Real-World Treatment Utilization Patterns, Discontinuation and Healthcare Resource Utilization of First-Line Bruton Tyrosine Kinase Inhibitor Therapy in Chronic Lymphocytic Leukemia: Age-Related Disparity

PF585

Keri Yang,[1] Swetha Challagulla,[1] Po-Ya Chuang,[2] Wesley Furnback,[2] Sikander Ailawadhi[3]

[1]BeOne Medicines Ltd., USA, San Carlos, CA, USA; [2]Real Chemistry, New York, NY, USA; [3]Mayo Clinic, Jacksonville, FL, USA

CONCLUSIONS

- This real-world study demonstrated that patients with chronic lymphocytic leukemia (CLL) treated with zanubrutinib had longer time to discontinuation (TTD), lower discontinuation rates, and less healthcare resource utilization (HCRU) than those treated with acalabrutinib and ibrutinib across all patients and specifically in older patients ≥65 years
- Further studies on long-term and clinical outcomes are warranted

INTRODUCTION

- Chronic lymphocytic leukemia (CLL) tends to affect older individuals, and these patients often endure greater health disparities and treatment burdens than younger patients that may impact their access to healthcare resources, adherence to treatment regimens, and treatment outcomes[1]

OBJECTIVE

- This study aimed to assess treatment utilization patterns, discontinuation, and healthcare resource utilization (HCRU) of first line (1L) use of Bruton tyrosine kinase inhibitors (BTKis) in CLL, focusing on older patients ≥65 years

METHODS

- A retrospective observational study was conducted using the Symphony Integrated Dataverse (IDV®), a comprehensive, longitudinal, open-claims database

Inclusion Criteria

- Adult patients (≥18 years) diagnosed with CLL initiating a first-line (1L) BTKi treatment regimen (zanubrutinib, acalabrutinib, or ibrutinib) during the index period (Feb 2022 through Sep 2024)
- Continuous enrollment in the database for 30 days pre- and post-index date, defined as the date of BTKi initiation

Study Groups

- Patients with CLL initiating a BTKi were further categorized by age at index date, and examined by overall CLL patient group and by older patients ≥65 years group
- Three treatment groups were identifed by BTKi at index date (zanubrutinib, acalabrutinib, or ibrutinib) for the overall and ≥65 years groups

Study Measures

- Patient sociodemographic, clinical characteristics, and outcomes were examined in overall and older (≥65 years) patients with CLL by each BTKi group
- Treatment discontinuation rate for the index BTKi at 60, 90, 120, 150, 180, and 360 days and the median time to discontinuation (TTD) were estimated using the Kaplan-Meier method, censoring patients at the end of follow-up
- HCRU (outpatient, inpatient, and other services) was evaluated during time on treatment and over the follow-up period and reported as per-patient-per-year (PPPY)

RESULTS

Study Population

- A total of 6846 patients initiated a 1L BTKi for CLL during the index period and 5022 patients (73.4%) were ≥65 years old (**Table 1**)
- Patients with CLL receiving zanubrutinib were older (mean age: 70.9 years; 78.0% ≥65 years) than those receiving acalabrutinib (69.4 years; 71.0% ≥65 years) and ibrutinib (69.6 years; 73.3% ≥65 years) (P<.05)
- Payer type and geography were significantly different across the BTKi groups in both the overall and ≥65 years groups (P<.001)
- There were no significant differences across the BTKis regarding baseline comorbidities in the overall group and ≥65 years group

Time to Discontinuation (TTD)

- Patients receiving zanubrutinib had the longest estimated median TTD (zanubrutinib: 18.9 months, acalabrutinib: 17.8 months, ibrutinib: 14.5 months; P<.001) (**Figure 1A**)
- In older patients ≥65 years, zanubrutinib also had the longest median TTD (17.2 months), compared to 15.2 months for acalabrutinib and 12.6 months for ibrutinib (P<.001) (**Figure 1B**)
- The median follow-up time across all ages/≥65 years was longest for ibrutinib (21.4/20.8 months), followed by acalabrutinib (16.8/16.3 months) and zanubrutinib (10.1/9.6 months)

Treatment Discontinuation Rates

- Zanubrutinib had the lowest estimated discontinuation rates at 60- , 90-, 120-, 150-, 180-, and 360-days post-BTKi initiation in the overall (**Figure 2A**) and ≥65 years (**Figure 2B**) groups

Table 1. Baseline Characteristics

	All Patients (n=6846)			≥65 Years (n=5022)		
	Zanubrutinib (n=1853)	Acalabrutinib (n=3519)	Ibrutinib (n=1474)	Zanubrutinib (n=1445)	Acalabrutinib (n=2497)	Ibrutinib (n=1080)
Female, n (%)	696 (37.6%)	1286 (36.5%)	555 (37.7%)	563 (39.0%)	926 (37.1%)	430 (39.8%)
Age*						
Mean (SD)	70.9 (8.6)	69.4 (9.1)	69.6 (8.8)	76.6 (4.8)	74.3 (4.9)	74.0 (4.7)
Median (IQR)	73 (66, 79)	71 (63, 78)	71 (64, 78)	76 (71, 79)	76 (70, 78)	75 (70, 78)
Payer type, n (%)*						
Medicaid	73 (3.9%)	175 (5.0%)	105 (7.1%)	23 (1.6%)	35 (1.4%)	26 (2.4%)
Medicare	1069 (57.7%)	1940 (55.1%)	856 (58.1%)	1021 (70.7%)	1861 (74.5%)	804 (74.4%)
Commercial	633 (34.2%)	1257 (35.7%)	431 (29.2%)	334 (23.1%)	486 (19.5%)	180 (16.7%)
Other	11 (0.6%)	24 (0.7%)	49 (3.3%)	11 (0.8%)	11 (0.4%)	40 (3.7%)
Unknown	67 (3.6%)	123 (3.5%)	33 (2.2%)	56 (3.9%)	104 (4.2%)	30 (2.8%)
Geography, n (%)*						
Midwest	402 (21.7%)	867 (24.6%)	359 (24.4%)	298 (20.6%)	610 (24.4%)	272 (25.2%)
Northeast	358 (19.3%)	582 (16.5%)	194 (13.2%)	276 (19.1%)	422 (16.9%)	148 (13.7%)
South	708 (38.2%)	1304 (37.1%)	558 (37.9%)	555 (38.4%)	896 (35.9%)	405 (37.5%)
West	358 (19.3%)	718 (20.4%)	321 (21.8%)	294 (20.3%)	532 (21.3%)	225 (20.8%)
Other/missing	27 (1.5%)	48 (1.4%)	42 (2.8%)	22 (1.5%)	37 (1.5%)	30 (2.8%)
CCI, mean (SD)	2.3 (3.2)	2.2 (3.1)	2.0 (3.2)	2.3 (3.2)	2.2 (3.1)	2.1 (3.2)

*P<.05
CCI, Charlson Comorbidity Index; IQR, interquartile range; SD, standard deviation.

Figure 1. TTD

A. Overall CLL Patient Group

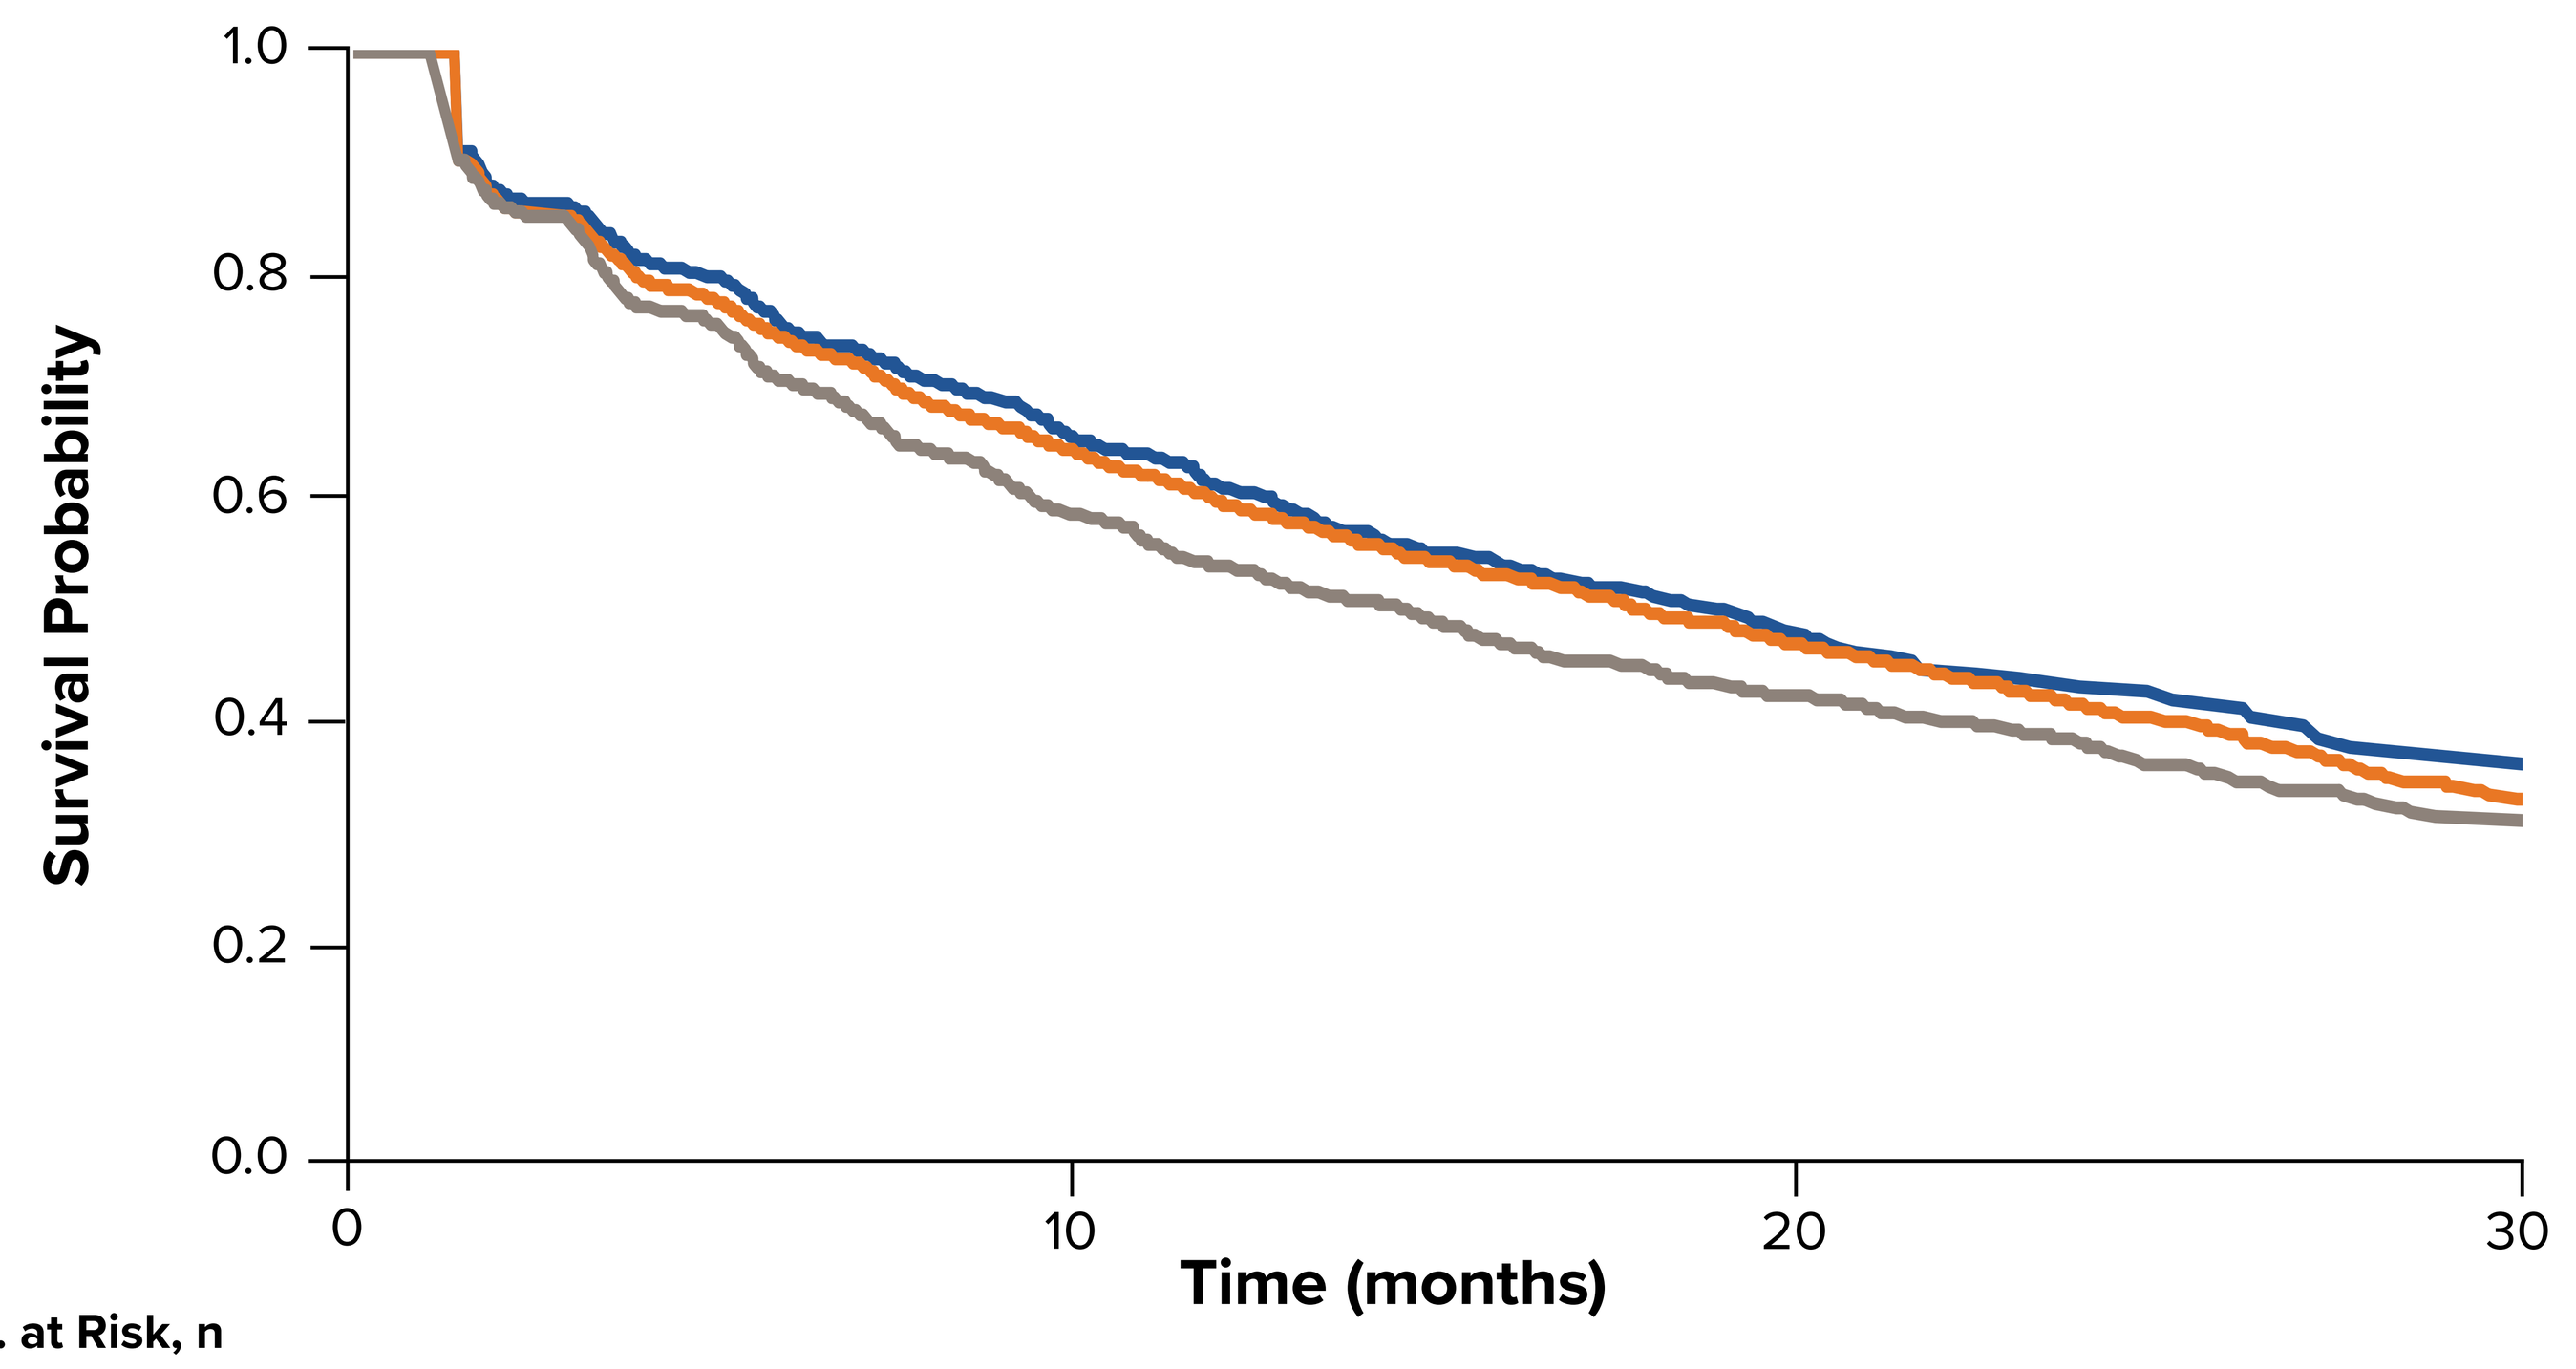

Survival Probability
1.0
0.8
0.6
0.4
0.2
0.0
0
10
20
30
Time (months)

No. at Risk, n				
Zanubrutinib	1853	712	155	23
Acalabrutinib	3519	1772	859	172
Ibrutinib	1474	751	439	107

B. ≥65 Years Group

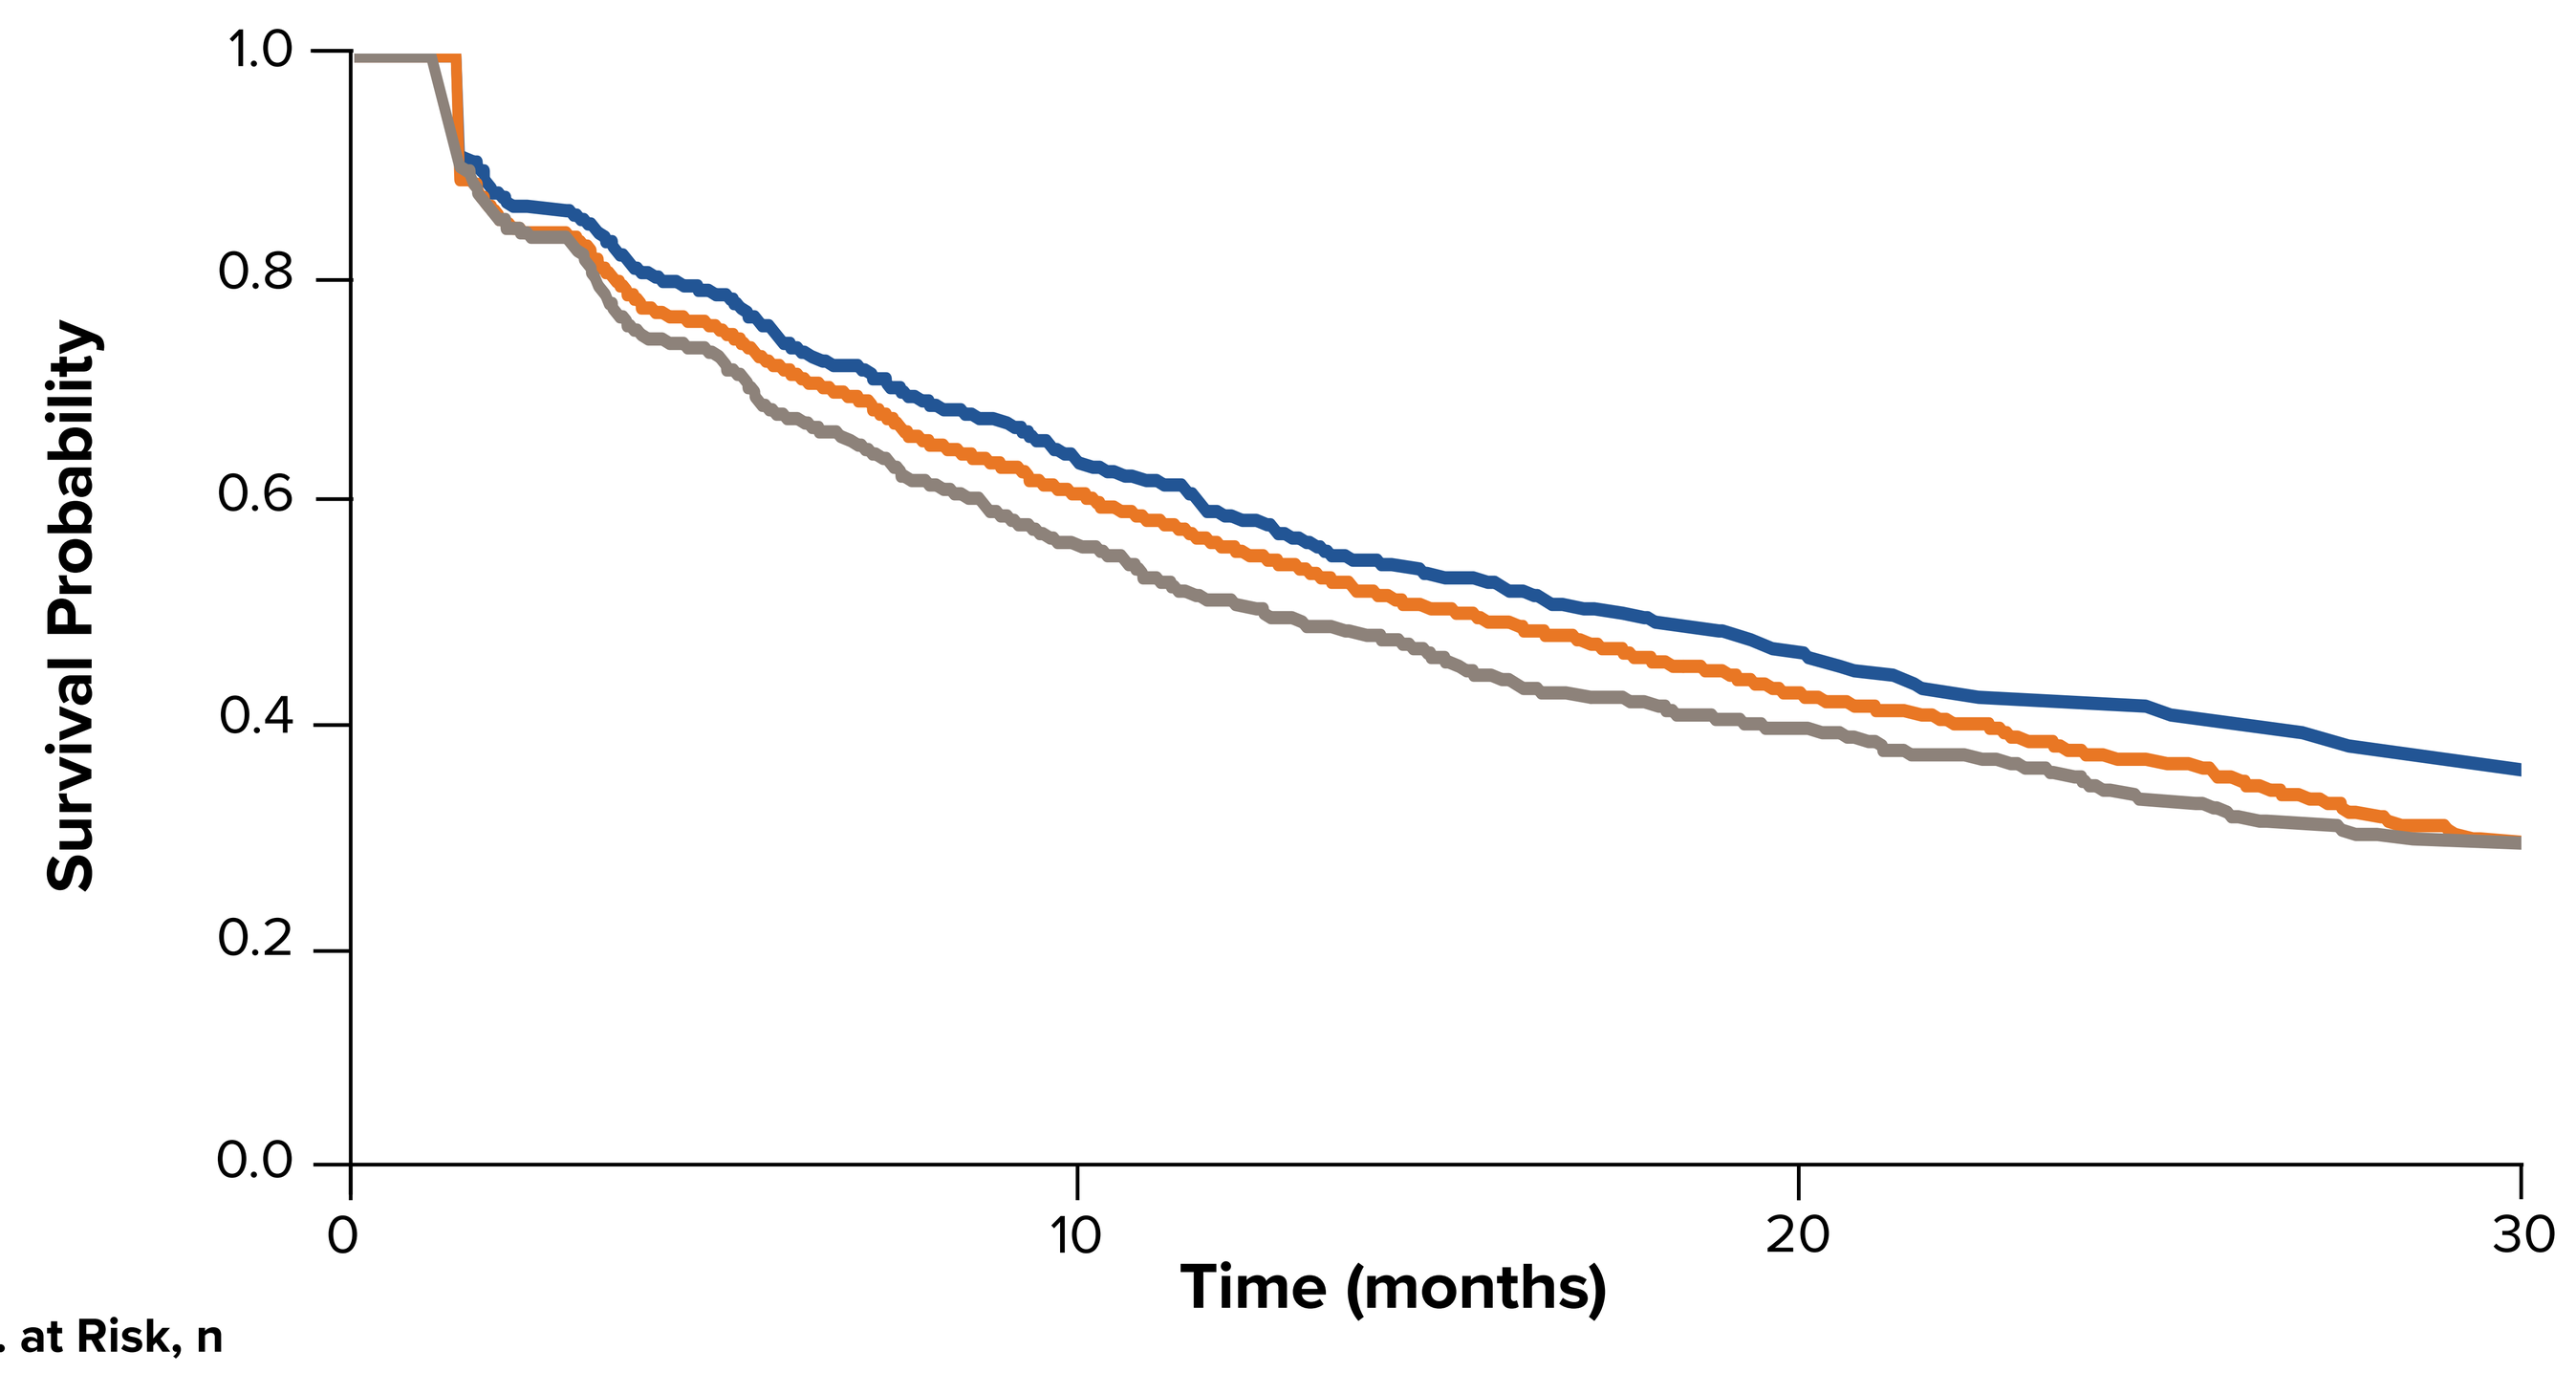

Survival Probability
1.0
0.8
0.6
0.4
0.2
0.0
0
10
20
30
Time (months)

No. at Risk, n				
Zanubrutinib	1445	523	111	16
Acalabrutinib	2497	1777	533	101
Ibrutinib	1080	525	290	60

CLL, chronic lymphocytic leukemia; TTD, time to discontinuation.

Figure 2. Treatment Discontinuation Rates

A. Overall CLL Patient Group

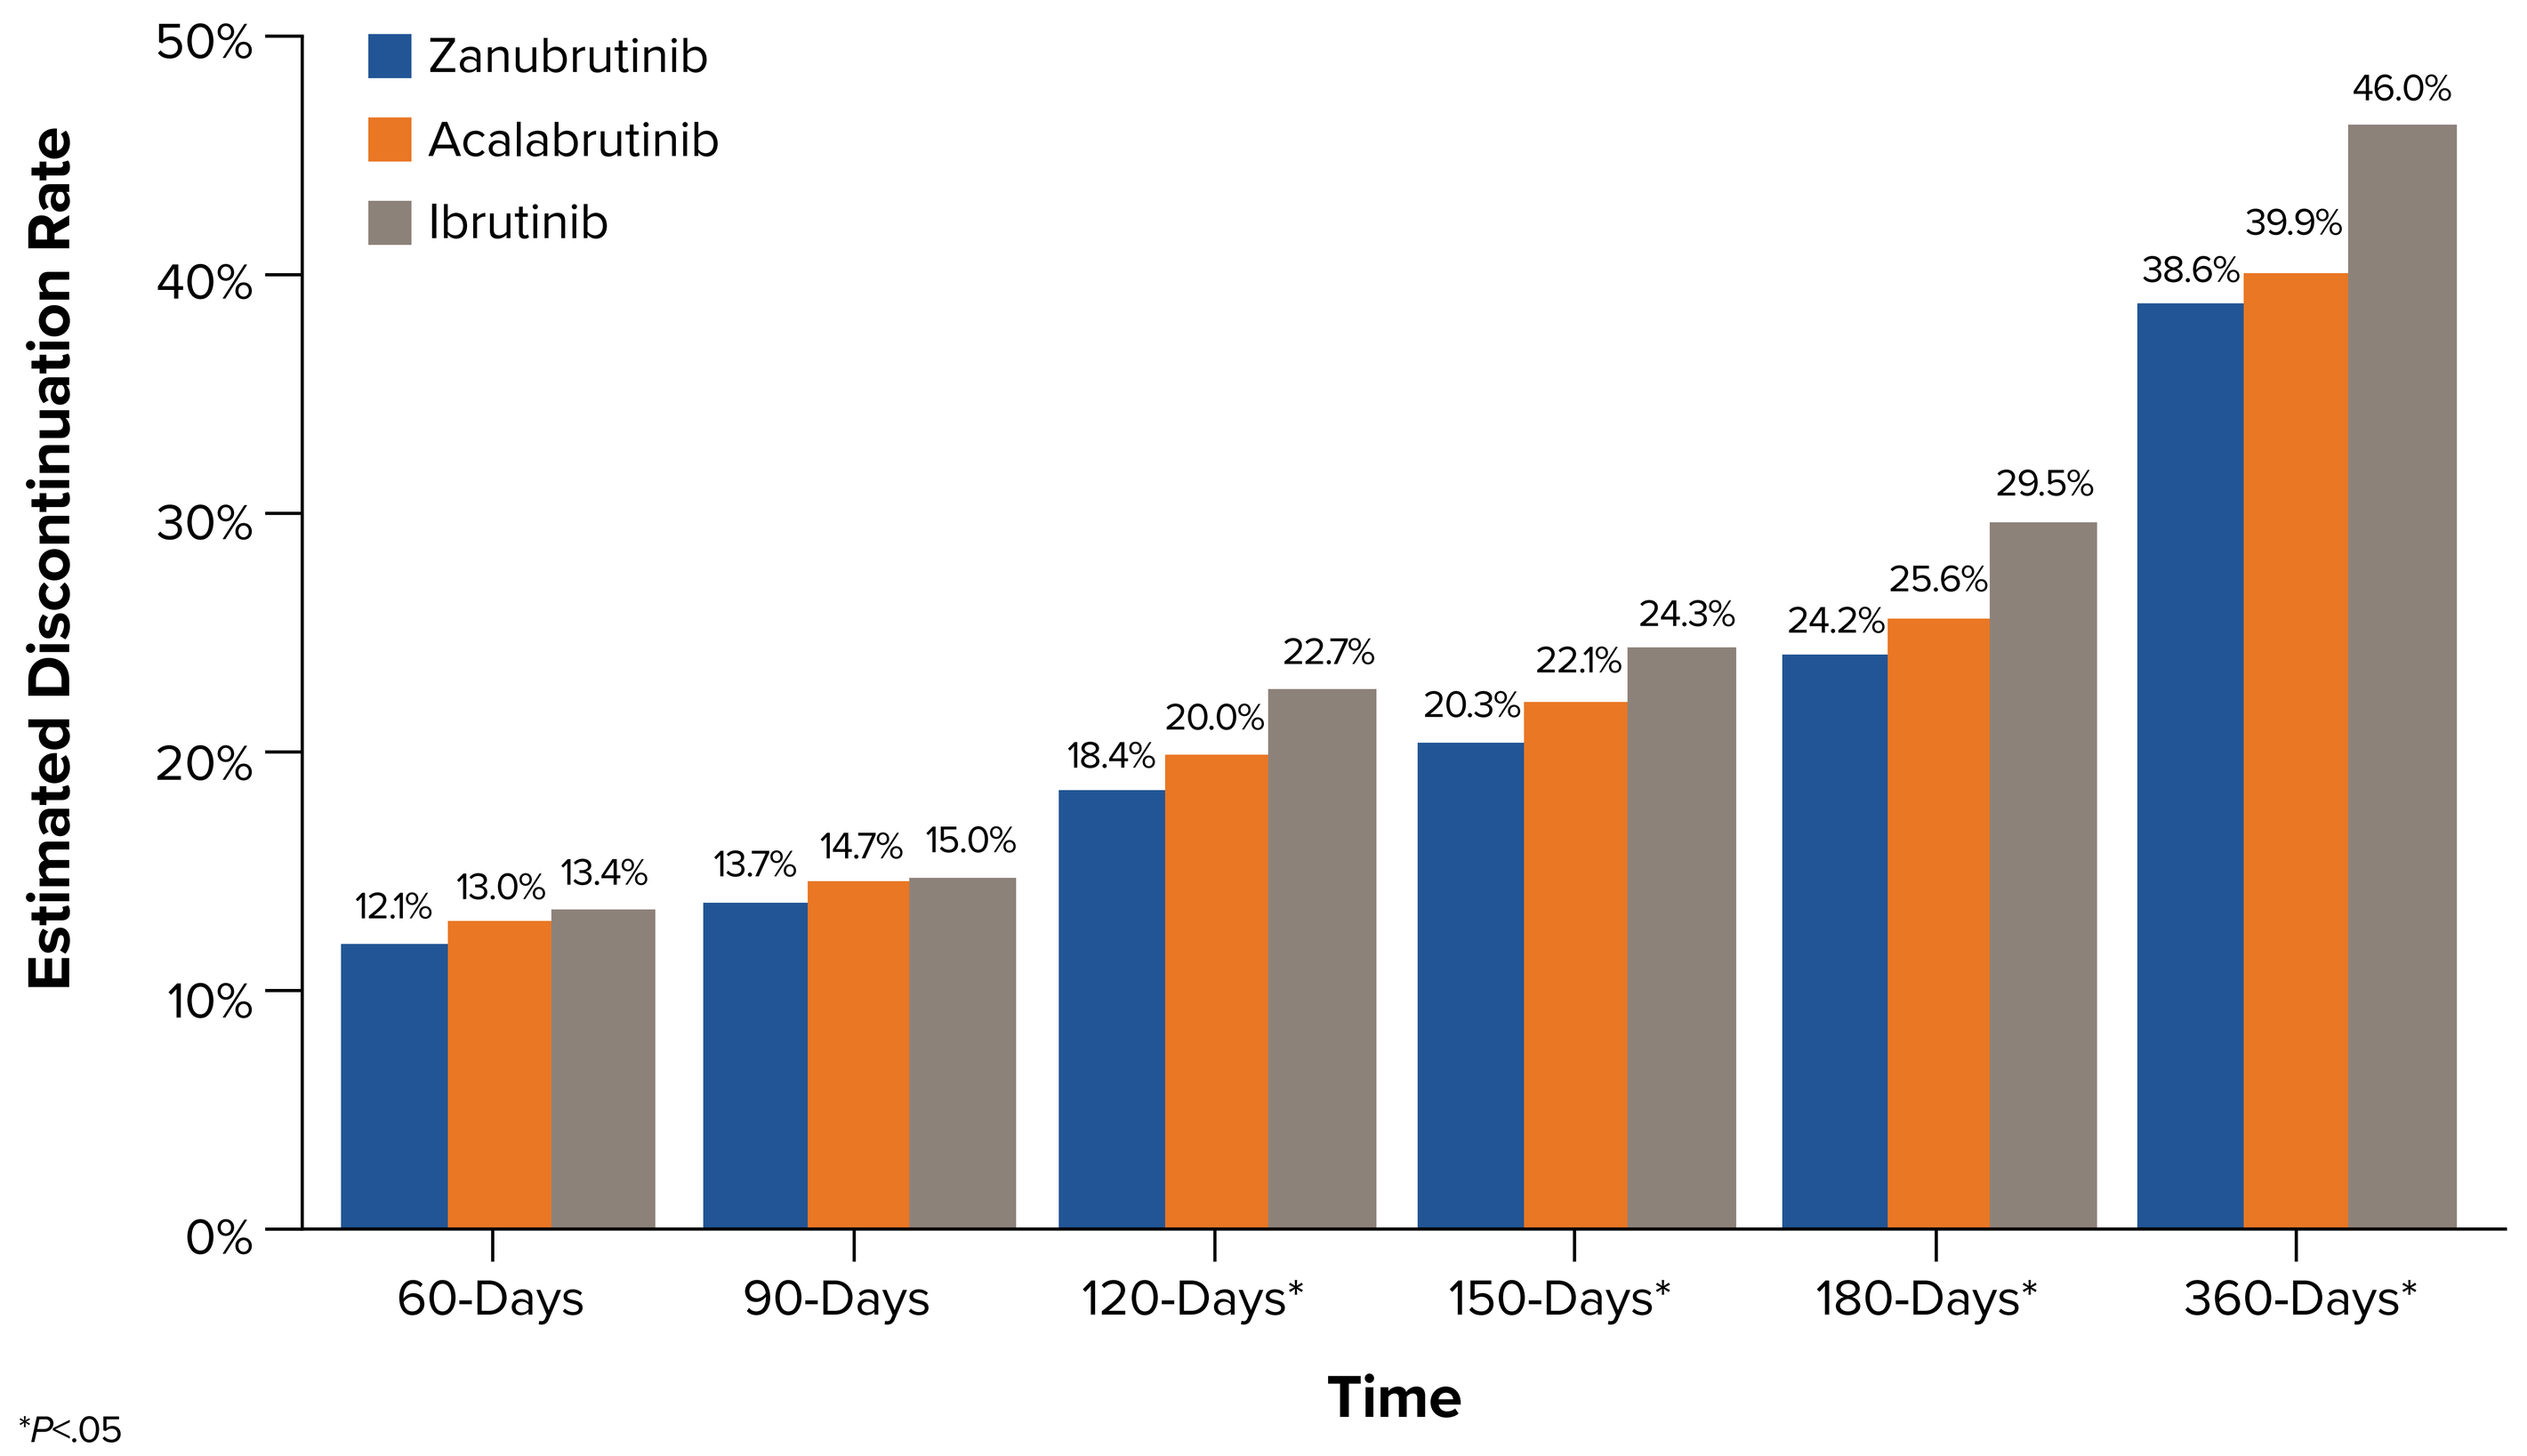

Estimated Discontinuation Rate
Zanubrutinib
Acalabrutinib
Ibrutinib
50%
40%
30%
20%
10%
0%
12.1% 13.0% 13.4%
13.7% 14.7% 15.0%
18.4% 20.0% 22.7%
20.3% 22.1% 24.3%
24.2% 25.6% 29.5%
38.6% 39.9% 46.0%
60-Days
90-Days
120-Days*
150-Days*
180-Days*
360-Days*
Time

B. ≥65 Years Group

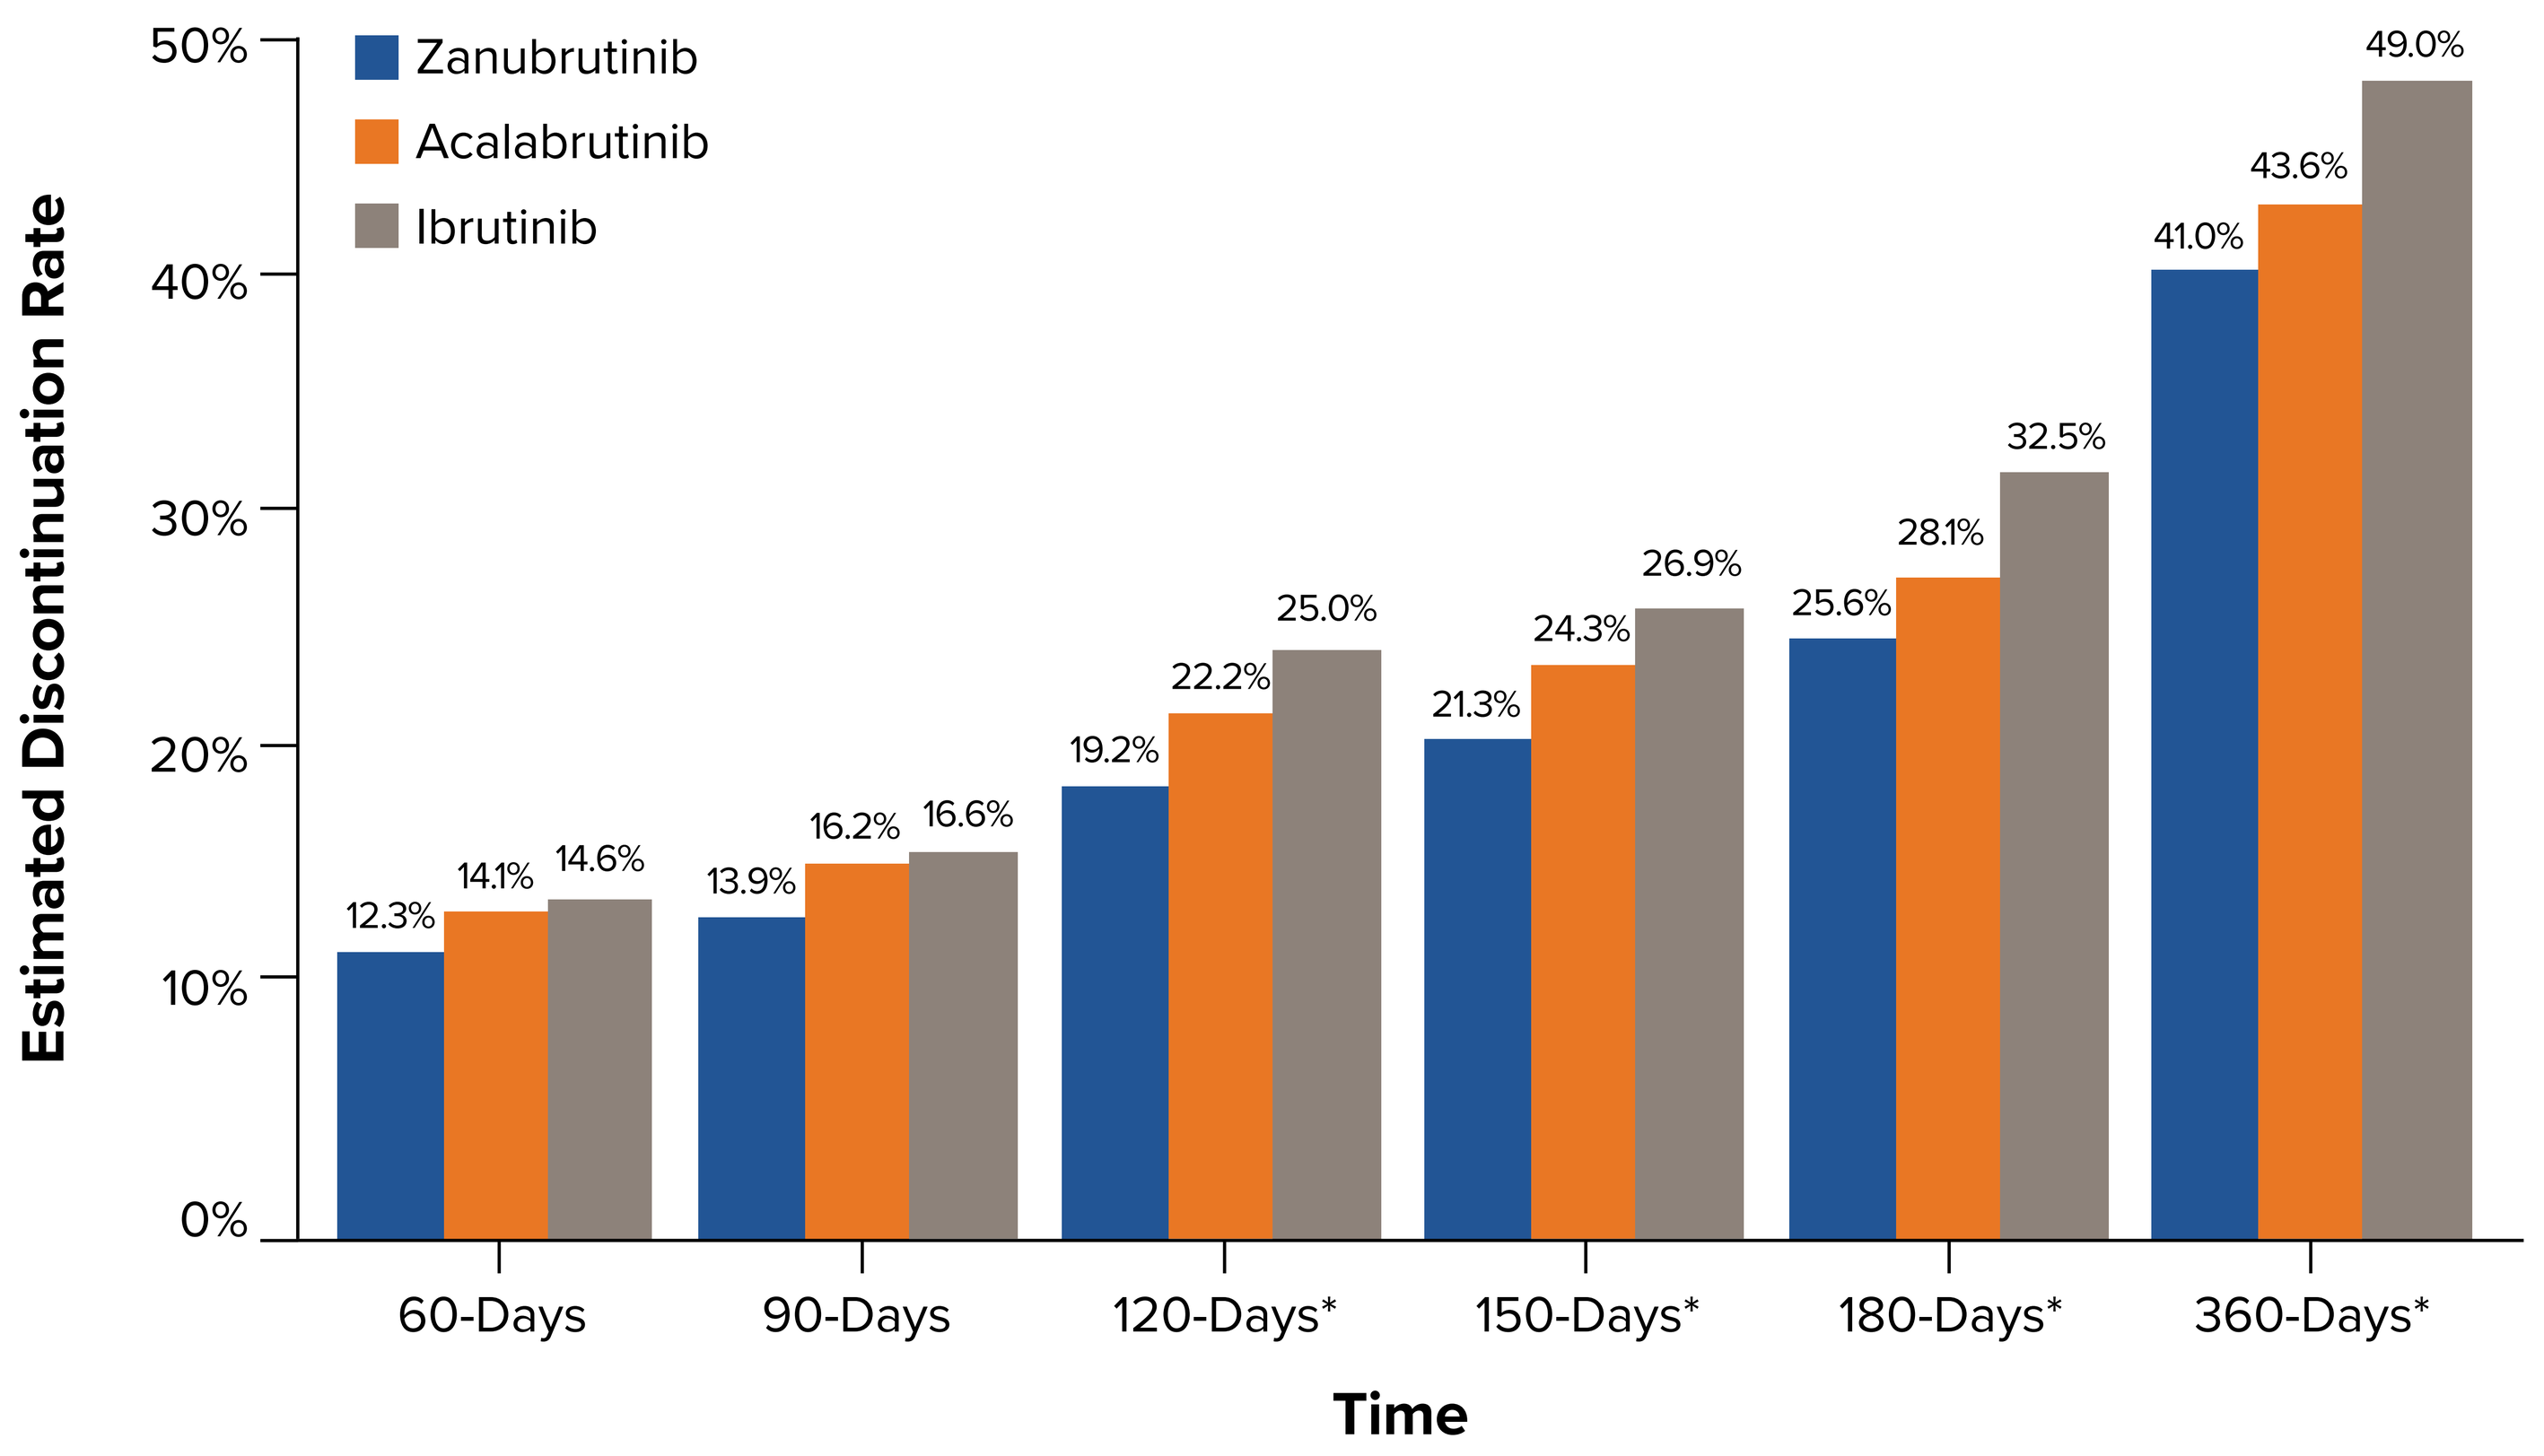

Estimated Discontinuation Rate
Zanubrutinib
Acalabrutinib
Ibrutinib
50%
40%
30%
20%
10%
0%
12.3% 14.1% 14.6%
13.9% 16.2% 16.6%
19.2% 22.2% 25.0%
21.3% 24.3% 26.9%
25.6% 28.1% 32.5%
41.0% 43.6% 49.0%
60-Days
90-Days
120-Days*
150-Days*
180-Days*
360-Days*
Time

*P<.05
CLL, chronic lymphocytic leukemia.

Healthcare Resource Utilization (HCRU)

- There were significant differences across the BTKi groups for inpatient visits PPPY in the overall group of patients with CLL and patients ≥65 years, with zanubrutinib having the lowest inpatient visits of the BTKis (zanubrutinib: 2.6, acalabrutinib: 3.6, ibrutinib: 4.6; P=.029 and zanubrutinib: 2.5, acalabrutinib: 4.3, ibrutinib: 5.3; P =.004, respectively) (**Figure 3**)

Figure 3. HCRU

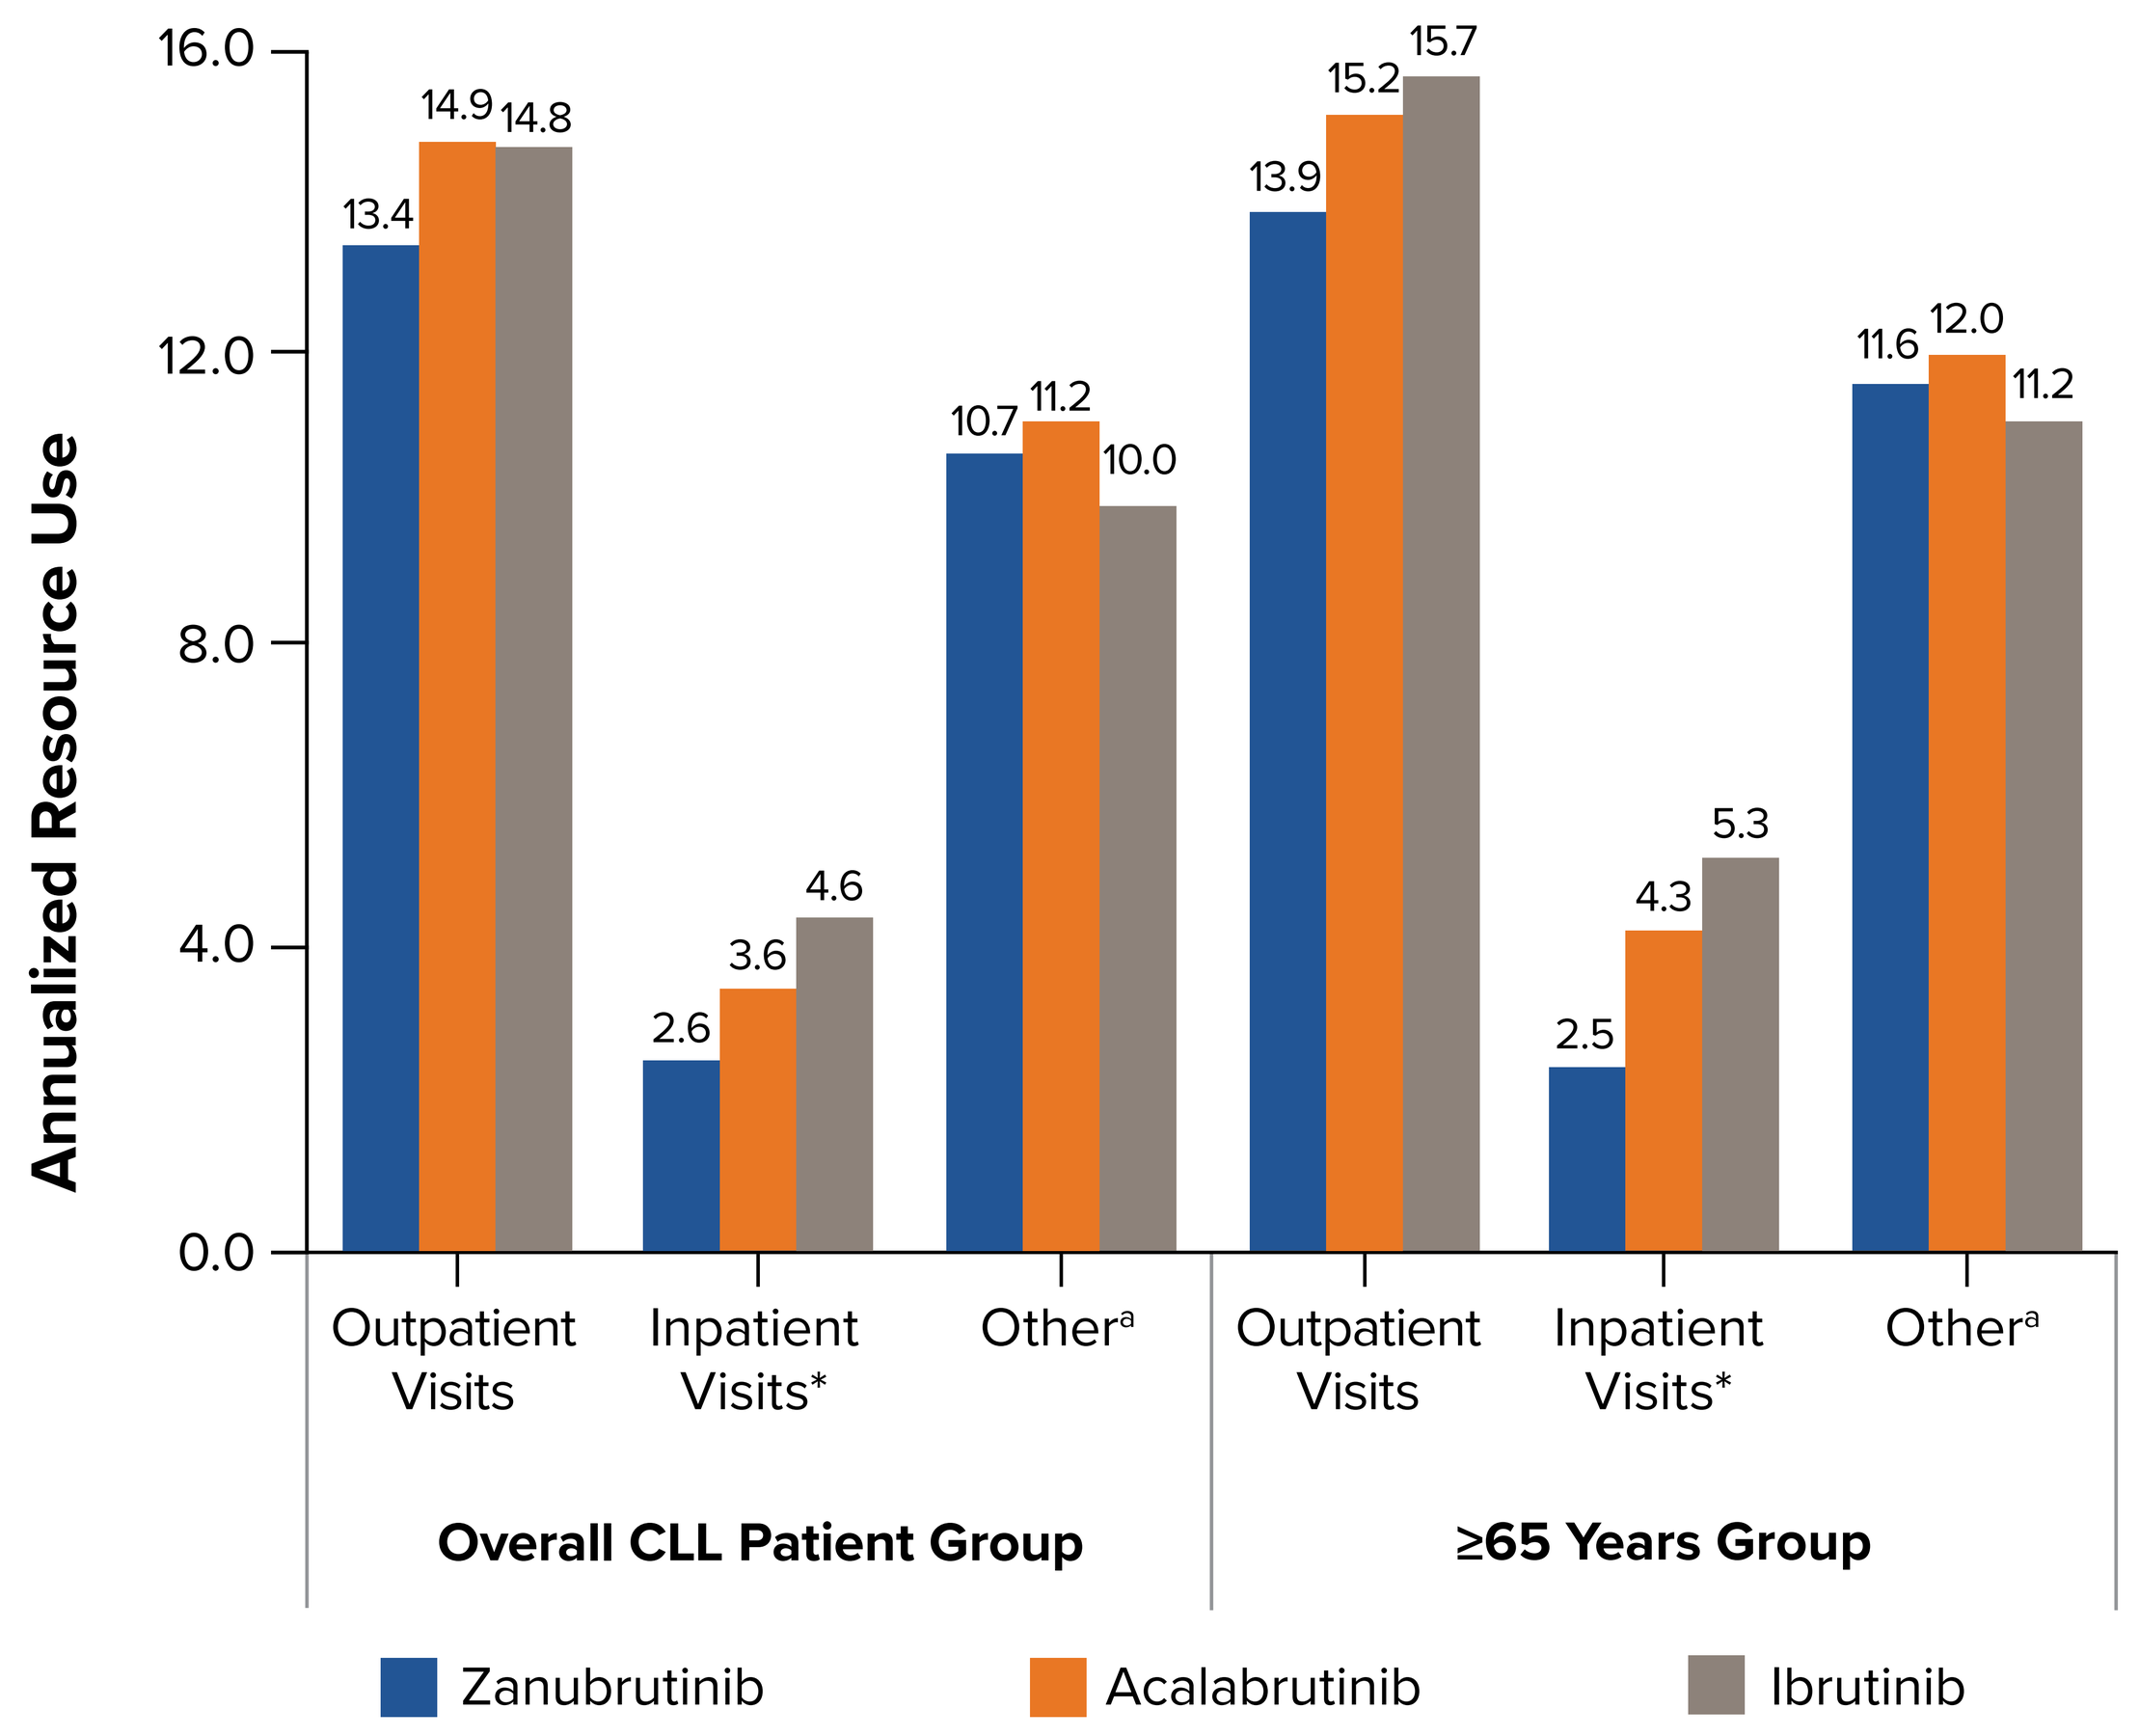

Annualized Resource Use
16.0
12.0
8.0
4.0
0.0
13.4 14.9 14.8
2.6 3.6 4.6
10.7 11.2 10.0
13.9 15.2 15.7
2.5 4.3 5.3
11.6 12.0 11.2
Outpatient Visits
Inpatient Visits*
Other[a]
Outpatient Visits
Inpatient Visits*
Other[a]
Overall CLL Patient Group
≥65 Years Group
Zanubrutinib
Acalabrutinib
Ibrutinib

*P<.05
[a]Includes Other Medical/Hospital Services
CLL, chronic lymphocytic leukemia; HCRU, healthcare resource utilization.

DISCUSSION

- This study addresses a critical gap in real-world evidence by focusing on older adults with CLL and providing valuable insights into age-related disparities in BTKi treatment outcomes
- The real-world data underscore the importance of treatment selection in mitigating age-related disparities and improving outcomes in a vulnerable patient population
- While this study included a diverse cross-section of patients with CLL in the United States, study limitations were inherent to the use of open claims databases in an observational study design

DISCLOSURES

KY: Employment and equity holder in BeOne. **SC**: Employment and equity holder in BeOne. **PC**: Employment in Real Chemistry. **WF**: Employment and equity holder in Real Chemistry. **SA**: Honoraria from Cellectar; consulting or advising roles for Amgen, BeOne, Bristol Myers Squibb, Cellectar, GSK, Johnson & Johnson, Regeneron, Pfizer, Sanofi, and Takeda; and research funding from AbbVie, Amgen, Ascentage, Bristol Myers Squibb, Cellectar, Genentech, GSK, Janssen, Johnson & Johnson, Pharmacyclics, Sanofi, and Xencor

REFERENCES

1. United Nations. Health Inequalities in Old Age. 2018.

ACKNOWLEDGEMENTS

This study was funded by BeOne Medicines Ltd.

Presented at EHA 2025; June 12-15, 2025; Milan, Italy