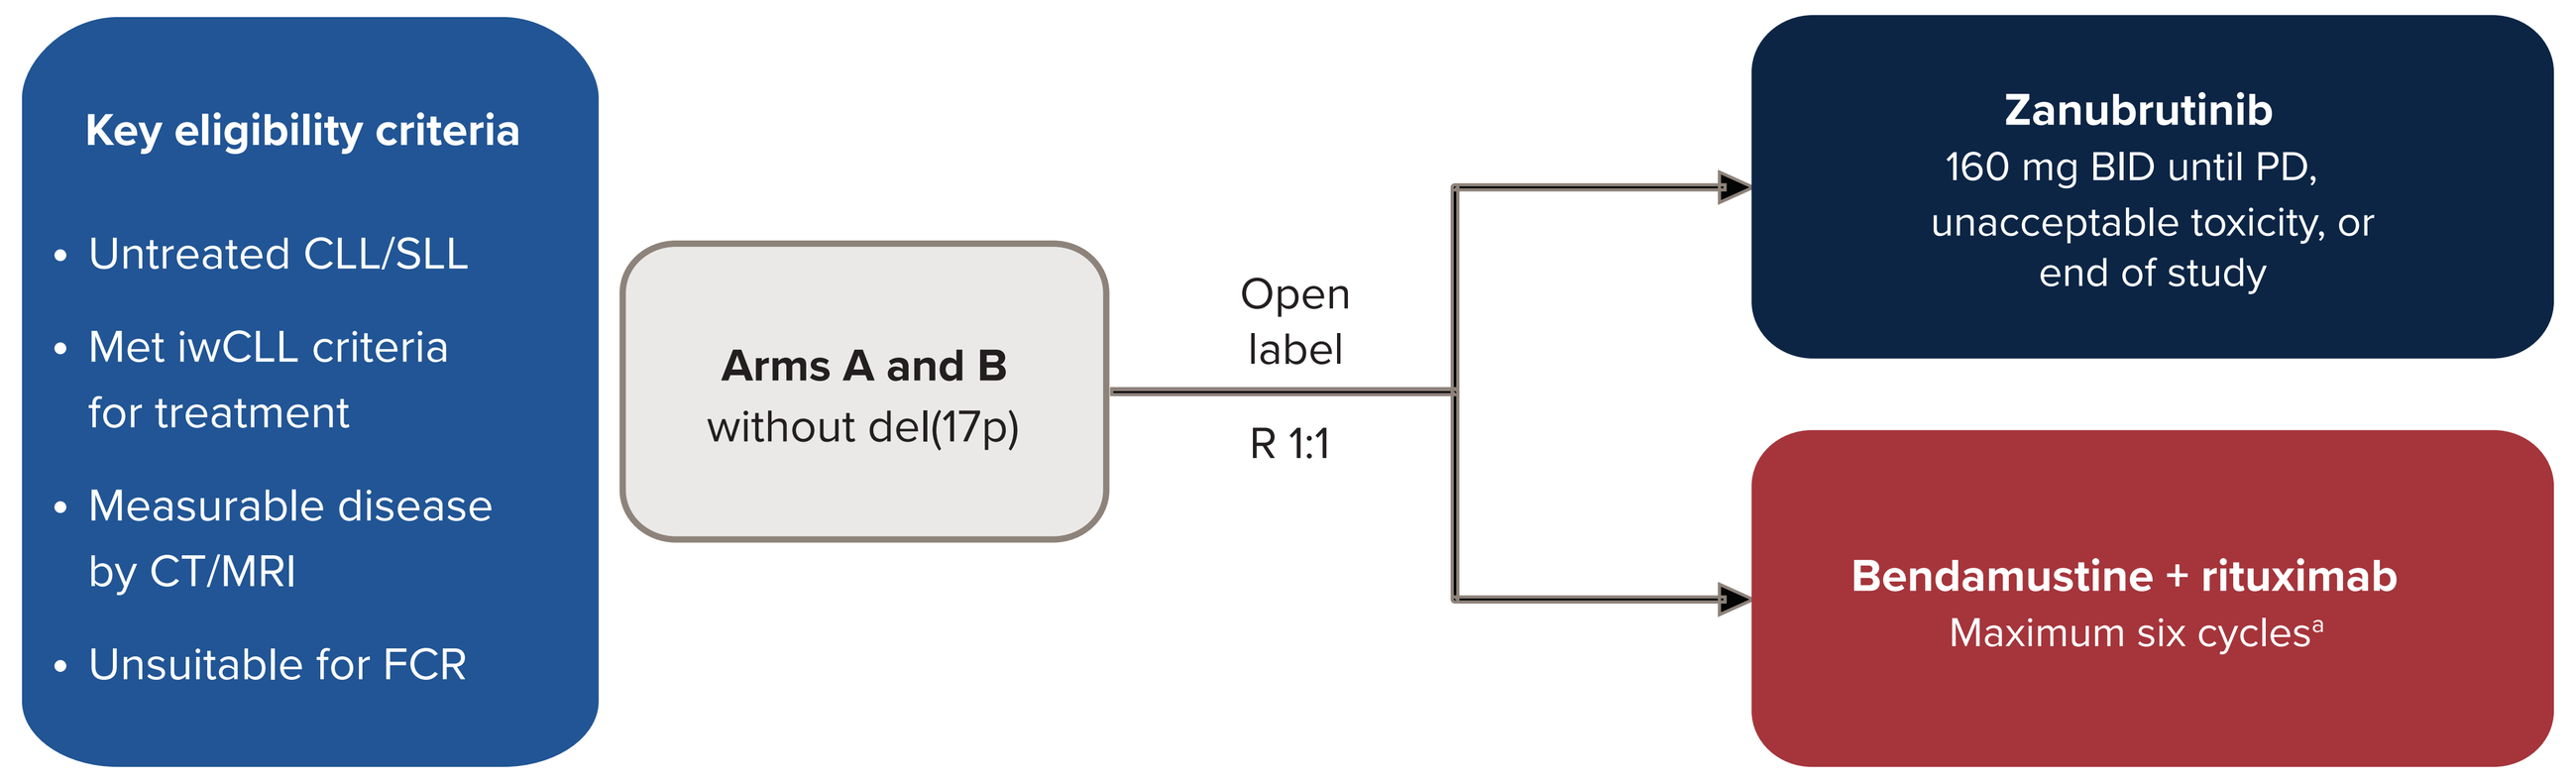
Key eligibility criteria
• Untreated CLL/SLL
• Met iwCLL criteria for treatment
• Measurable disease by CT/MRI
• Unsuitable for FCR
Arms A and B without del(17p)
Open label
R 1:1
Zanubrutinib
160 mg BID until PD, unacceptable toxicity, or end of study
Bendamustine + rituximab
Maximum six cycles[a]

[a]Bendamustine 90 mg/m² IV on days 1 and 2 for six cycles + rituximab 375 mg/m² IV the day before or on day 1 of cycle 1 and 500 mg/m² on day 1 of cycles 2-6.
Abbreviations: BID, twice daily; CLL, chronic lymphocytic leukemia; CT, computed tomography; FCR, fludarabine, cyclophosphamide, and rituximab; IV, intravenous; iwCLL, International Workshop on Chronic Lymphocytic Leukemia; MRI, magnetic resonance imaging; PD, progressive disease; R, randomized; SLL, small lymphocytic lymphoma.

RESULTS

Disposition and Baseline Characteristics

- In total, 479 patients were randomized: 241 to zanubrutinib and 238 to BR
- As of April 15, 2026, median follow-up was 84.01 months (range, 0.0-101.5); median treatment duration in Arm A was 82.50 months (range, 0.5-101.4)
- Overall, 130 patients (53.9%) were still receiving zanubrutinib
- In total, 69 patients (29%) from Arm B crossed over to receive zanubrutinib after disease progression
- Baseline demographics and disease characteristics were similar between the treatment arms (**Table 1**)
- Overall, 208/241 patients (86.3%) treated with zanubrutinib had not received subsequent therapy, and 36 patients had died without subsequent therapy

Table 1. Patient Demographics and Clinical Characteristics

	Arm A: zanubrutinib n=241	Arm B: BR n=238
Age, median (range), years	70 (40-86)	70 (35-87)
≥65 years, n (%)	198 (82)	195 (82)
Male, n (%)	154 (64)	144 (61)
ECOG PS 2, n (%)	15 (6)	20 (8)
Binet stage C, n (%)[a]	65 (29)	66 (30)
Bulky disease ≥5 cm, n (%)	69 (29)	73 (31)
***TP53* mutation, n (%)**		
Detected with VAF ≥1.0%	15 (6)	13 (5)
Not detected or VAF <1.0%	217 (90)	210 (88)
IGHV unmutated, n/N (%)	125/234 (53)	123/232 (53)
Complex karyotype (≥3 abnormalities), n/N (%)[b]	23/162 (14)	22/159 (14)

[a]Percentages are based on number of patients with CLL. [b]Patients with missing/insufficient metaphase activity were omitted from the complex karyotype analysis.
Abbreviations: BR, bendamustine and rituximab; CLL, chronic lymphocytic leukemia; ECOG PS, Eastern Cooperative Oncology Group performance status; IGHV, immunoglobulin heavy-chain variable region; VAF, variant allele frequency.

Efficacy

PFS

- Sustained PFS superiority with zanubrutinib vs BR was observed (HR, 0.28; 95% CI, 0.21-0.38; *P*<.0001), with 78-month PFS estimates of 71.8% (95% CI, 65.3-77.3) for zanubrutinib and 31.0% (95% CI, 24.3-37.9) for BR (**Supplementary Figure S1**)
- After adjustment for COVID-19, 78-month PFS estimates were 74.6% (95% CI, 68.1-79.9) for zanubrutinib and 31.4% (95% CI, 24.7-38.4) for BR (**Figure 2**)

Figure 2. COVID-Adjusted PFS

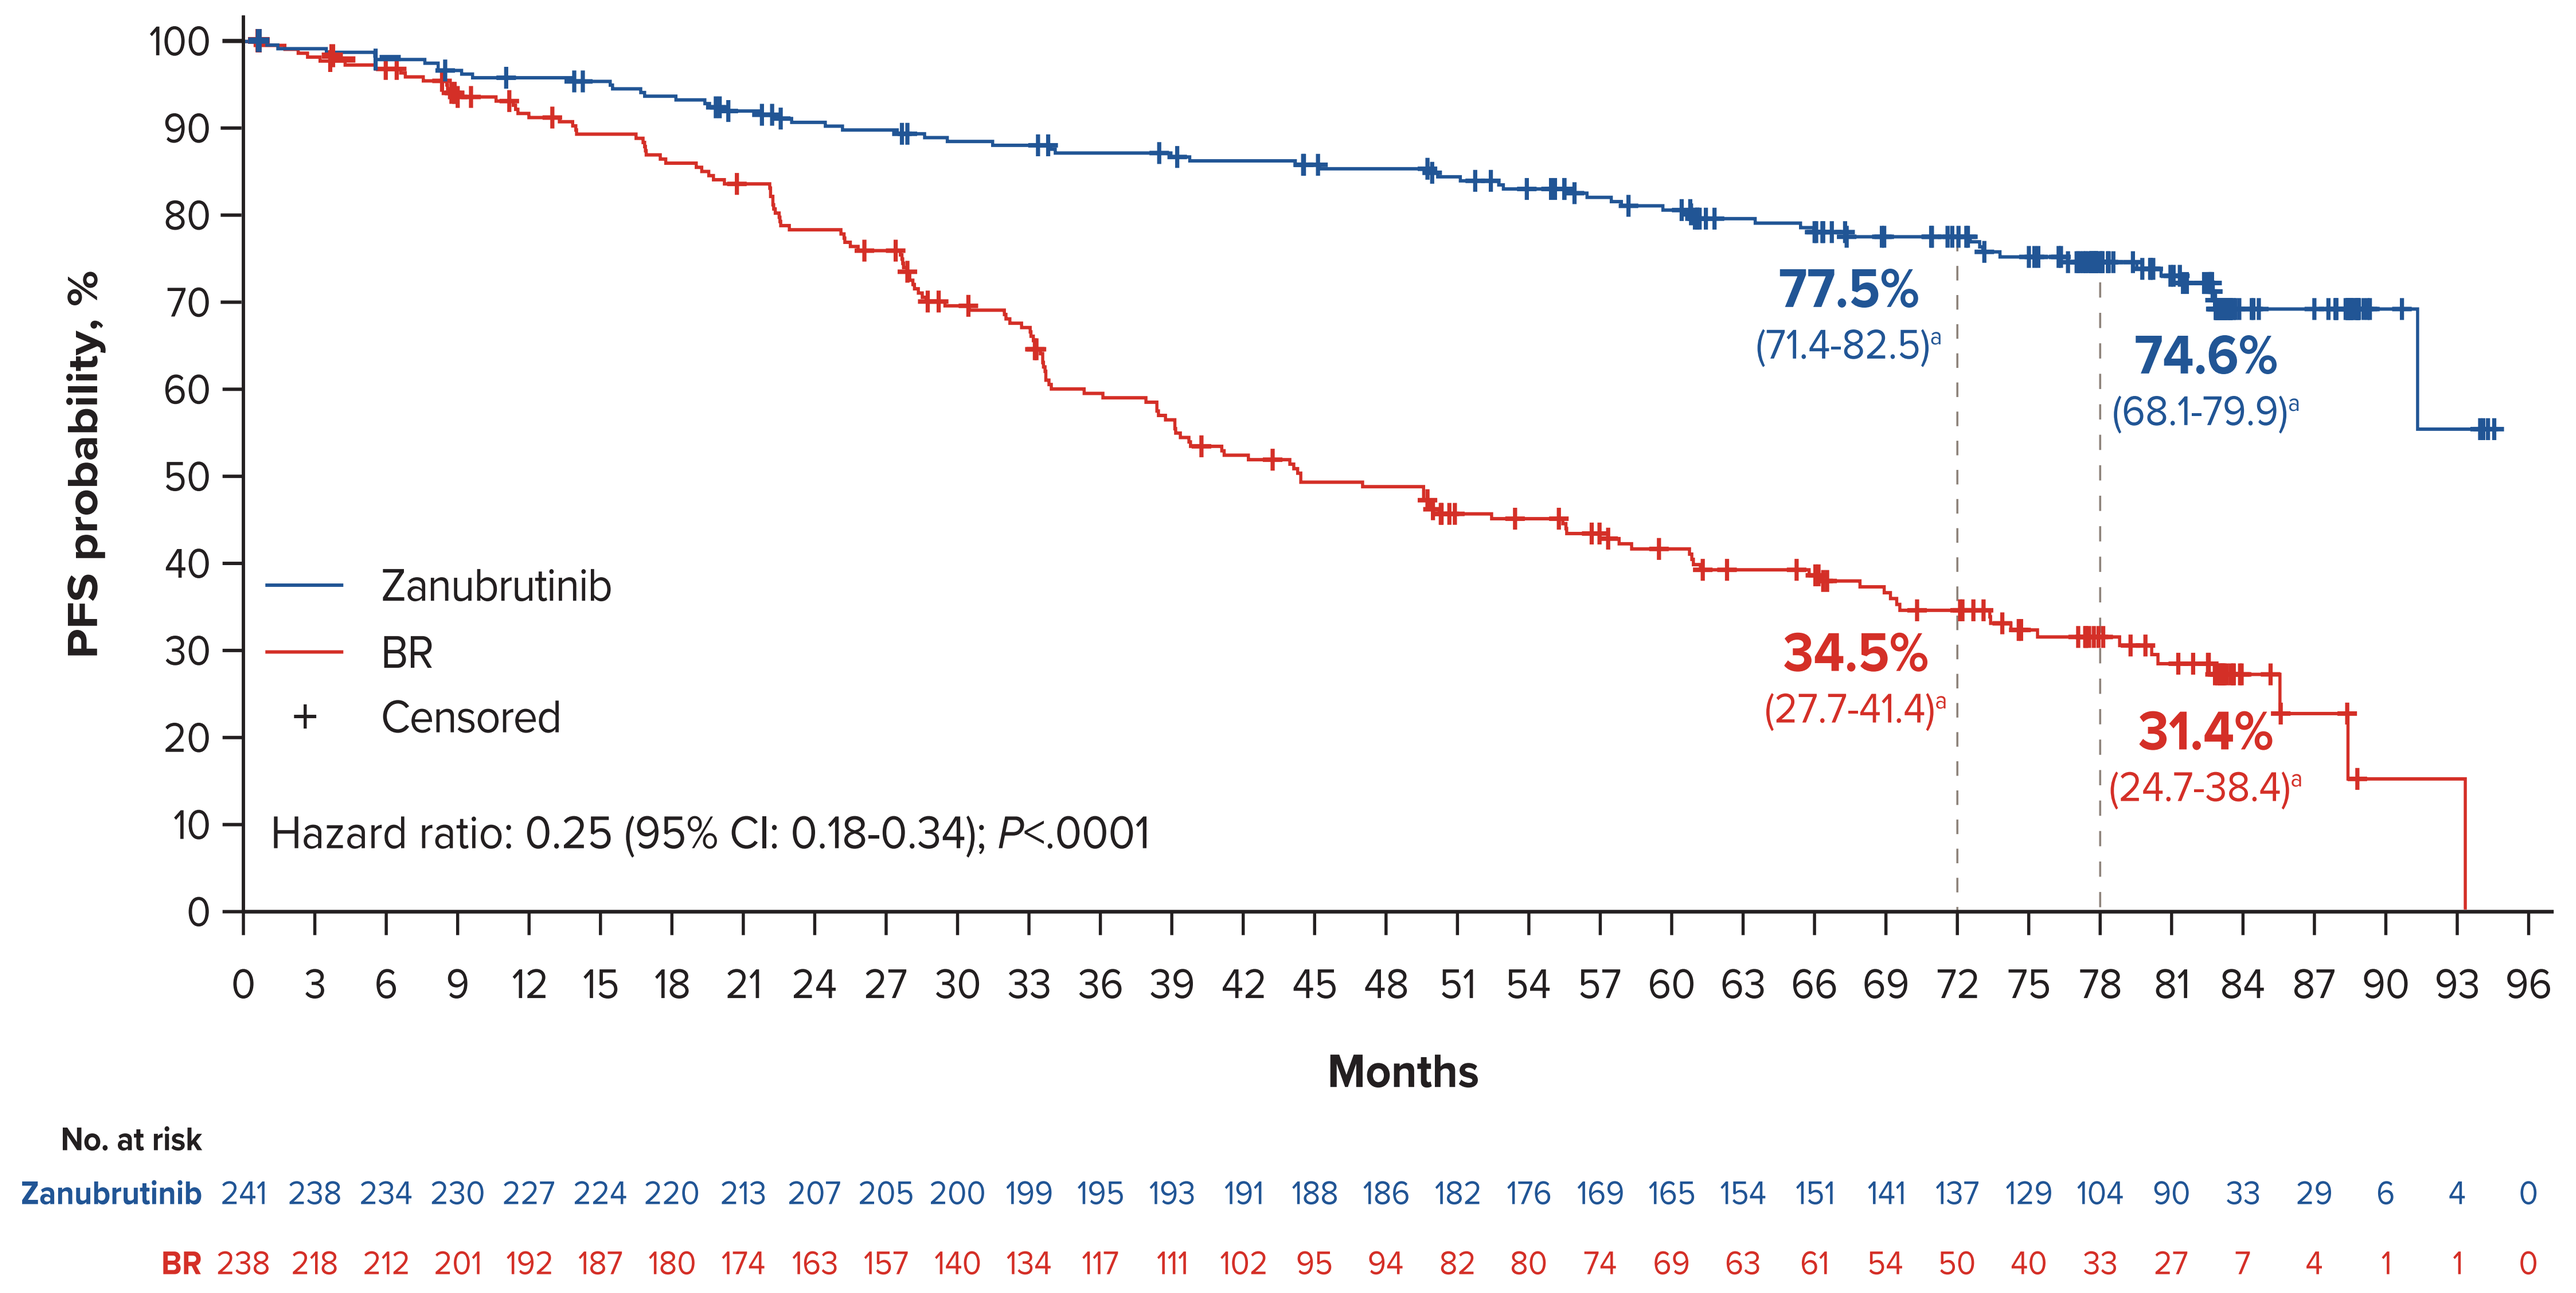
PFS probability, %
100
90
80
70
60
50
40
30
20
10
0
Zanubrutinib
BR
+ Censored
Hazard ratio: 0.25 (95% CI: 0.18-0.34); *P*<.0001
77.5%
(71.4-82.5)[a]
74.6%
(68.1-79.9)[a]
34.5%
(27.7-41.4)[a]
31.4%
(24.7-38.4)[a]
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45 48 51 54 57 60 63 66 69 72 75 78 81 84 87 90 93 96
Months
No. at risk
Zanubrutinib 241 238 234 230 227 224 220 213 207 205 200 199 195 193 191 188 186 182 176 169 165 154 151 141 137 129 104 90 33 29 6 4 0
BR 238 218 212 201 192 187 180 174 163 157 140 134 117 111 102 95 94 82 80 74 69 63 61 54 50 40 33 27 7 4 1 1 0

[a]95% CI values.
Abbreviation: BR, bendamustine and rituximab.

PFS by IGHV Status and Other Patient Subgroups

- Zanubrutinib demonstrated consistent PFS benefit over BR, regardless of IGHV status; in patients with IGHV-unmutated disease, zanubrutinib showed superior PFS compared with BR (HR, 0.25; *P*<.0001) and in IGHV-mutated disease similar results were seen with zanubrutinib vs BR (HR, 0.30; *P*<.0001) **(Supplementary Figure S2)**
 - Consistent results were seen after adjustment for COVID-19 (**Figure 3**)
- In addition, PFS superiority was consistent across all demographics and age groups (<65, ≥65 and <75, ≥75), as well as disease characteristics **(Supplementary Figure S3)**

Figure 3. COVID-Adjusted PFS by IGHV status

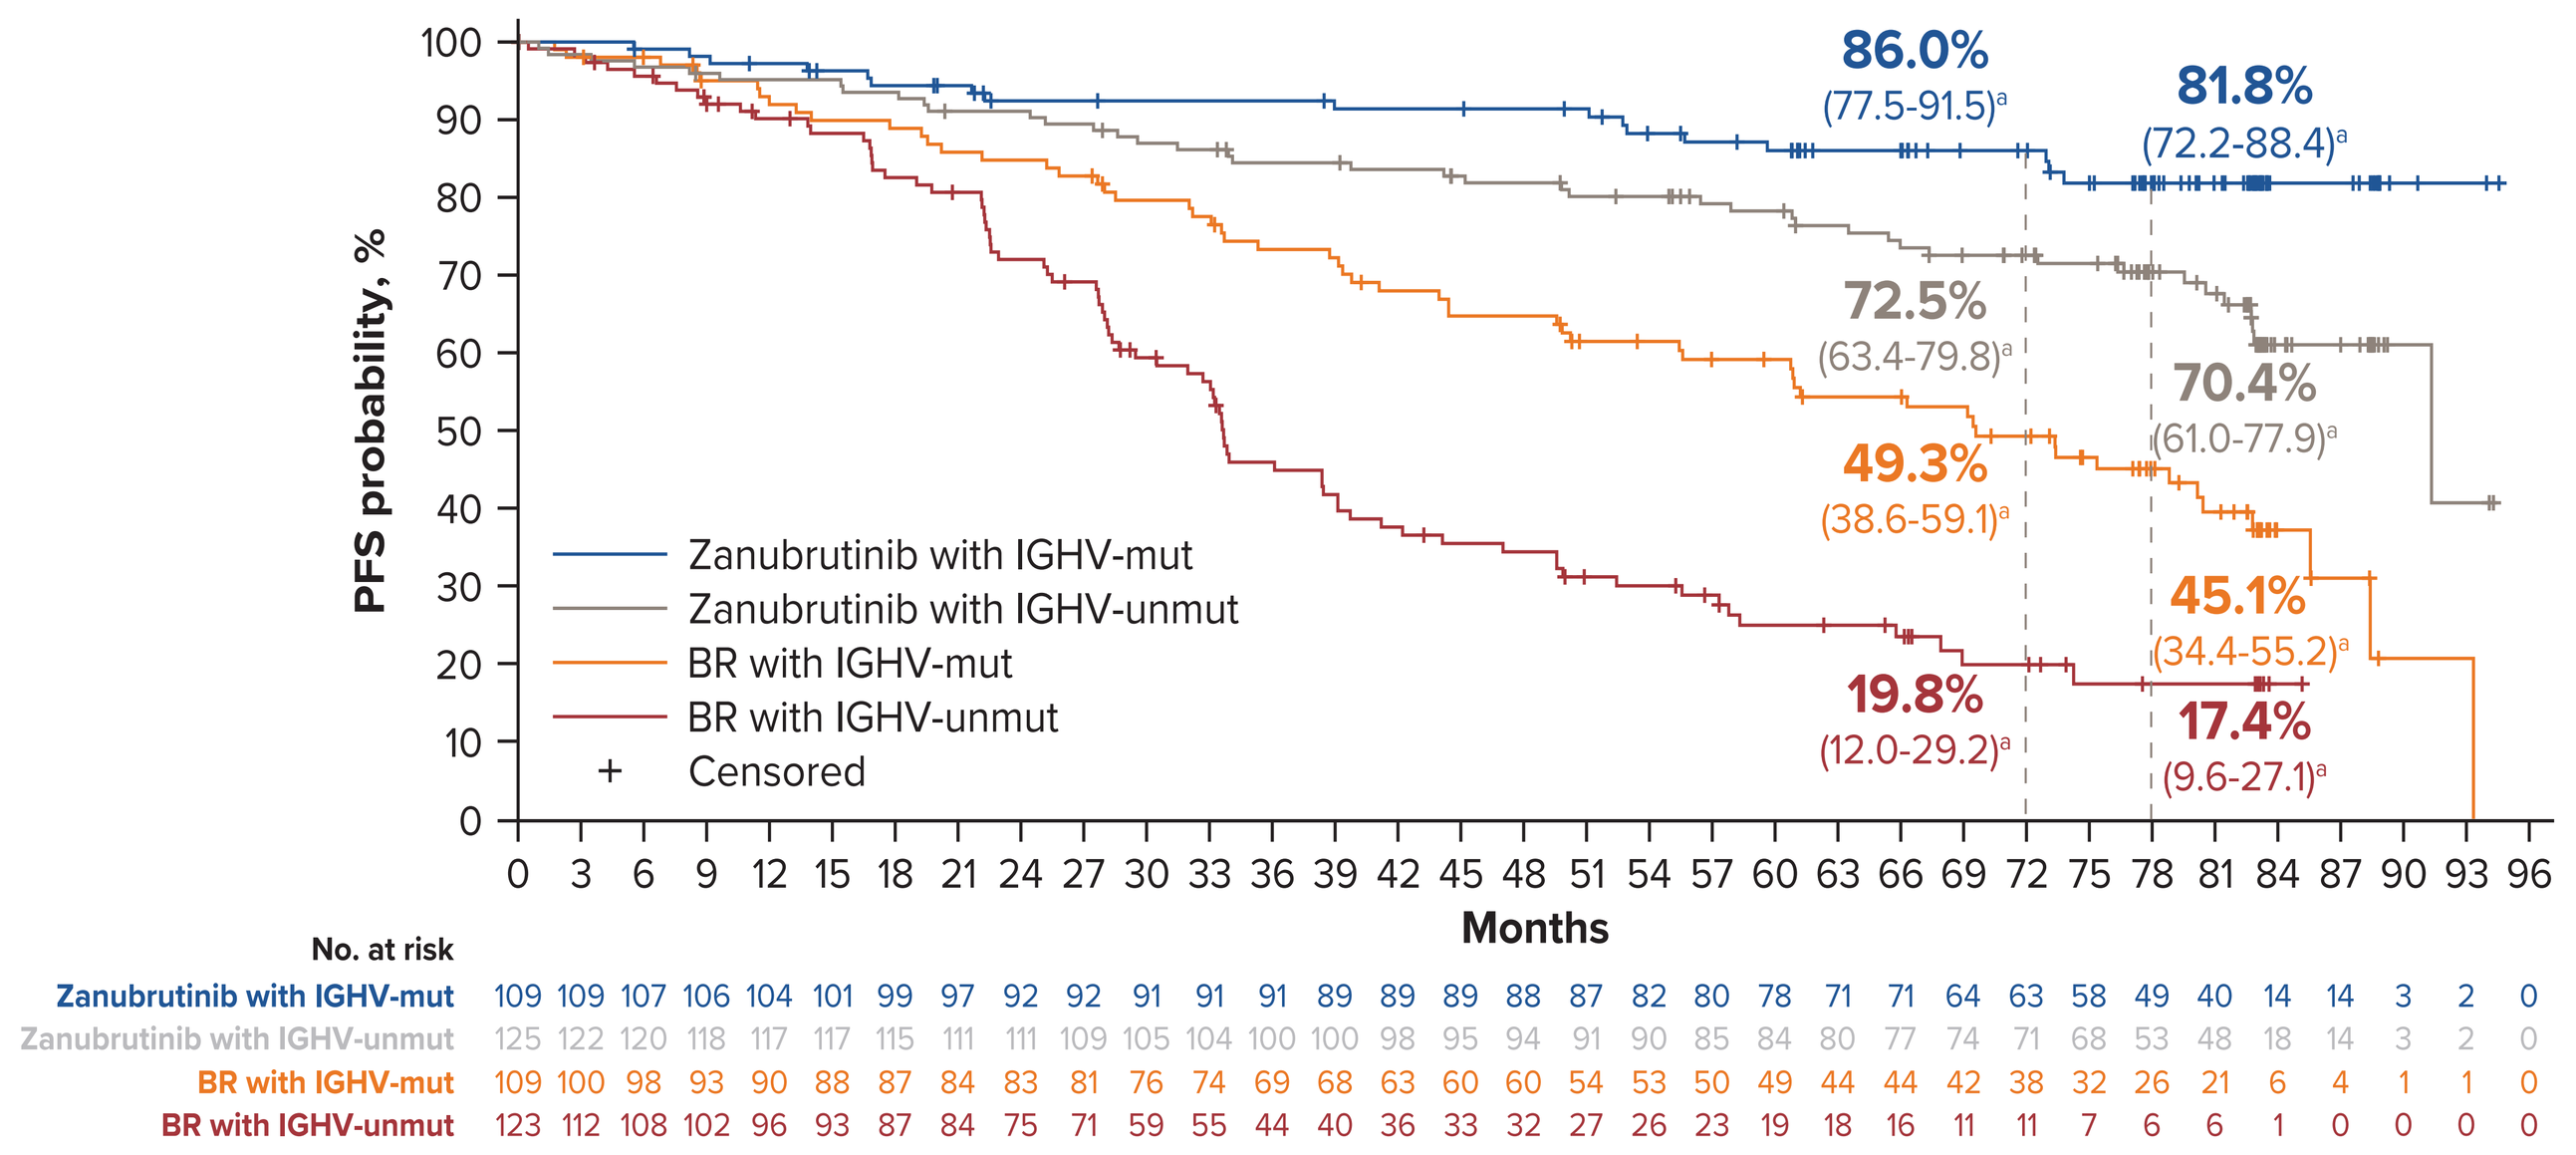
PFS probability, %
100
90
80
70
60
50
40
30
20
10
0
Zanubrutinib with IGHV-mut
Zanubrutinib with IGHV-unmut
BR with IGHV-mut
BR with IGHV-unmut
+ Censored
86.0%
(77.5-91.5)[a]
81.8%
(72.2-88.4)[a]
72.5%
(63.4-79.8)[a]
70.4%
(61.0-77.9)[a]
49.3%
(38.6-59.1)[a]
45.1%
(34.4-55.2)[a]
19.8%
(12.0-29.2)[a]
17.4%
(9.6-27.1)[a]
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45 48 51 54 57 60 63 66 69 72 75 78 81 84 87 90 93 96
Months
No. at risk
Zanubrutinib with IGHV-mut 109 109 107 106 104 101 99 97 92 92 91 91 91 89 89 89 88 87 82 80 78 71 71 64 63 58 49 40 14 14 3 2 0
Zanubrutinib with IGHV-unmut 125 122 120 118 117 117 115 111 111 109 105 104 100 100 98 95 94 91 90 85 84 80 77 74 71 68 53 48 18 14 3 2 0
BR with IGHV-mut 109 100 98 93 90 88 87 84 83 81 76 74 69 68 63 60 60 54 53 50 49 44 44 42 38 32 26 21 6 4 1 1 0
BR with IGHV-unmut 123 112 108 102 96 93 87 84 75 71 59 55 44 40 36 33 32 27 26 23 19 18 16 11 11 7 6 6 1 0 0 0 0

[a]95% CI values.
Abbreviation: BR, bendamustine and rituximab.

TTNT, TTNT-D and PFS2

- TTNT significantly favored zanubrutinib over BR (HR, 0.24; 95% CI, 0.16-0.35; *P*<.0001); 78-month TTNT rates were 87.0% (95% CI, 81.7-90.9) vs 53.0% (95% CI, 45.4-60.0), respectively (**Figure 4A**)
- TTNT-D significantly favored zanubrutinib over BR (HR, 0.37; 95% CI, 0.27-0.50; *P*<.0001); 78-month TTNT-D rates were 73.7% (95% CI, 67.5-78.9) vs 42.2% (95% CI, 35.4-48.9), respectively
- PFS2 favored zanubrutinib over BR (HR, 0.70; 95% CI, 0.47-1.02), with 78-month PFS2 estimates of 81.3% (95% CI, 75.6-85.8) vs 74.4% (95% CI, 67.8-79.8), respectively (**Figure 4B**); COVID-19–adjusted 78-month PFS2 estimates were 84.7% (95% CI, 79.2-88.8) and 76.4% (95% CI, 69.9-81.7), respectively

Figure 4. TTNT and PFS2

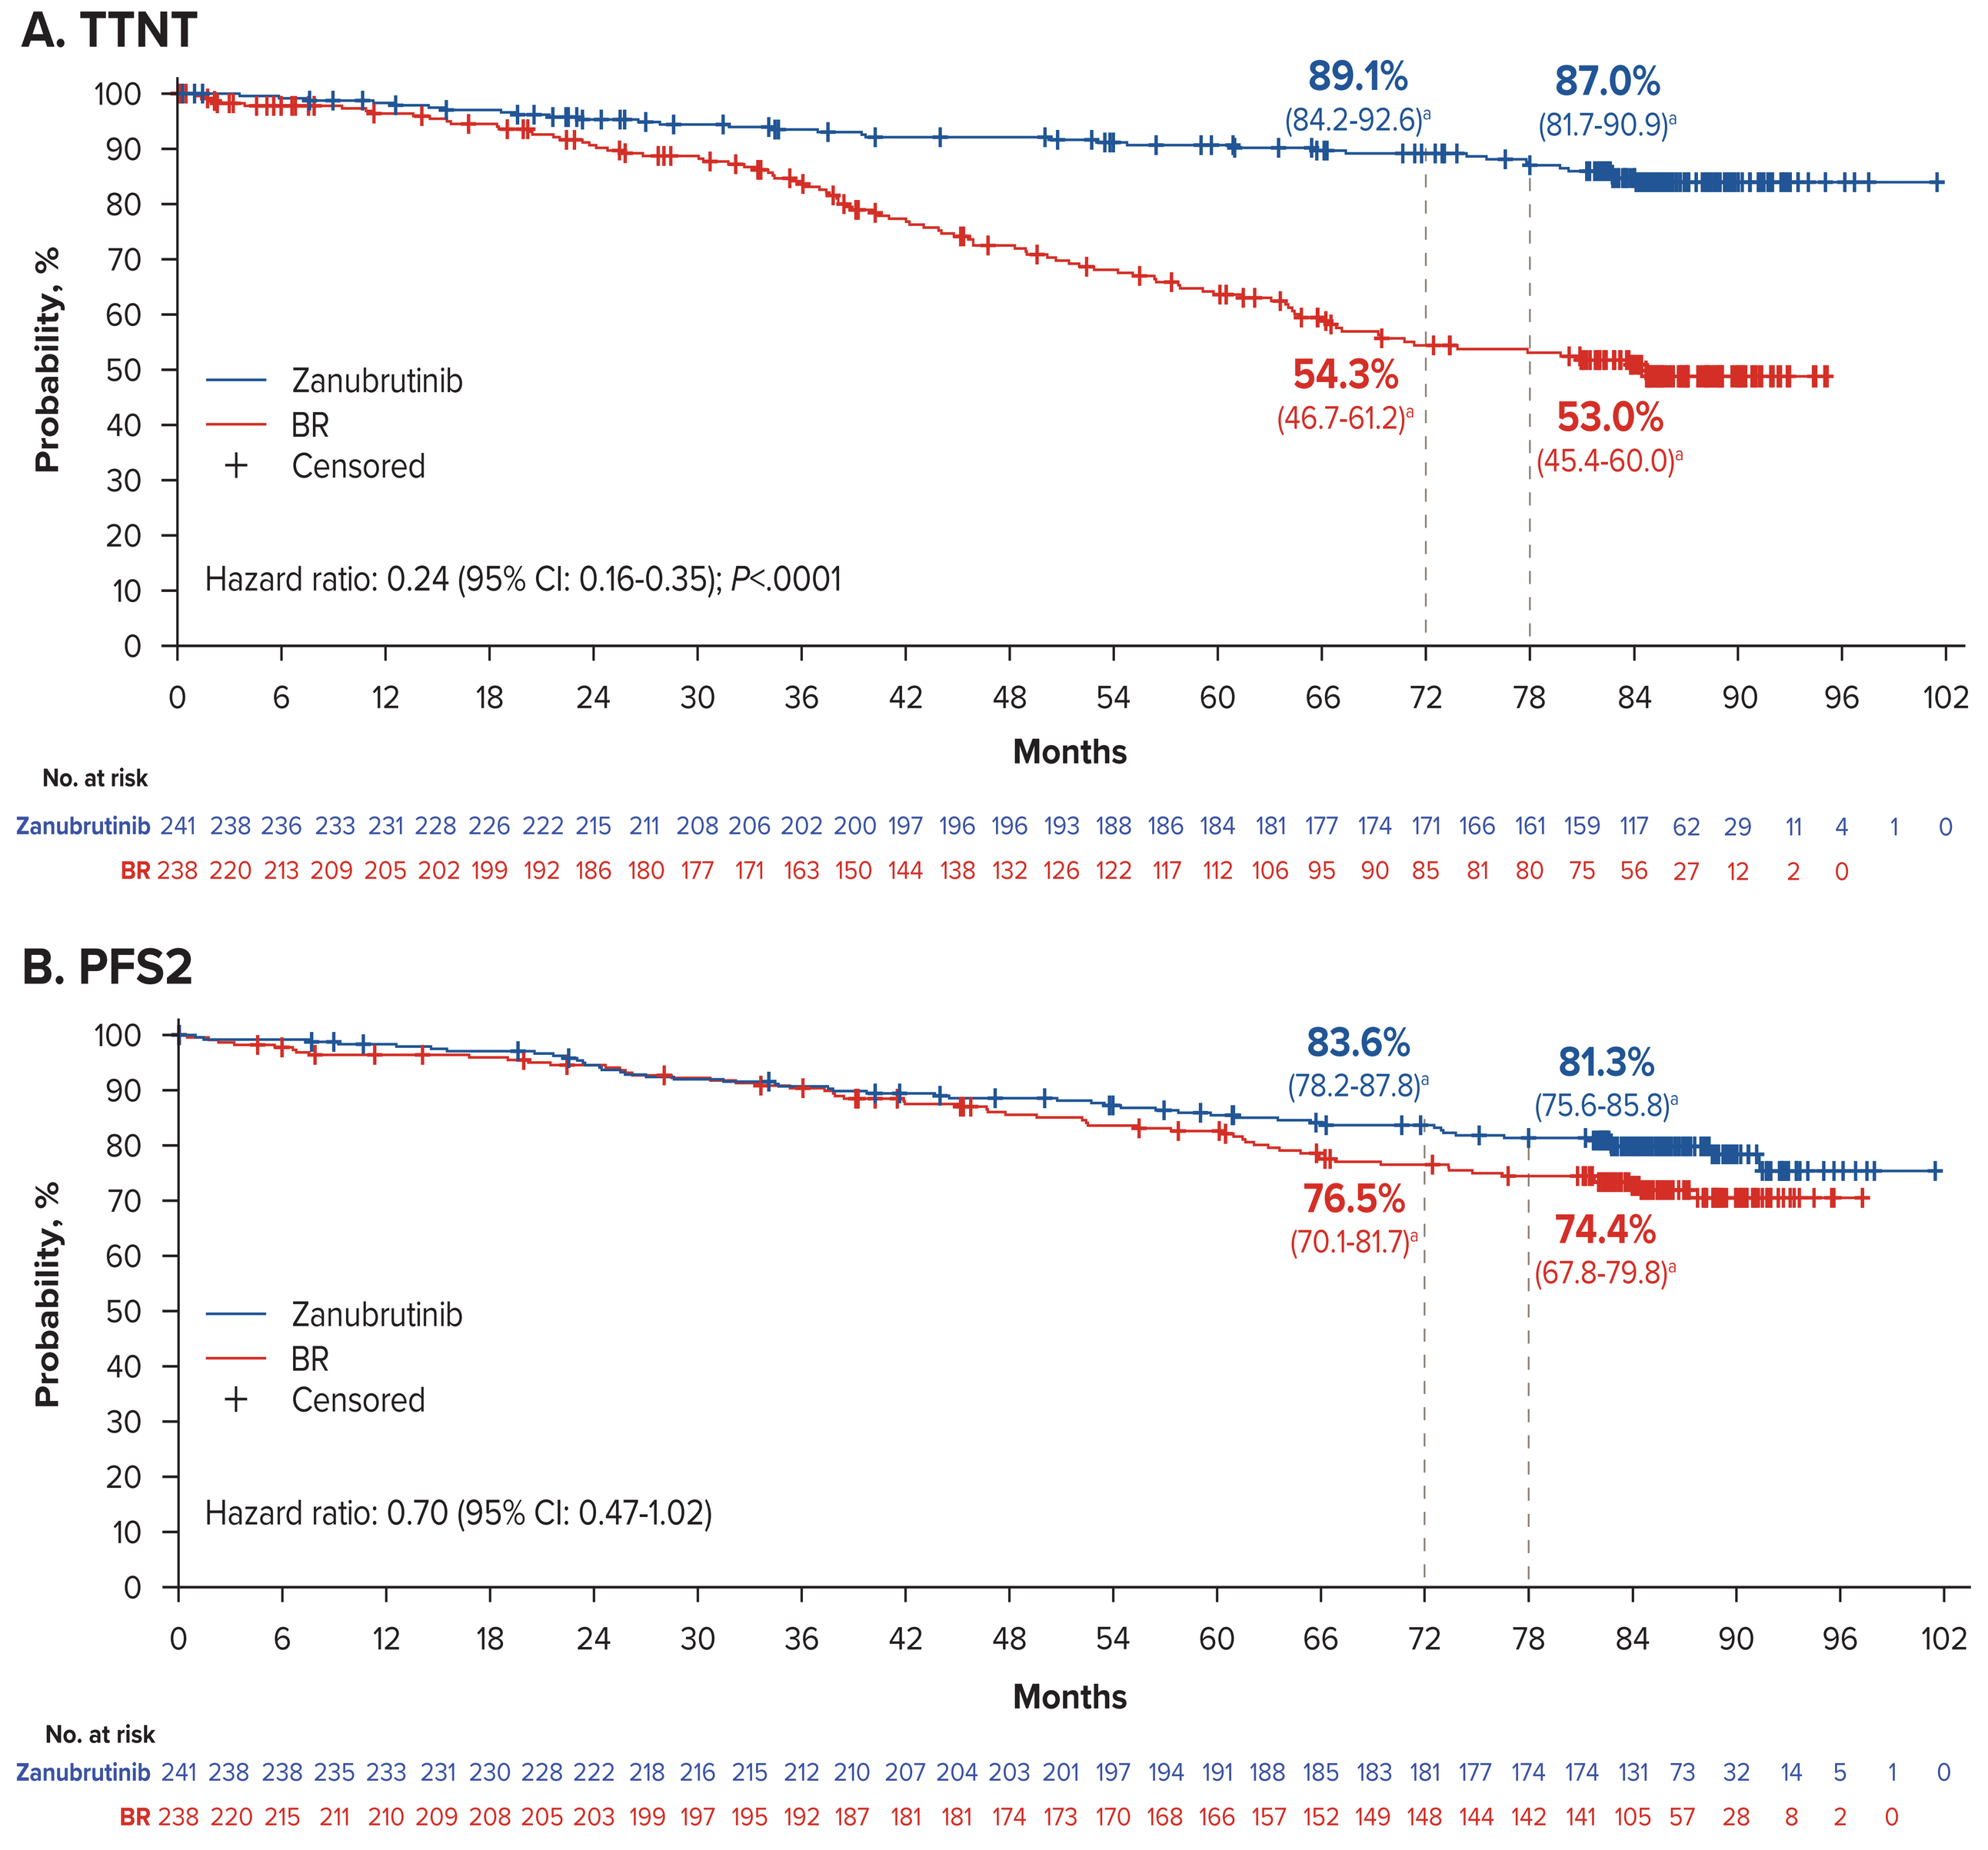
A. TTNT
Probability, %
Zanubrutinib
BR
+ Censored
Hazard ratio: 0.24 (95% CI: 0.16-0.35); *P*<.0001
89.1%
(84.2-92.6)[a]
87.0%
(81.7-90.9)[a]
54.3%
(46.7-61.2)[a]
53.0%
(45.4-60.0)[a]
0 6 12 18 24 30 36 42 48 54 60 66 72 78 84 90 96 102
Months
No. at risk
Zanubrutinib 241 238 236 233 231 228 226 222 215 211 208 206 202 200 197 196 196 193 188 186 184 181 177 174 171 166 161 159 117 62 29 11 4 1 0
BR 238 220 213 209 205 202 199 192 186 180 177 171 163 150 144 138 132 126 122 117 112 106 95 90 85 81 80 75 56 27 12 2 0
B. PFS2
Probability, %
Zanubrutinib
BR
+ Censored
Hazard ratio: 0.70 (95% CI: 0.47-1.02)
83.6%
(78.2-87.8)[a]
81.3%
(75.6-85.8)[a]
76.5%
(70.1-81.7)[a]
74.4%
(67.8-79.8)[a]
0 6 12 18 24 30 36 42 48 54 60 66 72 78 84 90 96 102
Months
No. at risk
Zanubrutinib 241 238 238 235 233 231 230 228 222 218 216 215 212 210 207 204 203 201 197 194 191 188 185 183 181 177 174 174 131 73 32 14 5 1 0
BR 238 220 215 211 210 209 208 205 203 199 197 195 192 187 181 181 174 173 170 168 166 157 152 149 148 144 142 141 105 57 28 8 2 0

[a]95% CI values.
Abbreviation: BR, bendamustine and rituximab.

OS

- Zanubrutinib demonstrated numerical OS benefit vs BR (HR, 0.84; 95% CI, 0.56-1.25; *P*=.1916); COVID-19–adjusted values were similar (HR, 0.74; 95% CI, 0.48-1.15; *P*=.0918)

Subsequent Therapy for CLL

- Overall, 26/241 zanubrutinib-treated patients (10.8%) and 88/238 BR-treated patients (37.0%) had progressive disease after study treatment and received subsequent therapy
 - Of the 26 zanubrutinib-treated patients receiving subsequent therapy, 50.0% received B-cell lymphoma 2 inhibitor (BCL2i)-based regimens (**Figure 5A; Supplementary Table S1**)
 - Of the 88 BR-treated patients receiving subsequent therapy, most patients (89.8%) received BTK inhibitor-based therapy (including 76 patients who received subsequent zanubrutinib) (**Figure 5B; Supplementary Table S1**)
 - In the zanubrutinib-treated patients who received BCL2i-based regimens (n=13), median follow-up was 38.9 months from the initiation of next-line therapy; among these patients, 9 (69.2%) remained alive and without progressive disease, three died (23.1%), and one had progressive disease (7.7%) (**Figure 5C**)

Figure 5. Subsequent CLL Therapy after First Progression

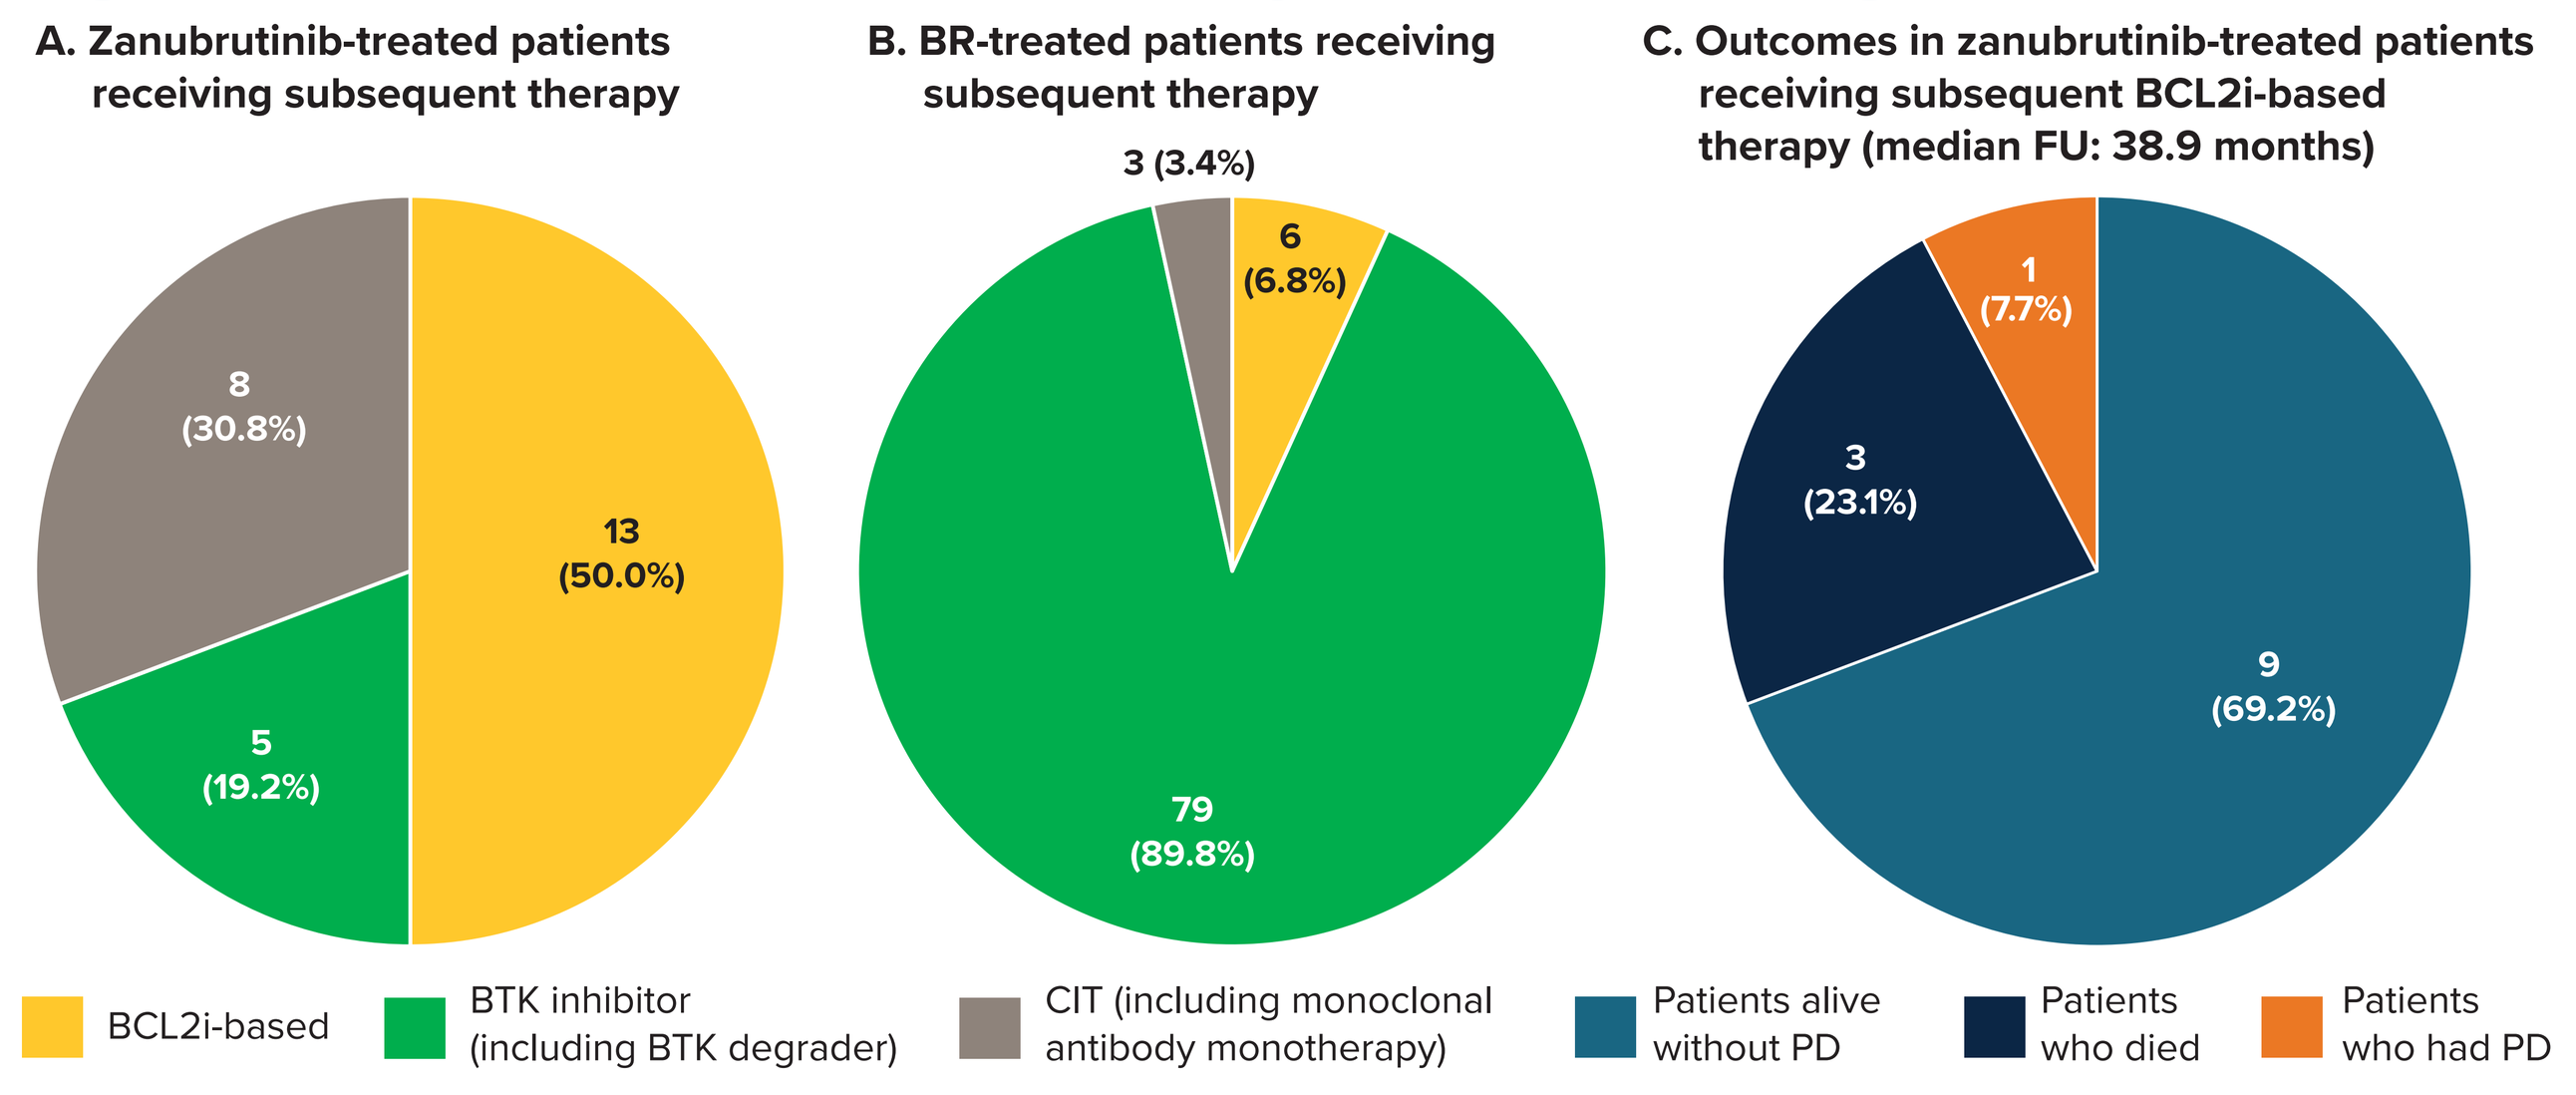
A. Zanubrutinib-treated patients receiving subsequent therapy
13
(50.0%)
5
(19.2%)
8
(30.8%)
B. BR-treated patients receiving subsequent therapy
3 (3.4%)
6
(6.8%)
79
(89.8%)
C. Outcomes in zanubrutinib-treated patients receiving subsequent BCL2i-based therapy (median FU: 38.9 months)
1
(7.7%)
3
(23.1%)
9
(69.2%)
BCL2i-based
BTK inhibitor (including BTK degrader)
CIT (including monoclonal antibody monotherapy)
Patients alive without PD
Patients who died
Patients who had PD

Abbreviations: CIT, chemoimmunotherapy; FU, follow-up; PD, progressive disease.

Safety

- All grade treatment-emergent adverse events (TEAEs) occurred in 233 (97.1%) and 214 (94.3%) of zanubrutinib-treated patients and BR-treated patients, respectively; Grade ≥3 TEAEs occurred in 176 (73.3%) and 169 (74.4%) patients, respectively
 - Treatment-emergent and post-treatment adverse events of special interest (≥15% of patients) are shown in (**Supplementary Table S2**)
- TEAEs led to death in 25 (10.4%) (zanubrutinib) and 7 (3.1%) (BR) patients
- The exposure-adjusted incidence rate (person per 100 persons-months) for selected treatment-emergent and post-treatment AEs are shown in **Table 2**

Table 2. Treatment-Emergent and Post-Treatment AEs

EAIRs, persons per 100 person-months[a]	Arm A: zanubrutinib n=240	Arm B: BR n=227
Anemia	0.17	0.56
Atrial fibrillation and flutter	0.16	0.10
Hemorrhage	1.49	0.33
Major hemorrhage	0.16	0.05
Hypertension	0.44	0.34
Infections	3.37	3.27
Neutropenia	0.31	2.79
Second primary malignancies	0.46	0.40
Thrombocytopenia	0.11	0.46

[a]EAIRs were calculated as the number of patients with an event in each TEAE category divided by the total time from the first dose date to the first event date or the exposure time if no event occurred.
Abbreviations: BR, bendamustine and rituximab; EAIR, exposure-adjusted incidence rate; TEAE, treatment-emergent adverse event.

REFERENCES

1. Guo Y, et al. *J Med Chem.* 2019;62(17):7923-7940.
2. Tam CS, et al. *Expert Rev Clin Pharmacol.* 2021;14(11):1329-1344.
3. Brown JR, et al. *N Engl J Med.* 2023;388(4):319-332.
4. Tam CS, et al. *Lancet Oncol.* 2022;23(8):1031-1043.
5. Shadman M, et al. *J Clin Oncol.* 2025;43(7):780-787.
6. Tam CS, et al. *Haematologica.* 2021;106(9):2354-2363.
7. Tam CS, et al. ASH 2025. Poster #2129.
8. Tedeschi A, et al. iwCLL 2025. Poster #1421.

ACKNOWLEDGMENTS

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This study was sponsored by BeOne Medicines, Ltd. Medical writing support was provided by Manoshi Nath, MSc, of Nucleus Global, an Inizio company, and supported by BeOne Medicines.

DISCLOSURES

CST: Honoraria: BeOne Medicines, Ltd, Janssen, AbbVie, AstraZeneca, Gilead, Lilly, Merck; Research funding: BeOne Medicines, Ltd, Janssen, AbbVie. **BSK:** Consulting or advisory role: AbbVie, BeOne Medicines, Ltd, AstraZeneca, Roche/Genentech, ADC Therapeutics, Incyte, Lilly, Merck, Pfizer, GSK; Research funding: BeOne Medicines, Ltd, Roche/Genentech (payable to institution). **TM:** Honoraria: BeOne Medicines, Ltd, AstraZeneca, Sobi, Roche, Janssen, AbbVie, Lilly; Consultant: AbbVie, BeOne Medicines, Ltd, Sobi, Alexion, Novartis, Janssen, AstraZeneca, Lilly, Roche; Research grants: Janssen, AbbVie; Travel, accommodations, or expenses: Alexion, BeOne Medicines, Ltd, AbbVie, Janssen, AstraZeneca; Advisory board: AbbVie, BeOne Medicines, Ltd, AstraZeneca, Janssen. **TR:** Honoraria and research funding: BeOne Medicines, Ltd, Johnson & Johnson, AbbVie, GSK, Takeda, Regeneron; Consulting or advisory role: Johnson & Johnson, AbbVie, BeOne Medicines, Ltd, AstraZeneca, Gilead, Octapharma; Travel, accommodations, expenses: BeOne Medicines, Ltd, AstraZeneca, Johnson & Johnson. **JRB:** Consultant: AbbVie, Acerta/AstraZeneca, Alloplex Biotherapeutics, BeOne Medicines, Ltd, BMS, EcoR1, Galapagos NV, Genentech/Roche, Grifols Worldwide Operations, InnoCare Pharma, Lilly, Magnet Biomedicine, Merck, Nurix Therapeutics, Pharmacyclics; Research funding: BeOne Medicines, Ltd, Gilead, iOnctura, Lilly, MEI Pharma, Nagoon Therapeutics; Royalties: UpToDate; Data Safety Monitoring Board for Grifols Therapeutics. **PG:** Honoraria, consulting or advisory role and research funding: AbbVie, AstraZeneca, BeOne Medicines, Ltd, BMS, Johnson & Johnson, Lilly, MSD, Roche. **SSO:** Honoraria: AbbVie, AstraZeneca, BeOne Medicines, Ltd, Gilead, Janssen; Consulting or advisory role: AbbVie, AstraZeneca, BeOne Medicines, Ltd, Janssen; Research funding (payable to institution): AbbVie, AstraZeneca, BeOne Medicines, Ltd, Gilead, Janssen, Novartis, Pharmacyclics, Roche, Takeda; Other relationship (member of safety and data monitoring committee): Merck. **PAW:** No disclosures. **ML:** Consulting or advisory role: Janssen, BeOne Medicines, Ltd, Sobi; Speakers bureau: Janssen, BeOne Medicines, Ltd, AbbVie. **IWF:** Employment: Tennessee Oncology; Board of Directors or advisory committee: Vincerx; Consulting or advisory role (all payments made to physician): AbbVie, BeOne Medicines, Ltd, Genentech, Genmab, Kite Pharma, Vincerx; Research funding (all payments made to institution): AbbVie, AstraZeneca, BeOne Medicines, Ltd, BMS, Celgene, City of Hope National Medical Center, Epizyme, Fate Therapeutics, Genentech, Gilead Sciences, IGM Biosciences, InnoCare Pharma, Incyte, Janssen, Kite Pharma Pharma, Lilly, Marker Therapeutics, Merck, MorphoSys, Myeloid Therapeutics, Novartis, Nurix, Pfizer, Roche, Seattle Genetics, TG Therapeutics, Vincerx Pharma, 2seventy bio. **WJ:** Research funding: AbbVie, AstraZeneca, BeOne Medicines, Ltd, Lilly. **JS:** Employment and may own stock: BeOne Medicines, Ltd. **ML:** Employment: BeOne Medicines, Ltd (Feb 2025 onwards), Roche (until 2025); Stock or other ownership: BeOne Medicines, Ltd; Roche. **JH:** Employment: BeOne Medicines, Ltd (currently), Genentech; Stocks or other ownership: BeOne Medicines, Ltd, Roche. **MS:** Consulting, advisory boards, steering committees, or data safety monitoring committees: AbbVie, Ascentage, Genentech, AstraZeneca, Genmab, Janssen, BeOne Medicines, Ltd, BMS, Morphosys/Incyte, Kite Pharma, Lilly, Fate Therapeutics, Nurix, Merck, Pfizer, Pierre Fabre; Research funding: Genentech, BeOne Medicines, Ltd, AstraZeneca, Genmab, Morphosys/Incyte, Sana Biotechnology, Nurix, Merck, Janssen; Employment (spouse): BMS; Stock options: Koi Biotherapeutics.

CORRESPONDENCE: Constantine S. Tam, constantine.tam@alfred.org.au

Presented at the EHA2026 Congress; June 11-14, 2026; Stockholm, Sweden
Data originally presented at the 2026 ASCO Annual Meeting; May 29-June 2, 2026; Chicago, IL, USA. Abstract 7046