TISLELIZUMAB IN COMBINATION WITH CHEMOTHERAPY FOR THE TREATMENT OF CHINESE PATIENTS WITH ESOPHAGEAL SQUAMOUS CELL CARCINOMA (ESCC): SAFETY AND TOLERABILTY RESULTS FROM ONE COHORT OF AN ONGOING PHASE 2 STUDY

Nong Xu[1], Xianglin Yuan[2], Buhai Wang[3], Yuxian Bai[4], Enxiao Li[5], Yanyan Li[6], Huanli Wang[6], Jianming Xu[7]

[1]The First Affiliated Hospital of Zhejiang University, Hangzhou, China; [2]Tongji Hospital, Wuhan, China; [3]Northern Jiangsu People's Hospital, Yangzhou, China; [4]Harbin Medical University Cancer Hospital, Harbin, China; [5]First Affiliated Hospital of Xi'an Jiao Tong University, Xi'an, China; [6]BeiGene (Beijing) Co., Ltd., Beijing, China; [7]307 Hospital of the Chinese People's Liberation Army, Beijing, China

BOARD E4: Abstract 14

Gastrointestinal Cancers Symposium of the American Society of Clinical Oncology

January 17–19, 2019, San Francisco, CA

BACKGROUND

- Esophageal cancer is the seventh most common cancer worldwide and the sixth most common cause of cancer-related deaths[1]
- Globally, esophageal squamous cell cancer (ESCC) remains the predominant histological subtype and accounts for most deaths from esophageal cancer; it has a higher prevalence in Asian countries[2]
- Monoclonal antibodies (mAbs) against immune checkpoint inhibitory receptors, such as programmed cell death-1 (PD-1), have demonstrated promising antitumor activity across multiple malignancies,[3] including ESCC[4]
- Tislelizumab (BGB-A317) is an investigational humanized IgG4 mAb with high affinity and binding specificity against PD-1[5]
- Tislelizumab was specifically engineered to minimize FcγR binding on macrophages in order to abrogate antibody-dependent phagocytosis (Figure 1),[5] a potential mechanism of T-cell clearance and potential resistance to anti-PD-1 therapy[6]
- Previous reports from early phase studies suggest tislelizumab was generally well tolerated and had antitumor activity in patients with advanced solid tumors[7–9]

Figure 1: Tislelizumab Mechanism of Action

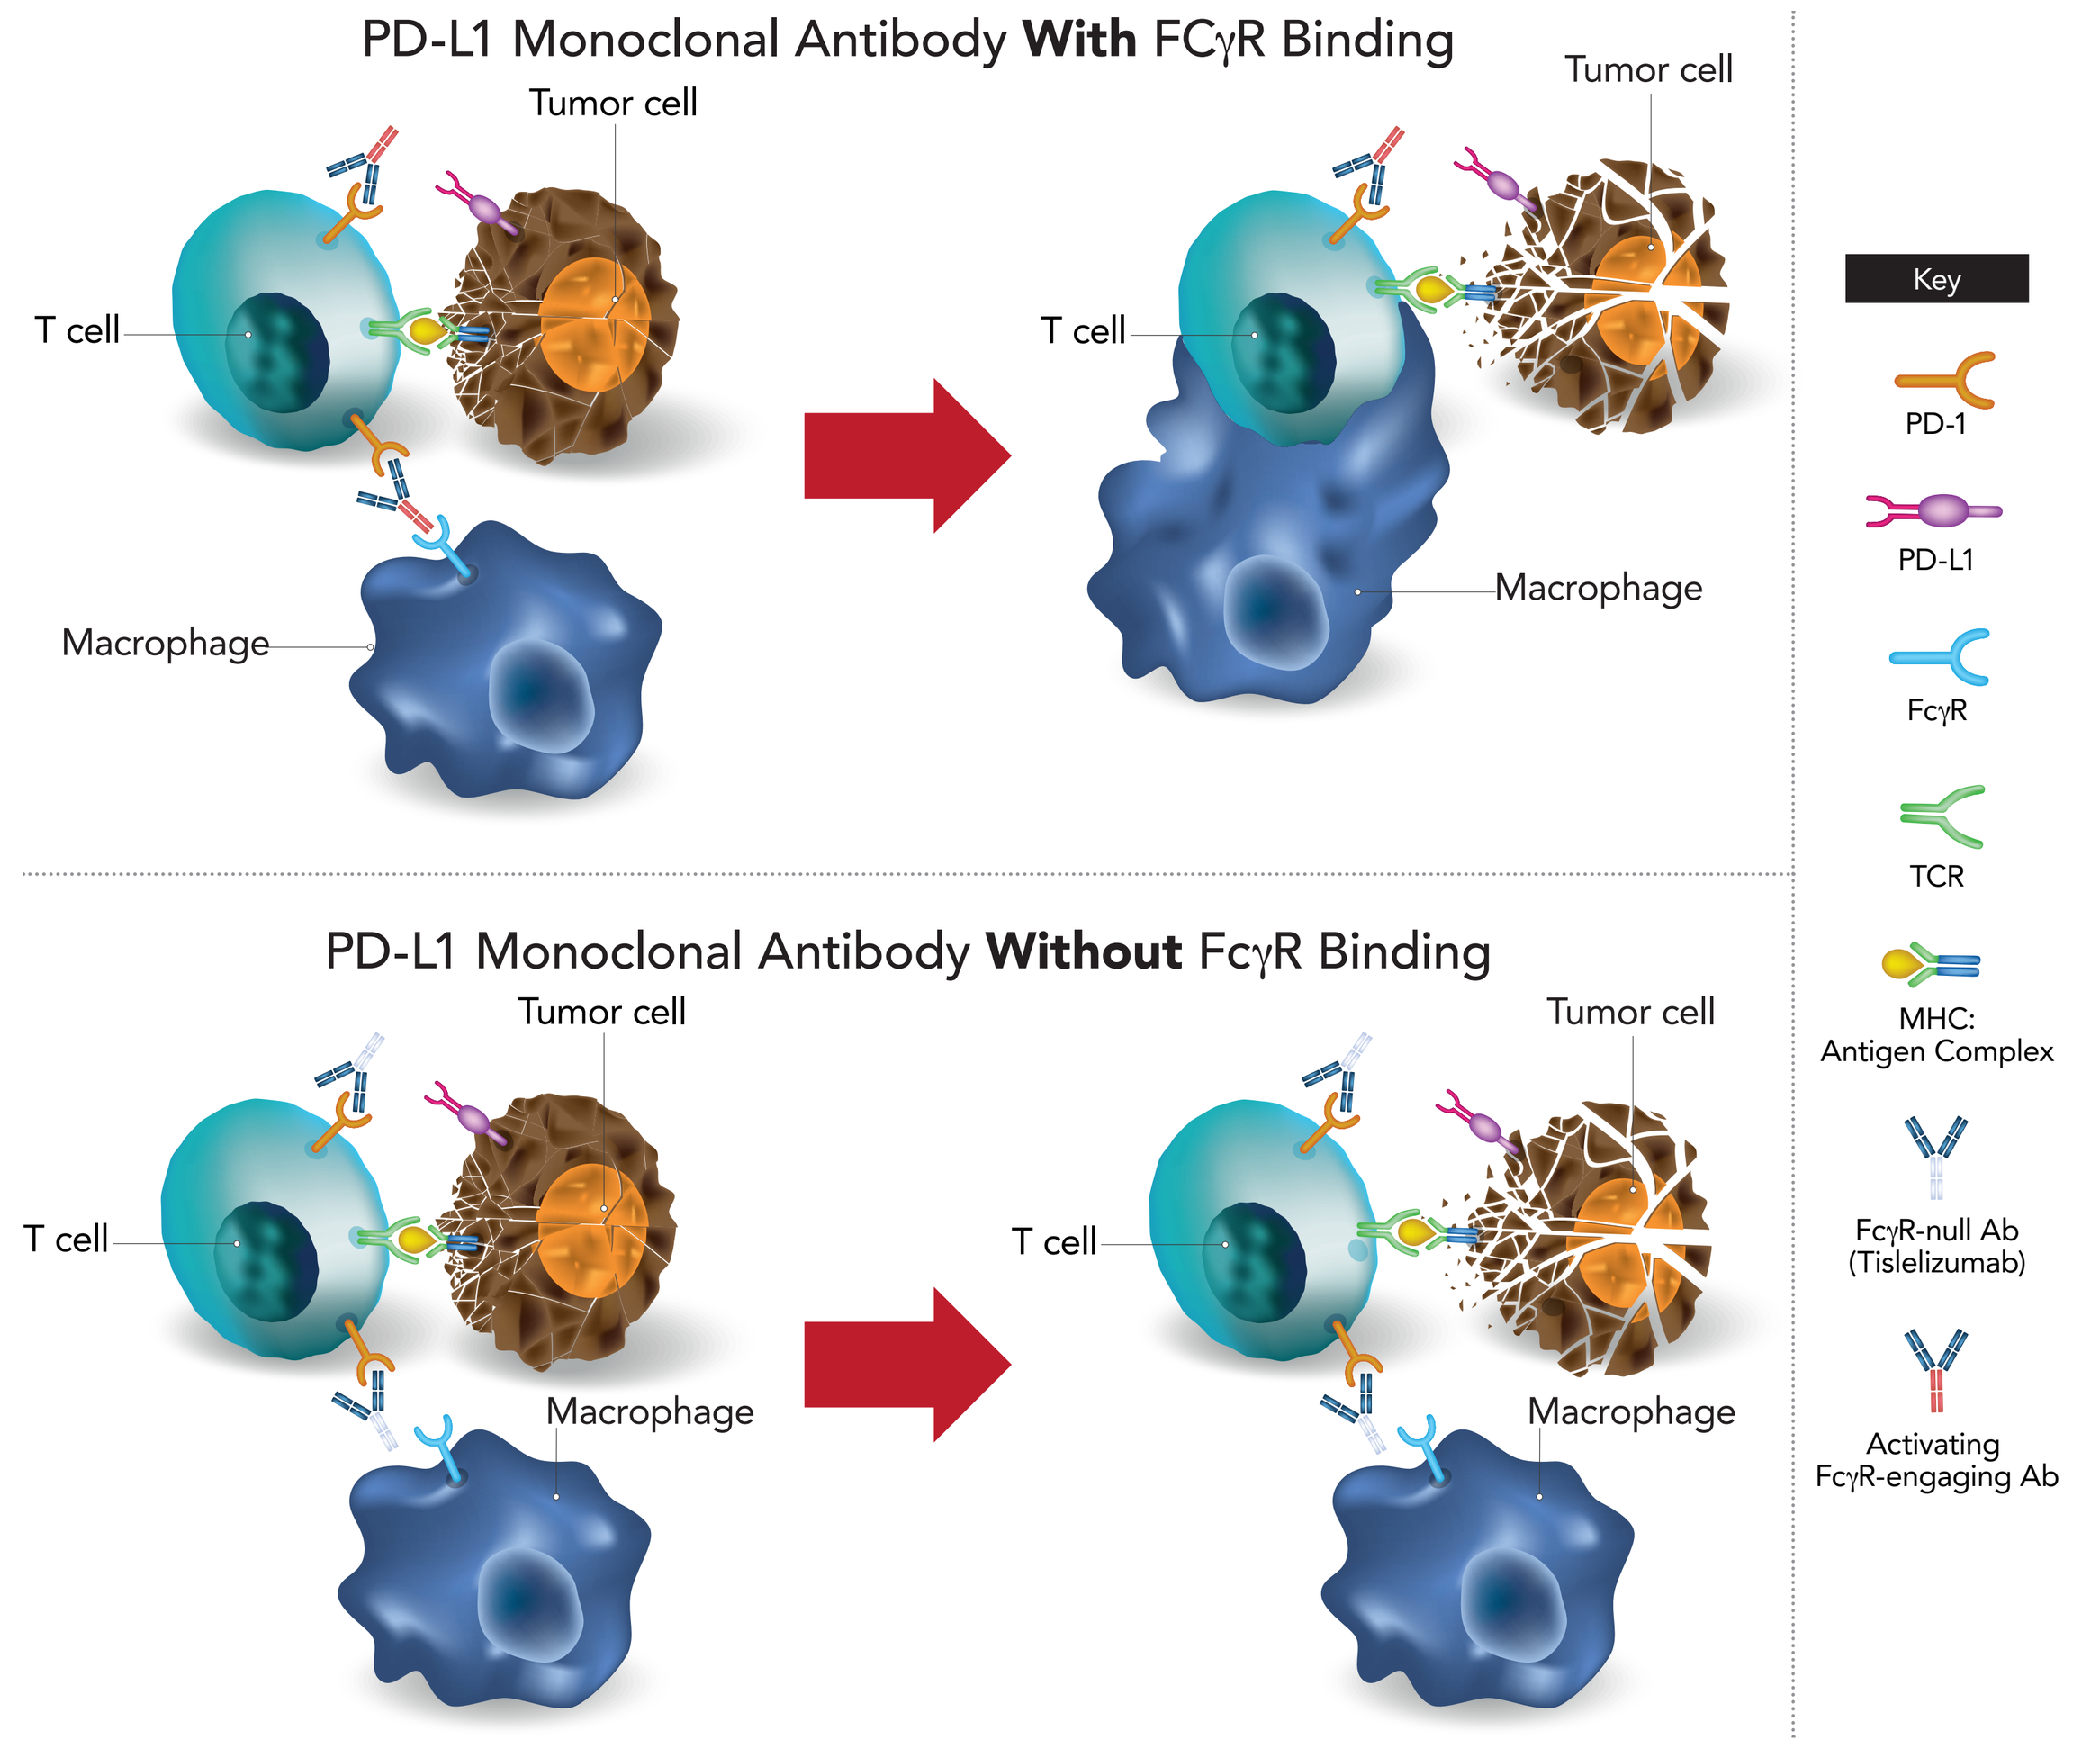
PD-L1 Monoclonal Antibody **With** FCγR Binding
Tumor cell
T cell
Macrophage
Tumor cell
T cell
Macrophage
PD-L1 Monoclonal Antibody **Without** FcγR Binding
Tumor cell
T cell
Macrophage
Tumor cell
T cell
Macrophage
Key
PD-1
PD-L1
FcγR
TCR
MHC: Antigen Complex
FcγR-null Ab (Tislelizumab)
Activating FcγR-engaging Ab

Abbreviations: Ab, antibody; MHC, major histocompatibility complex; PD-1, programmed cell death-1; PD-L1, programmed cell death-1 ligand; TCR, T-cell receptor.

METHODS

- The study design of this two-cohort phase 2 study (NCT03469557) is detailed in Figure 2
- Adult patients with histologically or cytologically confirmed HER2-negative gastric cancer (GC) or gastroesophageal junction (GEJ) cancer or ESCC were eligible for inclusion in the study

Figure 2: Study Design

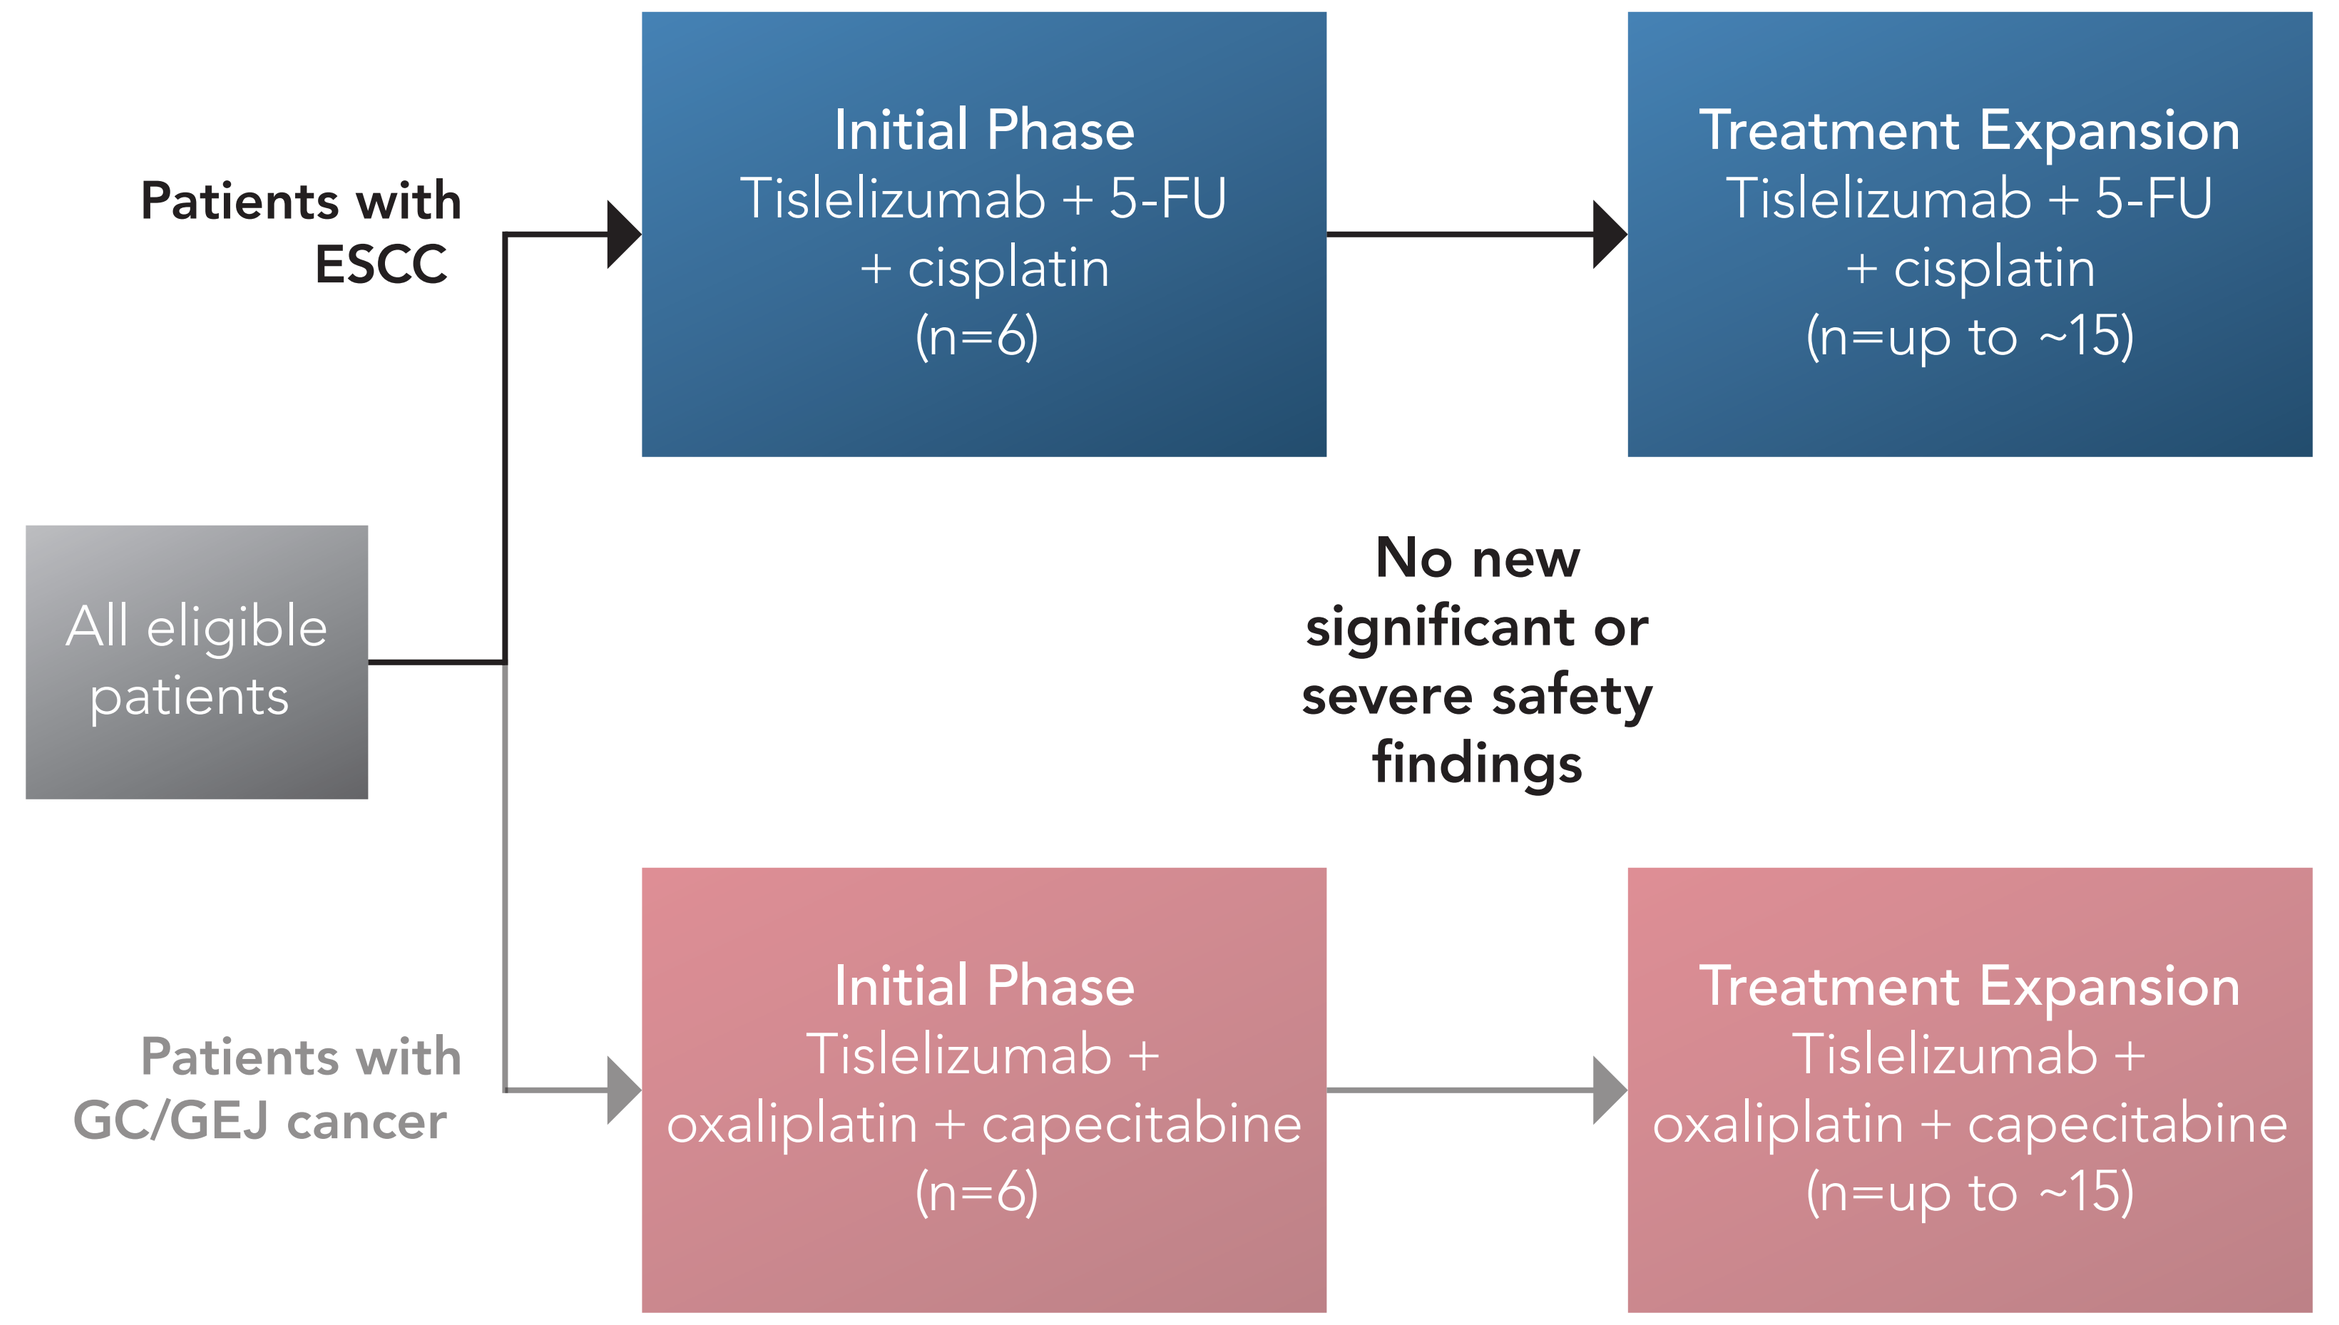
Patients with ESCC
Initial Phase
Tislelizumab + 5-FU + cisplatin
(n=6)
Treatment Expansion
Tislelizumab + 5-FU + cisplatin
(n=up to ~15)
All eligible patients
No new significant or severe safety findings
Patients with GC/GEJ cancer
Initial Phase
Tislelizumab + oxaliplatin + capecitabine
(n=6)
Treatment Expansion
Tislelizumab + oxaliplatin + capecitabine
(n=up to ~15)

Abbreviations: ESCC, esophageal squamous cell carcinoma; 5-FU, 5-fluorouracil; GC, gastric cancer; GEJ, gastroesophageal junction.

 - Patients must have ≥1 measurable or evaluable lesion considered inoperable, locally advanced, or metastatic at the time of enrollment; must not be amenable to radiation therapy alone; and must not have received prior systemic therapy for advanced or metastatic disease (prior neoadjuvant or adjuvant therapy including chemoradiation was allowed, provided it was completed ≥6 months prior to enrollment)
 - Patients were excluded if they had a history of severe hypersensitivity reactions to other mAbs or platinum agents such as fluorouracil or cisplatin
 - Patients were excluded if they had previously received PD-1, PD-L1, or PD-L2 antibody therapy for any indication
 - Patients who had a prior malignancy that was active within the previous 2 years, except for ESCC, and who had locally recurring cancers that had undergone curative intent treatment, such as basal or squamous cell skin cancer, superficial bladder cancer, or carcinoma in situ of the cervix or breast, were excluded
- Patients with ESCC were treated with tislelizumab (200 mg IV Q3W), cisplatin (80 mg/m² IV Q3W), and fluorouracil (800 mg/m²/d, Days 1–5 IV Q3W)
 - Cisplatin and 5-FU were administered for ≤6 cycles; tislelizumab will be administered until disease progression, intolerable toxicity, or treatment discontinuation
- Safety and tolerability were assessed through monitoring of adverse events (AEs), defined and graded according to CTCAE v4.03; tumor responses were assessed by investigators based on RECIST v1.1 criteria every 9 weeks during the first year, and every 12 weeks thereafter
- Here, we present the safety and tolerability profile of tislelizumab in combination with chemotherapy in the cohort of patients with ESCC
 - Assessment of the antitumor activity of tislelizumab in the ESCC cohort is ongoing
 - Safety, tolerability, and antitumor activity data for the cohort of patients with GC is presented at BOARD E1: Abstract 11

RESULTS

Patient Disposition

- As of 13 June 2018, 15 patients with ESCC had enrolled in this study
- Most patients were male (93%), had an Eastern Cooperative Oncology Group (ECOG) status of 1 (73%), and stage IV disease (53%) (Table 1)
- The median treatment duration was 108 days (range: 21–201)
 - Mean relative dose intensity was 0.92 for tislelizumab, 0.91 for cisplatin, and 0.78 for 5-FU

Table 1: Demographics and Baseline Disease Characteristics of Patients With ESCC

		ESCC Population (N=15)
Median age, years		61
Sex, n (%)	Male	14 (93)
	Female	1 (7)
ECOG status, n (%)	0	4 (27)
	1	11 (73)
Tumor stage, n (%)	0–II	0
	III	6 (40)
	IV	8 (53)
	Not applicable[a]	1 (7)
Prior systemic anticancer therapy regimens, n (%)	Neoadjuvant chemotherapy	1 (7)
	Adjuvant chemotherapy	2 (13)
	Targeted therapy	0
	Concurrent radiochemotherapy	0
	Radiation therapy	1 (7)
Prior surgery related to current cancer, n (%)		4 (27)

[a]One patient was staged as TxN1Mo.
Abbreviations: ECOG, Eastern Cooperative Oncology Group; ESCC, esophageal squamous cell carcinoma.

Safety and Tolerability

- Treatment with tislelizumab, in combination with chemotherapy, was generally well tolerated in patients with ESCC
- A summary of the safety and tolerability profile of tislelizumab in combination with chemotherapy is provided in Table 2
 - Grade ≥3 chemotherapy-related AEs were reported in seven patients; one patient reported grade ≥3 tislelizumab-related AEs
- A total of four patients discontinued study treatment due to AEs
 - Three patients discontinued due to chemotherapy or tislelizumab-related AEs (grade 2 pneumonitis, grade 3 increase in aspartate aminotransferase, grade 3 lung infection)
 - One patient discontinued due to an AE unrelated to study treatment (grade 3 tracheal fistula)

Table 2: Summary of Adverse Events Reported With Tislelizumab Combined With Chemotherapy in Patients With ESCC

	ESCC Population (N=15)
Patient reporting at least ≥1 treatment-related AE	15 (100)
Grade ≥3 treatment-related AE	7 (47)
Serious AE	5 (33)
Treatment-related serious AE	4 (27)
AE leading to treatment discontinuation	4 (27)
AE leading to death[a]	1 (7)
Treatment-related AE leading to death[a]	1 (7)

[a]One patient experienced grade 5 abnormal hepatic function considered mainly related to progressive disease, and possibly related to chemotherapy, tislelizumab, or underlying type B hepatitis.
Abbreviations: AE, adverse event; ESCC, esophageal squamous cell cancer.

Table 3: Adverse Events Considered Related to Treatment Occurring in >2 Patients With ESCC

	ESCC Population (N=15)					
	Chemotherapy-related		Tislelizumab-related		Any treatment-related[a]	
	All grades	Grades ≥3	All grades	Grades ≥3	All grades	Grades ≥3
Decreased appetite	9 (60)	1 (7)	2 (13)	0	9 (60)	1 (7)
Nausea	8 (53)	0	0	0	8 (53)	0
Anemia	6 (40)	1 (7)	0	0	6 (40)	1 (7)
Decreased white blood cell count	6 (40)	1 (7)	0	0	6 (40)	1 (7)
Decreased neutrophil count	5 (33)	1 (7)	0	0	5 (33)	1 (7)
Vomiting	5 (33)	3 (20)	0	0	5 (33)	3 (20)
Asthenia	4 (27)	1 (7)	2 (13)	0	4 (27)	1 (7)
Hypoalbuminemia	4 (27)	0	0	0	4 (27)	0
Thrombocytopenia	4 (27)	1 (7)	0	0	4 (27)	1 (7)

Data presented as n (%). [a]Adverse events could be considered related to more than one treatment.
Abbreviation: ESCC, esophageal squamous cell cancer.

- Adverse events related to chemotherapy occurred in 14 of the 15 patients with ESCC; AEs considered related to tislelizumab occurred in eight patients
- Adverse events reported in this cohort (Table 3) were consistent with the known tolerability profile of PD-1 inhibitors in combination with chemotherapy
 - Most of the reported AEs were considered related to chemotherapy and were mild-to-moderate in severity
- A total of five patients experienced ≥1 serious AE
 - Tracheal fistula was not considered related to treatment
 - Abnormal hepatic function was considered related to both chemotherapy and tislelizumab
 - Pneumonitis was considered at least possibly related to tislelizumab
 - All other serious AEs were considered related to chemotherapy
- One patient experienced grade 5 abnormal hepatic function, which was considered mainly related to progressive disease
 - The event was considered also possibly related to study treatment or underlying type B hepatitis

CONCLUSIONS

- First-line tislelizumab plus chemotherapy was generally a well-tolerated therapy in patients with ESCC
- Reported AEs were consistent with the known tolerability profile of PD-1 inhibitors in combination with chemotherapy
 - Most AEs were mild or moderate in severity
 - All four AEs leading to study discontinuation were manageable; two were considered related to tislelizumab
- The preliminary safety and tolerability profile from this study support continued development of tislelizumab in patients with ESCC
 - A global, randomized, phase 3 study of tislelizumab compared with chemotherapy as second-line treatment of advanced unresectable/metastatic ESCC is currently enrolling
 - A global, randomized, phase 3 study of tislelizumab in combination with chemotherapy as first-line treatment in patients with unresectable, locally advanced recurrent or metastatic ESCC is currently enrolling

REFERENCES

1. Bray F, Ferlay J, Soerjomataram I, Siegel RL, Torre LA, Jemal A. Global cancer statistics 2018: GLOBOCAN estimates of incidence and mortality worldwide for 36 cancers in 185 countries. *CA Cancer J Clin.* 2018;68(6):394–424.
2. Abbas G, Krasna M. Overview of esophageal cancer. *Ann Cardiothorac Surg.* 2017;6(2):131–136.
3. Topalian SL, Hodi FS, Brahmer JR, et al. Safety, activity, and immune correlates of anti-PD-1 antibody in cancer. *N Engl J Med.* 2012;366(26):2443–2454.
4. Doi T, Piha-Paul SA, Jalal SI, et al. Pembrolizumab (MK-3475) for patients (pts) with advanced esophageal carcinoma: Preliminary results from KEYNOTE-028. *J Clin Oncol.* 2015;33(1suppl 15):4010–4010.
5. Zhang T, Song X, Xu L, et al. The binding of an anti-PD-1 antibody to FcgammaRIota has a profound impact on its biological functions. *Cancer Immunol Immunother.* 2018;67(7):1079–1090.
6. Dahan R, Sega E, Engelhardt J, Selby M, Korman AJ, Ravetch JV. FcgammaRs modulate the anti-tumor activity of antibodies targeting the PD-1/PD-L1 axis. *Cancer Cell.* 2015;28(4):543.
7. Desai J, Millward M, Chao Y, et al. Preliminary results from subsets of patients (pts) with advanced gastric cancer (GC) and esophageal carcinoma (EC) in a dose-escalation/expansion study of BGB-A317, an anti-PD-1 monoclonal antibody (mAb). *Ann Oncol.* 2017;28(suppl 5):v122–v141.
8. Wu Y-L, Zhao J, Zhang Q, et al. Preliminary results with tislelizumab in Chinese patients with non-small cell lung cancer. Presented at: IASLC 19th World Conference on Lung Cancer; September 23–26, 2018; Toronto, Canada.
9. Zhao J, Wang Z, Ma Z, et al. Tislelizumab combined with chemotherapy as first-line treatment in chinese patients with advanced lung cancer. Presented at: IASLC 19th World Conference on Lung Cancer; September 23–26, 2018; Toronto, Canada.

ACKNOWLEDGMENTS

The authors wish to acknowledge the investigative center study staff and study patients, as well as recognize those from BeiGene who have substantially contributed to the development of this presentation. BeiGene, Ltd. provided financial support for this presentation, including writing and editorial assistance by Regina Switzer, PhD, of SuccinctChoice Medical Communications (Chicago, IL).

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Please address any questions or comments regarding this poster to Clinicaltrials@beigene.com