BTK-A428D is a Cross-resistant Mutation to Both BTK Inhibitors and Degraders

Haitao Wang[1]*, Lin Li[1]*, Xiaoyu Feng[2]*, Yiwen Wang[1], Weiwei Song[1], Shuang Peng[1], Yangyang Wang, Sijia Zhai[1], Ziyu Jia[1], Peng Chi[1], Teiko Sumiyoshi[3], Yangbo Yue[2], Wei Jin[1$], Zhirong Shen[2$]
1. BeiGene (Beijing) Co., Ltd., Beijing 102206, China; 2. BeiGene (Shanghai) Co., Ltd., Shanghai 200131, China;3. BeiGene USA Inc., San Mateo, CA USA
* Equal contribution; $ correspondence

INTRODUCTION

BTK inhibitors have revolutionized the management of chronic lymphocytic leukemia (CLL) and other B cell malignancies. However, acquired resistant mutation of BTK develops.

Mutations at Cysteine 481 (C481) result in resistance to covalent BTK inhibitors (BTKi). Non-covalent BTKis, such as pirtobrutinib, can overcome C481 mutations but are susceptible to other BTK mutations like V416L, T474I, and L528W.

Recently, a BTK mutation A428D was reported to confer resistance from the BGB-16673 trial and was also modeled to disrupt the binding of BTK to another BTK-degrader, NX-2127 [1]. This raises the question of whether A428D insensitivity is compound specific or common to BTK degraders and inhibitors currently in development and market. This study investigates the impact of BTK A428D on the response to approved BTK inhibitors and multiple degraders via preclinical models.

METHOD

TMD8 cells overexpressing BTK-wildtype, C481S, T474I, A428D, V416L, M437R mutations were generated by lentivirus transduction and utilized to evaluate their sensitivity to individual BTK degrader or inhibitor via *in vitro* CTG assay.

TMD8 BTK-A428D cell lines, which were generated by CRISPR-Cas9 mediated *in situ* gene editing, were used for BTK degradation examination by ELISA, BCR signalling evaluation by Western blot, and *in vivo* tumour growth inhibition (TGI) assessment via xenograft models.

REFERENCES

[1] Wong, R.L., Choi, M.Y., Wang, HY. et al. Mutation in Bruton Tyrosine Kinase (BTK) A428D confers resistance To BTK-degrader therapy in chronic lymphocytic leukemia. Leukemia 38, 1818–1821 (2024). https://doi.org/10.1038/s41375-024-02317-4

RESULTS

Figure 1. BTK-A428D is a cross resistant mutation to BTK inhibitors and degraders

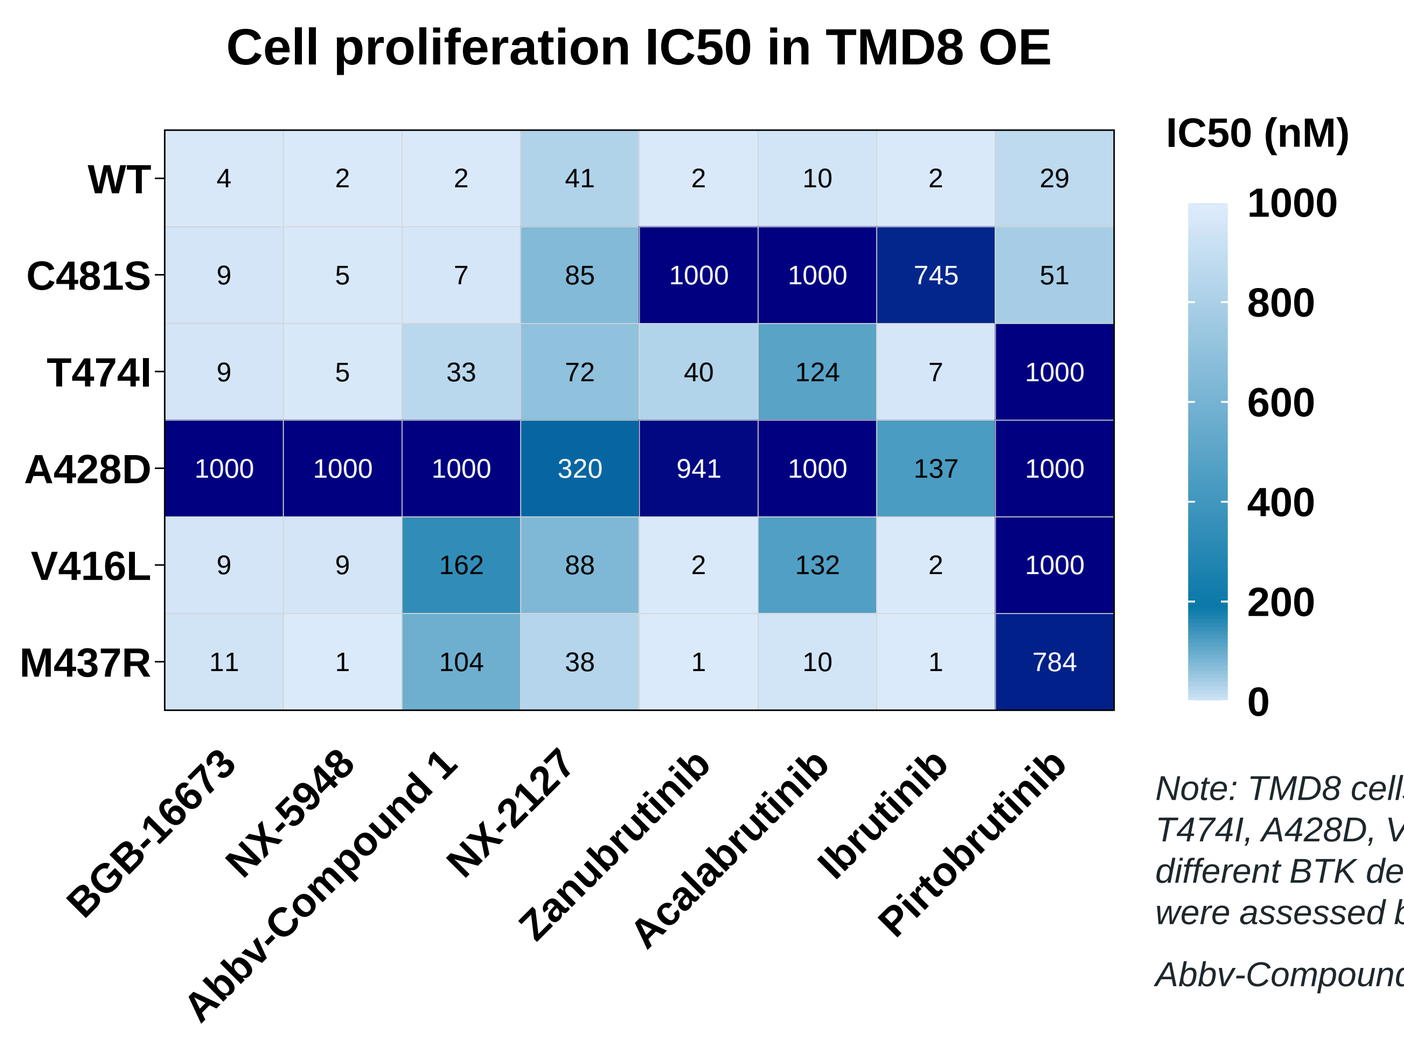
Cell proliferation IC50 in TMD8 OE
IC50 (nM)
WT
C481S
T474I
A428D
V416L
M437R
BGB-16673
NX-5948
Abbv-Compound 1
NX-2127
Zanubrutinib
Acalabrutinib
Ibrutinib
Pirtobrutinib

Note: TMD8 cells overexpressing BTK-wildtype, C481S, T474I, A428D, V416L, M437R mutations were treated by different BTK degrader or inhibitors, and the cell viability were assessed by CTG assay.

Abbv-Compound 1 is example 1 in WO 2023183811 A1

Among the four tested BTK degraders, which are BGB-16673, NX-2127, NX-5948, and Abbv-Compound 1: **1)** BGB-16673 and NX-5948 potently inhibited the growth of TMD8 cells overexpressing BTK-C481S, T474I, V416L, M437R mutations measured by *in vitro* CTG assay; **2)** BGB-16673 is more potent than Abbv-Compound 1 against BTK-T474I, -V416L, and -M437R overexpressing TMD8 cells; **3)** NX-2127 is generally less potent in all the mutations. However, TMD8 cells overexpressing BTK-A428D are resistant to all tested BTK degraders and inhibitors (ibrutinib, acalabrutinib, zanubrutinib, pirtobrutinib).

Figure 2. TMD8 cells with BTK-A428D knock in (KI) mutation confirmed the resistance to BTK inhibitors and degraders

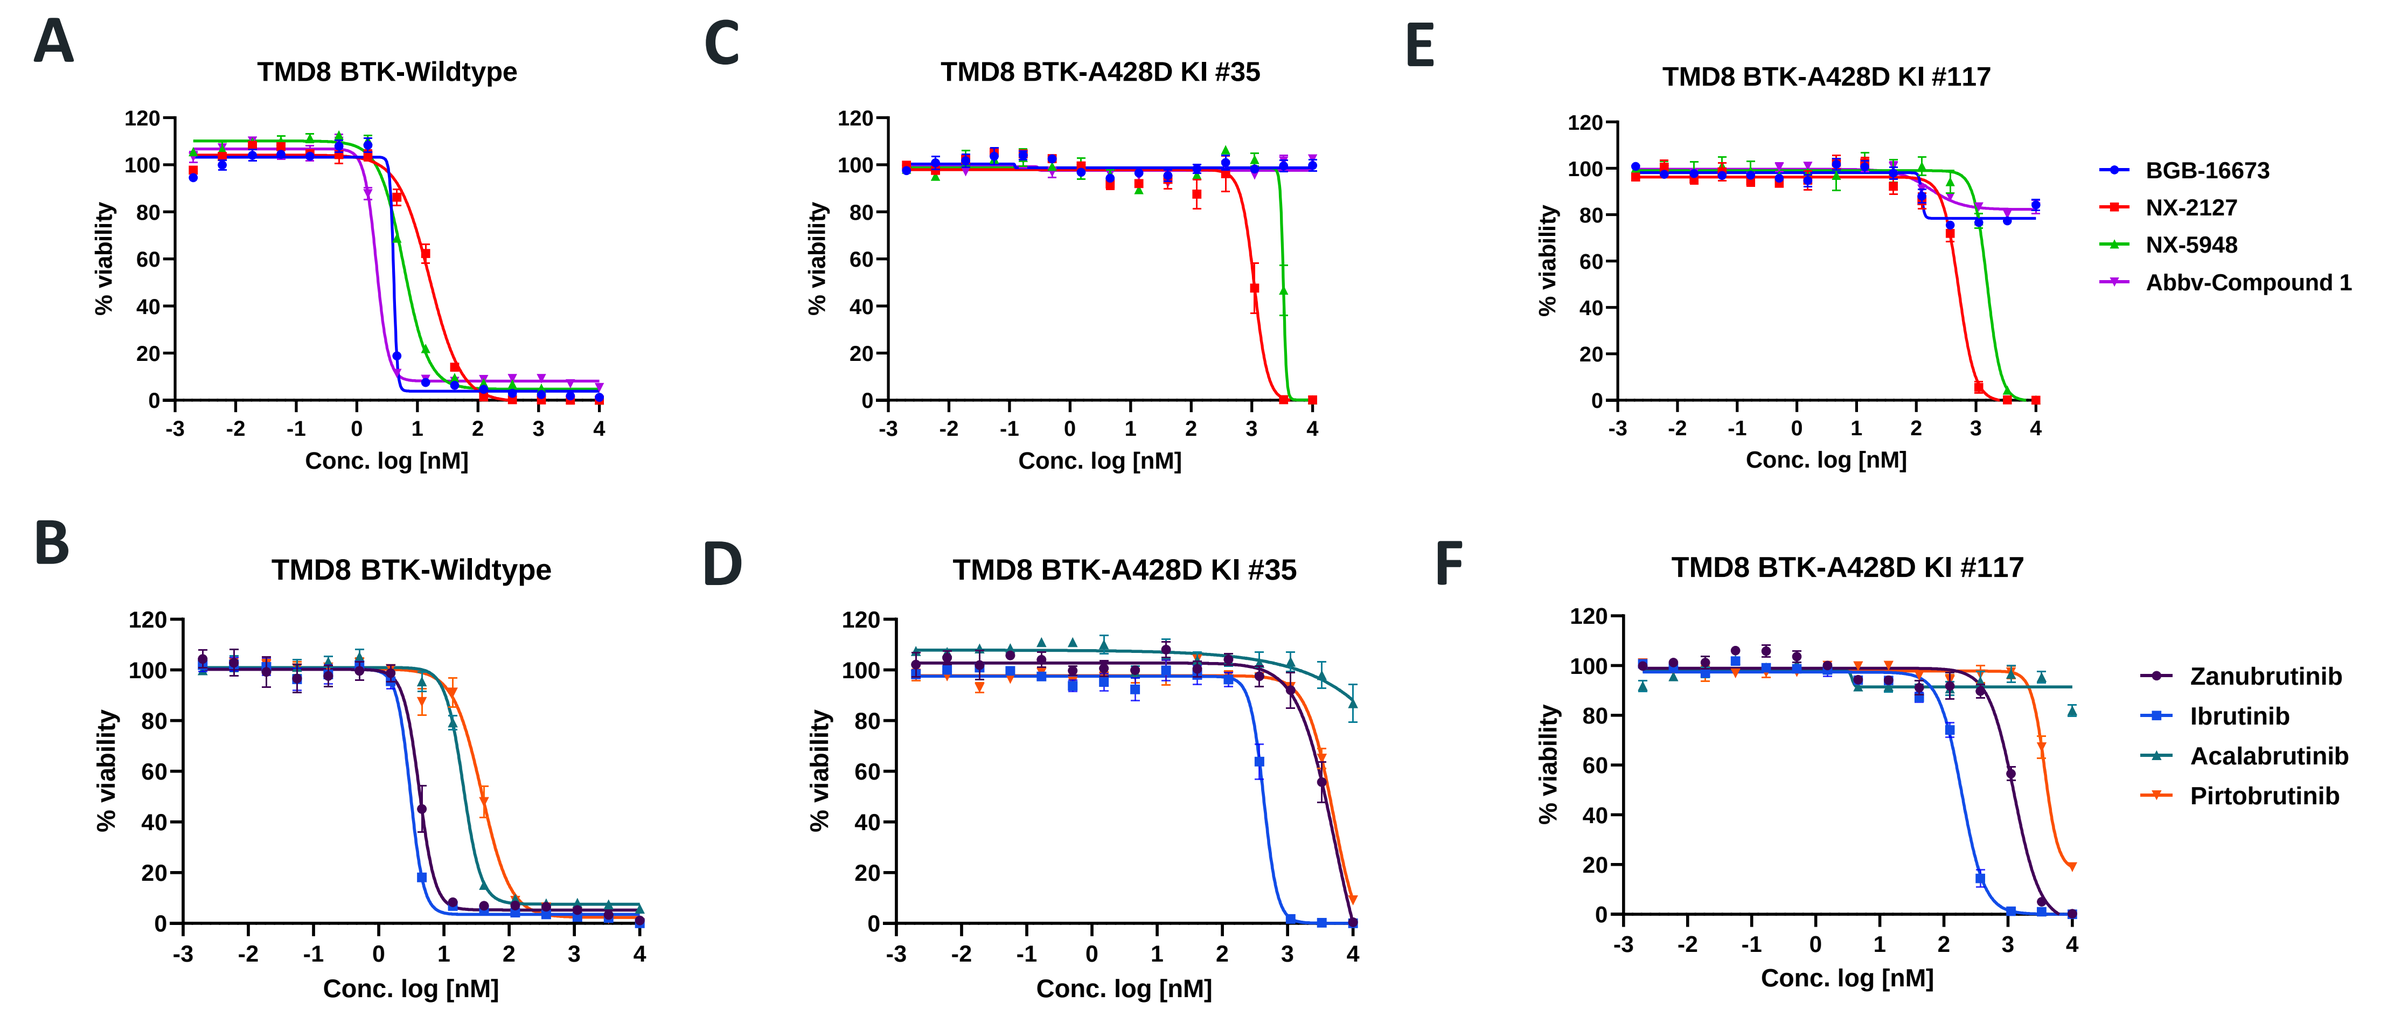
A
TMD8 BTK-Wildtype
C
TMD8 BTK-A428D KI #35
E
TMD8 BTK-A428D KI #117
BGB-16673
NX-2127
NX-5948
Abbv-Compound 1
B
TMD8 BTK-Wildtype
D
TMD8 BTK-A428D KI #35
F
TMD8 BTK-A428D KI #117
Zanubrutinib
Ibrutinib
Acalabrutinib
Pirtobrutinib
% viability
Conc. log [nM]

G

TMD8 Cells	IC50 (nM)							
	BGB-16673	NX-2127	NX-5948	Abbv-Compound1	Zanu	Ibru	Acala	Pirto
BTK-WT	4.058	15.87	5.921	2.108	4.186	3.044	19.75	38.07
BTK-A428D KI 35#	>10,000	1099	3312	>10,000	5225	431.8	>10,000	4975
BTK-A428D KI 117#	>10,000	512.6	1548	>10,000	1256	189.6	>10,000	3742

Two TMD8 BTK-A428D cell lines generated by CRISPR-Cas9 mediated *in situ* mutation were treated by BTK degraders and inhibitors to confirm their response to different drugs. **A-B).** In parental TMD8 cells expressing wildtype BTK, cellular growth was efficiently inhibited by both BTK degraders and inhibitors; **C-F).** Neither four BTK degraders, nor the four tested BTK inhibitors could efficiently inhibit the growth of two TMD8 BTK-A428D knock in cell clones (Clone #35 and Clone #117), confirming their resistance to BTK-A428D mutations observed in OE cells in Figure 1; **G).** A table showing the number of half maximum inhibition concentration (IC50) of different compounds from A to F.

Figure 3. BTK degradation by degraders is blocked by A428D mutation

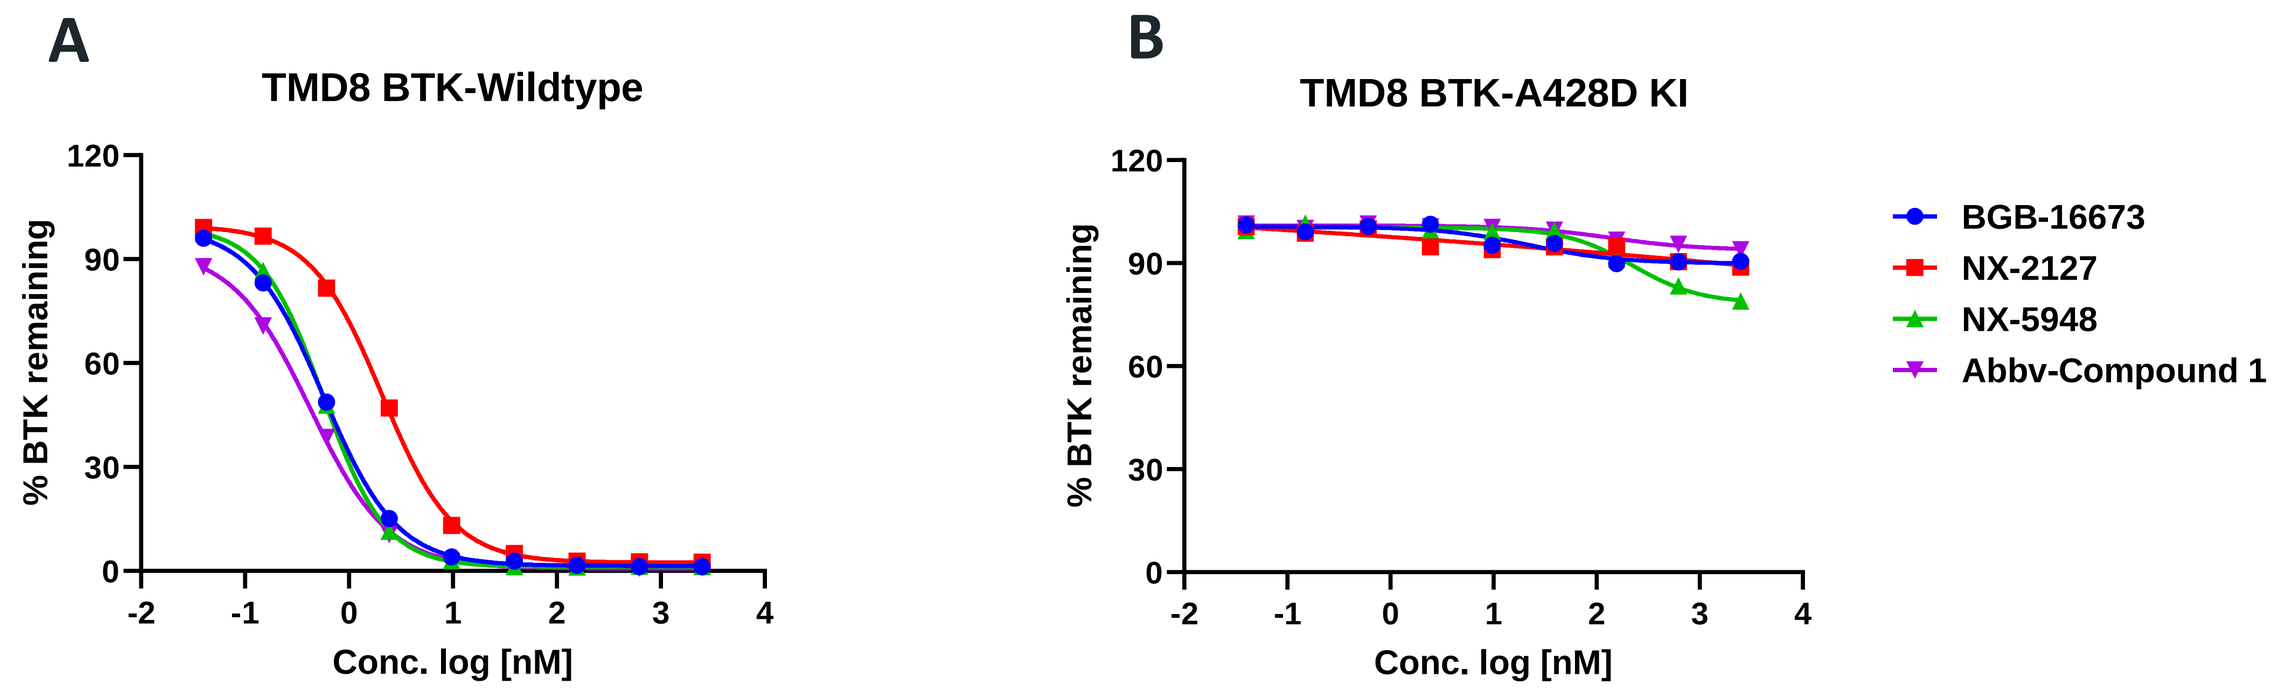
A
TMD8 BTK-Wildtype
B
TMD8 BTK-A428D KI
% BTK remaining
Conc. log [nM]
BGB-16673
NX-2127
NX-5948
Abbv-Compound 1

C

	TMD8 BTK-Wildtype				TMD8 BTK-A428D KI			
	BGB-16673	NX-2127	NX-5948	Abbv-Compound 1	BGB-16673	NX-2127	NX-5948	Abbv-Compound 1
DC50(nM)	0.5713	2.081	0.5647	0.4252	>1000	>1000	>1000	>1000

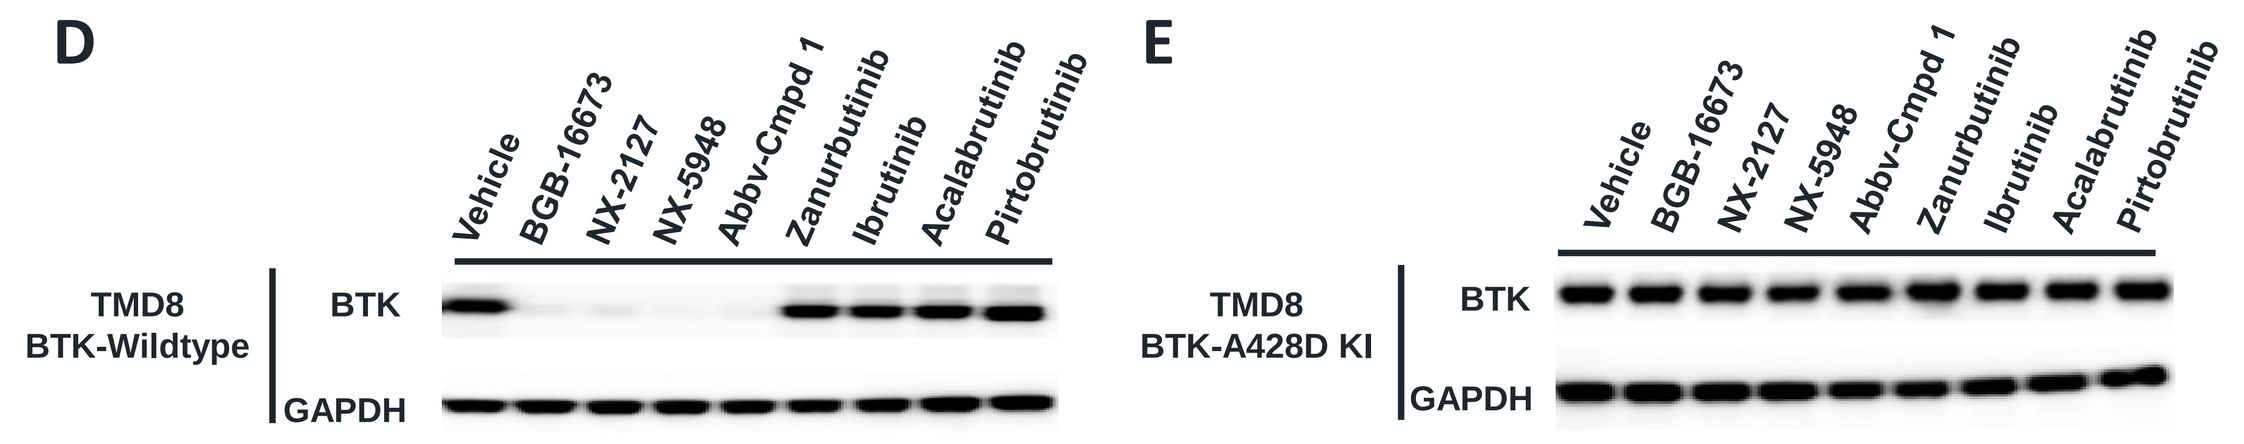
D
E
TMD8 BTK-Wildtype
TMD8 BTK-A428D KI
BTK
GAPDH
Vehicle
BGB-16673
NX-2127
NX-5948
Abbv-Cmpd 1
Zanurbutinib
Ibrutinib
Acalabrutinib
Pirtobrutinib

BTK degradation were examined by ELISA and western blot assay in TMD8 wildtype and BTK-A428D knock-in cells. **A).** BGB-16673, NX-5948, and Abbv-Compound 1 potently degraded wildtype BTK in TMD8 cells, NX-2127 also efficiently degraded wildtype BTK, but showed lower potency compared with other degraders. **B).** None of the BTK degraders showed significant degradation on BTK-A428D protein, even at 1,000 nM. **C).** A table shows the half degradation concentration (DC50) in figure A, B. **D-E).** TMD8 wildtype or BTK-A428D KI cells were treated by different compounds at 100nM for 24h, then protein lysates were used for western blot. Significant BTK degradation was only observed by different BTK degraders in wildtype cells, and the degradation was blocked in BTK-A428D KI cells. As expected, no BTK degradation was observed for BTK inhibitors.

Figure 4. BTK-A428D is a kinase-impaired mutation, but maintains BCR downstream signaling

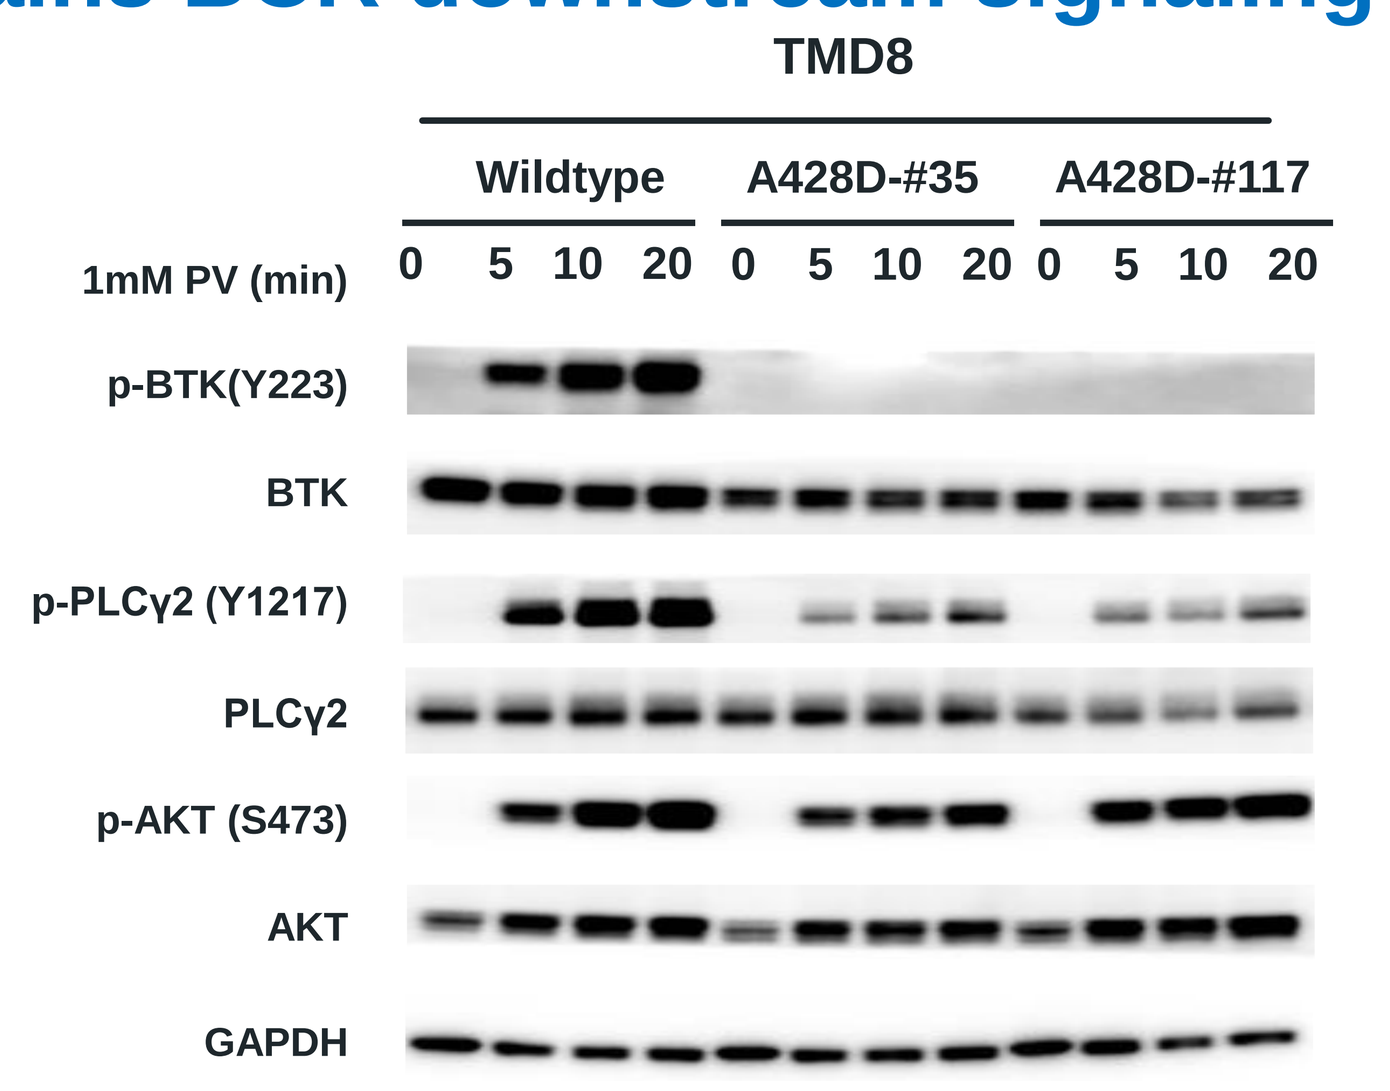
TMD8
Wildtype
A428D-#35
A428D-#117
1mM PV (min)
0 5 10 20 0 5 10 20 0 5 10 20
p-BTK(Y223)
BTK
p-PLCγ2 (Y1217)
PLCγ2
p-AKT (S473)
AKT
GAPDH

TMD8 wildtype and two BTK-A428D *in situ* mutated clones (Clone #35 and Clone #117) were used to examine BTK kinase activity with or without 1mM pervanadate (PV) stimulation. A time-dependent increase of phosphorylation signal at BTK-Y223, PLCγ2 - Y1217, and AKT-S473 were observed in TMD8 BTK-wildtype cells, suggesting an intact kinase activity of BTK and its downstream signals. However, no phosphorylation of BTK-Y223 can be observed in BTK-A428D cells, indicating A428D is a kinase impaired mutation. Consistently, phosphorylation at PLCγ2 is also significantly abrogated. Phosphorylation at AKT-S473 is less disrupted, suggesting the BCR downstream signaling retains in mutant.

Figure 5. BTK inhibitors and degraders are ineffective in inhibiting BTK downstream signaling in BTK-A428D mutated cells

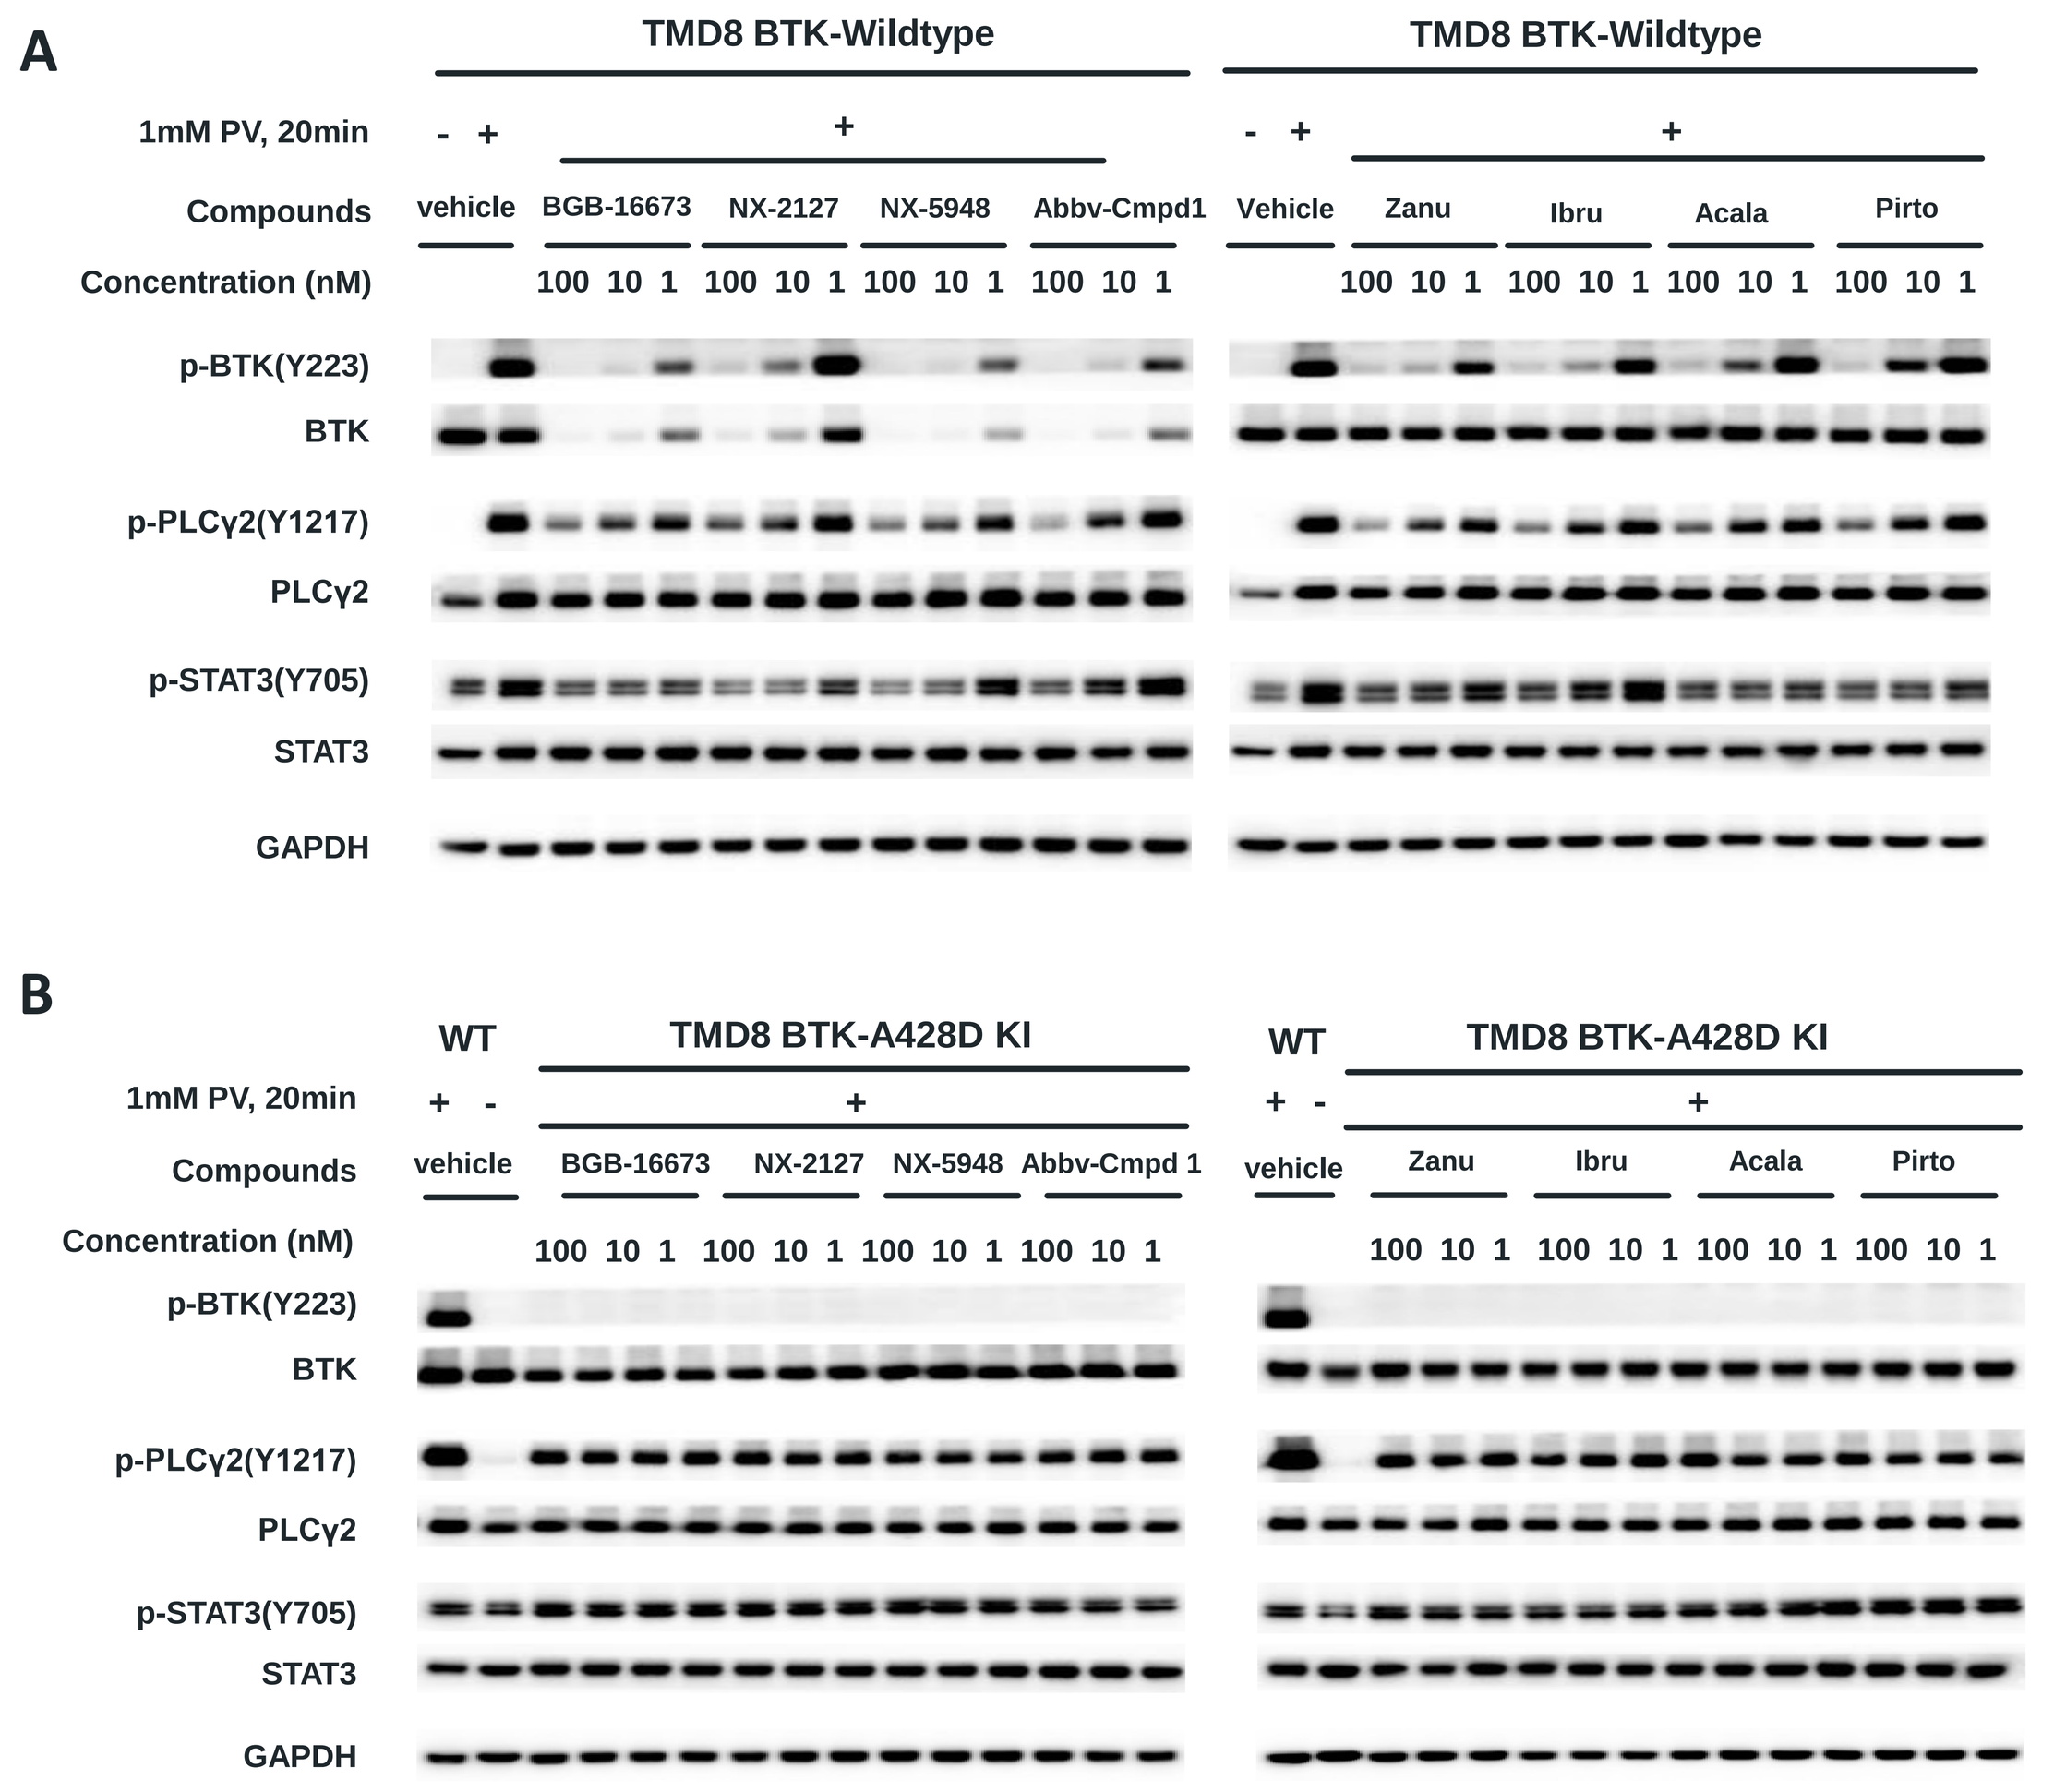
A
TMD8 BTK-Wildtype
TMD8 BTK-Wildtype
1mM PV, 20min
Compounds
Concentration (nM)
vehicle
BGB-16673
NX-2127
NX-5948
Abbv-Cmpd1
Vehicle
Zanu
Ibru
Acala
Pirto
p-BTK(Y223)
BTK
p-PLCγ2(Y1217)
PLCγ2
p-STAT3(Y705)
STAT3
GAPDH
B
WT
TMD8 BTK-A428D KI
WT
TMD8 BTK-A428D KI
Abbv-Cmpd 1

TMD8 wildtype and BTK-A428D knock-in cells were treated with compounds at indicated concentrations for 24 hours, then cells were stimulated with 1mM PV for 20min. Cell lysates were used for western blot. **A).** BTK was dose dependently degraded by BTK degraders (BGB-16673, NX-5948, NX-2127, Abbv-Compound 1) but not BTK inhibitors (zanu, ibru, acala, pirto). The phosphorylation of BTK, PLCγ2 and STAT3 were dose dependently inhibited by both BTK degraders and inhibitors, suggesting efficient inhibition of BTK and its downstream signal by both compounds. **B).** No obvious degradation of BTK-A428D was observed by different degraders, indicating the mutant is resistant to all BTK degraders. No BTK-Y223 phosphorylation can be observed, confirming it a kinase impaired mutation. None of the compounds significantly inhibited the phosphorylation of PLCγ2 and STAT3, suggesting they are ineffective in inhibiting downstream BCR signals in A428D mutant cells.

Figure 6. BTK inhibitors and degraders showed no obvious anti-tumor activity in BTK-A428D xenografts

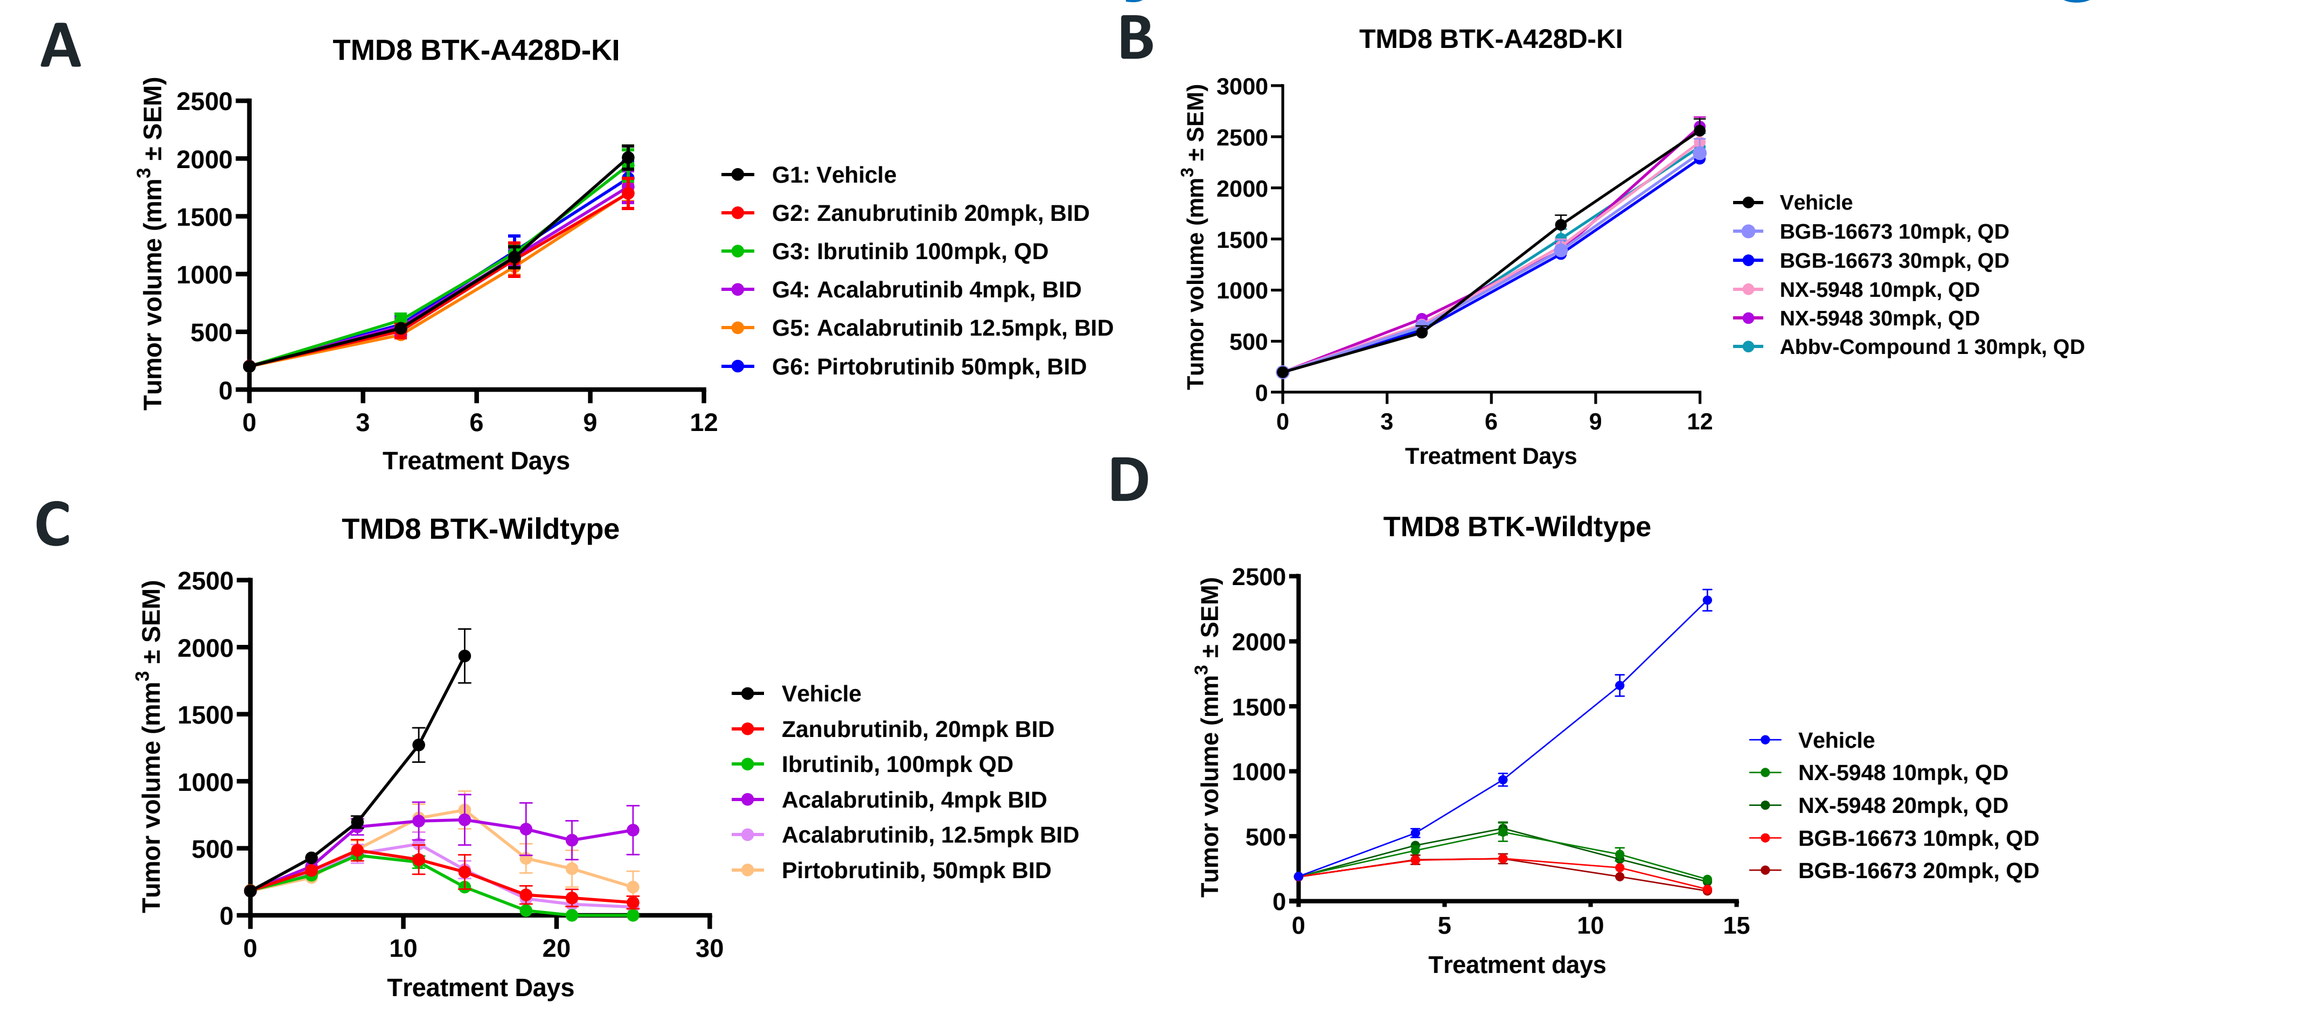
A
TMD8 BTK-A428D-KI
G1: Vehicle
G2: Zanubrutinib 20mpk, BID
G3: Ibrutinib 100mpk, QD
G4: Acalabrutinib 4mpk, BID
G5: Acalabrutinib 12.5mpk, BID
G6: Pirtobrutinib 50mpk, BID
B
TMD8 BTK-A428D-KI
Vehicle
BGB-16673 10mpk, QD
BGB-16673 30mpk, QD
NX-5948 10mpk, QD
NX-5948 30mpk, QD
Abbv-Compound 1 30mpk, QD
C
TMD8 BTK-Wildtype
Vehicle
Zanubrutinib, 20mpk BID
Ibrutinib, 100mpk QD
Acalabrutinib, 4mpk BID
Acalabrutinib, 12.5mpk BID
Pirtobrutinib, 50mpk BID
D
TMD8 BTK-Wildtype
Vehicle
NX-5948 10mpk, QD
NX-5948 20mpk, QD
BGB-16673 10mpk, QD
BGB-16673 20mpk, QD
Tumor volume (mm³ ± SEM)
Treatment Days

Note: zanubrutinib, ibrutinib, acalabrutinib at 20mpk BID, 100mg QD, 4mpk BID are their clinic relevant doses in mouse. High dose of pirtobrutinib and degraders were used based on published data.

A-B). No anti-tumor activity was observed for both BTK degraders and inhibitors in TMD8 BTK-A428D xenografts *in vivo*. **C-D).** Compounds showed significant anti-tumor activity in wildtype xenografts.

CONCLUSIONS

BTK-A428D is a mutation that exhibits cross-resistance to multiple BTK degraders (BGB-16673, NX-2127, NX-5948, Abbv-Compound 1) and inhibitors (ibrutinib, acalabrutinib, zanubrutinib, and pirtobrutinib) both *in vitro* and *in vivo*, as supported by evidence from multiple aspects.

ACKNOWLEDGEMENT

We thank Dr. Yongchun Tong and Dr. Lanjun Xu for the help in the poster.