A Phase 1 Study of BGB-24714, a Second Mitochondrial-Derived Activator of Caspases Mimetic, as Monotherapy or in Combination With Chemotherapy or Concurrent Chemoradiotherapy in Solid Tumors

CT120

Andreas Varkaris,[1] Erika Hamilton,[2] Jermaine Coward,[3] Rafael Santana-Davila,[4] Jinming Yu,[5] Byoung Chul Cho,[6] Jung Yun Lee,[6] Yongsheng Li,[7] Sanjeev Deva,[8] Youngjoo Lee,[9] Huai Liu,[10] Yuping Sun,[5] Zhe Yang,[11] Wen Xu,[12] Sook Ryun Park,[13] Se-Hoon Lee,[14] Andrew Z. Wang,[15] Hui Shao,[16] Jinhui Zhang,[17] Zhaoyin Zhu,[18] Runzhe Chen,[18] Hua-Xin Gao,[18] Judy S. Wang[19]

[1]Massachusetts General Hospital, Boston, MA, USA; [2]Sarah Cannon Research Institute, Nashville, TN, USA; [3]ICON Cancer Centre, South Brisbane, QLD, Australia; [4]Fred Hutch Cancer Center, Seattle, WA, USA; [5]Shandong Cancer Hospital, Jinan, Shandong, China; [6]Severance Hospital, Yonsei University Health System, Seoul, Republic of Korea; [7]Chongqing Cancer Hospital, Shapingba District, Chongqing, China; [8]Auckland City Hospital, Grafton, Auckland, New Zealand; [9]National Cancer Center, Goyang-si, Gyeonggi-do, Republic of Korea; [10]Hunan Cancer Hospital, Changsha, Hunan, China; [11]Shandong Provincial Hospital, Jinan, Shandong, China; [12]Princess Alexandra Hospital, Woolloongabba, QLD, Australia; [13]Asan Medical Center, Seoul, Republic of Korea; [14]Samsung Medical Center, Seoul, Republic of Korea; [15]University of Texas Southwestern Medical Center, Dallas, TX, USA; [16]BeOne Medicines, Ltd, Beijing, China; [17]BeOne Medicines, Ltd, Shanghai, China; [18]BeOne Medicines, Ltd, San Carlos, CA, USA; [19]Florida Cancer Specialists and Sarah Cannon Research Institute, Sarasota, FL, USA

CONCLUSIONS

- BGB-24714 demonstrated antitumor activity with a manageable safety profile in patients with advanced or metastatic solid tumors

INTRODUCTION

- Apoptosis modulation may enhance the efficacy of standard-of-care chemotherapy and concurrent chemoradiotherapy in solid tumors[1-3]
- BGB-24714, an oral selective second mitochondrial-derived activator of caspases (SMAC) mimetic that antagonizes Inhibitor of Apoptosis Proteins (IAPs), has demonstrated promising preclinical antitumor activity[4]
- This open-label, multicenter, first-in-human phase 1 study evaluated BGB-24714 as monotherapy or in combination with chemotherapy or concurrent chemoradiotherapy in patients with advanced or metastatic solid tumors (NCT05381909)

METHODS

- The study included dose-escalation (1a) and -expansion (1b) phases (**Figure 1**)
- Primary endpoints were safety/tolerability (phase 1a) and preliminary antitumor activity (phase 1b)
- Nine dose levels (30 mg to 900 mg oral once daily [QD]) of BGB-24714 were assessed in phase 1a Part A monotherapy (n=68) and 7 dose levels (60 mg to 650 mg oral QD) in phase 1a Parts B to D (n=64)
 - Two dosing regimens were evaluated, one with 1 week off and the other with no weeks off
- The phase 1b cohorts received the recommended dose for expansion (RDFE), which was 300 mg

Figure 1. Study Design

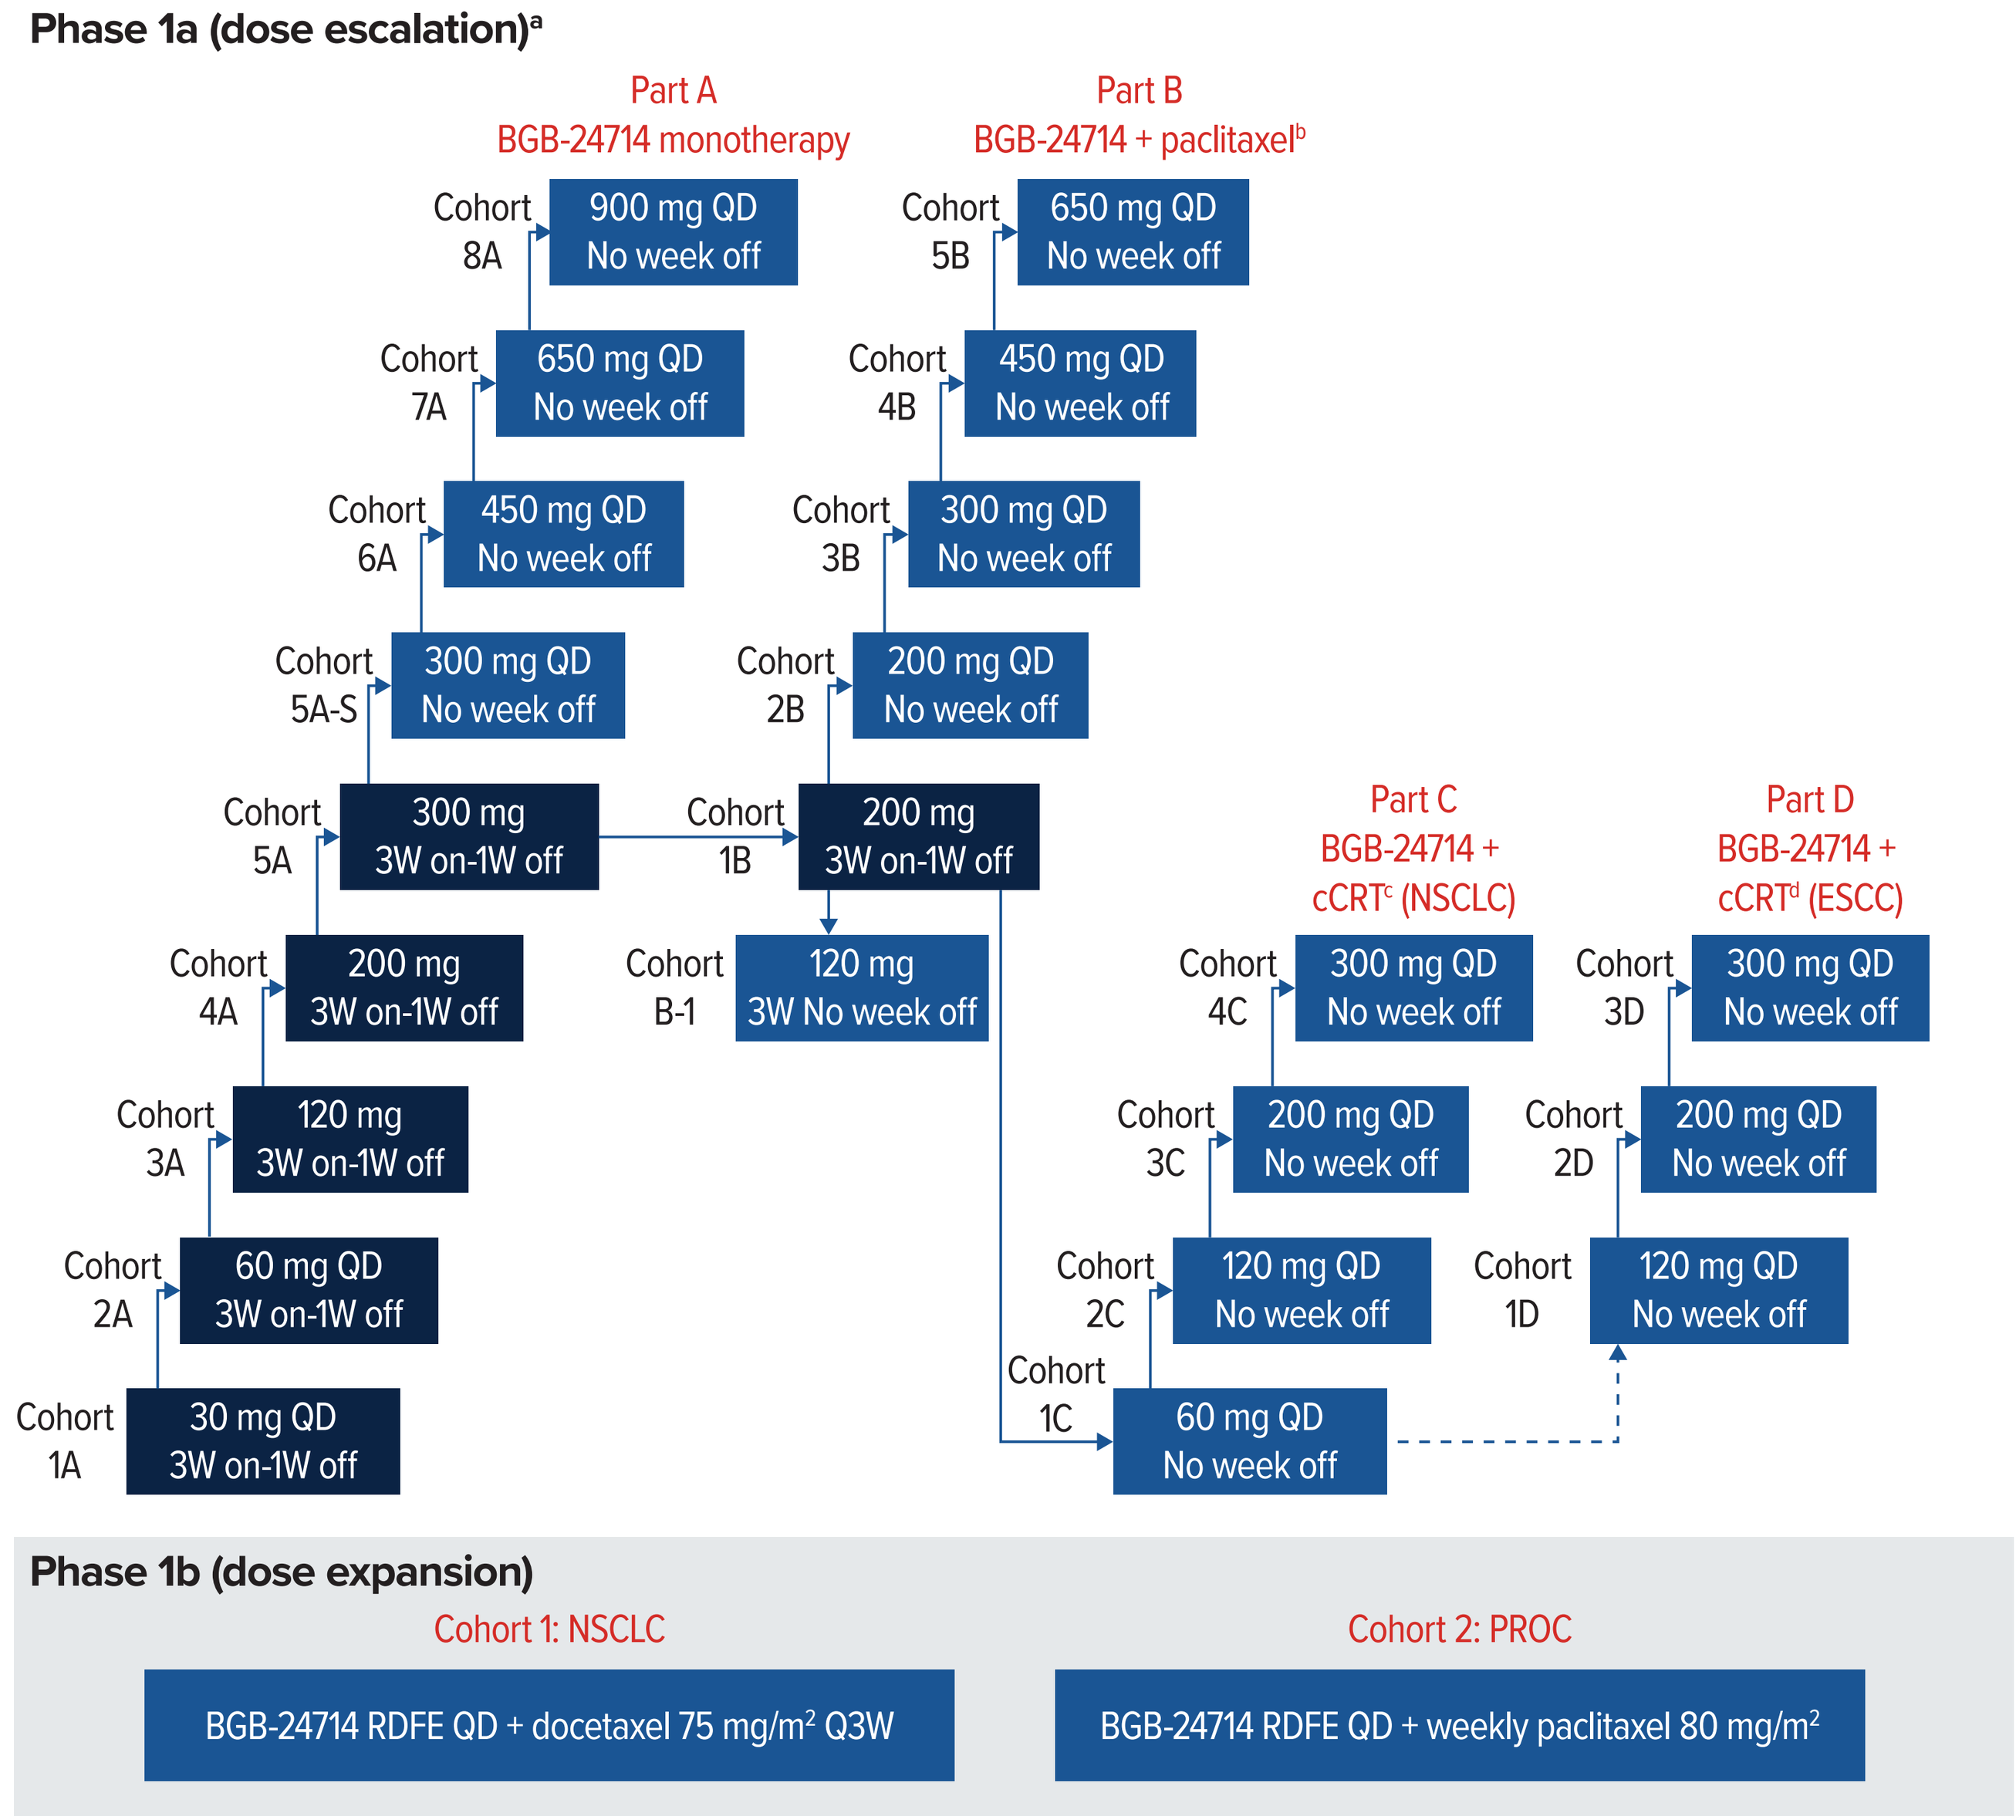
Phase 1a (dose escalation)[a]
Part A
BGB-24714 monotherapy
Part B
BGB-24714 + paclitaxel[b]
Cohort 8A 900 mg QD No week off
Cohort 7A 650 mg QD No week off
Cohort 6A 450 mg QD No week off
Cohort 5A-S 300 mg QD No week off
Cohort 5A 300 mg 3W on-1W off
Cohort 4A 200 mg 3W on-1W off
Cohort 3A 120 mg 3W on-1W off
Cohort 2A 60 mg QD 3W on-1W off
Cohort 1A 30 mg QD 3W on-1W off
Cohort 5B 650 mg QD No week off
Cohort 4B 450 mg QD No week off
Cohort 3B 300 mg QD No week off
Cohort 2B 200 mg QD No week off
Cohort 1B 200 mg 3W on-1W off
Cohort B-1 120 mg 3W No week off
Part C
BGB-24714 + cCRT[c] (NSCLC)
Part D
BGB-24714 + cCRT[d] (ESCC)
Cohort 4C 300 mg QD No week off
Cohort 3C 200 mg QD No week off
Cohort 2C 120 mg QD No week off
Cohort 1C 60 mg QD No week off
Cohort 3D 300 mg QD No week off
Cohort 2D 200 mg QD No week off
Cohort 1D 120 mg QD No week off
Phase 1b (dose expansion)
Cohort 1: NSCLC
BGB-24714 RDFE QD + docetaxel 75 mg/m² Q3W
Cohort 2: PROC
BGB-24714 RDFE QD + weekly paclitaxel 80 mg/m²

Dark blue represents 3 weeks on-1 week off and light blue represents no weeks off. BGB-24714 is administered orally QD. [a]In phase 1a, N≥3 for all cohorts. [b]Paclitaxel (80 mg/m²) is administered by intravenous infusion on Days 1, 8, and 15 of every 28-day treatment cycle. [c]Paclitaxel (45-50 mg/m²) and carboplatin (AUC2) is administered weekly, by intravenous infusion, on Days 1, 8, 15, and 22 of each 28-day treatment cycle for 6 weeks. [d]Paclitaxel (50 mg/m²) and carboplatin (AUC2) is administered weekly, by intravenous infusion, on Days 1, 8, 15 and 22 of each 28-day treatment cycle for 5 weeks.
Abbreviations: AUC, area under curve; cCRT, concurrent chemoradiotherapy; ESCC, esophageal squamous cell carcinoma; NSCLC, non-small cell lung cancer; PROC, platinum-resistant ovarian cancer; Q3W, every 3 weeks; W, week.

RESULTS

Patients

- As of July 25, 2025, 157 patients were enrolled and treated (132 in phase 1a dose escalation Parts A to D; 25 in phase 1b [11 NSCLC; 14 PROC])
- Patient characteristics are summarized in **Table 1**
- Median (range) study follow-up time was 3.8 (1.2-27.1) months in Part A monotherapy, 4.1 (1.4-13.3) months in Part B, 11.8 (2.0-16.4) months in Part C, 7.6 (6.8-11.1) months in Part D, 8.1 (2.1-10.9) months for phase 1b NSCLC cohort, and 6.3 (2.5-10.7) months for phase 1b PROC cohort

Table 1. Demographics and Baseline Characteristics

	Phase 1a				Phase 1b	
	Part A BGB-24714 monotherapy[a] (n=68)	Part B BGB-24714 + paclitaxel[b] (n=48)	Part C BGB-24714 + cCRT NSCLC (n=11)	Part D BGB-24714 + cCRT ESCC (n=5)	BGB-24714 + chemotherapy NSCLC (n=11)	BGB-24714 + chemotherapy PROC (n=14)
Age, median (range)	63 (31-82)	58 (31-88)	68 (50-84)	63 (61-68)	65 (42-73)	64 (43-81)
Sex, n (%)						
Female	53 (77.9)	29 (60.4)	3 (27.3)	1 (20.0)	3 (27.3)	14 (100)
Male	15 (22.1)	19 (39.6)	8 (72.7)	4 (80.0)	8 (72.7)	0
Race, n (%)						
Asian	20 (29.4)	4 (8.3)	6 (54.5)	5 (100)	6 (54.4)	11 (78.6)
Black or African American	3 (4.4)	0	0	0	0	0
White	44 (64.7)	43 (89.6)	5 (45.5)	0	4 (36.4)	3 (21.4)
Other/unknown	1 (1.5)	1 (2.1)	0	0	1 (9.1)	0
Ethnicity, n (%)						
Hispanic or Latino	1 (1.5)	2 (4.2)	0	0	0	1 (7.1)
Not Hispanic or Latino	64 (94.1)	44 (91.7)	11 (100)	5 (100)	10 (90.9)	13 (92.9)
Not reported/unknown	3 (4.4)	2 (4.2)	0	0	1 (9.1)	0
Prior lines of systemic therapy, median (range)	3 (1-8)	3 (1-11)	0	0	2 (1-4)	4 (2-4)

[a]Tumor types included adenoid cystic carcinoma (n=1), appendiceal cancer (n=1), breast cancer (n=8), cervical cancer (n=3), cholangiocarcinoma (n=1), colorectal cancer (n=5), endometrial cancer (n=9), endothelial tumor (n=1), head and neck cancer (n=4), leiomyosarcoma (n=1), melanoma (n=1), NSCLC (n=12), ovarian cancer (n=12), pancreatic cancer (n=4), sarcoma (n=1), small cell lung cancer (n=2), thymoma (n=1), and vulvar cancer (n=1). [b]Tumor types include adenoid cystic carcinoma (n=4), ampullary cancer (n=1), anal cancer (n=1), appendiceal cancer (n=2), bladder cancer (n=1), breast cancer (n=7), cervical cancer (n=1), cholangiocarcinoma (n=1), chordoma (n=1), colorectal cancer (n=3), endometrial cancer (n=2), esophageal cancer (n=1), gastric cancer (n=1), head and neck cancer (n=4), kidney cancer, (n=1), leiomyosarcoma (n=2), Merkel cell carcinoma (n=1), myoepithelial cancer (n=1), myxofibrosarcoma (n=1), myxoid liposarcoma (n=1), NSCLC (n=5), ovarian cancer (n=4), spindle cell sarcoma (n=1) and urothelial cancer (n=1).

Safety

- Treatment-emergent adverse events (TEAEs) and BGB-24714-related TEAEs are summarized in **Table 2**
- The most common BGB-24714-related TEAEs varied by study part and combination regimen. These BGB-24714-related TEAEs were predominantly low grade: nausea (33.8%) in phase 1a Part A monotherapy, diarrhea (25%) in Part B, neutrophil count decreased (43.8%) in Parts C/D, diarrhea and fatigue (45.5% each) in phase 1b-NSCLC, and stomatitis, alanine aminotransferase (ALT)/aspartate aminotransferase (AST) increased (28.6% each) in phase 1b-PROC
- The most common grade ≥3 BGB-24714-related TEAEs were ALT increased (5.9%) in phase 1a Part A monotherapy, lipase increased and fatigue (6.3% each) in Part B, neutrophil count decreased (31.3%) in Parts C/D, anemia (27.3%) in phase 1b-NSCLC, and neutrophil count decreased (28.6%) in phase 1b-PROC
- The most common serious BGB-24714-related TEAEs (≥5%) were pneumonitis (6.3%) in phase 1a Part B, diarrhea (18.2%) in phase 1b-NSCLC cohort, and febrile neutropenia and fatigue (7.1% each) in phase 1b-PROC cohort
- All TEAEs leading to treatment discontinuation were reported in single patients, with no clustering of events in any group, except the following: pneumonitis was reported in 3 patients each in Part B (6.3%) and phase 1b (12.0%), and also in 1 patient in Part A monotherapy (1.5%). In Parts C/D, neutrophil count decreased was reported in 3 patients (18.8%) and white blood cell count decreased was reported in 2 patients (12.5%)
 - Treatment discontinuation due to BGB-24714-related TEAEs occurred in 10.3% of patients in phase 1a Part A monotherapy, 12.5% in Part B, 27.3% in Part C, and 0% in Part D. In phase 1b, discontinuation rates were 18.2% in NSCLC and 28.6% in PROC
- Dose-limiting toxicities were grade 3 ALT/AST increased (n=2) and grade 3 gamma-glutamyltransferase increased (n=2) in Part A monotherapy, grade 3 febrile neutropenia (n=2) in Part B, and grade 3 esophagitis (n=1) in Part C
- One patient treated with 120 mg BGB-24714 + paclitaxel with no week off had a fatal TEAE of pneumonitis, which was evaluated as related to BGB-24714 and paclitaxel
- No maximum tolerated dose was reached in any part of the study

Table 2. Safety Summary

Parameter, n (%)	Phase 1a Part A BGB-24714 monotherapy (n=68)	Phase 1a Part B BGB-24714 + paclitaxel (n=48)	Phase 1a Part C BGB-24714 + cCRT NSCLC (n=11)	Phase 1a Part D BGB-24714 + cCRT ESCC (n=5)	Phase 1b BGB-24714 + chemotherapy NSCLC (n=11)	Phase 1b BGB-24714 + chemotherapy PROC (n=14)
Any TEAE	65 (95.6)	48 (100)	11 (100)	5 (100)	11 (100)	14 (100)
Grade ≥3	35 (51.5)	32 (66.7)	8 (72.7)	3 (60.0)	10 (90.9)	13 (92.9)
Serious	18 (26.5)	22 (45.8)	3 (27.3)	0	7 (63.6)	8 (57.1)
Discontinuation	8 (11.8)	7 (14.6)	4 (36.4)	0	2 (18.2)	5 (35.7)
Leading to death	4 (5.9)	4 (8.3)	0	0	0	0
BGB-24714-related TEAE	54 (79.4)	38 (79.2)	7 (63.6)	4 (80.0)	10 (90.9)	14 (100)
Grade ≥3	21 (30.9)	16 (33.3)	6 (54.5)	3 (60.0)	8 (72.7)	9 (64.3)
Serious	4 (5.9)	6 (12.5)	0	0	4 (36.4)	1 (7.1)
Discontinuation	7 (10.3)	6 (12.5)	3 (27.3)	0	2 (18.2)	4 (28.6)
Leading to death	0	1 (2.1)	0	0	0	0

Adverse events were graded for severity using CTCAE v5.0. BGB-24714-related TEAEs include those events considered by the investigator to be related or with missing assessment of the causal relationship.
Abbreviation: CTCAE v5.0, Common Terminology Criteria for Adverse Events, version 5.0.

Pharmacodynamics

- Pharmacodynamics showed rapid and sustained cIAP1 degradation in a dose-dependent manner, demonstrating strong target engagement in the apoptosis regulatory pathway (**Figure 2**)
 - Approximately 50% degradation was observed on Day 2 at the 60 mg dose; near complete cIAP1 degradation was achieved by Day 2 and maintained thereafter at doses ≥200 mg
 - More detailed translational pharmacodynamic effects are reported in poster CT121[5]

Figure 2. cIAP1 Degradation by BGB-24714

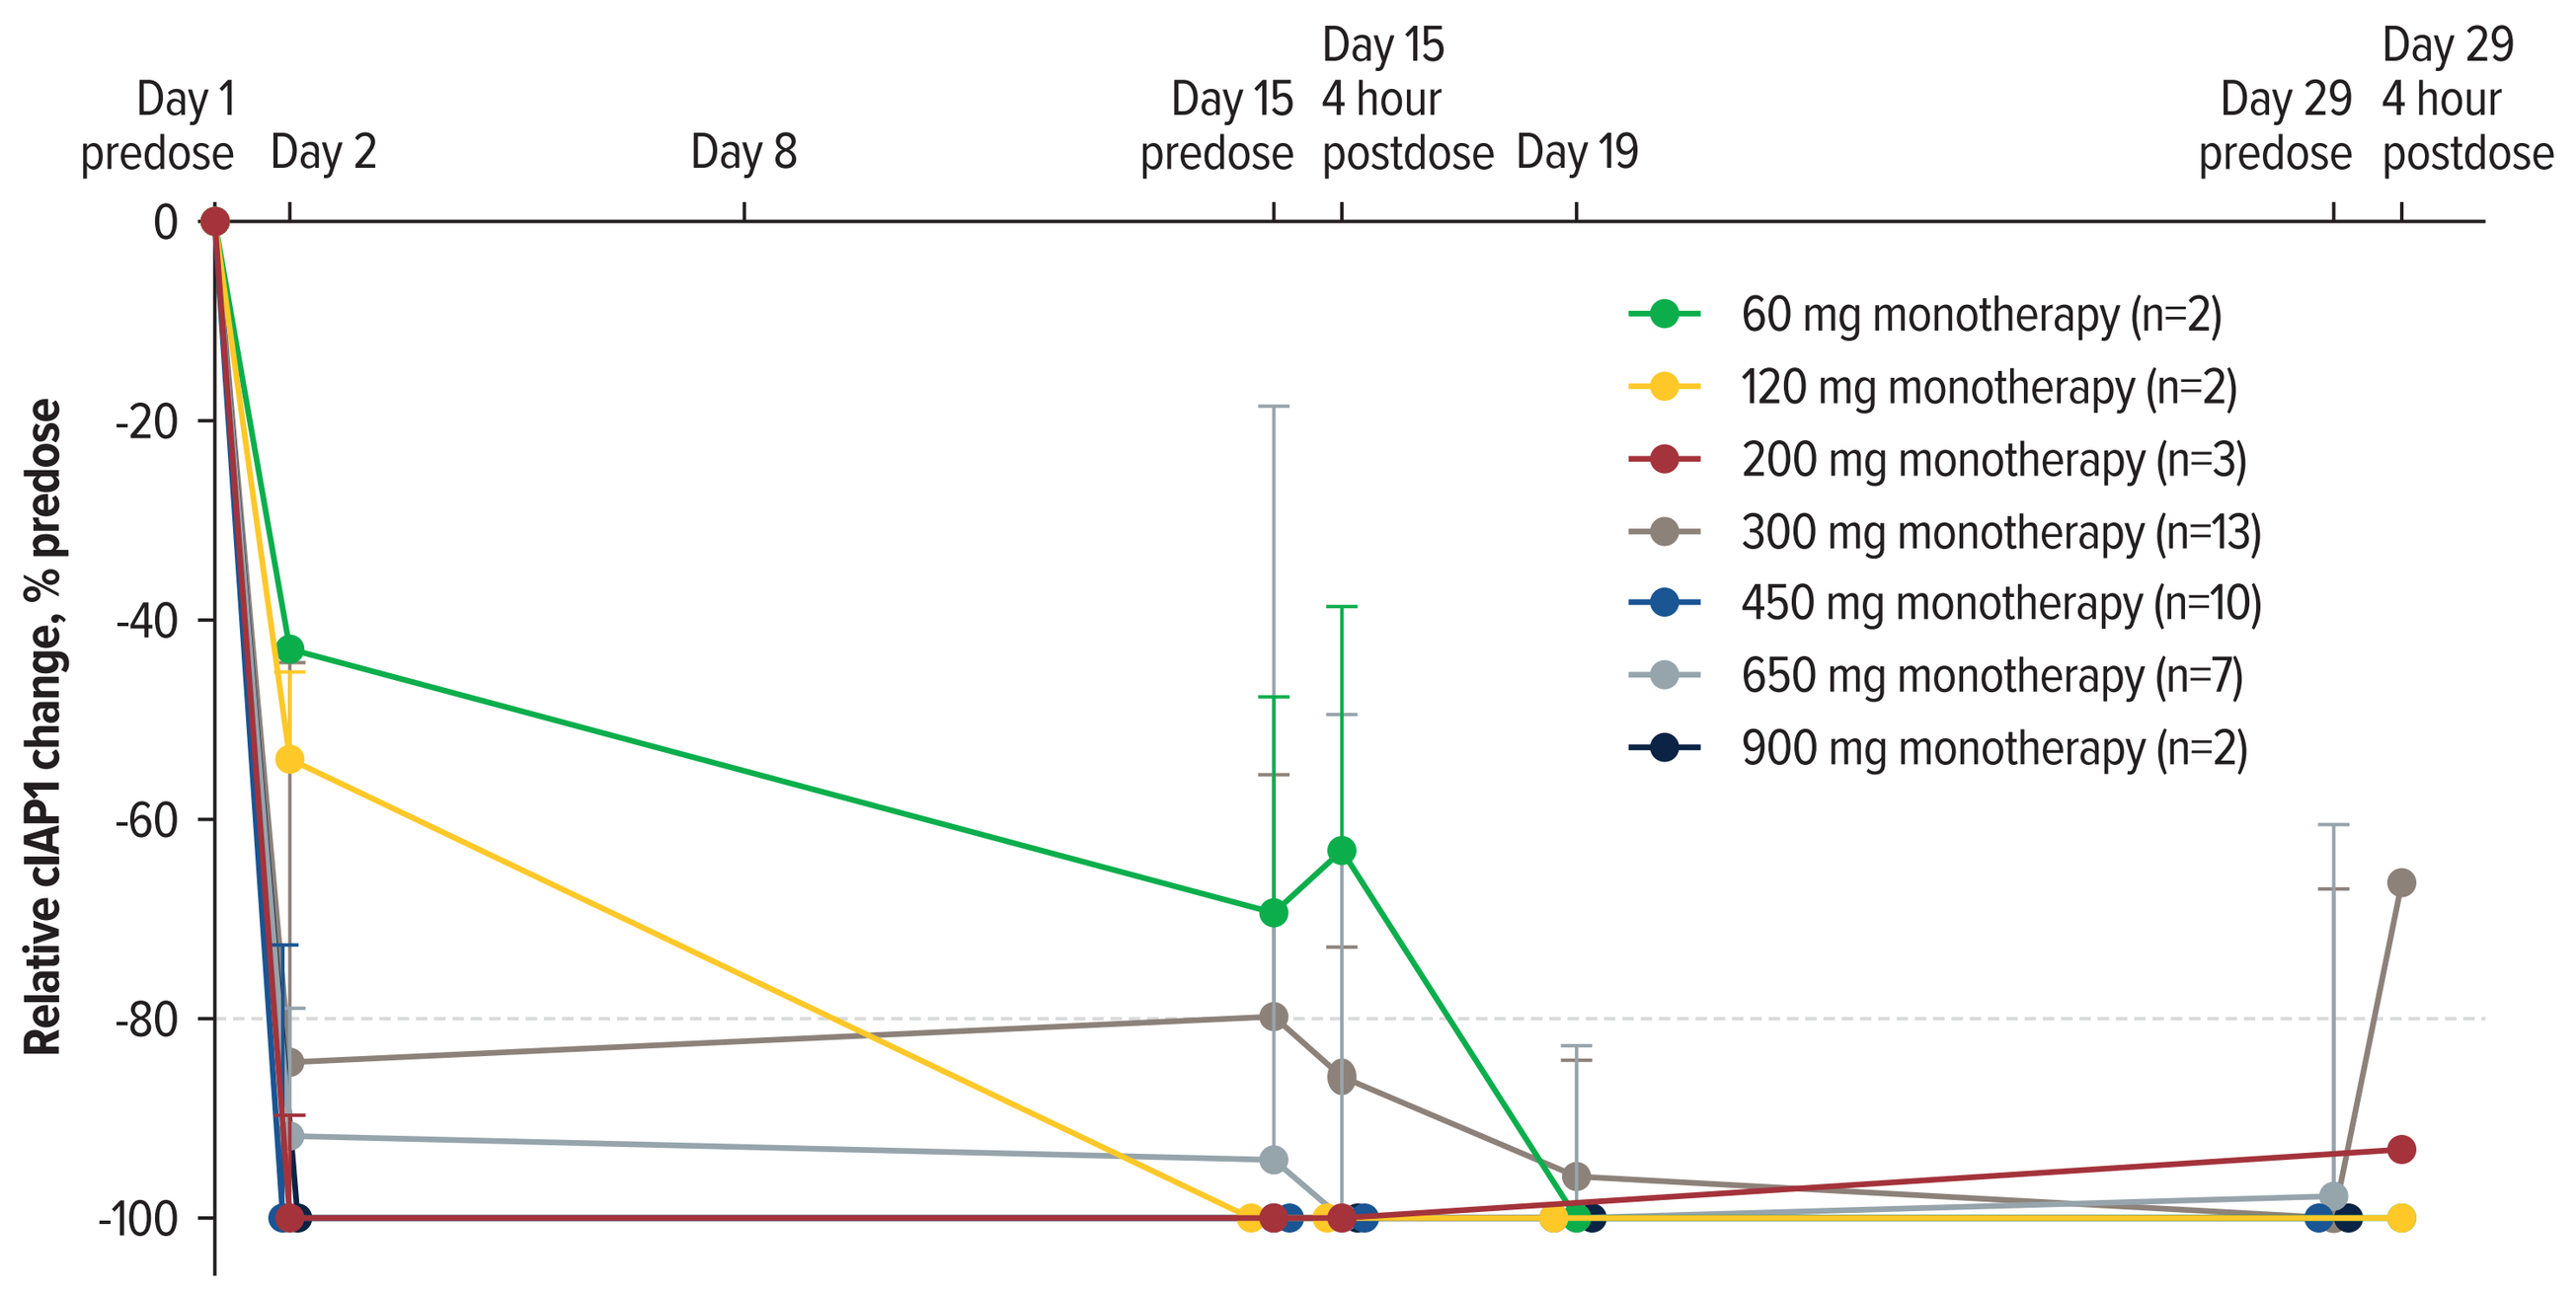
Day 1 predose
Day 2
Day 8
Day 15 predose
Day 15 4 hour postdose
Day 19
Day 29 predose
Day 29 4 hour postdose
Relative cIAP1 change, % predose
0
-20
-40
-60
-80
-100
60 mg monotherapy (n=2)
120 mg monotherapy (n=2)
200 mg monotherapy (n=3)
300 mg monotherapy (n=13)
450 mg monotherapy (n=10)
650 mg monotherapy (n=7)
900 mg monotherapy (n=2)

Median (IQR) cIAP1 level measured in peripheral PBMC.
Abbreviations: IQR, interquartile range; PBMC, peripheral blood mononuclear cell.

Antitumor Activity

- Confirmed treatment response is shown in **Table 3** and **Figure 3**
 - Part A monotherapy: unconfirmed PR was observed in 2 patients with endometrial cancer
 - Part B: 9 unconfirmed PRs, including 6 confirmed PRs were observed across multiple tumor types, including NSCLC, PROC, and breast cancer
 - Part C: 6 confirmed PRs
 - Part D: 2 confirmed PRs
 - Phase 1b: no confirmed PRs in the NSCLC cohort and 4 confirmed PRs in the PROC cohort
- In phase 1b, median (95% confidence interval [CI]) progression-free survival (PFS) was 5.3 months (0.8-not evaluable [NE]) for NSCLC and 5.3 months (1.7-NE) for PROC

Table 3. Confirmed Treatment Response in Efficacy-Evaluable Patients

	Phase 1a				Phase 1b	
	Part A BGB-24714 monotherapy (n=65)	Part B BGB-24714 + paclitaxel (n=47)	Part C BGB-24714 + cCRT NSCLC (n=11)	Part D BGB-24714 + cCRT ESCC (n=3)	BGB-24714 + chemotherapy NSCLC (n=10)	BGB-24714 + chemotherapy PROC (n=14)
Objective response rate (CR + PR), n (%)	0	6 (12.8)	6 (54.4)	2 (66.7)	0	4 (28.6)
95% CI[a]	0.0-5.5	4.8-25.7	23.4-83.3	9.4-99.2	0.0-30.8	8.4-58.1
Disease control rate (CR + PR + SD), n (%)	26 (40.0)	28 (59.6)	9 (81.8)	3 (100)	7 (70.0)	12 (85.7)
95% CI[a]	28.0-52.9	44.3-73.6	48.2-97.7	29.2-100.0	34.8-93.3	57.2-98.2
Clinical benefit rate (CR + PR + durable SD[b]), n (%)	6 (9.2)	11 (23.4)	7 (63.6)	2 (66.7)	1 (10.0)	5 (35.7)
95% CI[a]	3.5-19.0	12.3-38.0	30.8-89.1	9.4-99.2	0.3-44.5	12.8-64.9
Duration of confirmed response (months), median (95% CI[c])	NA	4.7 (3.5-NE)	12.2 (NE-NE)	6.5 (NE-NE)	NA	3.7 (3.5-NE)

[a]95% CI was estimated using the Clopper-Pearson method. [b]SD lasting ≥24 weeks. [c]95% CI was estimated using the Brookmeyer and Crowley method with log-log transformation.
Abbreviations: CR, complete response; NA, not applicable; PR, partial response; SD, stable disease.

Figure 3. Confirmed Best Percent Change From Baseline in Target Lesion Sum of Diameters

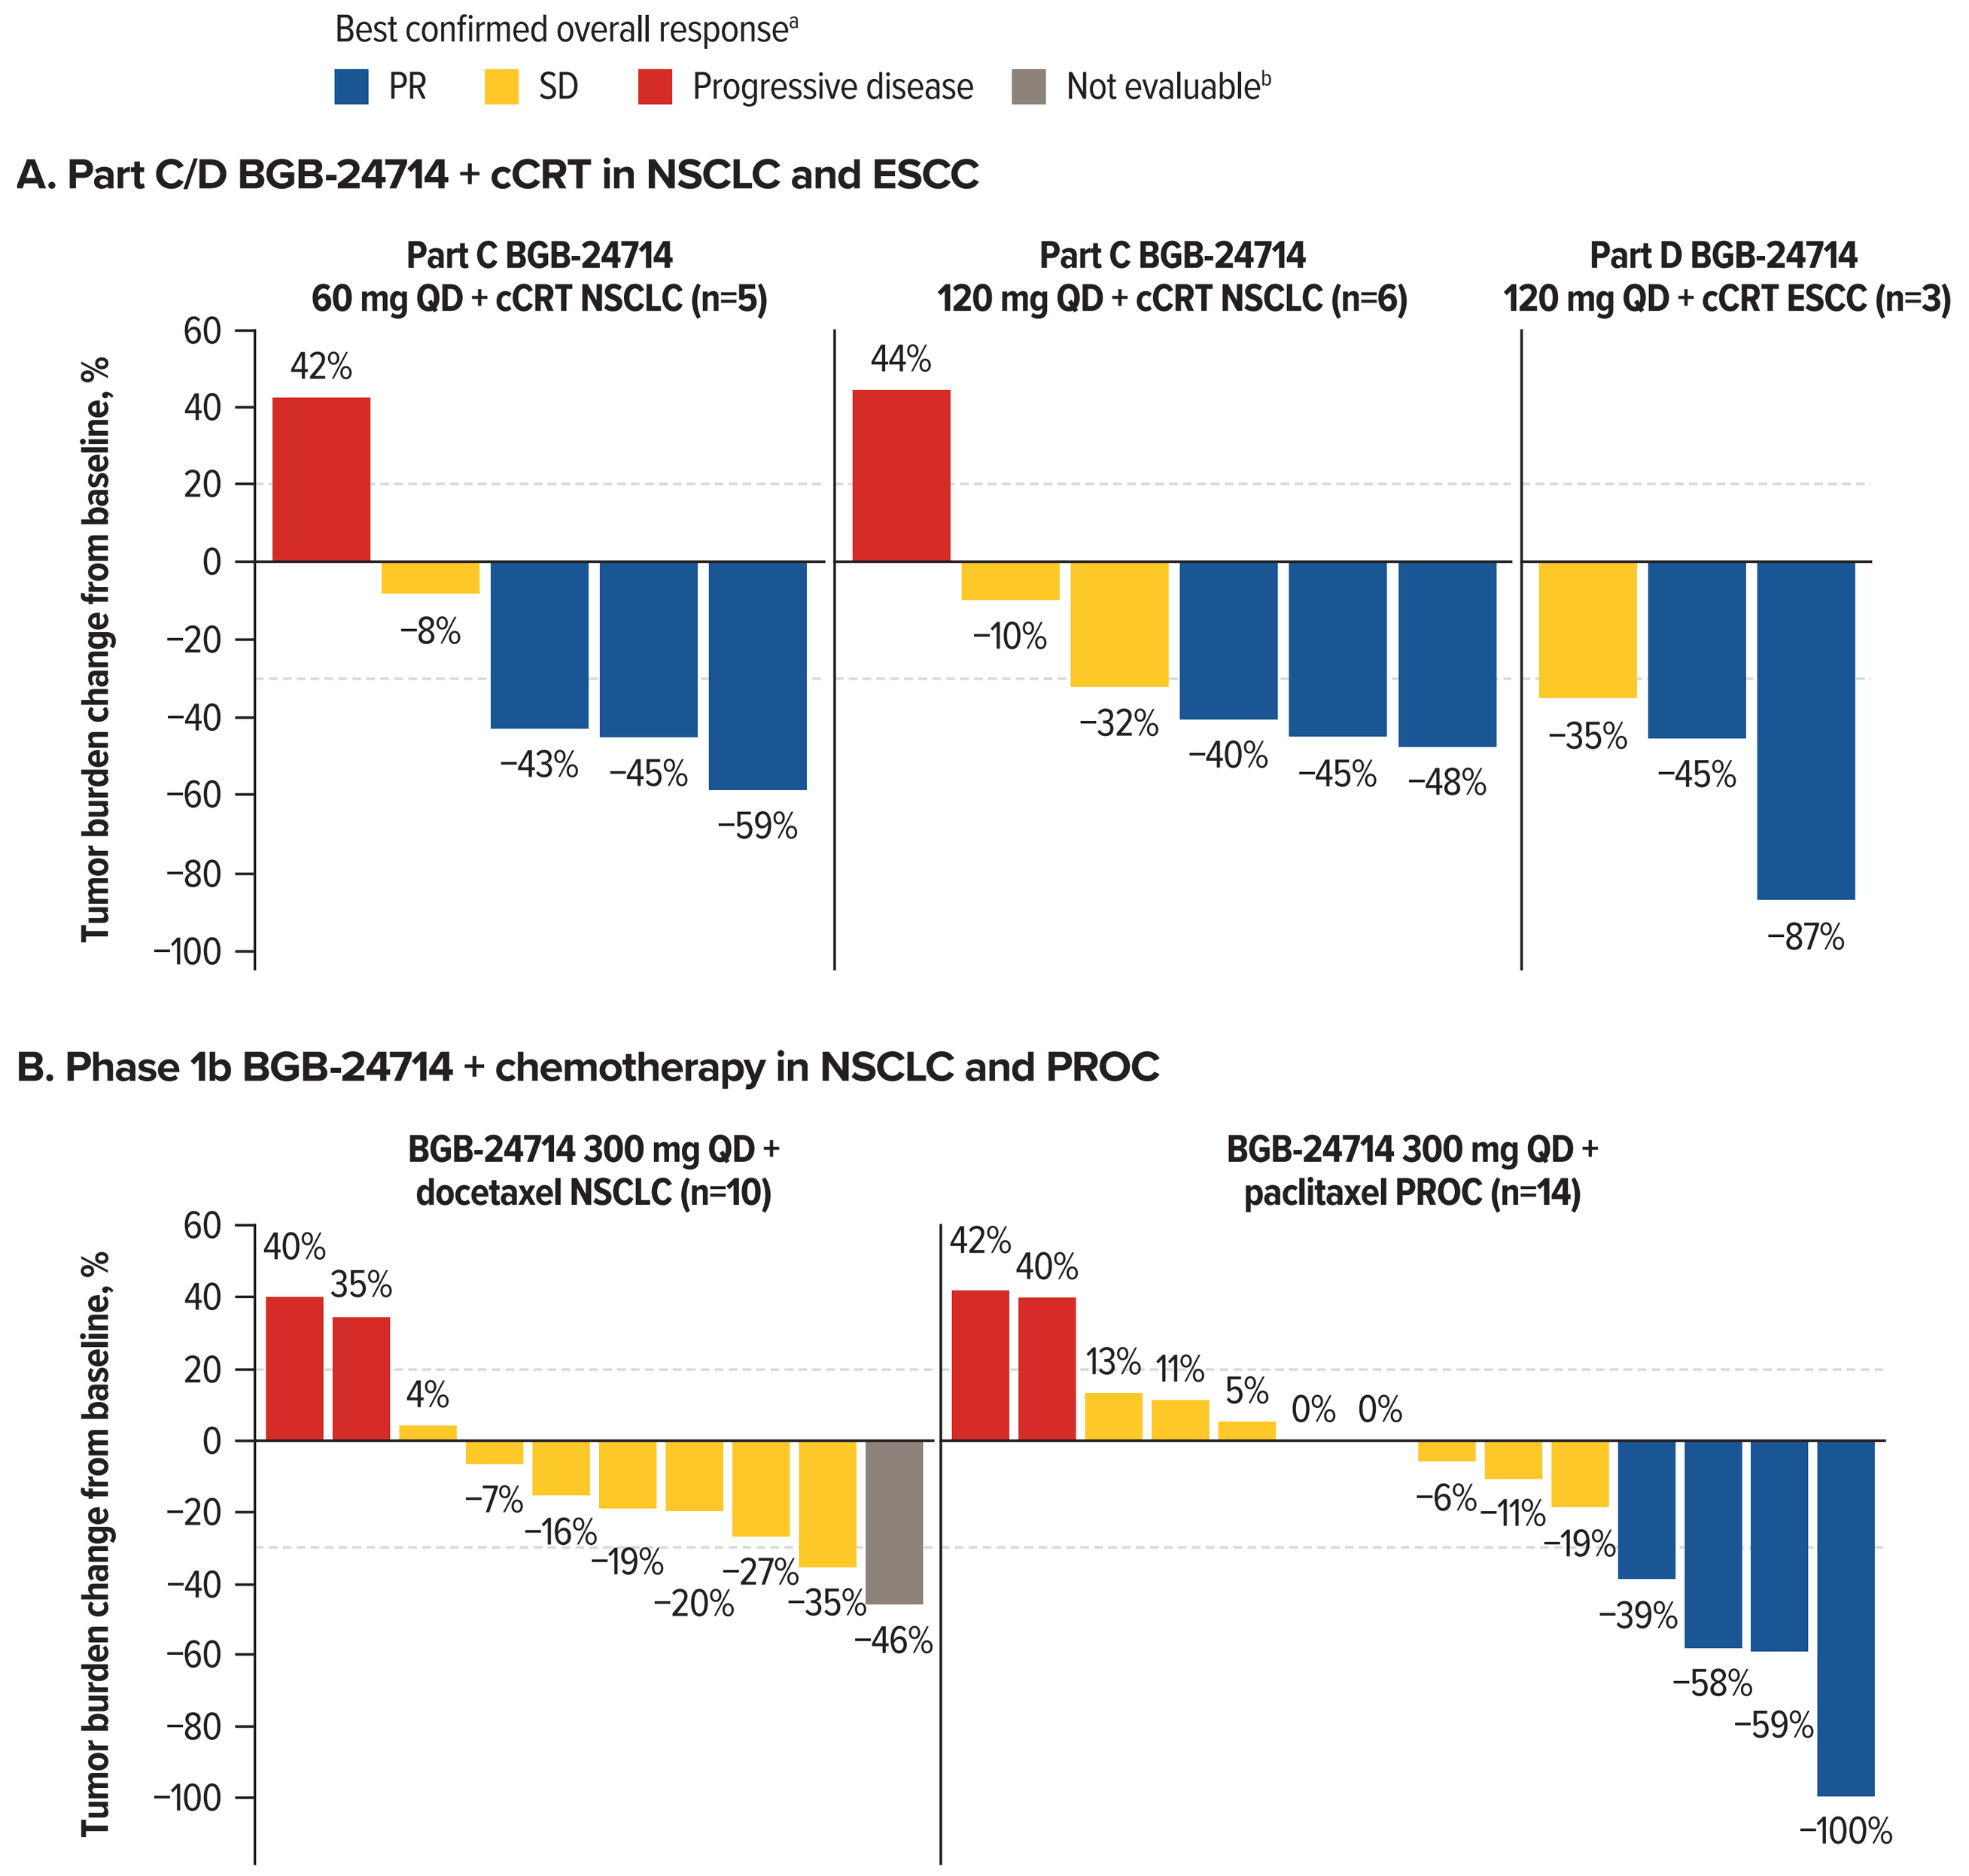
Best confirmed overall response[a]
PR
SD
Progressive disease
Not evaluable[b]
A. Part C/D BGB-24714 + cCRT in NSCLC and ESCC
Part C BGB-24714 60 mg QD + cCRT NSCLC (n=5)
Part C BGB-24714 120 mg QD + cCRT NSCLC (n=6)
Part D BGB-24714 120 mg QD + cCRT ESCC (n=3)
Tumor burden change from baseline, %
42% −8% −43% −45% −59%
44% −10% −32% −40% −45% −48%
−35% −45% −87%
B. Phase 1b BGB-24714 + chemotherapy in NSCLC and PROC
BGB-24714 300 mg QD + docetaxel NSCLC (n=10)
BGB-24714 300 mg QD + paclitaxel PROC (n=14)
Tumor burden change from baseline, %
40% 35% 4% −7% −16% −19% −20% −27% −35% −46%
42% 40% 13% 11% 5% 0% 0% −6% −11% −19% −39% −58% −59% −100%

[a]Confirmed response means that the initial observation of tumor response is verified by a follow-up scan. [b]One patient had a single tumor assessment and PR was not confirmed.

REFERENCES

1. Morrish E, et al. *Cells*. 2020;9:406.
2. Liu FR, et al. *ESMO Open*. 2024;9:103651.
3. Patel MR, et al. *Cancer Med*. 2025;14:e71451.
4. Li J, et al. *Cancer Res*. 2023;83:Abstract nr 6158.
5. Zhang J, et al. *Cancer Res.* 2026; AACR 2026 Poster CT121

DISCLOSURES

HXG: Employment: BeOne Medicines; Stock: BeOne Medicines.

ACKNOWLEDGMENTS

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This study was sponsored by BeOne Medicines, Ltd. Medical writing was provided by Teal M. Brechtel, PhD of Amiculum, and supported by BeOne Medicines.

PRESENTER: Hua-Xin Gao, hua-xin.gao@beonemed.com
CORRESPONDENCE: Judy S. Wang, JSWang@flcancer.com

Presented at American Association for Cancer Research Annual Meeting, San Diego, CA, USA, April 17-22, 2026