A Meta-Analysis Investigating Response Rates With Continuous Bruton Tyrosine Kinase Inhibitor Monotherapies in the Treatment of B-Cell Lymphomas

258

Pier Luigi Zinzani,[1] Rhys Williams,[2] Mei Xue,[2] Binod Neupane,[3] Priccila Zuchinali,[3] Heather Burnett,[4] Leyla Mohseninejad,[5] Keri Yang[2]

[1]Institute of Hematology "Seràgnoli", University of Bologna, Bologna, Italy; [2] BeOne Medicines Ltd, San Carlos, CA, USA; [3]Evidera, Montreal, QC, Canada; [4]Heather Burnett HEOR, Toronto, Canada; [5]BeOne Medicines Ltd, Schiphol, the Netherlands

CONCLUSIONS

- Zanubrutinib demonstrated significantly higher complete response (CR) and overall response rates (ORRs) compared with acalabrutinib and ibrutinib across B-cell lymphoma (BCL) indications
- Within each BCL indication, zanubrutinib demonstrated either similar, numerically higher, or statistically significantly higher response rates
- These findings suggest that zanubrutinib may offer a more effective treatment option for patients across BCL indications

INTRODUCTION

- Bruton tyrosine kinase (BTK) inhibitor (zanubrutinib, acalabrutinib, and ibrutinib) monotherapy has led to improved outcomes in patients with B-cell lymphomas (BCLs), including chronic lymphocytic leukemia (CLL), Waldenström macroglobulinemia, marginal zone lymphoma (MZL), mantle cell lymphoma (MCL), and Richter transformation[1]
- While the efficacy of each BTK inhibitor is understood within each individial BCL indication, this meta-analysis aims to compare response rates associated with BTK inhibitor monotherapy across BCL indications at the treatment naive (TN) and/or relapsed/refractory (R/R) stage

METHODS

- A systematic literature review was performed to identify clinical trials reporting CR rates or ORRs in patients with at least one type of BCL treated with zanubrutinib, acalabrutinib, or ibrutinib monotherapies
- Response rates, at similar follow-up time points (with a maximum difference of 12 months) and longest available follow-up time points, were extracted from each study and pooled across all applicable studies reporting data for zanubrutinib, acalabrutinib, or ibrutinib; investigator-assessed (INV) response rates were prioritized when available

Statistical Methods

- Results were presented as odds ratios with corresponding 95% confidence intervals; odds ratios >1 favor zanubrutinib over comparator BTK inhibitors
- Odds ratios were estimated for zanubrutinib vs each comparator BTK inhibitor (acalabrutinib or ibrutinib) for each response outcome in each BCL indication
- The odds ratios (OR) were then meta-analyzed across BCL indications using a random-effects model to account for variability between studies
- Between-study heterogeneity was assessed using I^2, the *P* value from the Q test, and τ (the standard deviation of underlying effects across studies)

RESULTS

- In total, 22 trials assessing 3599 patients were included for analysis across 4 BCL indications (**Table 1**)[3-27]
 - Fifteen trials (17 treatment arms) assessed patients with relapsed/refractory (R/R) disease or those who had been previously treated,[3-16, 26] while 4 trials (5 treatment arms) assessed treatment-naive (TN) patients,[17-23] and 3 trials (4 treatment arms) included a mixed population[24,25,27]
 - Sixteen trials reported INV response outcomes and 3 only reported IRC outcomes for the matching follow-up periods

Table 1. Characteristics of Trials Included in the Analysis by BCL Indication

Trials, n	22
Patients, n	3599
Treatment arms, n (%)	26
Zanubrutinib monotherapy	8 (31)
Ibrutinib monotherapy	11 (42)
Acalabrutinib monotherapy	7 (27)
Treatment status, n (%)	
Treatment naive	5 (19)
Relapsed/refractory	17 (65)
Mixed	4 (16)
Tumor type, n (%)	
CLL/SLL	11 (42)
MCL	6 (23)
MZL	4 (16)
WM	5 (19)

- The meta-analysis showed that zanubrutinib was associated with statistically significant improvements in both CR and ORR compared with acalabrutinib and ibrutinib across different BCL indications
- **Complete response rates** (**Figure 1**):
 - The pooled estimates of the to be consistent should be OR (95% CI) CR rates were 1.80 (1.03-3.13) for zanubrutinib vs acalabrutinib and 2.85 (1.16-7.04) for zanubrutinib vs ibrutinib
 - In R/R MCL, zanubrutinib demonstrated statistically superior efficacy over both acalabrutinib and ibrutinib for CR, with OR (95% CI) of 3.33 (1.91-5.81) and 9.53 (5.45-16.66), respectively; in R/R MZL, zanubrutinib showed superior efficacy over ibrutinib for CR, with an OR (95% CI) of 3.32 (1.28-8.61)
- **Overall response rates** (**Figure 2**):
 - The pooled estimates of the OR (95% CI) for ORR were 1.59 (1.0003-2.53) for zanubrutinib vs acalabrutinib and 2.25 (1.40-3.61) for zanubrutinib vs ibrutinib
 - In R/R MCL and R/R MZL, zanubrutinib showed superior ORR over ibrutinib, with OR (95% CI) of 2.23 (1.21, 4.12) and 2.39 (1.18, 4.85), respectively

Statistical Heterogeneity

- Moderate to high statistical heterogeneity was observed in the meta-analyses
- The I^2 values, which represent the percentage of variance due to heterogeneity, were:
 - CR rates: 68.5% for zanubrutinib vs acalabrutinib and 85.6% for zanubrutinib vs ibrutinib
 - ORR: 51.5% for zanubrutinib vs acalabrutinib and 49.2% for zanubrutinib vs ibrutinib

Figure 1. Odds Ratios for Complete Response Comparisons Across BCL Indications

Zanubrutinib vs acalabrutinib

Indication	N	zanu CR (95% CI)	aca CR (95% CI)	Odds ratio (95% CI)
R/R CLL	3	10.70 (7.78, 14.54)	8.25 (5.04, 13.23)	1.33 (0.71, 2.51)
R/R MCL	3	68.60 (58.08, 77.50)	39.62 (32.29, 47.44)	3.33 (1.91, 5.81)
R/R MZL	2	25.80 (16.80, 37.45)	12.50 (5.30, 26.72)	2.43 (0.82, 7.19)
TN CLL	3	20.70 (16.05, 26.28)	19.00 (13.90, 25.42)	1.11 (0.68, 1.81)
Pooled odds ratio				**1.80 (1.03, 3.13)**

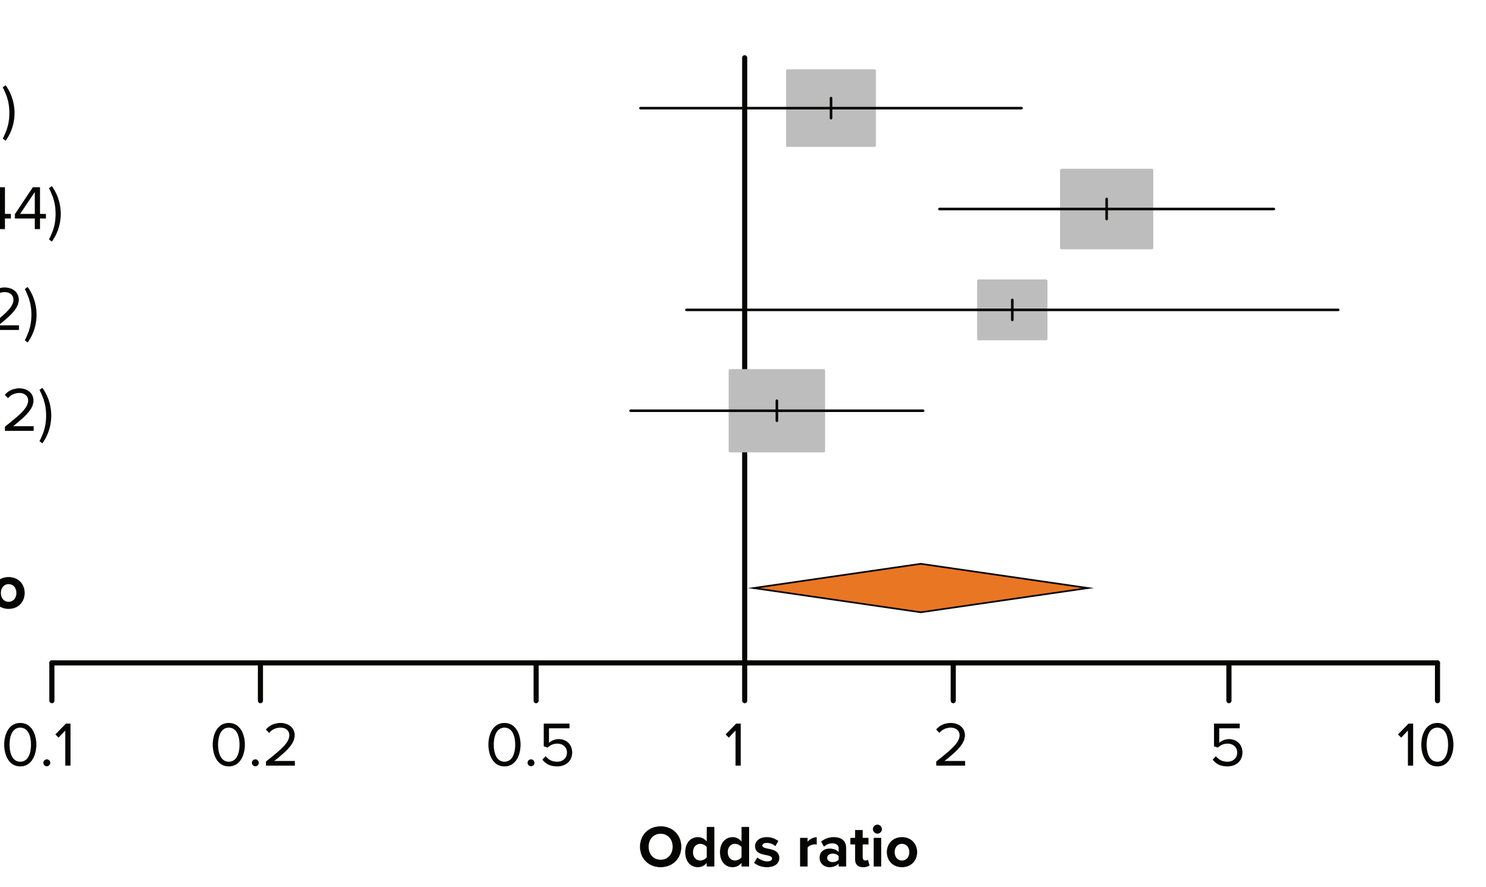
Complete response rate
0.1
0.2
0.5
1
2
5
10
Odds ratio

Zanubrutinib vs ibrutinib

Indication	N	zanu CR (95% CI)	ibr CR (95% CI)	Odds ratio (95% CI)
R/R CLL	4	10.70 (7.78, 14.54)	7.86 (5.83, 10.52)	1.40 (0.87, 2.26)
R/R MCL	2	77.90 (67.93, 85.44)	27.00 (22.72, 31.75)	9.53 (5.45, 16.66)
R/R MZL	3	26.95 (18.60, 37.33)	10.00 (4.65, 20.20)	3.32 (1.28, 8.61)
TN CLL	2	16.54 (13.00, 20.81)	13.54 (3.70, 38.95)	1.27 (0.30, 5.30)
WM	3	0.99 (0.20, 4.70)	0.50 (0.03, 7.48)	1.98 (0.08, 48.95)
Pooled odds ratio				**2.85 (1.16, 7.04)**

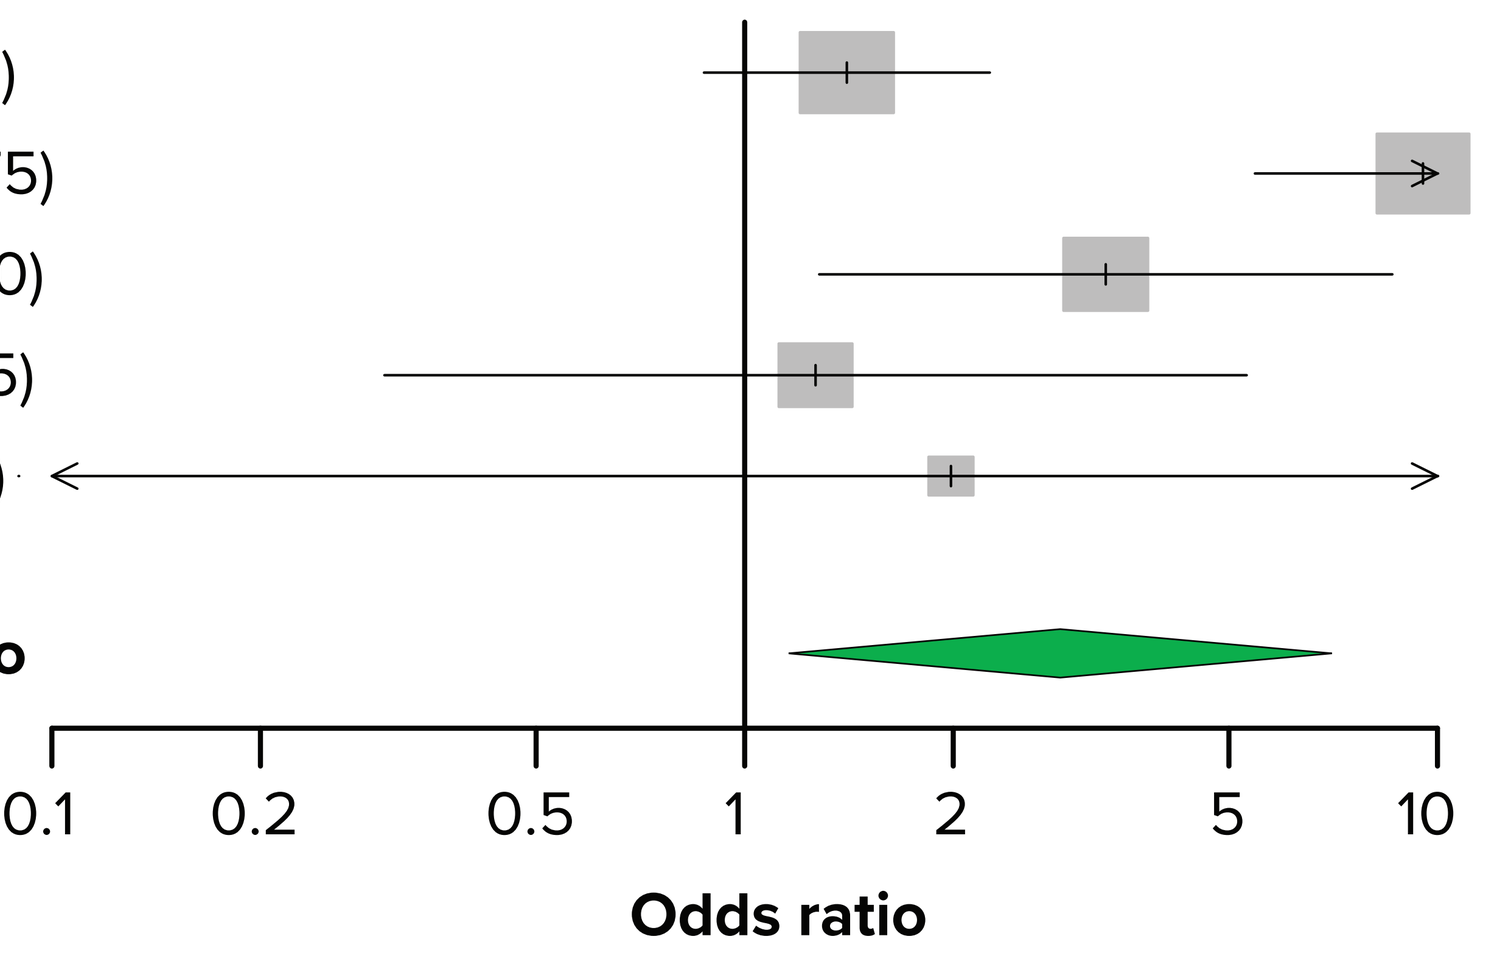
Complete response rate
0.1
0.2
0.5
1
2
5
10
Odds ratio

Abbreviations: aca, acalabrutinib; BCL, B-cell lymphoma; CI, confidence interval; CLL, chronic lymphocytic leukemia; CR, complete response; ibr, ibrutinib; MCL, mantle cell lymphoma; MZL, marginal zone lymphoma; N, number of trials or cohorts pooled; OR, odds ratio; R/R, relapsed/refractory; RE, random effects; TN, treatment naïve; WM: Waldenström macroglobulinemia; zanu, zanubrutinib.

Figure 2. Odds Ratios for Overall Response Comparisons Across BCL Indications

Zanubrutinib vs acalabrutinib

Indication	N	zanu ORR (95% CI)	aca ORR (95% CI)	Odds ratio (95% CI)
R/R CLL	2	85.60 (81.36, 89.00)	85.20 (81.45, 88.30)	1.03 (0.69, 1.56)
R/R MCL	3	84.00 (74.68, 90.33)	79.24 (72.17, 84.89)	1.38 (0.69, 2.75)
R/R MZL	2	68.20 (56.28, 78.13)	52.50 (37.27, 67.28)	1.94 (0.87, 4.33)
TN CLL	3	97.50 (94.56, 98.87)	90.00 (84.67, 93.62)	4.33 (1.68, 11.15)
WM	3	94.71 (90.18, 97.21)	93.00 (86.30, 96.55)	1.35 (0.50, 3.67)
Pooled odds ratio				**1.59 (1.0003, 2.53)**

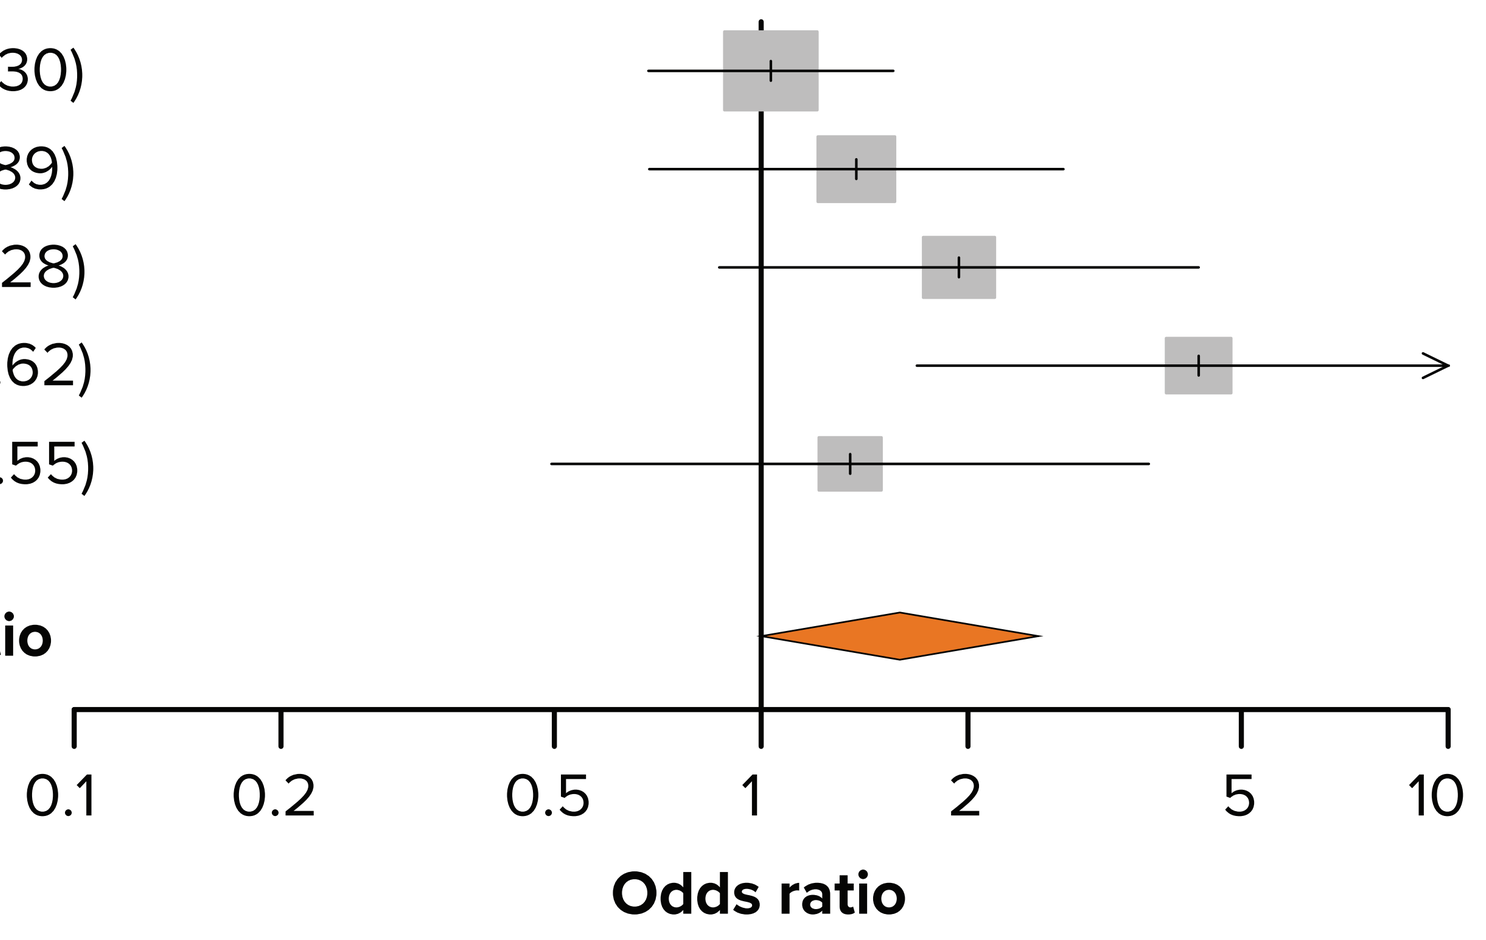
Overall response rate
0.1
0.2
0.5
1
2
5
10
Odds ratio

Zanubrutinib vs ibrutinib

Indication	N	zanu ORR (95% CI)	ibr ORR (95% CI)	Odds ratio (95% CI)
R/R CLL	4	85.60 (81.36, 89.00)	82.77 (70.44, 90.64)	1.24 (0.58, 2.66)
R/R MCL	2	83.70 (74.34, 90.10)	69.70 (64.82, 74.17)	2.23 (1.21, 4.12)
R/R MZL	3	76.73 (66.64, 84.47)	58.00 (45.57, 69.49)	2.39 (1.18, 4.85)
TN CLL	2	97.11 (94.71, 98.44)	86.00 (79.10, 90.88)	5.47 (2.47, 12.12)
WM	4	94.71 (90.18, 97.21)	92.19 (86.11, 95.75)	1.52 (0.60, 3.83)
Pooled odds ratio				**2.25 (1.40, 3.61)**

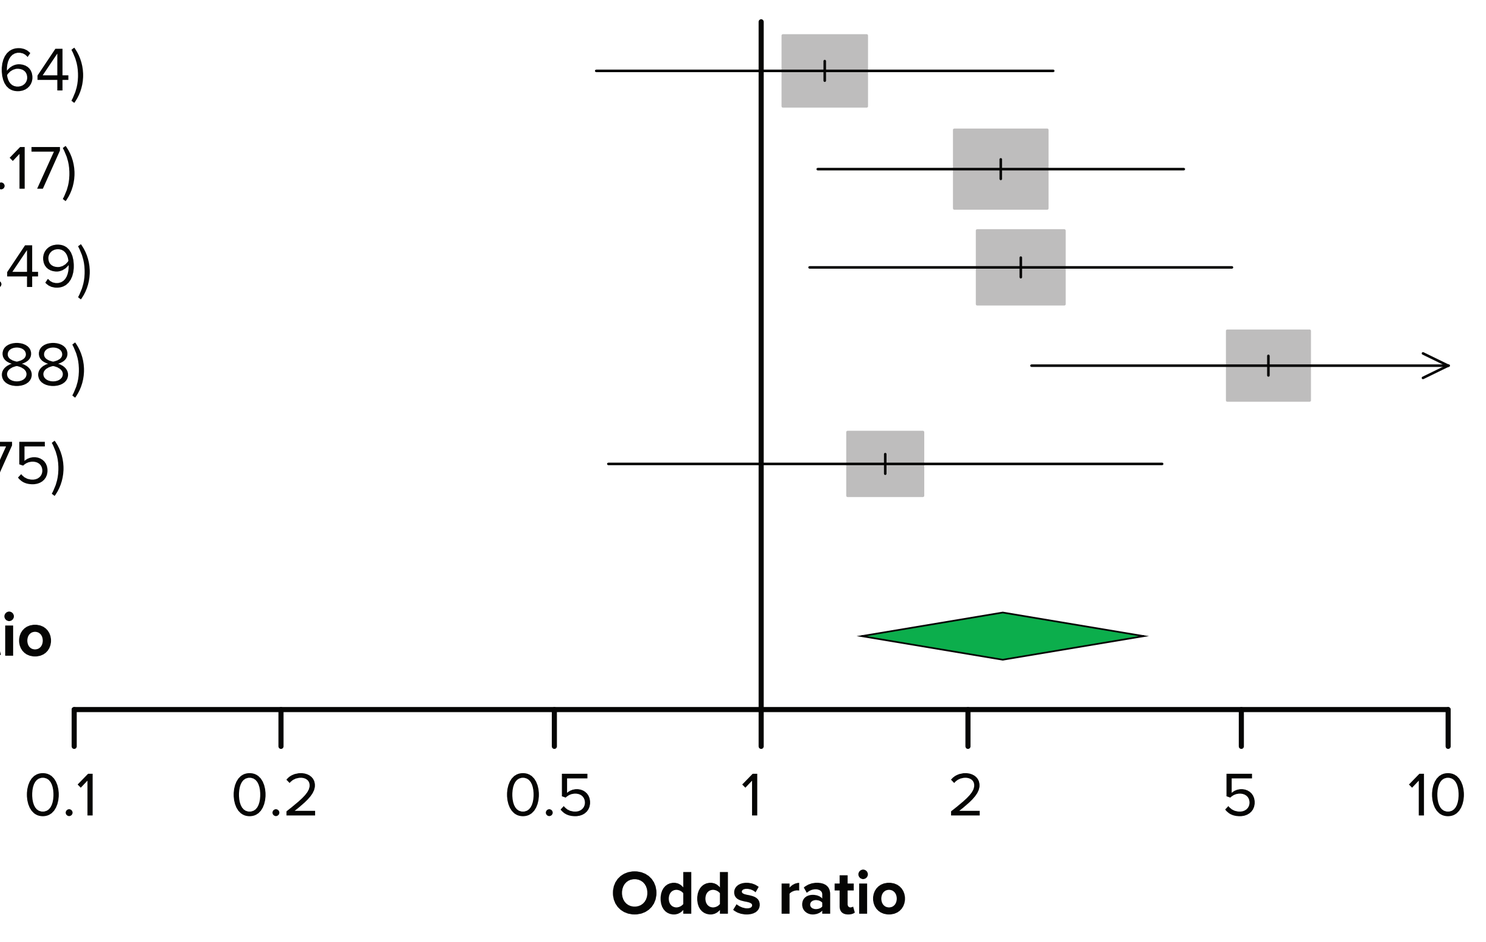
Overall response rate
0.1
0.2
0.5
1
2
5
10
Odds ratio

Abbreviations: aca, acalabrutinib; BCL, B-cell lymphoma; CI, confidence interval; CLL, chronic lymphocytic leukemia; CR, complete response; ibr, ibrutinib; MCL, mantle cell lymphoma; MZL, marginal zone lymphoma; N, number of trials or cohorts pooled; OR, odds ratio; R/R, relapsed/refractory; RE, random effects; TN, treatment naïve; WM: Waldenström macroglobulinemia; zanu, zanubrutinib.

DISCUSSION

- This analysis indicates that zanubrutinib is associated with higher response rates across BCL indications compared with acalabrutinib and ibrutinib
- Within each indication, zanubrutinib consistently demonstrated either numerically higher or statistically superior response rates; these findings suggest that zanubrutinib offers a more effective treatment option for patients with BCL
- The observed heterogeneity is likely driven by differences in key study characteristics across trials, including patient populations, mutation status, indication, line of therapy, and follow-up duration
- This variability suggests that the relative efficacy of zanubrutinib compared with other BTK inhibitors may differ by indication; nonetheless, zanubrutinib generally demonstrated superior efficacy and was favored in the majority of indications assessed

REFERENCES

1. Fares A, et al. *Int J Mol Sci.* 2024;25(4):2208-2221.
2. Tam C, Thompson PA. *Blood Adv.* 2024;8(9):2300-2309.
3. Ferrant E, et al. SFH 2024.
4. Byrd JC, et al. *J Clin Oncol.* 2017;35(26):3048-3055.
5. Byrd JC, et al. *J Clin Oncol.* 2021;39(31):3441-3452.
6. Ghia P, et al. *Hemasphere.* 2022;6(12):e801.
7. Song Y, et al. *Clin Cancer Res.* 2020;26(16):4216-4224.
8. Song Y, et al. *Blood.* 2022;139(21):3148-3158.
9. Rule S, et al. *Haematologica.* 2019;104(5):e211-e214.
10. Wang M, et al. *Lancet.* 2018;391(10121):659-667.
11. Song Y, et al. *Leuk Lymphoma.* 2024;65(4):867-875.
12. Phillips T, et al. *Blood Adv.* 2022;6(11):3472-3479.
13. Opat S, et al. *Clin Cancer Res.* 2021;27(21):5753-5763.
14. Opat S, et al. *Blood.* 2022;139(22):3278-3289.
15. Noy A, et al. *Blood Adv.* 2020;4(22):5773-5784.
16. Strati P, et al. *Br J Haematol.* 2022;196(3):682-686.
17. Munir T, et al. *HemaSphere.* 2023;7(6):e915.
18. Shadman M, et al. *J Clin Oncol.* 2025;43(10):1123-1134.
19. Tam CS, et al. *Lancet Oncol.* 2022;23(8):1031-1043.
20. Woyach JA, et al. *N Engl J Med.* 2018;379(26):2517-2528.
21. Burger JA, et al. *N Engl J Med.* 2015;373(25):2425-2437.
22. Barr PM, et al. *Blood Adv.* 2022;6(11):3440-3450.
23. Sharman JP, et al. *Blood.* 2023;142(Suppl 1):636.
24. Trotman J, et al. *Blood.* 2020;136(18):2027-2037.
25. Tam CS, et al. *Blood.* 2020;136(18):2038-2050.
26. Dimopoulos MA, et al. *Lancet Oncol.* 2017;18(5):602-612.
27. Owen RG, et al. *Lancet Haematol.* 2020;7(2):e112-e121.

DISCLOSURES

PLZ: Honoraria and Speakers bureau: Kyowa Kirin, Roche, AbbVie, BeOne Medicines Ltd, BMS, Gilead, Novartis, Incyte, Sobi. **RW, MX, LM, KY:** Employment and own stock: BeOne Medicines Ltd. **PZ:** Employment: Evidera. **HB:** Employment and stock: AstraZeneca; Consulting or advisory role: Regeneron. **BN:** No disclosure.

ACKNOWLEDGMENTS

This study was sponsored by BeOne Medicines Ltd. Editorial assistance was provided by Adam Ruth, PhD, Nucleus Global, an Inizio company, and supported by BeOne Medicines.

CORRESPONDENCE: Pier Luigi Zinzani, pierluigi.zinzani@unibo.it

Presented at the International Conference on Malignant Lymphoma (ICML); June 17-21, 2025; Lugano, Switzerland