RATIONALE-307 Long-term Outcomes: First-line Tislelizumab (TIS) Plus Chemotherapy (chemo) vs Chemo Alone for Advanced Squamous (sq) NSCLC

Zhijie Wang[1], Xinmin Yu[2], Jun Zhao[3], Yan Yu[4], Jingxun Wu[5], Rui Ma[6], Zhiyong Ma[7], Jiuwei Cui[8], Na Zhao[9], Liang Liang[10], Jinghui Fan[10], Jie Wang[1]

[1]Department of Medical Oncology, Cancer Hospital Chinese Academy of Medical Sciences and Peking Union Medical College, Beijing, China; [2]Department of Medical Oncology, Zhejiang Cancer Hospital, Institute of Basic Medicine and Cancer (IBMC), Chinese Academy of Sciences, Hangzhou, China; [3]Department of Medical Oncology, Beijing Cancer Hospital, Beijing, China; [4]Department of Medical Oncology, Harbin Medical University Cancer Hospital, Harbin, China; [5]Department of Medical Oncology, The First Affiliated Hospital of Xiamen University, Xiamen, China; [6]Department of Chest Internal Medicine, Liaoning Cancer Hospital & Institute, Shenyang, China; [7]Department of Medical Oncology, The Affiliated Cancer Hospital of Zhengzhou University/Henan Cancer Hospital, Zhengzhou, China; [8]Cancer Center Department, The First Hospital of Jilin University, Changchun, China; [9]BeiGene (Shanghai) Co., Ltd., Shanghai, China; [10]BeiGene (Beijing) Co., Ltd., Beijing, China

FPN: 1323P

13-17 SEP 2024
ESMO CONGRESS Barcelona Spain

BACKGROUND

- Tislelizumab is a humanized monoclonal antibody with high affinity and binding specificity for PD-1, and was specifically engineered to minimize Fcγ receptor binding on macrophages.[1,2] It has demonstrated survival benefits across a variety of advanced solid tumors, including non-small cell lung cancer (NSCLC).[3-7]
- In the phase 3 RATIONALE-307 study (NCT03594747), tislelizumab + chemotherapy (chemo) significantly extended progression-free survival (PFS) vs chemo as first-line treatment in patients with advanced squamous NSCLC.[3]
- This study led to the approval of tislelizumab + chemo as a standard of care for squamous NSCLC in the first-line setting in China and Europe.
- Here we report the updated outcomes of RATIONALE-307 with approximately 4 years of follow-up.

METHODS

- Eligible patients with previously untreated advanced squamous NSCLC were randomized 1:1:1 to receive tislelizumab + chemo (two arms: tislelizumab + PC [paclitaxel and carboplatin], tislelizumab + nPC [nab-paclitaxel and carboplatin]) or chemo (PC) alone **(Fig. 1).**
- Patients allocated to the chemo arm were allowed to cross over to tislelizumab monotherapy upon disease progression.
- Patients with long-term exposure (LTE) to tislelizumab were defined as those who received ≥35 cycles of tislelizumab treatment.
- Biomarker testing was performed on baseline tumor samples, including PD-L1 protein expression (VENTANA PD-L1 [SP263] assay), tumor mutational burden (TMB), genomic alterations (OncoScreen Plus), and gene expression profiling (EdgeSeq Precision IO Panel).

Figure 1. Study design

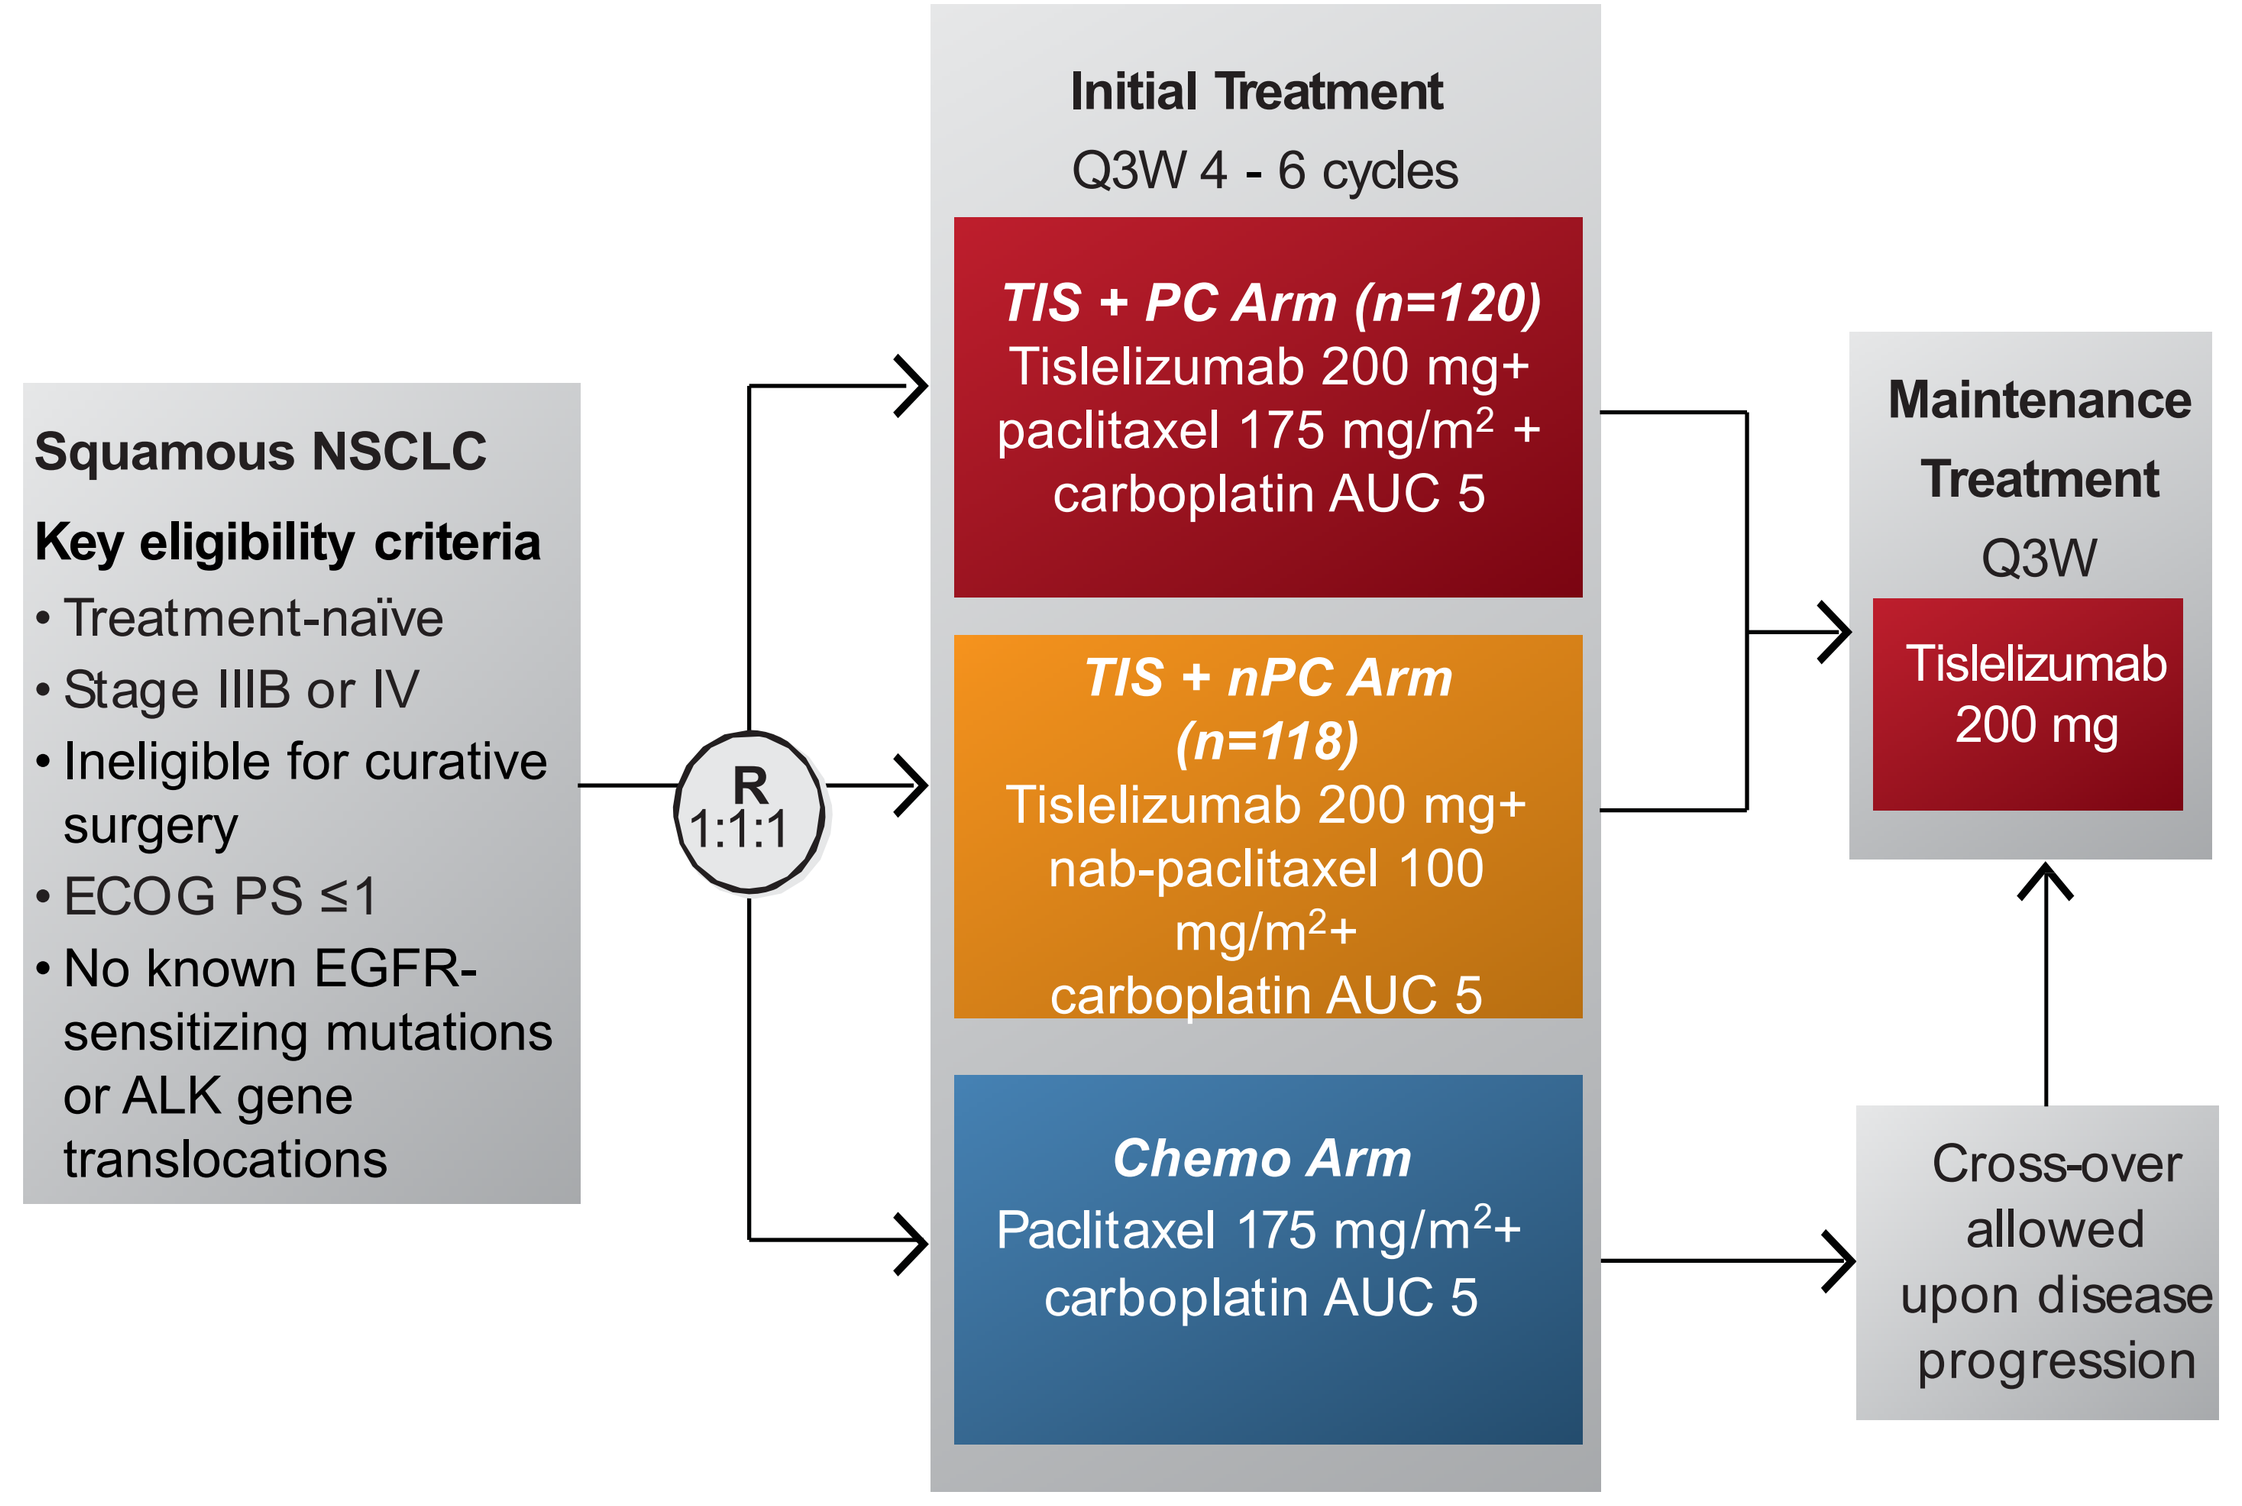

Squamous NSCLC
Key eligibility criteria
• Treatment-naïve
• Stage IIIB or IV
• Ineligible for curative surgery
• ECOG PS ≤1
• No known EGFR-sensitizing mutations or ALK gene translocations
R 1:1:1
Initial Treatment
Q3W 4 - 6 cycles
TIS + PC Arm (n=120)
Tislelizumab 200 mg+ paclitaxel 175 mg/m² + carboplatin AUC 5
TIS + nPC Arm (n=118)
Tislelizumab 200 mg+ nab-paclitaxel 100 mg/m²+ carboplatin AUC 5
Chemo Arm
Paclitaxel 175 mg/m²+ carboplatin AUC 5
Maintenance Treatment
Q3W
Tislelizumab 200 mg
Cross-over allowed upon disease progression

Stratification factors
- Stage (IIIB vs IV)
- PD-L1 TC (<1% vs 1%-49% vs ≥50%)

- **Primary endpoint: PFS per IRC**
- **Key secondary endpoints: ORR, DoR, OS, and safety**

Abbreviations

AUC, Area Under the Curve; Chemo, chemotherapy; CI, confidence interval; D, day; DoR, duration of response; ECOG PS, Eastern Cooperative Oncology Group; HR, hazard ratio; im-AE, immune-mediated adverse event; IRC, independent review committee; ITT, intention-to-treat; LTE, long-term exposure; mo, months; nPC, nab-paclitaxel and carboplatin; NSCLC, non-small cell lung cancer; ORR, objective response rate; OS, overall survival; PC, paclitaxel and carboplatin; PD, progressive disease; PD-L1, programmed death-ligand 1; PFS, progression-free survival; Q3W, every 3 weeks; R, randomized; RECIST, Response Evaluation Criteria in Solid Tumors; sq, squamous; TC, tumor cell; TIS, tislelizumab; TMB, tumor mutation burden.

References

1. Zhang T, et al. *Cancer Immunol Immunother*. 2018;67:1079-1090.
2. Hong Y, et al. *FEBS Open Bio*. 2021;11:782-792.
3. Wang J, et al. *JAMA Oncol*. 2021 May 1;7(5):709-717
4. Lu S, et al. *J Thorac Oncol*. 2021 Sep;16(9):1512-1522.
5. Xu J, et al. *Lancet Oncol*. 2023; 24: 483–95.
6. Qiu M, et al. *BMJ*. 2024 May 28:385:e078876.
7. Cheng Y, et al. *J Thorac Oncol*. 2024 Jul;19(7):1073-1085.

Acknowledgments: We would like to thank the investigators, the site support staff, and especially the patients for participating in this study. This study was sponsored by BeiGene.

Declaration of interests: Dr Zhijie Wang declares no potential conflict of interest.

CONCLUSIONS

- **Tislelizumab + chemo maintained long-term clinical meaningful survival benefits compared with chemo alone, with no new safety signals, despite a high in-study cross-over rate of 58.7%**
 - **4-yr OS rates: 32.2% with tislelizumab + PC vs 26.0% with tislelizumab + nPC vs 19.2% with chemo alone**
- **Patients with long-term exposure (≥35 cycles) to tislelizumab achieved high ORR and long-term survival, with higher expression of PD-L1 and T cell inflammation signature, as well as a FAT1 enriched mutational profile**

RESULTS

Patients

- 360 patients were randomly assigned (tislelizumab + PC, n=120; tislelizumab + nPC, n=119; chemo, n=121).
- As of April 28, 2023, median time from randomization to data cutoff was 50.3 mo (range, 46.5-57.0).

Updated efficacy and safety

- Clinically meaningful OS improvement with tislelizumab + chemo was well maintained vs. chemo alone with extended follow-up
 - Tislelizumab + PC vs. chemo: median 26.1 mo vs 19.4 mo; HR 0.67 (95% CI, 0.49 - 0.92) **(Fig. 2A)**
 - Tislelizumab + nPC vs. chemo: median 23.3 mo vs 19.4 mo; HR 0.82 (95% CI, 0.60 - 1.11) **(Fig. 2B)**
- OS benefit with tislelizumab + chemo was sustained, with 4-year OS rates of 32.2% (95% CI, 23.8 - 40.9) and 26.0% (95% CI, 18.3 - 34.4), versus 19.2% (95% CI, 12.0 - 27.7), respectively (**Fig. 2**).
- In the chemo arm, 77 (63.6%) patients received subsequent anti-PD-(L)1 therapy, including 71 (58.7%) who crossed over to tislelizumab in-study; analyses adjusted for in-study cross-over effect with the two-stage method further confirmed the OS benefits with tislelizumab + chemo (cross-over adjusted median OS of chemo: 16.0 mo; HRs 0.53 and 0.65, respectively; **Fig. 3A & B**).
- PFS benefits with tislelizumab + chemo vs. chemo were also maintained at this updated analysis (**Fig. 4**).
- Tislelizumab plus chemotherapy was tolerable, with no new safety signals identified with extended follow-up.

Figure 2. Kaplan-Meier curves of OS

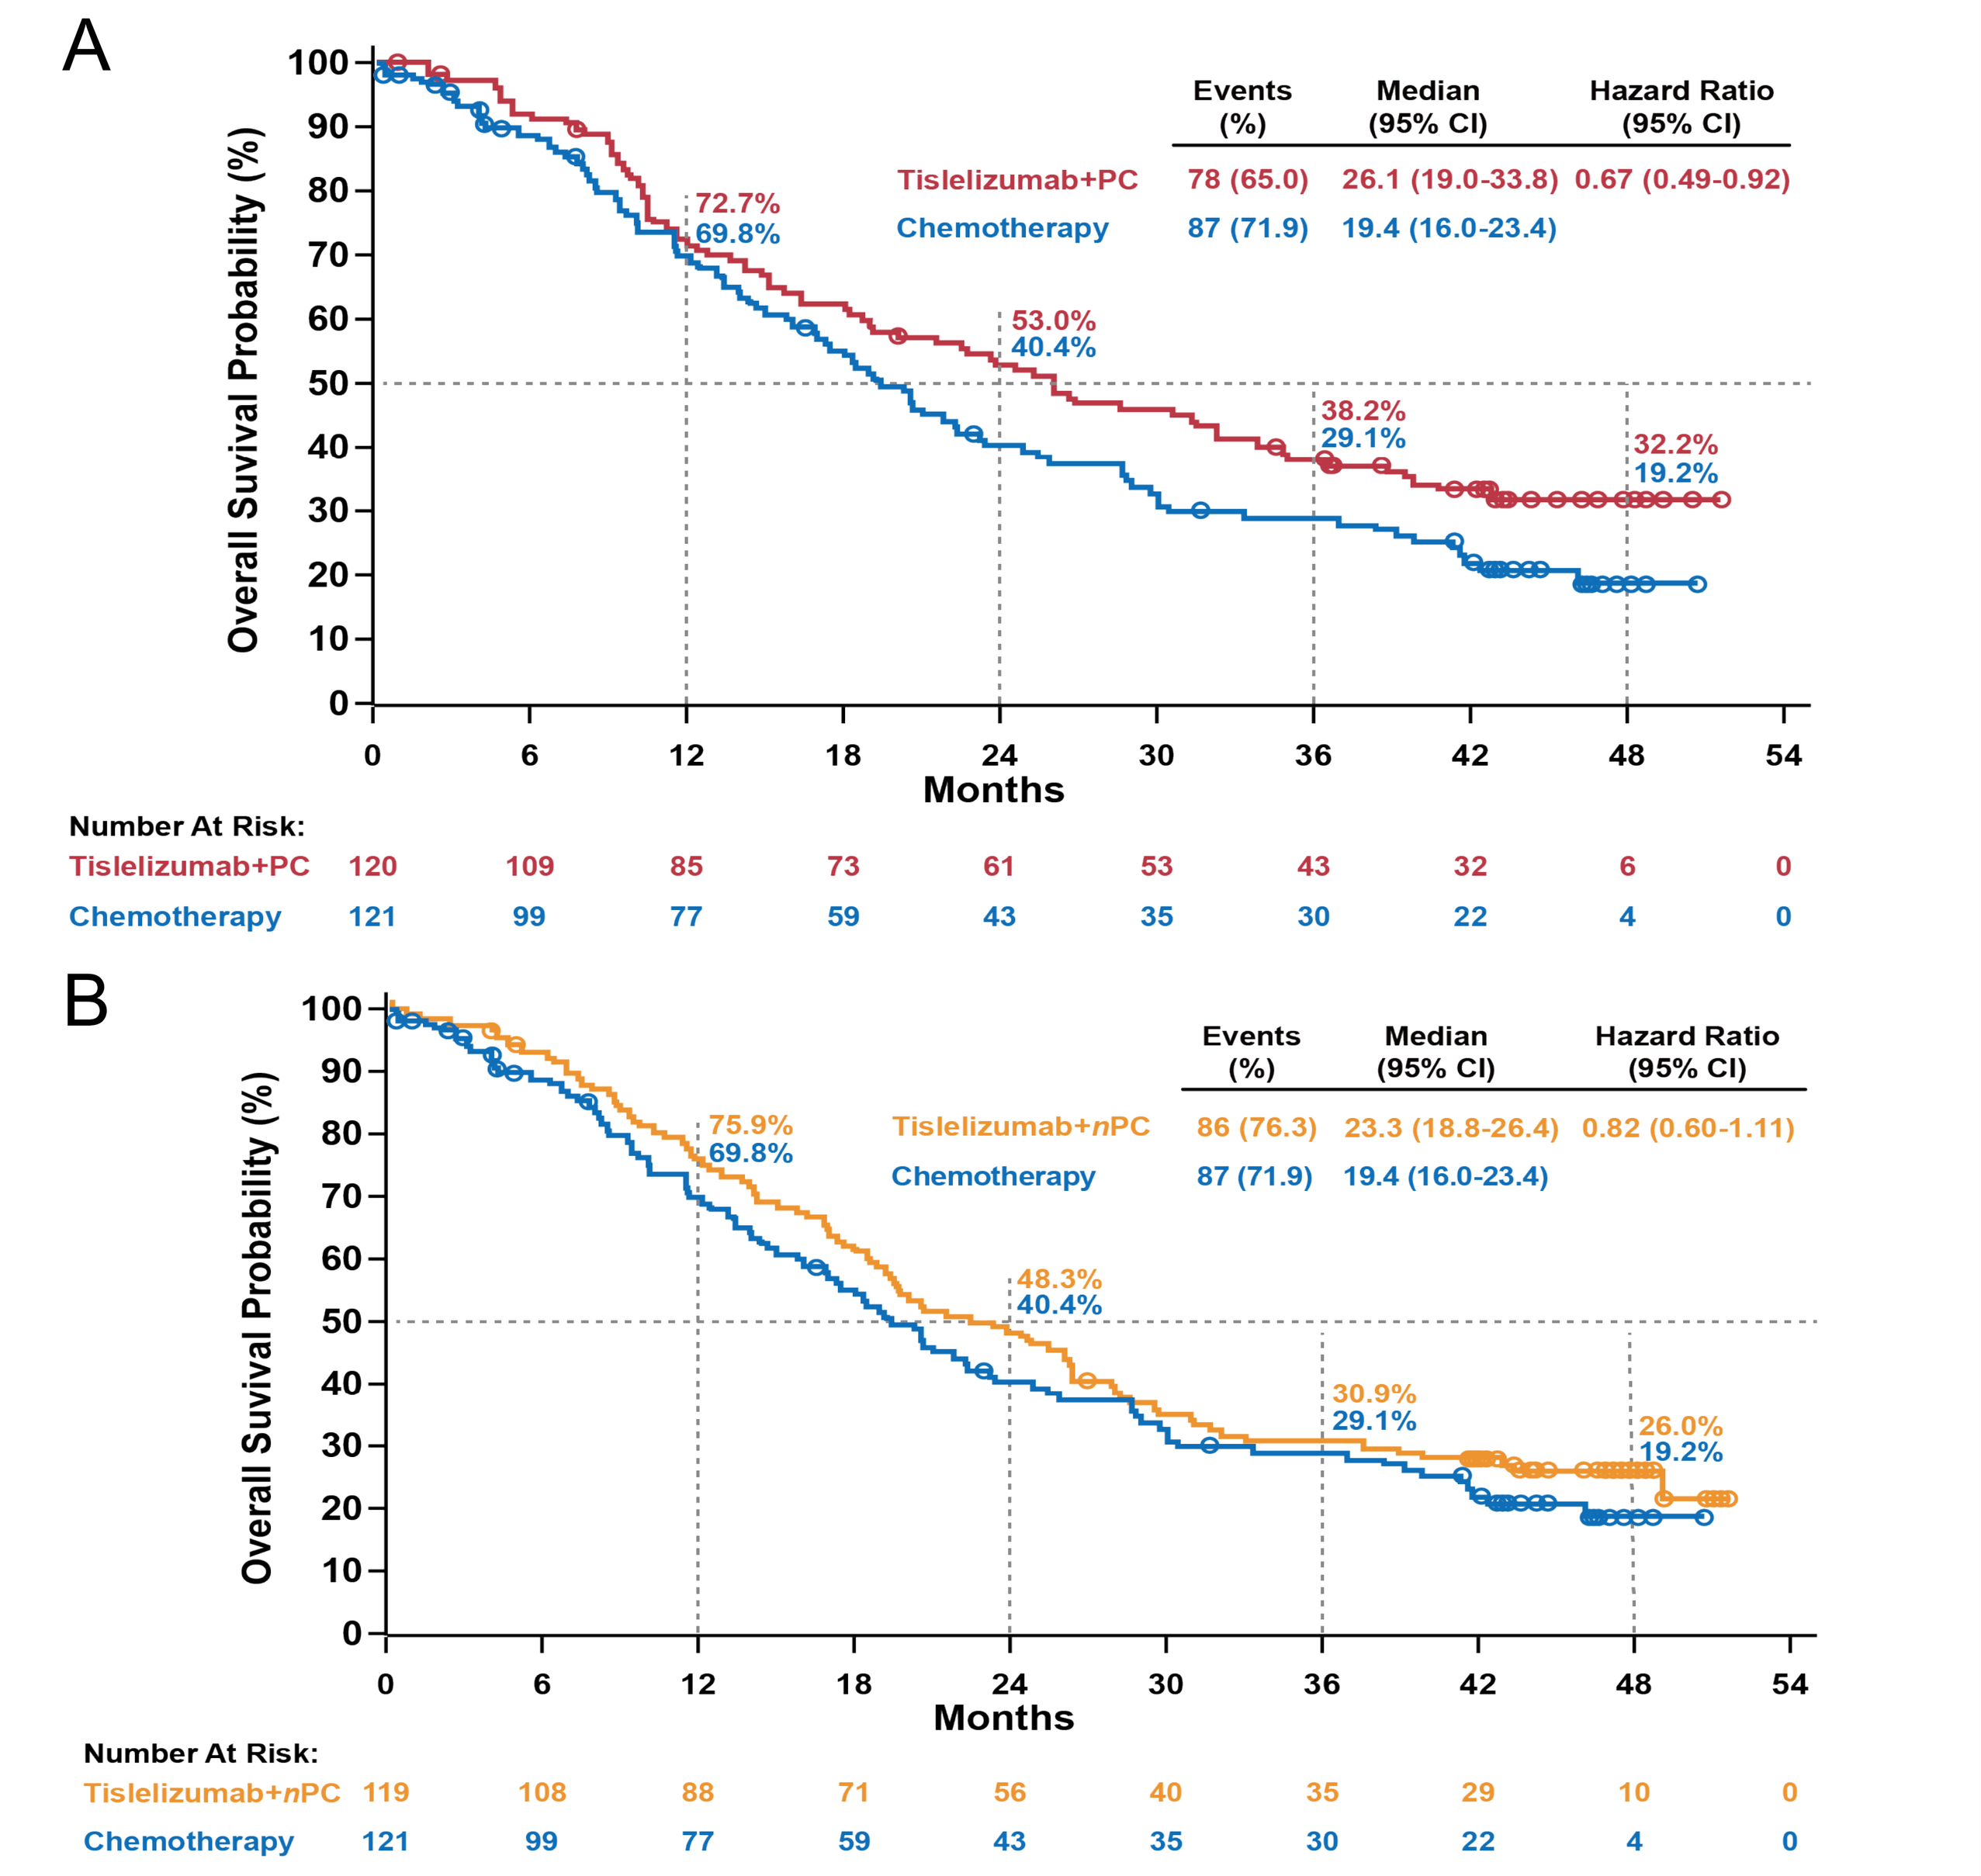

A
Overall Suvival Probability (%)
Events (%) | Median (95% CI) | Hazard Ratio (95% CI)
Tislelizumab+PC 78 (65.0) 26.1 (19.0-33.8) 0.67 (0.49-0.92)
Chemotherapy 87 (71.9) 19.4 (16.0-23.4)
72.7% 69.8%
53.0% 40.4%
38.2% 29.1%
32.2% 19.2%
Months
Number At Risk:
Tislelizumab+PC 120 109 85 73 61 53 43 32 6 0
Chemotherapy 121 99 77 59 43 35 30 22 4 0
B
Events (%) | Median (95% CI) | Hazard Ratio (95% CI)
Tislelizumab+nPC 86 (76.3) 23.3 (18.8-26.4) 0.82 (0.60-1.11)
Chemotherapy 87 (71.9) 19.4 (16.0-23.4)
75.9% 69.8%
48.3% 40.4%
30.9% 29.1%
26.0% 19.2%
Number At Risk:
Tislelizumab+nPC 119 108 88 71 56 40 35 29 10 0
Chemotherapy 121 99 77 59 43 35 30 22 4 0

Figure 3. Kaplan-Meier curves of two-stage-adjusted OS

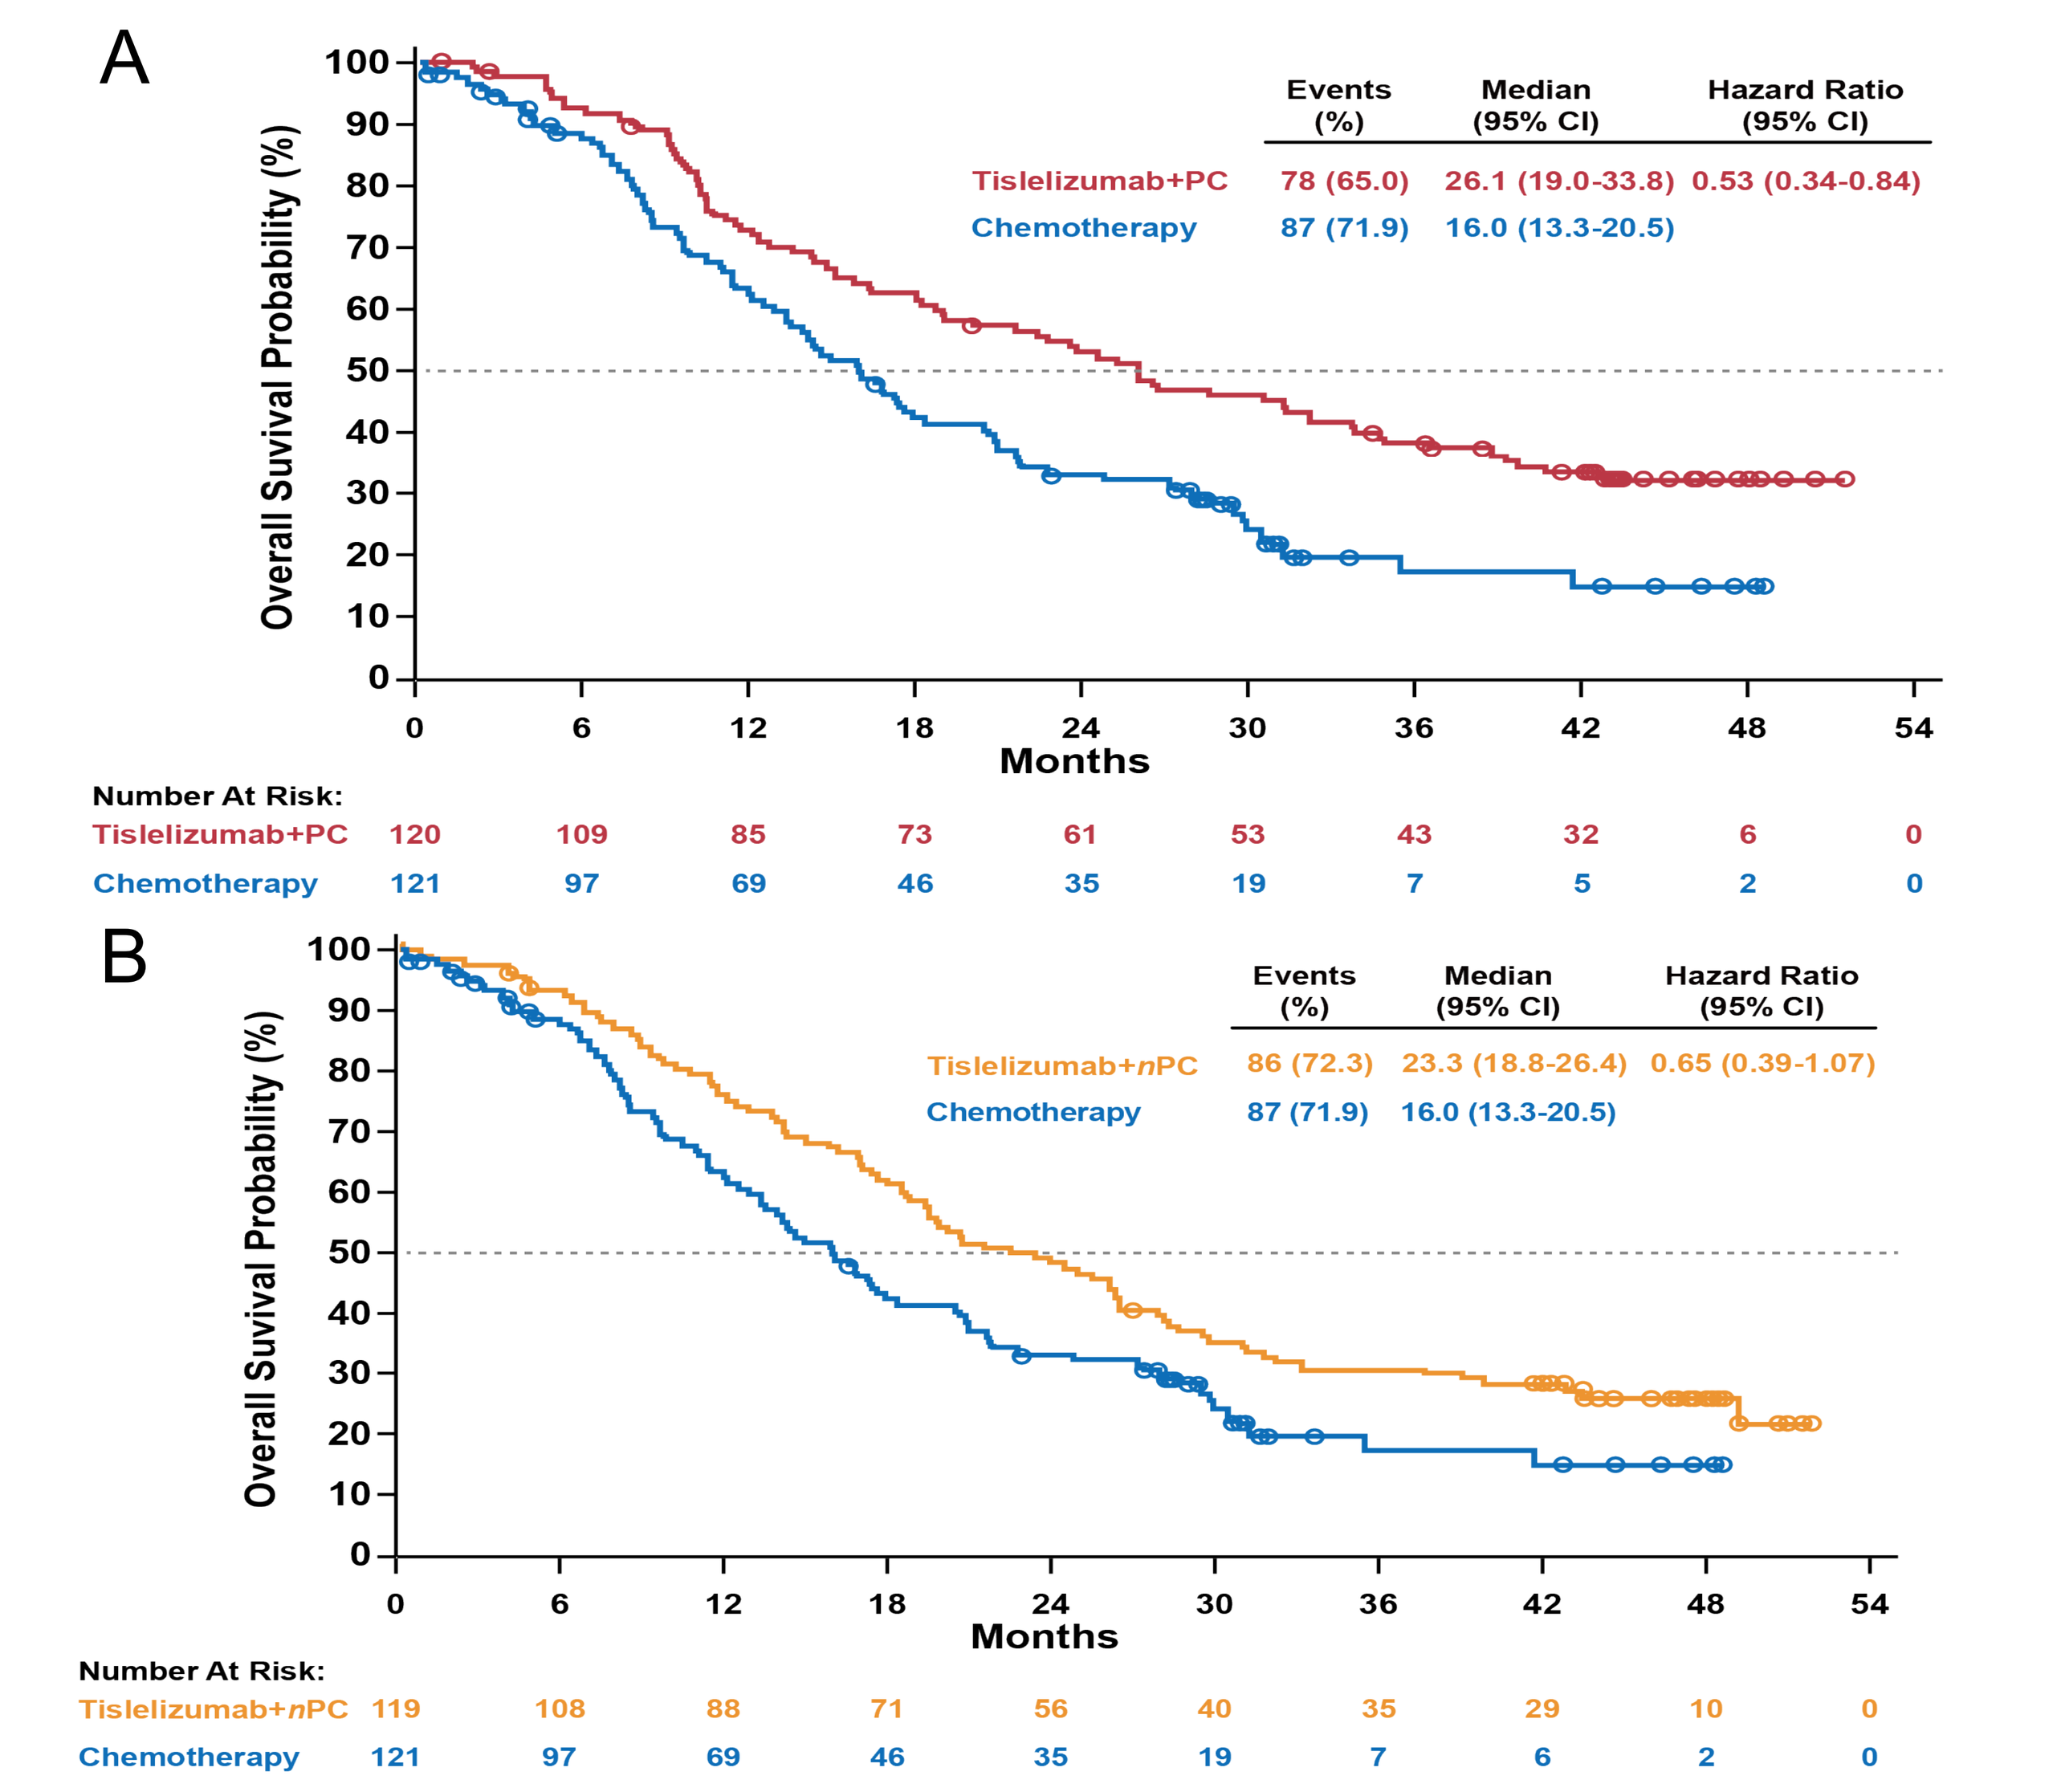

A
Overall Suvival Probability (%)
Events (%) | Median (95% CI) | Hazard Ratio (95% CI)
Tislelizumab+PC 78 (65.0) 26.1 (19.0-33.8) 0.53 (0.34-0.84)
Chemotherapy 87 (71.9) 16.0 (13.3-20.5)
Months
Number At Risk:
Tislelizumab+PC 120 109 85 73 61 53 43 32 6 0
Chemotherapy 121 97 69 46 35 19 7 5 2 0
B
Events (%) | Median (95% CI) | Hazard Ratio (95% CI)
Tislelizumab+nPC 86 (72.3) 23.3 (18.8-26.4) 0.65 (0.39-1.07)
Chemotherapy 87 (71.9) 16.0 (13.3-20.5)
Number At Risk:
Tislelizumab+nPC 119 108 88 71 56 40 35 29 10 0
Chemotherapy 121 97 69 46 35 19 7 6 2 0

Figure 4. Kaplan-Meier curves of PFS

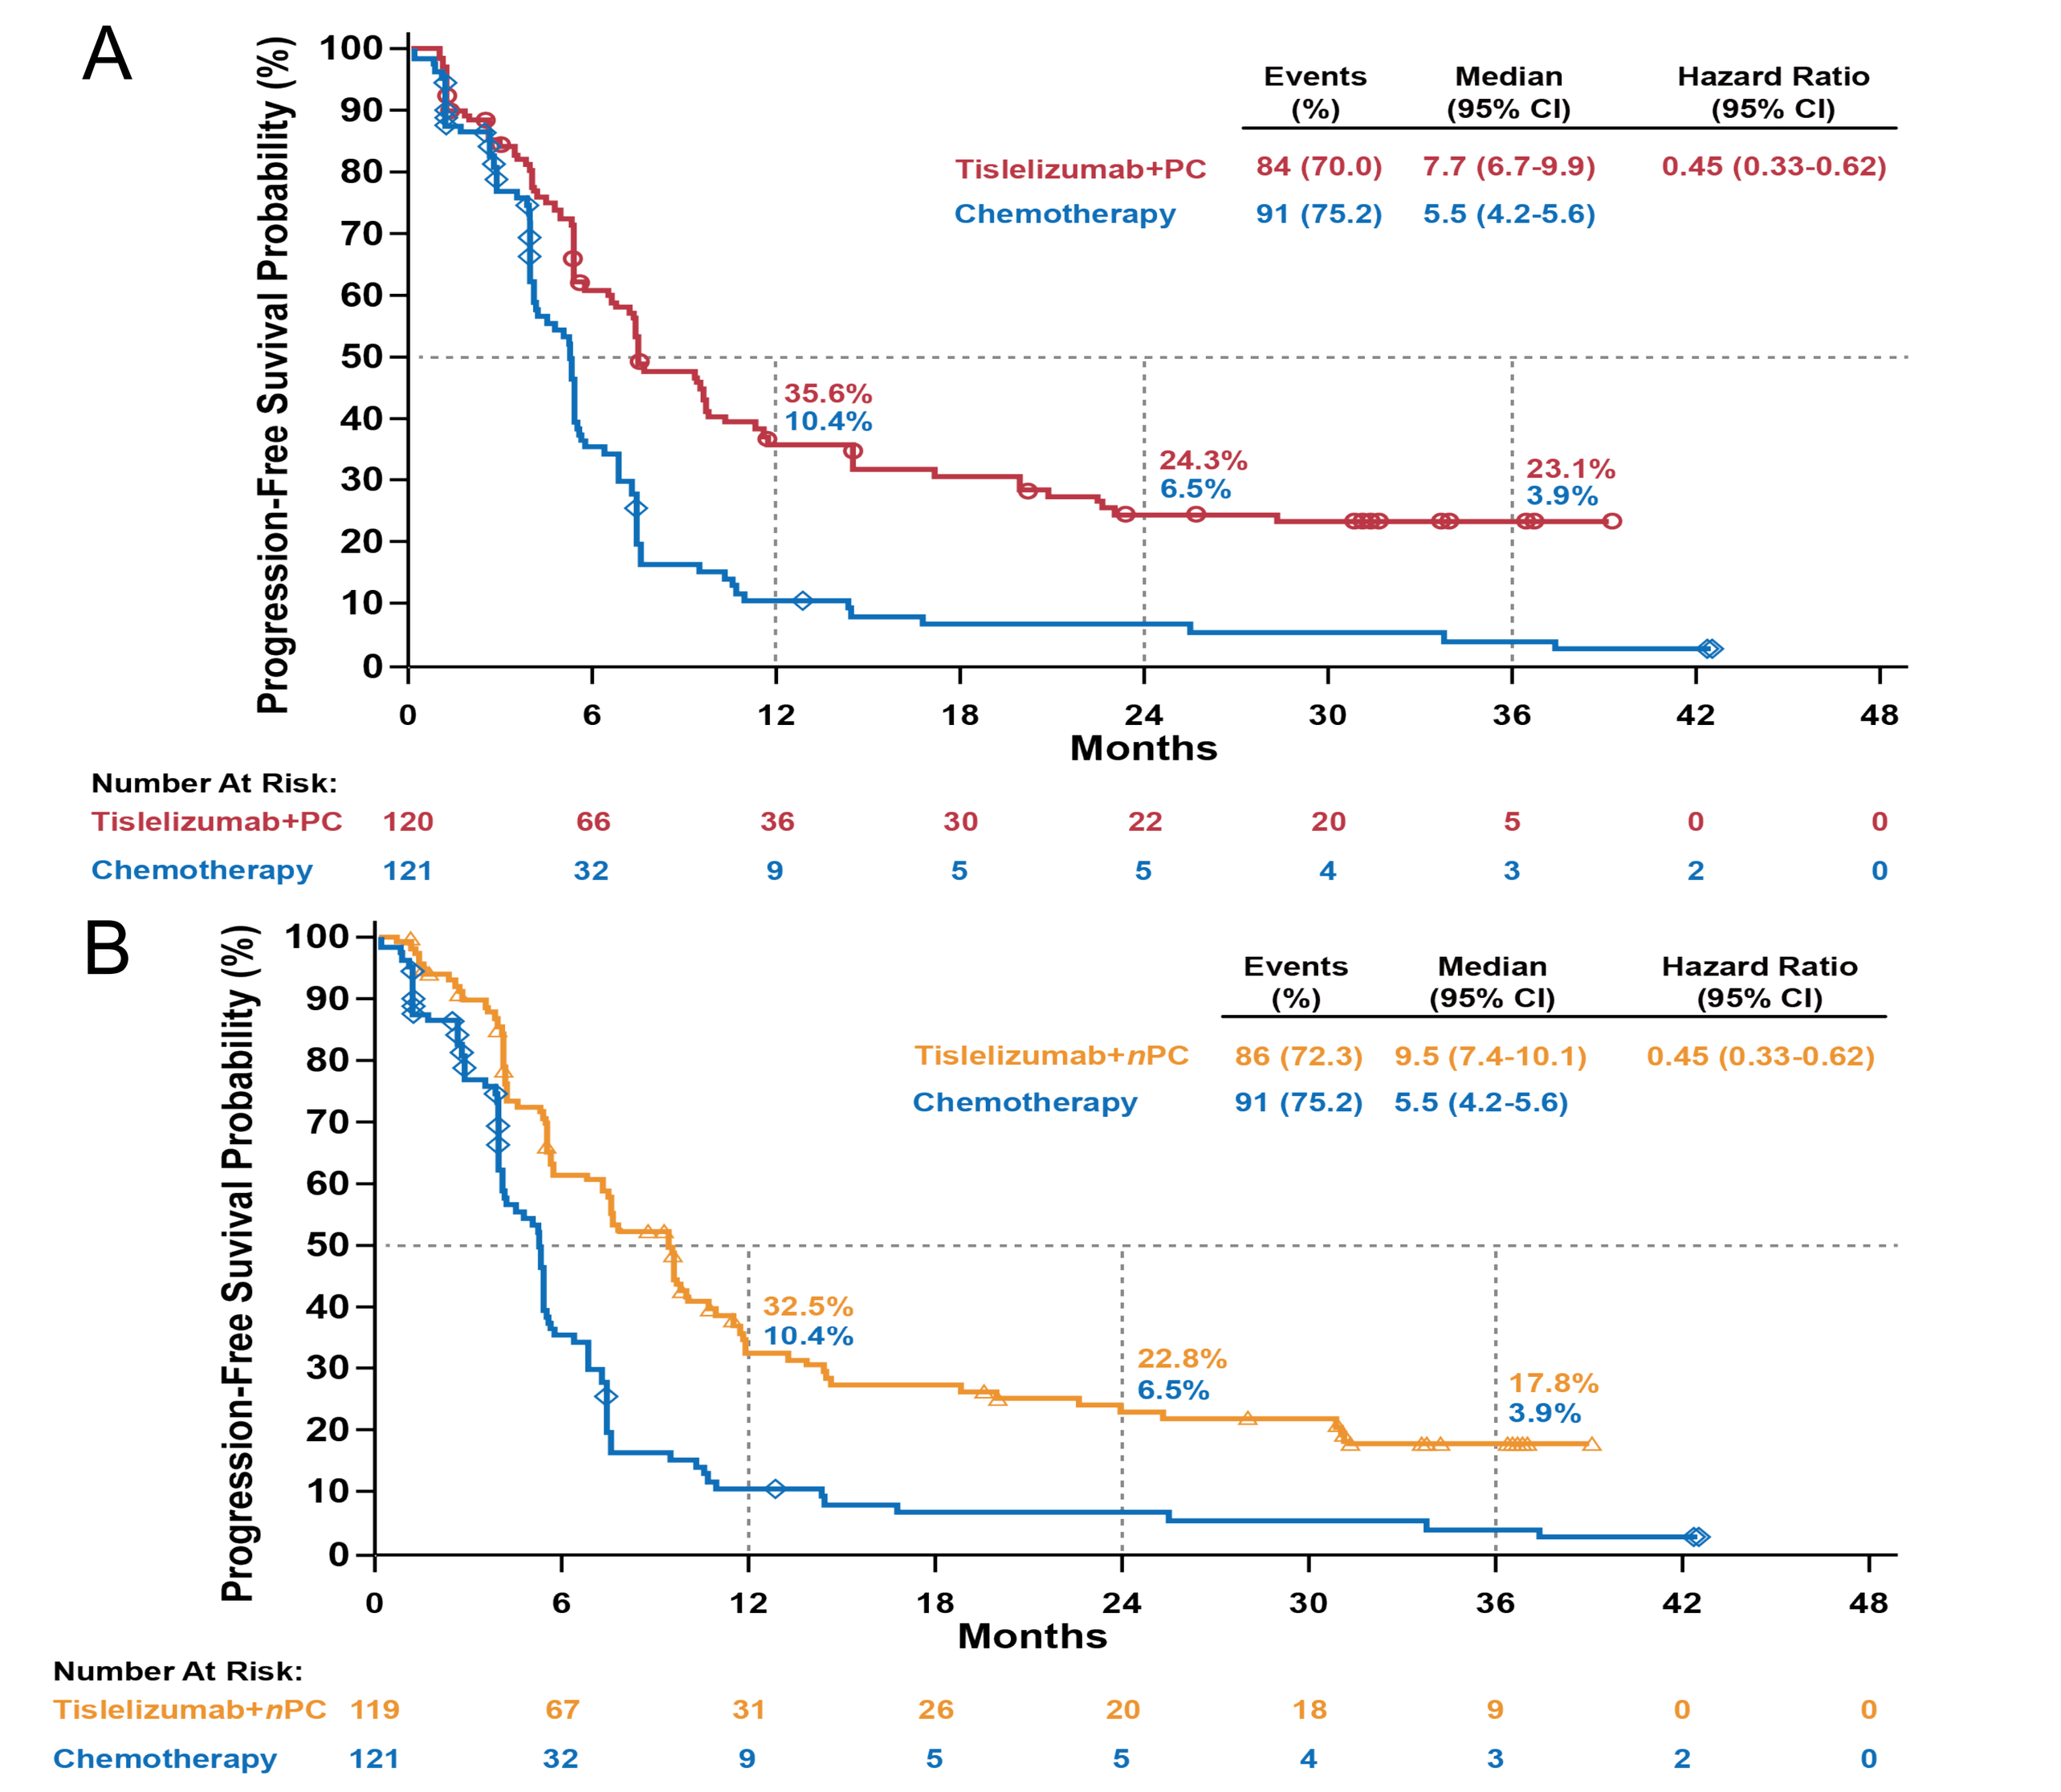

A
Progression-Free Suvival Probability (%)
Events (%) | Median (95% CI) | Hazard Ratio (95% CI)
Tislelizumab+PC 84 (70.0) 7.7 (6.7-9.9) 0.45 (0.33-0.62)
Chemotherapy 91 (75.2) 5.5 (4.2-5.6)
35.6% 10.4%
24.3% 6.5%
23.1% 3.9%
Months
Number At Risk:
Tislelizumab+PC 120 66 36 30 22 20 5 0 0
Chemotherapy 121 32 9 5 5 4 3 2 0
B
Events (%) | Median (95% CI) | Hazard Ratio (95% CI)
Tislelizumab+nPC 86 (72.3) 9.5 (7.4-10.1) 0.45 (0.33-0.62)
Chemotherapy 91 (75.2) 5.5 (4.2-5.6)
32.5% 10.4%
22.8% 6.5%
17.8% 3.9%
Number At Risk:
Tislelizumab+nPC 119 67 31 26 20 18 9 0 0
Chemotherapy 121 32 9 5 5 4 3 2 0

Outcomes in patients who received ≥35 cycles of tislelizumab

- A total of 42 (17.6%) patients received ≥35 cycles of tislelizumab (LTE) in tislelizumab + chemo arms, with a median of 58 (range 37-71) treatment cycles .
- 73.8% of patients had PD-L1 TC ≥1% (TC ≥50%, 50.0%), and 26.2% patients had PD-L1 TC <1%. Other baseline characteristics were generally similar to the total ITT population of tislelizumab plus chemo arms.
- ORR was 100% (CR, n=11; PR, n=31; **Fig. 6A**), and median DOR was not reached.
- Median OS was not reached. 4-year OS rate was 97.5% (95% CI, 83.5%-99.6%) (**Fig. 6B**).
- The profile of imAEs in LTE patients was similar to the overall population of tislelizumab + chemo arms.

Figure 6. Swimmer plot and Kaplan–Meier curve for OS of LTE patients

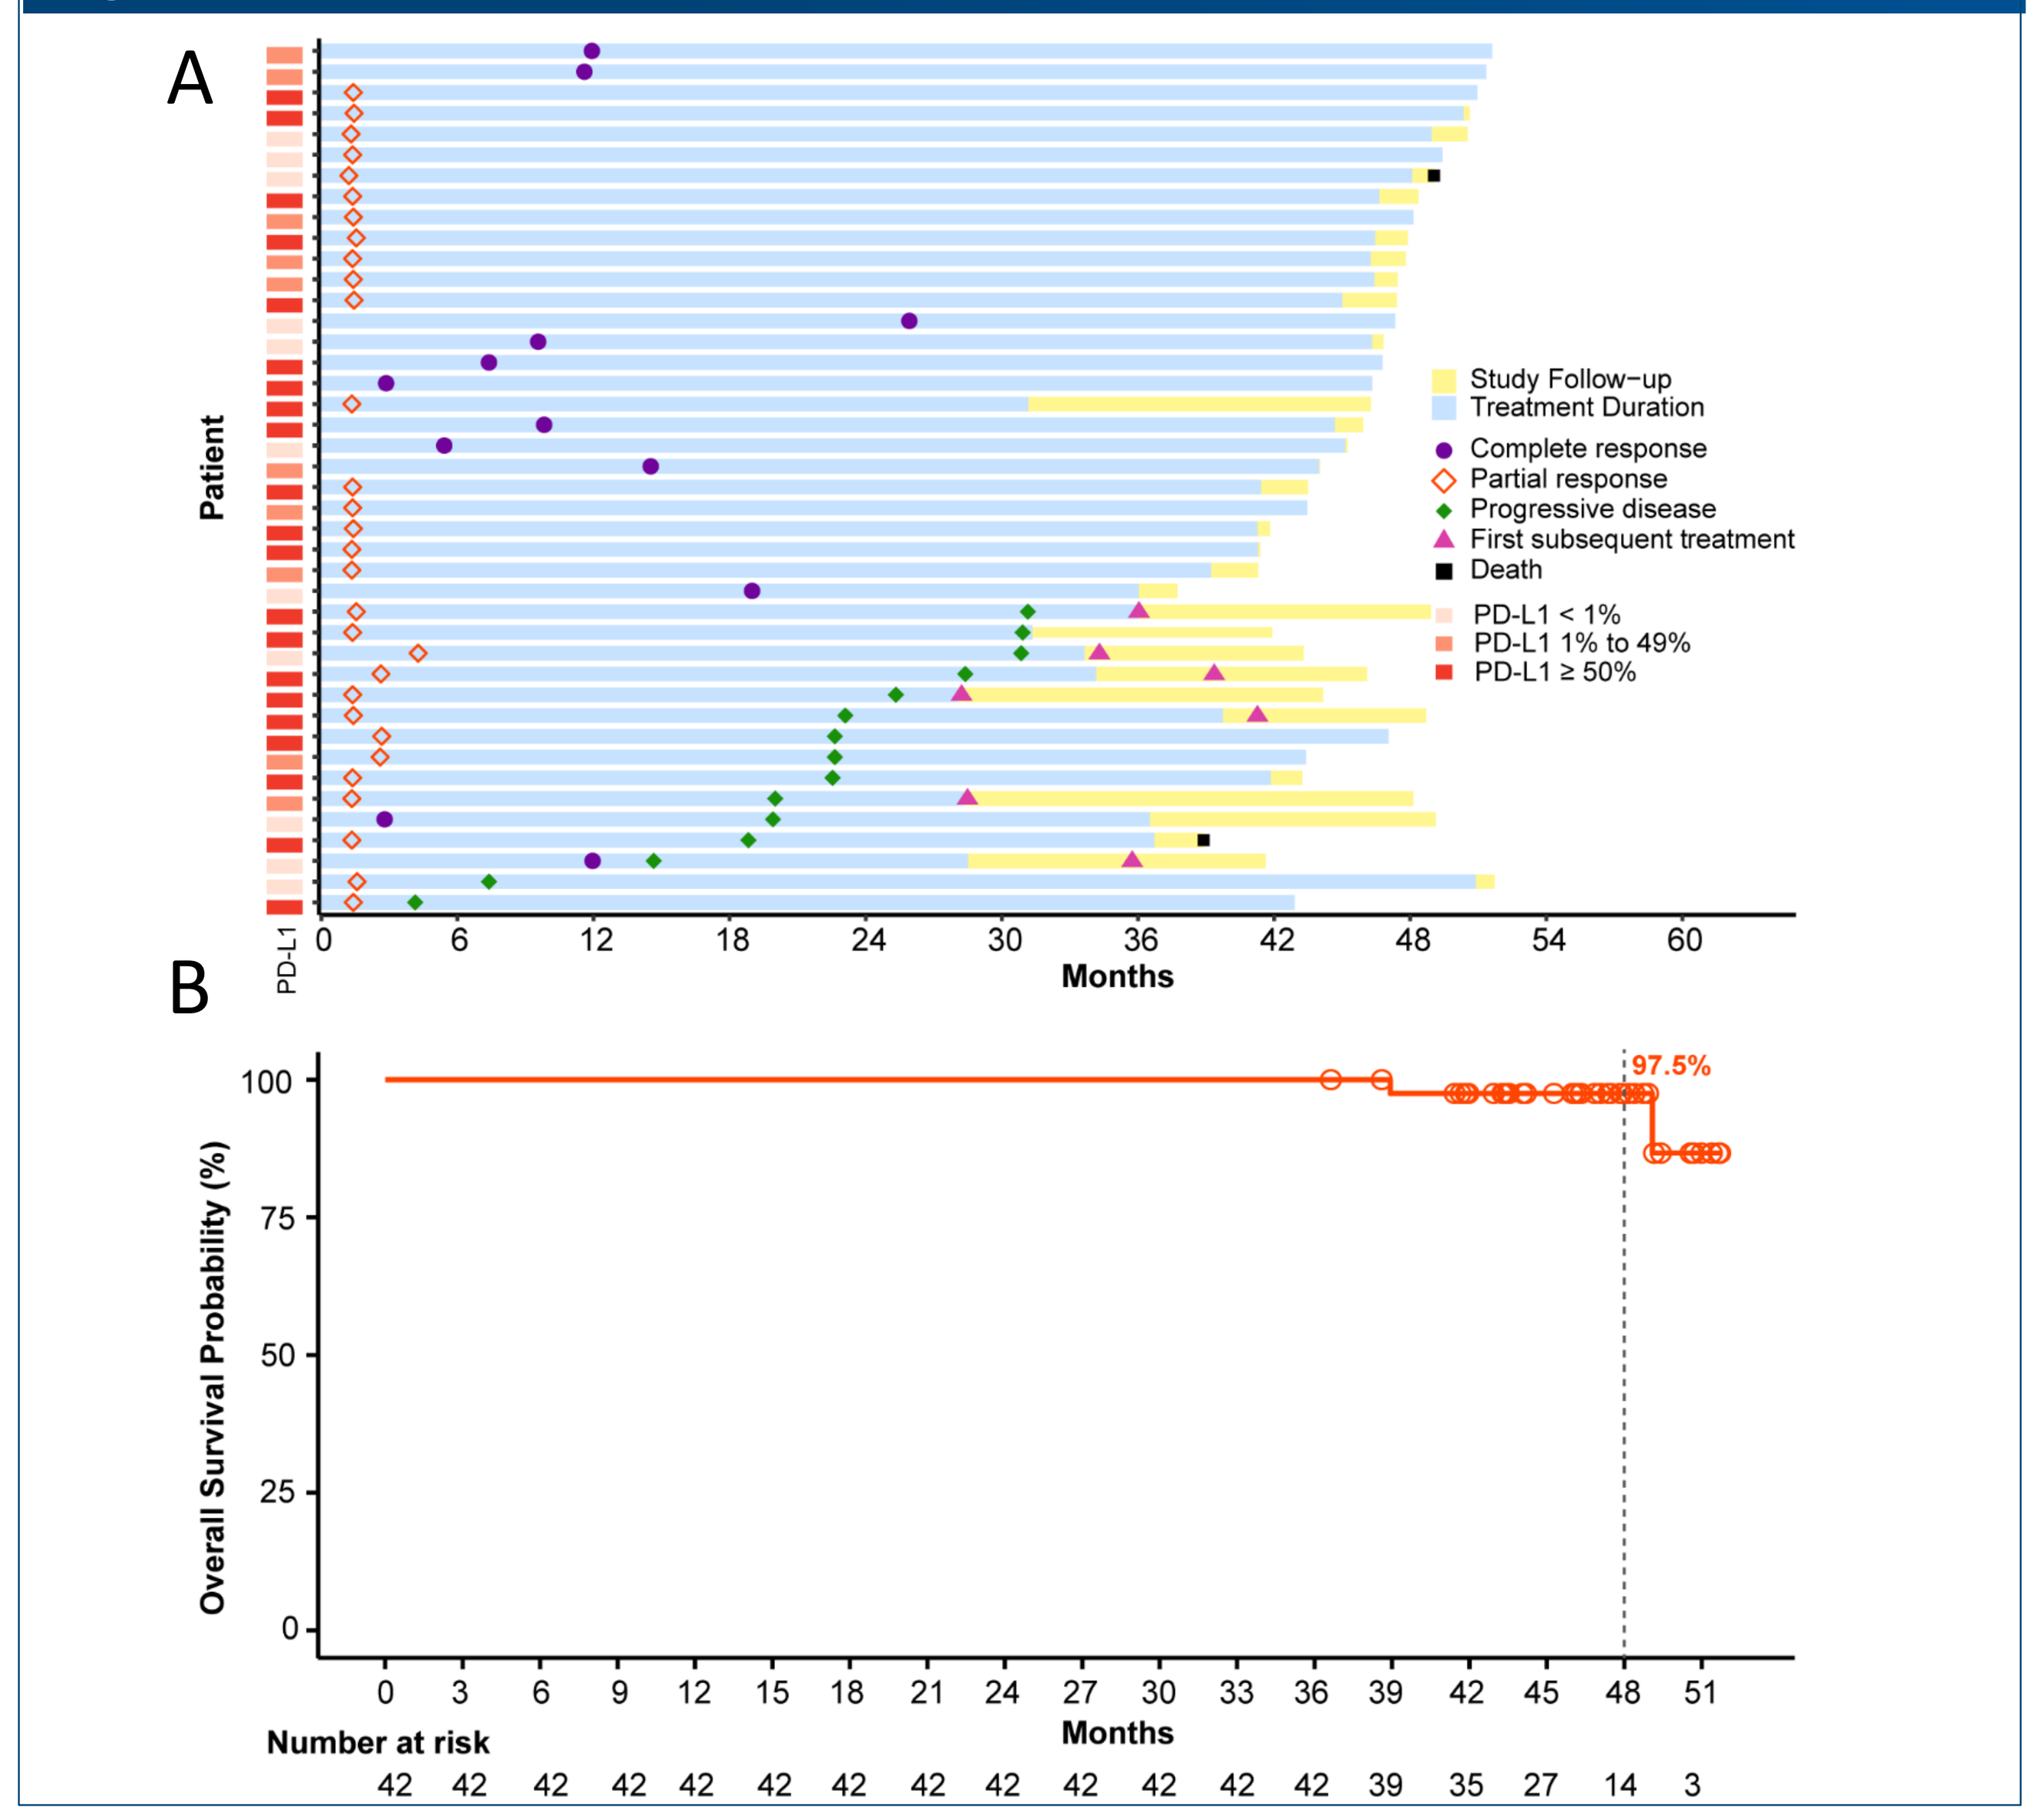

A
Patient
PD-L1
Months
Study Follow-up
Treatment Duration
Complete response
Partial response
Progressive disease
First subsequent treatment
Death
PD-L1 < 1%
PD-L1 1% to 49%
PD-L1 ≥ 50%
B
Overall Survival Probability (%)
97.5%
Months
Number at risk
42 42 42 42 42 42 42 42 42 42 42 42 42 39 35 27 14 3

Molecular features of LTE patients in tislelizumab + chemo arms

- The baseline tumor tissues from LTE patients had higher PD-L1 expression and T cell inflammation signature levels, while the TMB status was similar between LTE vs non-LTE **(Fig.7A)**
- FAT1 alterations were enriched in LTE vs non-LTE (34.6% vs 15.4%, P=0.028); A numerically lower frequency was observed in NFE2L2, KEAP1, STK11, JAK1/2, and B2M in LTE vs non-LTE **(Fig.7B)**

Figure 7. Molecular features in LTE patients in tislelizumab + chemo arms

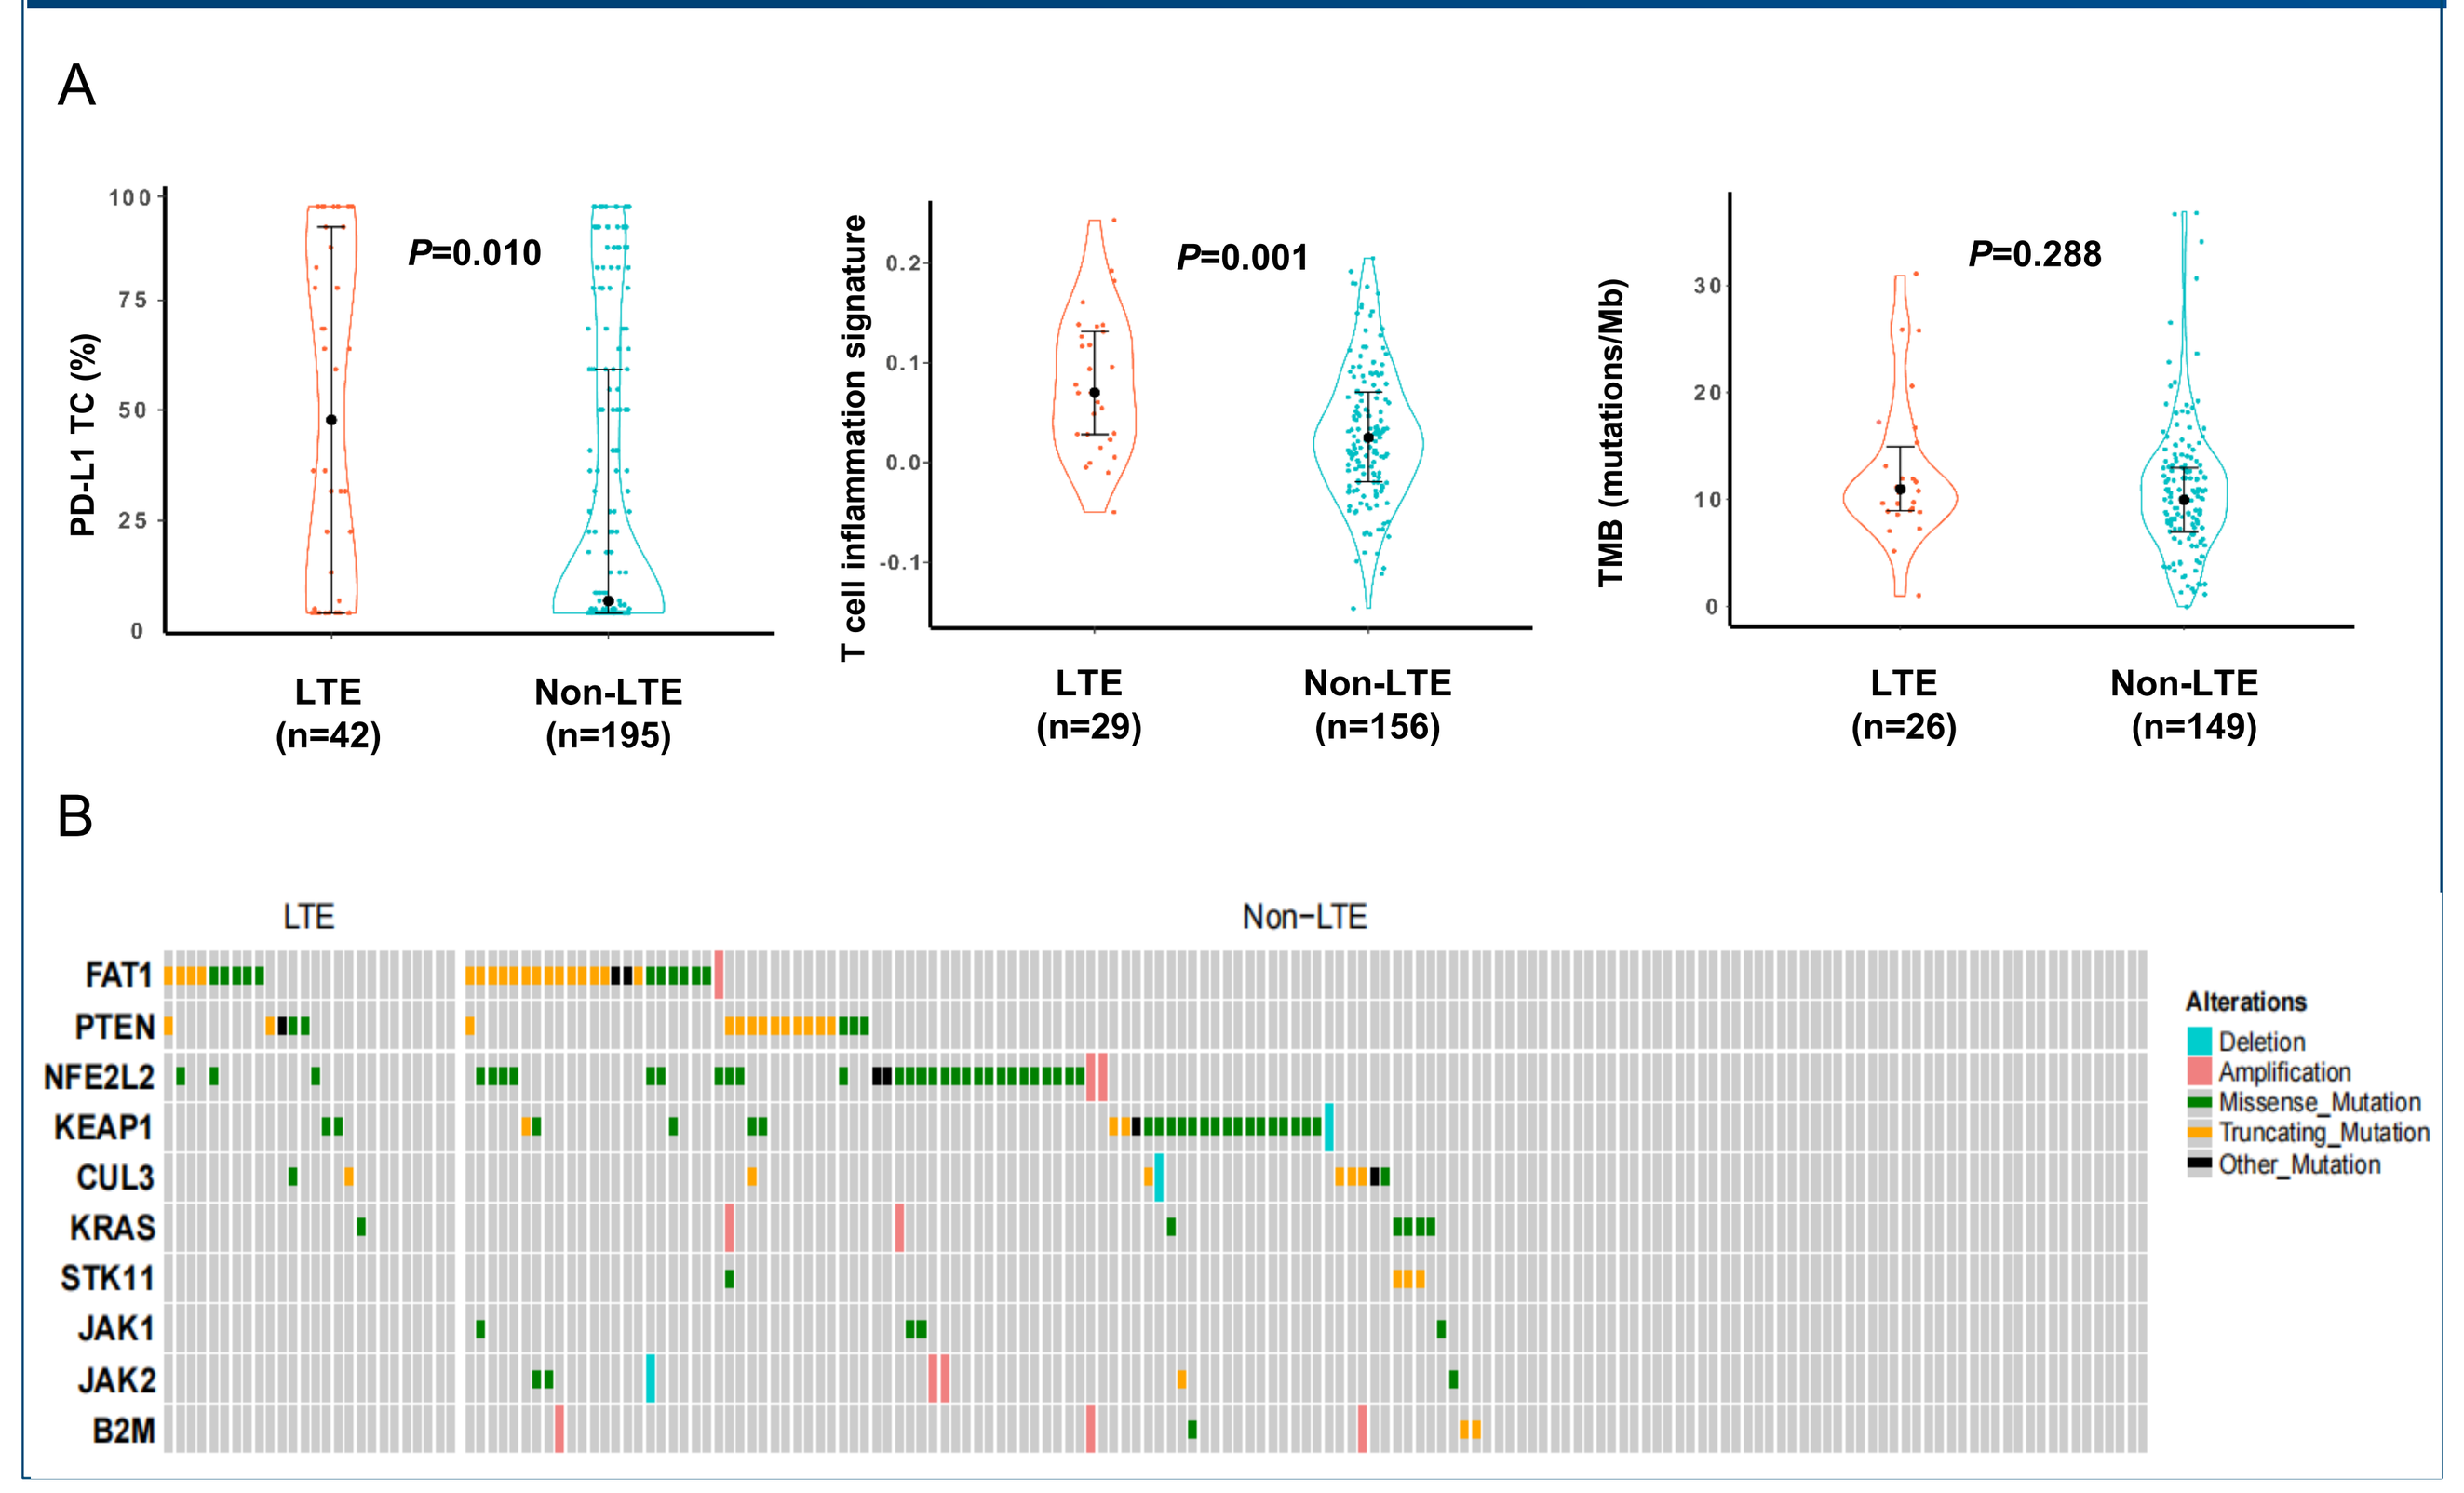

A
PD-L1 TC (%)
P=0.010
LTE (n=42)
Non-LTE (n=195)
T cell inflammation signature
P=0.001
LTE (n=29)
Non-LTE (n=156)
TMB (mutations/Mb)
P=0.288
LTE (n=26)
Non-LTE (n=149)
B
LTE
Non-LTE
FAT1
PTEN
NFE2L2
KEAP1
CUL3
KRAS
STK11
JAK1
JAK2
B2M
Alterations
Deletion
Amplification
Missense_Mutation
Truncating_Mutation
Other_Mutation