Real-World Chronic Lymphocytic Leukemia/Small Lymphocytic Lymphoma Treatment Patterns at Florida Cancer Specialists & Research Institute Among Patients Receiving Zanubrutinib Immediately Following Prior BTKi Therapy

Amanda Warner, MS, BSN, RN[1], Xiaoliang Wang, PhD, MPH[2], Ashley Hernandez, BSN, RN[1], Christian Okitondo, MS[1], Karen Carr, PharmD[2], Gregory A. Maglinte, PhD, MPH[2], Judah Friedman, MD[1]

[1]Florida Cancer Specialists & Research Institute, Tampa, FL, USA; [2]BeOne Medicines, Ltd., San Carlos, CA, USA

CONCLUSIONS

- In a real-world setting, in patients diagnosed with CLL/SLL and treated with a BTKi, TLAEs were the most common reason for switching from a prior BTKi to zanubrutinib
- The TLAEs that commonly resulted in treatment discontinuation were atrial fibrillation, rash, fatigue, nausea, and vomiting
- Once initiated on zanubrutinib, recurrence of prior TLAEs was not common, cardiac TLAEs did not recur, and patients were likely to remain on treatment to the end of follow-up independent of the prior BTKi
- Median treatment duration was longer for zanubrutinib (37.2 months) than either ibrutinib or acalabrutinib (28.2 and 6.0 months, respectively)
- Overall, the findings were consistent with clinical trial research that has demonstrated that zanubrutinib is safe and effective in treating CLL/SLL, even among patients with prior BTKi therapy

INTRODUCTION

- Since their introduction, the Bruton tyrosine kinase inhibitors (BTKis) have improved outcomes for patients with various B-cell malignancies and are currently a preferred treatment recommended by NCCN guidelines[1-2]
- Exploring treatment switching in patients with B-cell malignancies had become increasingly important due to advances in the second generation of BTKis[3]
- In 2023, a phase 2 clinical trial (NCT04116437) demonstrated that treatment with zanubrutinib was a safe and effective option for patients with B-cell malignancies previously treated with but intolerant of another BTKi (ibrutinib, acalabrutinib)[4-5]
- Little real-world evidence, however, has been available to demonstrate the value of zanubrutinib in patients switching from another BTKi
- Examining patients diagnosed with chronic lymphocytic leukemia or small lymphocytic leukemia (CLL/SLL) and switched to zanubrutinib immediately following treatment with another BTKi at the Florida Cancer Specialists & Research Institute (FCS) offers the opportunity to address this question

OBJECTIVES

- To describe real-world demographic and clinical characteristics, treatment patterns, and clinical outcomes, including treatment-limiting adverse events (TLAEs) and reasons for BTKi discontinuation, in patients with CLL/SLL who initiated zanubrutinib following switch from ibrutinib or acalabrutinib
- To assess treatment discontinuation and TLAEs following zanubrutinib initiation after treatment switching

METHODS

Population and Sample

- Patients were identified from electronic medical records (EMR) and enterprise data warehouse records from FCS
- FCS is one of the largest medical oncology/hematology practices in Florida with over 90 facilities across the state, serving nearly 100,000 new patients annually, and with over 500 providers
- Eligible patients were diagnosed with CLL or SLL (ICD-10 C91.1 or C83.0) and initiated on zanubrutinib between November 1, 2019 and March 1, 2025
- Patients were included in the analysis if they received another BTKi (ibrutinib or acalabrutinib) immediately prior to zanubrutinib treatment
- The index date was the start of zanubrutinib treatment. Patients were followed until zanubrutinib discontinuation, disenrollment from the network, death, or end of the study period on June 1, 2025 (**Figure 1**)

Outcomes

- Patient demographic and clinical characteristics, including age, gender, ethnicity, insurance type, ECOG status, and Rai stage at diagnosis, were collected and examined overall and by treatment cohort
- Treatment history, including previous lines of therapy, prior BTKi therapy, TLAEs associated with the previous BTKi, and reason for discontinuation of prior BTKi, were examined
- For zanubrutinib, time on treatment, reason for discontinuation, and TLAEs were also examined
- Disease control rate (DCR) was defined as the percentage of patients with advanced cancer whose therapeutic intervention has led to a complete response, partial response, or stable disease[6]

Figure 1. Patient Identification Timeline

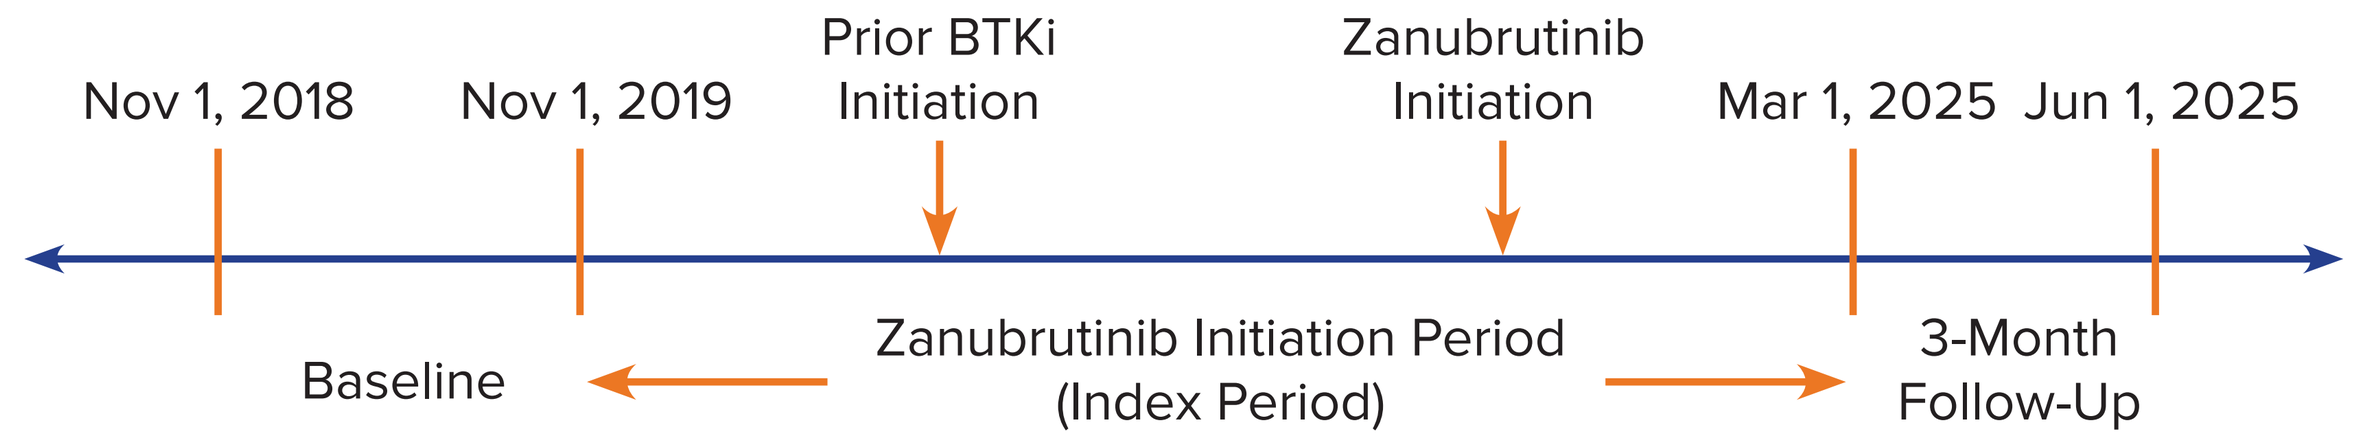

Nov 1, 2018
Nov 1, 2019
Prior BTKi Initiation
Zanubrutinib Initiation
Mar 1, 2025
Jun 1, 2025
Baseline
Zanubrutinib Initiation Period (Index Period)
3-Month Follow-Up

BTKi, Bruton tyrosine kinase inhibitor.

Study Design

- This was a retrospective observational study
- Once identified and selected, patients were stratified into cohorts based on their immediate prior BTKi

RESULTS

Demographic and Clinical Characteristics

- A total of 100 patients met the inclusion criteria for the study (**Table 1**)

Table 1. Baseline Demographics and Clinical Characteristics

		Immediate Prior BTKi	
Variable	Overall N=100	Acalabrutinib N=46	Ibrutinib N=54
Age at Index			
Median (IQR)	75.7 (67.3, 81.0)	76.1 (69.3, 80.9)	75.0 (65.2, 81.2)
Age at First BTKi			
Median (IQR)	71.5 (64.2, 77.5)	74.7 (67.7, 77.5)	69.7 (62.9, 77.1)
Gender			
Female	43 (43.0%)	22 (47.8%)	21 (38.9%)
Male	57 (57.0%)	24 (52.2%)	33 (61.1%)
Race			
Black or African American	3 (3.0%)	1 (2.2%)	2 (3.7%)
White	84 (84.0%)	40 (87.0%)	44 (81.5%)
Other	10 (10.0%)	3 (6.5%)	7 (13.0%)
Unknown	3 (3.0%)	2 (4.3%)	1 (1.9%)
Ethnicity			
Hispanic or Latino	4 (4.0%)	2 (4.3%)	2 (3.7%)
Not Hispanic or Latino	88 (88.0%)	39 (84.8%)	49 (90.7%)
Unknown	8 (8.0%)	5 (10.9%)	3 (5.6%)
Insurance Type			
Commercial	9 (9.0%)	4 (8.7%)	5 (9.3%)
Medicare	88 (88.0%)	39 (84.8%)	49 (90.7%)
Medicaid	2 (2.0%)	2 (4.3%)	0 (0.0%)
Government	1 (1.0%)	1 (2.2%)	0 (0.0%)
Rai Stage or Binet Stage at Diagnosis			
0	43 (43.0%)	19 (41.3%)	24 (44.4%)
I	28 (28.0%)	13 (28.3%)	15 (27.8%)
II	12 (12.0%)	5 (10.9%)	7 (13.0%)
III	3 (3.0%)	2 (4.3%)	1 (1.9%)
IV	5 (5.0%)	3 (6.5%)	2 (3.7%)
A	3 (3.0%)	1 (2.2%)	2 (3.7%)
B	4 (4.0%)	2 (4.3%)	2 (3.7%)
C	2 (2.0%)	1 (2.2%)	1 (1.9%)
ECOG PS at index			
0-1	72 (72.0%)	32 (69.6%)	40 (74.1%)
2-4	8 (8.0%)	3 (6.5%)	5 (9.3%)
Unknown	20 (20.0%)	11 (23.9%)	9 (16.7%)

BTKi, Bruton tyrosine kinase inhibitor; ECOG PS, Eastern Cooperative Oncology Group performance status; IQR, interquartile range.

Treatment History

- Immediately prior to treatment with zanubrutinib, 54.0% patients were treated with ibrutinib and 46.0% were treated with acalabrutinib (**Table 2**)
- For most patients (78.0%), zanubrutinib was the second-line treatment. Patients treated with ibrutinib were less likely to have received additional previous lines of treatment compared to those treated with acalabrutinib (14.9% vs 30.4%, respectively) (**Figure 2**)
- When ibrutinib or acalabrutinib was given as prior therapy, in any line of treatment, it was primarily prescribed as monotherapy (96.3% vs 84.8%) (**Table 2**)

Table 2. Prior BTKi Treatment Characteristics

		Immediate Prior BTKi	
Variable	Overall N=100	Acalabrutinib N=46	Ibrutinib N=54
Number of prior LOTs			
1	78 (78.0%)	32 (69.6%)	46 (85.2%)
2	12 (12.0%)	9 (19.6%)	3 (5.6%)
3	8 (8.0%)	3 (6.5%)	5 (9.3%)
4	2 (2.0%)	2 (4.3%)	0 (0.0%)
Number of prior BTKi			
1	89 (89.0%)	37 (80.4%)	52 (96.3%)
2	11 (11.0%)	9 (19.6%)	2 (3.7%)
Prior BTKi[a]			
Ibrutinib monotherapy	61 (61.0%)	9 (19.6%)	52 (96.3%)
Ibrutinib combination therapy	6 (6.0%)	0 (0.0%)	6 (11.1%)
Acalabrutinib monotherapy	41 (41.0%)	39 (84.8%)	2 (3.7%)
Acalabrutinib combination therapy	10 (10.0%)	10 (21.7%)	0 (0.0%)

[a]Categories are not mutually exclusive.
BTKi, Bruton tyrosine kinase inhibitor; LOT, line of therapy.

Figure 2. Treatment Pathways to Zanubrutinib

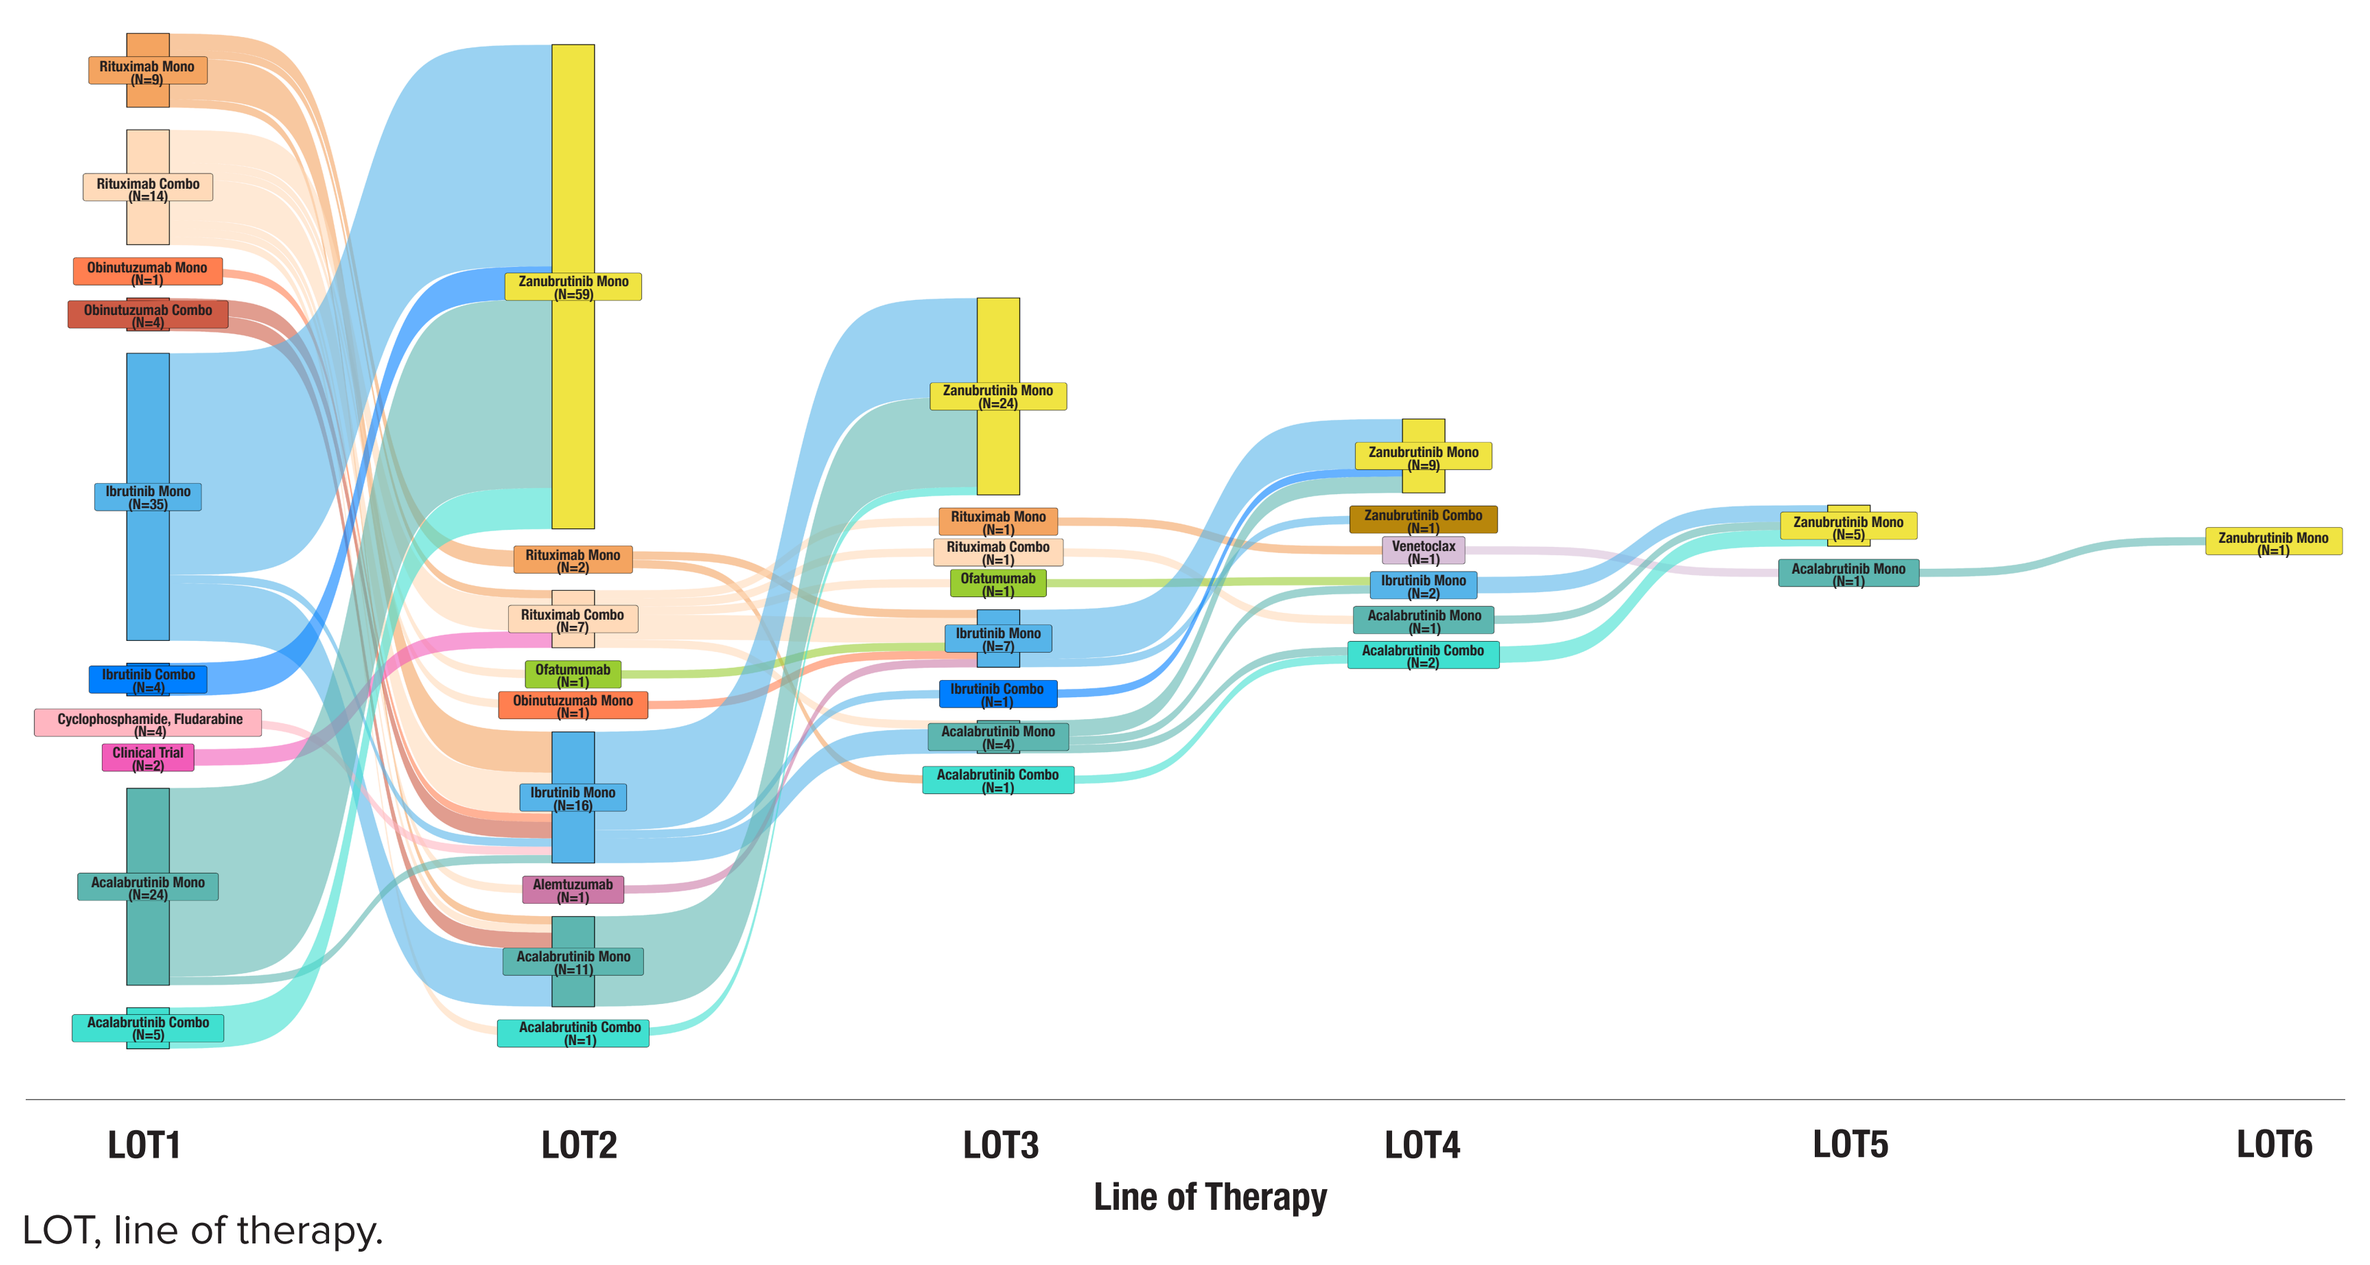

LOT1
LOT2
LOT3
LOT4
LOT5
LOT6
Line of Therapy

LOT, line of therapy.

Discontinuation of Prior BTKi

- The most common reason for discontinuation of all prior BTKi was TLAEs (76.0%), followed by physician/patient choice (17.0%), disease progression (12.0%), and other (9.0%) (**Figure 3**)
- Overall, 65.0% of patients discontinued the immediate prior BTKi due to TLAEs. The rate of discontinuation due to TLAEs was similar by cohort, 68.5% treated with ibrutinib and 60.9% treated with acalabrutinib
- Median treatment duration for ibrutinib was 28.2 months (IQR: 9.3-56.7) and for acalabrutinib, 6.0 months (IQR: 2.0-14.3)
- For patients who discontinued the immediate prior BTKi due to TLAEs, the most common reasons were atrial fibrillation (23.1%), rash (15.4%), fatigue (13.8%), nausea (9.2%), and vomiting (6.2%)
- Among patients who discontinued immediate prior ibrutinib due to TLAEs, the most common AEs were atrial fibrillation (40.5%), rash (10.8%), and fatigue (10.8%)
- Among patients who discontinued immediate prior acalabrutinib due to TLAEs, the most common AEs were rash (21.4%), nausea (17.9%), fatigue (17.9%), headaches (10.7%), diarrhea (19.7%), and vomiting (10.7%)

Figure 3. Reason for Discontinuation of Any Prior BTKi[a,b]

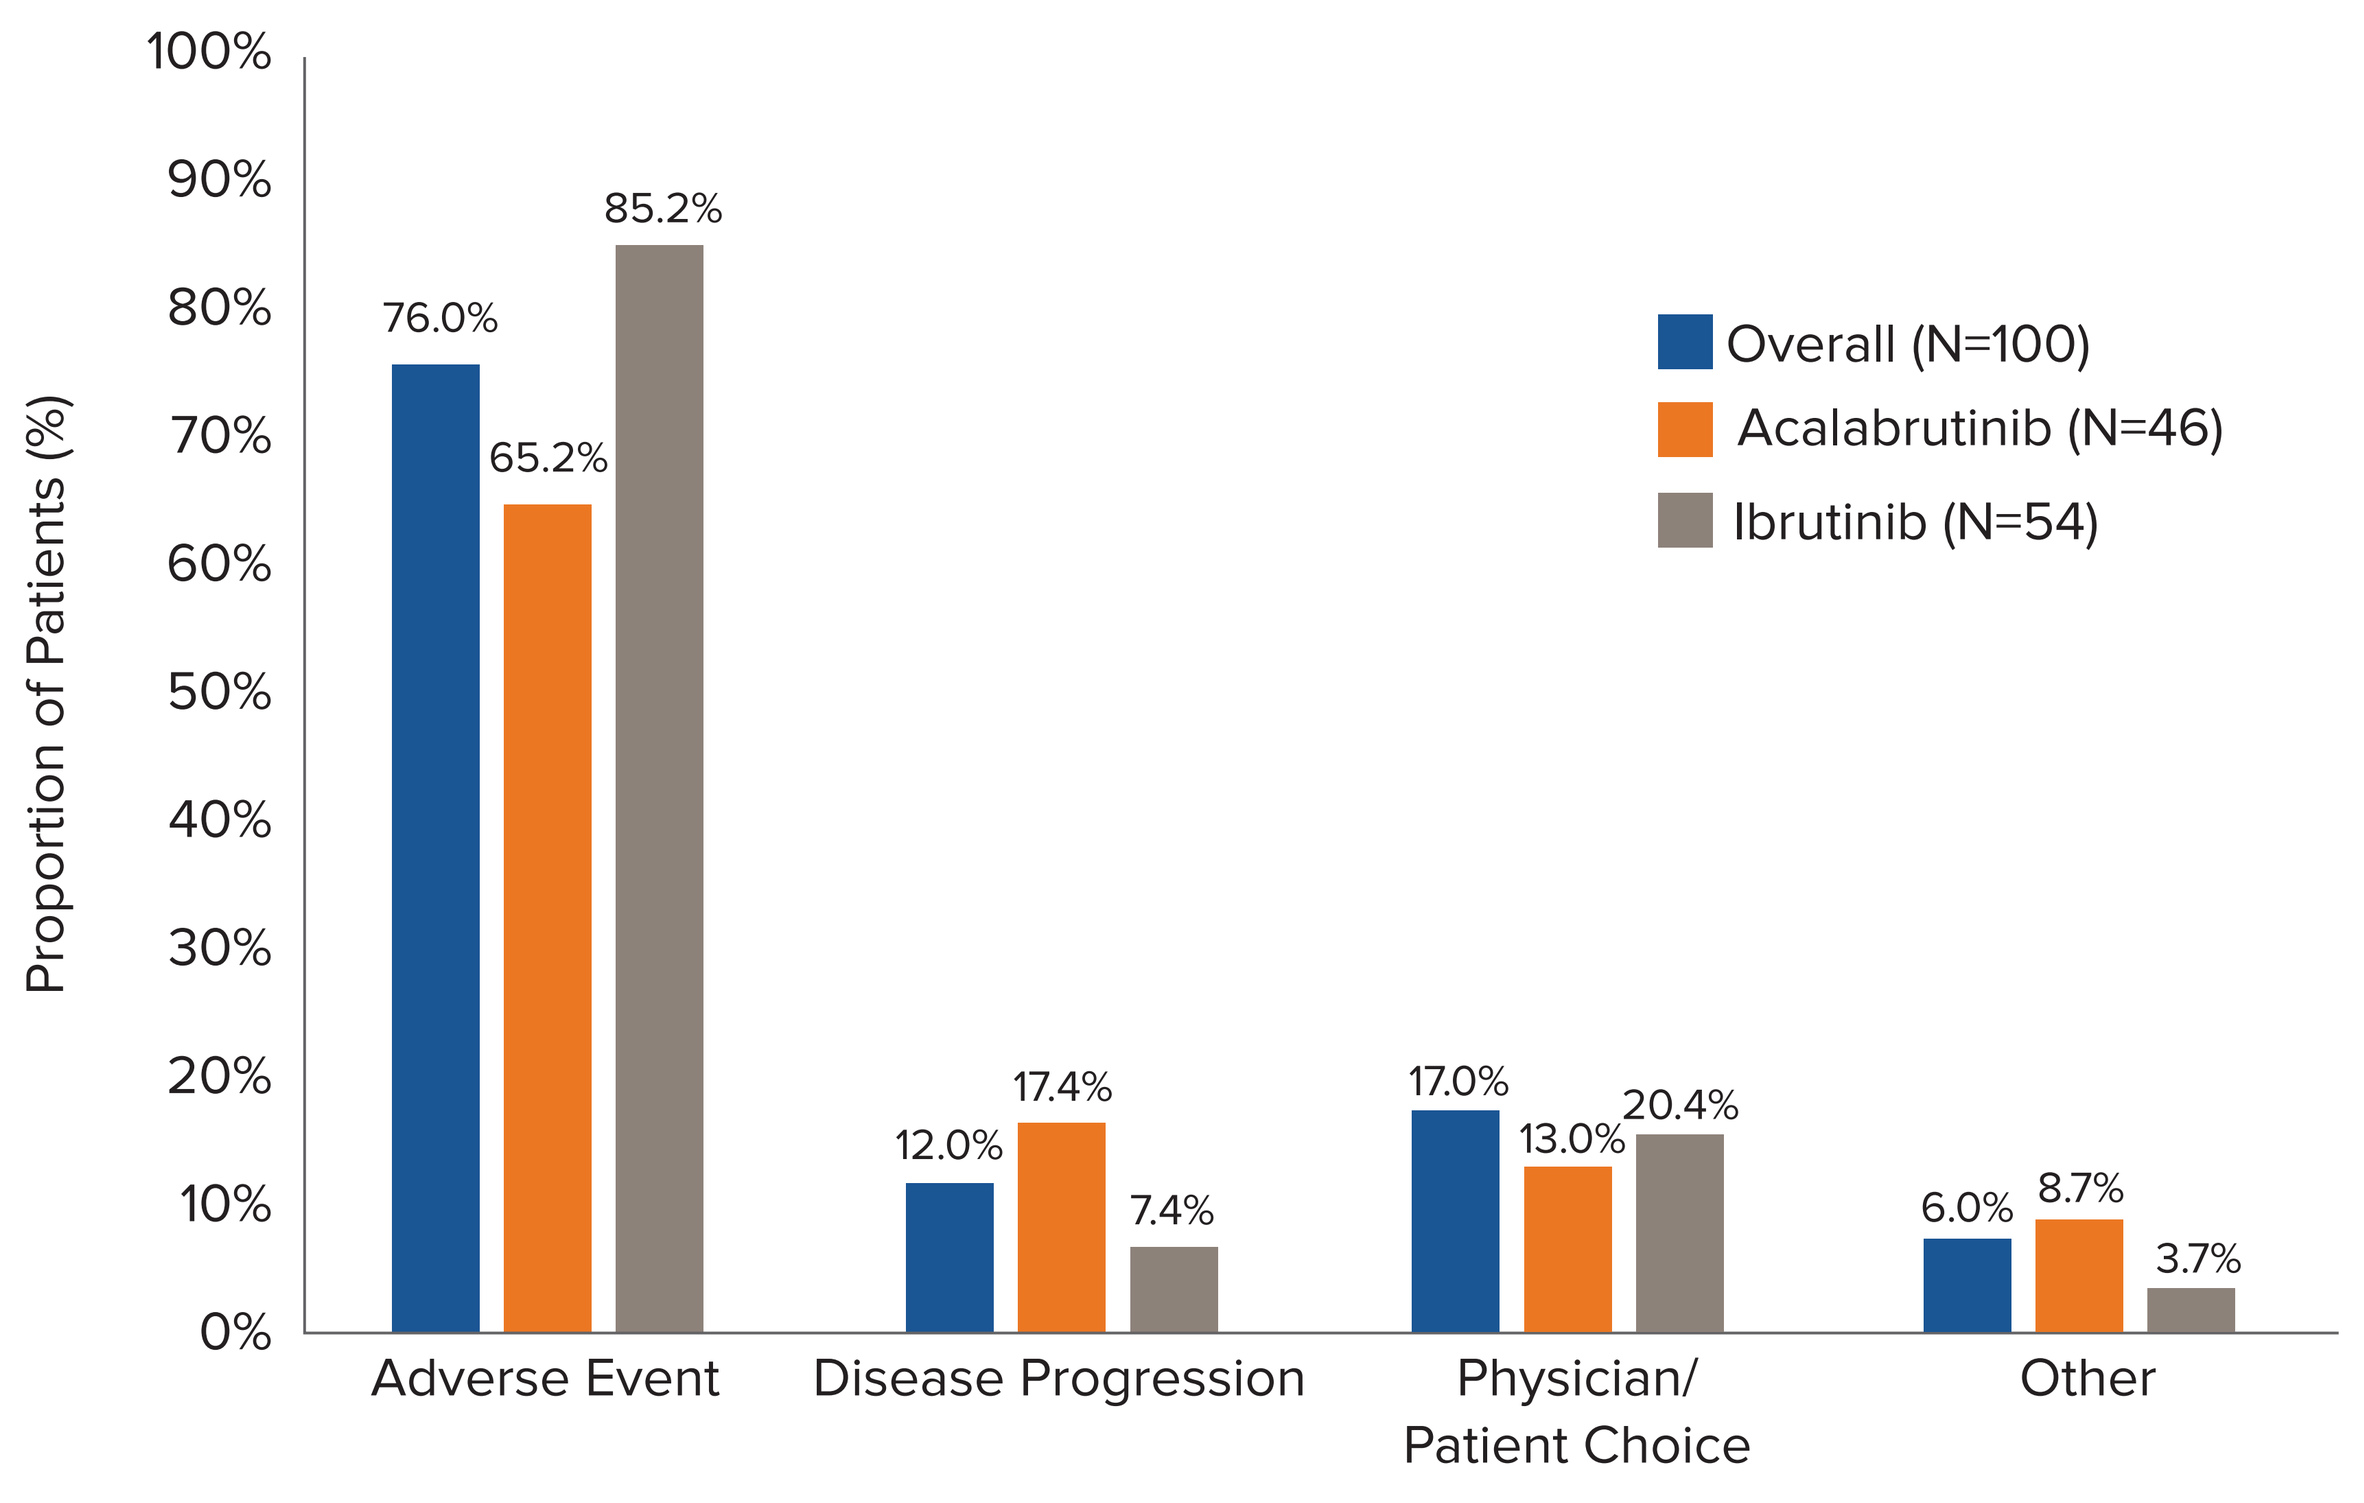

Proportion of Patients (%)
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
76.0%
65.2%
85.2%
12.0%
17.4%
7.4%
17.0%
13.0%
20.4%
6.0%
8.7%
3.7%
Adverse Event
Disease Progression
Physician/ Patient Choice
Other
Overall (N=100)
Acalabrutinib (N=46)
Ibrutinib (N=54)

[a]Among patients who discontinued; [b]Not mutually exclusive categories.
BTKi, Bruton tyrosine kinase inhibitor.

Outcomes for Zanubrutinib

- Median duration of zanubrutinib was 37.2 months (95% confidence interval: 17.3-NA)
- Of the 100 patients initiated on zanubrutinib, 58.0% remained on treatment until the end of the follow-up period. This finding did not differ by immediate prior BTKi therapy (ibrutinib: 57.4%; acalabrutinib: 58.7%)
- Of the 42.0% of patients who discontinued zanubrutinib, the most common reasons for discontinuation included TLAEs (38.1%), disease progression (21.4%), and physician/patient choice (16.6%). The most common TLAE leading to zanubrutinib discontinuation was rash (18.8%) (**Figure 4**)
- Recurrence of TLAEs associated with the prior BTKi after switching to zanubrutinib was not common, and there were no recurrences of cardiac TLAEs (**Figure 5**)
- Zanubrutinib provided an efficacy benefit in patients, as measured by a DCR of 82.0%, regardless of reason for discontinuation from prior BTKi (prior ibrutinib DCR was 84.0% and prior acalabrutinib DCR was 72.5%)

Figure 4. Reason for Discontinuation of Zanubrutinib[a]

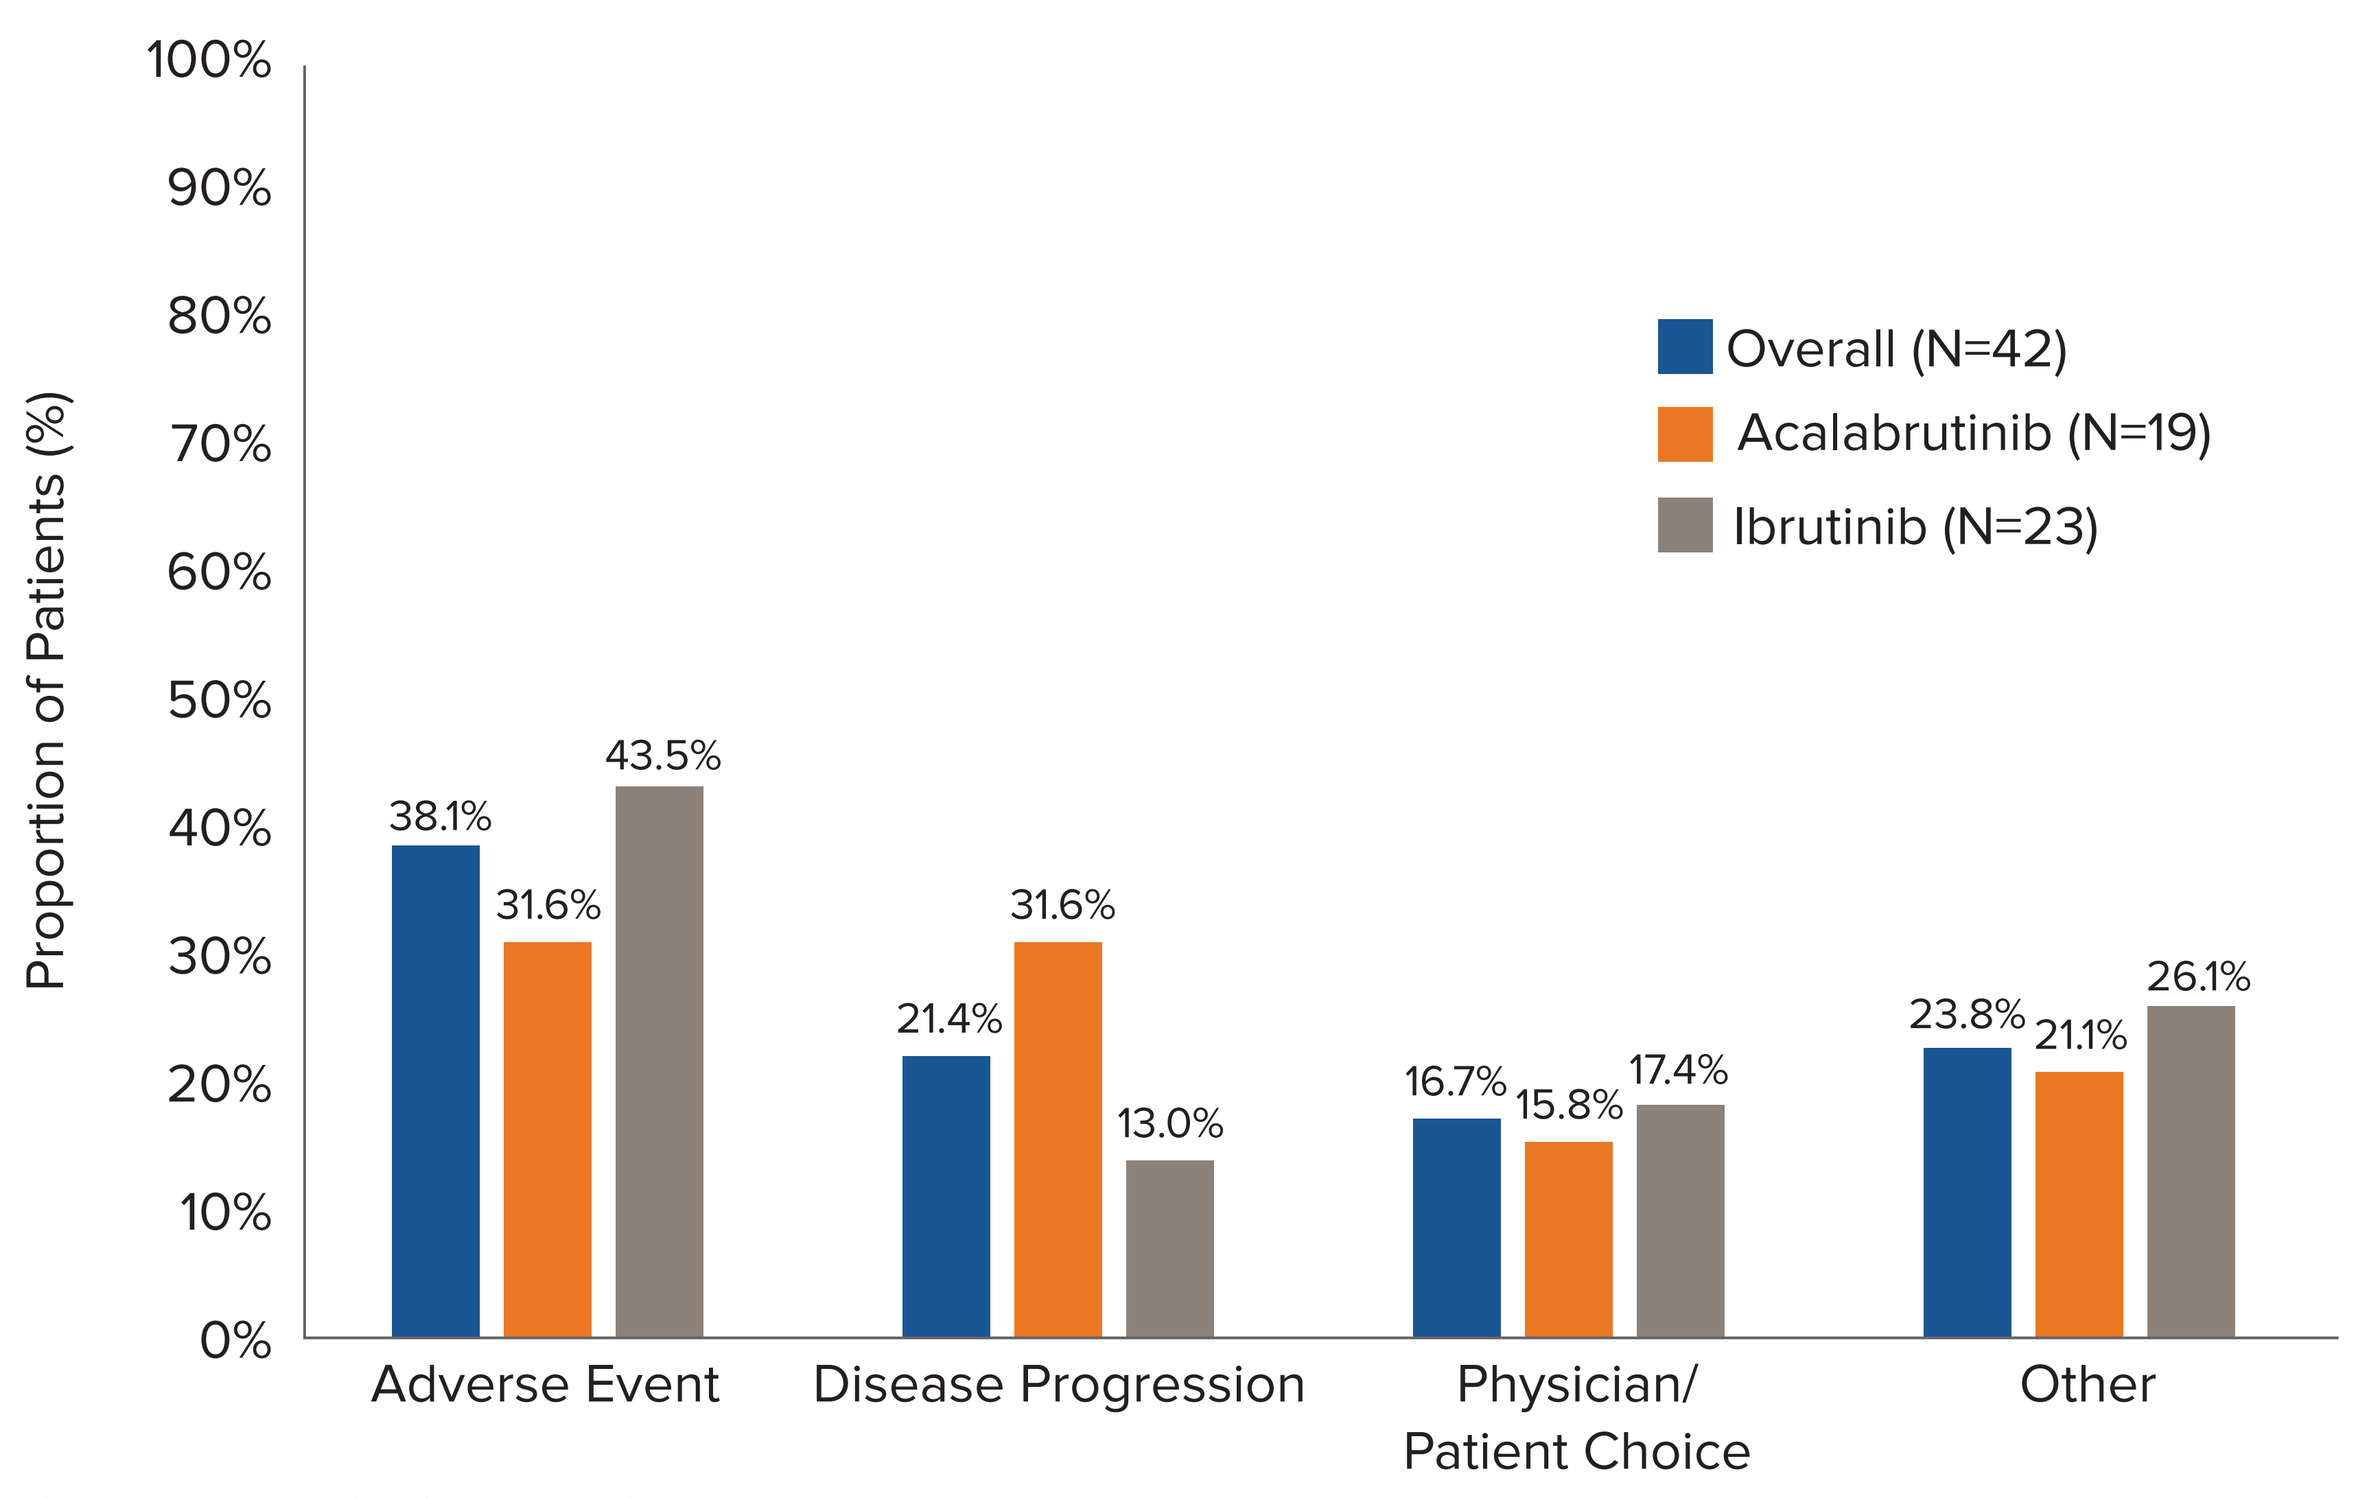

Proportion of Patients (%)
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
38.1%
31.6%
43.5%
21.4%
31.6%
13.0%
16.7%
15.8%
17.4%
23.8%
21.1%
26.1%
Adverse Event
Disease Progression
Physician/ Patient Choice
Other
Overall (N=42)
Acalabrutinib (N=19)
Ibrutinib (N=23)

[a]Among patients who discontinued.

Figure 5. Recurrence of TLAEs on Zanubrutinib

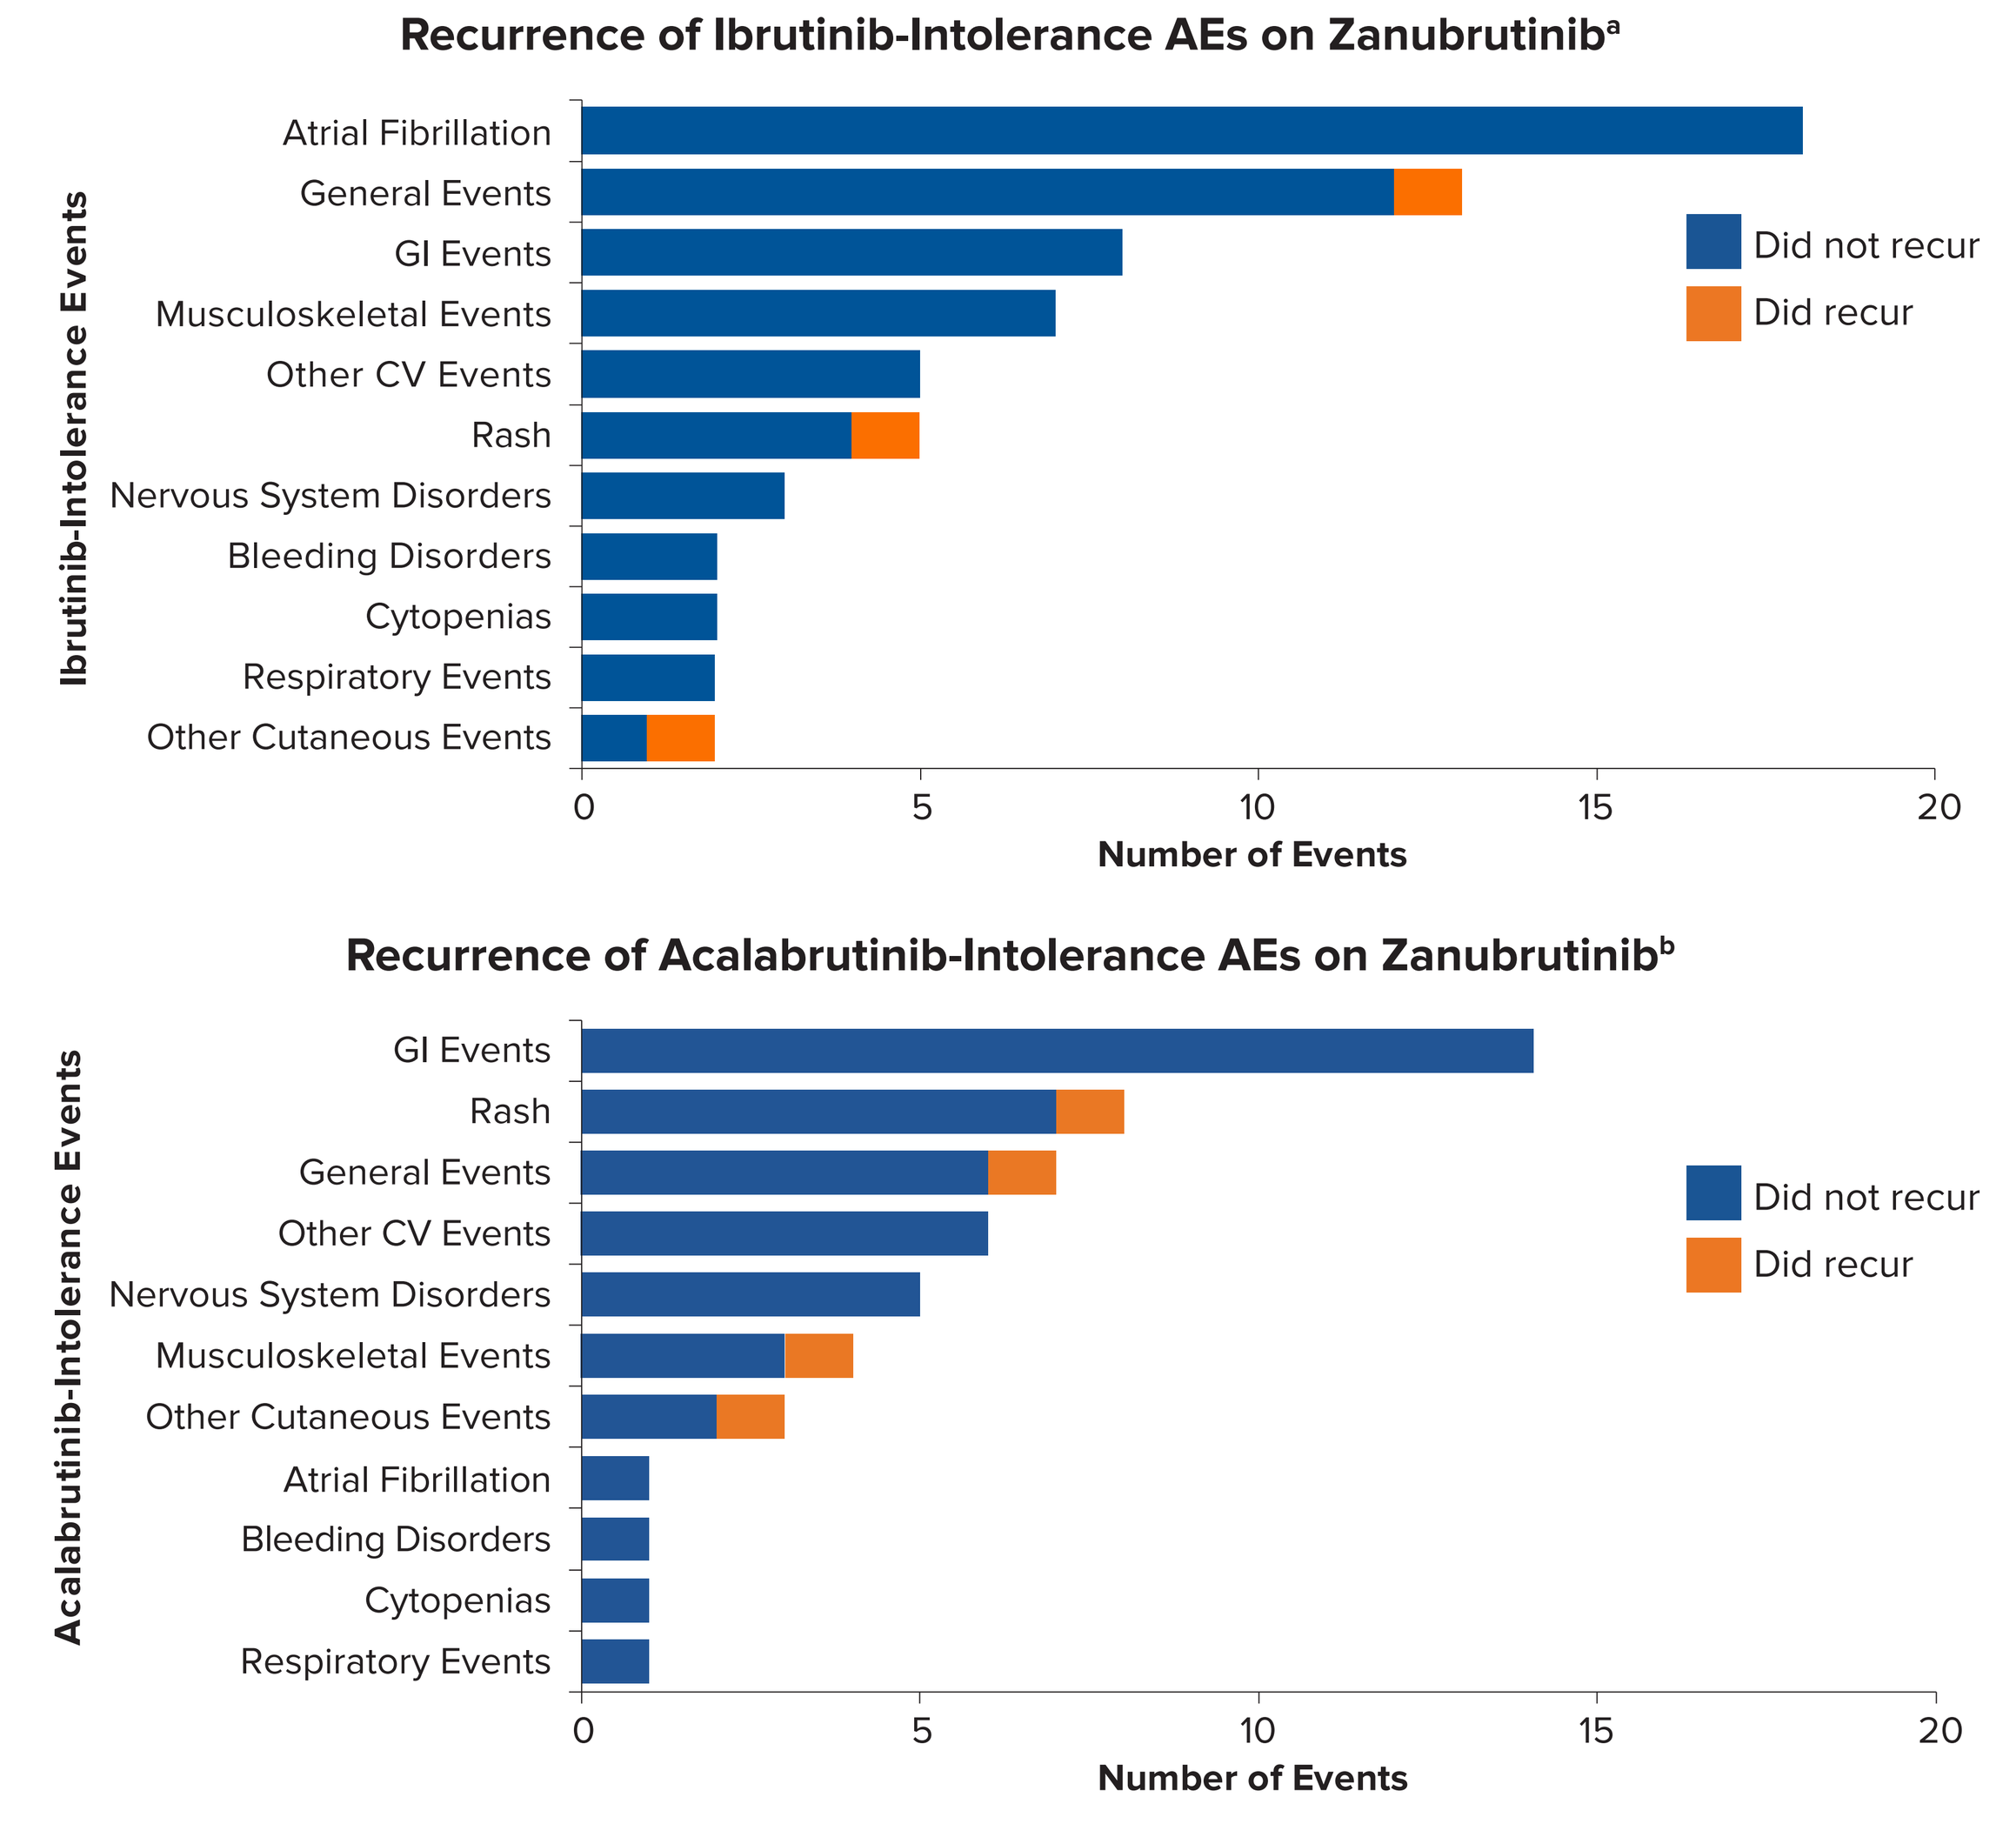

Recurrence of Ibrutinib-Intolerance AEs on Zanubrutinib[a]
Ibrutinib-Intolerance Events
Atrial Fibrillation
General Events
GI Events
Musculoskeletal Events
Other CV Events
Rash
Nervous System Disorders
Bleeding Disorders
Cytopenias
Respiratory Events
Other Cutaneous Events
0
5
10
15
20
Number of Events
Did not recur
Did recur
Recurrence of Acalabrutinib-Intolerance AEs on Zanubrutinib[b]
Acalabrutinib-Intolerance Events
GI Events
Rash
General Events
Other CV Events
Nervous System Disorders
Musculoskeletal Events
Other Cutaneous Events
Atrial Fibrillation
Bleeding Disorders
Cytopenias
Respiratory Events
0
5
10
15
20
Number of Events
Did not recur
Did recur

[a]General events: chills, fatigue, lower extremity edema, poor tolerance, weakness; GI events: abdominal pain, dysphagia, hyponatremia, mucositis, nausea, vomiting; Musculoskeletal events: arthralgias, bradykinesia, leg cramps, muscle spasm, myalgias, unsteady gait; Other CV events: cardiac issues, heart disease, hypertension, palpitations, transient ischemic attack; Cytopenias: cytopenias, neutropenia; Bleeding disorders: bleeding, gastrointestinal bleeding; Nervous system disorders: confusion, dizziness, paranoia; Respiratory events: cough, dyspnea; Other cutaneous events: ecchymosis.
[b]GI events: abdominal pain, diarrhea, gastrointestinal symptoms, nausea, vomiting; Rash: rash, maculopapular rash; General events: bilateral lower extremity edema, fatigue; Other CV events: cardiac arrhythmias, palpitations, syncope; Nervous system disorders: headaches, mental changes, migraines; Musculoskeletal events: arthralgias, myalgias; Other cutaneous events: ecchymosis, pruritus; Bleeding disorders: intramuscular hematoma; Cytopenias: thrombocytopenia; Respiratory events: dyspnea.
AE, adverse event; BTKi, Bruton tyrosine kinase inhibitor; CV, cardiovascular; GI, gastrointestinal; TLAE, treatment-limiting adverse event.

DISCUSSION

- The results from this real-world study provide important insight into the value of zanubrutinib for physicians treating patients with CLL/SLL who are intolerant to ibrutinib or acalabrutinib
- Possible limitations include the retrospective observational design and the single practice setting, thus limiting generalizability

REFERENCES

1. Robak T, et al. *Cancers* (Basel). 2022;14(3):771.
2. National Comprehensive Cancer Network. NCCN Guideline for Chronic Lymphocytic Leukemia/Small Lymphocytic Lymphoma Version 2.2026. https://www.nccn.org/professionals/physician_gls/pdf/cll.pdf. Accessed 1/28/2026.
3. Muñoz J, et al. *Oncologist.* 2023;28(4):309-318. Correction published *Oncologist* 2023 June;28(6):e487.
4. Shadman M, et al. *Lancet Haematol.* 2023;10(1):e35-e45.
5. Shadman M, et al. *Blood Adv.* 2025;9(16):4100-4110.
6. Delgado A, et al. *Am J Cancer Res.* 2021;11(4):1121-1131.

DISCLOSURES

- **AW:** Consulting fees from Cellectar Biosciences Inc. (individual consultant).
- **XW, KC,** and **GAM:** Employment and equity holder in BeOne.
- **AH:** No disclosure to report.
- **CO:** No disclosure to report.
- **JF:** Stock ownership in Amgen, Abbott, and AbbVie; speakers bureau participation for Jazz Pharmaceuticals and AstraZeneca.

ACKNOWLEDGMENTS

- Medical writing support provided by SNELL Medical Communication, Inc. This study was funded by BeOne Medicines, Ltd.

CORRESPONDENCE: Amanda Warner, amanda.warner@flcancer.com

Presented at HOPA 2026; March 25-27, 2026, New Orleans, LA, USA.
Previously presented at ASH 2025; December 6-9, 2025, Orlando, FL, USA.