RATIONALE-306 Subgroup Analysis of Patients With Advanced/Metastatic Esophageal Squamous Cell Cancer and Tumor Area Positivity Score ≥5%

395P

David Tougeron,[1*] Jianming Xu,[2] Eric Raymond,[3] Richard Hubner,[4] Ken Kato,[5] Lucjan Wyrwicz,[6] Eric Van Cutsem,[7] Sook Ryun Park,[8] Paula Jimenez Fonseca,[9] Takashi Kojima,[10] Ryu Ishihara,[11] Antonio Avallone,[12] Roberto Pazo-Cid,[13] Kaijun Wang,[14] Sebastian Yan,[15] Jingwen Shi,[15] Xuan Kong,[16] Harry H. Yoon,[17] Florian Lordick[18]

[1]Department of Hepato-Gastroenterology, CHU de Poitiers, Poitiers, France; [2]Department of Gastrointestinal Oncology, Fifth Medical Center, Chinese PLA General Hospital, Beijing, China; [3]Department of Medical Oncology, Paris Saint-Joseph Hospital, Paris, France; [4]Department of Medical Oncology, The Christie NHS Foundation Trust, and Division of Cancer Sciences, University of Manchester, Manchester, UK; [5]Department of Head and Neck, Esophageal Medical Oncology, National Cancer Center Hospital, Tokyo, Japan; [6]Department of Oncology and Radiotherapy, Maria Sklodowska-Curie National Cancer Research Institute, Warsaw, Poland; [7]Division of Digestive Oncology, University Hospitals Gasthuisberg, Leuven, and KU Leuven, Leuven, Belgium; [8]Department of Oncology, Asan Medical Center, University of Ulsan College of Medicine, Seoul, Republic of Korea; [9]Department of Medical Oncology, Central University Hospital of Asturias, ISPA, Oviedo, Spain; [10]Department of Gastrointestinal Oncology, National Cancer Center Hospital East, Chiba, Japan; [11]Department of Gastrointestinal Oncology, Osaka International Cancer Institute, Osaka, Japan; [12]Department of Medical Oncology, National Cancer Institute IRCCS G. Pascale Foundation, Naples, Italy; [13]Department of Medical Oncology, Hospital Universitario Miguel Servet, Zaragoza, Spain; [14]Global Statistics and Data Science, BeOne Medicines, Ltd., San Carlos, CA, USA; [15]Clinical Development, BeOne Medicines, Ltd., Beijing, China; [16]Clinical Biomarker, BeOne Medicines, Ltd., Shanghai, China; [17]Department of Oncology, Mayo Clinic, Rochester, MN, USA; [18]Department of Medicine II (Oncology, Gastroenterology, Hepatology, Pulmonology, and Infectious Diseases), Leipzig University Medical Center, and University Cancer Center Leipzig (UCCL), Leipzig, Germany. *Lead and presenting author.

CONCLUSIONS

- The median overall survival (OS) of 19.1 months with tislelizumab plus chemotherapy in the tumor programmed death-ligand 1 (PD-L1) ≥5% population sets a new bar for efficacy in this group of patients with advanced/metastatic esophageal squamous cell carcinoma (ESCC) and should be considered in shared decision-making
- Efficacy benefits and safety outcomes remained consistent with primary analysis and 3-year follow-up data, showing sustained improvement with no new safety signals

INTRODUCTION

- Tislelizumab, in combination with platinum-based chemotherapy, is approved by the European Medicines Agency for the first-line treatment of adults with unresectable, locally advanced or metastatic ESCC with tumor PD-L1 Tumor Area Positivity (TAP) score ≥5%[1]
- In the CheckMate 648 trial, patients with unresectable advanced, recurrent or metastatic ESCC and tumor PD-L1 combined positive score (CPS) ≥5 showed an improvement in OS with nivolumab plus chemotherapy vs chemotherapy alone (hazard ratio [HR]=0.78) after a 29-month minimum follow-up[2]
- The RATIONALE-306 trial demonstrated significant OS benefit for first-line treatment with tislelizumab plus chemotherapy compared with placebo plus chemotherapy in patients with advanced ESCC, both at primary analysis[3] and at the minimum 3-year follow-up[4]
 - At the 3-year follow-up, OS results showed a stratified HR of 0.70 for all patients in the intent-to-treat (ITT) population[4]
 - In patients with tumor PD-L1 TAP score ≥10% and ≥5%, the HRs were 0.70[4] and 0.62, respectively, at the 3-year follow-up
- Here, we report the study close-out analysis (August 22, 2024) of RATIONALE-306 in the subgroup with tumor PD-L1 TAP score ≥5%

METHODS

- RATIONALE-306 (NCT03783442) is a randomized, double-blind, global, phase 3 trial evaluating the efficacy and safety of tislelizumab plus chemotherapy, compared with placebo plus chemotherapy, as a first-line treatment for metastatic or unresectable ESCC (**Figure 1**)

Figure 1. Study Design

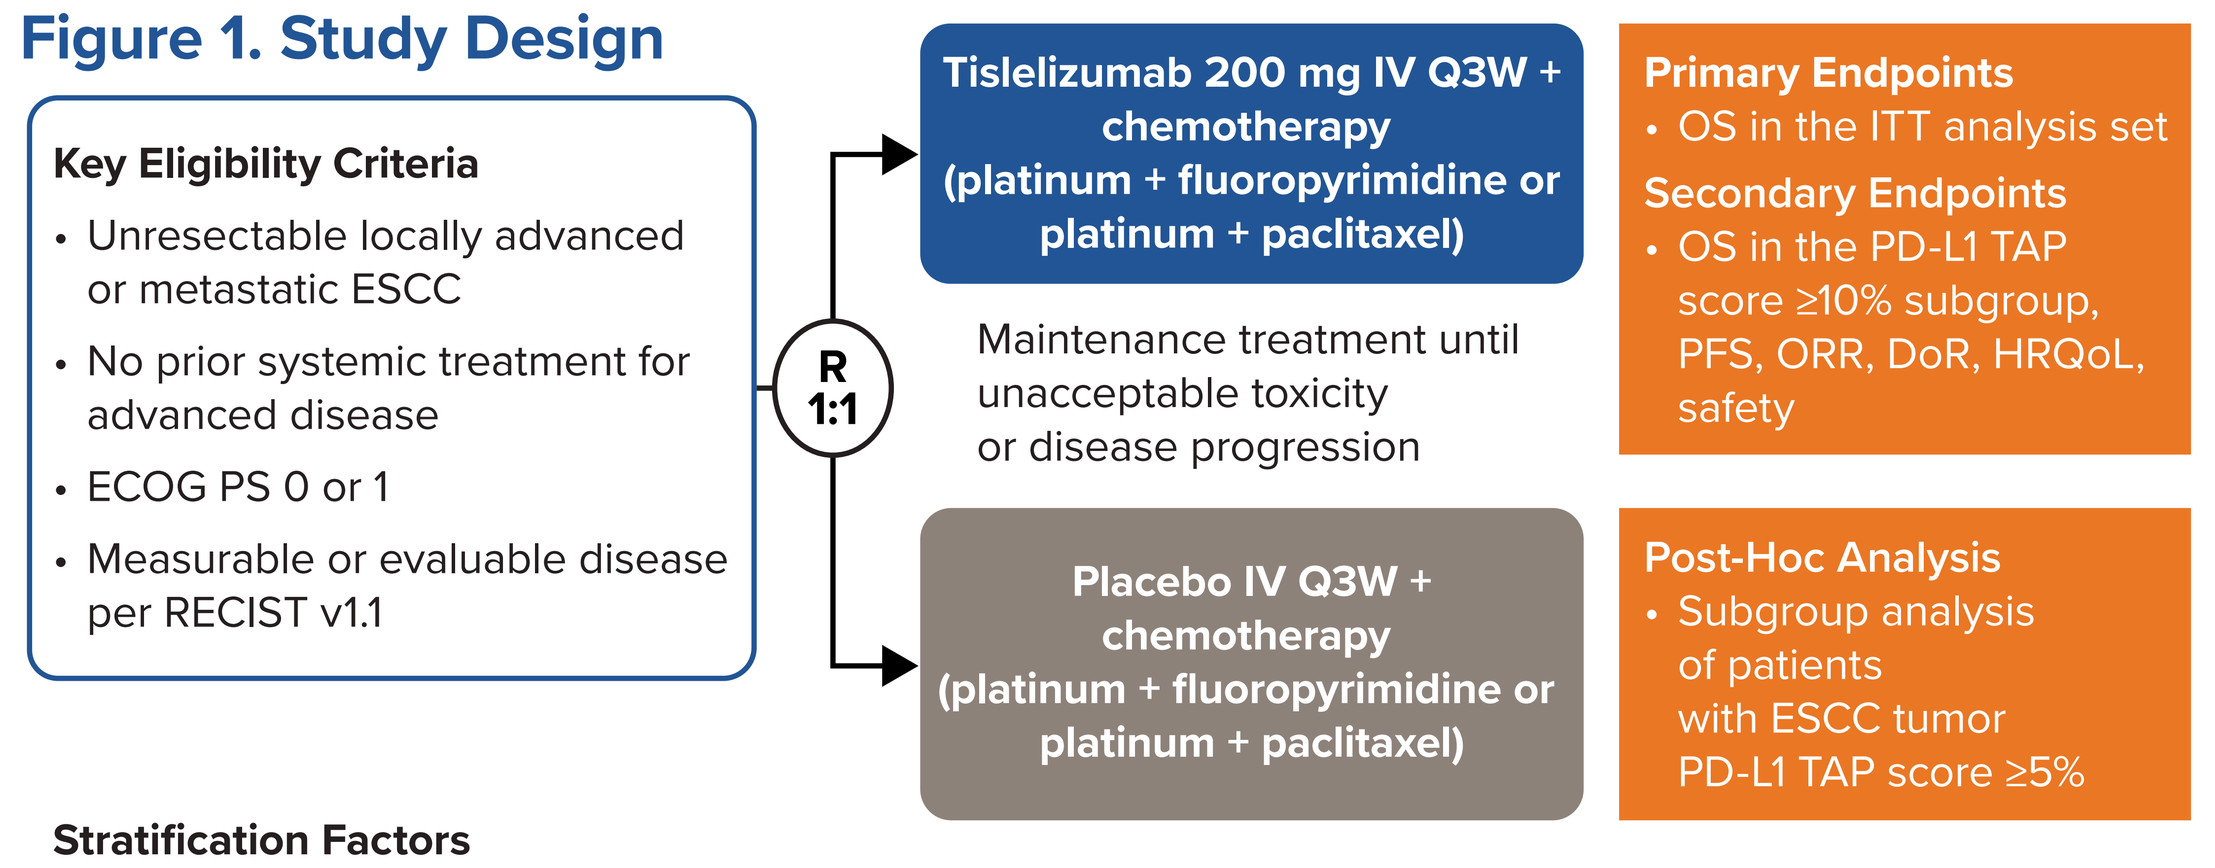
Key Eligibility Criteria
• Unresectable locally advanced or metastatic ESCC
• No prior systemic treatment for advanced disease
• ECOG PS 0 or 1
• Measurable or evaluable disease per RECIST v1.1
R 1:1
Tislelizumab 200 mg IV Q3W + chemotherapy (platinum + fluoropyrimidine or platinum + paclitaxel)
Maintenance treatment until unacceptable toxicity or disease progression
Placebo IV Q3W + chemotherapy (platinum + fluoropyrimidine or platinum + paclitaxel)
Primary Endpoints
• OS in the ITT analysis set
Secondary Endpoints
• OS in the PD-L1 TAP score ≥10% subgroup, PFS, ORR, DoR, HRQoL, safety
Post-Hoc Analysis
• Subgroup analysis of patients with ESCC tumor PD-L1 TAP score ≥5%

Stratification Factors
- Geographic region (Asia [excluding Japan] vs Japan vs rest of world)
- Prior definitive therapy (yes vs no)
- Investigator-chosen chemotherapy (platinum + fluoropyrimidine vs platinum + paclitaxel)

Abbreviations: DoR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; HRQoL, health-related quality of life; ITT, intent-to-treat; IV, intravenous; ORR, objective response rate; PFS, progression-free survival; Q3W, once every 3 weeks; R, randomized; RECIST, Response Evaluation Criteria in Solid Tumours.

METHODS (CONT.)

- Patients were enrolled regardless of their tumor PD-L1 expression at screening
 - Samples were stained for PD-L1 using the VENTANA PD-L1 (SP263) Assay (Roche) and expression was determined by TAP score
 - For exploratory purposes, pathologists in the central laboratory scored the same stained samples according to CPS

RESULTS

Patient Disposition and Baseline Characteristics

- Of 649 patients randomized (tislelizumab plus chemotherapy n=326; placebo plus chemotherapy n=323), 358 (55.2%) had a tumor PD-L1 TAP score ≥5% (tislelizumab plus chemotherapy n=172; placebo plus chemotherapy n=186) (**Table 1**)
- Baseline characteristics of patients with tumor PD-L1 TAP score ≥5% were consistent with the ITT population
- At data cutoff (August 22, 2024), the minimum study follow-up time was 45.2 months (range: 0.4-63.6)
- In the tislelizumab plus chemotherapy arm, 106 patients (61.6%) vs 126 (67.7%) in the placebo plus chemotherapy arm received post-treatment systemic therapy, of whom 27 (25.5%) vs 44 (34.9%), respectively, had systemic immunotherapy

Table 1. Patient Demographic and Baseline Characteristics

	Tislelizumab Plus Chemotherapy (n=172)	Placebo Plus Chemotherapy (n=186)
Median age, years (range)	62.0 (41-81)	64.0 (40-84)
≥65, n (%)	72 (41.9)	88 (47.3)
Male, n (%)	141 (82.0)	163 (87.6)
ECOG PS, n (%)		
0	50 (29.1)	57 (30.6)
1	122 (70.9)	129 (69.4)
Primary site of esophageal cancer, n (%)		
Cervical	9 (5.2)	9 (4.8)
Upper thoracic	30 (17.4)	38 (20.4)
Middle thoracic	74 (43.0)	78 (41.9)
Lower thoracic	59 (34.3)	61 (32.8)
Metastatic disease at study entry, n (%)	147 (85.5)	166 (89.2)
Number of metastatic sites at study entry, n (%)		
0	25 (14.5)	20 (10.8)
1	75 (43.6)	87 (46.8)
2	46 (26.7)	45 (24.2)
>2	26 (15.1)	34 (18.3)
Patients who received at least one prior definitive therapy, n (%)[a]	68 (39.5)	73 (39.2)
Prior anticancer systemic therapy, n (%)	46 (26.7)	59 (31.7)
Prior radiotherapy, n (%)	35 (20.3)	41 (22.0)
Prior anticancer surgery, n (%)	54 (31.4)	60 (32.3)

[a]A patient was counted only once within each category but may be counted in multiple categories.

Efficacy

- Clinically meaningful improvements in OS (**Figure 2A**) and investigator-assessed PFS (**Figure 2B**) were observed with tislelizumab plus chemotherapy compared to placebo plus chemotherapy
 - OS benefit was observed across all prespecified subgroups, as shown in **Figure 3**

RESULTS (CONT.)

- A higher ORR and improved DoR were observed with tislelizumab plus chemotherapy vs placebo plus chemotherapy, as shown in **Table 2**

Figure 2. Kaplan–Meier Plots of (A) OS and (B) PFS by Investigator

A

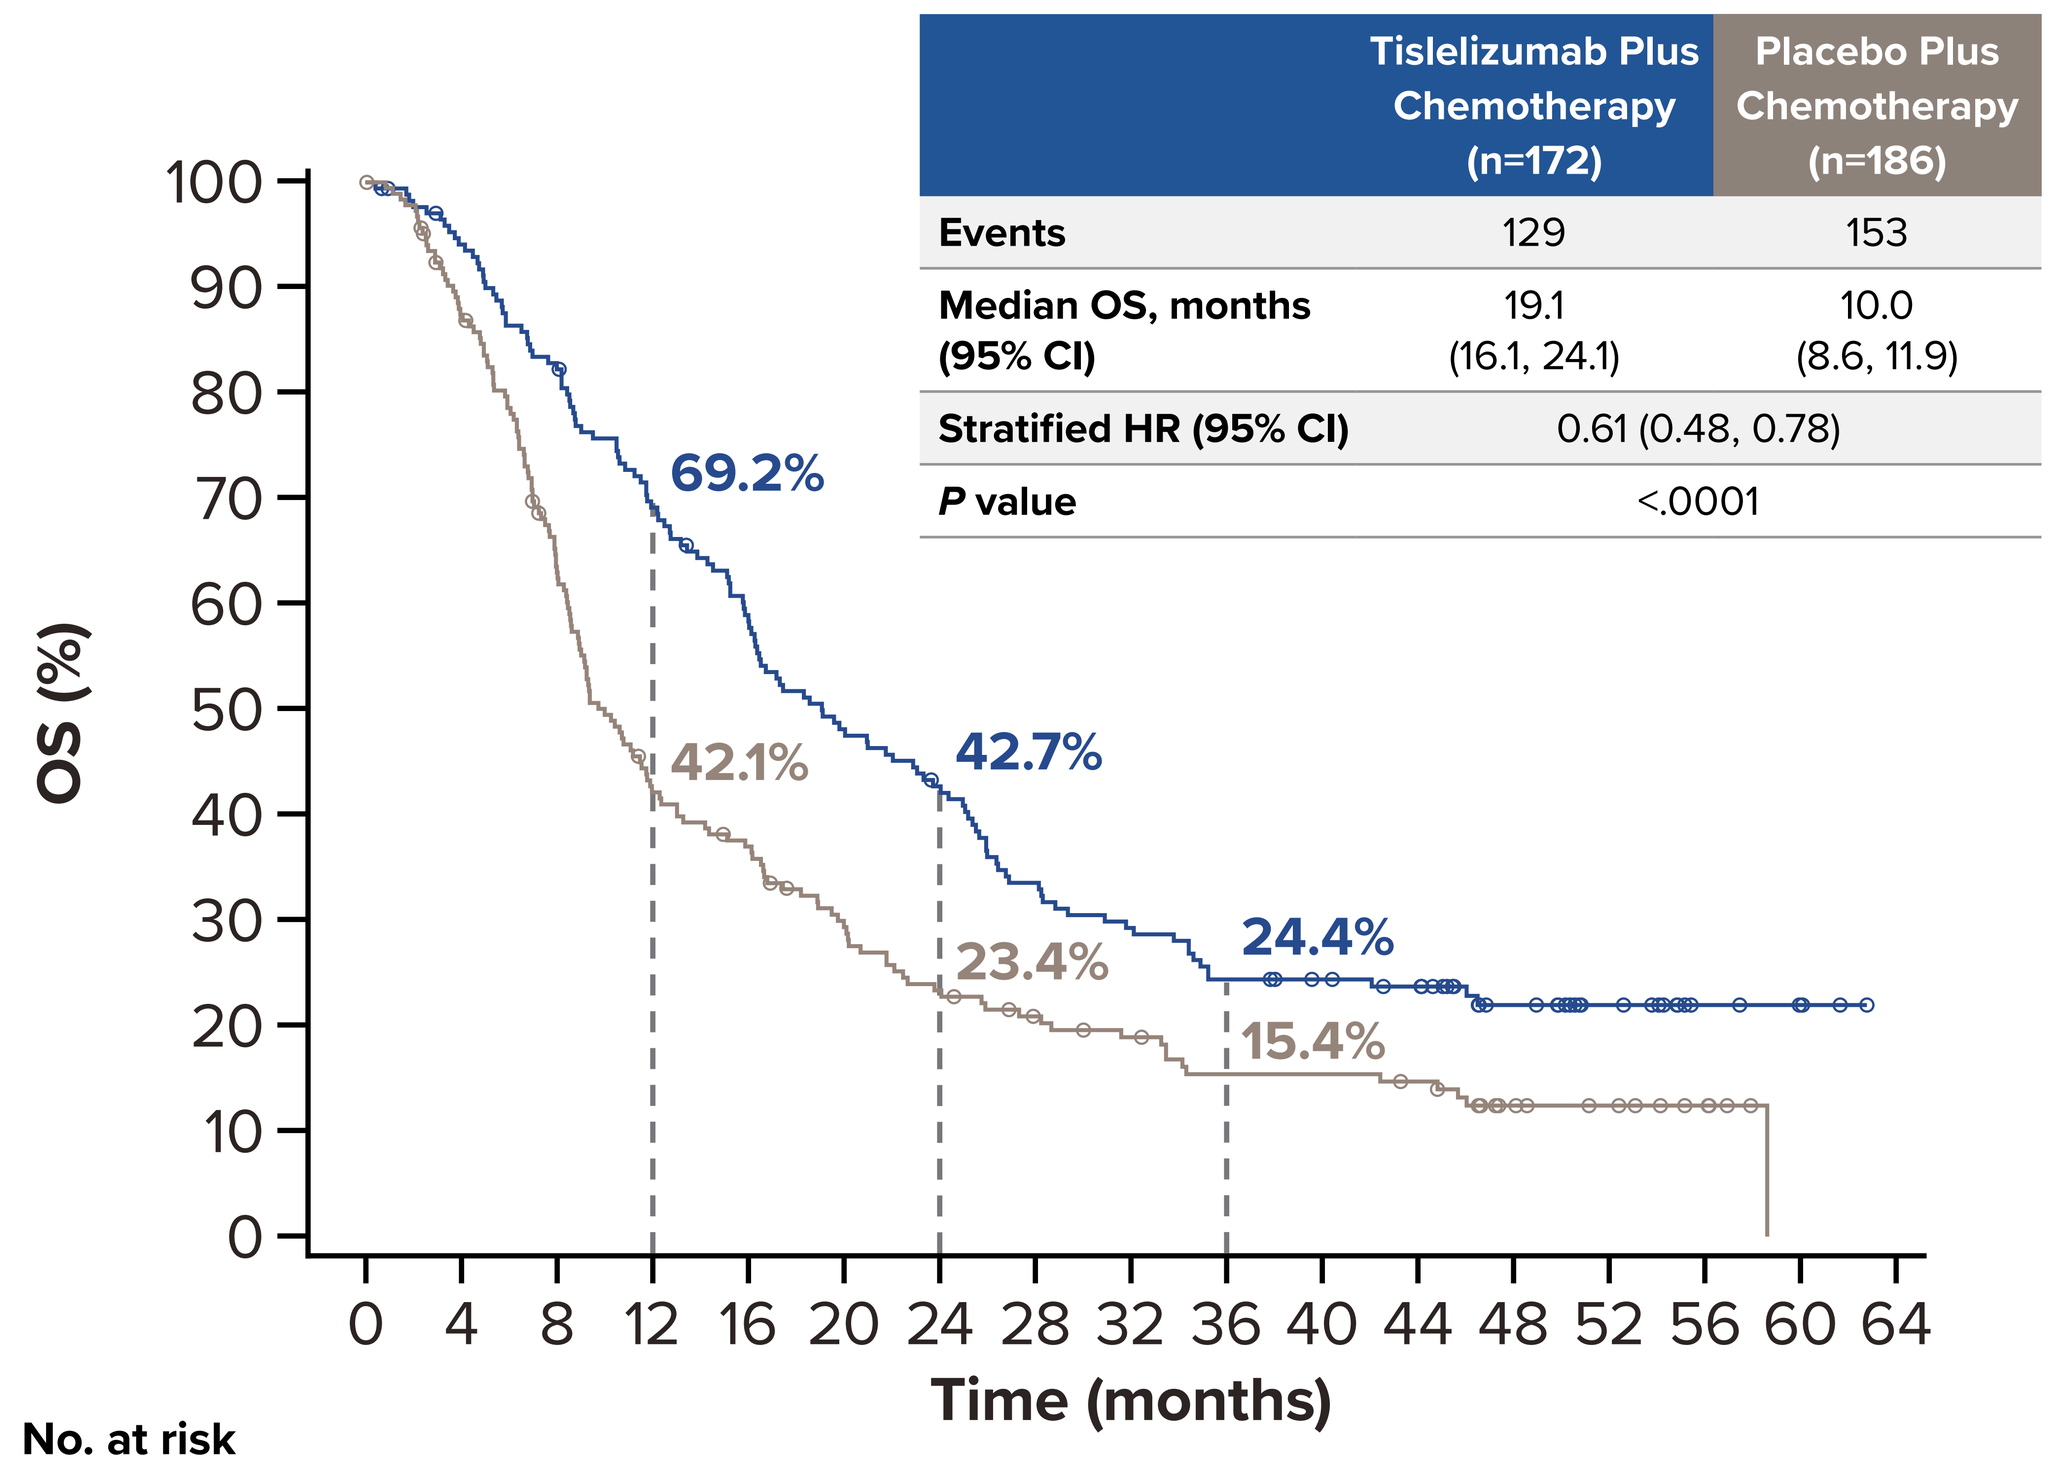
OS
OS (%)
Time (months)
69.2%
42.1%
42.7%
23.4%
24.4%
15.4%

	Tislelizumab Plus Chemotherapy (n=172)	Placebo Plus Chemotherapy (n=186)
Events	129	153
Median OS, months (95% CI)	19.1 (16.1, 24.1)	10.0 (8.6, 11.9)
Stratified HR (95% CI)	0.61 (0.48, 0.78)	
***P* value**	<.0001	

No. at risk	0	4	8	12	16	20	24	28	32	36	40	44	48	52	56	60	64
Tislelizumab plus chemotherapy	172	159	140	116	98	80	70	55	48	40	37	34	22	13	5	4	0
Placebo plus chemotherapy	186	159	113	74	64	50	39	32	28	22	22	20	12	9	5	0	0

B

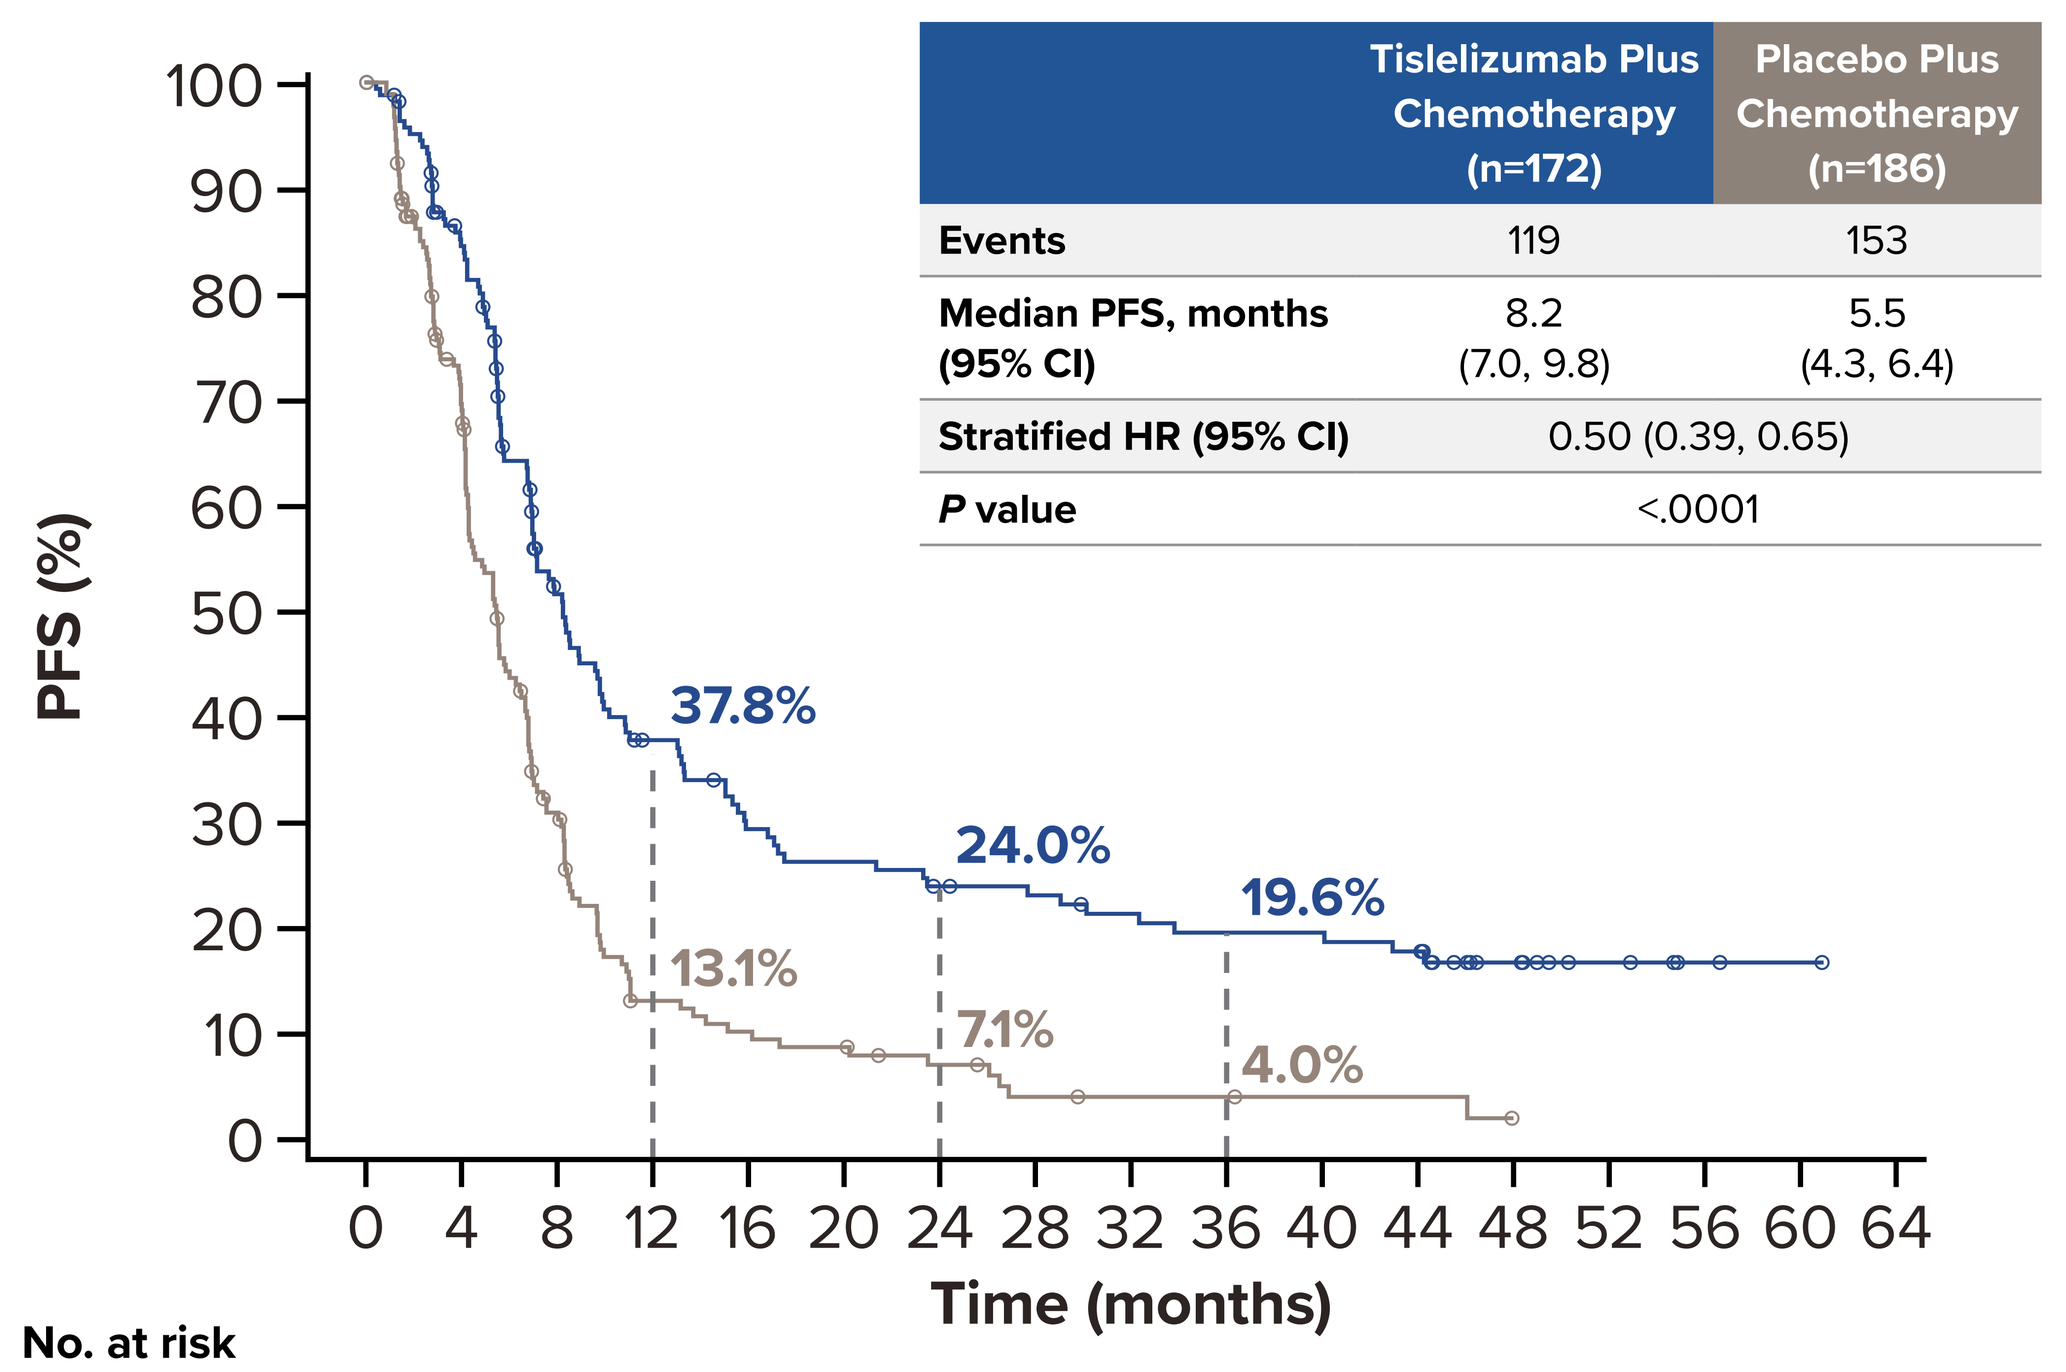
PFS by Investigator
PFS (%)
Time (months)
37.8%
13.1%
24.0%
7.1%
19.6%
4.0%

	Tislelizumab Plus Chemotherapy (n=172)	Placebo Plus Chemotherapy (n=186)
Events	119	153
Median PFS, months (95% CI)	8.2 (7.0, 9.8)	5.5 (4.3, 6.4)
Stratified HR (95% CI)	0.50 (0.39, 0.65)	
***P* value**	<.0001	

No. at risk	0	4	8	12	16	20	24	28	32	36	40	44	48	52	56	60	64
Tislelizumab plus chemotherapy	172	132	71	50	38	34	29	27	24	22	22	20	10	5	2	1	0
Placebo plus chemotherapy	186	115	47	18	14	12	8	4	3	3	2	2	0	0	0	0	0

Medians and other quartiles were estimated by Kaplan–Meier method, with 95% CIs estimated using the method of Brookmeyer and Crowley. OS rates (cumulative probability of OS) were estimated by Kaplan–Meier method, with 95% CIs estimated using Greenwood's formula. One-sided *P* value was estimated from a log-rank test stratified by pooled geographic region (Asia vs rest of world) per IRT, prior definitive therapy (yes vs no) per IRT, and chemotherapy option (investigator choice) per IRT. HR was based on a Cox regression model including treatment as a covariate and pooled geographic region (Asia vs rest of world) per IRT, prior definitive therapy (yes vs no) per IRT, and chemotherapy (investigator choice) per IRT as strata.
Abbreviations: CI, confidence interval; IRT, interactive response technology; PD, progressive disease.

PD-L1 TAP Score vs CPS Concordance

- TAP score 5% and CPS 5 cutoffs had 84.9% overall percent agreement[5], showing substantial concordance (**Figure 4**)

Figure 3. Forest Plot of OS by Subgroup Analysis

Subgroup	Event/Total: Tislelizumab Plus Chemotherapy	Event/Total: Placebo Plus Chemotherapy	Unstratified HR (95% CI)
Overall	129/172	153/186	0.62 (0.49, 0.78)
Age			
<65	74/100	77/98	0.66 (0.48, 0.91)
≥65	55/72	76/88	0.58 (0.41, 0.82)
Sex			
Male	111/141	136/163	0.68 (0.53, 0.87)
Female	18/31	17/23	0.45 (0.23, 0.87)
Smoking status			
Former/current smoker	102/131	113/132	0.60 (0.46, 0.78)
Non-smoker	23/35	32/45	0.70 (0.41, 1.20)
ECOG PS			
0	36/50	43/57	0.61 (0.39, 0.95)
1	93/122	110/129	0.62 (0.47, 0.82)
Geographic region analysis			
Asia	99/133	122/147	0.61 (0.47, 0.80)
Rest of world	30/39	31/39	0.65 (0.39, 1.08)
Race			
Asian and other	99/133	124/150	0.62 (0.47, 0.80)
White	30/39	29/36	0.62 (0.37, 1.04)
Disease status at study entry			
Metastatic	113/147	137/166	0.66 (0.51, 0.85)
Locally advanced	16/25	16/20	0.42 (0.21, 0.86)
Prior definitive therapy – IRT			
Yes	47/70	68/80	0.46 (0.32, 0.67)
No	82/102	85/106	0.76 (0.56, 1.03)

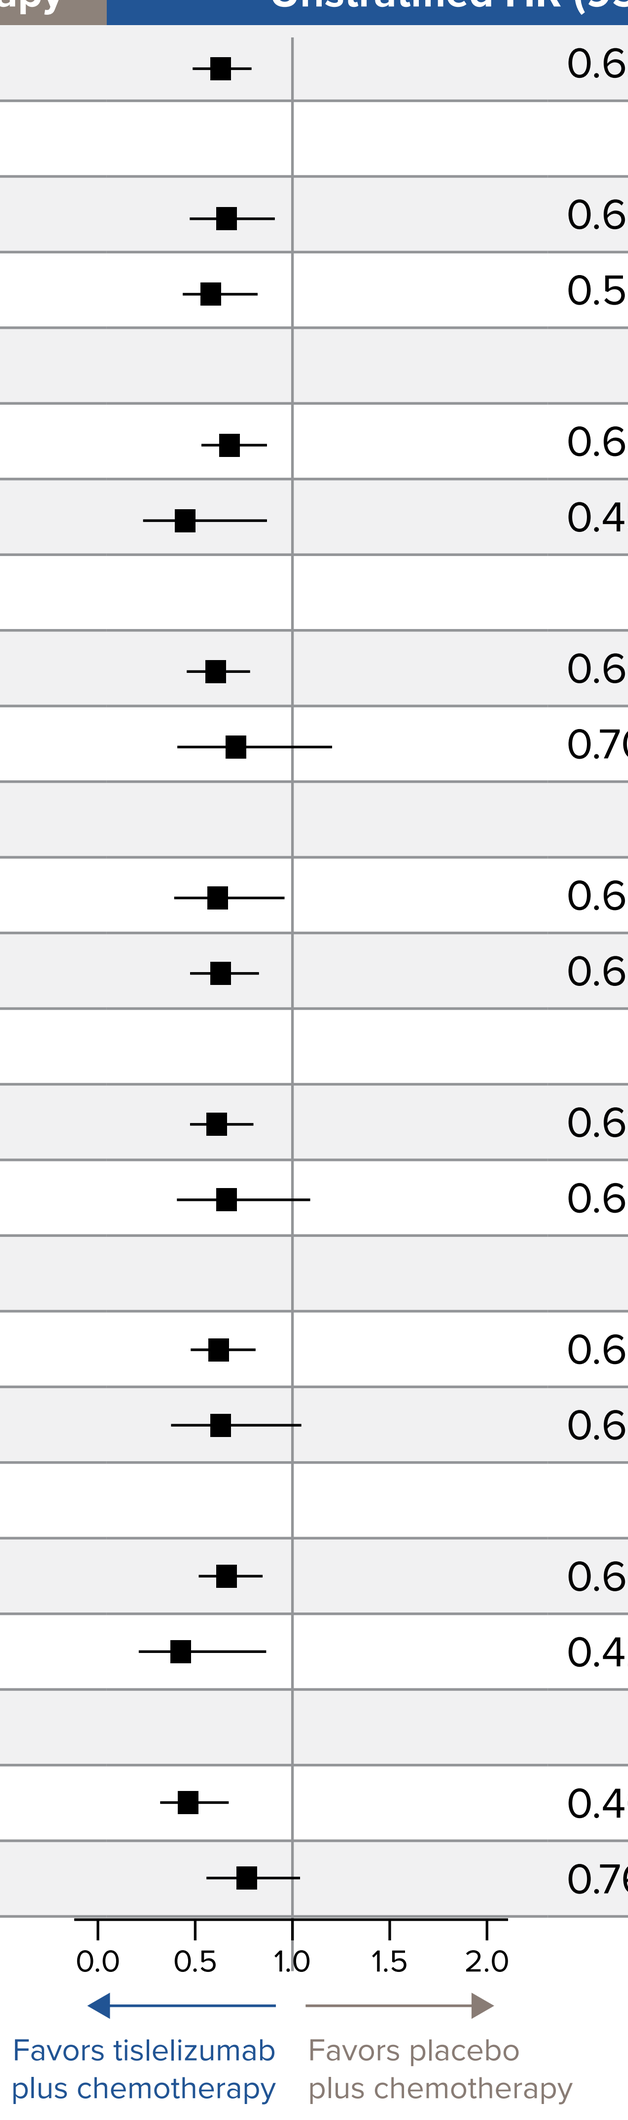
0.0
0.5
1.0
1.5
2.0
Favors tislelizumab plus chemotherapy
Favors placebo plus chemotherapy

HR was based on an unstratified Cox regression model including treatment as covariate. The race subcategory "Other" includes American Indian or Alaska Native, Not Reported, and Unknown.
Abbreviations: CRF, case report form.

Table 2. Efficacy Outcomes

	Tislelizumab Plus Chemotherapy (n=172)	Placebo Plus Chemotherapy (n=186)
ORR[a], n (%) [95% CI][b]	123 (71.5) [64.1, 78.1]	77 (41.4) [34.2, 48.8]
Median DoR[c], months [95% CI][d]	7.1 [5.8, 9.7]	5.4 [4.1, 5.8]

[a]Investigator assessed. ORR is unconfirmed and defined as the proportion of patients with PR or CR, as assessed by the investigator per RECIST v1.1. [b]95% CI was estimated using the Clopper–Pearson method. [c]Duration of response analysis included patients with unconfirmed objective response. Medians and other quartiles were estimated by Kaplan–Meier method. [d]95% CI was estimated using the method of Brookmeyer and Crowley.
Abbreviations: CR, complete response; PR, partial response.

Figure 4. PD-L1 TAP Score vs CPS Concordance

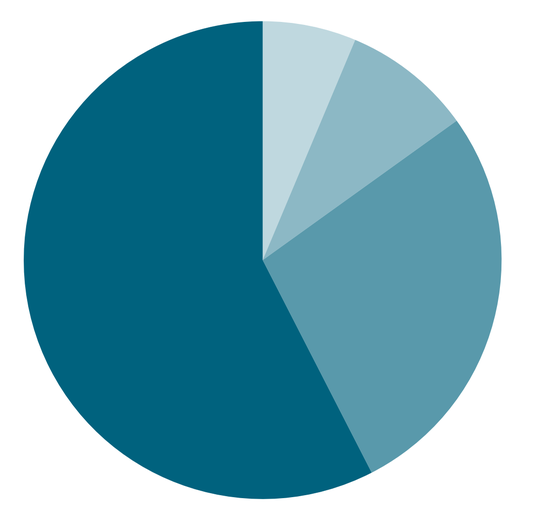
TAP ≥5%/CPS ≥5: n=309 (57.5%)
TAP <5%/CPS <5: n=147 (27.4%)
TAP ≥5%/CPS <5: n=47 (8.8%)
TAP <5%/CPS ≥5: n=34 (6.3%)

TAP/CPS[5] 5%/5	n/N	Agreement, % (95% CI
PPA	309/343	90.1 (87, 93)
NPA	147/194	75.8 (70, 81)
OPA	456/537	84.9 (82, 88)
Cohen's Kappa (95% CI)	0.67 (0.60, 0.73)	

Strength of agreement (Kappa)

Slight (0.01-0.20)	Fair (0.21-0.40)	Moderate (0.41-0.60)	Substantial (0.61-0.80)	Almost perfect (0.81-1.0)

Abbreviations: NPA, negative percent agreement; OPA, overall percent agreement; PPA, positive percent agreement.

Safety/Tolerability Profile

- Treatment-related adverse events (TRAEs) were observed in most patients across both cohorts, with similar any-grade rates. A higher incidence of grade ≥3 and serious TRAEs were observed with tislelizumab plus chemotherapy (**Table 3**)
 - Grade ≥3 TRAEs occurring in ≥10% of patients with tislelizumab plus chemotherapy vs placebo plus chemotherapy included decreased neutrophil count (35.1% vs 31.9%), decreased white blood cell count (12.3% vs 17.8%), and anemia (13.5% vs 11.4%)
- More TRAEs leading to death (2.9% vs 1.6%) and treatment-emergent adverse events (TEAEs) leading to any treatment discontinuation (34.5% vs 23.2%) occurred in the tislelizumab plus chemotherapy arm than in the placebo plus chemotherapy arm
- Immune-mediated adverse events (imAEs) were observed in both arms. The incidence of grade ≥3 imAEs was higher with tislelizumab plus chemotherapy vs placebo plus chemotherapy (8.8% vs 2.2%) (**Table 3**)

Table 3. Safety Summary (Safety Analysis Set)

	Tislelizumab Plus Chemotherapy (n=171)	Placebo Plus Chemotherapy (n=185)
Patients with ≥1 TRAEs, n (%)	167 (97.7)	182 (98.4)
Grade ≥3	120 (70.2)	123 (66.5)
Serious	50 (29.2)	38 (20.5)
Leading to death	5 (2.9)	3 (1.6)
Patients with ≥1 TEAEs leading to any treatment discontinuation, n (%)	59 (34.5)	43 (23.2)
Patients with any imAEs, n (%)	73 (42.7)	40 (21.6)
Grade ≥3	15 (8.8)	4 (2.2)

Adverse event grades were evaluated based on Common Terminology Criteria for Adverse Events version 4.03. Adverse event terms were coded using Medical Dictionary for Drug Regulatory Affairs version 24.0. TRAEs include events considered by the investigator to be related to study treatments or with missing assessment of the causal relationship. Deaths here exclude those caused by disease under study.

REFERENCES

1. European Medicines Agency. Tevimbra 100 mg concentrate for solution for infusion. Summary of product characteristics. https://www.ema.europa.eu/en/documents/product-information/tevimbra-epar-product-information_en.pdf. Accessed: December 19, 2024.
2. Kato K, et al. *Cancer Med*. 2024;13:e7235.
3. Xu J, et al. *Lancet Oncol*. 2023;24:483-495.
4. Yoon HH, et al. *J Clin Oncol*. 2024;42(Suppl 16):4032.
5. Moehler M, et al. *Mod Pathol*. 2025. doi.org/10.1016/j.modpat.2025.100793.

DISCLOSURES

DT: Consulting fees/honoraria from Amgen, AstraZeneca, Incyte, Merck Serono, MSD, Novartis, Pierre Fabre, Roche, Bristol Myers Squibb, Takeda, and Servier Laboratories.

ACKNOWLEDGMENTS

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This study was sponsored by BeOne Medicines, Ltd. Medical writing support was provided by Nitya Venkataraman, PhD, of Parexel, and supported by BeOne Medicines.

CORRESPONDENCE: David Tougeron, david.tougeron@chu-poitiers.fr

Presented at ESMO GI, July 2-5, 2025, Barcelona, Spain