Zanubrutinib (Zanu) Overcomes BTK-V416L Resistance in B Cell Lymphoma Models

Haitao Wang[1]*, Xiaoyu Feng[3]*, Jinhong Ren[1], Lanyue Bai[3], Lin Li[1], Chao Cui[3], Lei Shi[3], Yang Li[1], Yangyang Wang[3], Wenjing Zhang[3], Jessica Li[2], Teiko Sumiyoshi[2], Yangbo Yue[3], Wei Jin[1]#, Zhirong Shen[3]#

1. BeiGene (Beijing) Co., Ltd., Beijing 102206, China; 2. BeiGene USA Inc., San Mateo, CA USA; 3. BeiGene (Shanghai) Co., Ltd., Shanghai, China;
* These authors contributed equally; # Corresponding author

#abstract 6769

Introduction

The second-generation BTK inhibitors (BTKis), e.g., zanu, acalabrutinib (acala), have further transformed the therapy landscape of chronic lymphocytic leukemia (CLL). However, acquired mutations in BTK (mainly at position C481) results in covalent BTKi resistance and disease progression. Non-covalent BTKi e.g., pirtobrutinib (pirto) can overcome C481 mutations, but is susceptible to other BTK mutations like the recently described kinase impaired V416L and L528W mutations, which both maintain downstream signaling. However, the impact of V416L on BTKi therapy remains not well characterized. Here, we investigated how V416L affects acala, zanu, and pirto response in B cancer cells.

Method

TMD8 BTK V416L cell lines were generated by CRISPR-Cas9 mediated gene editing. Kinase activity and phosphorylation of BTK Y223 were evaluated in V416L and wildtype (WT) TMD8 cells by Western blot. Structural modeling of human BTK was performed. Viability of TMD8 cells with V416L or WT was measured with CellTiter-Glo assay. Tumor growth inhibition (TGI) was measured in V416L and WT xenograft models. BTK occupancy in TMD8 cells was measured via ELISA with biotinylated probes.

Results

1. BTK-V416L encodes a kinase with impaired enzymatic activity compared to WT BTK

A

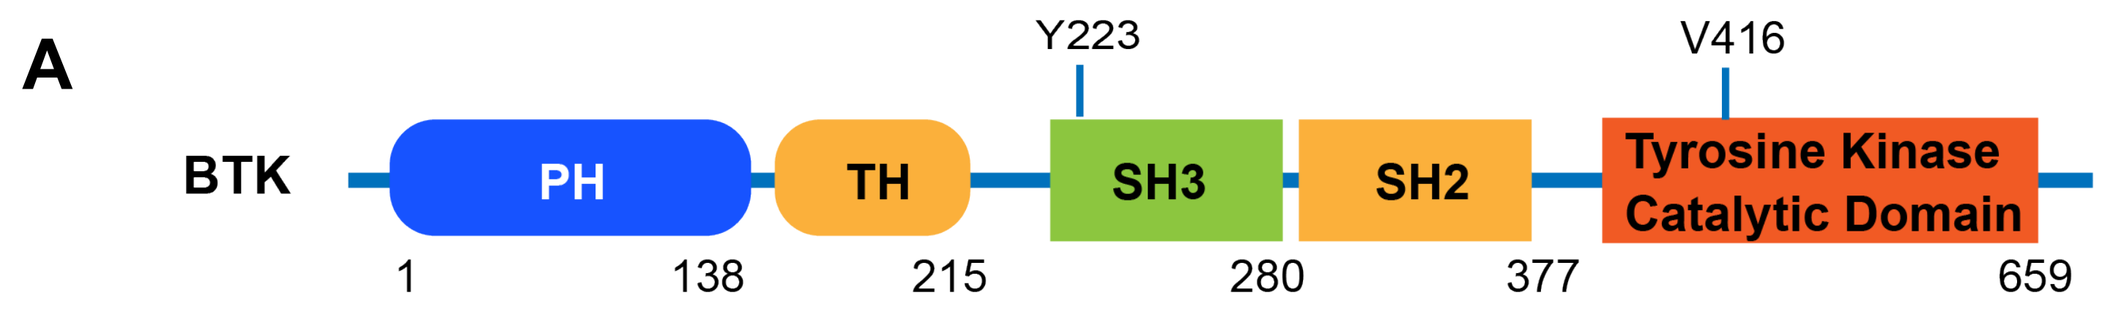
BTK
PH
TH
SH3
SH2
Tyrosine Kinase Catalytic Domain
Y223
V416
1
138
215
280
377
659

B

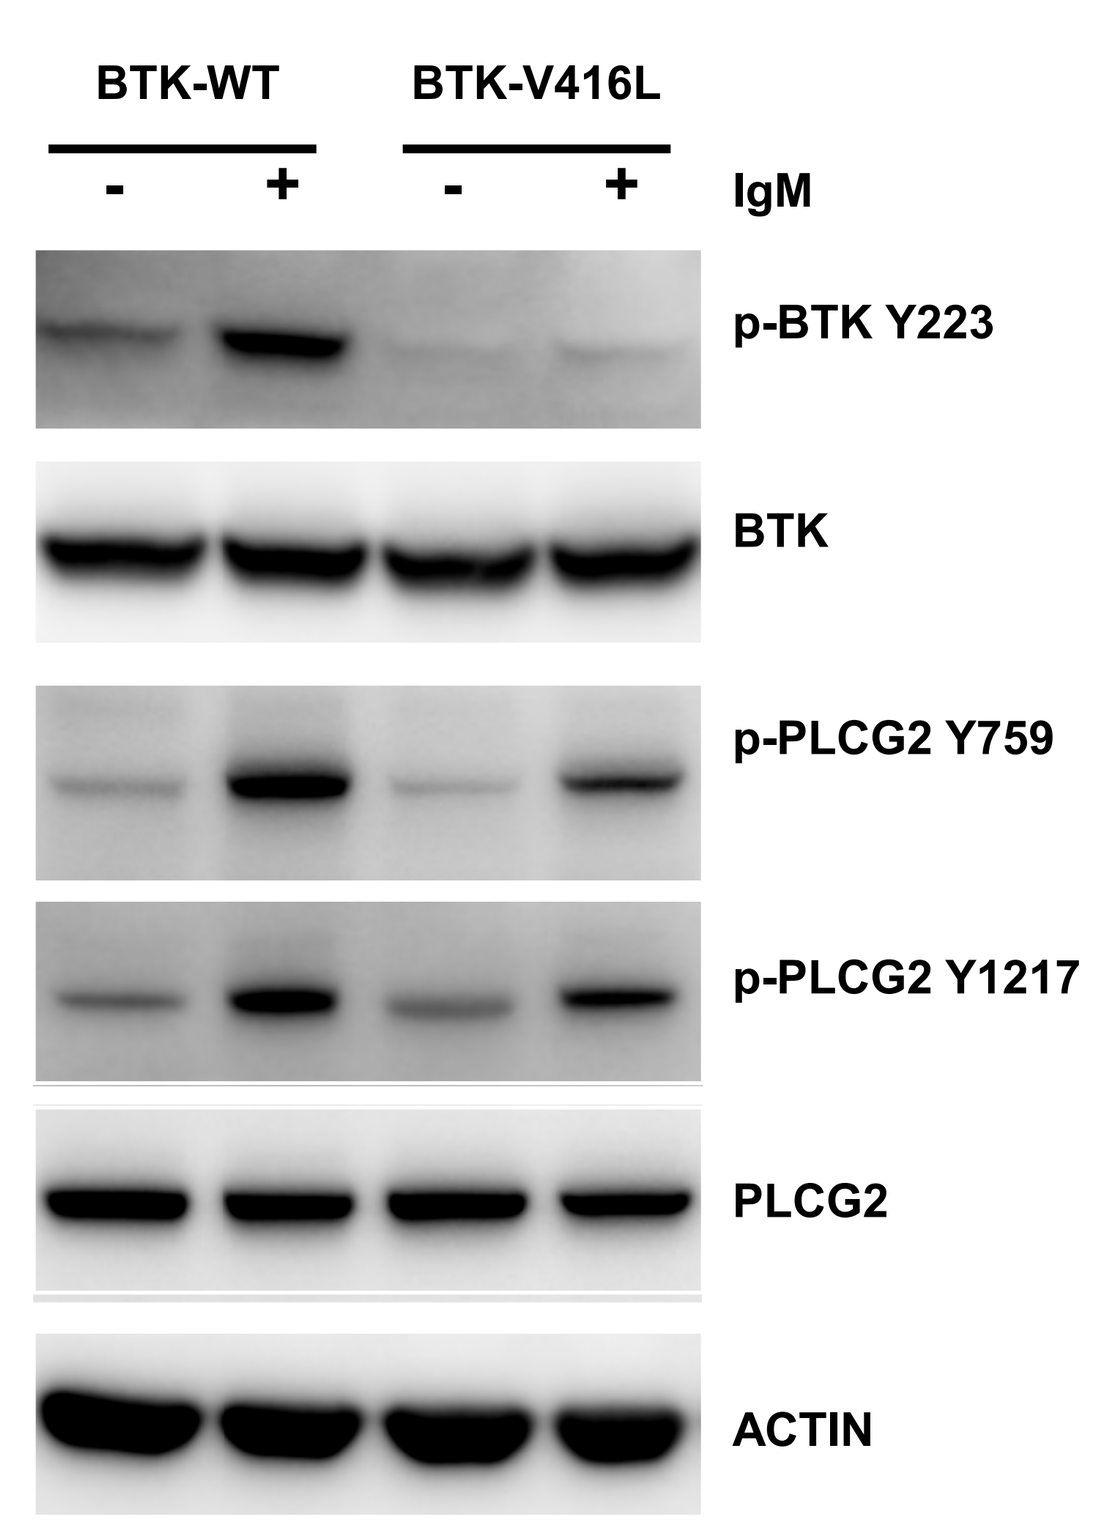
BTK-WT
BTK-V416L
- + - +
IgM
p-BTK Y223
BTK
p-PLCG2 Y759
p-PLCG2 Y1217
PLCG2
ACTIN

C

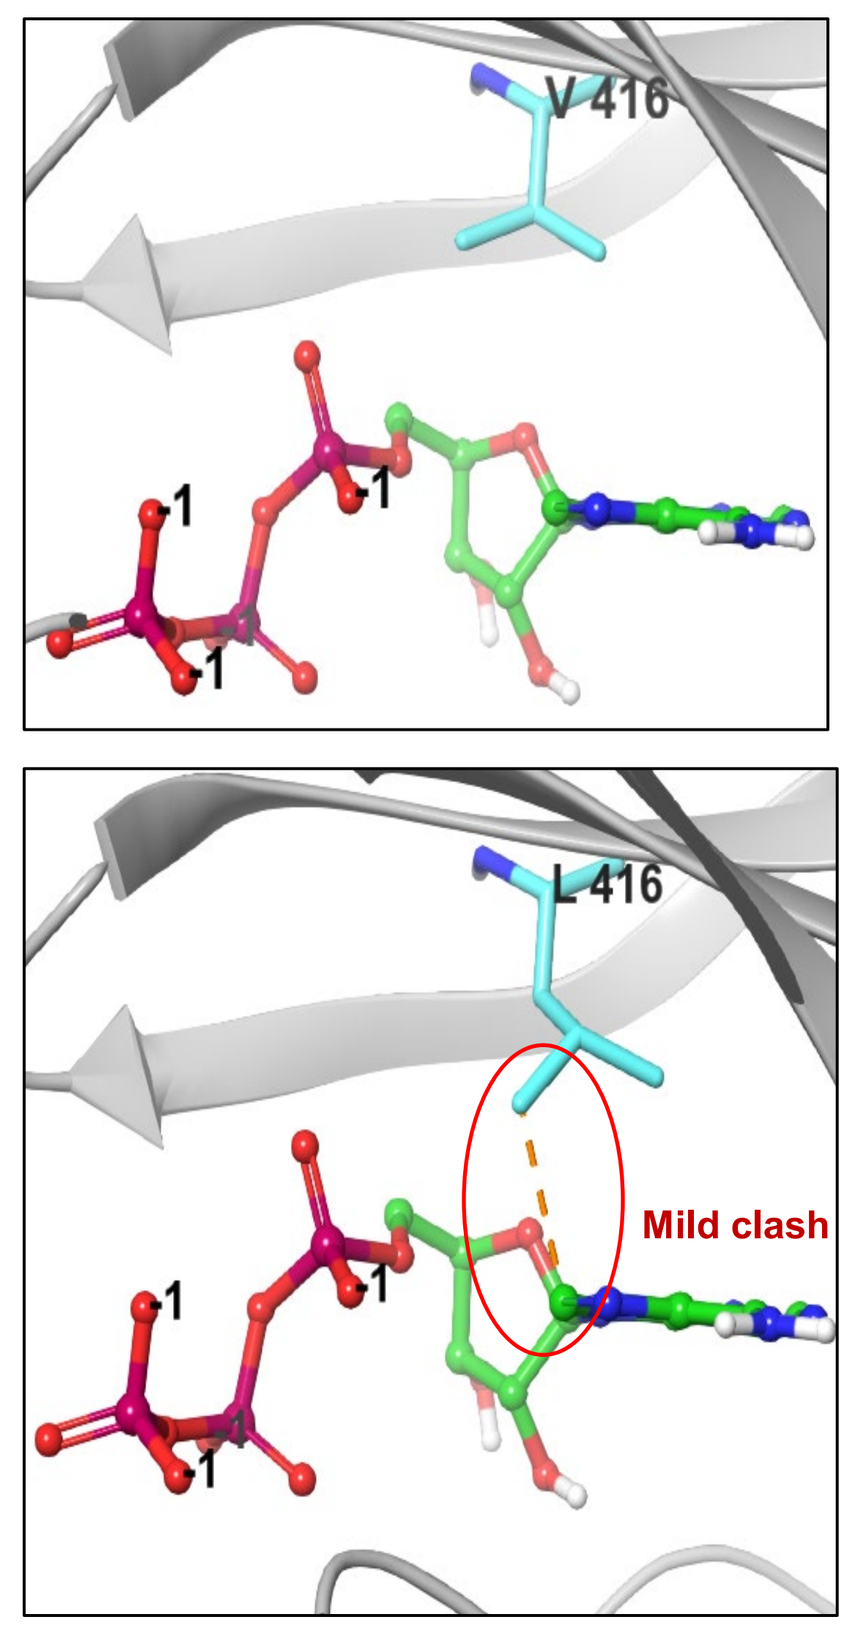
V 416
L 416
Mild clash

Note: TMD8 BTK-V416L is an in situ mutated cell line

Figure 1 A). A schematic representation of BTK domains and V416 locus; **B).** TMD8 WT and BTK-V416L cells were first stimulated by IgM antibody and then used for western blot to examine BTK and PLCG2 protein and their phosphorylation level. Autophosphorylation of BTK-V416L at Y223 is significantly reduced compared with wt BTK; **C)**. Structural modeling predicts that V416L mutation leads to binding clash between BTK and ATP, supporting it as a kinase-impaired mutation.

2. Zanu is extremely potent in killing BTK-V416L expressing TMD8 cells *in vitro*

A.

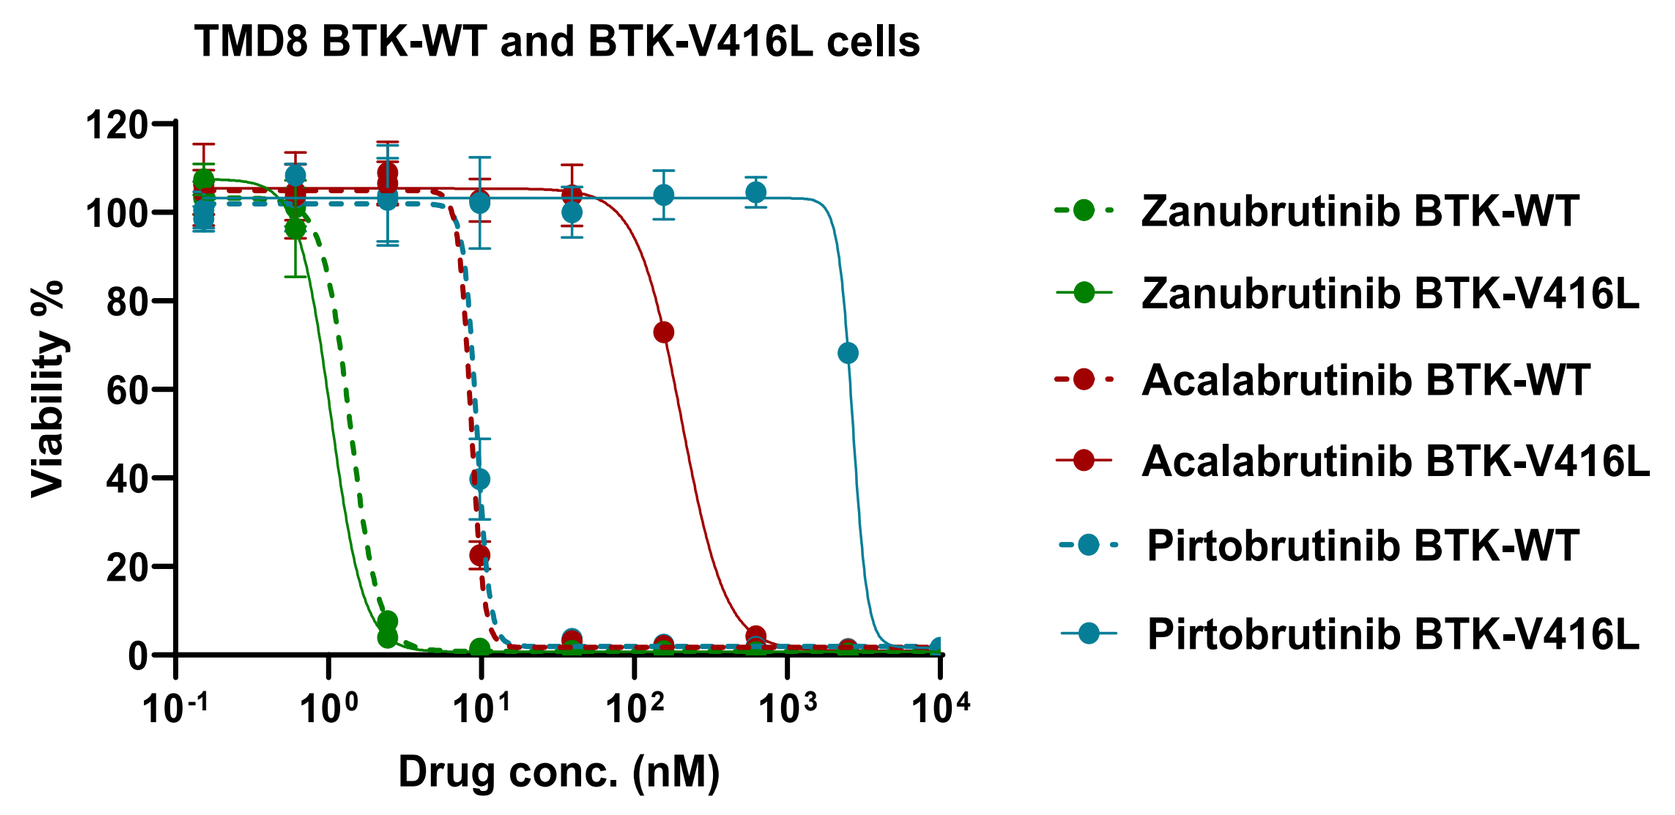
TMD8 BTK-WT and BTK-V416L cells
Viability %
Drug conc. (nM)
Zanubrutinib BTK-WT
Zanubrutinib BTK-V416L
Acalabrutinib BTK-WT
Acalabrutinib BTK-V416L
Pirtobrutinib BTK-WT
Pirtobrutinib BTK-V416L

B.

IC50 (nM)	Zanubrutinib	Acalabrutinib	Pirtobrutinib
TMD8 BTK-WT	1.3	7.7	9.4
TMD8 BTK-V416L	0.8	144.4	2074.8

Note: TMD8 BTK-V416L is an in situ mutated cell line

Figure 2 A). WT or V416L BTK expressing TMD8 cells were treated with zanubrutinib or acalabrutinib at indicated concentrations for 5 days, cell viability was examined by CellTiter-Glo luminescent cell viability assay; **B).** A table showing IC50 number from figure 2A. BTK-V416L mutation does not affect the potency of zanubrutinib but results in significant resistance to acalabrutinib and pirtobrutinib.

3. Zanu drives complete regression of BTK-V416L mutant tumors *in vivo,* while no obvious efficacy observed for acala and pirto

A.

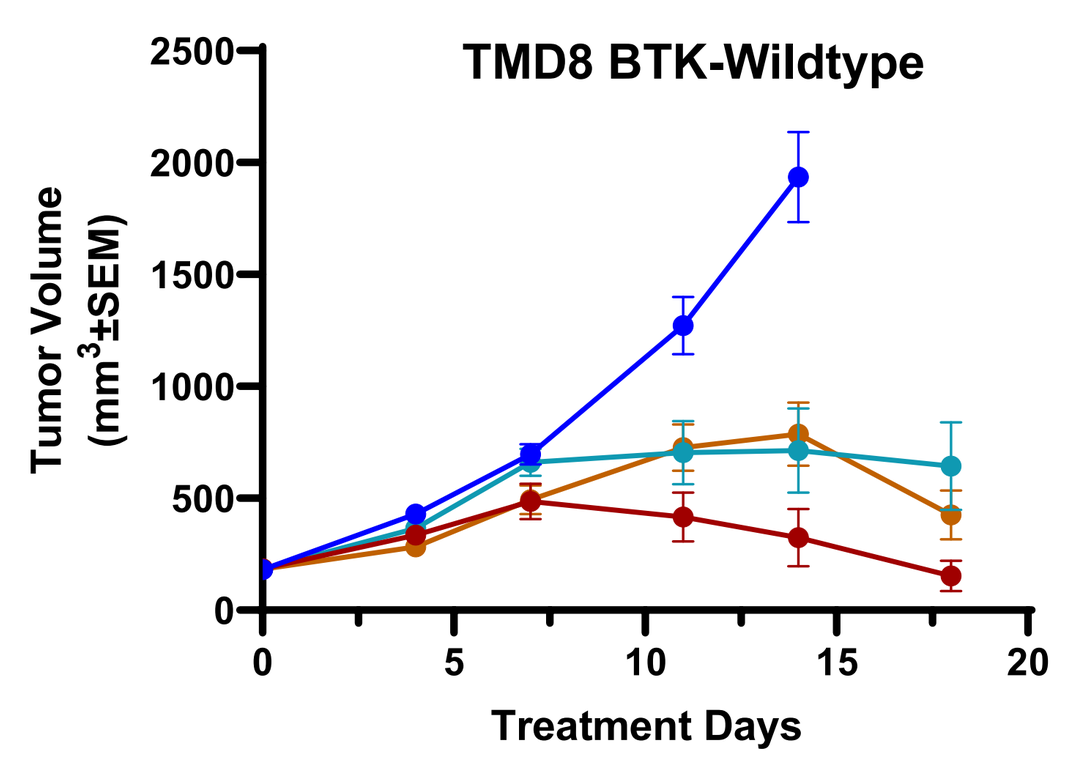
TMD8 BTK-Wildtype
Tumor Volume (mm3±SEM)
Treatment Days

Note: The exposure of 20mpk BID Zanubrutinib in mice is close to exposure of 160mg BID in human, while the exposure of 4mpk BID acalabrutinib in mice is close to the exposure of 100mg BID inhuman.

	TGI (%), D14	p-value (compared with vehicle)
Vehicle	na	na
Zanubrutinib, 20mpk BID	92	<0.001
Acalabrutinib, 4mpk BID	70	<0.001
Pirtobrutinib, 50mpk BID	66	<0.001

B.

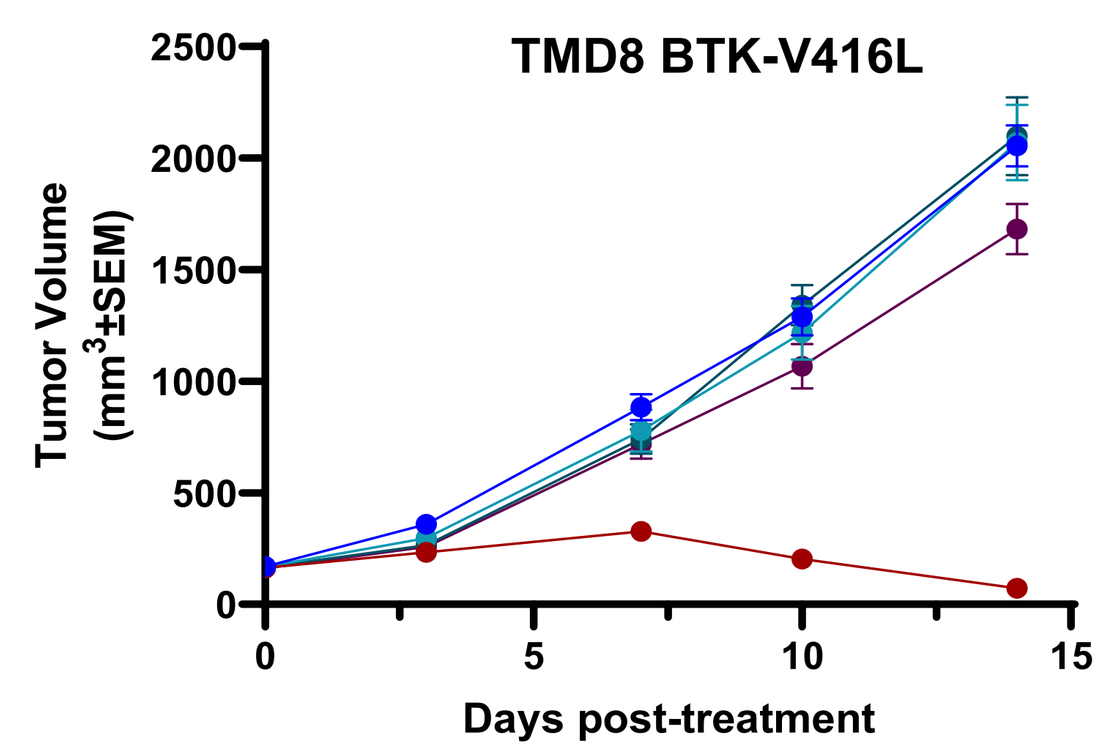
TMD8 BTK-V416L
Tumor Volume (mm3±SEM)
Days post-treatment

	TGI(%), D14	p-value (compared with vehicle)
Vehicle	na	na
Zanubrutinib , 20mpk BID	104.58	<0.0001
Acalabrutinib, 4mpk BID	-0.84	ns
Acalabrutinib, 12.5mpk BID	-2.39	ns
Pirtobrutinib, 50mpk BID	19.35	ns

Figure 3. TMD8 cells expressing WT or V416L mutant BTK were inoculated subcutaneously into female NCG mice for *in vivo* efficacy evaluation. **A).** At clinically relevant doses (20 mpk BID for zanu and 4 mpk BID for acala), both drugs and pirto induce significant tumor regression in TMD8 WT xenografts, with zanu demonstrating better efficacy; **B).** Administration of 20 mpk BID zanu resulted in significant regression of TMD8 V416L xenografts, while acala at 4 mpk BID did not achieve significant TGI. Neither a higher dose of acala (12 mpk BID) nor 50 mpk BID of pirto showed significant efficacy.

4. Structural simulation suggests mild clashes in the binding of acalaor pirto with BTK-V416L

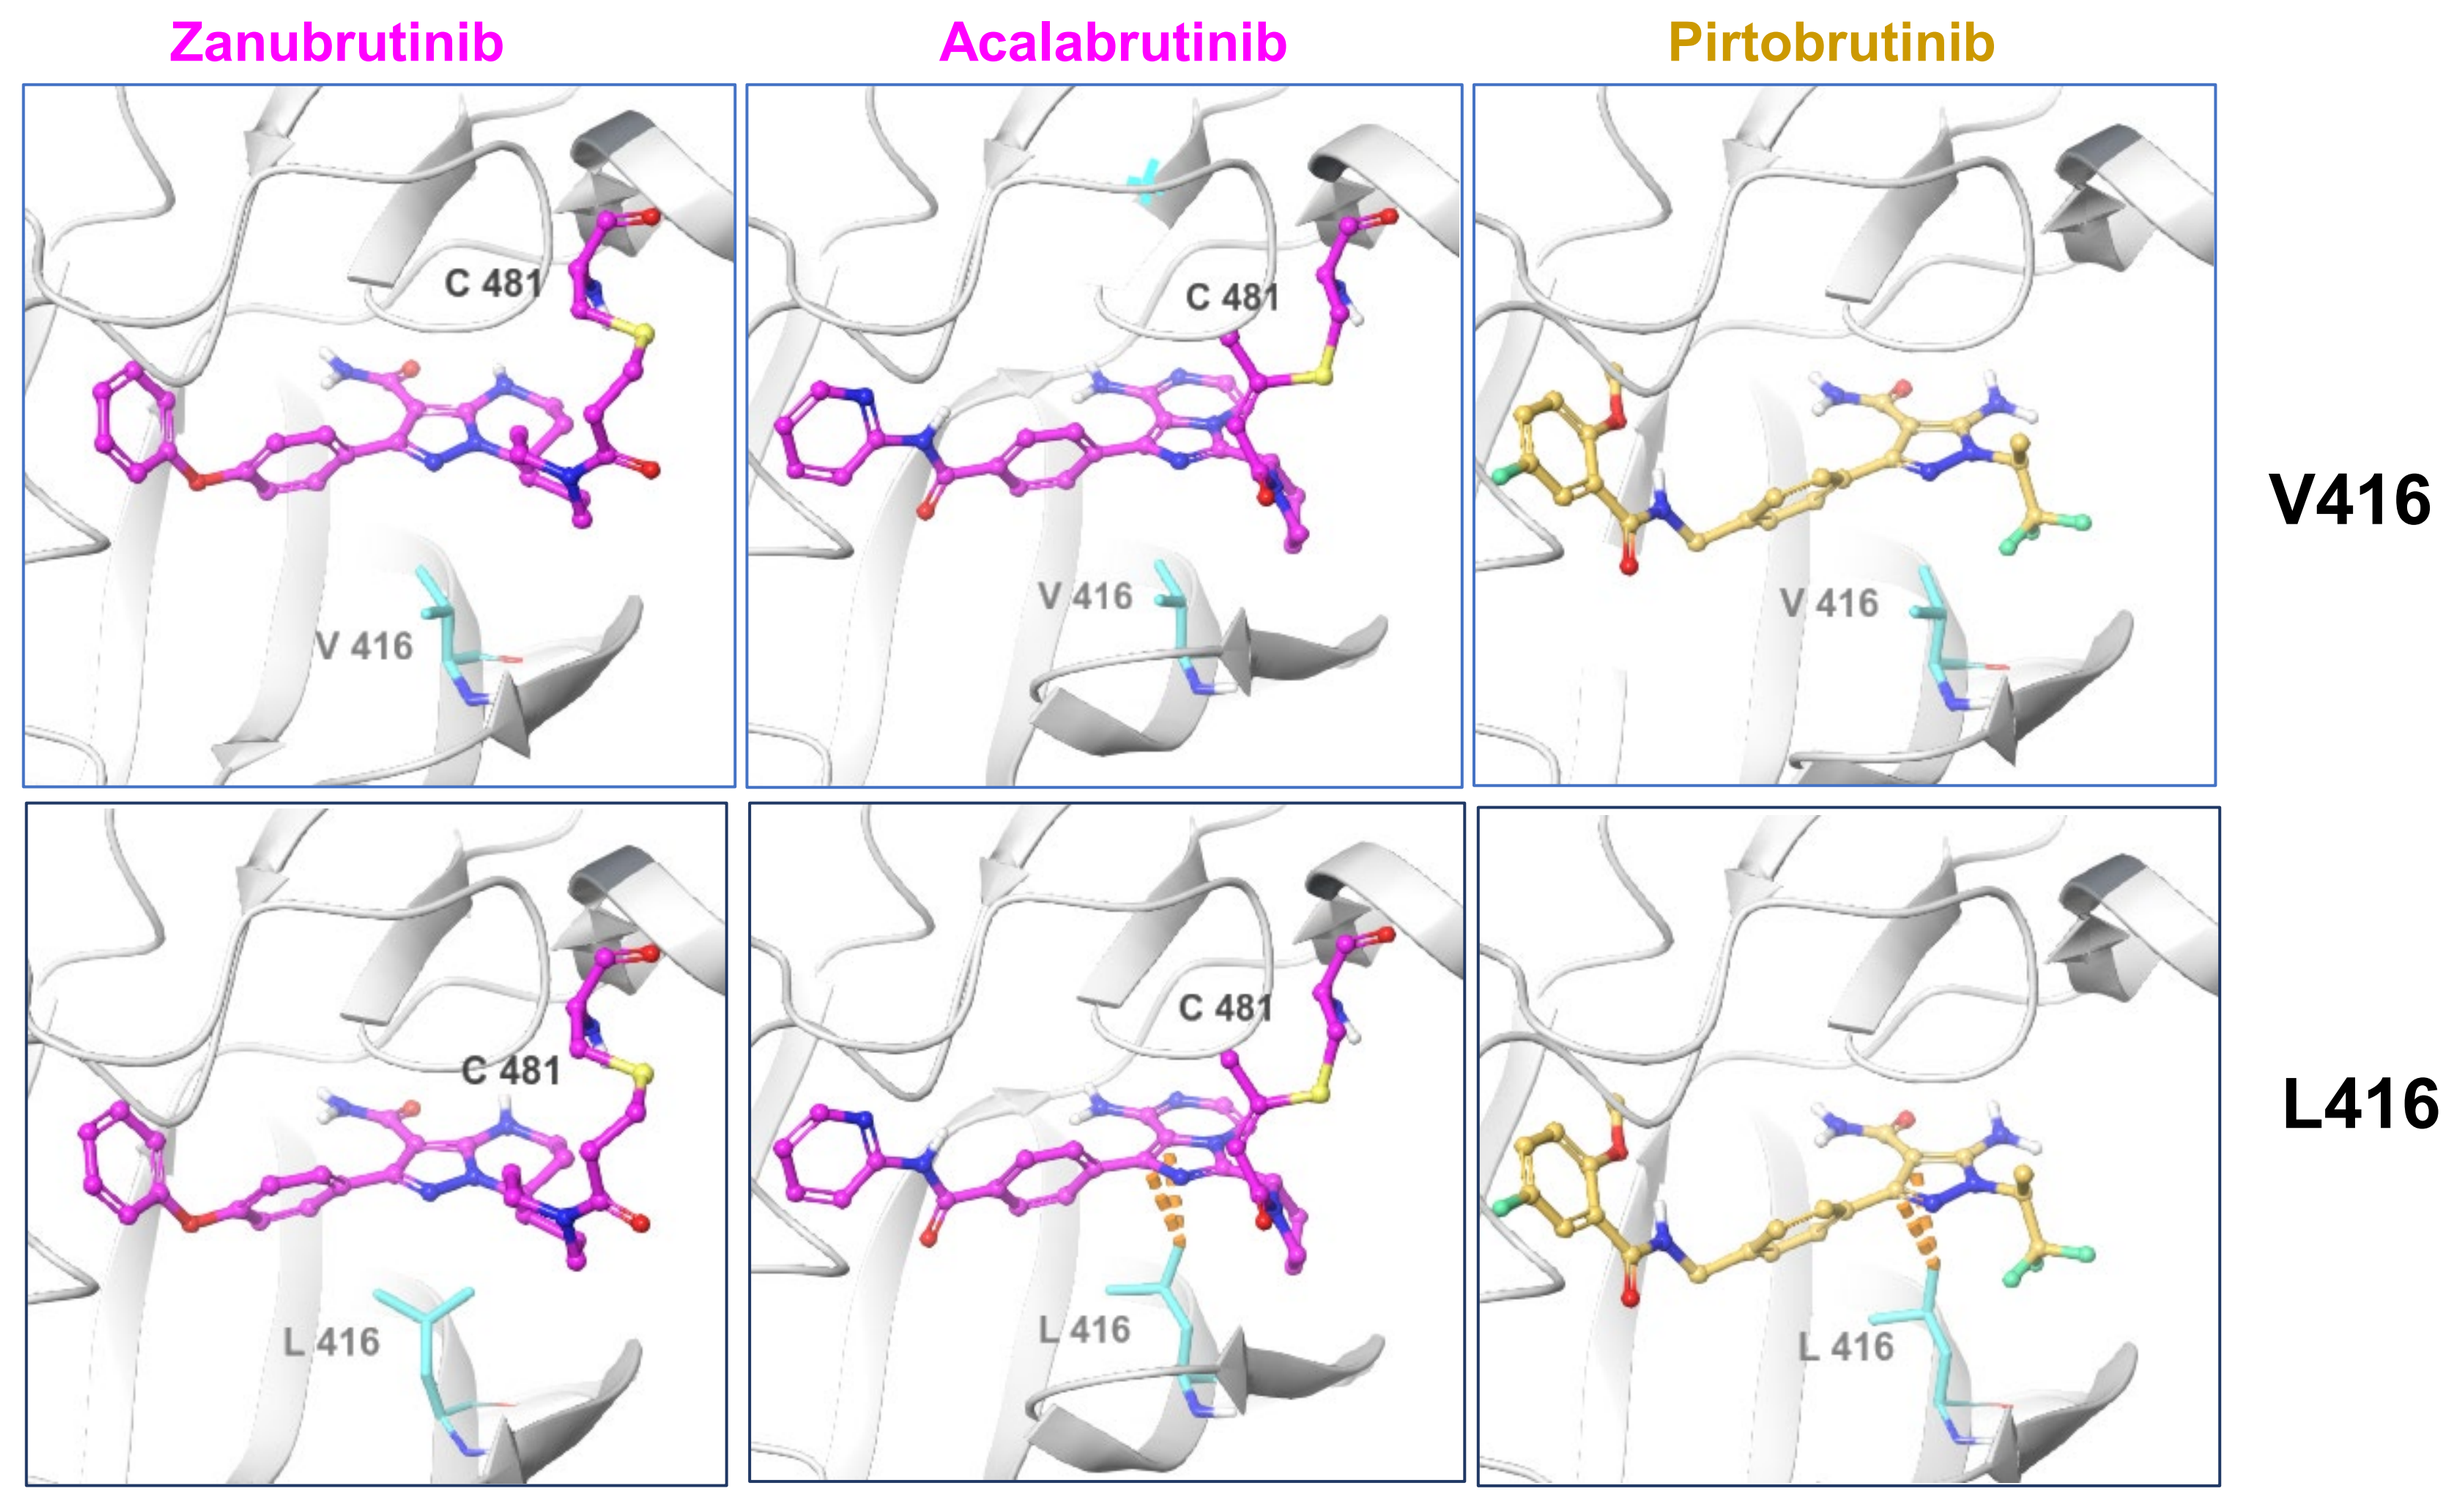
Zanubrutinib
Acalabrutinib
Pirtobrutinib
C 481
V 416
L 416
V416
L416

Figure 4. Computational structural modeling predicts that V416L mutation in BTK creates steric clashes with acala and pirto at the binding site, whereas zanu binding remains unaffected.

5. Zanu efficiently binds to BTK-V416L as evidenced by BTK occupancy in TMD8 cells *in vitro*

A.

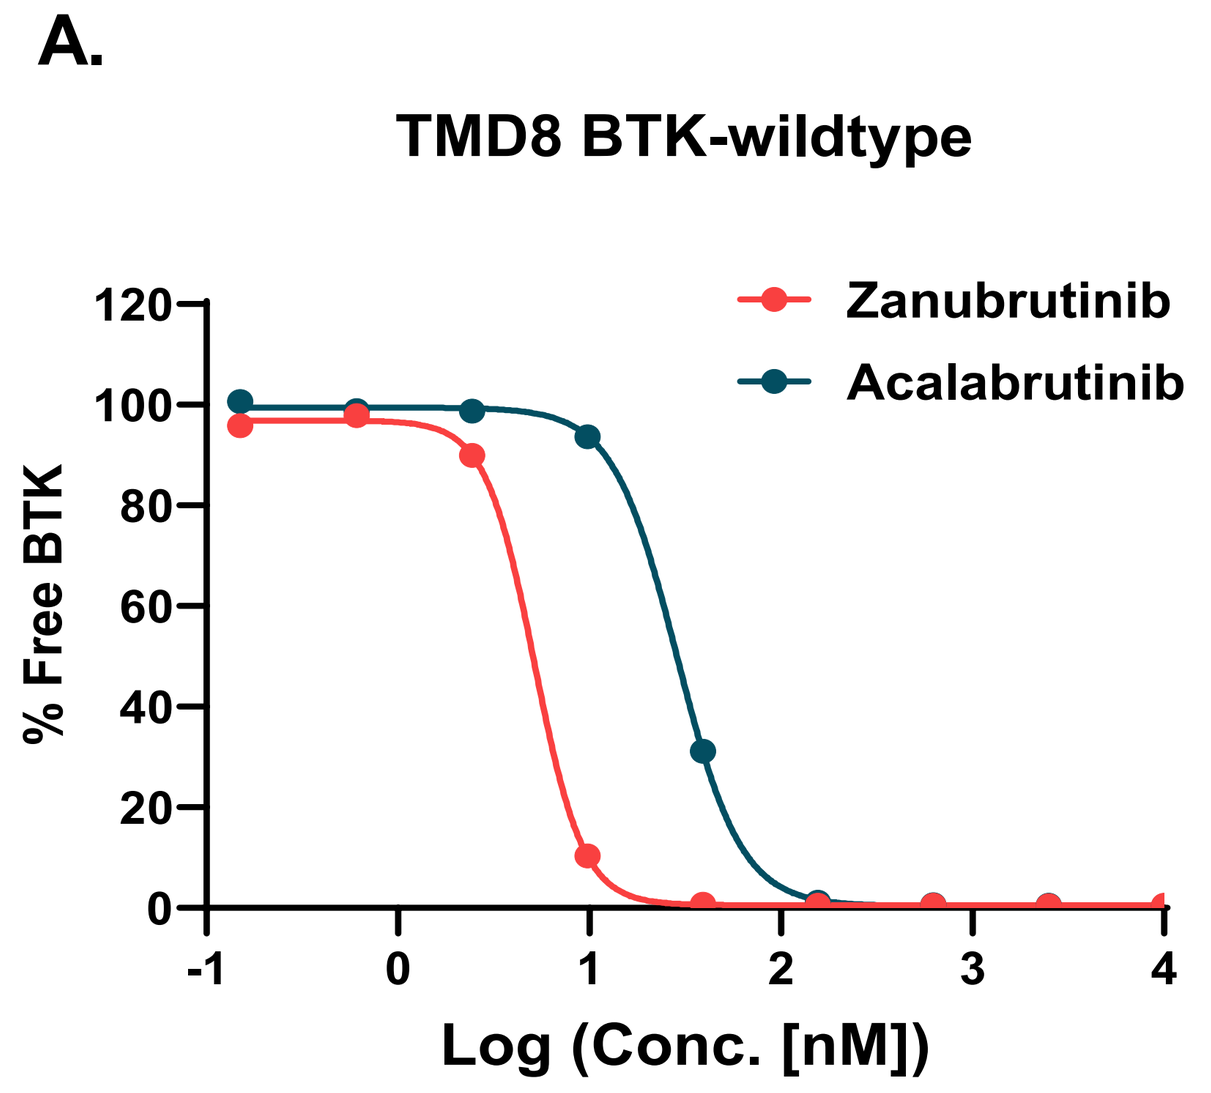
TMD8 BTK-wildtype
% Free BTK
Log (Conc. [nM])
Zanubrutinib
Acalabrutinib

B.

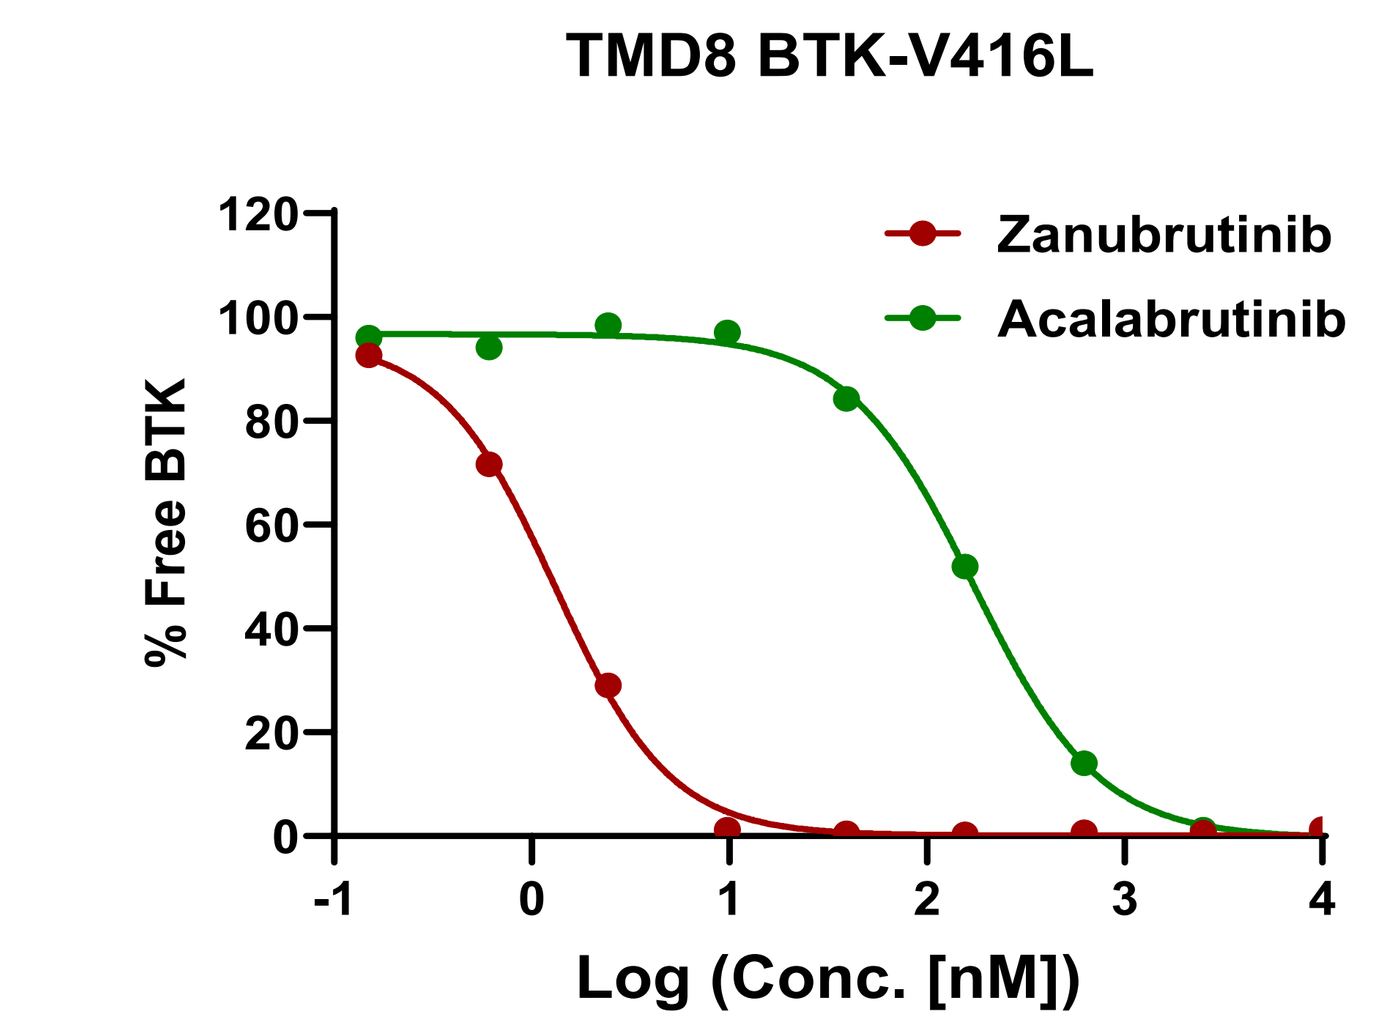
TMD8 BTK-V416L
% Free BTK
Log (Conc. [nM])
Zanubrutinib
Acalabrutinib

Figure 5. TMD8 BTK-WT and BTK-V416L mutant cells were treated with zanu or acala for 2h. Free BTK were examined using an enzyme-linked immunosorbent assay (ELISA) with biotinylated probes. **A).** Zanu is more potent than acala in binding to WT BTK in TMD8 cells; **B).** V416L mutation significantly disrupted the binding with acala, while the binding with zanu remains unaffected

6. Zanu, but not acala or pirto, efficiently inhibits BTK phosphorylation and its downstream signal in TMD8 cells

A.

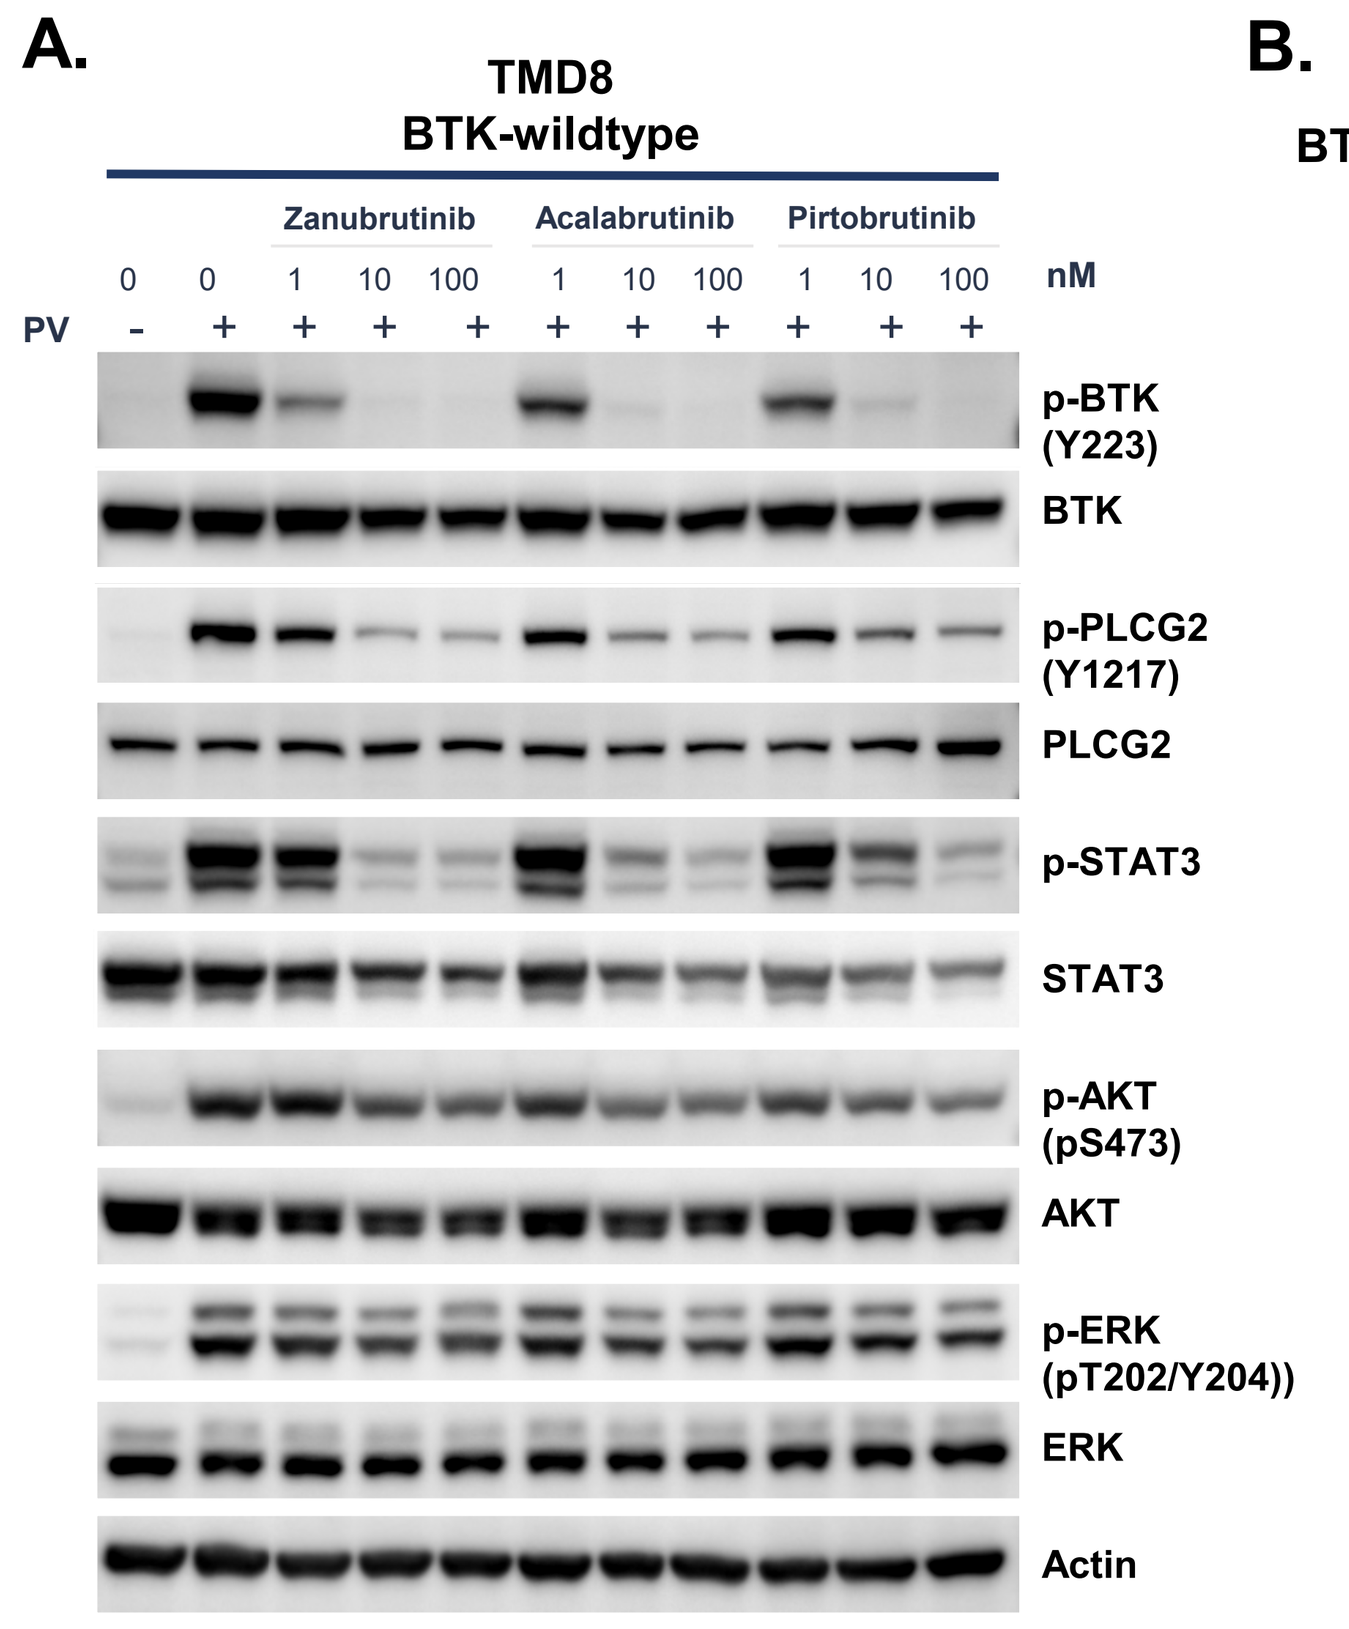
TMD8 BTK-wildtype
Zanubrutinib
Acalabrutinib
Pirtobrutinib
nM
PV
p-BTK (Y223)
BTK
p-PLCG2 (Y1217)
PLCG2
p-STAT3
STAT3
p-AKT (pS473)
AKT
p-ERK (pT202/Y204))
ERK
Actin

B.

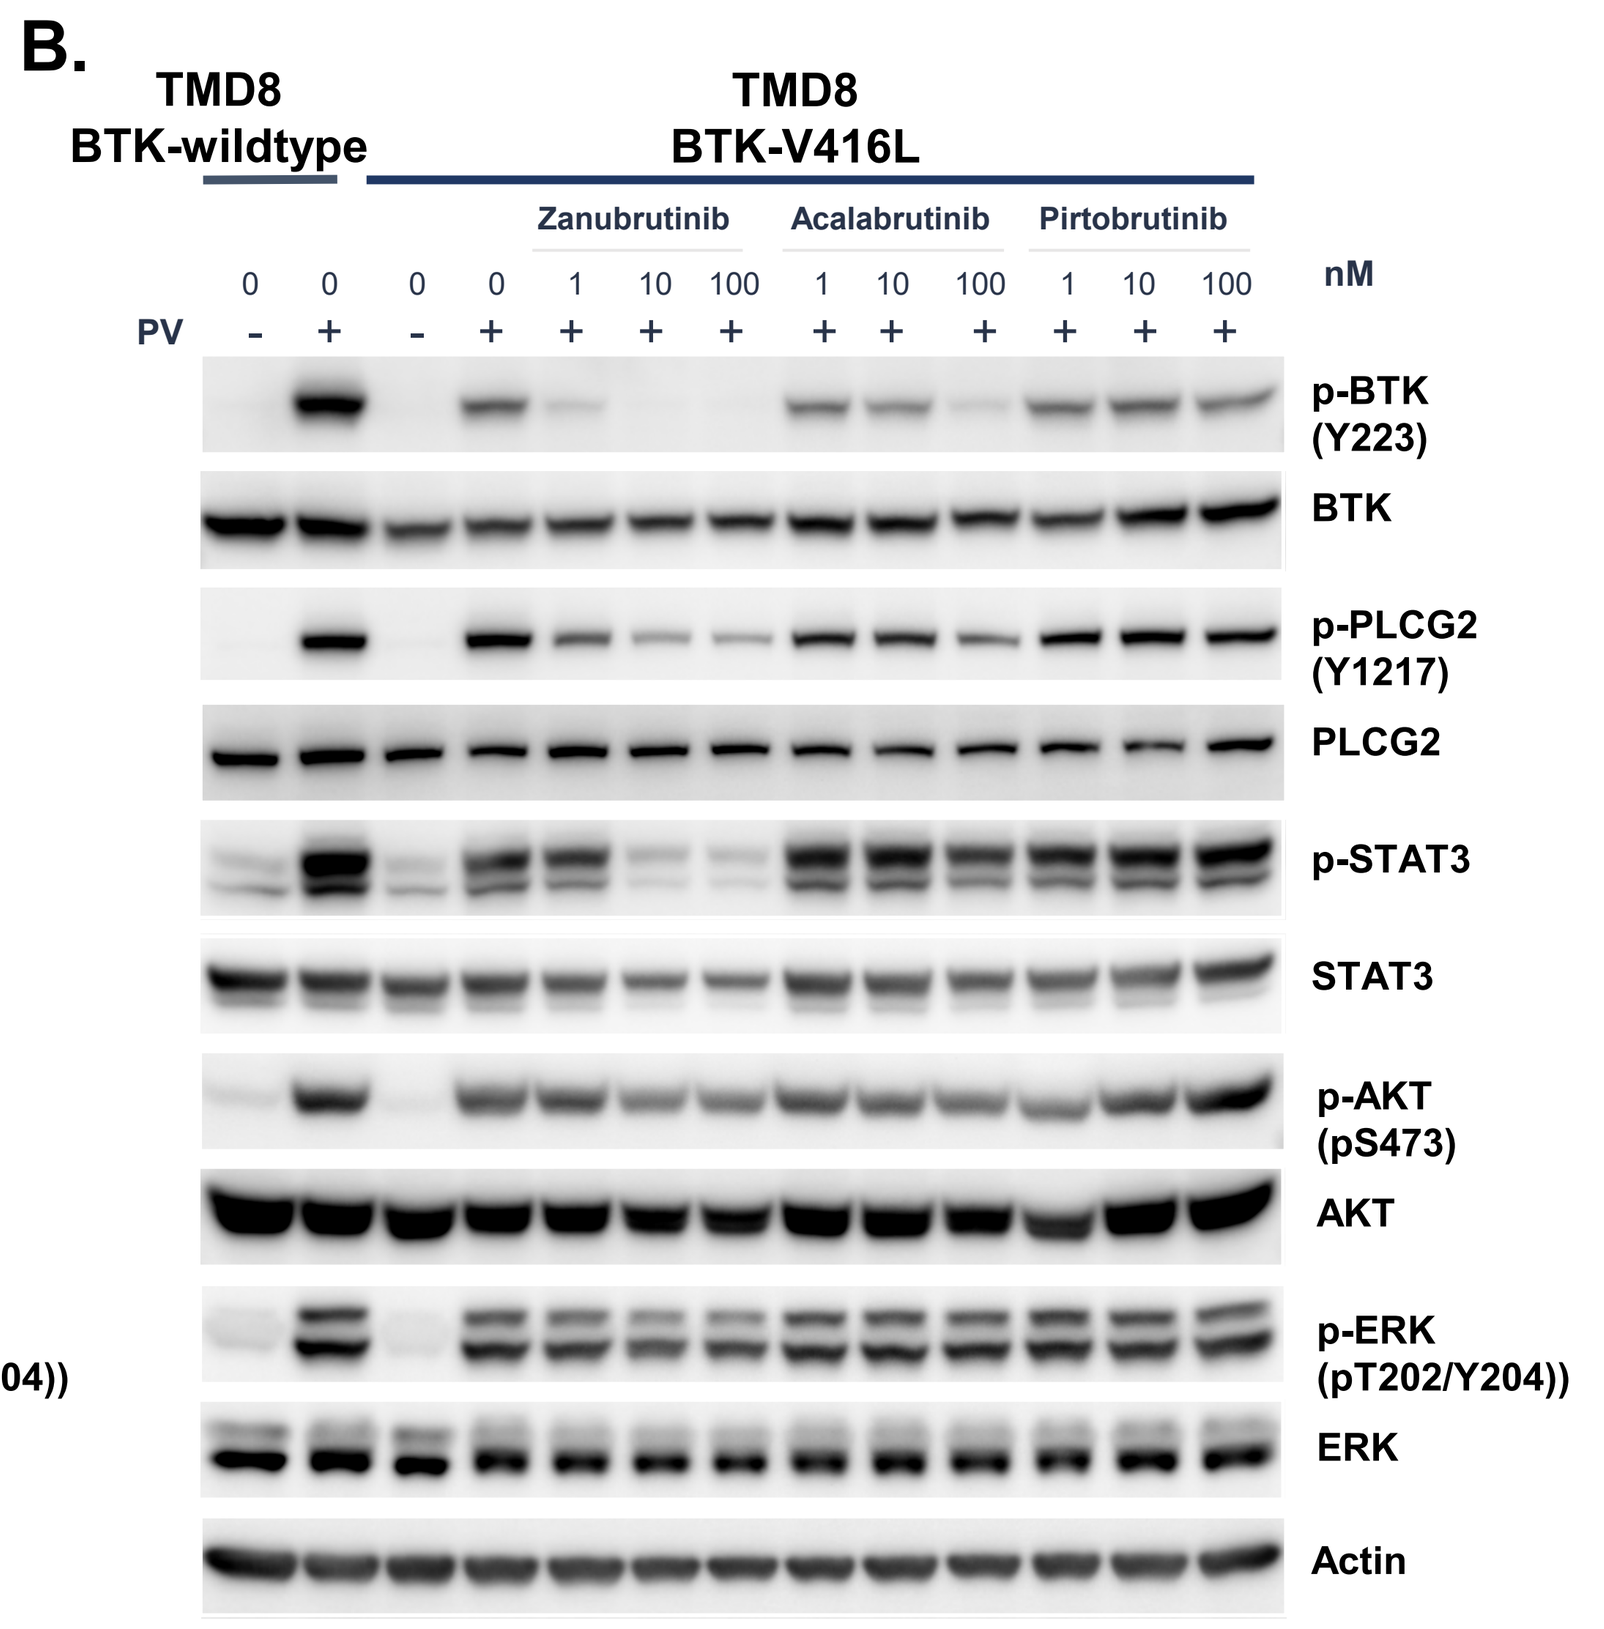
TMD8 BTK-wildtype
TMD8 BTK-V416L
Zanubrutinib
Acalabrutinib
Pirtobrutinib
nM
PV
p-BTK (Y223)
BTK
p-PLCG2 (Y1217)
PLCG2
p-STAT3
STAT3
p-AKT (pS473)
AKT
p-ERK (pT202/Y204))
ERK
Actin

Figure 6. Cells were treated with BTK inhibitors for 24 h, then incubated with pervanadate (PV) for 20 minutes. The whole cell lysate was prepared for western blot. **A).** In WT TMD8 cells, all BTK inhibitors inhibited the kinase activity of BTK and its downstream signal dose-dependently; **B).** In BTK-V416L expressing cells, only zanu but not acala or pirto potently inhibited BTK Y223 phosphorylation and downstream signals.

7. *In vivo* pharmacodynamics (PD) confirms that zanu but not acala is more efficient in occupying BTK-V416L mutated tumors

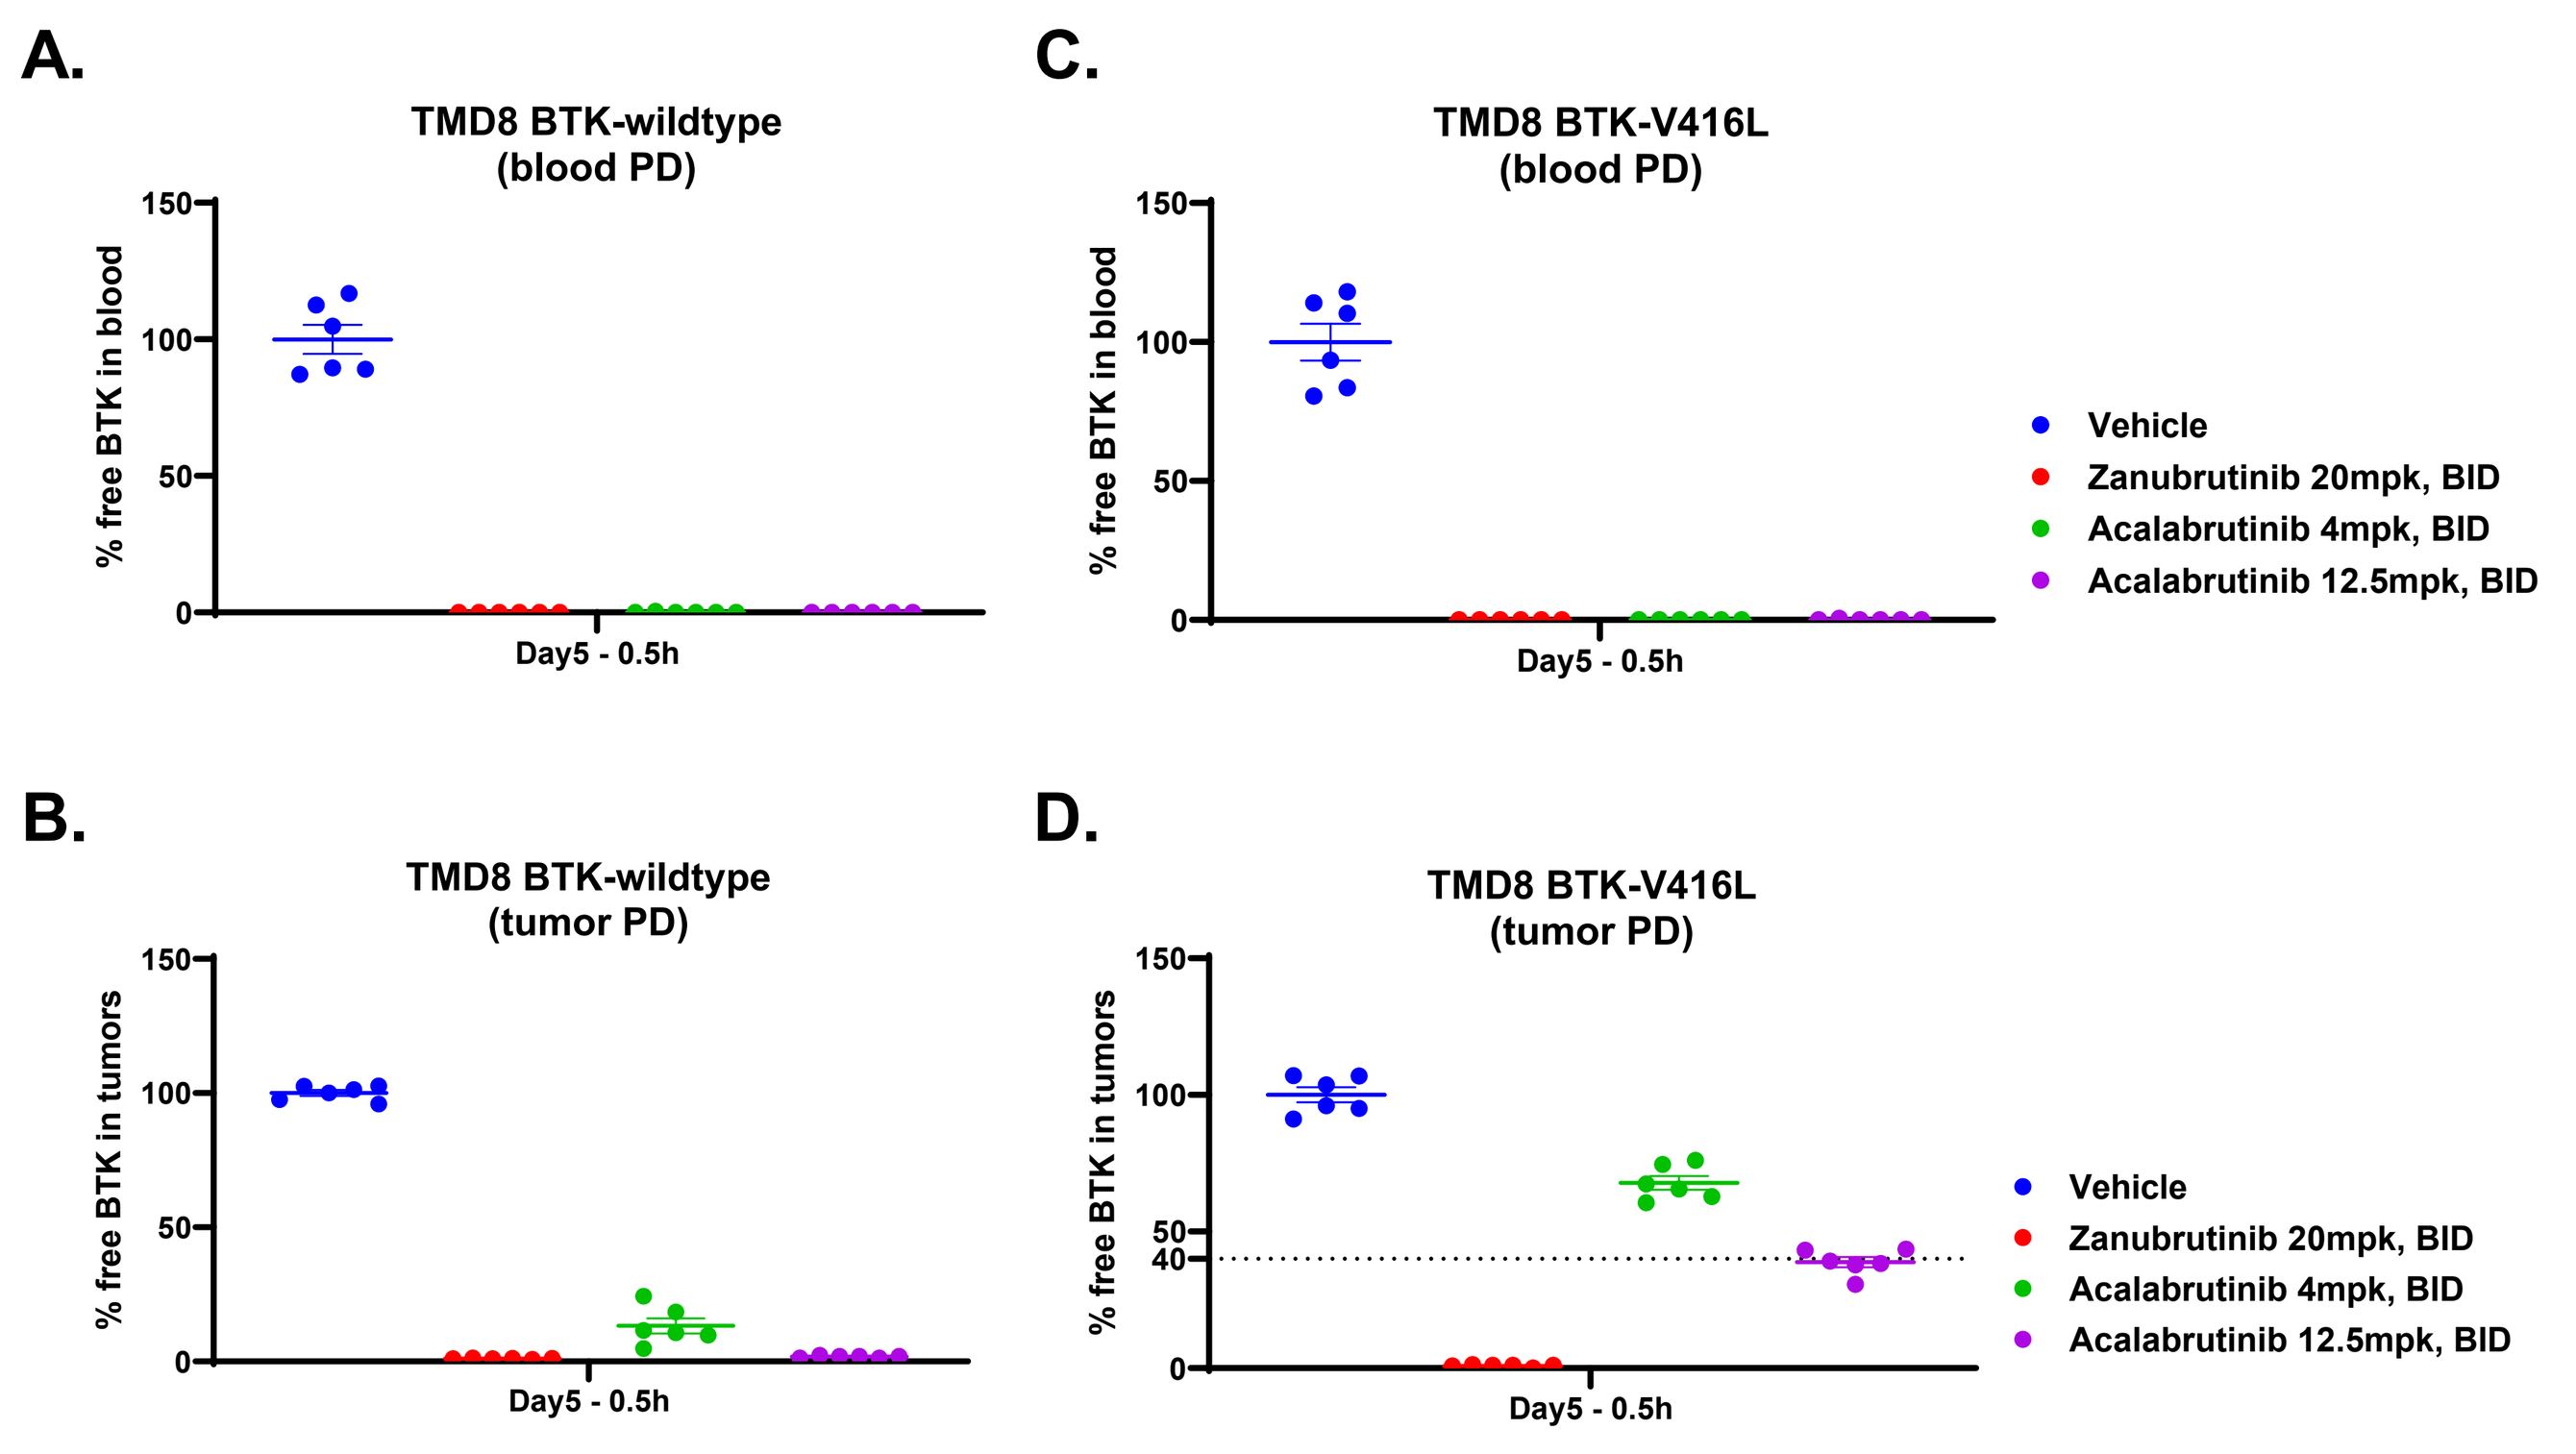
A.
TMD8 BTK-wildtype (blood PD)
% free BTK in blood
Day5 - 0.5h
C.
TMD8 BTK-V416L (blood PD)
B.
TMD8 BTK-wildtype (tumor PD)
% free BTK in tumors
D.
TMD8 BTK-V416L (tumor PD)
Vehicle
Zanubrutinib 20mpk, BID
Acalabrutinib 4mpk, BID
Acalabrutinib 12.5mpk, BID

Figure 7. Cancer cells were inoculated subcutaneously into NOG mice for tumor growth of ~2 weeks. Then mice were grouped based on their body weight and tumor volumes and dosed by zanu or acala at indicated doses for 4 days. On day5, blood and tumors were sampled 0.5 hours after 1st dosing. **A) & C).** In mouse blood, where all BTK are WT, fully BTK occupation by zanu and acala was observed; **B).** In WT tumors, at their clinically relevant doses, zanu demonstrated complete BTK occupancy while acala did not. Acala showed complete binding at 12.5mpk BID dose; **D).** In V416L mutated tumors, zanu's occupation of BTK remains full, while acala's are significantly blocked.

Conclusion

These data demonstrate that zanu retains potent antitumor activity against TMD8 cells expressing BTK-V416L, whereas acala and pirto may be attenuated by steric clashes. Thus, we hypothesize that clonal expansion of BTK-V416L is less likely to occur in patients with B cell malignancies (*e.g.* CLL) treated with zanu, whereas it may lead to resistance in patients treated with acala or pirto. This hypothesis should be validated in clinical studies.

AACR Annual Meeting 2025 April 25-30, 2025. Chicago, Illinois

AACR 2025